Cladribine Tablets Maintain or Improve Cognition Over 4 Years in People With Relapsing Multiple Sclerosis: Pooled Analyses of the CLARIFY-MS and MAGNIFY-MS Extension Studies

Bruno Brochet, Eva Kubala Havrdova, Krzysztof Selmaj, Klaus Schmierer, *Tahmidul Islam, Andrzej Smyk*, Nicola De Stefano

This work was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945).
Authors in italics are employees of EMD Serono and the healthcare business of Merck KGaA, Darmstadt, Germany, or its affiliates. Please scan the QR code below for full affiliations.

SCAN FOR POSTER PDF
For personal use only and may not be reproduced without written permission of the authors

RESEARCH IN CONTEXT

1. **Short-course CladT delivers sustained cognitive improvements over 4 years in PwRMS.**
2. **Greater cognitive improvements observed in younger PwRMS (aged <50 years), supporting early use of CladT for maximum benefit, complementing its beneficial effects on conventional clinical outcomes.**

OBJECTIVES

To assess the long-term effects of cladribine tablets (CladT) on cognitive function across a large population of people with relapsing multiple sclerosis (PwRMS).

INTRODUCTION

- Cognitive deterioration in PwRMS often begins early and worsens over time, with cognitive processing speed (CPS) frequently affected.[1]
- The Symbol Digit Modalities Test (SDMT) is a validated measure of CPS in multiple sclerosis (MS).[2]
- The CLARIFY-MS and MAGNIFY-MS parent studies and their extensions showed stable or improved cognitive function for up to 4 years following treatment with short-course CladT.[3,4]

METHODS

- 4-year pooled analysis from an integrated database of the 2-year CLARIFY-MS and MAGNIFY-MS parent studies and their respective 2-year treatment-free extension studies.

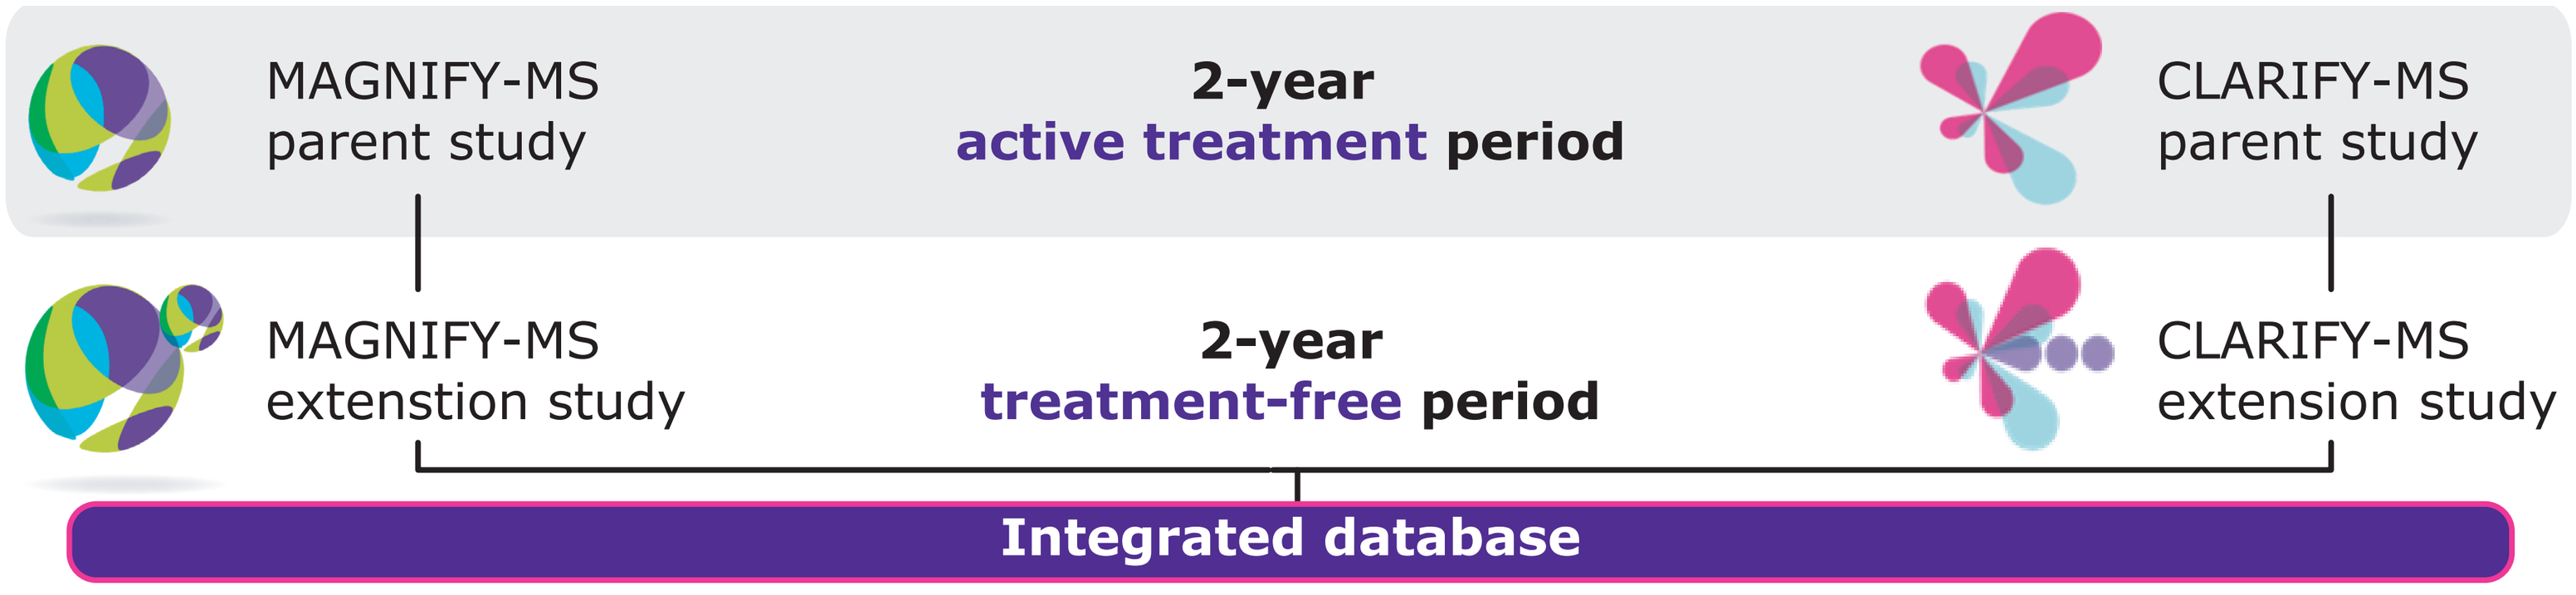
MAGNIFY-MS parent study
2-year active treatment period
CLARIFY-MS parent study
MAGNIFY-MS extension study
2-year treatment-free period
CLARIFY-MS extension study
Integrated database

