This is a reprint from European Society for Medical Oncology Congress 2025 (ESMO 2025), which was originally presented in Berlin, Germany on October 17-21, 2025; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

962P

Precemtabart tocentecan (M9140), an anti-CEACAM5 ADC with exatecan payload, in patients with metastatic colorectal cancer: updated clinical data from the Phase 1 PROCEADE-CRC-01 study

Scott Kopetz[1], Rocio Garcia-Carbonero[2], Sae-Won Han[3], Ildefonso Ismael Rodriguez-Rivera[4], Jose Carlos Ruffinelli Rodriguez[5], Valentina Boni[6], Mario Vieito Villar[7], Maria de Miguel[8], Athanasios Pallis[9], Christina Habermehl[10], Ken Kato[11]

[1]GI Medical Oncology Department, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Medical Oncology, Hospital Universitario 12 de Octubre, Imas12, Universidad Complutense de Madrid, Madrid, Spain; [3]Department of Internal Medicine, Seoul National University Hospital, Seoul, Republic of Korea; [4]Oncology, NEXT Oncology, San Antonio, USA; [5]Medical Oncology Department, ICO - Institut Català d'Oncologia l'Hospitalet (Hospital Duran i Reynals), L'Hospitalet De Llobregat, Spain; [6]Department of Oncology, Hospital Universitario Quirónsalud Madrid, Pozuelo De Alarcon, Spain; [7]Department of Medical Oncology, Vall d'Hebron University Hospital, Barcelona, Spain; [8]Centro Integral Oncologico Clara Campal, START Madrid-CIOCC, Madrid, Spain; [9]Global Clinical Development, Merck Santé S.A.S., Lyon, France, an affiliate of Merck KGaA, Darmstadt, Germany; [10]Global Biostatistics, Merck KGaA, Darmstadt, Germany; [11]Department of Gastrointestinal Medical Oncology, National Cancer Center - Tsukiji Campus, Chuo-ku, Japan

PROCEADE CRC-01 PHASE 1

SCOPE

PROCEADE-CRC-01 (NCT05464030; Phase 1) evaluates the clinical activity and safety of the anti-CEACAM5 TOP1i-payload ADC precemtabart tocentecan (Precem-TcT) in patients with heavily pretreated mCRC. We present OS alongside updated PFS, ORR, and safety data from the dose optimization (2.8 mg/kg Q3W or 2.4 mg/kg Q3W Precem-TcT)

CONCLUSIONS

- In this population of 60 patients with heavily pretreated mCRC (all had received prior irinotecan), Precem-TcT has demonstrated promising efficacy with a predictable and manageable safety profile
- The trial is ongoing. At data cutoff (02 Sep 2025) in the 2.8 mg/kg Q3W arm:
 - o After a median follow-up of 13.1 months:
 - – mOS was NR (95% CI: 8.7–NE)
 - – mPFS was 6.9 months (95% CI: 4.4–9.5); 6-month PFS rate was 64.3%
 - o ORR was 31.0% (95% CI: 15.3–50.8; confirmed: 20.7% [95% CI: 8.0–39.7])
 - o No ILDs – AEs commonly associated with ADCs – were observed; GI TRAEs were mild
 - o Grade ≥3 TRAEs were mainly hematologic
- Notably higher ORR at 2.8 mg/kg Q3W vs 2.4 mg/kg Q3W, and overall comparable safety and consistent PK profiles between the dose levels tested, support 2.8 mg/kg Q3W as the recommended dose for further development
- OS, PFS, and ORR with Precem-TcT benchmark favorably with monotherapy (mOS 6.4–7.8 months; mPFS 1.9–3.7 months; ORR 1–2%)[1-4]; or combination (SUNLIGHT: mOS 10.8 months; mPFS 5.6 months; 6-month PFS rate: 43%; ORR 6.1%)[5] SoCs approved for 3L+ mCRC
- Encouraging efficacy and predictable, manageable safety support further clinical development of Precem-TcT at 2.8 mg/kg Q3W in mCRC and other CEACAM5-expressing solid tumors (PROCEADE-PanTumor; NCT06710132)

GET POSTER PDF

Copies of this poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

SUPPLEMENTARY DATA

Please scan this QR code to view supplementary data associated with this poster.

INTRODUCTION

- CEACAM5 is overexpressed in ~90% of CRCs, with limited expression in healthy tissues[1-3]
- Precemtabart tocentecan (Precem-TcT) is the first anti-CEACAM5 ADC with a ß-glucuronide linker-payload combination in clinical development that selectively delivers exatecan, a potent TOP1i, to CEACAM5-overexpressing tumor cells (**Figure 1**)[4]
- In the dose escalation of the Phase 1 PROCEADE-CRC-01 study in patients with heavily pretreated mCRC (including irinotecan), Precem-TcT showed promising early efficacy and a predictable, manageable safety profile – without any cases of ILD or ocular toxicities[5,6]
- We present data from the dose optimization (**Figure 2**)

Figure 1. MoA of Precem-TcT

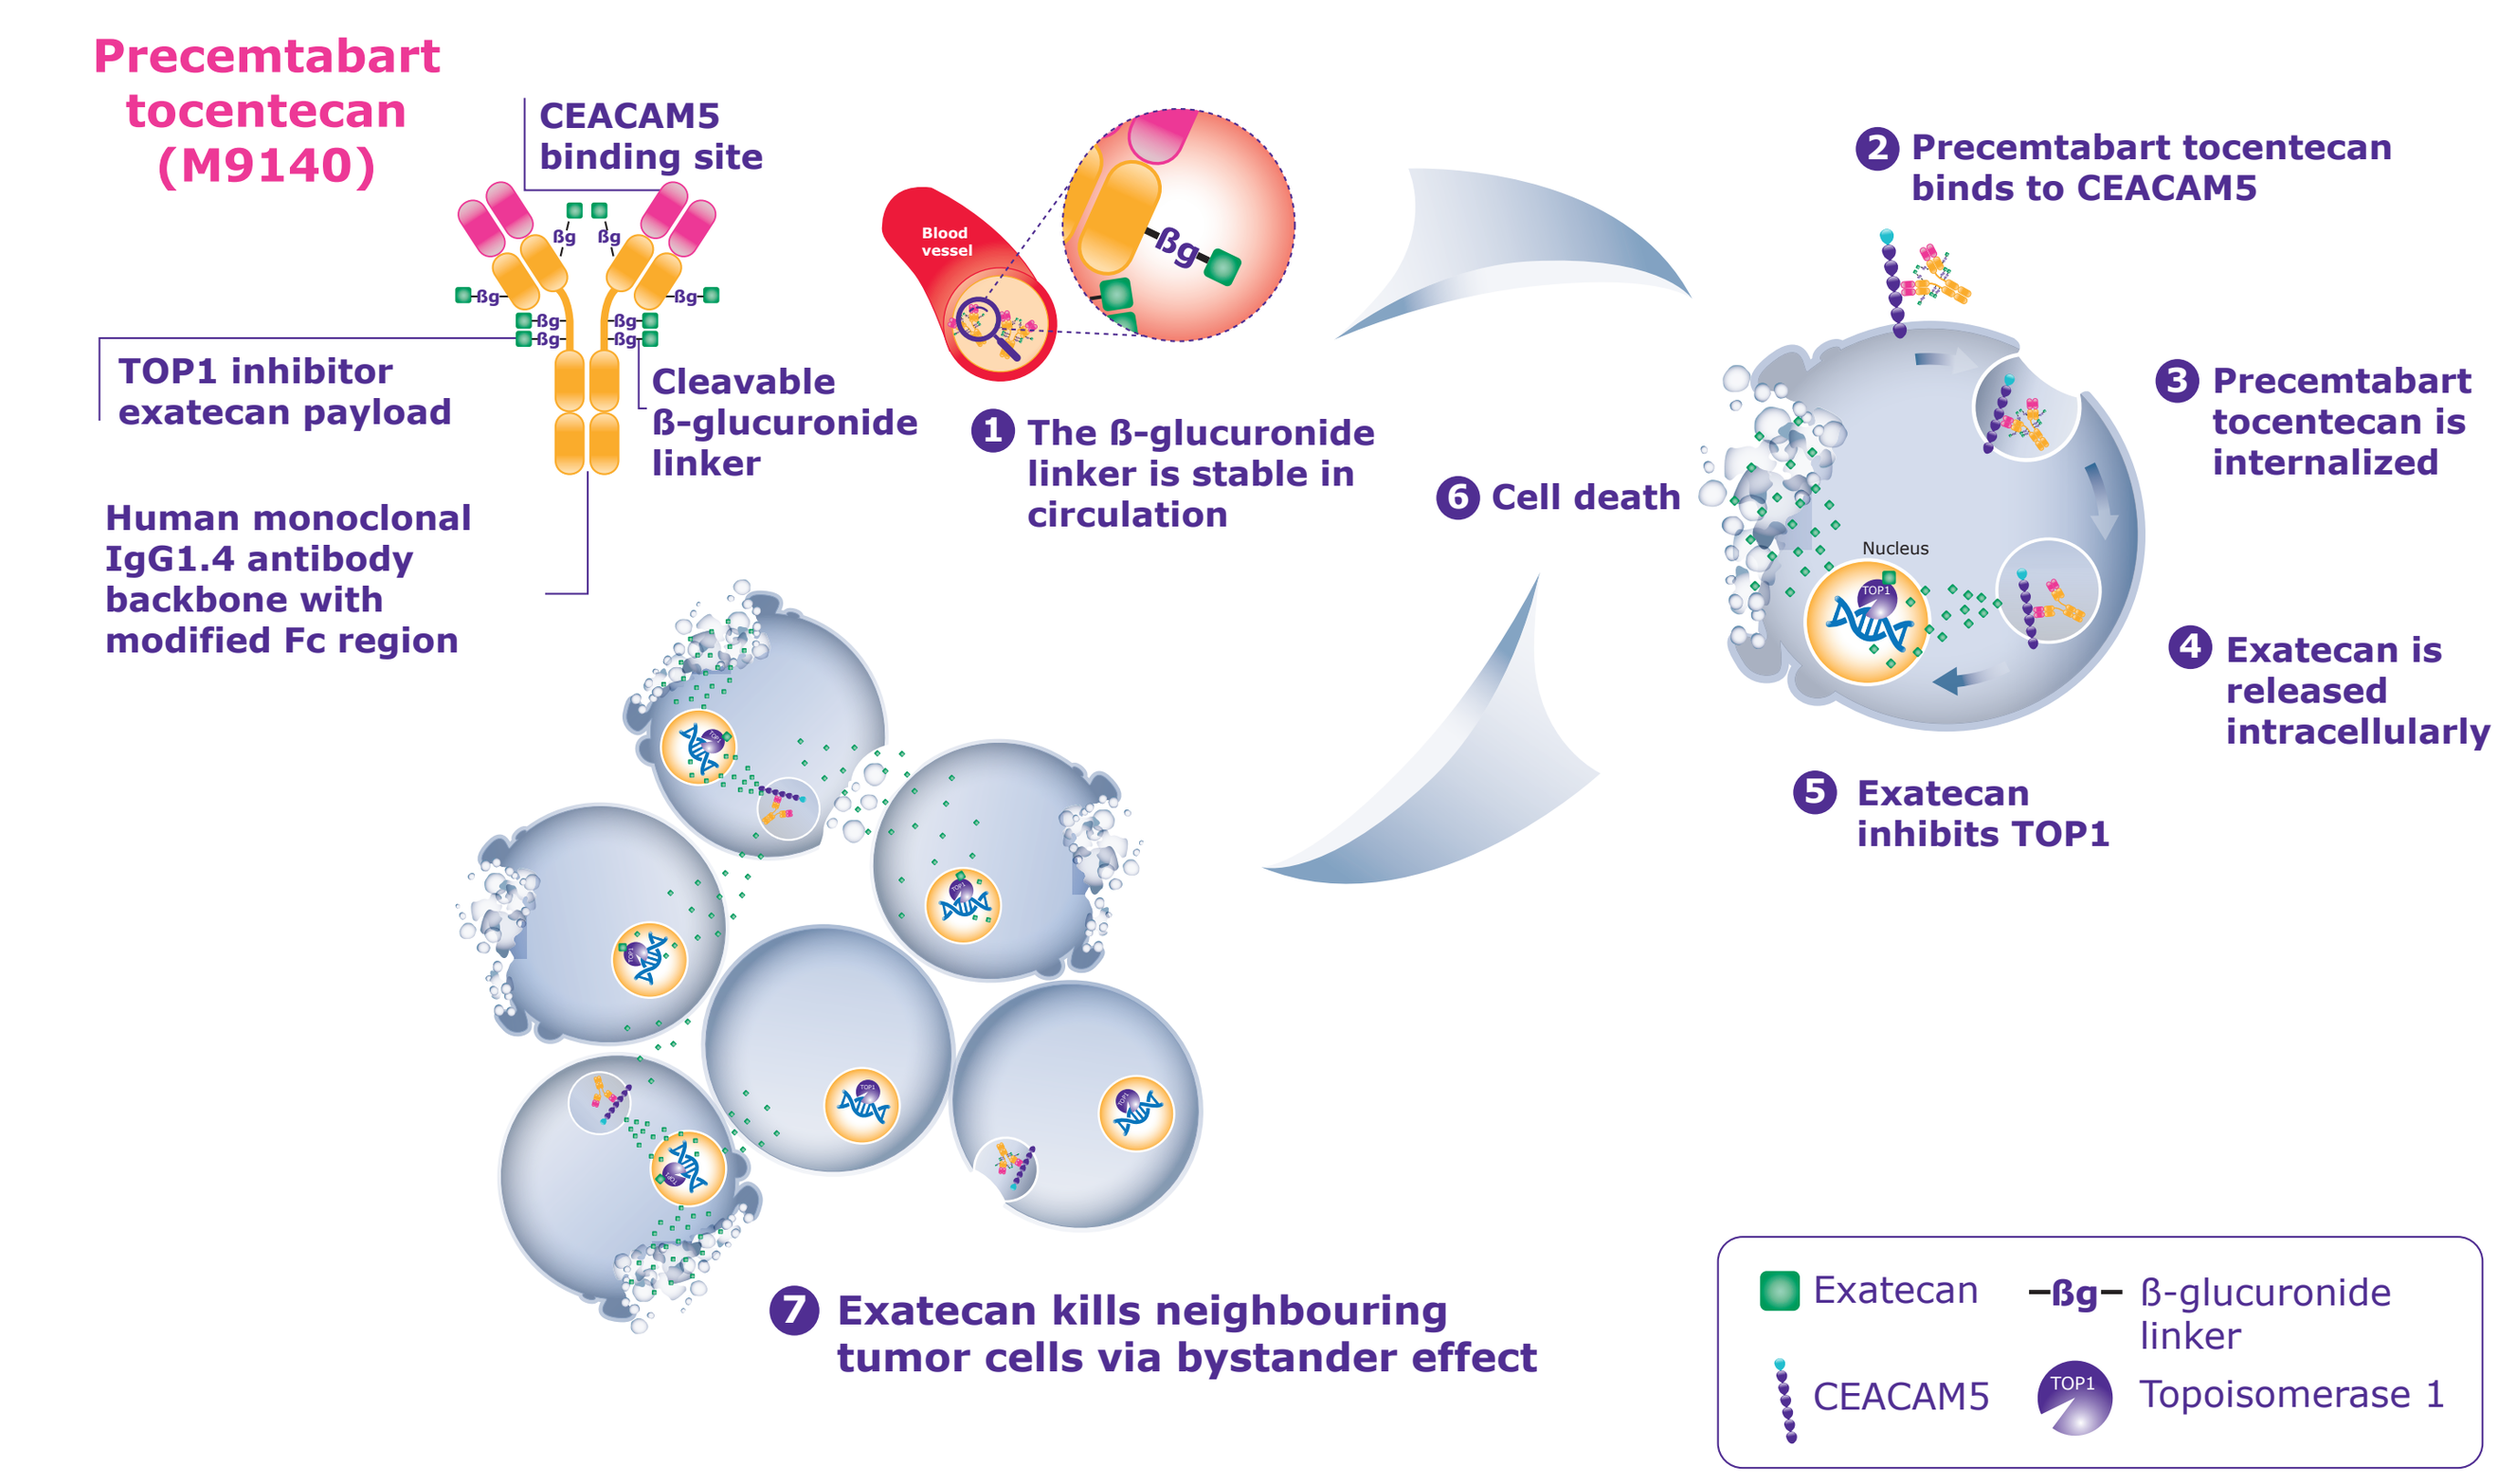
Precemtabart tocentecan (M9140)
CEACAM5 binding site
TOP1 inhibitor exatecan payload
Cleavable ß-glucuronide linker
Human monoclonal IgG1.4 antibody backbone with modified Fc region
1 The ß-glucuronide linker is stable in circulation
2 Precemtabart tocentecan binds to CEACAM5
3 Precemtabart tocentecan is internalized
4 Exatecan is released intracellularly
5 Exatecan inhibits TOP1
6 Cell death
7 Exatecan kills neighbouring tumor cells via bystander effect
Nucleus
Exatecan
ß-glucuronide linker
CEACAM5
Topoisomerase 1

