Effectiveness and Safety of Switching to Cladribine Tablets in US Patients With RMS Experiencing High Disease Activity or Disease Breakthrough Despite Current DMT Use

Barry Hendin, Jeffrey Kaplan, Sam Hooshmand, Angel Chinea, MaryAnn Picone, Daniel Bandari, *Julie Korich, Julie Aldridge,* Ralf Koelbach, *Christina Caon*, Regina Berkovich

This work was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Authors in italics are employees of EMD Serono or its affiliates. Please scan the QR code below for full affiliations.

SCAN FOR POSTER PDF

For personal use only and may not be reproduced without written permission of the authors

RESEARCH IN CONTEXT

These real-world data support the safety and effectiveness of cladribine tablets for managing MS disease activity over 2 years in patients with relapsing MS following high-disease activity or disease breakthrough on other high-efficacy therapies.

OBJECTIVES

To evaluate the real-world effectiveness and safety of switching to cladribine tablets (CladT) in a United States (US) cohort of patients with relapsing multiple sclerosis (RMS) experiencing high disease activity (HDA) or disease breakthrough despite ongoing treatment with other disease-modifying therapies (DMTs).

INTRODUCTION

Despite the availability of multiple DMTs, some patients with RMS (PwRMS) continue to experience disease progression while on treatment, requiring a switch to a more efficacious DMT.[1]

CladT, a short-course high-efficacy therapy, are approved in the US for RMS.[2] Existing evidence from the CladT development program and phase 4 studies have demonstrated its efficacy and safety in PwRMS.[3-7] However, real-world evidence for PwRMS in the US switching to CladT after experiencing HDA or disease breakthrough despite ongoing treatment with high-efficacy therapy is limited.

METHODS

- This real–world, retrospective and prospective, single-cohort study was conducted across 17 US centers (**Supplementary Figure 1**), and enrolled 61 PwRMS who switched to CladT (on/before January 31, 2022) from moderate- or high-efficacy DMTs due to HDA/ disease breakthrough.
- HDA was defined as any relapses, magnetic resonance imaging (MRI) activity (T1 gadolinium-enhancing [Gd+] and/or new/enlarging T2 lesions), and/or disability progression ≤12 months before CladT initiation.

Outcome Type	Outcomes[b]	Statistics
Primary outcome[a]	Annualized relapse rate (ARR) at Month 24	Descriptive
Secondary outcomes[a]	ARR at Month 12 At Months 12 and 24: PwRMS free from MRI activity, modified Expanded Disability Status Scale (mEDSS) scores Safety assessments and lymphocyte dynamics over 24 months	

[a]After index date (date of CladT initiation). [b]Data on relapses, MRI activity, and mEDSS was obtained through routine collection.

RESULTS

Figure 1. Baseline demographics and disease characteristics (N=61)

Age
median (IQR) **48.0** (42.0-56.0) years
<50 years, **55.7%** (n=34)
≥50 years, **44.3%** (n=27)

Female
72.1% (n=44)

MS Type
RRMS **83.6%** (n=51)
aSPMS **16.4%** (n=10)

Duration from MS diagnosis to CladT initiation
Mean (SD) **12.35** (9.3) years

aSPMS, active secondary progressive multiple sclerosis; CladT, cladribine tablets; IQR, interquartile range; MS, multiple sclerosis; RRMS, relapsing-remitting MS; SD, standard deviation.

Please refer **Supplementary Table 1** for detailed baseline characteristics.

Figure 2. The majority of PwRMS switched from monoclonal antibodies to CladT

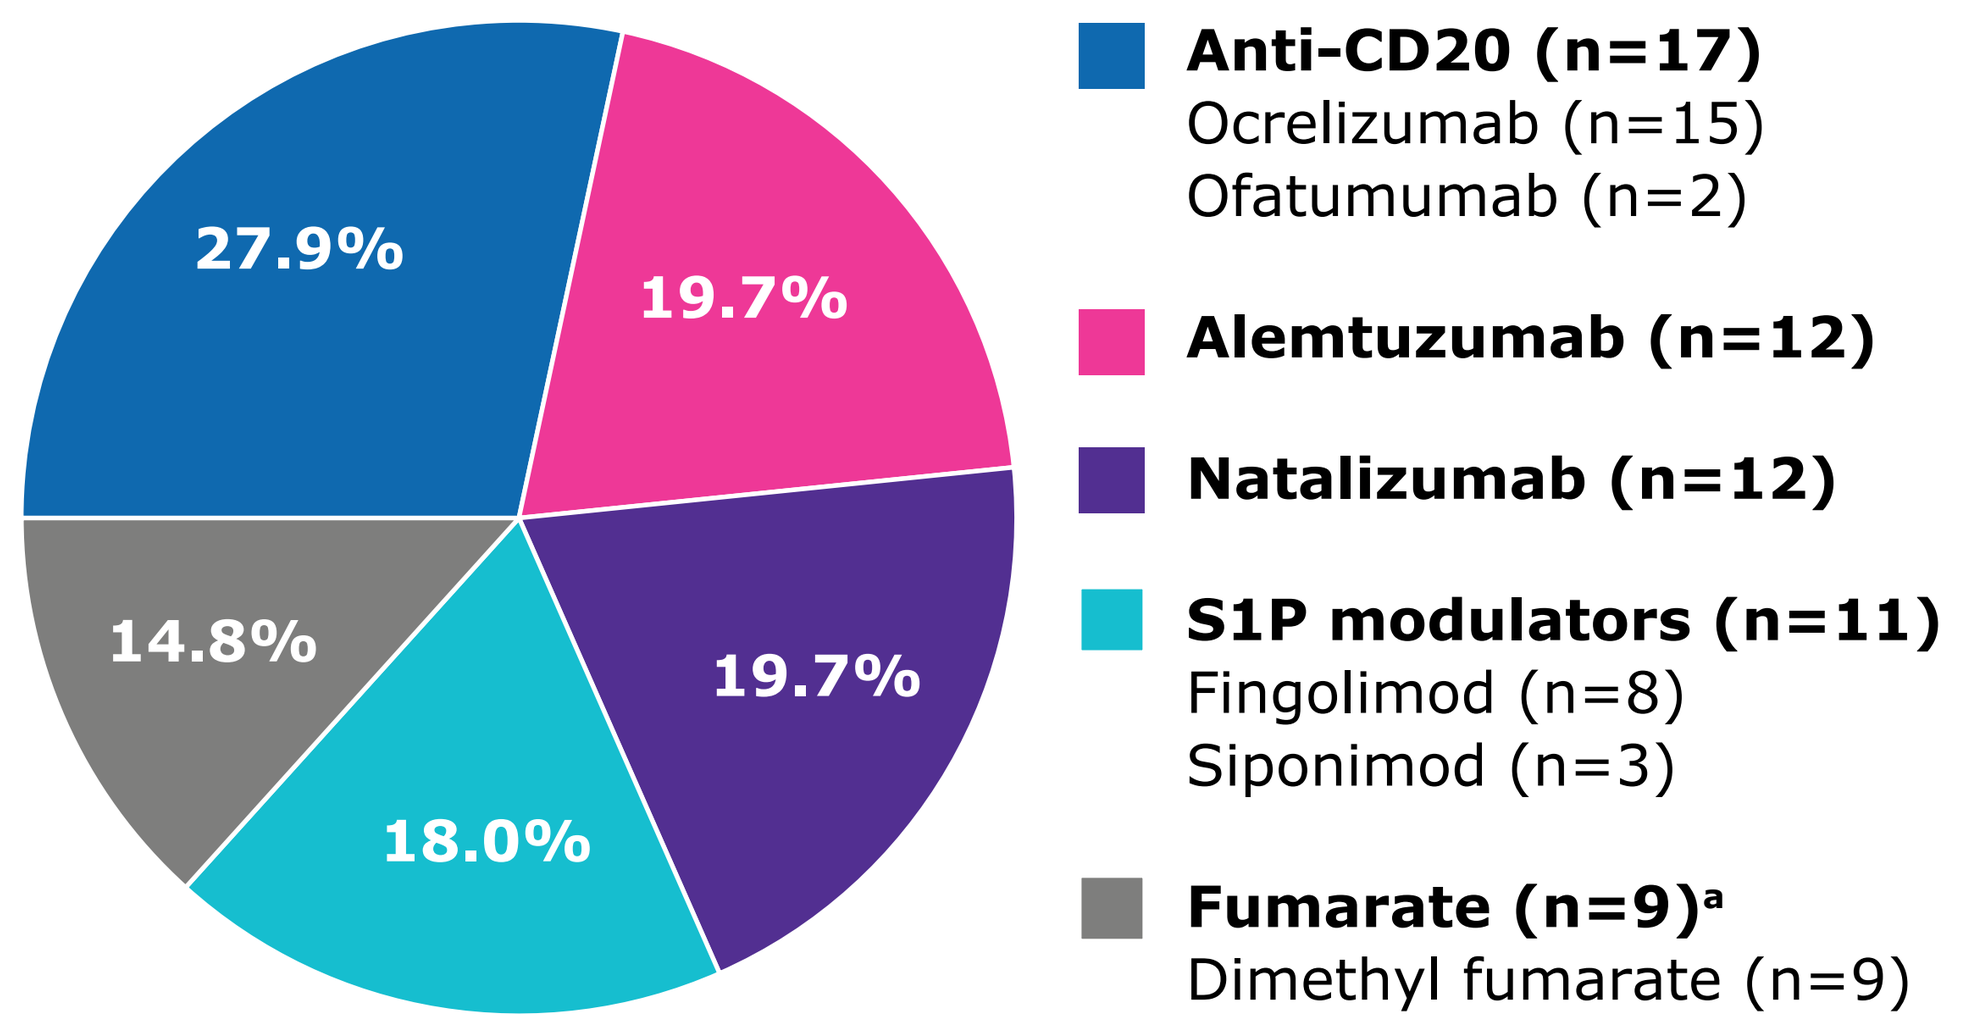
27.9%
19.7%
14.8%
19.7%
18.0%
Anti-CD20 (n=17)
Ocrelizumab (n=15)
Ofatumumab (n=2)
Alemtuzumab (n=12)
Natalizumab (n=12)
S1P modulators (n=11)
Fingolimod (n=8)
Siponimod (n=3)
Fumarate (n=9)a
Dimethyl fumarate (n=9)