- PwRMS received short-course CladT (3.5 mg/kg cumulative dose) during Years 1 and 2, followed by a treatment-free period during Years 3 and 4.

Assessment	Statistical methodology
Overall change in SDMT score from baseline[a] to Month 48	• Adjusted LS mean changes in SDMT scores with 95% CI • Assessed using MMRM adjusting for baseline score, age, EDSS (≤3 vs >3), and visit
4- and 8-point clinically meaningful improvements in SDMT scores from baseline[a] to Month 48	• Presented as number and proportion of PwRMS with improvement, stability, or worsening in SDMT scores
Subgroup analysis Age: <50 years vs ≥50 years Baseline SDMT scores[b]: Low (<53 points) vs High (≥53 points)	• Adjusted LS mean changes in SDMT scores using MMRM and clinically meaningful thresholds applied to all subgroups

[a]The baseline is defined as the last non-missing assessment prior to or at treatment start date. [b]Baseline SDMT subgroups were defined using the median score of 53 points as the cutoff point. 'Low' and 'High' refers to lower and higher than the median score (53 points) for the group.

CI, confidence interval; EDSS, Expanded Disability Status Scale; LS, least square; MMRM, Mixed Effect Model for Repeated Measures; SDMT, Symbol Digit Modalities Test.

RESULTS

- This analysis utilized 4-year data pooled from 499 PwRMS with active disease at baseline, participating in the CLARIFY-MS extension (n=280) and MAGNIFY-MS extension (n=219) studies.

Mean age: 37.9 years
Female: 68.7%
Median SDMT: 53 points

Key demographics and characteristics

Age, n (%)		Baseline SDMT[a], n (%)	
<50 years	434 (87.0)	Low (<53)	242 (48.9)
≥50 years	65 (13.0)	High (≥53)	253 (51.1)

Subgroup patient population

n, number of patients; SDMT, Symbol Digit Modalities Test. [a]Baseline SDMT data was missing for 4 PwRMS.

Refer to **Supplementary Tables 1** and **2** for demographics and baseline characteristics of the subgroups by age and baseline SDMT score.

Mean change from baseline in SDMT scores

- At Month 48, statistically significant improvements in LS mean SDMT scores were observed in the total population (+1.79 points; p<0.0001) and in PwRMS aged <50 years (+2.22 points; p<0.0001), while improvements in those aged ≥50 years (+0.81 points; p=0.7806) were not statistically significant (**Figure 1**).