METHODS

Figure 2. Study design of PROCEADE-CRC-01

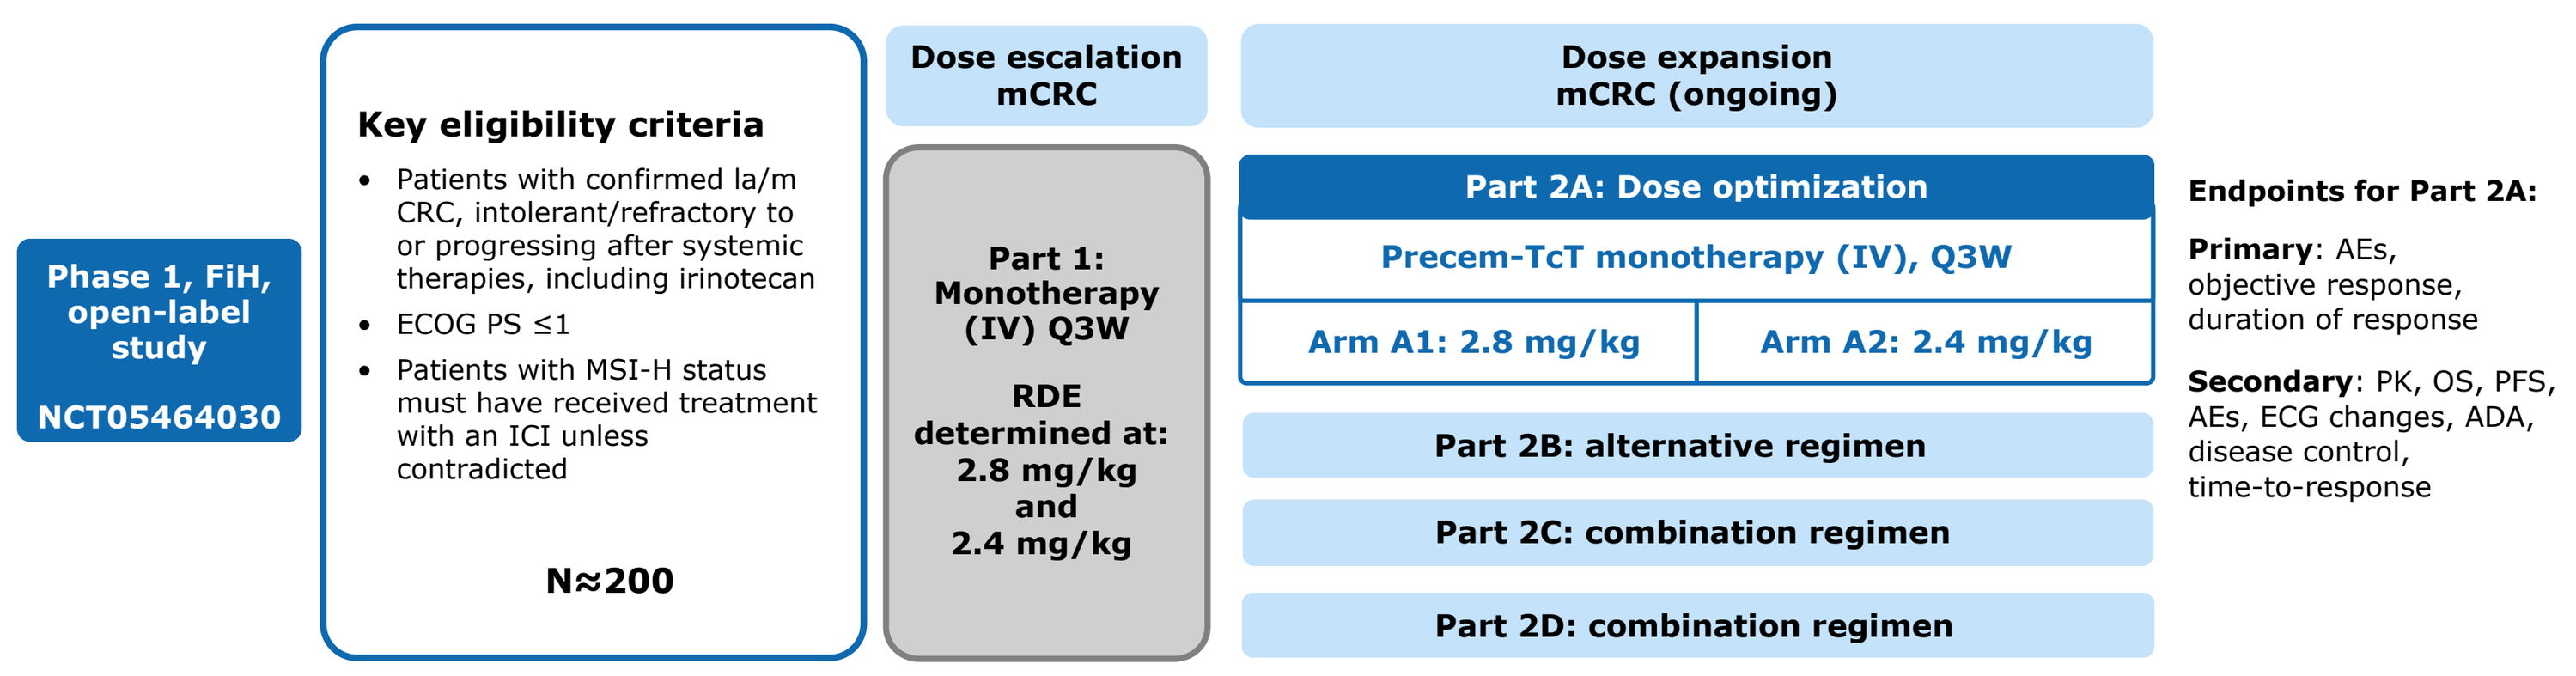
Phase 1, FiH, open-label study
NCT05464030
Key eligibility criteria
Patients with confirmed la/m CRC, intolerant/refractory to or progressing after systemic therapies, including irinotecan
ECOG PS ≤1
Patients with MSI-H status must have received treatment with an ICI unless contradicted
N≈200
Dose escalation mCRC
Part 1: Monotherapy (IV) Q3W
RDE determined at: 2.8 mg/kg and 2.4 mg/kg
Dose expansion mCRC (ongoing)
Part 2A: Dose optimization
Precem-TcT monotherapy (IV), Q3W
Arm A1: 2.8 mg/kg
Arm A2: 2.4 mg/kg
Part 2B: alternative regimen
Part 2C: combination regimen
Part 2D: combination regimen
Endpoints for Part 2A:
Primary: AEs, objective response, duration of response
Secondary: PK, OS, PFS, AEs, ECG changes, ADA, disease control, time-to-response

RESULTS

- All patients were irinotecan, oxaliplatin, 5-FU, and bevacizumab pretreated; median age was 60.0 years (range, 29–80); ~50% were female; and baseline characteristics were overall well-balanced between the two DLs (**Table S1**)
- As of 02 Sep 2025, 5 patients (8.3%) remained on treatment (**Table 1**)
- Mean duration of therapy was 6.5 ± 4.03 months, with 60% of patients having received >6 cycles of Precem-TcT therapy (**Table 1**)
 - o Eight patients have received treatment for >1 year, of whom 4 were still on treatment at data cutoff

Table 1. Treatment overview

	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Patients still in trial, n (%)	17 (58.6)	13 (41.9)	30 (50.0)
Treatment ongoing, n (%)	2 (6.9)	3 (9.7)	5 (8.3)
Treatment completed/ discontinued, n (%)	27 (93.1)	28 (90.3)	55 (91.7)
Withdrawal by patient[a]	1 (3.4)	0 (0.0)	1 (1.7)
Death	0 (0.0)	1 (3.2)	1 (1.7)
Progressive disease	25 (86.2)	24 (77.4)	49 (81.7)
Patients with ≥1 dose reduction, n (%)	11 (37.9)	6 (19.4)	17 (28.3)
Off treatment and in follow-up, n (%)	15 (51.7)	10 (32.3)	25 (41.7)
Duration of Precem-TcT therapy, Mean ± SD (months)	6.8 ± 3.88	6.3 ± 4.21	6.5 ± 4.03
Total number of infusions received, Mean ± SD	9.1 ± 5.42	8.5 ± 5.69	8.8 ± 5.52
4–6	5 (17.2)	7 (22.6)	12 (20.0)
>6	18 (62.1)	18 (58.1)	36 (60.0)

[a]Patient went to hospice

RESULTS

EFFICACY

- At 2.8 mg/kg Q3W, ORR was 31.0% (95% CI: 15.3–50.8; confirmed: 20.7% [95% CI: 8.0–39.7]) and DCR was 44.8% (95% CI: 26.4–64.3) (**Table 2, Figure S1**)
- Response did not appear to be impacted by *KRAS/NRAS/BRAF* mutation status, or by CEACAM5 expression status (**Figure 3**)
 - o All patients with available IHC data had high CEACAM5 expression levels
- At 2.8 mg/kg Q3W, after a median follow-up of 13.1 months, mOS was NR (95% CI: 8.7–NE) (**Figure 4**), mPFS was 6.9 months (95% CI: 4.4–9.5), and the 6-month PFS rate was 64.3% (95% CI: 43.8–78.9) (**Figure 5**)

Table 2. Treatment response

Parameter	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Objective response rate[a], %			
Unconfirmed (95% CI)	31.0 (15.3–50.8); n=9	12.9 (3.6–29.8); n=4	21.7 (12.1–34.2); n=13
of which confirmed (95% CI)	20.7 (8.0–39.7); n=6	6.5 (0.8–21.4); n=2	13.3 (5.9–24.6); n=8
Best overall response, unconfirmed, n (%)			
Partial response	9 (31.0)	4 (12.9)	13 (21.7)
Stable disease	13 (44.8)	20 (64.5)	33 (55.0)
Progressive disease	6 (20.7)	6 (19.4)	12 (20.0)
Not evaluable	1 (3.4)	1 (3.2)	2 (3.3)
DCR (SD+PR) at Week 24 (95% CI)[b], %	44.8 (26.4–64.3); n=13	41.9 (24.5–60.9); n=13	43.3 (30.6–56.8); n=26

Overall response according to RECIST v1.1 as assessed by Investigators. [a]Best overall response assessment of CR or PR. [b]DCR defined as having an overall response assessment of CR, PR, SD, or non-CR/non-PD at Week 24 visit or later, prior to documented PD.