Number of prior DMTs
Median (IQR), **3.0 (1.0–5.0)**

Switched to CladT due to inadequate response to previous therapy
96.7% (n=59)

[a]n=1 received dimethyl fumarate in the year before starting CladT but switched to glatiramer acetate for 2 months due to disease activity, ultimately discontinued glatiramer acetate and switched to CladT.

Anti-CD20, anti-cluster of differentiation 20; CladT, cladribine tablets; DMT, disease-modifying therapy; IQR, interquartile range; MS, multiple sclerosis; S1P, sphingosine 1-phosphate; PwRMS, patients with relapsing multiple sclerosis.

Figure 3. ARR was low at Month 24 after switching to CladT

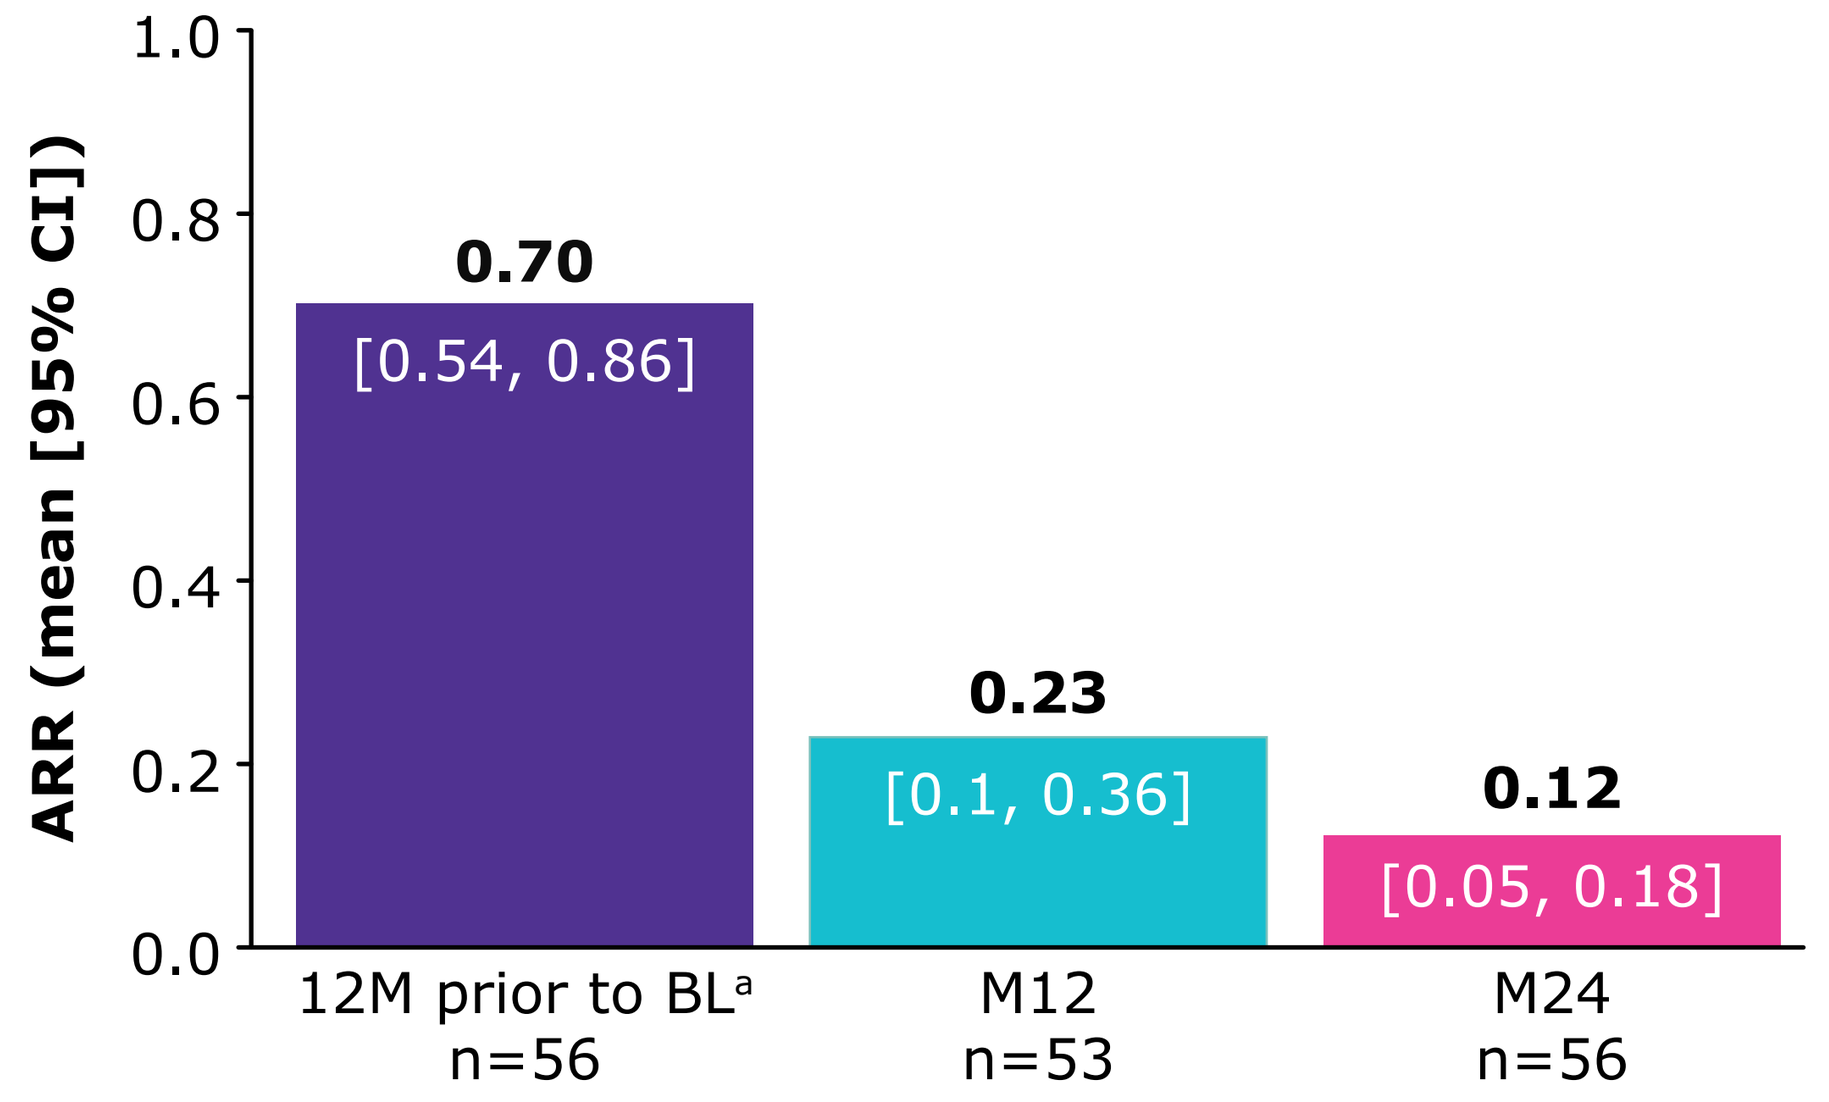
ARR (mean [95% CI])
1.0
0.8
0.6
0.4
0.2
0.0
0.70
[0.54, 0.86]
0.23
[0.1, 0.36]
0.12
[0.05, 0.18]
12M prior to BLa
n=56
M12
n=53
M24
n=56