Figure 1. Overall improvement in CPS at Month 48 (versus baseline) with greater benefits in younger PwRMS (<50 years)

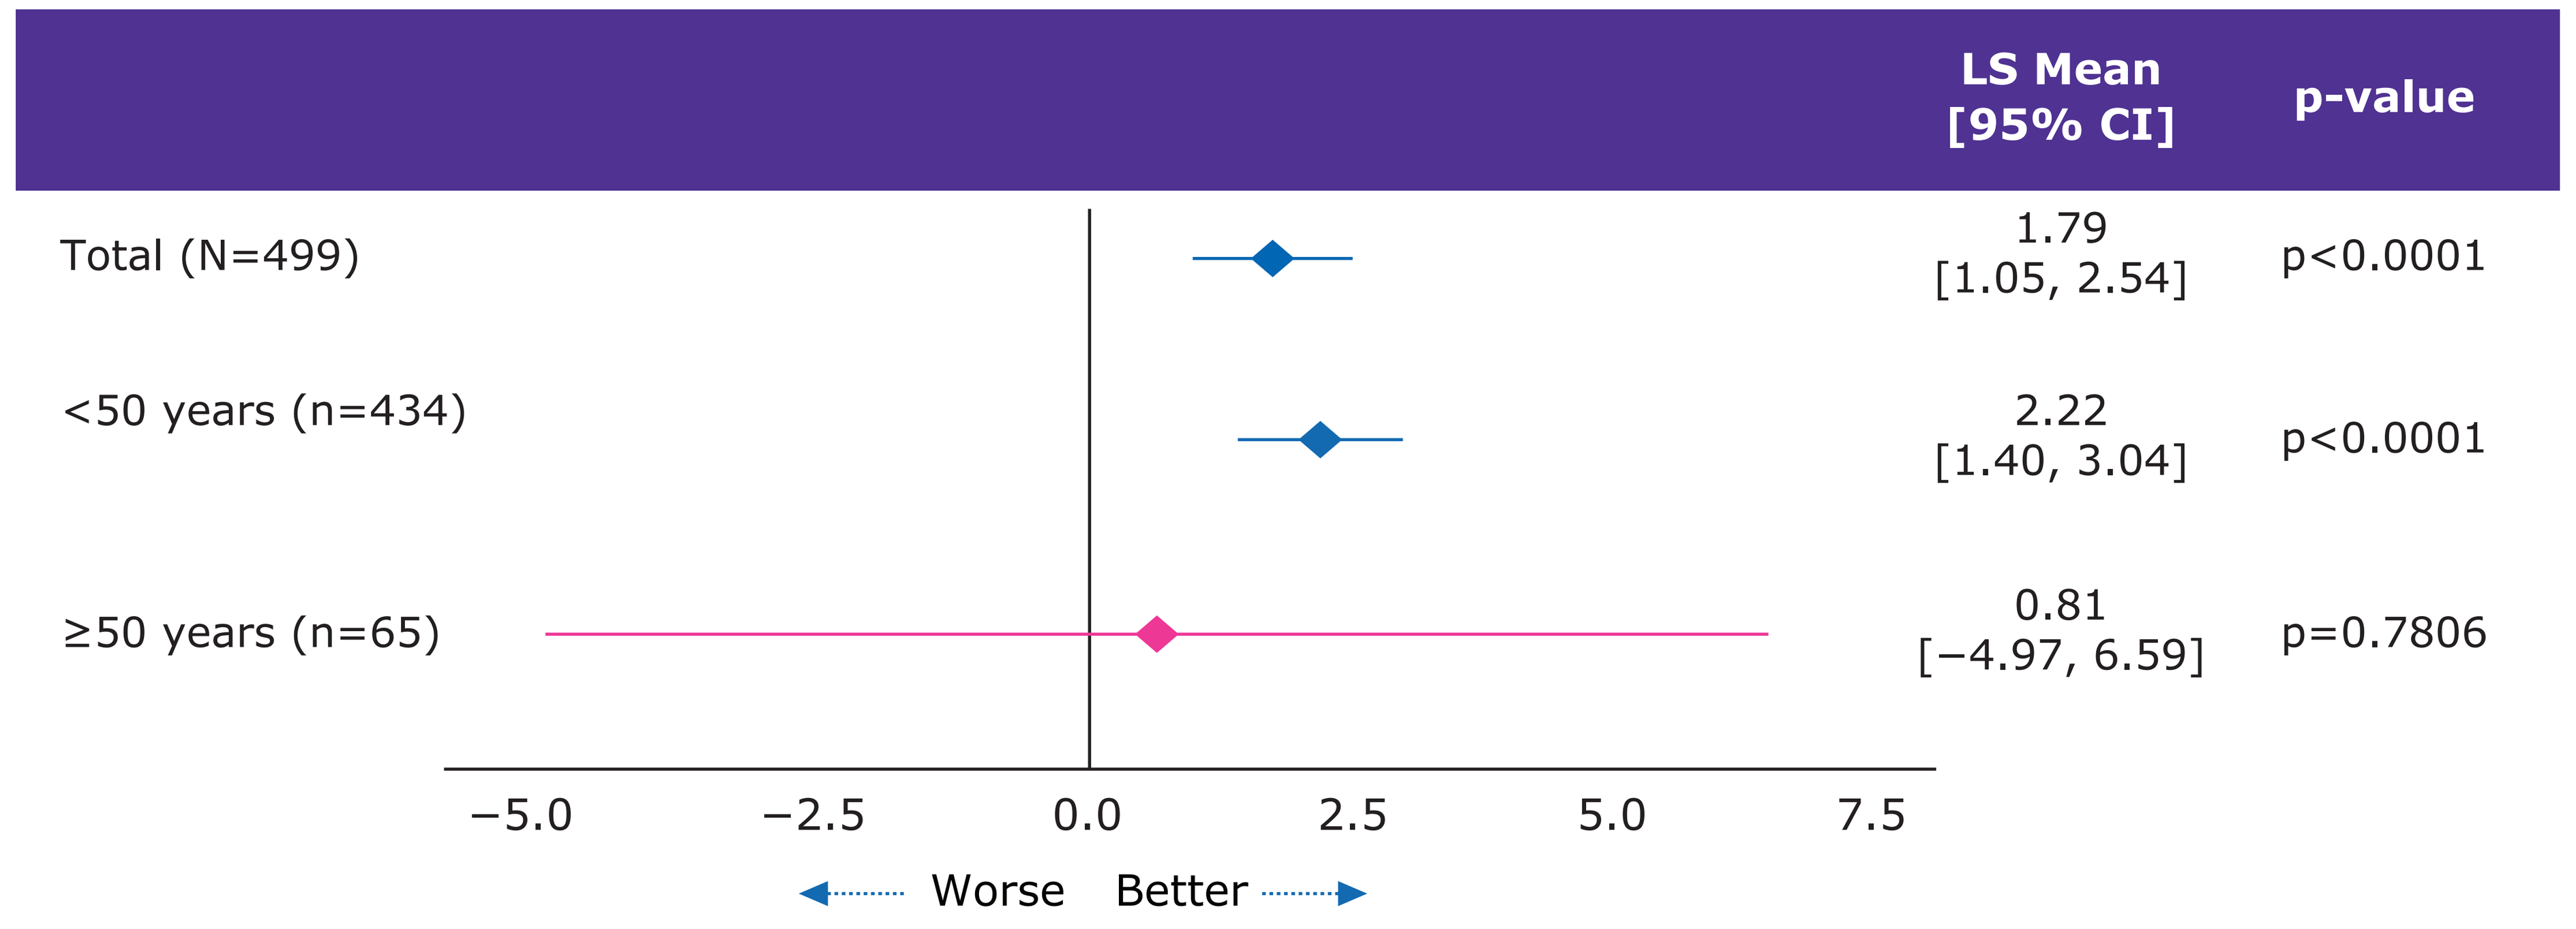
LS Mean [95% CI]
p-value
Total (N=499)
1.79 [1.05, 2.54]
p<0.0001
<50 years (n=434)
2.22 [1.40, 3.04]
p<0.0001
≥50 years (n=65)
0.81 [−4.97, 6.59]
p=0.7806
−5.0
−2.5
0.0
2.5
5.0
7.5
Worse
Better