Figure 3. Objective response

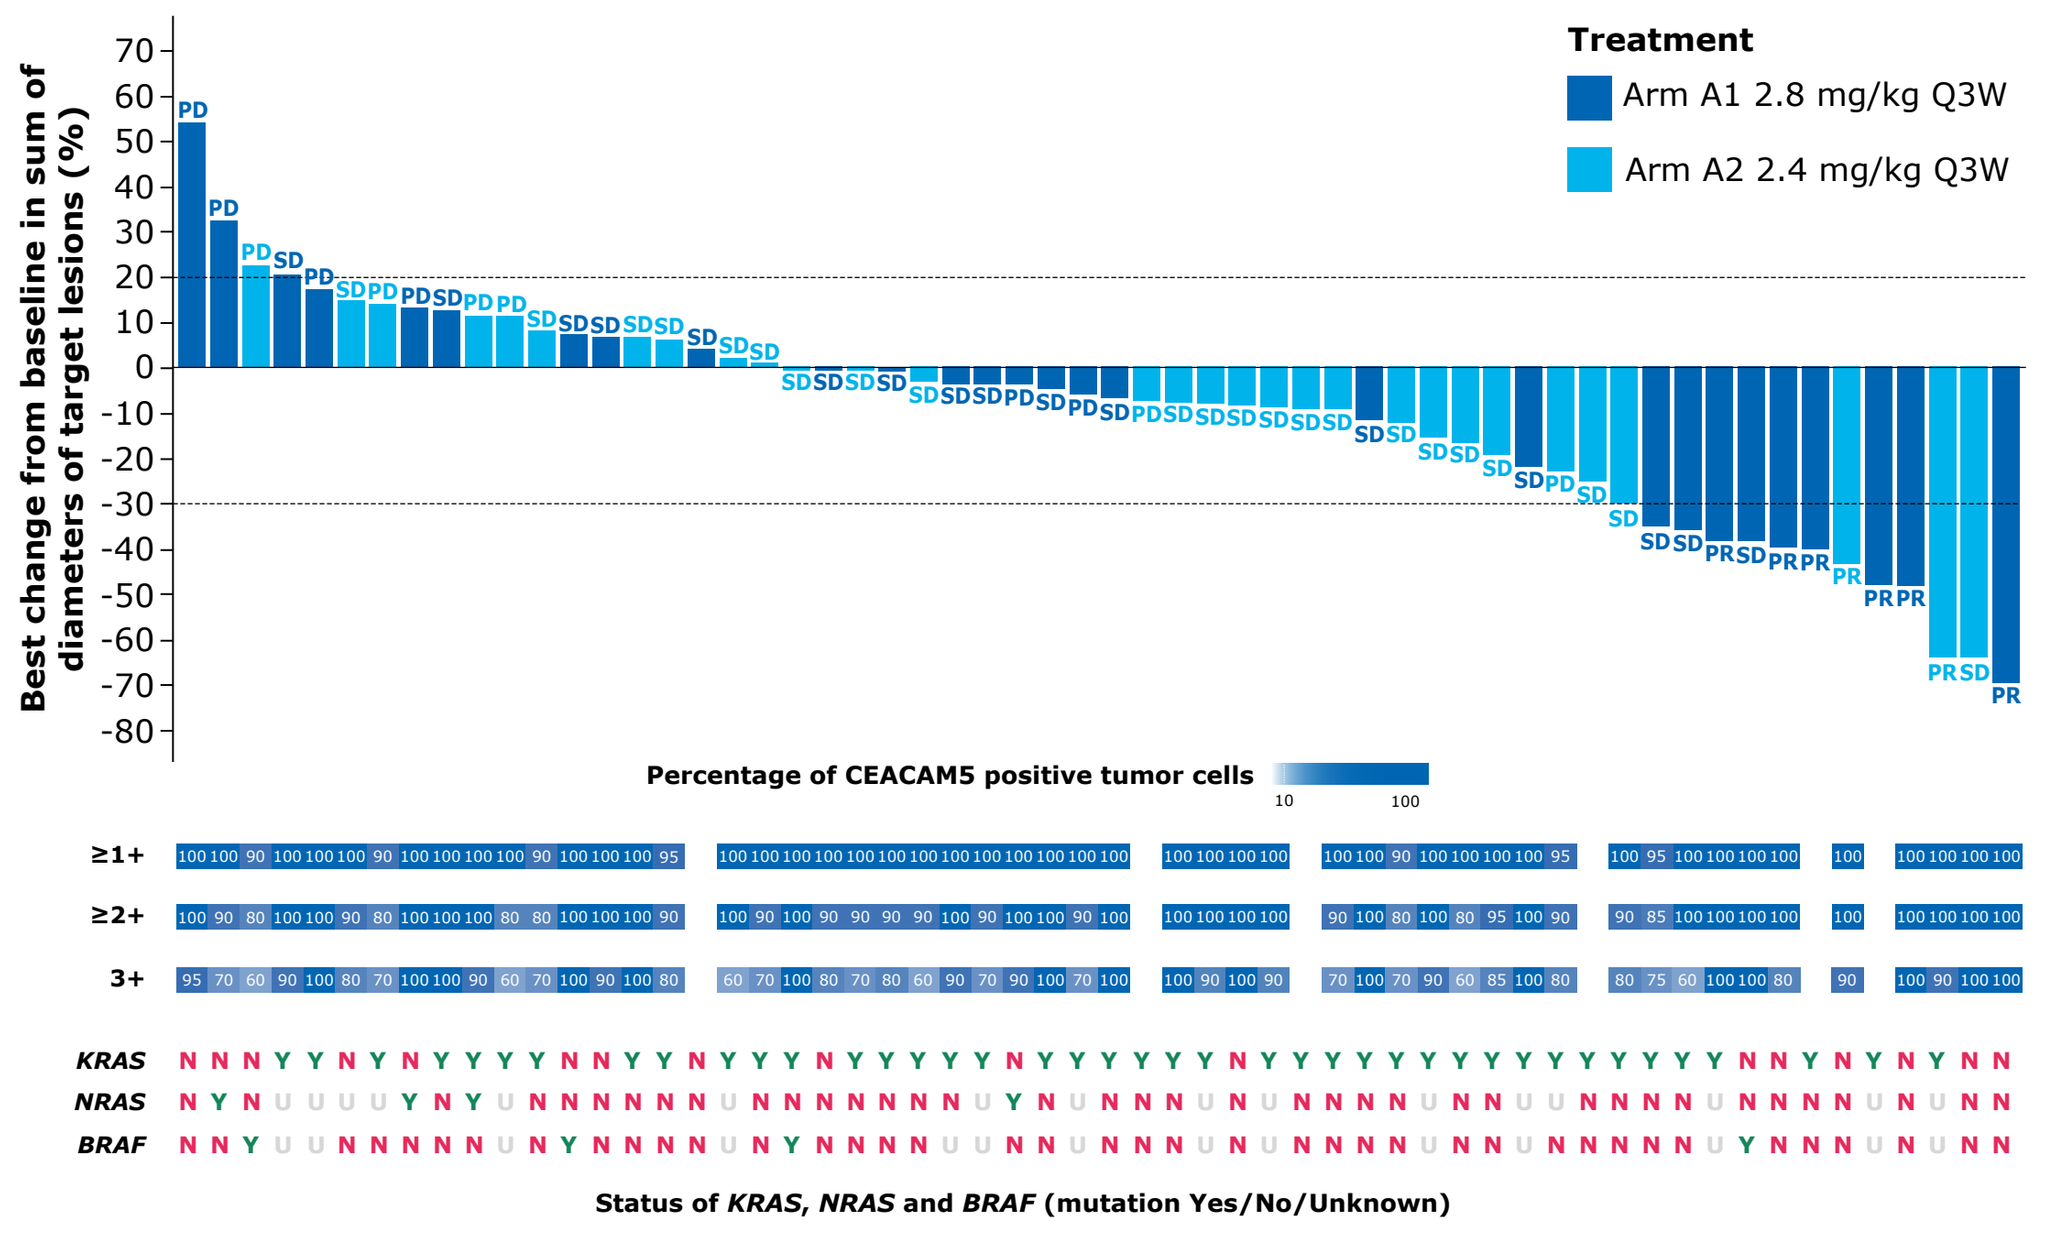
Treatment
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Best change from baseline in sum of diameters of target lesions (%)
70
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
Percentage of CEACAM5 positive tumor cells
≥1+
≥2+
3+
KRAS
NRAS
BRAF
Status of KRAS, NRAS and BRAF (mutation Yes/No/Unknown)

Figure 4. OS

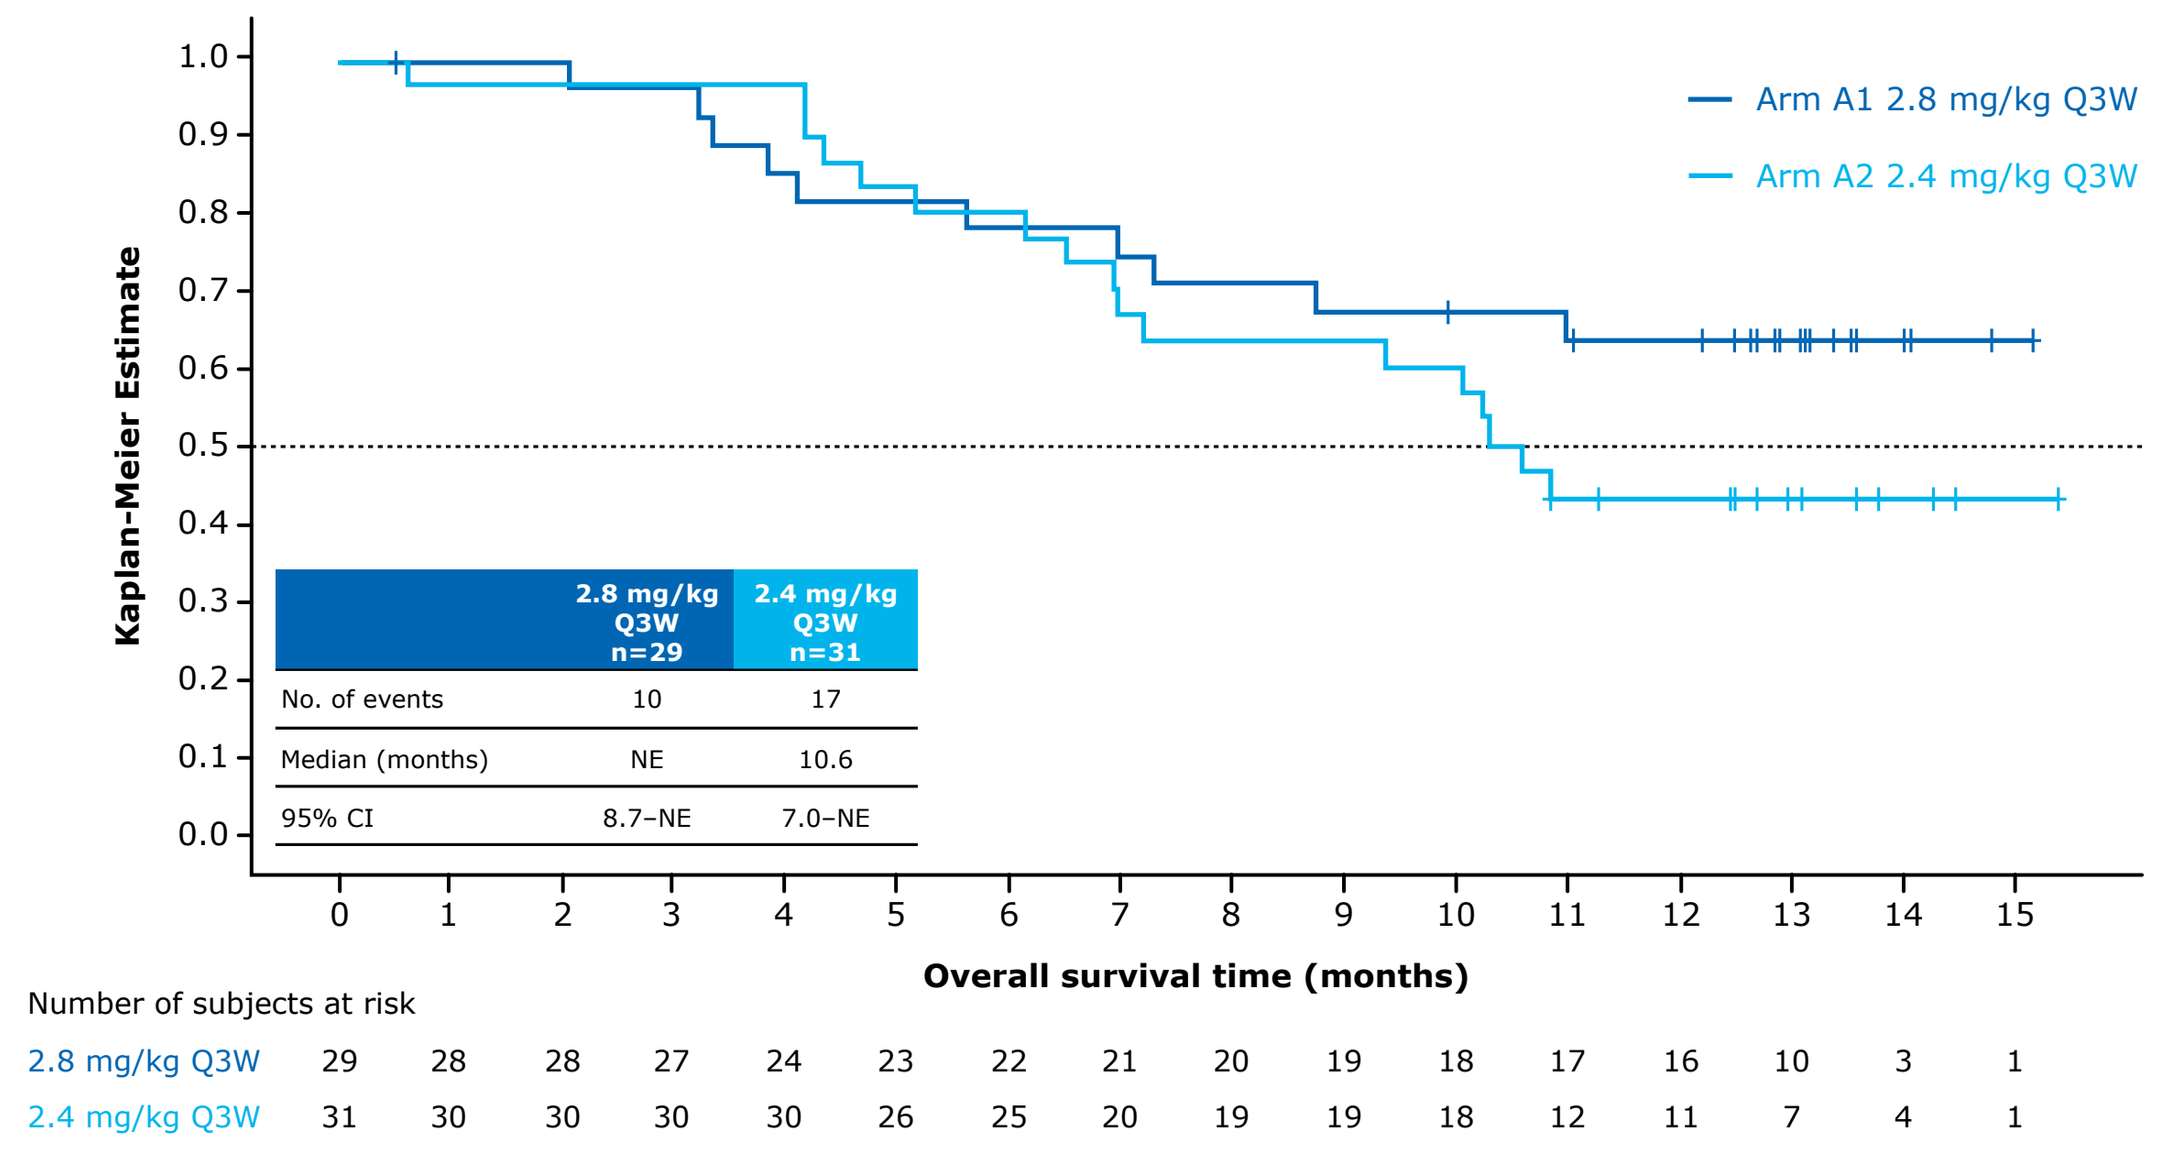
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Kaplan-Meier Estimate
	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31
No. of events	10	17
Median (months)	NE	10.6
95% CI	8.7–NE	7.0–NE
Overall survival time (months)
Number of subjects at risk
2.8 mg/kg Q3W 29 28 28 27 24 23 22 21 20 19 18 17 16 10 3 1
2.4 mg/kg Q3W 31 30 30 30 30 26 25 20 19 19 18 12 11 7 4 1

