Comparative Assessment of Switching, Healthcare Resource Utilization, and Cost Outcomes for Cladribine Tablets Versus Other Oral Disease-Modifying Therapies Among US Patients with Multiple Sclerosis Over 4 Years

Ajay Gupta, Joanna P. MacEwan, Jeffrey Anderson, Xin Zhao, Sonia Kim, Andy Surinach, Yu Hong, *Amy L. Phillips, Xiaoxue Chen*

This work was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Authors in italics are employees of Merck or its affiliates. Please scan the QR code below for full affiliations.

SCAN FOR POSTER PDF

For personal use only and may not be reproduced without written permission of the authors

RESEARCH IN CONTEXT

1. **The comparative findings of cladribine tablets (CladT) versus other disease modifying therapies (DMTs) highlight the role of CladT as a valuable treatment option in people with multiple sclerosis (PwMS) owing to fewer treatment switches and lower medical costs.**
2. **Further research should evaluate outcomes in patient subgroups to better understand the clinical utility of different DMTs in heterogeneous MS populations.**

OBJECTIVES

To evaluate treatment switching, healthcare resource utilization (HCRU), and cost outcomes among PwMS treated with CladT vs fingolimod (FTY), dimethyl fumarate (DMF), and teriflunomide (TER) over 4 years in the United States (US).

INTRODUCTION

As treatment options for MS increase, comparative effectiveness data for DMTs are essential.[1]

However, real-world data on switching, HCRU, and costs related to CladT vs other DMTs remain limited.

METHODS

Study Design: Retrospective study using US claims data from the Komodo Healthcare Map™ database from April 1, 2018, to March 31, 2024.

Inclusion Criteria:

Adult PwMS with ≥2 claims for MS ≥30 days apart and ≥1 claim for CladT, FTY, DMF, or TER (Index date = date of the first claim for a DMT of interest).

Continuous enrollment for 12 months pre- and 48 months post-index.

Exclusion Criteria:

Pregnancy diagnosis during 12 months pre- or 24 months post-index.

No prior claims for the index DMT during the 12 months pre-index (new user design).

POS code indicating hospice, inpatient mental health, or inpatient rehabilitation during 12 months pre-index.

Any cladribine infusion claims, multiple treatments on the index date or excessive claims[a] for CladT.

Study Outcomes: Proportion of PwMS switching to non-index DMT, time to switch, all-cause HCRU, and healthcare costs over a 4-year post-index period.

Statistical methods:

Comparisons	CladT vs FTY; CladT vs DMF; CladT vs TER
Method	Propensity score (PS)[b] weighted pairwise comparisons
Key covariates	Age, index year, sex, region, insurance type, CCI, MS comorbidities, relapse history, MS severity, prior DMT, dalfampridine use
Handling imbalance in baseline characteristics	Covariates with SMD >0.1 included in GLM and Cox models
Analysis framework	Intention-to-treat; outcomes assigned to index DMT over 4 years

[a]threshold: >6 claims may indicate data quality issues according to clinical input. [b]calculated using the weighting of odds approach to estimate the average treatment among the treated.
CladT, cladribine tablets; CCI, Charlson Comorbidity Index; DMF, dimethylfumarate; DMT, disease modifying therapy; FTY, fingolimod; GLM, generalized linear model; MS, multiple sclerosis; POS, place of service; SMD, standardized mean difference; TER, teriflunomide.

RESULTS

- Overall, 3,038 PwMS met the inclusion criteria (CladT: 140, DMF: 1,465, FTY: 454, and TER: 979).
- At the index, the mean age ranged from 42 (standard deviation [SD]: 11) years in the FTY cohort to 50 (11) years in the TER cohort and >70% of the PwMS were female. (**Supplementary Table 1**).
- After PS weighting, most baseline characteristics across the treatment groups were balanced (**Table 1**).

Table 1. Baseline demographic and clinical characteristics (weighted)

Characteristics	CladT (n=140)	FTY (Weighted n=137/ ESS=141)	FTY vs. CladT SMD	DMF (Weighted n=141/ ESS=536)	DMF vs. CladT SMD	TER (Weighted n=140/ ESS=491)	TER vs. CladT SMD
Age (years), mean (SD)	48 (10)	48 (12)	0.03	48 (12)	<0.01	47 (12)	0.02
Sex, n (%)			0.07		<0.01		0.01
Female	107 (76)	101 (74)		108 (77)		107 (76)	
Male	33 (24)	36 (26)		33 (23)		34 (24)	
Index year, n (%)			0.05		0.02		0.03
2019	68 (49)	64 (46)		67 (48)		66 (47)	
2020	72 (51)	74 (54)		74 (52)		74 (53)	
Region, n (%)			0.05		0.05		0.04
Midwest	37 (26)	33 (24)		39 (28)		36 (26)	
Northeast	47 (34)	47 (34)		49 (35)		49 (35)	
South	40 (29)	40 (29)		37 (26)		39 (28)	
West	16 (11)	17 (12)		16 (11)		16 (11)	
Insurance type, n (%)			0.05		0.02		0.02
Commercial	87 (62)	88 (64)		87 (62)		87 (62)	
Medicare	38 (27)	35 (25)		38 (27)		37 (27)	
Medicaid	15 (11)	14 (11)		16 (11)		16 (11)	
CCI, mean (SD)	0.67 (1.22)	0.68 (1.25)	0.01	0.68 (1.17)	0.01	0.67 (1.29)	<0.01
MS severity score, mean (SD)	5.4 (4.0)	5.1 (4.1)	0.08	5.4 (4.2)	0.02	5.3 (4.0)	0.01
MS relapse, n (%)	55 (39)	55 (40)	0.02	58 (41)	0.04	56 (40)	0.01
MS-related hospitalization, n (%)	6 (4.3)	21 (15)	0.46	16 (12)	0.34	13 (8.9)	0.22
Treatment history, n (%)							
Dalfampridine use	10 (7.1)	7 (5.2)	0.1	12 (8.2)	0.05	9 (6.5)	0.03
Prior DMT use	78 (56)	72 (53)	0.06	77 (55)	0.02	77 (55)	0.02
Platform injection therapies	30 (21)	54 (40)	0.45	66 (47)	0.6	62 (45)	0.52
Monoclonal antibodies	41 (29)	11 (8.3)	0.61	9 (6.6)	0.67	11 (7.7)	0.62
Oral medications	12 (8.6)	8 (5.8)	0.11	3 (2.0)	0.3	7 (5.2)	0.14