CI, confidence interval; CPS, cognitive processing speed; LS, least square; n, number of patients in the subgroup; N, total number of patients; PwRMS, people with relapsing multiple sclerosis.

SDMT scores at Month 48 using clinically meaningful ≥8- and ≥4-point thresholds

- At Month 48, most PwRMS had stable or improved SDMT scores versus baseline (**Figure 2**)
 - ≥4-point change: 74.4% stable/improved, 25.2% worsened
 - ≥8-point change: 86.6% stable/improved, 12.9% worsened
- Higher proportions of stability/improvement were observed in younger (<50 years) versus older (≥50 years) PwRMS and in those with low (<53) versus high (≥53) baseline SDMT scores (**Figure 2**)
 - <50 vs ≥50 years: ≥4-point change: 75.3% vs 68.3%; ≥8-point change: 87.4% vs 81.6%
 - Low vs high SDMT: ≥4-point change: 80.1% vs 69.6%; ≥8-point change: 89.6% vs 84.6%

Figure 2. Cognitive processing speed remained stable or improved in most PwRMS through Month 48

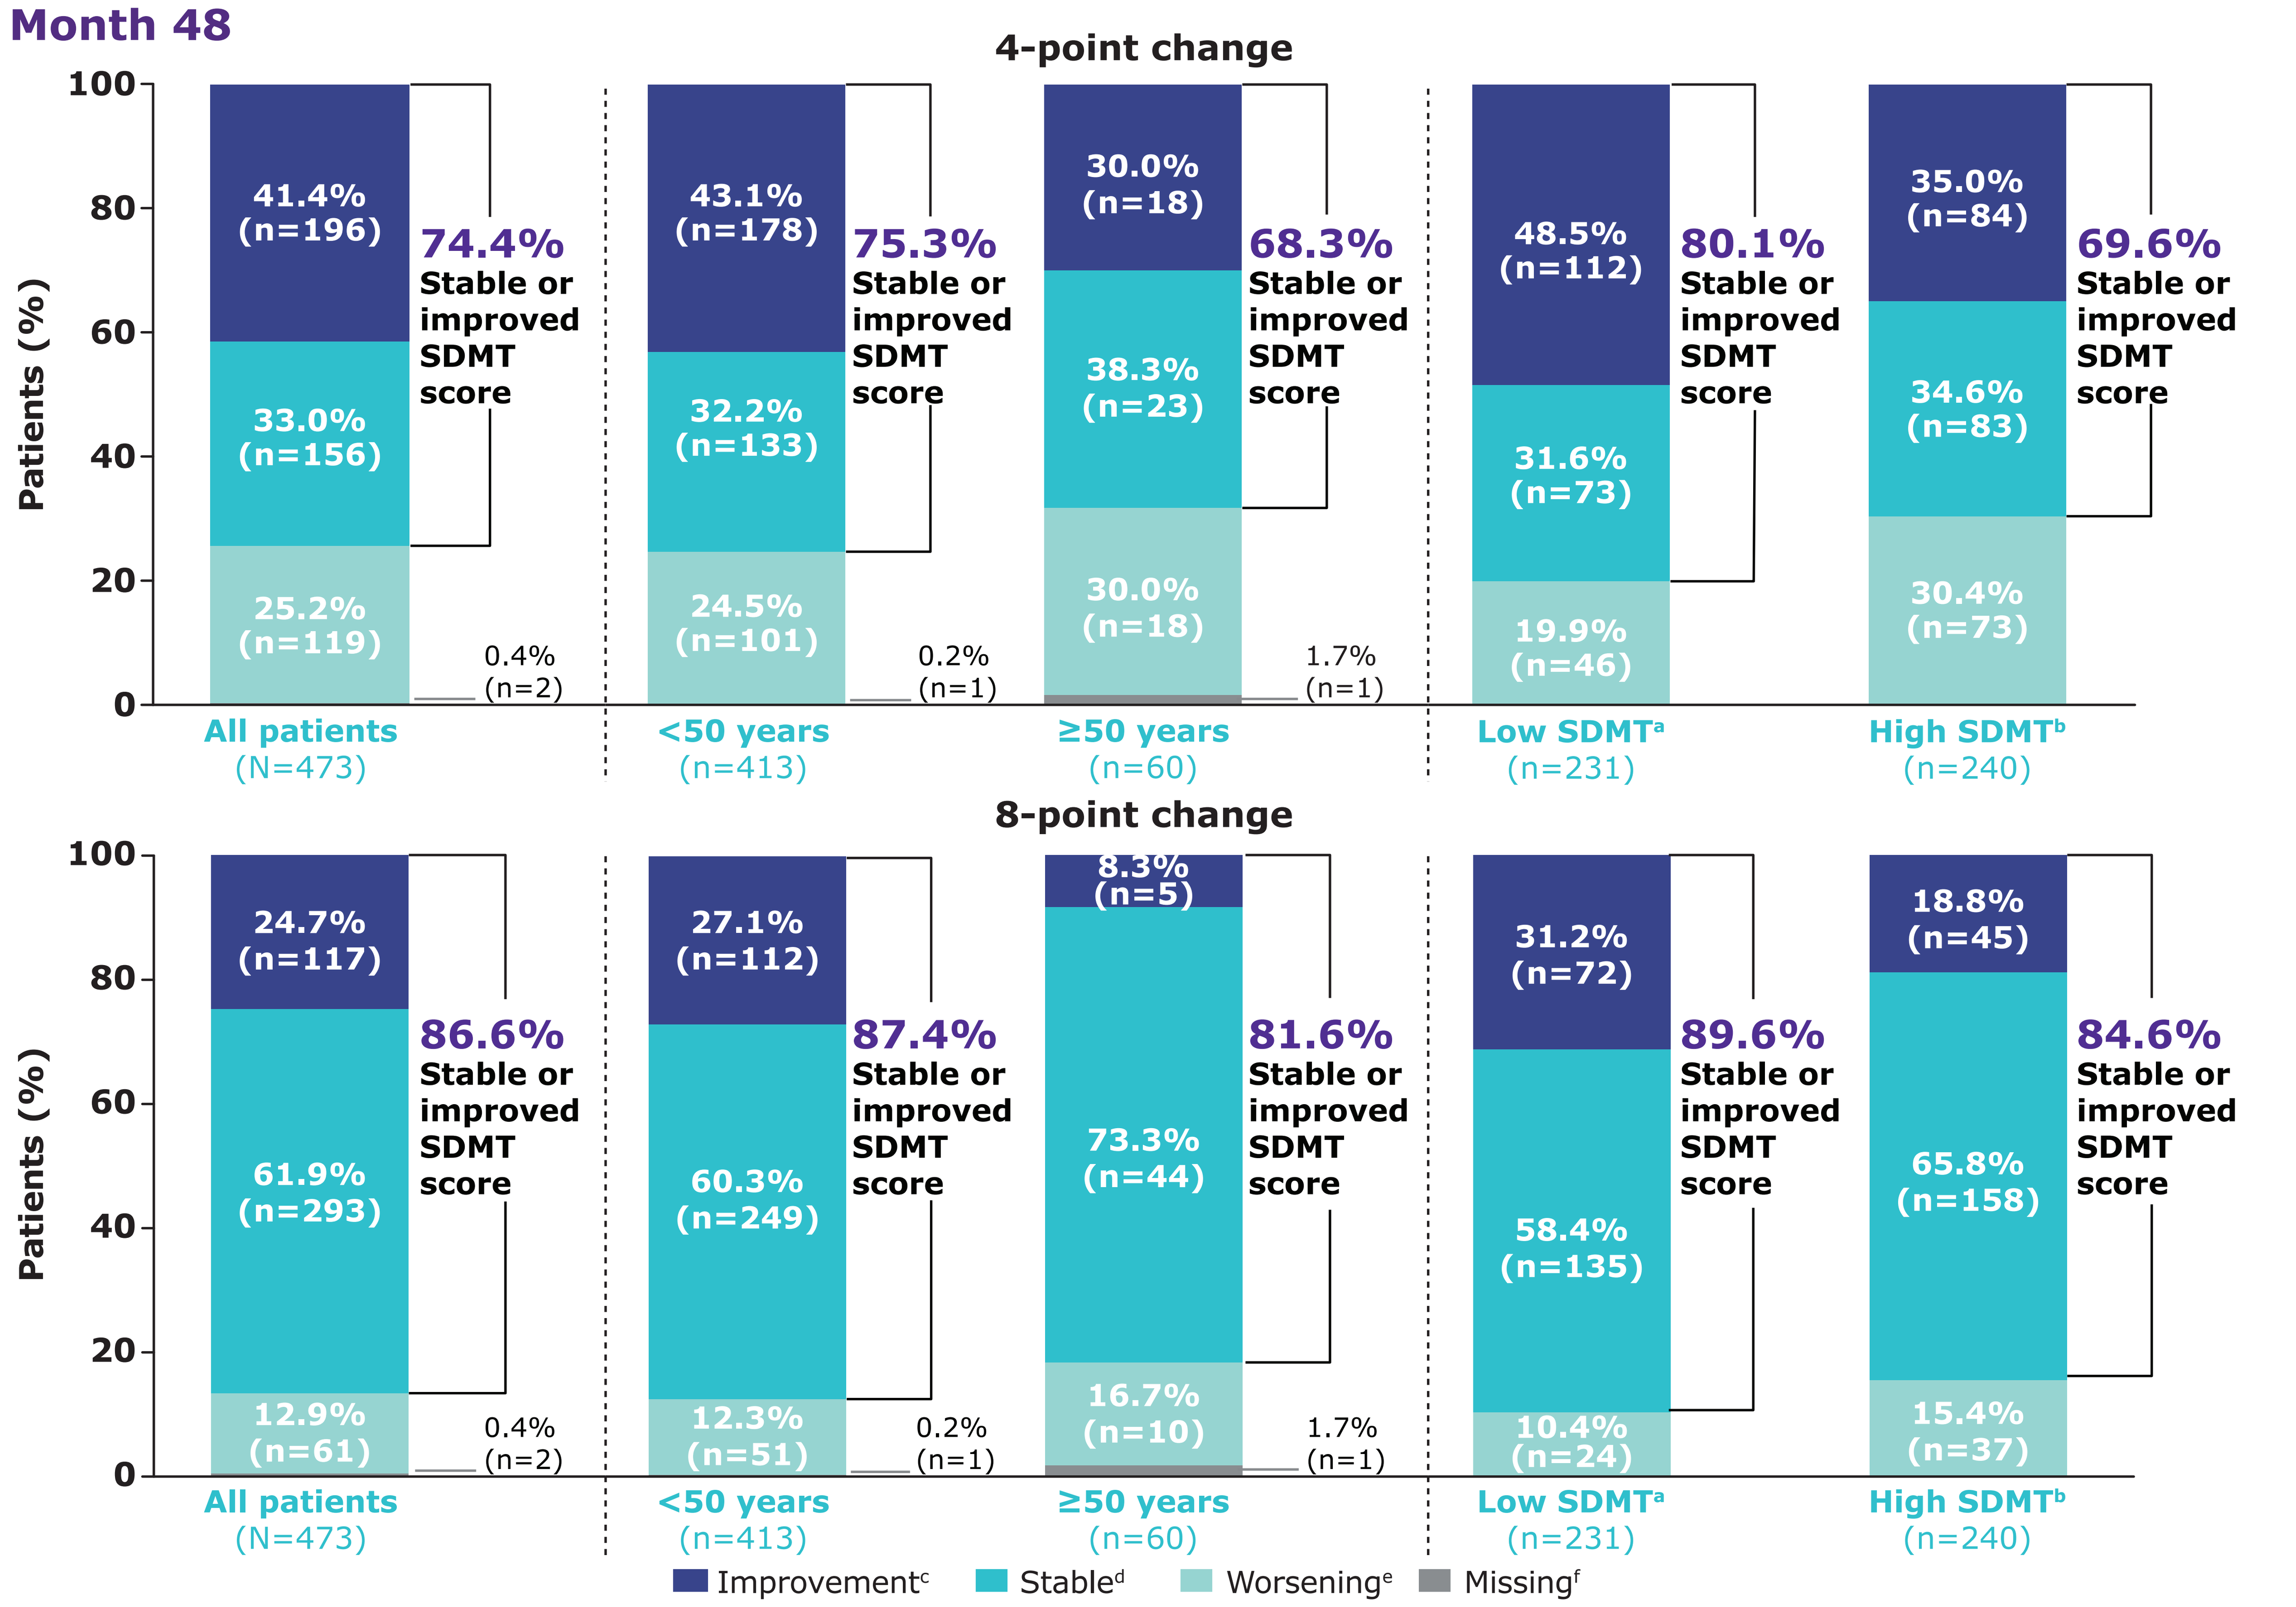
4-point change
Patients (%)
All patients (N=473): 41.4% (n=196); 33.0% (n=156); 25.2% (n=119); 0.4% (n=2); 74.4% Stable or improved SDMT score
<50 years (n=413): 43.1% (n=178); 32.2% (n=133); 24.5% (n=101); 0.2% (n=1); 75.3% Stable or improved SDMT score
≥50 years (n=60): 30.0% (n=18); 38.3% (n=23); 30.0% (n=18); 1.7% (n=1); 68.3% Stable or improved SDMT score
Low SDMT[a] (n=231): 48.5% (n=112); 31.6% (n=73); 19.9% (n=46); 80.1% Stable or improved SDMT score
High SDMT[b] (n=240): 35.0% (n=84); 34.6% (n=83); 30.4% (n=73); 69.6% Stable or improved SDMT score
8-point change
All patients (N=473): 24.7% (n=117); 61.9% (n=293); 12.9% (n=61); 0.4% (n=2); 86.6% Stable or improved SDMT score
<50 years (n=413): 27.1% (n=112); 60.3% (n=249); 12.3% (n=51); 0.2% (n=1); 87.4% Stable or improved SDMT score
≥50 years (n=60): 8.3% (n=5); 73.3% (n=44); 16.7% (n=10); 1.7% (n=1); 81.6% Stable or improved SDMT score
Low SDMT[a] (n=231): 31.2% (n=72); 58.4% (n=135); 10.4% (n=24); 89.6% Stable or improved SDMT score
High SDMT[b] (n=240): 18.8% (n=45); 65.8% (n=158); 15.4% (n=37); 84.6% Stable or improved SDMT score
Improvement[c]
Stable[d]
Worsening[e]
Missing[f]