Figure 5. PFS

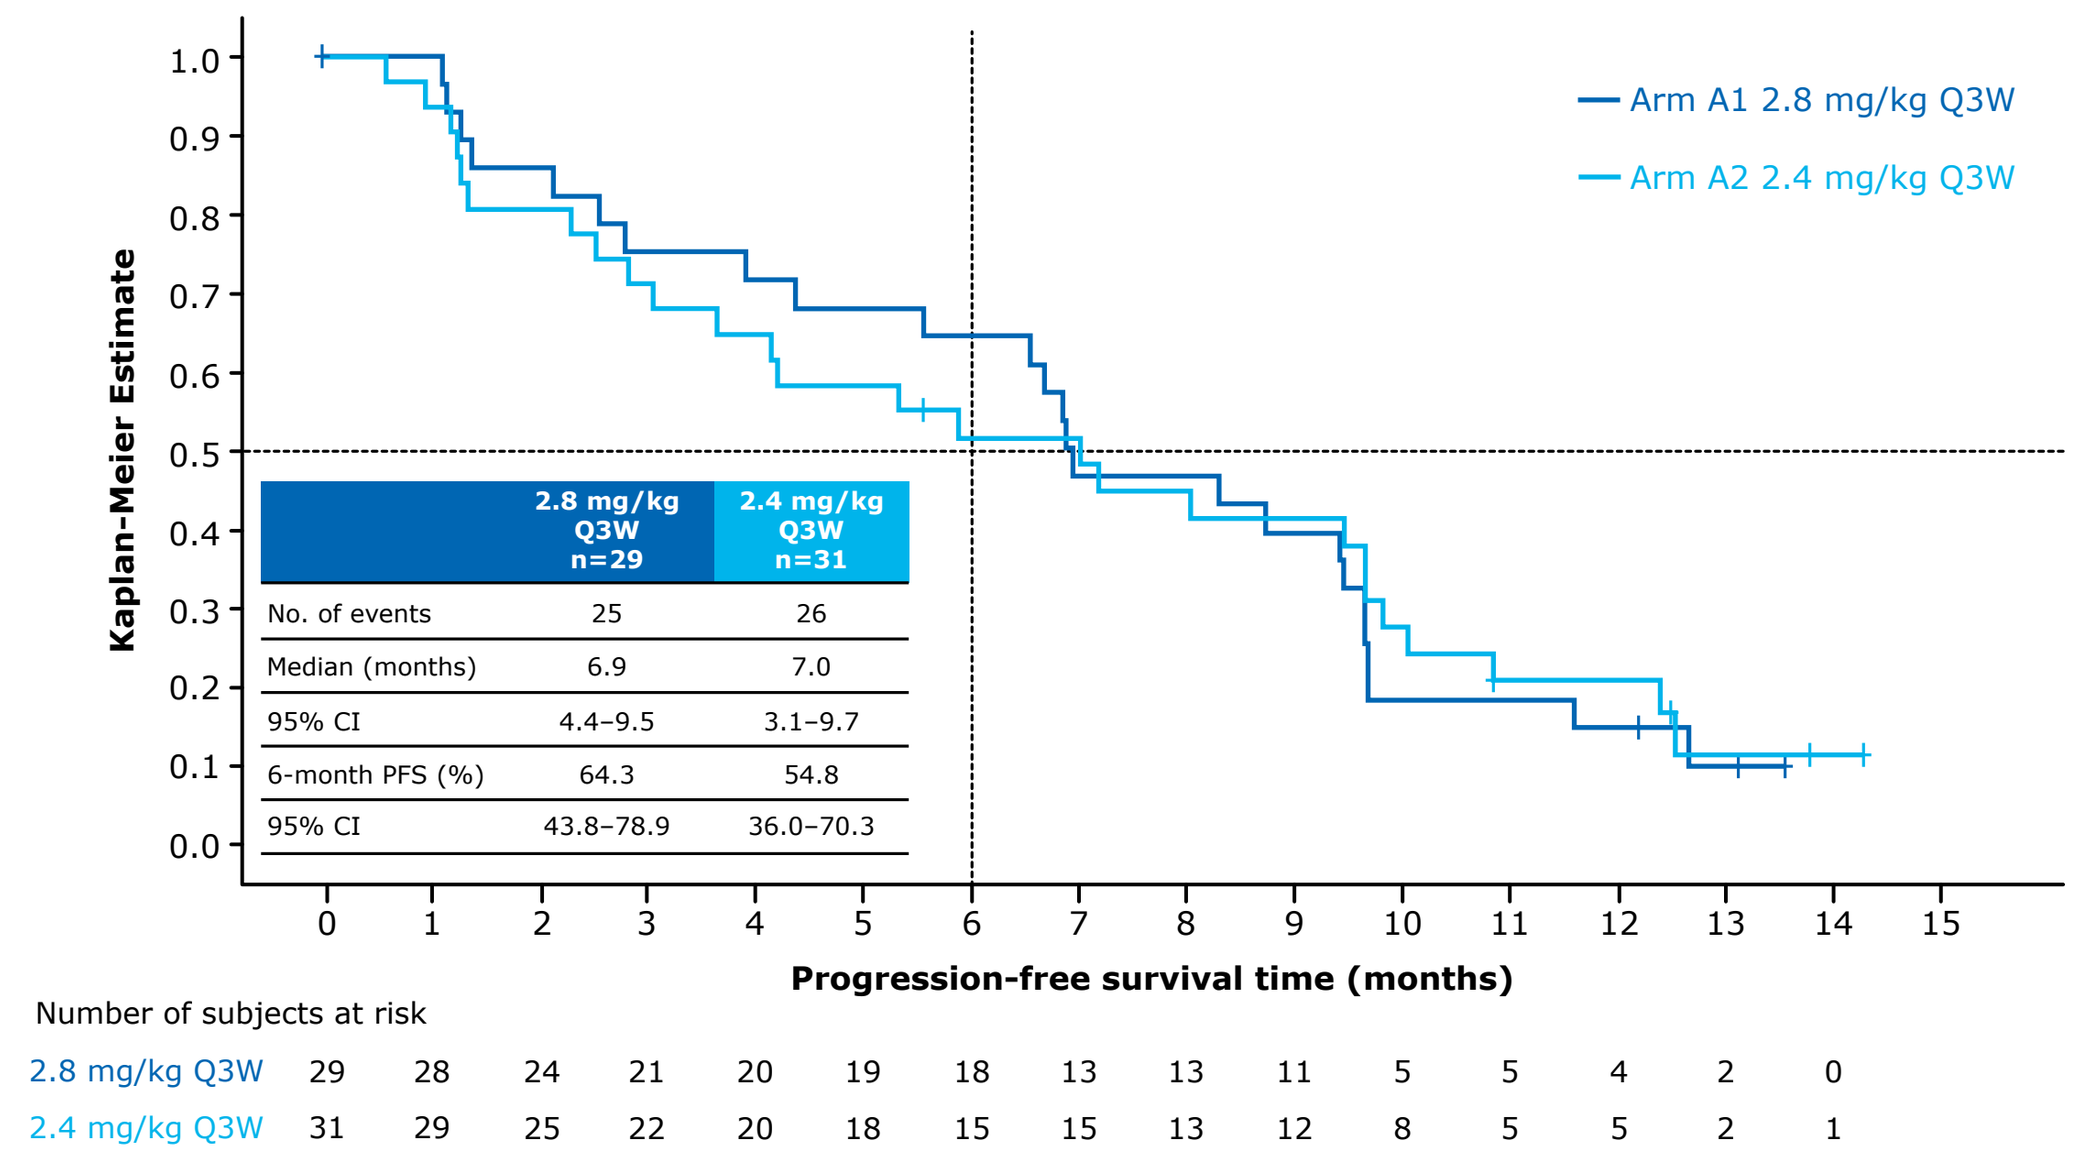
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Kaplan-Meier Estimate
	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31
No. of events	25	26
Median (months)	6.9	7.0
95% CI	4.4–9.5	3.1–9.7
6-month PFS (%)	64.3	54.8
95% CI	43.8–78.9	36.0–70.3
Progression-free survival time (months)
Number of subjects at risk
2.8 mg/kg Q3W 29 28 24 21 20 19 18 13 13 11 5 5 4 2 0
2.4 mg/kg Q3W 31 29 25 22 20 18 15 15 13 12 8 5 5 2 1

SAFETY

- Most common grade ≥3 TEAEs were hematologic* 60.0% (**Table S2**; **Figure S2**; **Figure S3)**
- Most common grade ≥3 TRAEs were neutropenia (43.3%) and anemia (40%) (**Figure 6**; **Table S3**)
- All GI TRAEs were grade 1-2; and there were no ILD events (**Table S3**)
- No Precem-TcT-related discontinuations or deaths were reported (**Table 3**)
- PK of conjugated antibody and unconjugated exatecan at 2.4 mg/kg and 2.8 mg/kg was generally consistent with data from the dose escalation phase (**Figure S4**)

*PTs for 'Blood and lymphatic system disorders' and 'Investigations' have been combined as composite terms for hematologic toxicities

Table 3. Safety overview

AEs, n (%)	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Any TEAE	29 (100.0)	31 (100.0)	60 (100.0)
Any grade ≥3 TEAE	22 (75.9)	21 (67.7)	43 (71.7)
Any grade ≥4 TEAE	5 (17.2)	7 (22.6)	12 (20.0)
Any Precem-TcT-related TEAE (TRAE)	29 (100.0)	30 (96.8)	59 (98.3)
Any grade ≥3 TRAE	20 (69.0)	17 (54.8)	37 (61.7)
Any grade ≥4 TRAE	5 (17.2)	5 (16.1)	10 (16.7)
Any TRAE leading to permanent discontinuation of Precem-TcT	0 (0.0)	0 (0.0)	0 (0.0)
Any serious TRAE	4 (13.8)	5 (16.1)	9 (15.0)
Any TEAE leading to death	0 (0.0)	2 (6.5)	2 (3.3)
Any TRAE leading to death	0 (0.0)	0 (0.0)	0 (0.0)

Figure 6. Most common TRAEs by grade

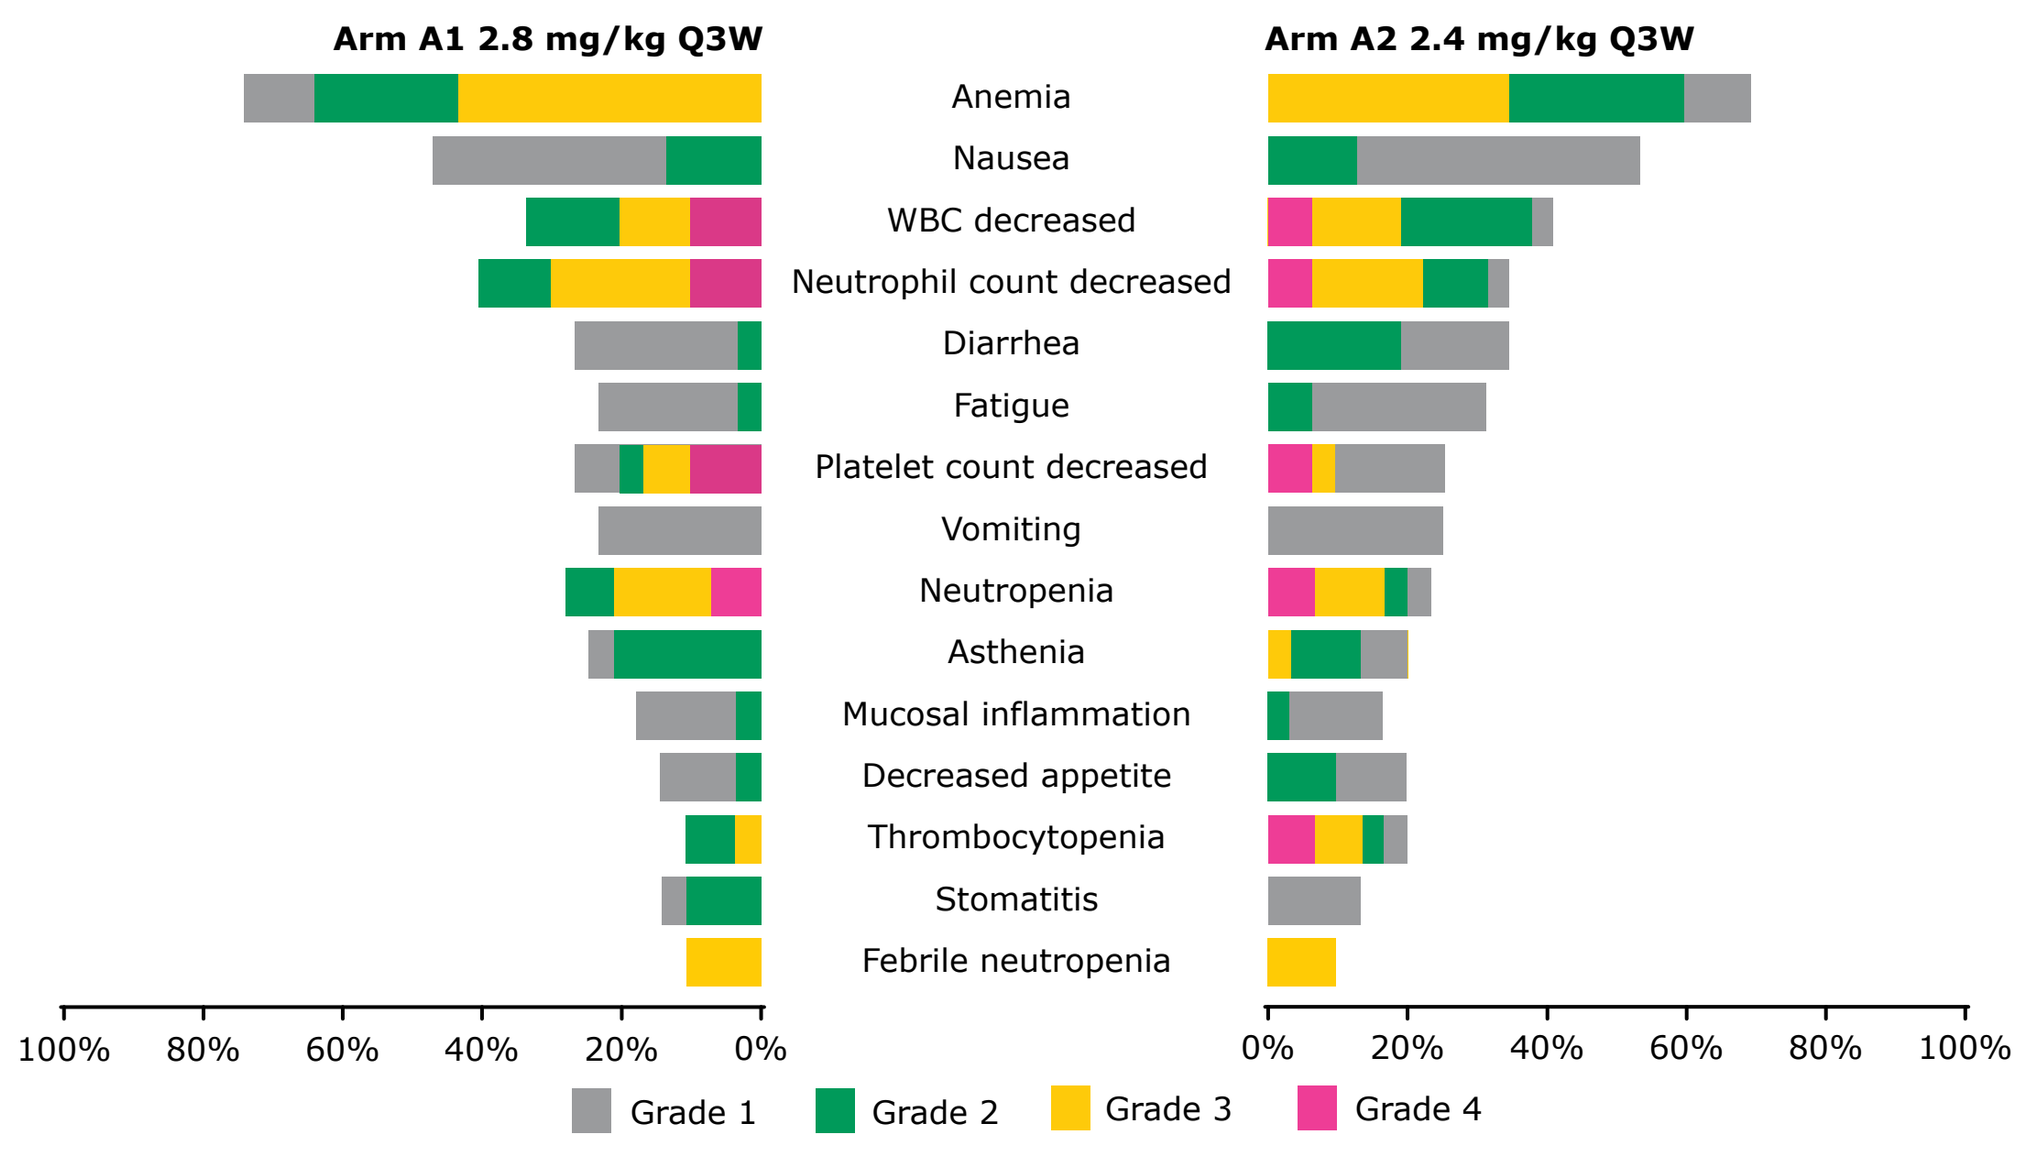
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Anemia
Nausea
WBC decreased
Neutrophil count decreased
Diarrhea
Fatigue
Platelet count decreased
Vomiting
Neutropenia
Asthenia
Mucosal inflammation
Decreased appetite
Thrombocytopenia
Stomatitis
Febrile neutropenia
100% 80% 60% 40% 20% 0%
0% 20% 40% 60% 80% 100%
Grade 1
Grade 2
Grade 3
Grade 4