CladT, cladribine tablets; CCI, Charlson Comorbidity Index; DMF, dimethyl fumarate; DMT, disease-modifying therapy; ESS, effective sample size; FTY, fingolimod; MS, multiple sclerosis; SD, standard deviation; SMD, standardized mean difference; TER, teriflunomide.

Treatment switching within 4 Years

- Fewer PwMS switched to other DMTs within 4 years following treatment with CladT (11%) vs FTY (42%), DMF (57%) and TER (42%) cohorts (**Supplementary table 2**). The odds ratios (OR) for switching were significantly higher for FTY, DMF and TER vs CladT (**Figure 1A**).
- Higher hazard ratios (HR) for treatment switching were observed for DMF (7.57 [95% CI: 4.59–12.48]), FTY (5.26 [95% CI: 3.02–9.17]) and TER (5.01 [95% CI: 3.02–8.31]), vs CladT (**Figure 1B**). Kaplan-Meier curves are presented in **Supplementary Figure 1**.

Figure 1A. ORs (95% CI) for treatment switching within 4 years vs CladT

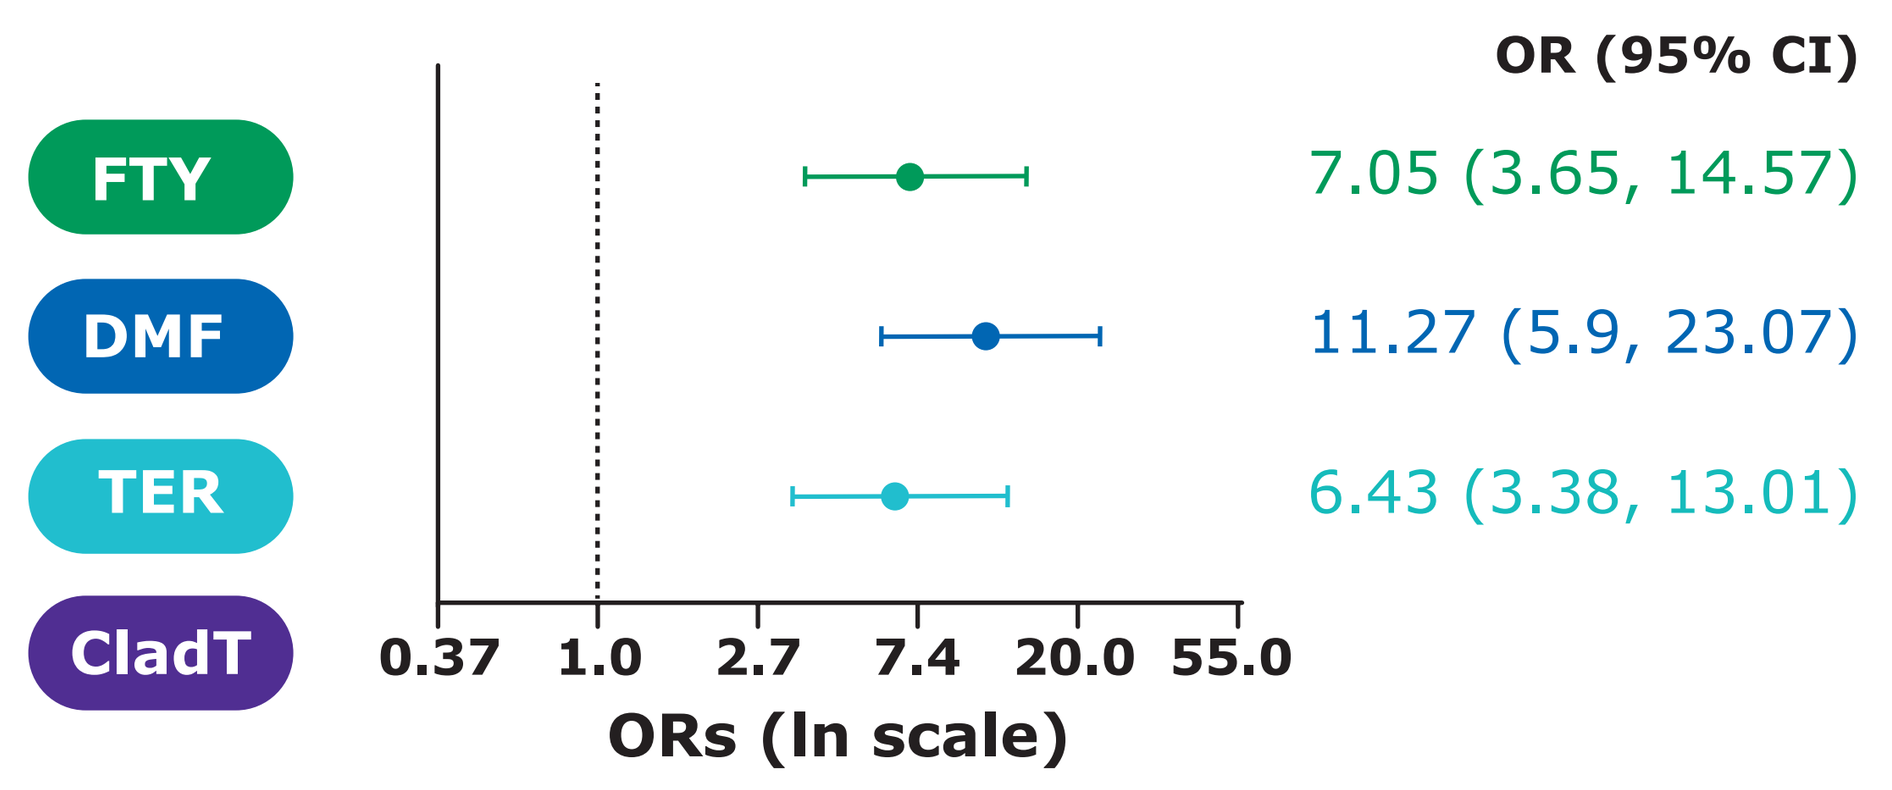
OR (95% CI)
FTY
DMF
TER
CladT
7.05 (3.65, 14.57)
11.27 (5.9, 23.07)
6.43 (3.38, 13.01)
0.37
1.0
2.7
7.4
20.0
55.0
ORs (ln scale)