Data shown for PwRMS with available 48-month visit data. [a]Low SDMT score: SDMT at baseline <53 points (median); [b]High SDMT score: SDMT at baseline ≥53 points (median); [c]Improvement: change from baseline in SDMT score of ≥4 or ≥8 points at the Month 48 visit; [d]Stable: >−4 or >−8 point change from baseline in SDMT at the Month 48 visit <4 or <8 points; [e]Worsening: change from baseline in SDMT score of ≤−4 or ≤−8 points at the Month 48 visit; [f]Missing: number of PwRMS for whom the visit is performed but the SDMT assessment is missing. Percentage is based on the number of PwRMS for whom the visit is performed which includes PwRMS with missing SDMT data. SDMT data were missing at Month 48 vs baseline for 0.4% (n=2) of PwRMS.

CPS, cognitive processing speed; n, number of patients in the subgroup; N, total number of patients; PwRMS, people with relapsing multiple sclerosis; SDMT, Symbol Digit Modalities Test

CONCLUSIONS

- **Overall, most PwRMS treated with CladT either improved or had stable cognitive function.**
- **Clinically meaningful improvements or stability in CPS were observed over 4 years, particularly in younger PwRMS (<50 years) and those with baseline SDMT score below 53 points.**
- **These findings suggest long-term benefits of CladT beyond its effects on conventional disability indices, especially when initiated earlier in the disease course.**

SCAN FOR AFFILIATIONS, DISCLOSURES, AND SUPPLEMENTARY MATERIALS

References: 1. Benedict RH, et al. Lancet Neurol. 2020;19(10):860-871.
2. Benedict RH, et al. Mult Scler. 2017;23(5):721-733.
3. Selmaj K, et al. ECTRIMS 2024 [P846].
4. De Stefano N, et al. ECTRIMS 2024 [P349].

For Reactive Medical Use Only

Presented at ACTRIMS Forum 2026 | February 5–7 | San Diego, California

SUPPLEMENTARY MATERIAL

Author Affiliations

Bruno Brochet[1], Eva Kubala Havrdova[2], Krzysztof Selmaj[3,4], Klaus Schmierer[5,6], Tahmidul Islam[7], Andrzej Smyk[8], Nicola De Stefano[9]

[1]INSERM U1215, University of Bordeaux, Bordeaux, France; [2]Charles University, First Medical Faculty, Prague, Czech Republic; [3]Center for Neurology, Lódź, Poland; [4]University of Warmia and Mazury, Olsztyn, Poland; [5]The Blizard Institute, Centre for Neuroscience, Surgery and Trauma, Faculty of Medicine and Dentistry, Queen Mary University of London, London, UK; [6]Clinical Board Medicine (Neuroscience), The Royal London Hospital, Barts Health NHS Trust, London, UK; [7]EMD Serono, Billerica, MA, USA; [8]The healthcare business of Merck KGaA, Darmstadt, Germany; [9]Department of Medicine, Surgery and Neuroscience, University of Siena, Siena, Italy

Author Disclosures

Bruno Brochet has received consultancy fees, speaker fees, research grants (non-personal), or honoraria from the healthcare business of Merck KGaA, Darmstadt, Germany, and Sanofi.