[a]Baseline ARR was calculated based on the number of relapses in the 12 months prior to CladT initiation.

ARR, annualized relapse rate; BL, baseline; CI, confidence interval; CladT, cladribine tablets; M, month.

Figure 4. Most PwRMS were relapse-free at Months 12 and 24 after switching to CladT[a]

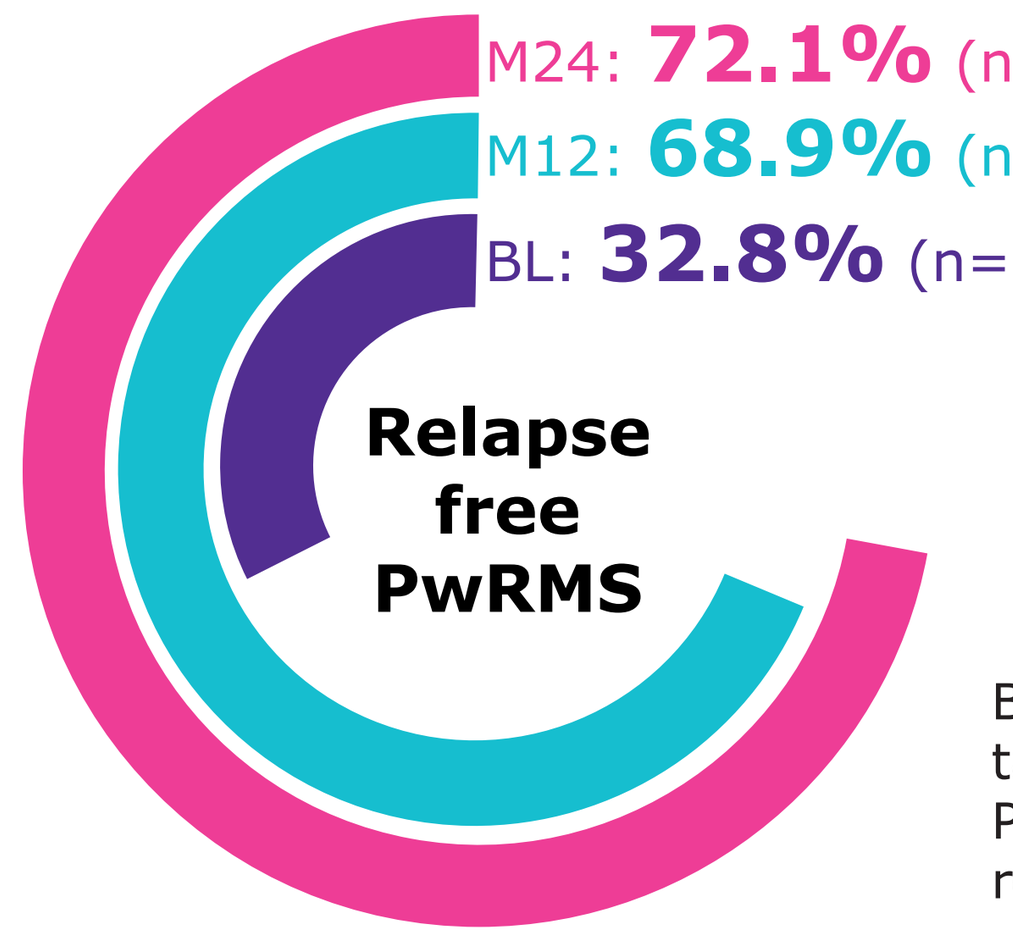
M24: 72.1% (n=44)
M12: 68.9% (n=42)
BL: 32.8% (n=20)
Relapse free PwRMS

BL, baseline; CladT, cladribine tablets; M, month; PwRMS, patients with relapsing multiple sclerosis.

[a] N=61; At BL, 5 (8.2%) PwRMS had missing data, and 36 (59.0%) experienced ≥ 1 relapse. At M12, 8 (13.1%) PwRMS had missing data, with 11 (18.0%) having ≥ 1 relapse. At M24, 5 (8.2%) PwRMS had missing data, and 12 (19.7%) had ≥ 1 relapse.

Please refer **Supplementary Table 2** for details on relapse counts.

Figure 5. Among PwRMS with available MRI scans, the majority were free from MRI activity at Months 12 and 24 after switching to CladT