Figure 1B. HRs (95% CI) for time to switch within 4 years vs CladT

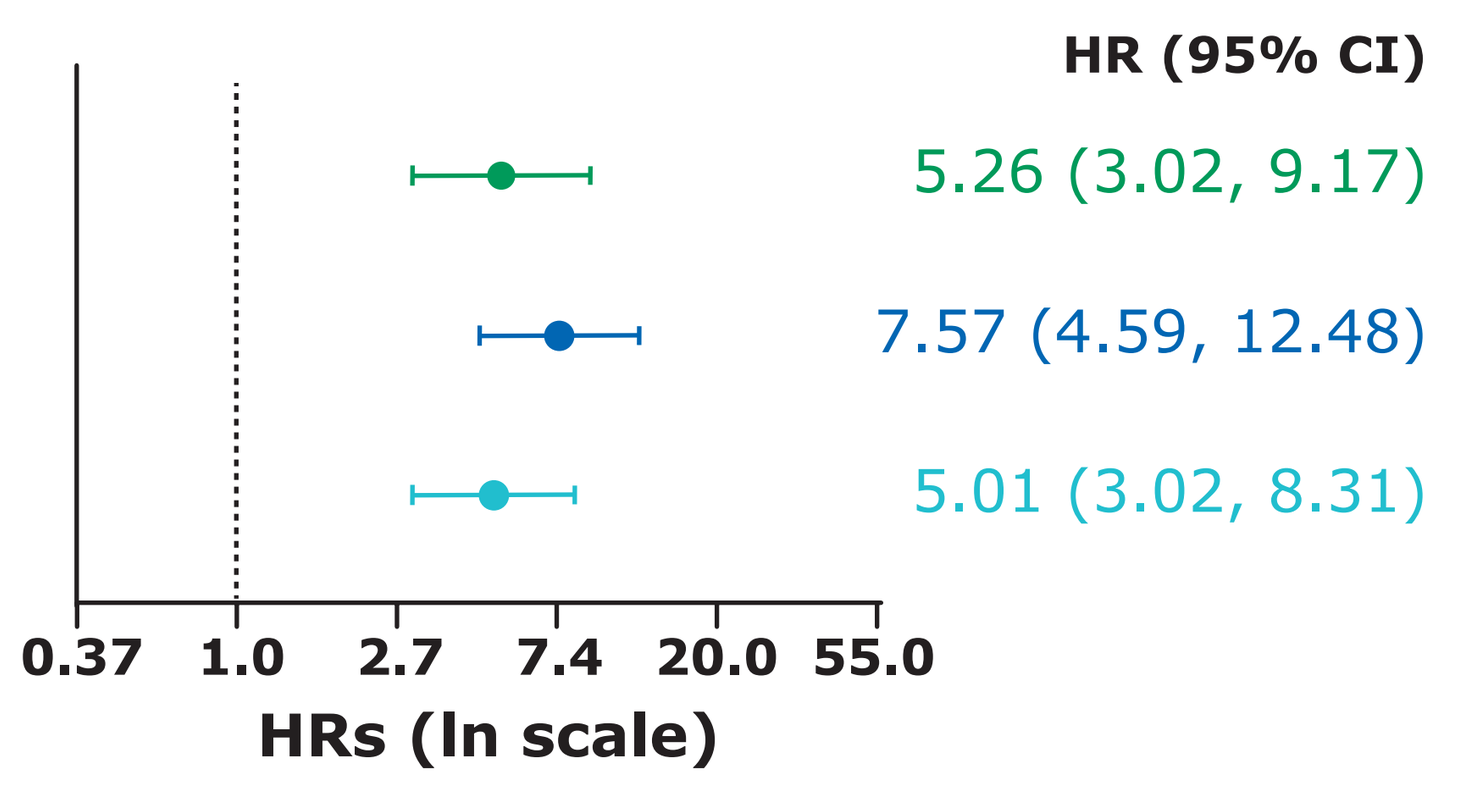
HR (95% CI)
5.26 (3.02, 9.17)
7.57 (4.59, 12.48)
5.01 (3.02, 8.31)
0.37
1.0
2.7
7.4
20.0
55.0
HRs (ln scale)