Presenting author (skopetz@mdanderson.org)

Abbreviations: 3L+, third or later line of therapy; 5-FU, 5-fluorouracil; ADA, anti-drug antibody; ADC, antibody-drug conjugate; AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; AUC, area under curve; CEACAM5, carcinoembryogenic antigen-related cell adhesion molecule 5; CI, confidence interval; CR, complete response; CRC, colorectal cancer; DCR, disease control rate; DL, dose level; ECG, electrocardiogram; ECOG PS, Eastern Cooperative Oncology Group performance status; FiH, first-in-human; GGT, gamma-glutamyl transferase; GI, gastrointestinal; ICI, immune checkpoint inhibitor; ILD, interstitial lung disease; IV, intravenous; MoA, mechanism of action; la/m, locally advanced or metastatic; mCRC, metastatic colorectal cancer; MSI-H, microsatellite instability-high; NE, not estimable; NR, not reached; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetics; PR, partial response; PT, Preferred Term; Q3W, once every three weeks, RDE, recommended dose for expansion; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SD, stable disease; SMQ, Standardized MedDRA Query; SoC, standard of care; TEAE, treatment-emergent adverse event; TOP1i, topoisomerase 1 inhibitor; TRAE, treatment-related adverse event; WBC, white blood cell.

References: 1. Dasari A, et al. *Lancet.* 2023;402:41-53; 2. Weinberg BA, et al. *Clin Adv Hematol Oncol.* 2016;14:630-638; 3. Mayer RJ, et al. *N Engl J Med.* 2015;372:1909-1919; 4. Grothey A, et al. *Lancet.* 2013;381:303-312; 5. Prager GW, et al. *N Engl J Med.* 2023;388:1657-1667; 6. Beauchemin N & Arabzadeh A. *Cancer Metastasis Rev.* 2013;32(3-4):643-671; 7. Zhang X, et al. *J Int Med Res.* 2020;48(9):300060520959478; 8. Decary S, et al. *Clin Cancer Res.* 2020;26(24):6589-6599; 9. Raab-Westphal S, et al. *Cancer Res.* 2024;84(6_suppl):2362; 10. Boni V, et al. *Ann Oncol.* 2024;35(2_suppl):S428-S481; 11. Kopetz S, et al. *J Clin Oncol.* 2024;42(16_suppl):3000.

Acknowledgements: The authors would like to thank patients, investigators, co-investigators, and the study teams at each of the participating centers. The authors would like to acknowledge the contributions of Abdul Siddiqui, who at the time of the study was affiliated with Global Patient Safety at Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany. The trial was funded by the healthcare business of Merck (CrossRef Funder ID: 10.13039/100009945). Medical writing assistance was provided by Ramya Gupta of Merck Specialities Pvt. Ltd, Bangalore, India, an affiliate of |the healthcare business of Merck KGaA, Darmstadt, Germany.

Disclosures: SK reports stock and other ownership interests in Frontier Medicines, Iylon, Lutris, Navire, Xilis, consulting or advisory role in Abbvie, Accademia Nazionale Di Medicina (ACCMED), Amal Therapeutics, Amgen, AstraZeneca/MedImmune, AVEO, Bayer Health, Bicara Therapeutics, Black Diamond Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb/Medarex, Cardiff Oncology, Carina Biotech, CureTeq, Merck, Endeavor BioMedicines, Flame Biosciences, Foundation Medicine, Frontier Medicines, Genentech, Genomic Health, Gilead Sciences, GlaxoSmithKline, HalioDx, Harbinger Oncology, Inc., Holy Stone Healthcare, Inivata, Ipsen, Iylon, Jacobio, Jazz Pharmaceuticals, Johnson & Johnson/Janssen, Lilly, Lutris, MSD, Mirati Therapeutics, NeoGenomics Laboratories, Novartis, Numab, Ono Pharmaceutical, Pfizer, Redx Pharma, Repare Therapeutics, Replimune, Roche, Sanofi, Servier, Tachyon Therapeutics, Taiho Pharmaceutical, Takeda, Tempus, Xilis, Zentalis, and research funding from Amgen, Array BioPharma, Biocartis, Daiichi Sankyo, Merck, Genentech/Roche, Guardant Health, Lilly, MedImmune, Novartis, Sanofi.

Abstract #7056 | Presented at the European Society of Medical Oncology (ESMO) Congress 2025 | Oct 17–21, 2025 | Berlin, Germany

962P

Precemtabart tocentecan (M9140), an anti-CEACAM5 ADC with exatecan payload, in patients with metastatic colorectal cancer: updated clinical data from the Phase 1 PROCEADE-CRC-01 study

Scott Kopetz[1], Rocio Garcia-Carbonero[2], Sae-Won Han[3], Ildefonso Ismael Rodriguez-Rivera[4], Jose Carlos Ruffinelli Rodriguez[5], Valentina Boni[6], Mario Vieito Villar[7], Maria de Miguel[8], Athanasios Pallis[9], Christina Habermehl[10], Ken Kato[11]

[1]GI Medical Oncology Department, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Medical Oncology, Hospital Universitario 12 de Octubre, Imas12, Universidad Complutense de Madrid, Madrid, Spain; [3]Department of Internal Medicine, Seoul National University Hospital, Seoul, Republic of Korea; [4]Oncology, NEXT Oncology, San Antonio, USA; [5]Medical Oncology Department, ICO - Institut Català d'Oncologia l'Hospitalet (Hospital Duran i Reynals), L'Hospitalet De Llobregat, Spain; [6]Department of Oncology, Hospital Universitario Quirónsalud Madrid, Pozuelo De Alarcon, Spain; [7]Department of Medical Oncology, Vall d'Hebron University Hospital, Barcelona, Spain; [8]Centro Integral Oncologico Clara Campal, START Madrid-CIOCC, Madrid, Spain; [9]Global Clinical Development, Merck Santé S.A.S., Lyon, France, an affiliate of Merck KGaA, Darmstadt, Germany; [10]Global Biostatistics, Merck KGaA, Darmstadt, Germany; [11]Department of Gastrointestinal Medical Oncology, National Cancer Center - Tsukiji Campus, Chuo-ku, Japan

SUPPLEMENTARY DATA

Table S1. Baseline and demographic characteristics

Characteristics, n (%)	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Age			
Median (range), years	60.0 (41.0–80.0)	61.0 (29.0–77.0)	60.0 (29.0–80.0)
Age categories			
<65 years	18 (62.1)	20 (64.5)	38 (63.3)
≥65 years	11 (37.9)	11 (35.5)	22 (36.7)
Sex			
Male	15 (51.7)	16 (51.6)	31 (51.7)
Female	14 (48.3)	15 (48.4)	29 (48.3)
Race			
Asian	9 (31.0)	13 (41.9)	22 (36.7)
White	17 (58.6)	17 (54.8)	34 (56.7)
Other	3 (10.3)	1 (3.2)	4 (6.7)
ECOG performance status			
0	21 (72.4)	23 (74.2)	44 (73.3)
1	8 (27.6)	8 (25.8)	16 (26.7)
Site of primary tumor			
Colon, left	10 (34.5)	5 (16.1)	15 (25.0)
Colon, right	9 (31.0)	14 (45.2)	23 (38.3)
Rectum	6 (20.7)	9 (29.0)	15 (25.0)
Time since initial cancer diagnosis			
Median (min–max), years	2.0 (0.8–7.6)	2.3 (0.2–8.6)	2.1 (0.2–8.6)
Time since metastatic disease diagnosis			
Median (min–max), years	1.6 (0.3–5.1)	1.6 (0.2–8.6)	1.6 (0.2–8.6)
Previous exposure to irinotecan	29 (100.0)	31 (100.0)	60 (100.0)