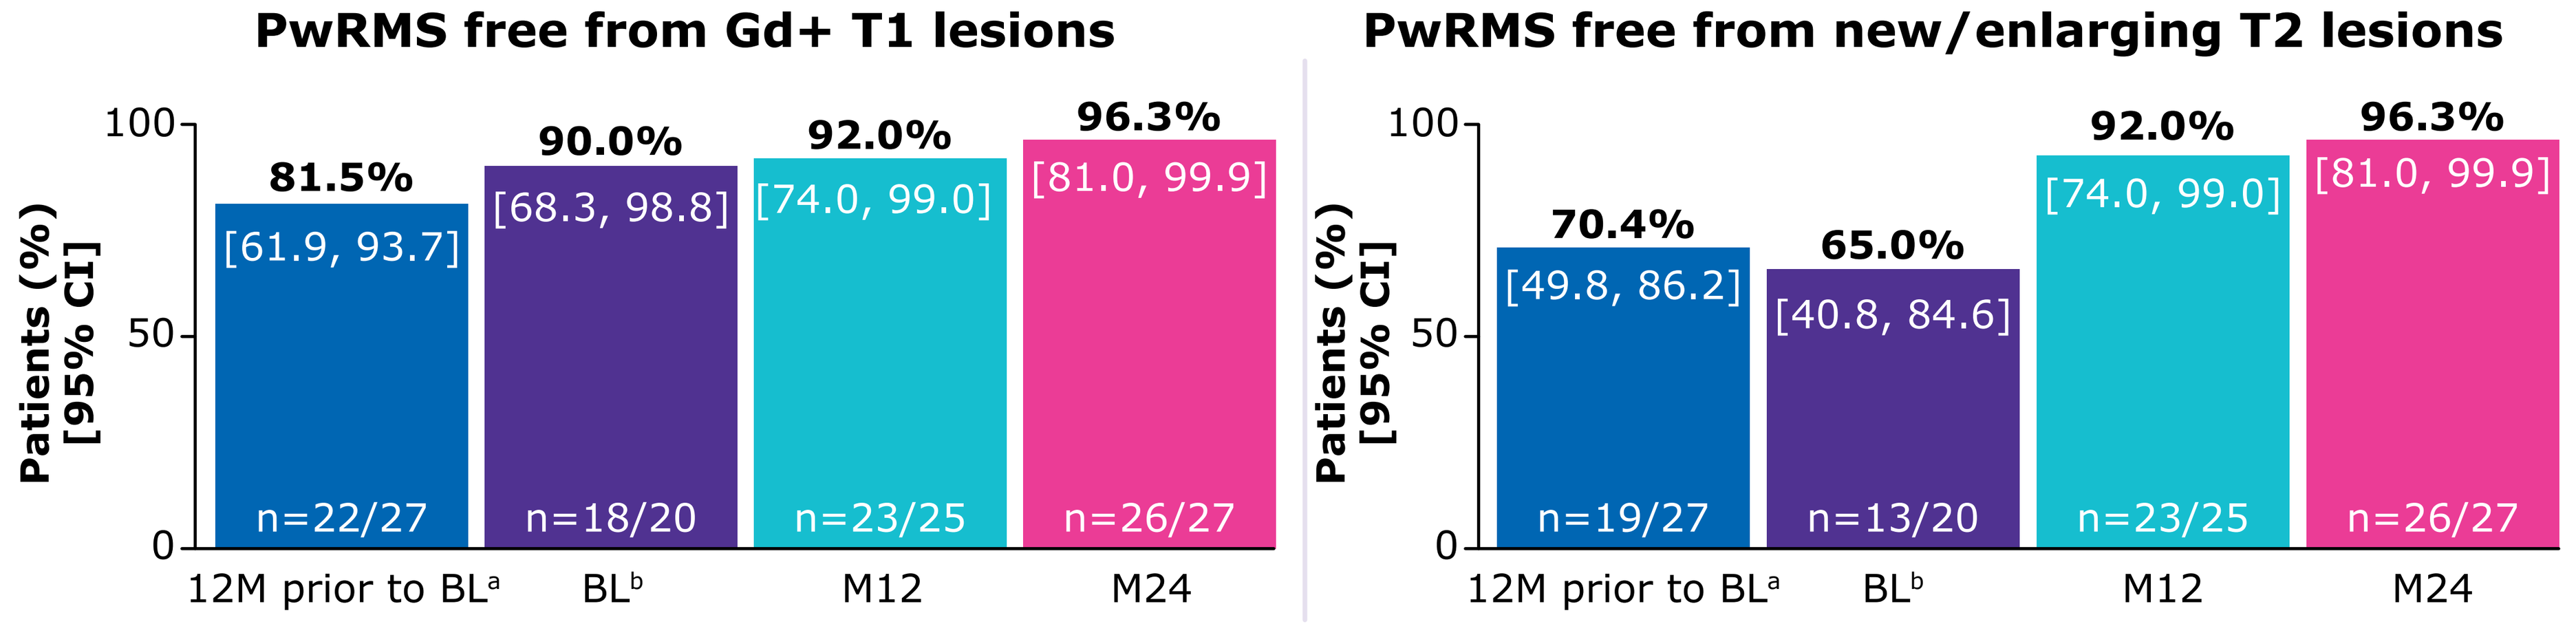
PwRMS free from Gd+ T1 lesions
Patients (%) [95% CI]
100
50
0
81.5%
[61.9, 93.7]
n=22/27
90.0%
[68.3, 98.8]
n=18/20
92.0%
[74.0, 99.0]
n=23/25
96.3%
[81.0, 99.9]
n=26/27
12M prior to BLa
BLb
M12
M24
PwRMS free from new/enlarging T2 lesions
Patients (%) [95% CI]
100
50
0
70.4%
[49.8, 86.2]
n=19/27
65.0%
[40.8, 84.6]
n=13/20
92.0%
[74.0, 99.0]
n=23/25
96.3%
[81.0, 99.9]
n=26/27
12M prior to BLa
BLb
M12
M24

MRI data availability was based on routine collection. [a]Visit occurring in the window 12 months prior to CladT start date to 3 months prior to CladT start date. [b]Baseline was defined as a visit occurring within the window from 3 months before the CladT start date to 1 month after the CladT start date.

BL, baseline; CI, confidence interval; CladT, cladribine tablets; Gd+, gadolinium-enhancing; M, month; MRI, magnetic resonance imaging; PwRMS, patients with relapsing multiple sclerosis.

- Among those with available data, the proportion of PwRMS free from any MS MRI activity after CladT initiation was 84.0% (n=21; 95% CI: 63.9, 95.5) at Month 12 and 84.0% (n=21; 95% CI: 75.7, 99.1) at Month 24.