CladT is used as the reference cohort for calculating OR and HR.
CI, confidence interval; CladT, cladribine tablets; DMF, dimethyl fumarate; FTY, fingolimod; HR, hazard ratio; ln, natural logarithm; OR, odds ratio; TER, teriflunomide.

HCRU and costs within 4 years

- All-cause outpatient visits were lower in the CladT cohort, vs the DMF and TER cohorts, but were similar to the FTY cohort. No significant differences were observed in emergency room visits and hospitalizations between the cohorts (**Table 2**).

Table 2. All-cause HCRU within 4 years (weighted[a])

Outcomes	CladT (n=140)	FTY (Weighted n=137/ESS=141)	IRR (95% CI)	DMF (Weighted n=141/ESS=536)	IRR (95% CI)	TER (Weighted n=140/ESS=491)	IRR (95% CI)
	Mean (SD)	Mean (SD)	FTY (ref=CladT)	Mean (SD)	DMF (ref=CladT)	Mean (SD)	TER (ref=CladT)
Inpatient admissions, PPPY	0.1 (0.3)	0.1 (0.3)	0.96 (0.45–2.12)	0.1 (0.4)	1.2 (0.68–2.51)	0.1 (0.3)	1.09 (0.64–2.33)
ER visits, PPPY	0.5 (1.0)	0.4 (0.7)	0.8 (0.53–1.34)	0.5 (1.0)	1.0 (0.67–1.52)	0.5 (0.8)	0.93 (0.65–1.35)
Outpatient visits, PPPY	23.7 (22.5)	26.7 (31.8)	1.2 (0.96–1.53)	29.4 (42.4)	1.32 (1.06–1.63)	26.9 (31.7)	1.19 (1.01–1.43)

95% CI was estimated using bootstrapping with 1,000 replicates. [a]All weighted models were further adjusted by baseline monoclonal antibody use and baseline oral/platform use.
CI, confidence interval; CladT, cladribine tablets; FTY, fingolimod; DMF, dimethyl fumarate; ER, emergency room; ESS, effective sample size; IRR, incidence rate ratio; HCRU, healthcare resource utilization; PPPY, per patient per year; SD, standard deviation; ref, reference; TER, teriflunomide.

- All-cause medical costs PPPY were lower in the CladT cohort ($13,377) vs FTY (adjusted mean difference [AMD]: $10,073 [95% CI: $5,006–$15,148]), DMF (AMD: $12,013 [95% CI: $7,490–$16,706]), and TER (AMD: $9,917 [95% CI: $6,558–$13,997]) cohorts.
- All-cause total costs PPPY were also lower in the CladT cohort ($53,007) vs FTY (AMD: $14,140 [95% CI: $7,181–$22,021), TER (AMD: $6,652 [95%CI: $567–$12,395]) cohorts and were similar to DMF cohort (AMD: $4,472 (95% CI: $−2,011–$11,111) (**Table 3**).

Table 3. All-cause costs within 4 years (weighted[a])

Outcomes	CladT (n=140)	FTY (Weighted n=137/ESS=141)	AMD (95% CI)	DMF (Weighted n=141/ESS=536)	AMD (95% CI)	TER (Weighted n=140/ESS=491)	AMD (95% CI)
	Mean (SD)	Mean (SD)	FTY (ref=CladT)	Mean (SD)	DMF (ref=CladT)	Mean (SD)	TER (ref=CladT)
Medical costs, PPPY	$13,377 ($17,305)	$21,724 ($27,380)	$10,073 ($5,006–$15,148)	$23,133 ($31,736)	$12,013 ($7,490–$16,706)	$21,542 ($31,023)	$9,917 ($6,558–$13,997)
Inpatient admissions, PPPY	$2,164 ($6,800)	$2,510 ($8,031)	$346 ($−1,299–$1,938)	$2,531 ($7,249)	$367 ($−1,063–$1,679)	$2,637 ($8,318)	$473 ($830–$1,663)
ER costs, PPPY	$653 ($1,553)	$515 ($1,527)	$−197 ($−570–$182)	$582 ($1,507)	$−72 ($−416–$290)	$579 ($1,231)	$-120 ($−455–$225)
Outpatient costs, PPPY	$10,553 ($12,668)	$18,619 ($24,827)	$9,367 ($5,223–$13,789)	$19,656 ($28,511)	$10,989 ($7,246–$14,998)	$18,010 $(26,998)	$8,910 ($6,115–$12,037)
Pharmacy costs, PPPY	$39,630 ($21,454)	$42,921 ($27,729)	$4,353 ($−1,125–$10,393)	$31,392 ($29,669)	$−6,751 ($−11,410–$−2,161)	$36,075 ($28,708)	$-3,181 ($−7,908–$1,135)
Total costs, PPPY	$53,007 ($29,681)	$64,646 ($33,640)	$14,140 ($7,181–$22,021)	$54,525 ($41,608)	$4,472 ($−2,011–$11,111)	$57,618 ($38,153)	$6,652 ($567–$12,395)