Figure S1. Treatment duration and response over time

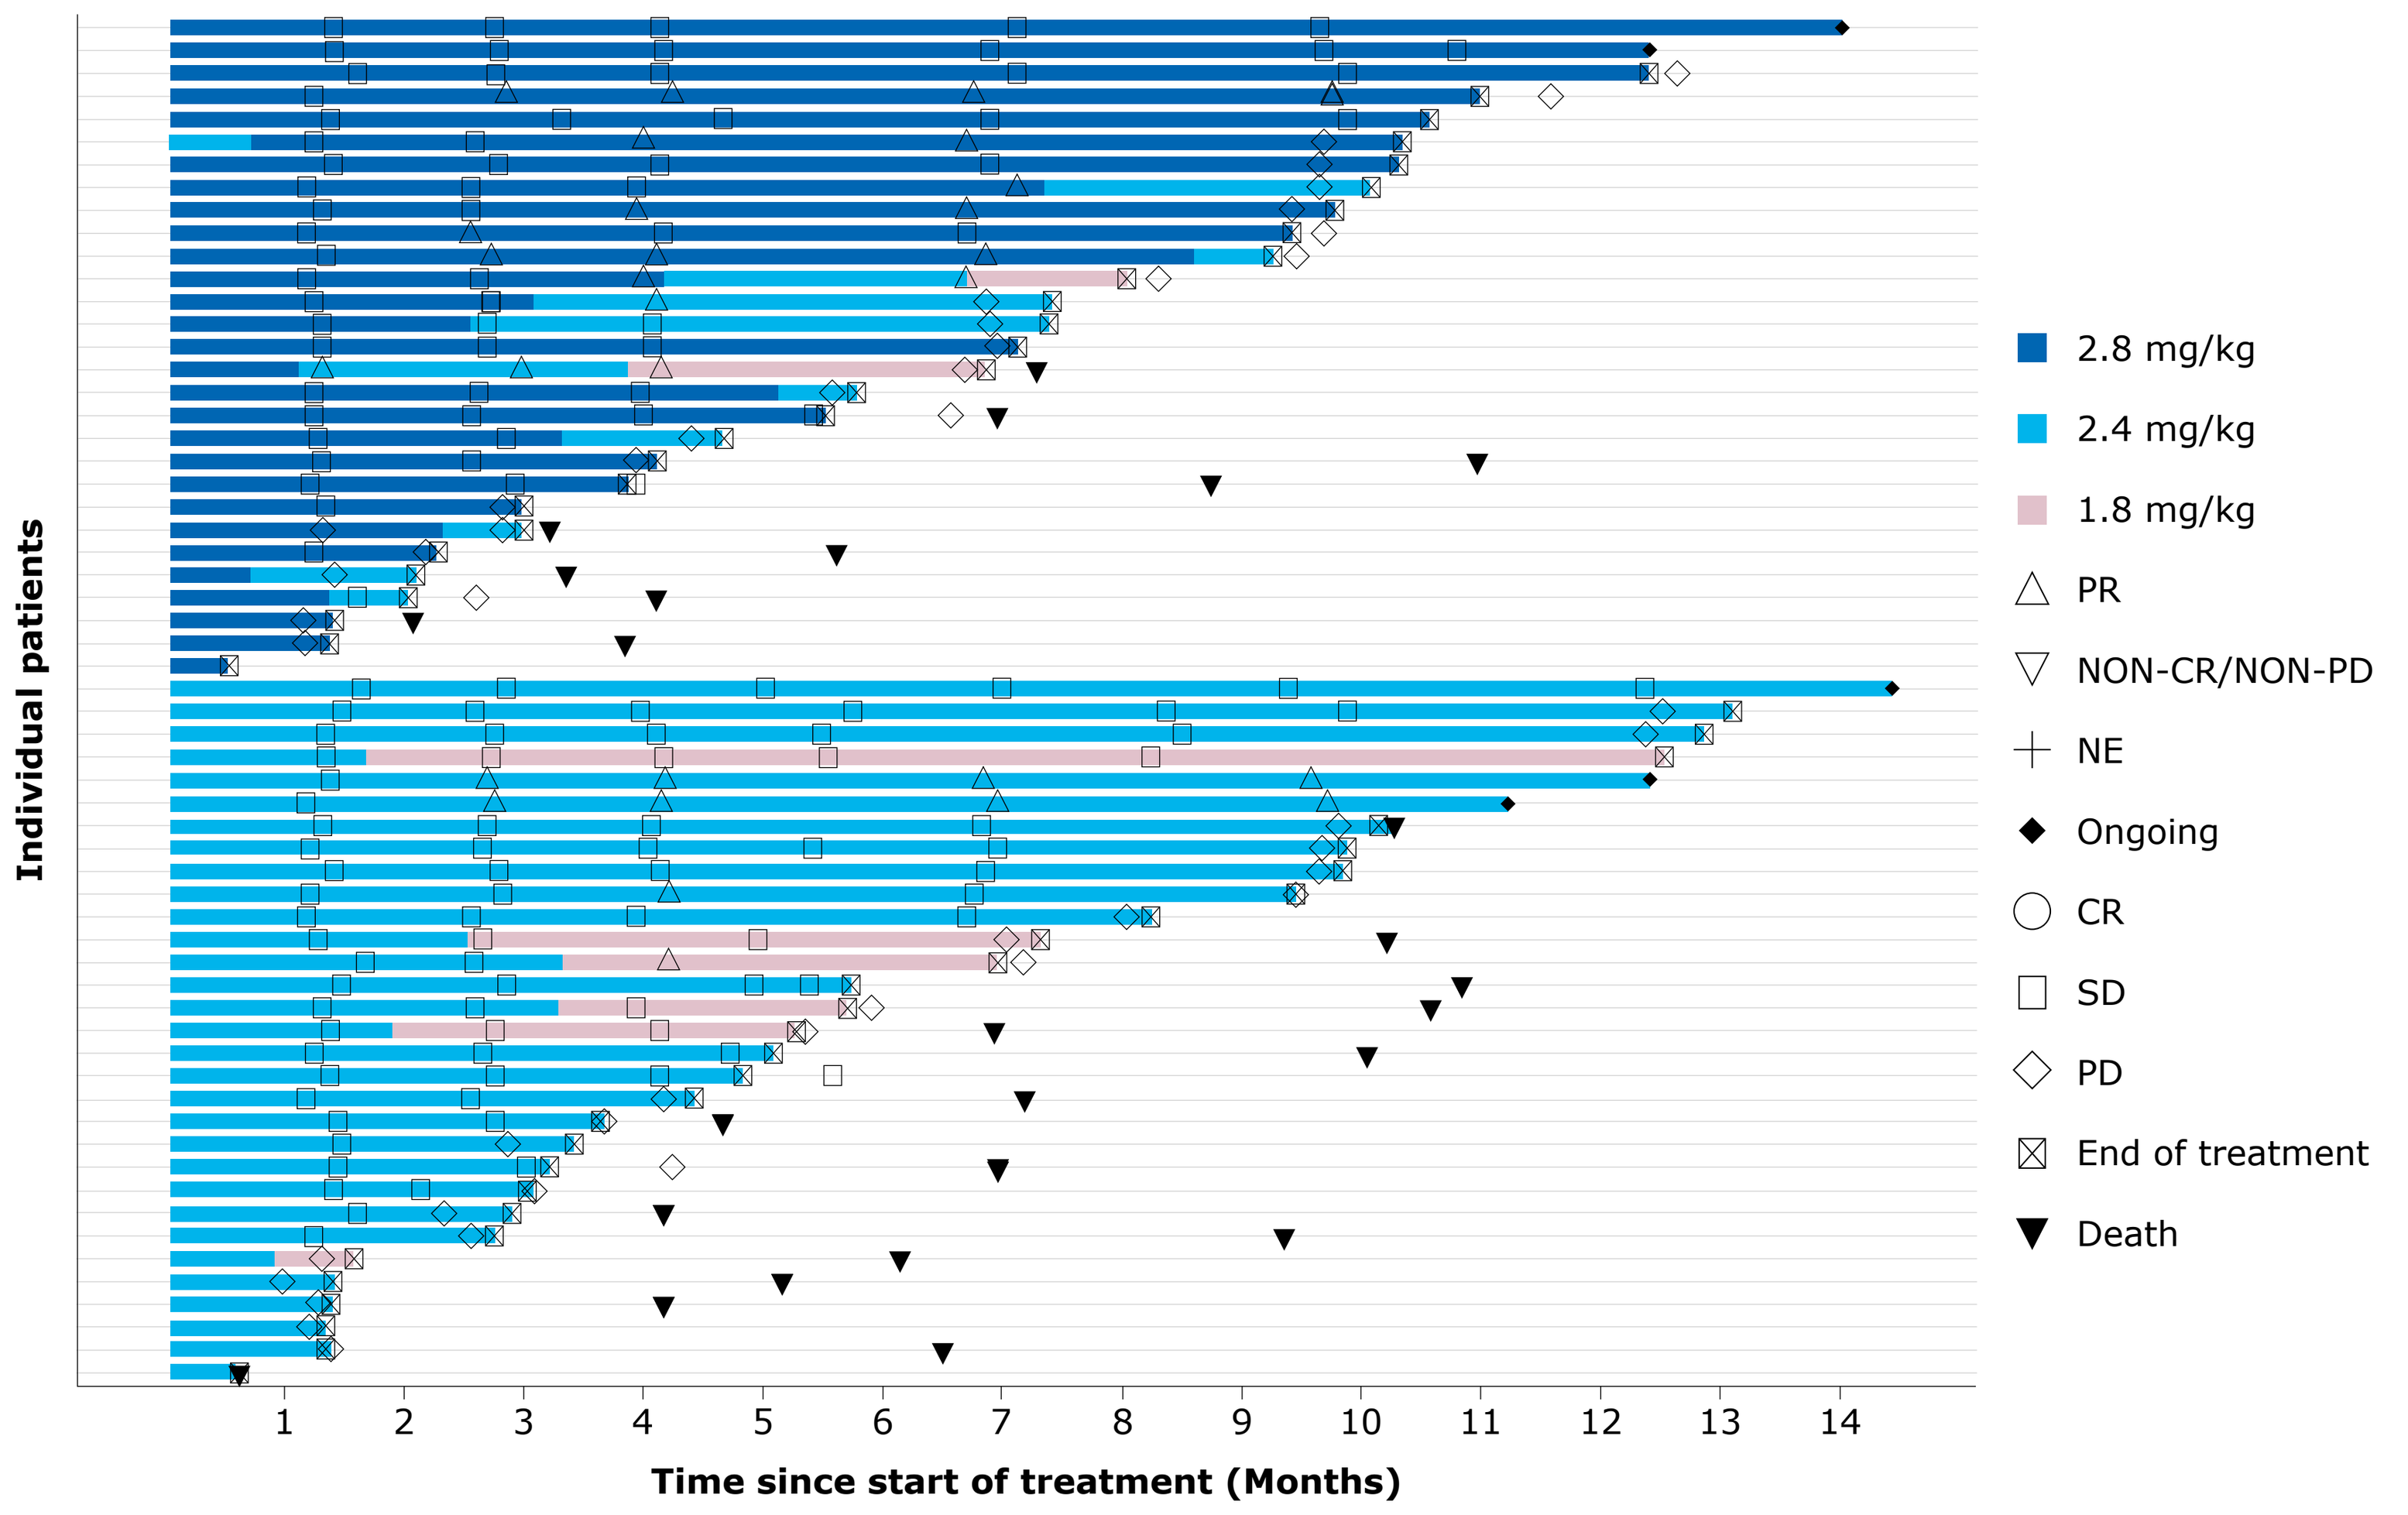
Individual patients
1 2 3 4 5 6 7 8 9 10 11 12 13 14
Time since start of treatment (Months)
2.8 mg/kg
2.4 mg/kg
1.8 mg/kg
PR
NON-CR/NON-PD
NE
Ongoing
CR
SD
PD
End of treatment
Death

Table S2. Selected[a] grade ≥3 TEAEs

TEAEs, n (%)	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Any grade ≥3 TEAE	22 (75.9)	21 (67.7)	43 (71.7)
Blood and lymphatic system disorders	17 (58.6)	15 (48.4)	32 (53.3)
Anemia	13 (44.8)	11 (35.5)	24 (40.0)
Neutropenia	6 (20.7)	5 (16.1)	11 (18.3)
Febrile neutropenia	3 (10.3)	3 (9.7)	6 (10.0)
Thrombocytopenia	1 (3.4)	4 (12.9)	5 (8.3)
Investigations	11 (37.9)	14 (45.2)	25 (41.7)
Neutrophil count decreased	9 (31.0)	7 (22.6)	16 (26.7)
WBC count decreased	6 (20.7)	6 (19.4)	12 (20.0)
Platelet count decreased	5 (17.2)	3 (9.7)	8 (13.3)
ALT increased	0 (0.0)	4 (12.9)	4 (6.7)
AST increased	1 (3.4)	3 (9.7)	4 (6.7)
GGT increased	0 (0.0)	4 (12.9)	4 (6.7)
Lymphocyte count decreased	1 (3.4)	2 (6.5)	3 (5.0)
Blood bilirubin increased	0 (0.0)	2 (6.5)	2 (3.3)
General disorders and administration site conditions	1 (3.4)	2 (6.5)	3 (5.0)
Asthenia	1 (3.4)	1 (3.2)	2 (3.3)
Nervous system disorders	1 (3.4)	1 (3.2)	2 (3.3)
Hepatobiliary disorders	2 (6.9)	5 (16.1)	7 (11.7)
Hepatic failure	1 (3.4)	1 (3.2)	2 (3.3)
GI disorders	3 (10.3)	2 (6.5)	5 (8.3)
Infections and infestations	5 (17.2)	0 (0.0)	5 (8.3)
Pneumonia	2 (6.9)	0 (0.0)	2 (3.3)