Figure 6. mEDSS scores[a] through 24 months after switching to CladT

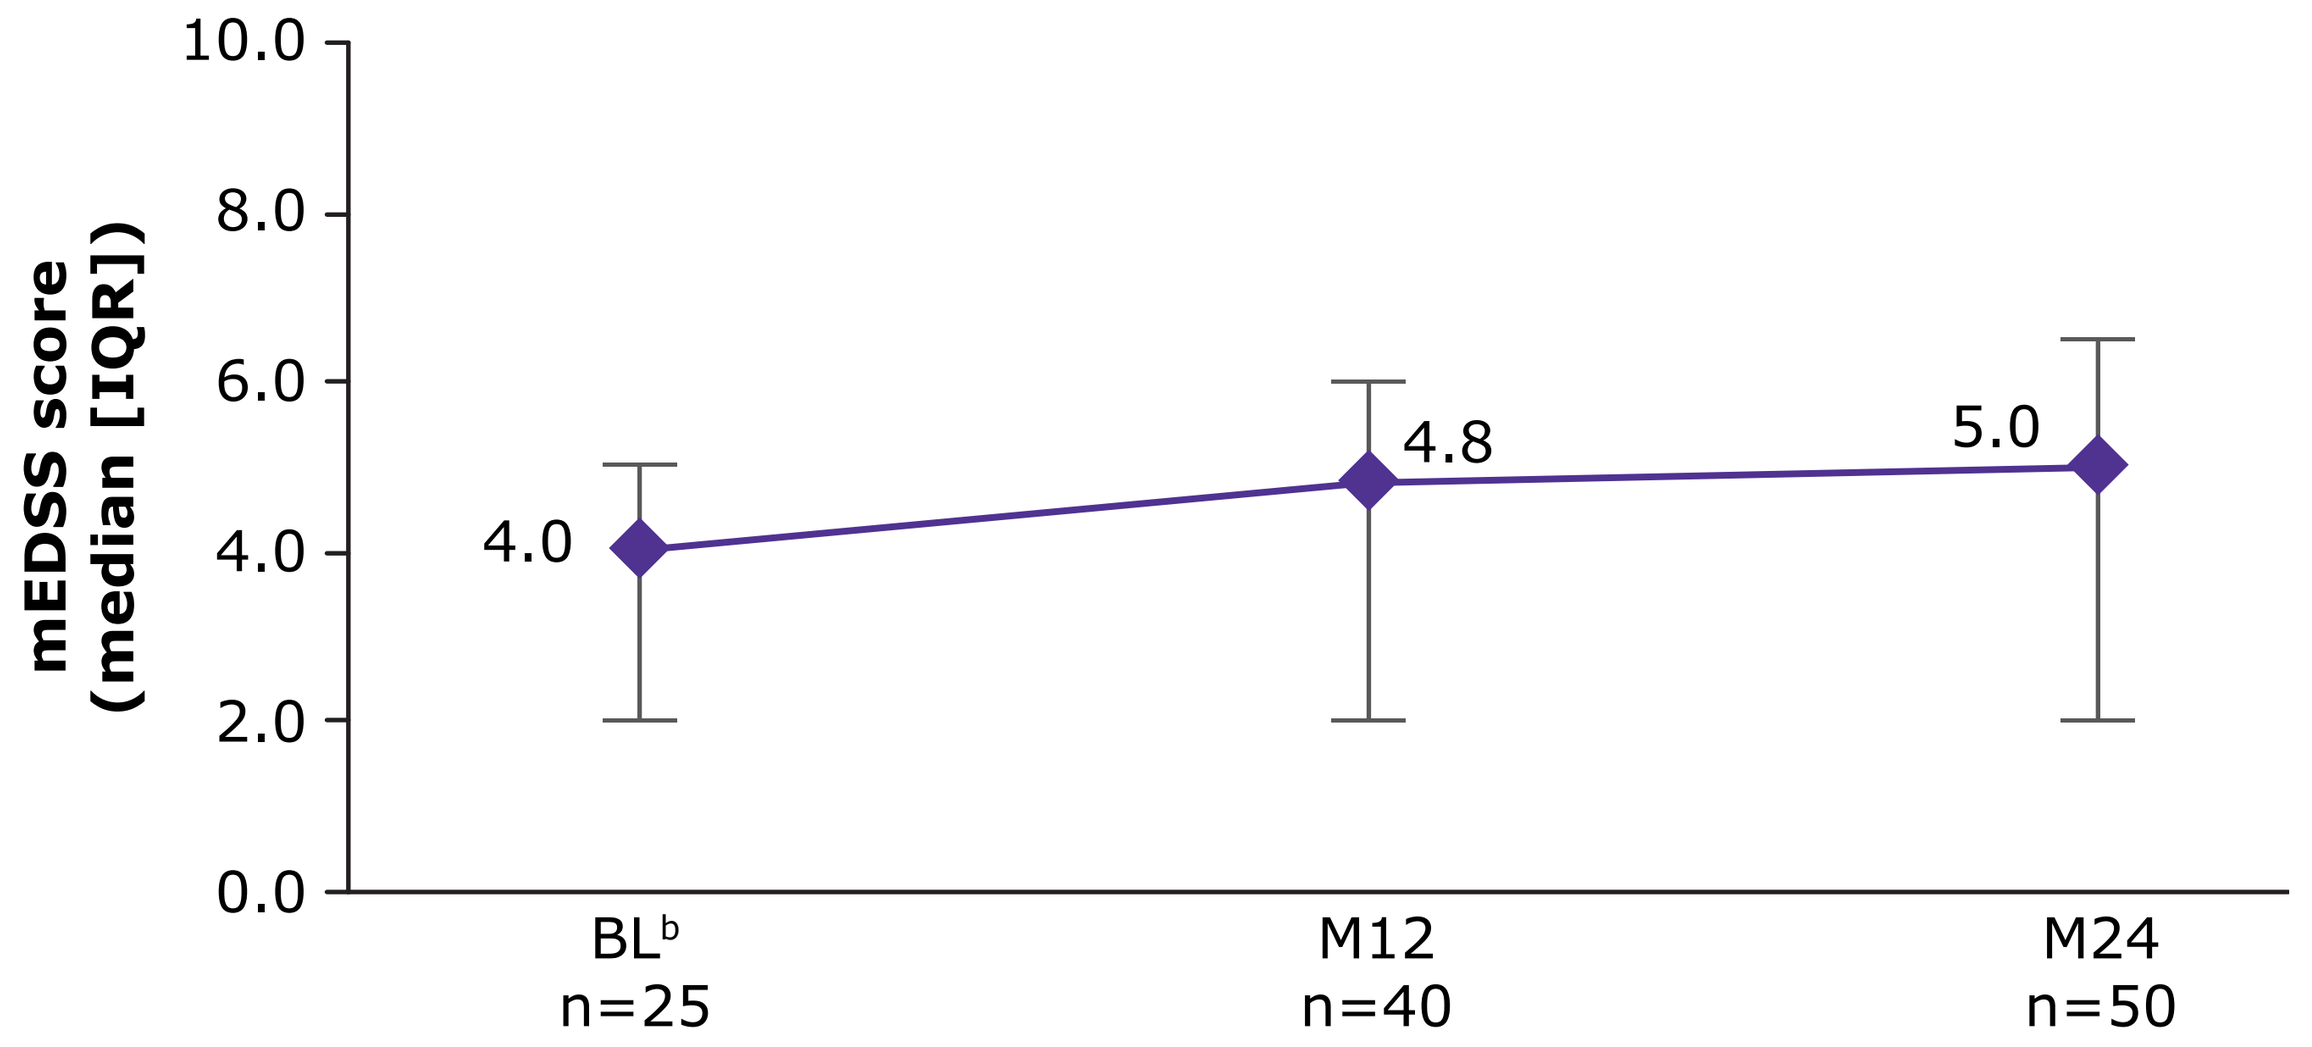
mEDSS score (median [IQR])
10.0
8.0
6.0
4.0
2.0
0.0
4.0
4.8
5.0
BLb
n=25
M12
n=40
M24
n=50

- Among PwRMS with data available at relevant timepoints, no changes in median (IQR) mEDSS scores were observed from baseline to Month 12 (n=20; 0.0 [-1.0, 0.0]) and from baseline to Month 24 (n=21; 0.0 [-0.5, 0.0]) after switching to CladT.

[a]mEDSS was developed to retrospectively capture EDSS scores using data from prior clinical documentation and has been validated as an accurate assessment of disability.[8] [b]Baseline was defined as a visit occurring within the window from 3 months before the CladT start date to 1 month after the CladT start date.

BL, baseline; CladT, cladribine tablets; IQR, interquartile range; M, month; mEDSS, modified Expanded Disability Status Scale.

Figure 7. Overall safety profile

Overall, adverse events (AEs) occurred in 45.9% (n=28) of PwRMS (**Supplementary Table 3**). One serious AE (MS pseudo-relapse) was reported.

Two PwRMS discontinued CladT due to rash and low absolute lymphocyte count.

No new safety signals and no malignancies or deaths were reported.

Most common AEs were:

- Lymphopenia n=14 (23.0%)
- Urinary tract infection n=4 (6.6%)
- Lymphocyte count decreased n=5 (8.2%)
- Muscular weakness n=3 (4.9%)