Eva Kubala Havrdova has received honoraria/research support from Actelion (Janssen/J&J), Biogen, Celgene (BMS), the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva; has served on advisory boards for Actelion (Janssen/J&J), Biogen, Celgene (BMS), the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi; and has been supported by the Czech Ministry of Education (project Cooperatio LF1, research area Neuroscience, and the project National Institute for Neurological Research (Programme EXCELES, ID Project No. LX22NPO5107), funded by the European Union-Next Generation EU.

Krzysztof Selmaj has received honoraria for speaking, consulting, and serving on advisory boards for Biogen, Celgene (BMS), the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and TG Therapeutics.

Klaus Schmierer has received research support from the National Institute for Health and Care Research, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sandoz, and Sanofi; speaking honoraria from, and/or served in an advisory role for, Biogen, EMD Serono, the healthcare business of Merck KGaA, Darmstadt, Germany, Neuraxpharm, Novartis, Roche, and Sanofi; and remuneration for teaching activities from AcadeMe and Medscape.

Tahmidul Islam is an employee of EMD Serono, Billerica, MA, USA.

Andrzej Smyk is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany.

Nicola De Stefano has received honoraria from Biogen Idec, Bristol Myers Squibb, Celgene, Genzyme, Immunic, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Teva for consulting services, speaking, and travel support. He serves on advisory boards for Biogen-Idec, Immunic, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi. He has received research support from the Italian Ministry of University and Research and Fondazione Italiana Sclerosi Multipla. He is the Associate Editor of Neurological Sciences and co-founder of Siena-Imaging S.r.l.

Acknowledgements

This work was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Saimithra Thammera and and Pritorthi Bhattacharjee of Merck Specialties Pvt. Ltd., Bengaluru, India, an affiliate of the healthcare business of Merck KGaA, Darmstadt, Germany.

SUPPLEMENTARY MATERIAL

Supplementary Table 1. Demographics and Baseline Characteristics - Subgroup by Age (<50 years vs ≥50 years)

Parameters	<50 years (n=434)	≥50 years (n=65)	Total (N=499)
Age (years)			
Mean±SD	35.5±8.04	54.5±4.44	37.9±9.99
Median (Q1, Q3)	36.0 (29.0, 42.0)	53.0 (51.0, 56.0)	38.0 (30.0, 45.0)
Female, n (%)	299 (68.9)	44 (67.7)	343 (68.7)
Race, n (%)			
White	331 (76.3)	48 (73.8)	379 (76.0)
Black or Africa American	1 (0.2)	0	1 (0.2)
Asian	2 (0.5)	0	2 (0.4)
Other	3 (0.7)	3 (4.6)	6 (1.2)
Not collected at this site	97 (22.4)	14 (21.5)	111 (22.2)
Ethnicity, n (%)			
Hispanic	16 (3.7)	5 (7.7)	21 (4.2)
Non-Hispanic	353 (81.3)	50 (76.9)	403 (80.8)
Not reported	65 (15.0)	10 (15.4)	75 (15.0)

n, number of patients in the subgroup; N, total number of patients; Q, quartile; SD, standard deviation.

Supplementary Table 2. Demographics and Baseline Characteristics - Subgroup by Baseline SDMT score (Low vs High)[a]

Parameters	Low SDMT score (n=242)	High SDMT score (n=253)	Total (N=499)
Age (years)			
Mean±SD	40.9±9.93	34.9±9.11	37.9±9.99
Median (Q1, Q3)	42.0 (34.0, 47.0)	34.0 (27.0, 42.0)	38.0 (30.0, 45.0)
Female, n (%)	153 (63.2)	187 (73.9)	343 (68.7)
Race, n (%)			
White	197 (81.4)	178 (70.4)	379 (76.0)
Black or Africa American	0	1 (0.4)	1 (0.2)
Asian	0	2 (0.8)	2 (0.4)
Other	5 (2.1)	1 (0.4)	6 (1.2)
Not collected at this site	40 (16.5)	71 (28.1)	111 (22.2)
Ethnicity, n (%)			
Hispanic	10 (4.1)	11 (4.3)	21 (4.2)
Non-Hispanic	201 (83.1)	198 (78.3)	403 (80.8)
Not reported	31 (12.8)	44 (17.4)	75 (15.0)

[a]Baseline SDMT subgroups were defined using the median score of 53 points as the cutoff point. ‘Low’ and ‘High’ refers to lower and higher than the median score (53 points) for the group.
n, number of patients in the subgroup; N, total number of patients; Q, quartile; SD, standard deviation.