[a]Preferred term ≥2 patients

Figure S2. Most common (≥10%) TEAEs by grade

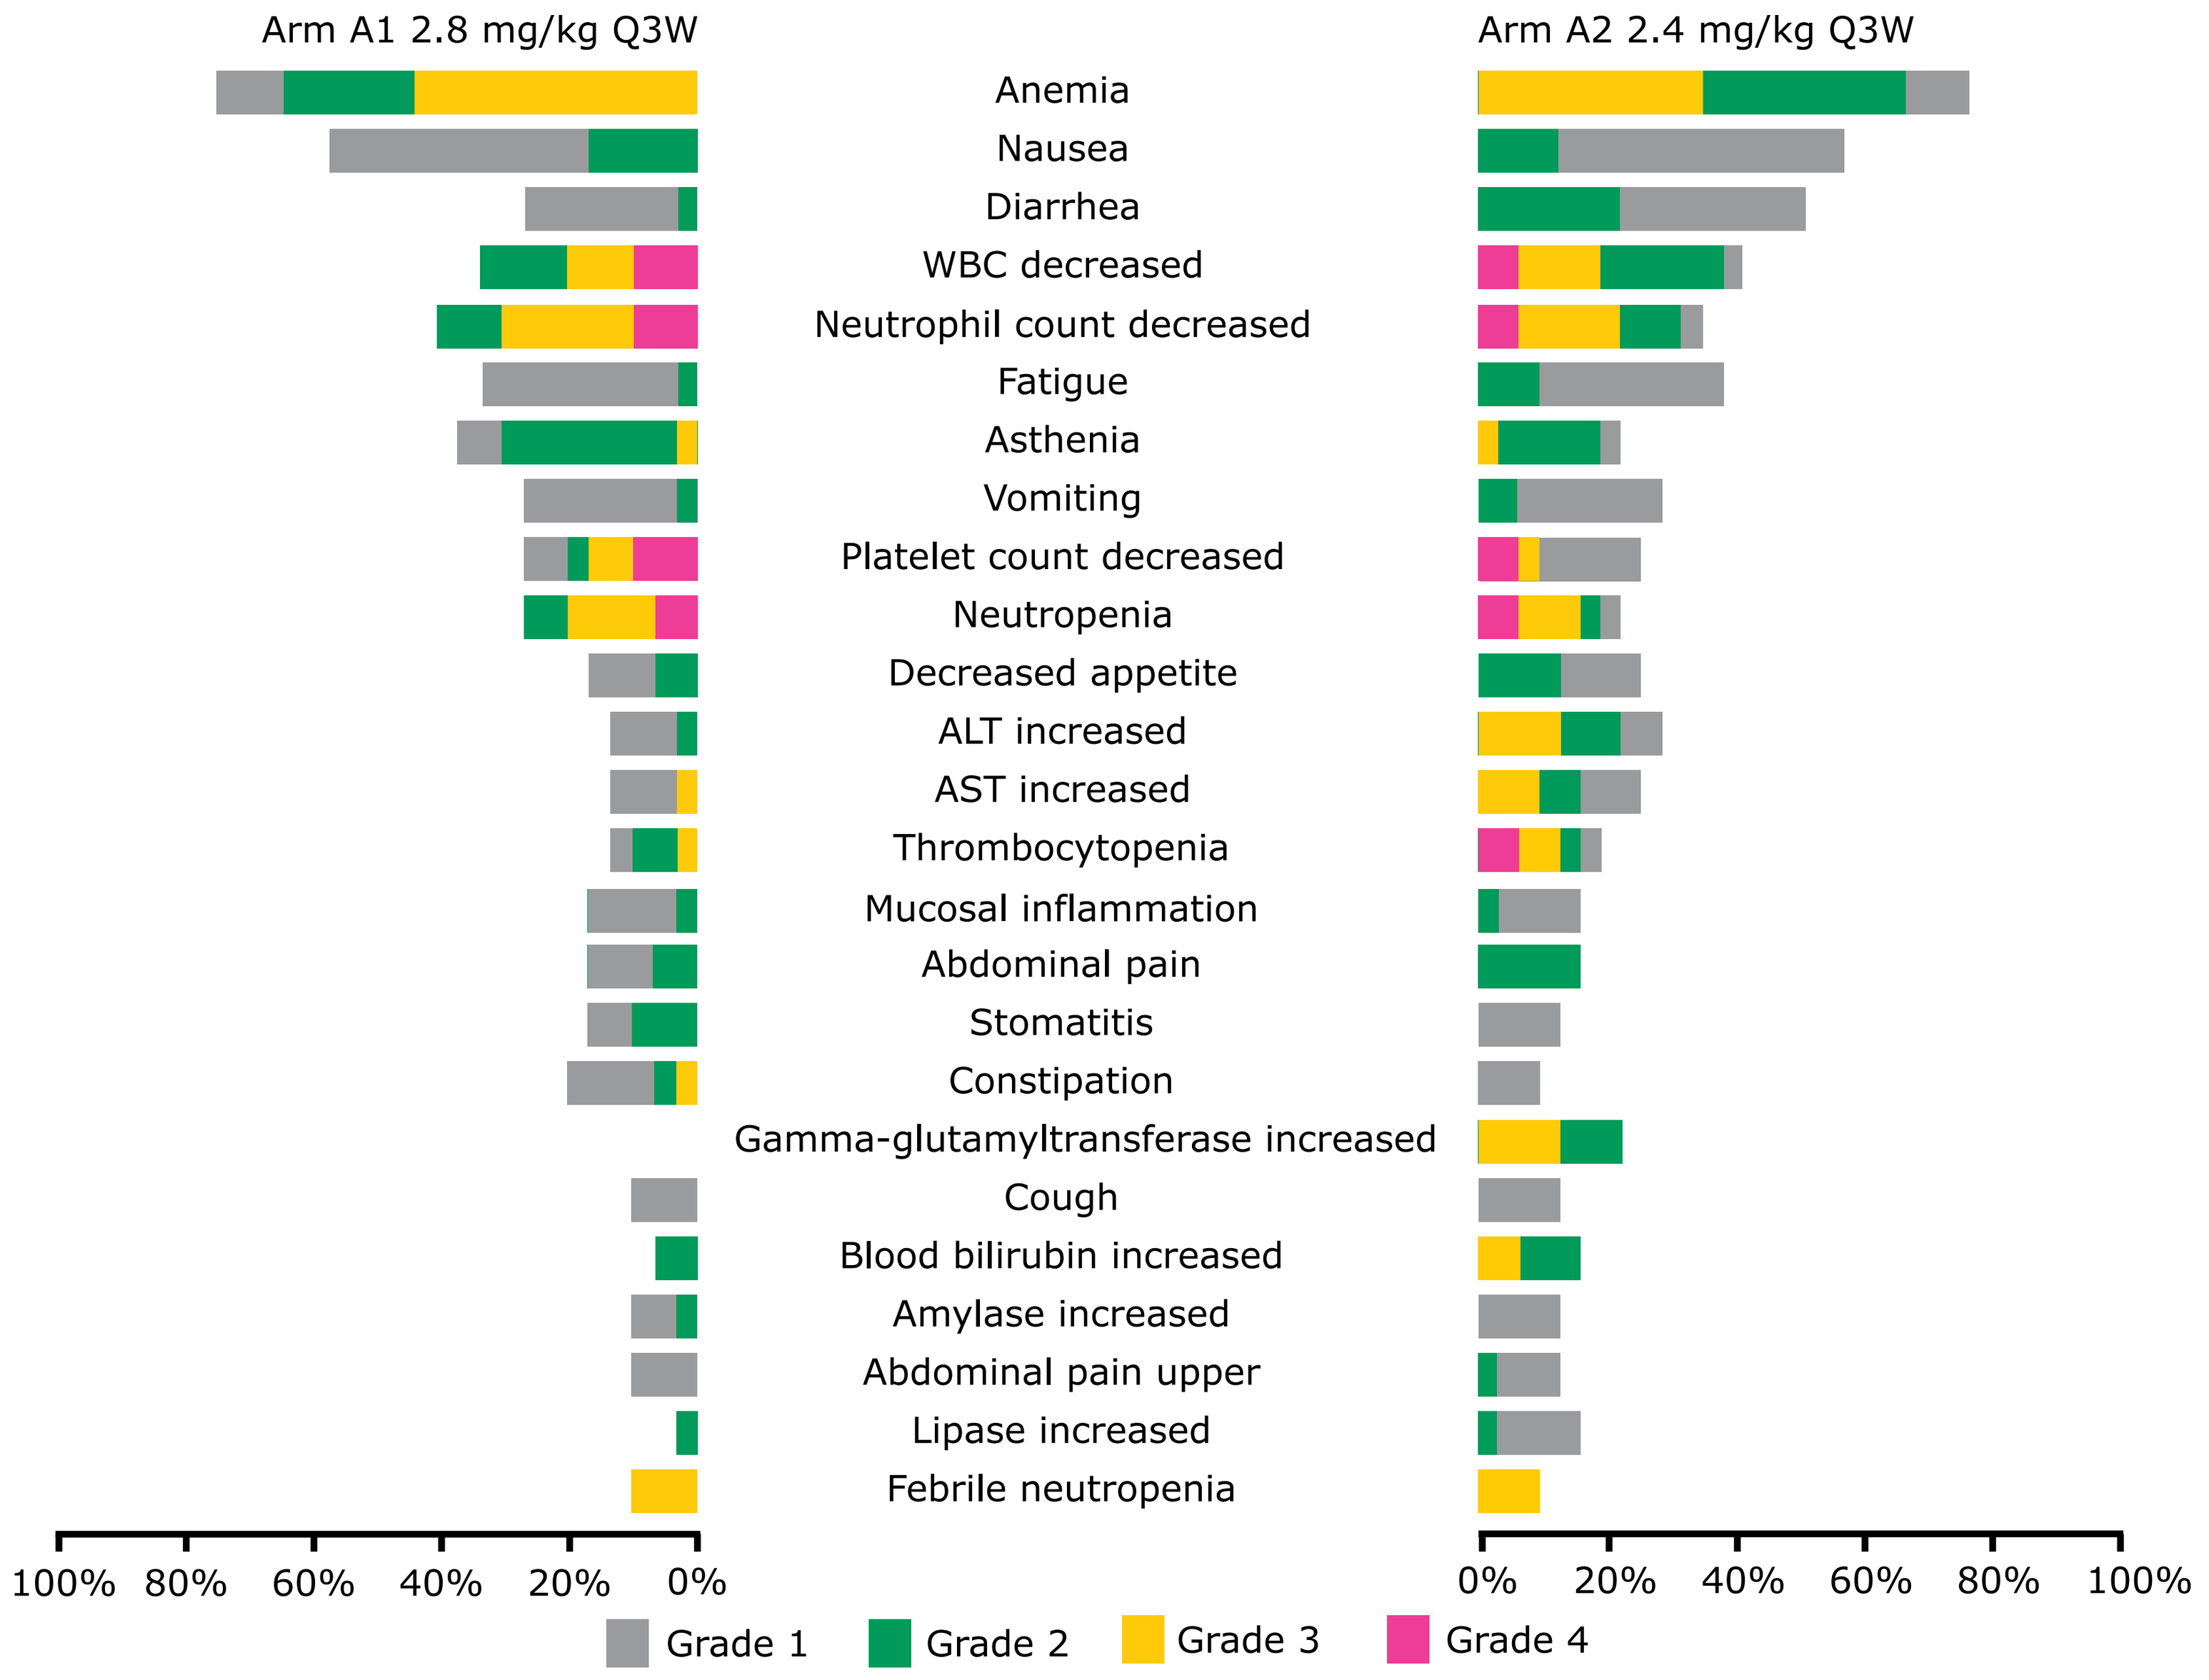
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Anemia
Nausea
Diarrhea
WBC decreased
Neutrophil count decreased
Fatigue
Asthenia
Vomiting
Platelet count decreased
Neutropenia
Decreased appetite
ALT increased
AST increased
Thrombocytopenia
Mucosal inflammation
Abdominal pain
Stomatitis
Constipation
Gamma-glutamyltransferase increased
Cough
Blood bilirubin increased
Amylase increased
Abdominal pain upper
Lipase increased
Febrile neutropenia
100% 80% 60% 40% 20% 0%
0% 20% 40% 60% 80% 100%
Grade 1
Grade 2
Grade 3
Grade 4

Table S3. All grade ≥3 TRAEs

TRAEs, n (%)	2.8 mg/kg Q3W n=29	2.4 mg/kg Q3W n=31	Total N=60
Any grade ≥3 TRAE	22 (75.9)	21 (67.7)	43 (71.7)
Blood and lymphatic system disorders	17 (58.6)	15 (48.4)	32 (53.3)
Anemia	13 (44.8)	11 (35.5)	24 (40.0)
Neutropenia	6 (20.7)	5 (16.1)	11 (18.3)
Febrile neutropenia	3 (10.3)	3 (9.7)	6 (10.0)
Thrombocytopenia	1 (3.4)	4 (12.9)	5 (8.3)
Pancytopenia	0 (0.0)	1 (3.2)	1 (1.7)
Investigations	10 (34.5)	8 (25.8)	18 (30.0)
Neutrophil count decreased	9 (31.0)	7 (22.6)	16 (26.7)
WBC count decreased	6 (20.7)	6 (19.4)	12 (20.0)
Platelet count decreased	5 (17.2)	3 (9.7)	8 (13.3)
Lymphocyte count decreased	1 (3.4)	1 (3.2)	2 (3.3)
Hepatobiliary disorders	0 (0.0)	1 (3.2)	1 (1.7)
Hepatic function abnormal	0 (0.0)	1 (3.2)	1 (1.7)

Figure S3. Hematologic TEAEs (blood and lymphatic system disorders)[a]

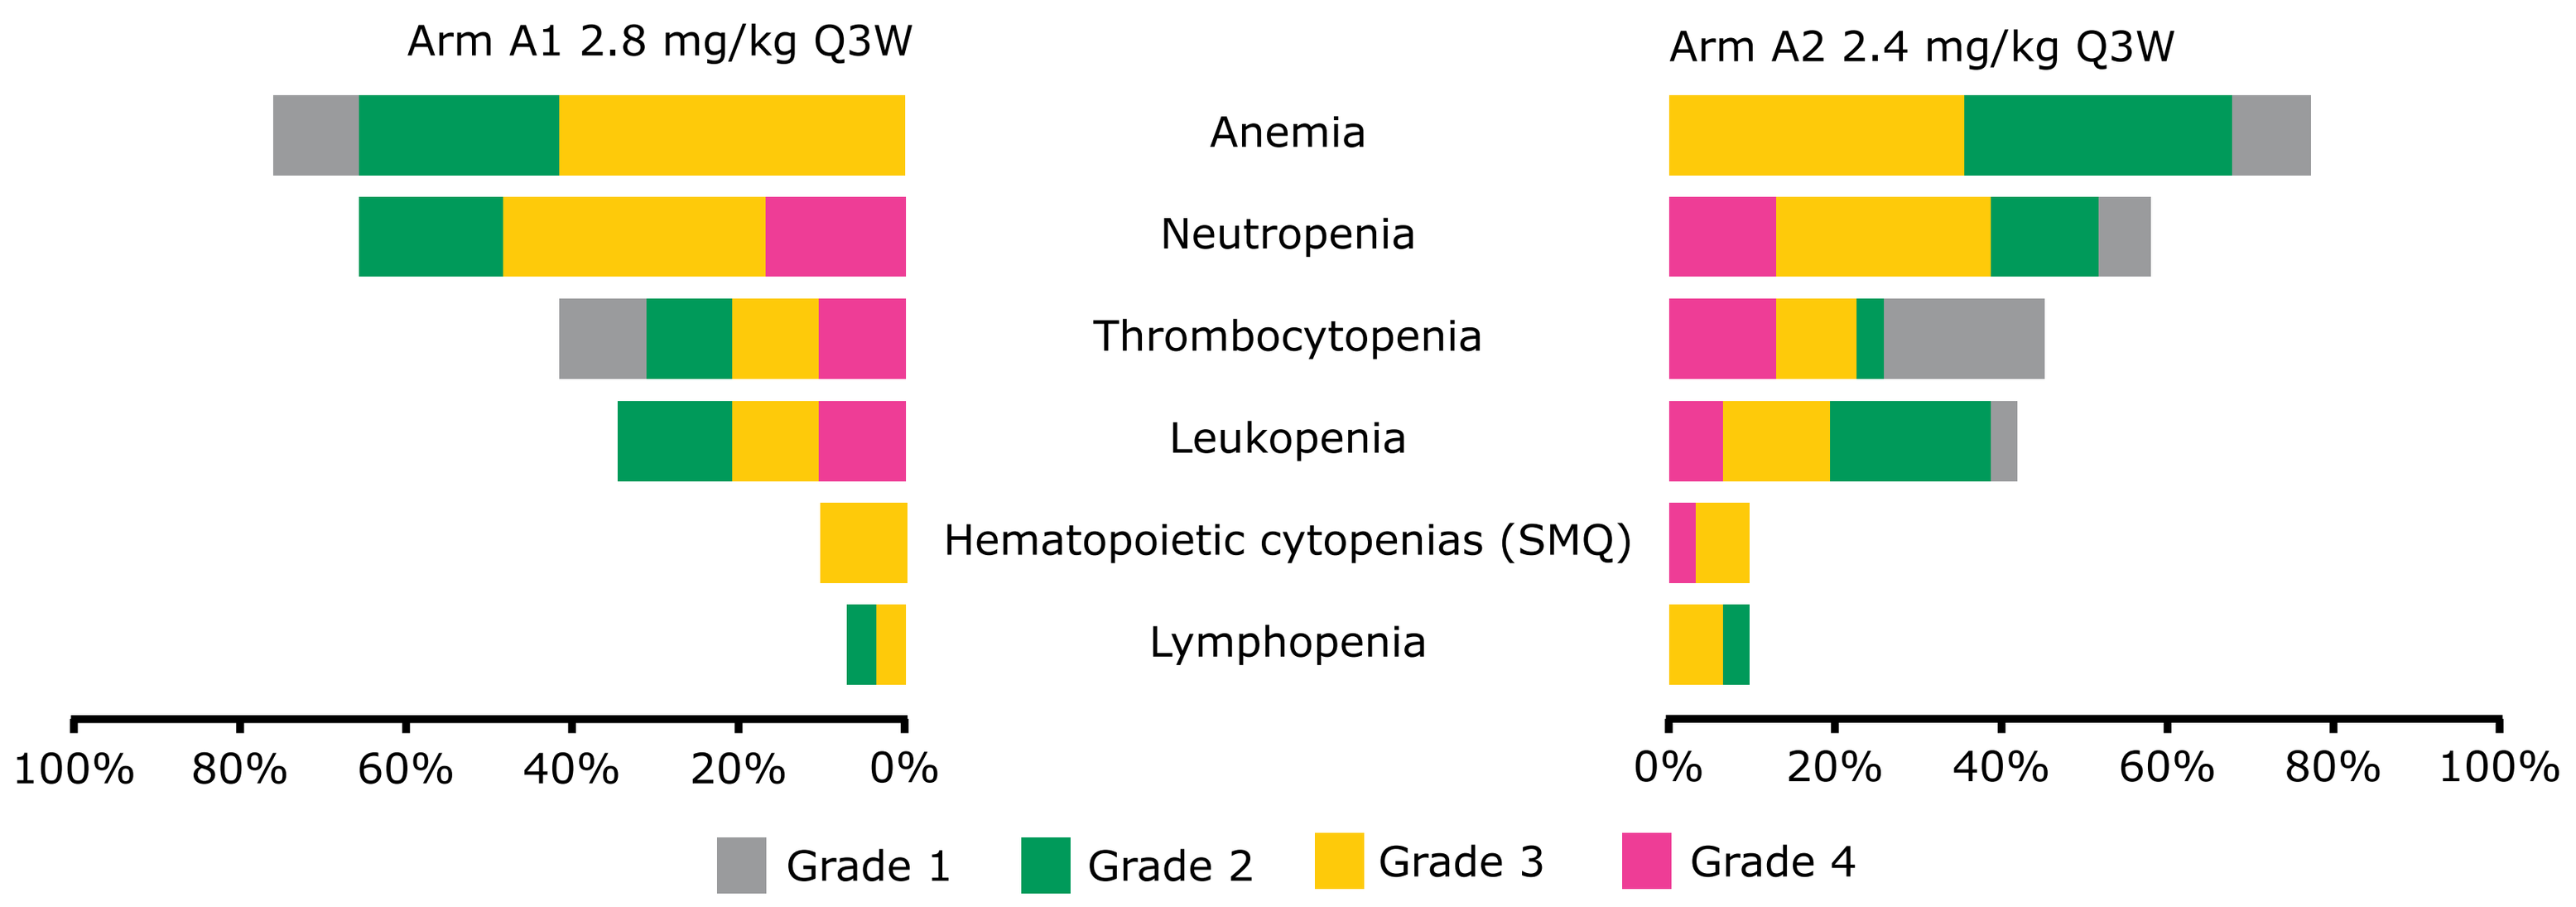
Arm A1 2.8 mg/kg Q3W
Arm A2 2.4 mg/kg Q3W
Anemia
Neutropenia
Thrombocytopenia
Leukopenia
Hematopoietic cytopenias (SMQ)
Lymphopenia
100% 80% 60% 40% 20% 0%
0% 20% 40% 60% 80% 100%
Grade 1
Grade 2
Grade 3
Grade 4

[a]Anemia: includes anaemia, haemoglobin decreased, RBC count decreased, and haematocrit decreased; neutropenia: includes neutropenia and neutrophil count decreased; thrombocytopenia: includes thrombocytopenia and platelet count decreased; leukopenia: includes leukopenia and WBC count decreased; lymphopenia: includes lymphopenia and lymphocyte count decreased; haematopoietic cytopenias excludes the Preferred Terms contained in the anaemia, neutropenia, thrombocytopenia, leukopenia, and lymphopenia.