Figure 8. Lymphocyte counts through 24 months after switching to CladT

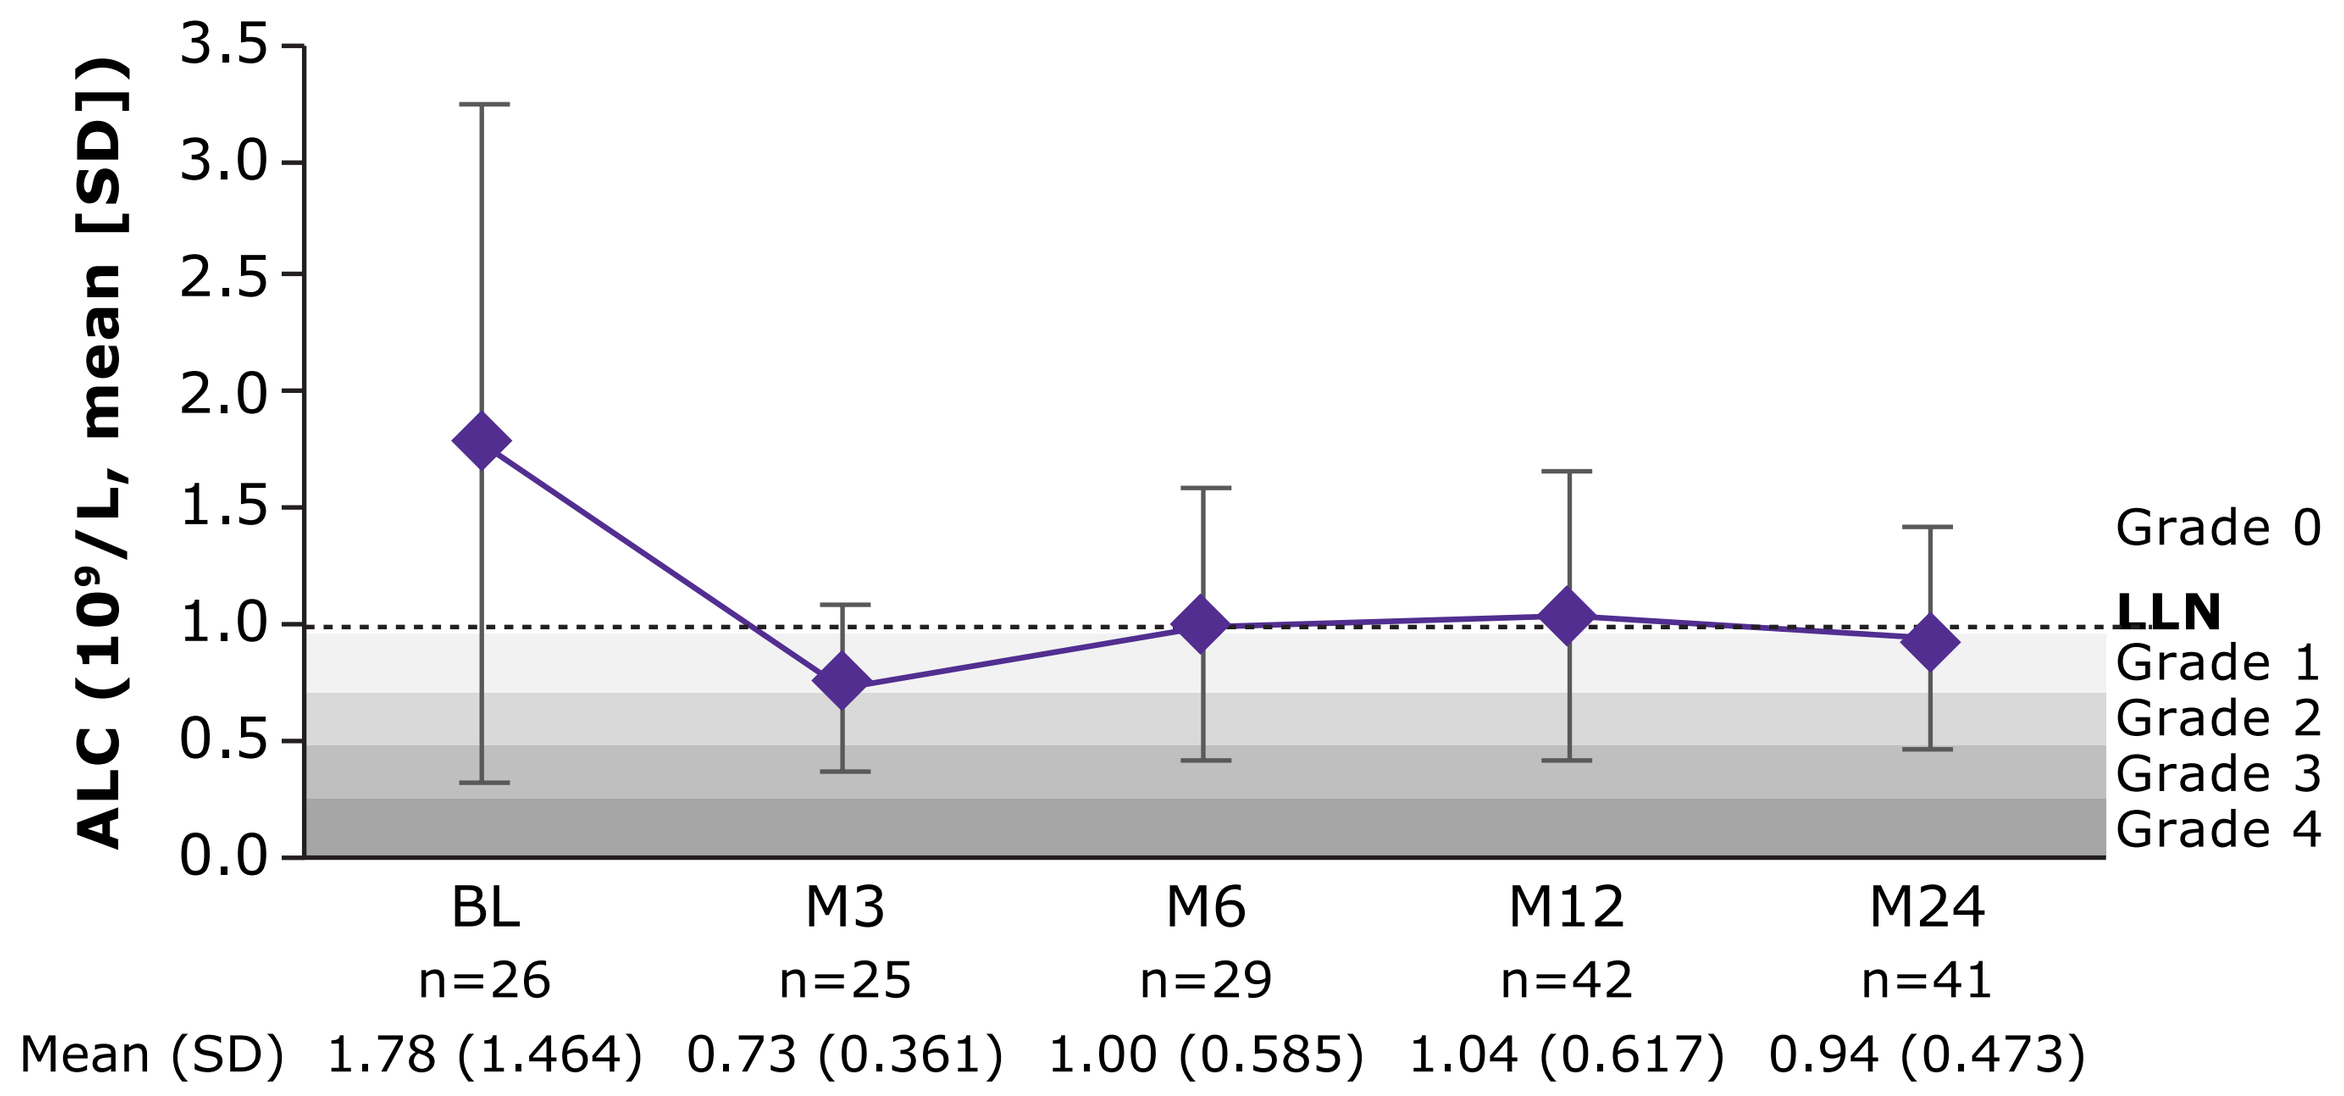
ALC (10^9/L, mean [SD])
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
Grade 0
LLN
Grade 1
Grade 2
Grade 3
Grade 4
BL
n=26
M3
n=25
M6
n=29
M12
n=42
M24
n=41
Mean (SD) 1.78 (1.464) 0.73 (0.361) 1.00 (0.585) 1.04 (0.617) 0.94 (0.473)

- After switching to CladT, ALC levels decreased from baseline to Month 3, recovered by Month 6, and were maintained through Month 24 in PwRMS with available lymphocyte data.

ALC, absolute lymphocyte count; BL, baseline; CladT, cladribine tablets; LLN, lower limit of normal; M, month; SD, standard deviation.

Limitations

- In this study, the data was collected per standard of care, during the COVID-19 pandemic, which resulted in missing data, that may limit the generalizability of the results.

CONCLUSIONS

- **ARR was low over 24 months in PwRMS who switched from moderate- to high-efficacy DMTs to CladT.**
- **Despite the small study size, these US real–world findings support the use of CladT as an effective and safe switching option for PwRMS experiencing HDA/disease breakthrough despite moderate- or high-efficacy DMT use.**

SCAN FOR AFFILIATIONS, DISCLOSURES, AND SUPPLEMENTARY MATERIALS

References:
1. Ziemssen T, *Mult Scler Relat Disord.* 2015;4(5):460-469.
2. Mavenclad®. Prescribing information. EMD Serono, Inc; January 2025.
3. Giovannoni G, *N Engl J Med.* 2010;362(5):416-426.
4. Giovannoni G, *Mult Scler.* 2018;24(12):1594-1604.
5. Giovannoni G, *Mult Scler.* 2019;25(6):819-827.
6. Brochet B, *Mult Scler.* 2023;29(14):1808-1818. 9:101884.
7. Schmierer K, *Ther Adv Neurol Disord.* 2025;18:17562864251351760.
8. Ciotti JR, *Mult Scler Relat Disord.* 2020;39:101884.