95% CI was estimated using bootstrapping with 1,000 replicates. [a]All weighted models were further adjusted by baseline monoclonal antibody use and baseline oral/platform use. Overall inpatient costs were not further adjusted to ensure successful bootstrap execution.
AMD, adjusted mean difference; CI, confidence interval; CladT, cladribine tablets; DMF, dimethyl fumarate; ER, emergency room; ESS, effective sample size; FTY, fingolimod; PPPY, per patient per year; SD, standard deviation; TER, teriflunomide.

CONCLUSIONS

- **PwMS treated with CladT had significantly fewer treatment switches and lower medical costs vs those treated with FTY, DMF, and TER.**

Limitations

- Requirement for ≥4 years of post-index continuous enrollment limits generalizability to all DMT initiators.
- Cost imputation was utilized due to incomplete cost data in the Komodo Health dataset, which may not accurately reflect true costs and could underestimate variance.
- Imbalances in baseline characteristics, such as prior DMT drug categories, remained after PS weighting and were adjusted for in the outcome model; however, residual confounding cannot be excluded.

SCAN FOR AFFILIATIONS, DISCLOSURES, AND SUPPLEMENTARY MATERIALS

Reference: 1. Rae-Grant A, et al. *Neurology*. 2018;90(17):789-800.

For Reactive Medical Use Only

Presented at the ACTRIMS Forum, 2026 | February 5–7 | San Diego, CA, USA

SUPPLEMENTARY MATERIALS

Author Affiliations

Ajay Gupta[1], Joanna P. MacEwan[2], Jeffrey Anderson[2], Xin Zhao[2], Sonia Kim[2], Andy Surinach[2], Yu Hong[2], *Amy L. Phillips*[3], *Xiaoxue Chen*[3]

[1]Indiana University Medical School, Ft Wayne, IN; [2]Genesis Research Group, Hoboken, NJ; [3]EMD Serono, Boston, MA, USA.

Author Disclosures

AG has received compensation for consulting or speaking from EMD Serono.

JPM, **JA, XZ, SK, YH** are employees of Genesis Research. Genesis Research received funding from EMD Serono.

AS was an employee of Genesis Research during the poster development.

ALP and **XC** are employees of EMD Serono, Boston, MA, USA.

Acknowledgments

This work was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Medical writing support was provided by Akshata Rao of Merck Specialties Pvt. Ltd., Bengaluru, India, an affiliate of Merck KGaA, Darmstadt, Germany.

SUPPLEMENTARY MATERIALS

Supplementary Table 1. Baseline demographic and clinical characteristics (unweighted)

Characteristics	CladT (n=140)	FTY (n=454)	SMD for FTY vs. CladT	DMF (n=1,465)	SMD for DMF vs. CladT	TER (n=979)	SMD for TER vs. CladT
Age (years), mean (SD)	48 (10)	42 (11)	0.51	46 (11)	0.18	50 (11)	0.25
Sex, n (%)			0.1		0.06		0.07
Female	107 (76)	328 (72)		1,082 (74)		778 (79)	
Male	33 (24)	126 (28)		383 (26)		201 (21)	
Index year, n (%)			0.57		0.35		0.4
2019	68 (49)	341 (75)		961 (66)		667 (68)	
2020	72 (51)	113 (25)		504 (34)		312 (32)	
Region, n (%)			0.14		0.18		0.15
Midwest	37 (26)	126 (28)		401 (27)		291 (30)	
Northeast	47 (34)	123 (27)		378 (26)		275 (28)	
South	40 (29)	146 (32)		470 (32)		318 (32)	
West	16 (11)	59 (13)		216 (15)		95 (9.7)	
Insurance type, n (%)			0.53		0.33		0.18
Commercial	87 (62)	346 (76)		1,064 (73)		663 (68)	
Medicare	38 (27)	35 (7.7)		207 (14)		192 (20)	
Medicaid	15 (11)	73 (16)		194 (13)		124 (13)	
CCI	0.67 (1.22)	0.46 (1.05)	0.19	0.42 (0.92)	0.23	0.55 (1.10)	0.1
MS severity score, mean (SD)	5.4 (4.0)	3.8 (3.4)	0.43	3.6 (3.2)	0.49	3.9 (3.4)	0.39
MS relapse, n (%)	55 (39)	153 (34)	0.12	450 (31)	0.18	303 (31)	0.18
MS-related hospitalization, n (%)	6 (4.3)	36 (7.9)	0.15	86 (5.9)	0.07	50 (5.1)	0.04
Treatment history, n (%)							
Dalfampridine use	10 (7.1)	7 (1.5)	0.28	34 (2.3)	0.23	48 (4.9)	0.09
Prior DMT use	78 (56)	125 (28)	0.6	422 (29)	0.57	428 (44)	0.24
Platform injection therapies	30 (21)	91 (20)	0.03	373 (25)	0.1	355 (36)	0.33
Monoclonal Antibodies	41 (29)	15 (3.3)	0.75	36 (2.5)	0.79	38 (3.9)	0.73
Oral Medications	12 (8.6)	22 (4.8)	0.15	23 (1.6)	0.32	51 (5.2)	0.13

CladT, Cladribine tablets; CCI, Charlson Comorbidity Index; DMF, dimethyl fumarate; DMT, disease modifying therapy; FTY, fingolimod; MS, multiple sclerosis; SMD, standardized mean difference; SD, standard deviation; TER, teriflunomide.