Figure S4. PK of Precem-TcT at 2.4 mg/kg and 2.8 mg/kg

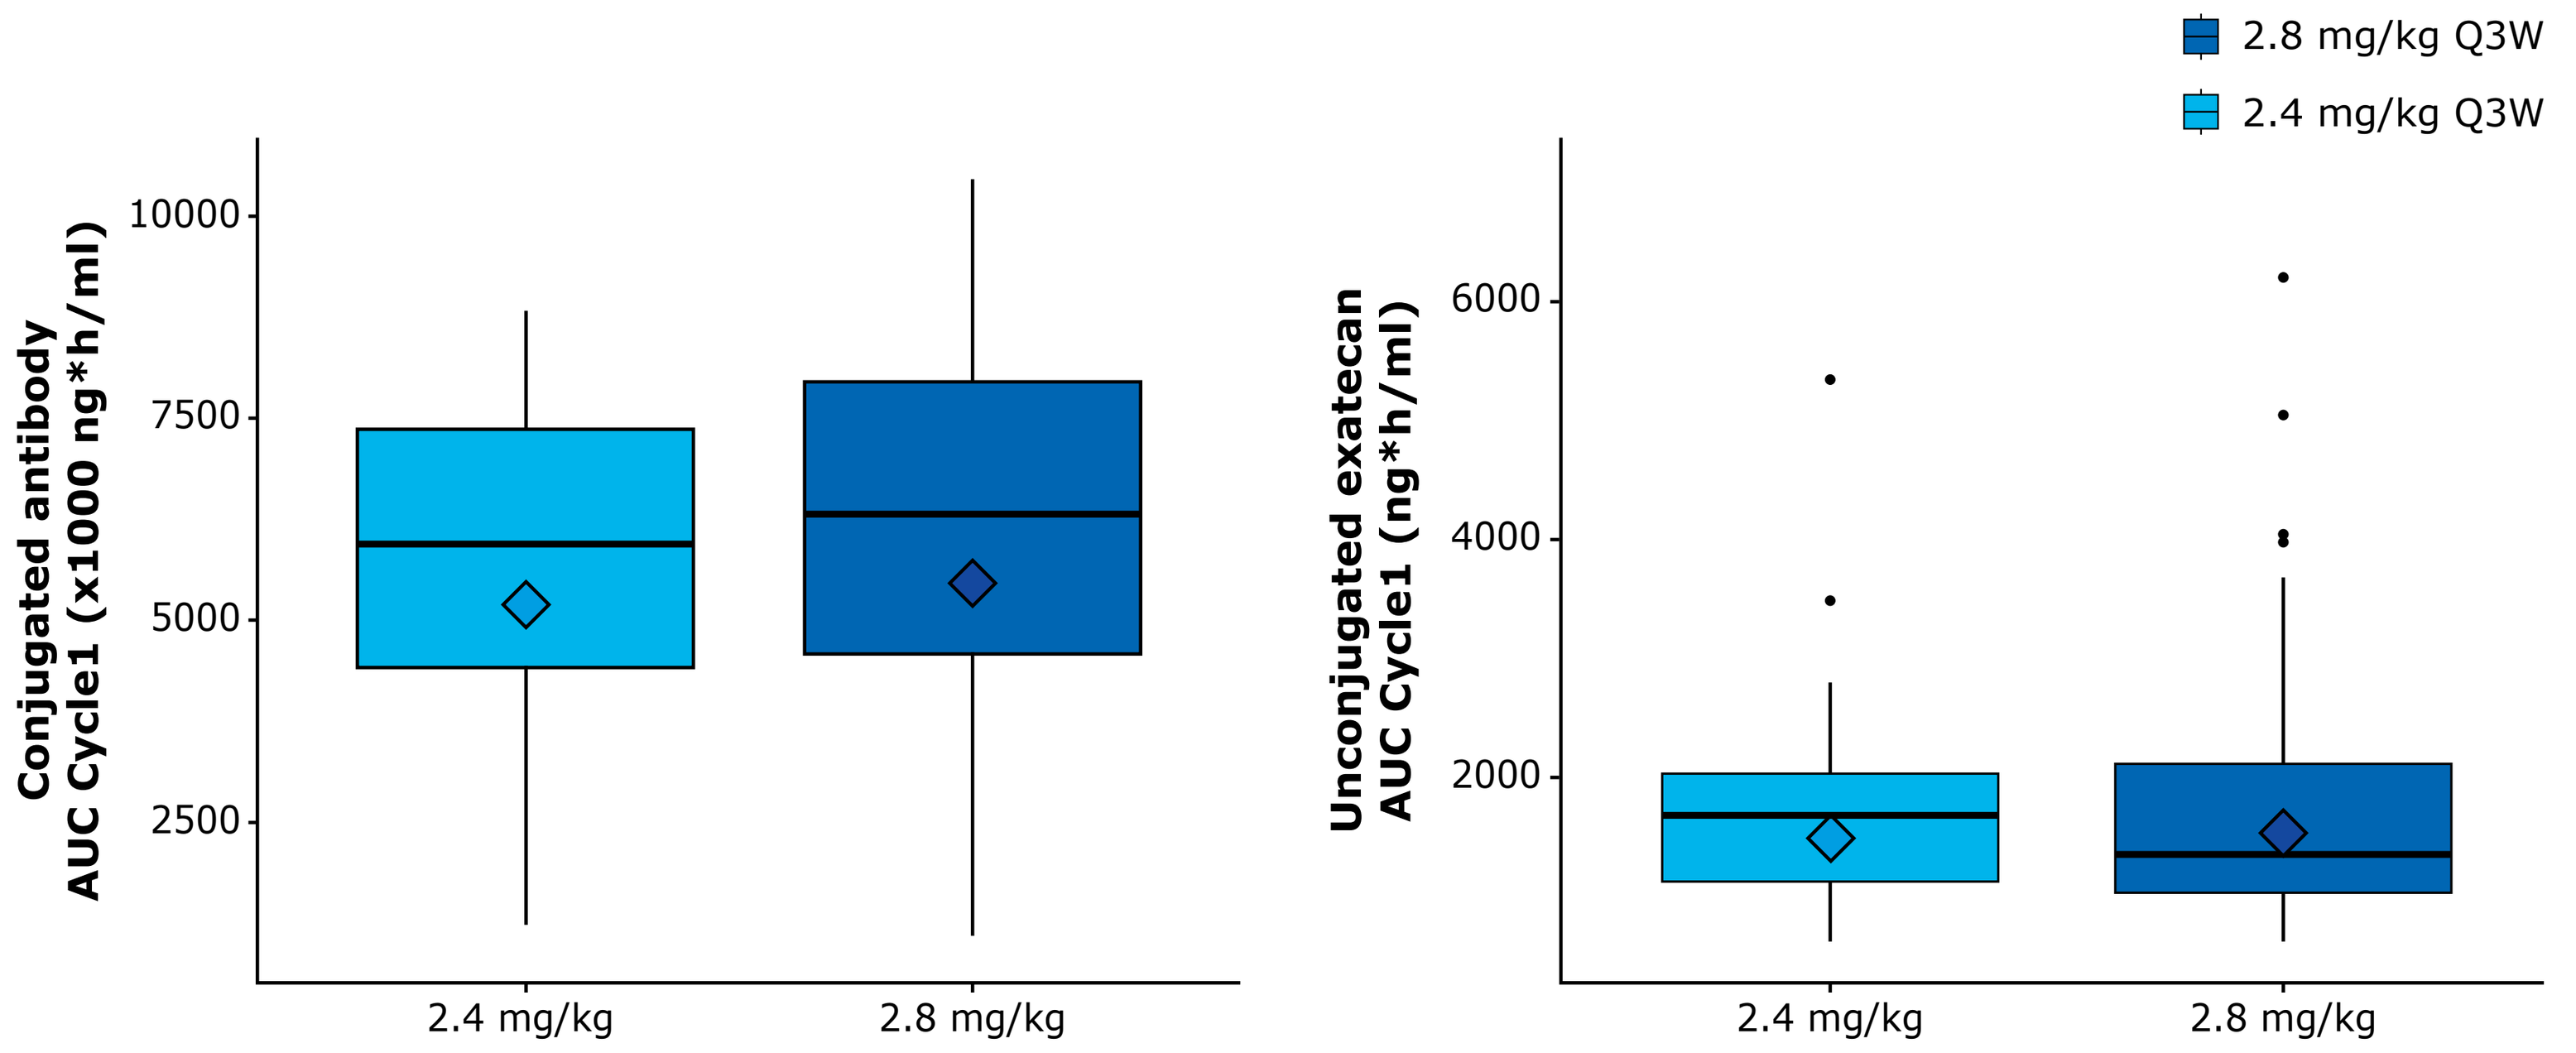
2.8 mg/kg Q3W
2.4 mg/kg Q3W
Conjugated antibody AUC Cycle1 (x1000 ng*h/ml)
10000
7500
5000
2500
2.4 mg/kg
2.8 mg/kg
Unconjugated exatecan AUC Cycle1 (ng*h/ml)
6000
4000
2000
2.4 mg/kg
2.8 mg/kg

Presenting author (skopetz@mdanderson.org)

Abbreviations: 3L+, third or later line of therapy; 5-FU, 5-fluorouracil; ADA, anti-drug antibody; ADC, antibody-drug conjugate; AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; AUC, area under curve; CEACAM5, carcinoembryogenic antigen-related cell adhesion molecule 5; CI, confidence interval; CR, complete response; CRC, colorectal cancer; DCR, disease control rate; DL, dose level; ECG, electrocardiogram; ECOG PS, Eastern Cooperative Oncology Group performance status; FiH, first-in-human; GGT, gamma-glutamyl transferase; GI, gastrointestinal; ICI, immune checkpoint inhibitor; ILD, interstitial lung disease; IV, intravenous; MoA, mechanism of action; la/m, locally advanced or metastatic; mCRC, metastatic colorectal cancer; MSI-H, microsatellite instability-high; NE, not estimable; NR, not reached; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetics; PR, partial response; PT, Preferred Term; Q3W, once every three weeks, RDE, recommended dose for expansion; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SD, stable disease; SMQ, Standardized MedDRA Query; SoC, standard of care; TEAE, treatment-emergent adverse event; TOP1i, topoisomerase 1 inhibitor; TRAE, treatment-related adverse event; WBC, white blood cell.

References: 1. Dasari A, et al. *Lancet.* 2023;402:41-53; 2. Weinberg BA, et al. *Clin Adv Hematol Oncol.* 2016;14:630-638; 3. Mayer RJ, et al. *N Engl J Med.* 2015;372:1909-1919; 4. Grothey A, et al. *Lancet.* 2013;381:303-312; 5. Prager GW, et al. *N Engl J Med.* 2023;388:1657-1667; 6. Beauchemin N & Arabzadeh A. *Cancer Metastasis Rev.* 2013;32(3-4):643-671; 7. Zhang X, et al. *J Int Med Res.* 2020;48(9):300060520959478; 8. Decary S, et al. *Clin Cancer Res.* 2020;26(24):6589-6599; 9. Raab-Westphal S, et al. *Cancer Res.* 2024;84(6_suppl):2362; 10. Boni V, et al. *Ann Oncol.* 2024;35(2_suppl):S428-S481; 11. Kopetz S, et al. *J Clin Oncol.* 2024;42(16_suppl):3000.

Acknowledgements: The authors would like to thank patients, investigators, co-investigators, and the study teams at each of the participating centers. The authors would like to acknowledge the contributions of Abdul Siddiqui, who at the time of the study was affiliated with Global Patient Safety at Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany. The trial was funded by the healthcare business of Merck (CrossRef Funder ID: 10.13039/100009945). Medical writing assistance was provided by Ramya Gupta of Merck Specialities Pvt. Ltd, Bangalore, India, an affiliate of |the healthcare business of Merck KGaA, Darmstadt, Germany.

Disclosures: SK reports stock and other ownership interests in Frontier Medicines, Iylon, Lutris, Navire, Xilis, consulting or advisory role in Abbvie, Accademia Nazionale Di Medicina (ACCMED), Amal Therapeutics, Amgen, AstraZeneca/MedImmune, AVEO, Bayer Health, Bicara Therapeutics, Black Diamond Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb/Medarex, Cardiff Oncology, Carina Biotech, CureTeq, Merck, Endeavor BioMedicines, Flame Biosciences, Foundation Medicine, Frontier Medicines, Genentech, Genomic Health, Gilead Sciences, GlaxoSmithKline, HalioDx, Harbinger Oncology, Inc., Holy Stone Healthcare, Inivata, Ipsen, Iylon, Jacobio, Jazz Pharmaceuticals, Johnson & Johnson/Janssen, Lilly, Lutris, MSD, Mirati Therapeutics, NeoGenomics Laboratories, Novartis, Numab, Ono Pharmaceutical, Pfizer, Redx Pharma, Repare Therapeutics, Replimune, Roche, Sanofi, Servier, Tachyon Therapeutics, Taiho Pharmaceutical, Takeda, Tempus, Xilis, Zentalis, and research funding from Amgen, Array BioPharma, Biocartis, Daiichi Sankyo, Merck, Genentech/Roche, Guardant Health, Lilly, MedImmune, Novartis, Sanofi.

Abstract #7056 | Presented at the European Society of Medical Oncology (ESMO) Congress 2025 | Oct 17–21, 2025 | Berlin, Germany