Presented at ACTRIMS Forum 2026 | February 5–7 | San Diego, California, USA

SUPPLEMENTARY MATERIALS

Author Affiliations

Barry Hendin[1], Jeffrey Kaplan[2], Sam Hooshmand[3], Angel Chinea[4], MaryAnn Picone[5], Daniel Bandari[6], Julie Korich[7], Julie Aldridge[8], Ralf Koelbach[9], Christina Caon[7], Regina Berkovich[10]

[1]Integrated Multiple Sclerosis Center, Phoenix, AZ, USA; [2]Kansas City Multiple Sclerosis and Headache Center, Overland Park, KS, USA; [3]Department of Neurology, Medical College of Wisconsin, Milwaukee, WI, USA; [4]Caribbean Center for Clinical Research, Caribbean Neurological Center, Guaynabo, Puerto Rico, USA; [5]Holy Name Multiple Sclerosis Center, Teaneck, NJ, USA; [6]Multiple Sclerosis Center of California & Research Group, Laguna Hills, CA, USA; [7]EMD Serono, Boston, MA, USA; [8]EMD Serono, Billerica, MA, USA; [9]Cytel Inc., Berlin, Germany; [10]MS Center and Research Institute, West Hollywood, CA, USA.

Author Disclosures

Barry Hendin has participated in meetings sponsored by and received honoraria (lectures, advisory boards, consultations) from Novartis, Genentech, Biogen, Sanofi, EMD Serono, Amgen, Bristol, Myers, Squibb, and Alexion.

Jeffrey Kaplan has received speaking and consulting fees from Abbvie, Amgen, Biogen, Bristol Myers Squibb, EMD Serono, Research and Speakers Bureau, Lundbeck, Mallinckrodt, Sanofi, and Vanda Pharmaceuticals. Advisory Boards and Research for Novartis.

Sam Hooshmand has received compensation for consulting and/or speaking engagements from Amgen, EMD Serono, Genentech, TG Therapeutics, and Sanofi-Genzyme. Research funding through Novartis and EMD Serono.

Angel Chinea is a speaker for Sanofi-Genzyme, Biogen, Teva, Novartis, Genentech, EMD Serono, the Healthcare business of Merck KGaA, Darmstadt, Germany in the US and Canada, Bristol Myers Squibb, and Allergan.

MaryAnn Picone has no conflicts of interest to disclose.

Daniel Bandari consulted/was a speaker for Biogen, Genentech, Genzyme, and EMD Serono, and received research support from Biogen, Genentech, and Genzyme.

Regina Berkovich received research funding from Biogen, Mallinckrodt, Novartis, Sanofi, and Teva; she is/has been a consultant for Alexion, Bayer, Biogen, Celgene, Genentech, Mallinckrodt, Novartis, and Sanofi.

Julie Korich and **Christina Caon** are employees of EMD Serono, Boston, MA, USA.

Julie Aldridge was an employee of EMD Serono, Billerica, MA, USA at the time of study.

Ralf Koelbach is an employee of Cytel Inc., Berlin, Germany, funded by EMD Serono, Boston, MA, USA.

Acknowledgements

This study was supported by EMD Serono (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Lakshmi Settu and Pritorthi Bhattacharjee of Merck Specialties Pvt. Ltd., Bengaluru, India, an affiliate of the healthcare business of Merck KGaA, Darmstadt, Germany.

SUPPLEMENTARY MATERIALS

Supplementary Figure 1: Study design, inclusion and exclusion criteria

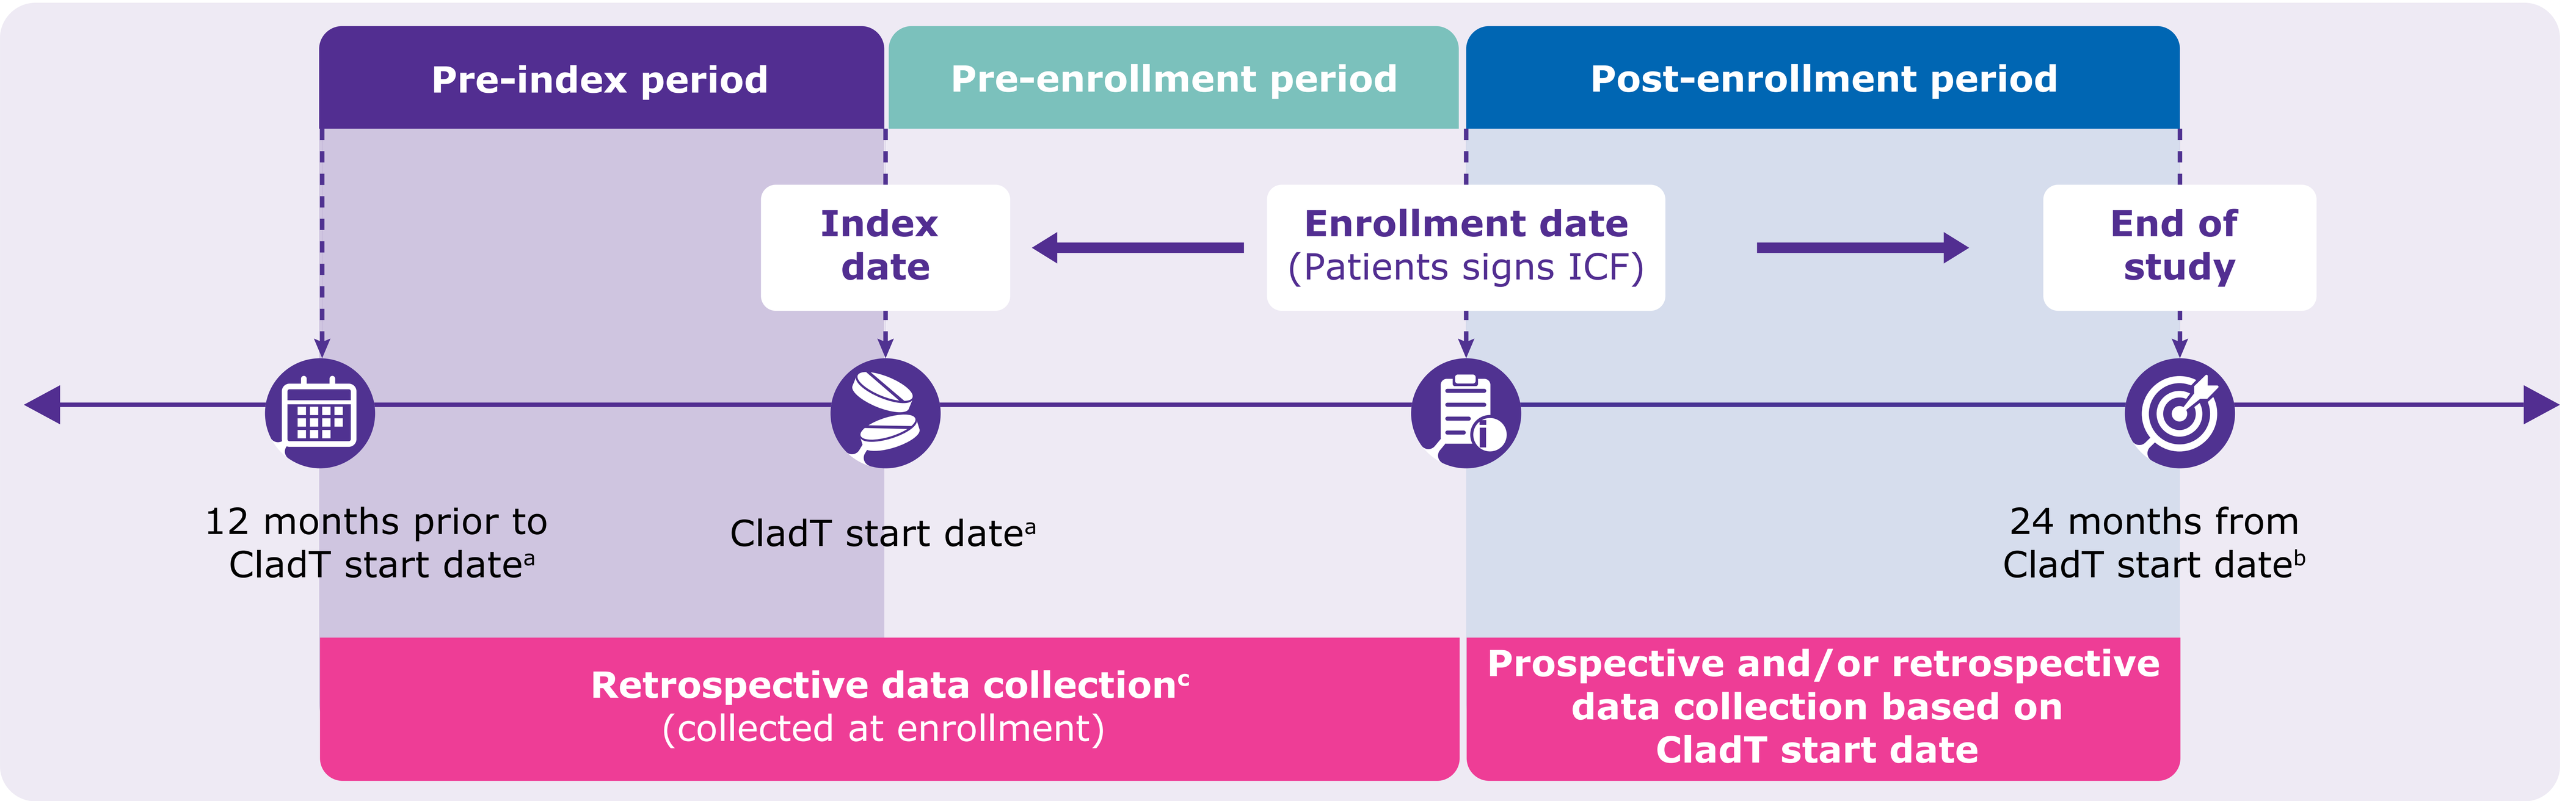
Pre-index period
Pre-enrollment period
Post-enrollment period
Index date
Enrollment date (Patients signs ICF)
End of study
12 months prior to CladT start date[a]
CladT start date[a]
24 months from CladT start date[b]
Retrospective data collection[c] (collected at enrollment)
Prospective and/or retrospective data collection based on CladT start date