Supplementary Table 2. Proportion of patients switching to non-index DMTs (weighted)

Name of DMT switched to, n (%)	CladT (n=140)	FTY (Weighted n=137/ ESS=141)	DMF (Weighted n=141/ ESS=536)	TER (Weighted n=140/ ESS=491)
Platform	4 (2.9%)	12 (8.5%)	9 (6.6%)	10 (7.0%)
Glatiramer	3 (2.1%)	6 (4.2%)	7 (4.9%)	7 (5.2%)
Interferon beta-1a	1 (0.7%)	4 (2.7%)	1 (0.9%)	2 (1.4%)
Interferon beta-1b	0 (0%)	0 (0%)	0 (0.1%)	0 (0.3%)
Peginterferon beta-1b	0 (0%)	2 (1.6%)	1 (0.7%)	0 (0.1%)
Monoclonal antibodies	8 (5.7%)	32 (23%)	29 (20%)	32 (23%)
Alemtuzumab	0 (0%)	0 (0%)	0 (<0.1%)	0 (0%)
Natalizumab	0 (0%)	4 (3.0%)	4 (2.7%)	4 (3.0%)
Ocrelizumab	5 (3.6%)	19 (14%)	18 (13%)	18 (13%)
Ofatumumab	3 (2.1%)	9 (6.3%)	7 (4.6%)	10 (6.8%)
Ublituximab	0 (0%)	0 (0%)	1 (0.5%)	0 (0.1%)
Oral	4 (2.9%)	15 (11%)	41 (29%)	17 (12%)
CladT	0 (0%)	3 (2.0%)	3 (2.1%)	4 (2.7%)
DMF	2 (1.4%)	4 (3.2%)	0 (0%)	4 (2.8%)
TER	0 (0%)	1 (0.8%)	7 (5.2%)	0 (0%)
FTY	0 (0%)	0 (0%)	4 (3.0%)	2 (1.2%)
Diroximel fumarate	0 (0%)	0 (0.3%)	22 (16%)	3 (2.4%)
Monomethyl fumarate	0 (0%)	0 (0%)	0 (0.2%)	0 (0%)
Ozanimod	2 (1.4%)	3 (2.0%)	2 (1.7%)	3 (1.8%)
Ponesimod	0 (0%)	0 (0%)	0 (0%)	0 (0%)
Siponimod	0 (0%)	4 (2.6%)	2 (1.4%)	2 (1.5%)

CladT, cladribine tablets; DMF, dimethyl fumarate; DMT, disease-modifying therapy; ESS, effective sample size; FTY, fingolimod; TER, teriflunomide.

Supplementary Figure 1. Kaplan-Meier curves for time to switch (weighted)