[a]CladT start date was on or before January 31, 2022. Prior 12 months was defined as a visit occurring in the window 12 months prior to CladT start date to 3 months prior to CladT start date. [b]Loss to follow-up or death, whichever occurred first. [c]Most of the data were collected retrospectively from chart review resulting in missing data.

CladT, cladribine tablets; ICF, informed consent form.

Inclusion criteria

- Patients aged ≥18 years
- Diagnosis of RMS (RRMS or aSPMS) and satisfy the approved indication for CladT as per USPI
- Active disease before switching to CladT while on moderate- or high-efficacy DMTs, including fumarates[a], S1P modulators[b], anti-CD20s[c], natalizumab or alemtuzumab. Active disease was defined as relapse disease activity, MRI activity (T1 Gd+ lesions, and/or new/enlarging T2 lesions), and/or disability progression within 12 months prior to CladT initiation[d]
- Initiated CladT according to routine clinical care on or before January 31, 2022

Exclusion criteria

- Diagnosis of CIS, naSPMS, or PPMS
- Previously treated with cladribine in forms other than oral (IV or SC)
- History of malignancy except for basal cell carcinoma that has been completely excised prior to study entry and shows no evidence of recurrence
- History of and/or current serious infections (HIV, hepatitis, and tuberculosis)
- Moderate or severe renal (creatinine clearance < 60 mL/ minute)/hepatic (Child-Pugh score > 6) impairment
- Pregnant patients, those planning to breastfeed while taking CladT, those whose partner planned to become pregnant within Year 1 or 6 months after the last dose of CladT in Year 2, or who was unwilling or unable to use contraception as per CladT USPI
- Participation in an interventional clinical trial since CladT initiation

[a]Fumarates: dimethyl fumarate, diroximel fumarate, and monomethyl fumarate. [b]S1P modulators: fingolimod, siponimod, ozanimod, and ponesimod. [c]Anti-CD20s: ocrelizumab and ofatumumab. [d]One exception were switches from alemtuzumab. As alemtuzumab is an immune reconstitution therapy, a patient was eligible if activity occurred any length of time after their last infusion but within 12 months of starting CladT.

anti-CD20, anti-cluster of differentiation 20; aSPMS, active secondary progressive MS; CIS, clinically isolated syndrome; CladT, cladribine tablets; DMTs, disease-modifying therapy; Gd+, gadolinium-enhancing; HIV, human immunodeficiency virus, IV, intravenous; MRI, magnetic resonance imaging; MS, multiple sclerosis; naSPMS, non–active secondary progressive MS; PPMS, primary progressive MS; RMS, relapsing MS; RRMS, relapsing–remitting MS; S1P, sphingosine-1-phosphate; SC, subcutaneous; USPI, United States prescribing information.

Supplementary Table 1. Baseline demographics and disease characteristics

Characteristics	Total (N=61)
Age (years)	
Mean (SD)	49 (10.1)
Median, (IQR)	48 (42–56)
<50 years, n (%)	34 (55.7)
≥50 years, n (%)	27 (44.3)
Sex, n (%)	
Male	17 (27.9)
Female	44 (72.1)
Race, n (%)	
White	50 (82.0)
Black or African American	8 (13.1)
Other Asian	1 (1.6)
Unknown/Unavailable	2 (3.3)
Ethnicity, n (%)	
Hispanic or Latino	6 (9.8)
Not Hispanic or not Latino	53 (86.9)
Type of MS, n (%)	
RRMS	51 (83.6)
aSPMS[a]	10 (16.4)
Duration from MS diagnosis to CladT initiation, (years)[b]	
Mean (SD)	12.35 (9.30)
Median, (IQR)	9.56 (5.42–18.01)
Medical history event or comorbidity	
Vitamin D deficiency	17 (27.9)
Insomnia	8 (13.1)
Hypertension	8 (13.1)
Fatigue	7 (11.5)
Others	14 (22.9)

[a]aSPMS: with relapses and/or evidence of new MRI activity during a specified period. [b]Duration was calculated as: (date of first intake of CladT–date of first MS diagnosis + 1)/365.25.

aSPMS, active secondary progressive multiple sclerosis; CI, confidence interval; CladT, cladribine tablets; IQR, interquartile range; MS, multiple sclerosis; RRMS, relapsing–remitting MS; SD, standard deviation.

Supplementary Table 2. Relapse counts (N=61)

Relapse count, n (%)	BL	M12	M24
Missing	5 (8.2)	8 (13.1)	5 (8.2)
0	20 (32.8)	42 (68.9)	44 (72.1)
1	34 (55.7)	10 (16.4)	11 (18.0)
2	1 (1.6)	1 (1.6)	1 (1.6)
≥3	1 (1.6)	0	0

BL, baseline; M, month.

Supplementary Table 3. Overall safety profile

Event, n (%)	Total (N=61)
Any AE	28 (45.9)
Any SAE	1 (1.6)
MS pseudo relapse	1 (1.6)
Any ADR	21 (34.4)
Any serious ADR	0
AEs leading to CladT discontinuation[a]	2 (3.2)
Pre-specified potential safety event (Severe lymphopenia, severe infections, progressive multifocal leukoencephalopathy, opportunistic infection, malignancies)	0
Most common AEs (>3.0% in the total population)	
Lymphopenia	14 (23.0)
Urinary tract infection	4 (6.6)
Lymphocyte count decreased	5 (8.2)
Muscular weakness	3 (4.9)

[a]Discontinued CladT treatment due to low ALC (n=1) and due to rash (n=1).

ADR, adverse drug reaction; AE, adverse event; ALC, absolute lymphocyte count; CladT, cladribine tablets; MS, multiple sclerosis; SAE, serious adverse event.

Supplement Table 4. Number of days between the last DMT administration and the first CladT treatment

Prior-DMT	Number of days median [IQR][a]
Alemtuzumab	794.0 [519.0; 1223.0]
Ofatumumab	113.5 [15.0; 212.0]
Ocrelizumab	105.0 [53.0; 200.0]
Natalizumab	70.0 [31.5; 131.5]
Fumarates	52.5 [17.5; 128.0]
S1P modulators	48.0 [7.0; 135.0]

[a]Number of days of washout is defined as the time between the last date of most recent prior DMT and the first date of CladT initiation.

CladT, Cladribine Tablets; DMT, disease-modifying therapies; S1P, sphingosine 1-phosphate.