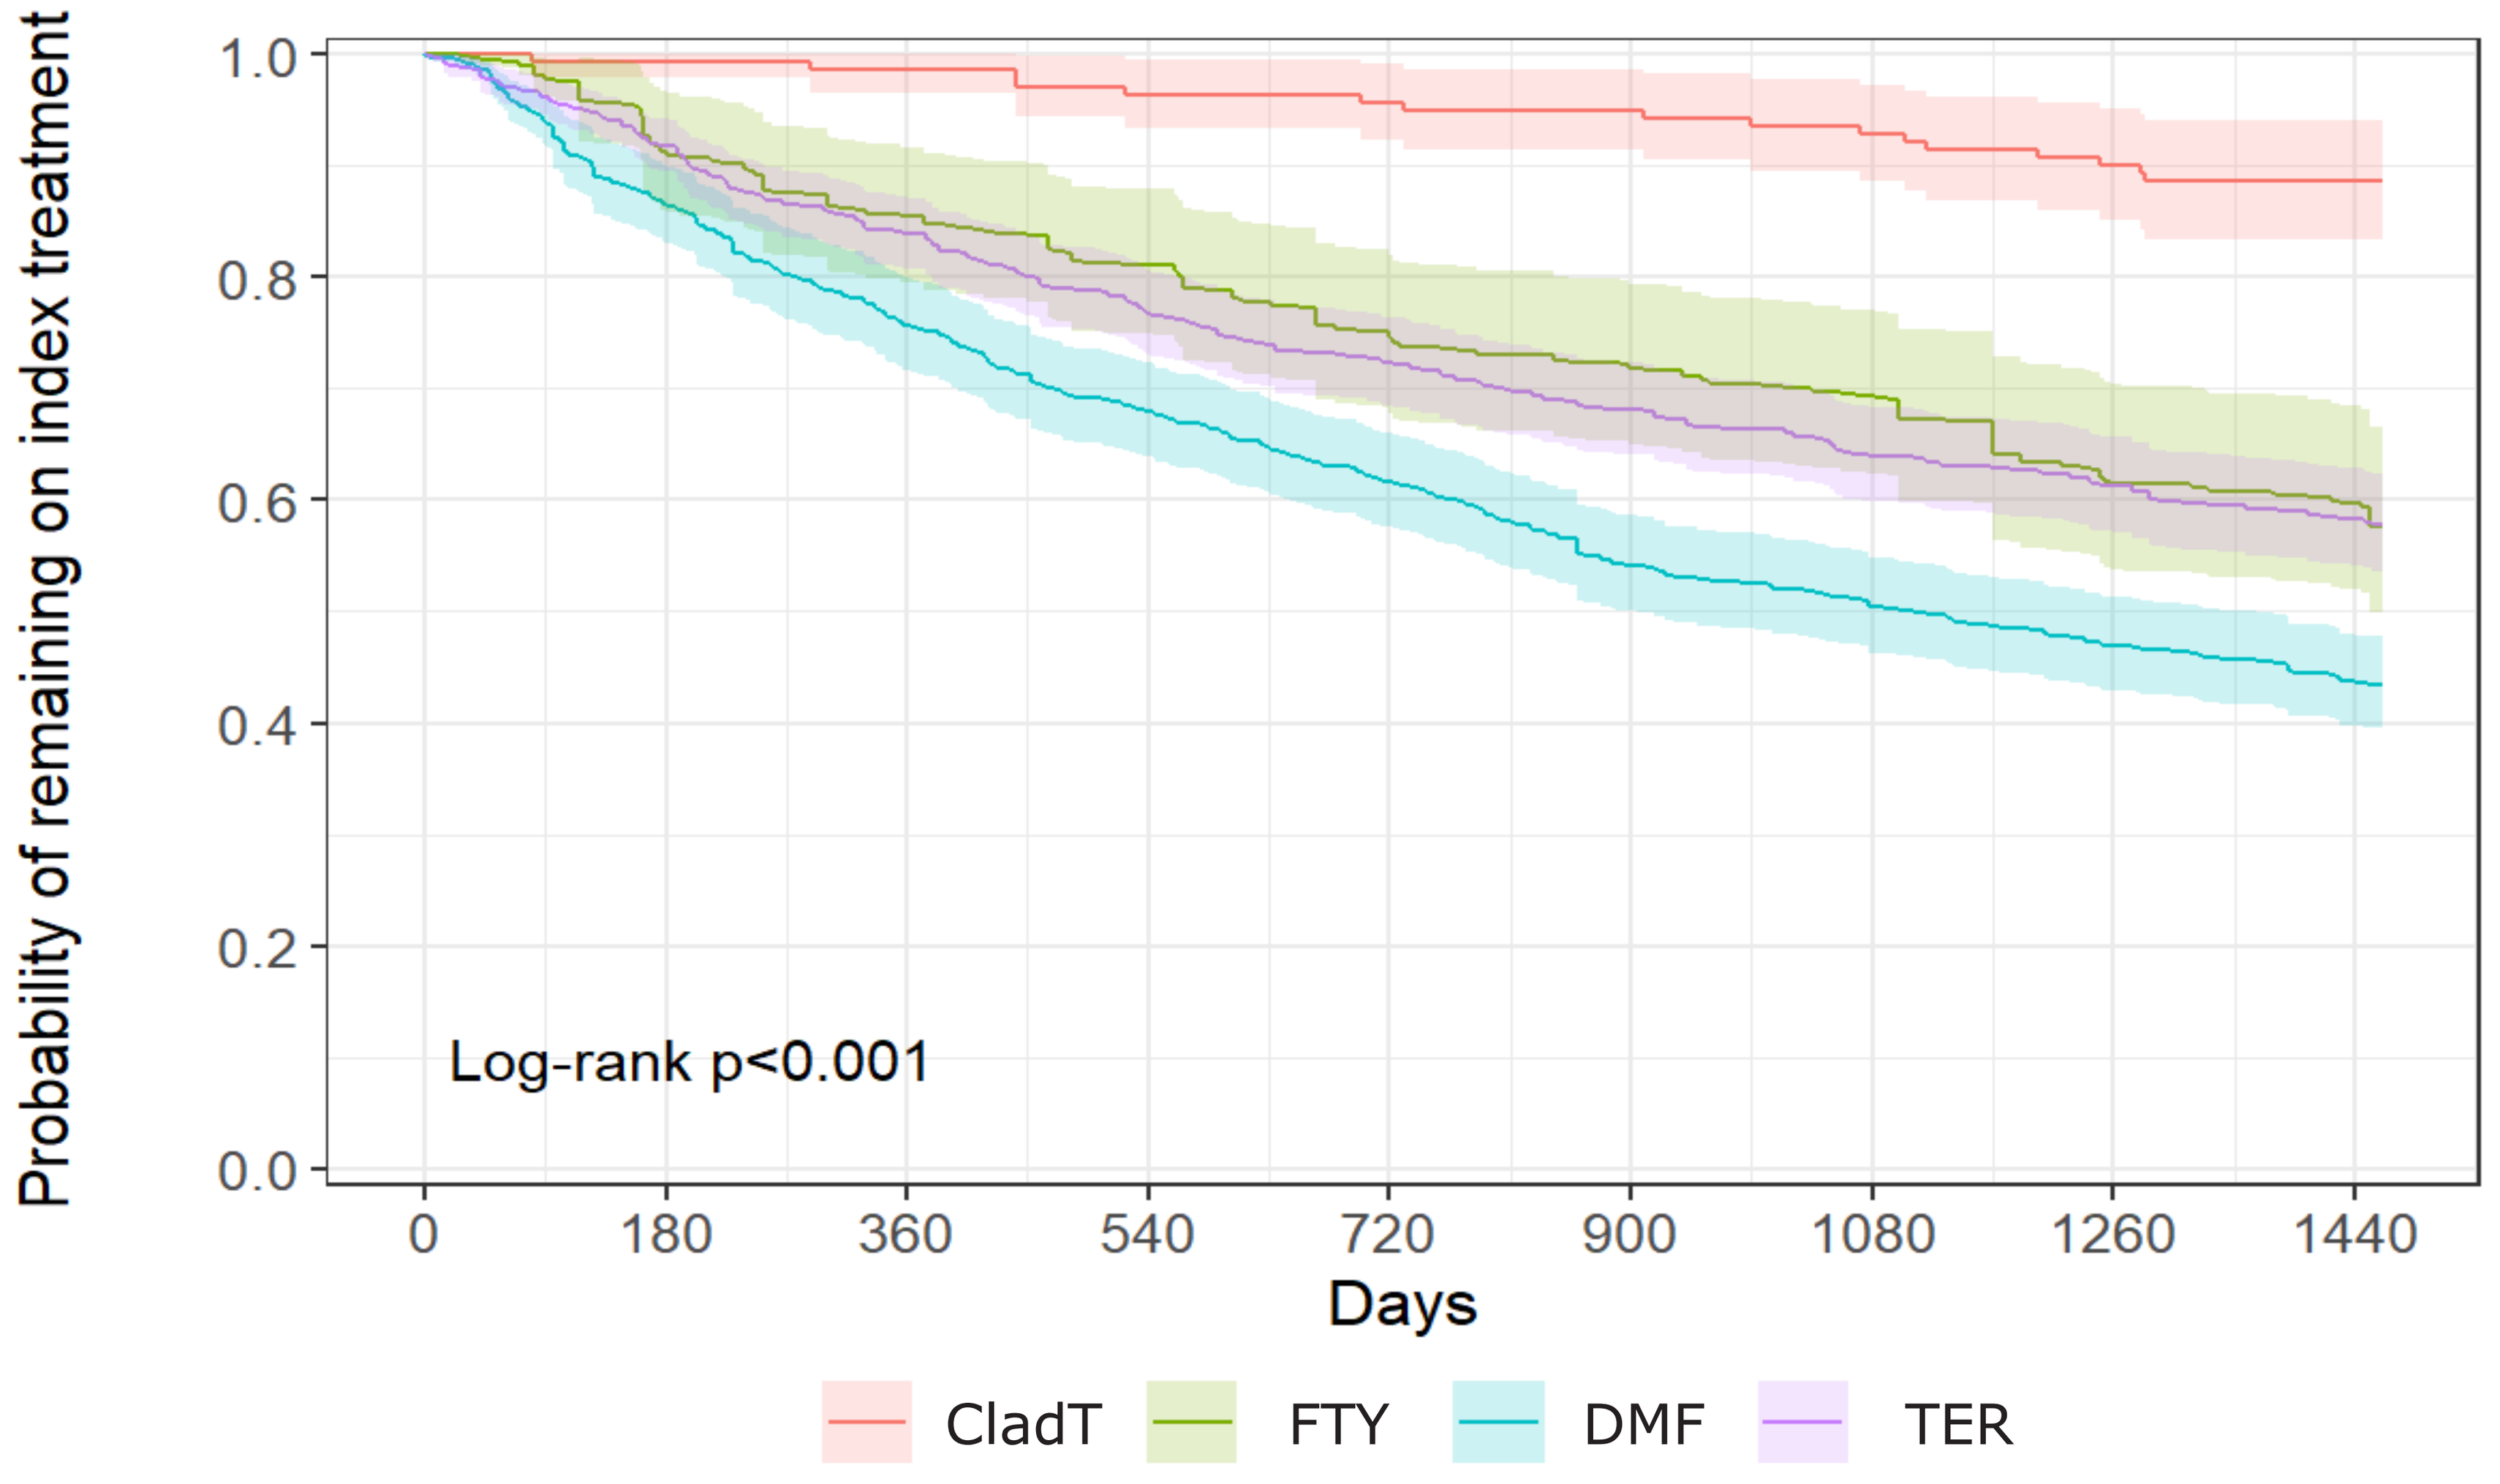
Probability of remaining on index treatment
1.0
0.8
0.6
0.4
0.2
0.0
Log-rank p<0.001
0
180
360
540
720
900
1080
1260
1440
Days
CladT
FTY
DMF
TER

At Risk

CladT	140	139	138	135	134	133	130	126	124
FTY	137	125	117	111	102	99	95	84	82
DMF	141	122	107	96	87	76	71	66	61
TER	140	129	118	108	101	96	90	86	82

The shaded portions indicate 95% confidence bands.
CladT, cladribine tablets; DMF, dimethyl fumarate; FTY, fingolimod; TER, teriflunomide.