Neurofilament Light Chain Levels and Disease Activity During Long-term Treatment of Relapsing Multiple Sclerosis With the Bruton's Tyrosine Kinase Inhibitor Evobrutinib

Jens Kuhle[1], Ludwig Kappos[2], Xavier Montalban[3], Pascal Benkert[4], Ying Li[5], Karthinathan Thangavelu[5], Yann Hyvert[6], Davorka Tomic[7]

[1]Neurologic Clinic and Policlinic, MS Center and Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB), University Hospital Basel, and University of Basel, Basel, Switzerland; [2]Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB), Departments of Medicine, Clinical Research and Biomedical Engineering, University Hospital Basel, and University of Basel, Switzerland; [3]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitario Vall d'Hebron, Barcelona, Spain; [4]Department of Clinical Research, University Hospital Basel, University of Basel, Basel, Switzerland; [5]EMD Serono, Billerica, MA, USA; [6]The healthcare business of Merck KGaA, Darmstadt, Germany; [7]Ares Trading SA, Eysins, Switzerland, an affiliate of Merck KGaA, Darmstadt, Germany.

GET POSTER PDF AND SUPPLEMENTARY DATA

Copies of this poster obtained through a QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

CONCLUSIONS

- **Evobrutinib maintained reduced NfL levels up to Week 144 approaching close to normal reference levels**
- **Low NfL levels were associated with improved MRI and relapse outcomes, supporting the role of NfL as a prognostic marker of disease activity and a potential surrogate marker of evobrutinib treatment response**

INTRODUCTION

- Evobrutinib is a highly selective, CNS-penetrating, covalent BTK inhibitor[1-3] and is currently undergoing Phase III trials (NCT04338022 and NCT04338061) for RMS
- In the DBP (Week 0–48) of a Phase II trial (NCT02975349) in RMS patients, evobrutinib led to significant reductions in T1 Gd+ lesions versus placebo at Week 24 (primary endpoint). In patients receiving evobrutinib 75 mg BID, the ARR at Week 24 and Week 48 was 0.08 and 0.11, respectively. Evobrutinib also reduced new/enlarging T2 lesions[4]
 - The efficacy of evobrutinib observed during the DBP was maintained in the OLE phase up to Week 192 (beginning at Week 48 from the DBP baseline)[5]
- Previous analyses of the Phase II trial also revealed sustained reductions in NfL levels, a biomarker of neuroaxonal damage, with evobrutinib treatment as early as Week 12 through to Week 144[6]

OBJECTIVE

To evaluate NfL levels as a prognostic and a potential surrogate marker of evobrutinib treatment response (MRI and relapse outcomes) in patients with RMS

METHODS

- Patients who were enrolled during the DBP could enter the OLE phase at Week 48 **(Figure 1)**
- NfL levels were measured over time in the pooled OLE safety population and reported as NfL Z-scores (age- and BMI-adjusted based on control population). NfL Z-score is an expression of SDs away from the mean NfL level in a control population[8]
- Based on the NfL Z-scores at Week 48 and Week 96, patients were stratified into those with NfL Z-scores <1 and ≥1
- MRI outcomes (number of T1 Gd+ and new/enlarging T2 lesions) at Week 48 and Week 96 and qualified relapses over Weeks 48–96 were evaluated using stratified Week 48 NfL Z-scores
- MRI outcomes at Week 96 and Week 144 and qualified relapses over Weeks 96–144 were also evaluated using stratified Week 96 NfL Z-scores

Figure 1 : Phase II NfL clinical analysis

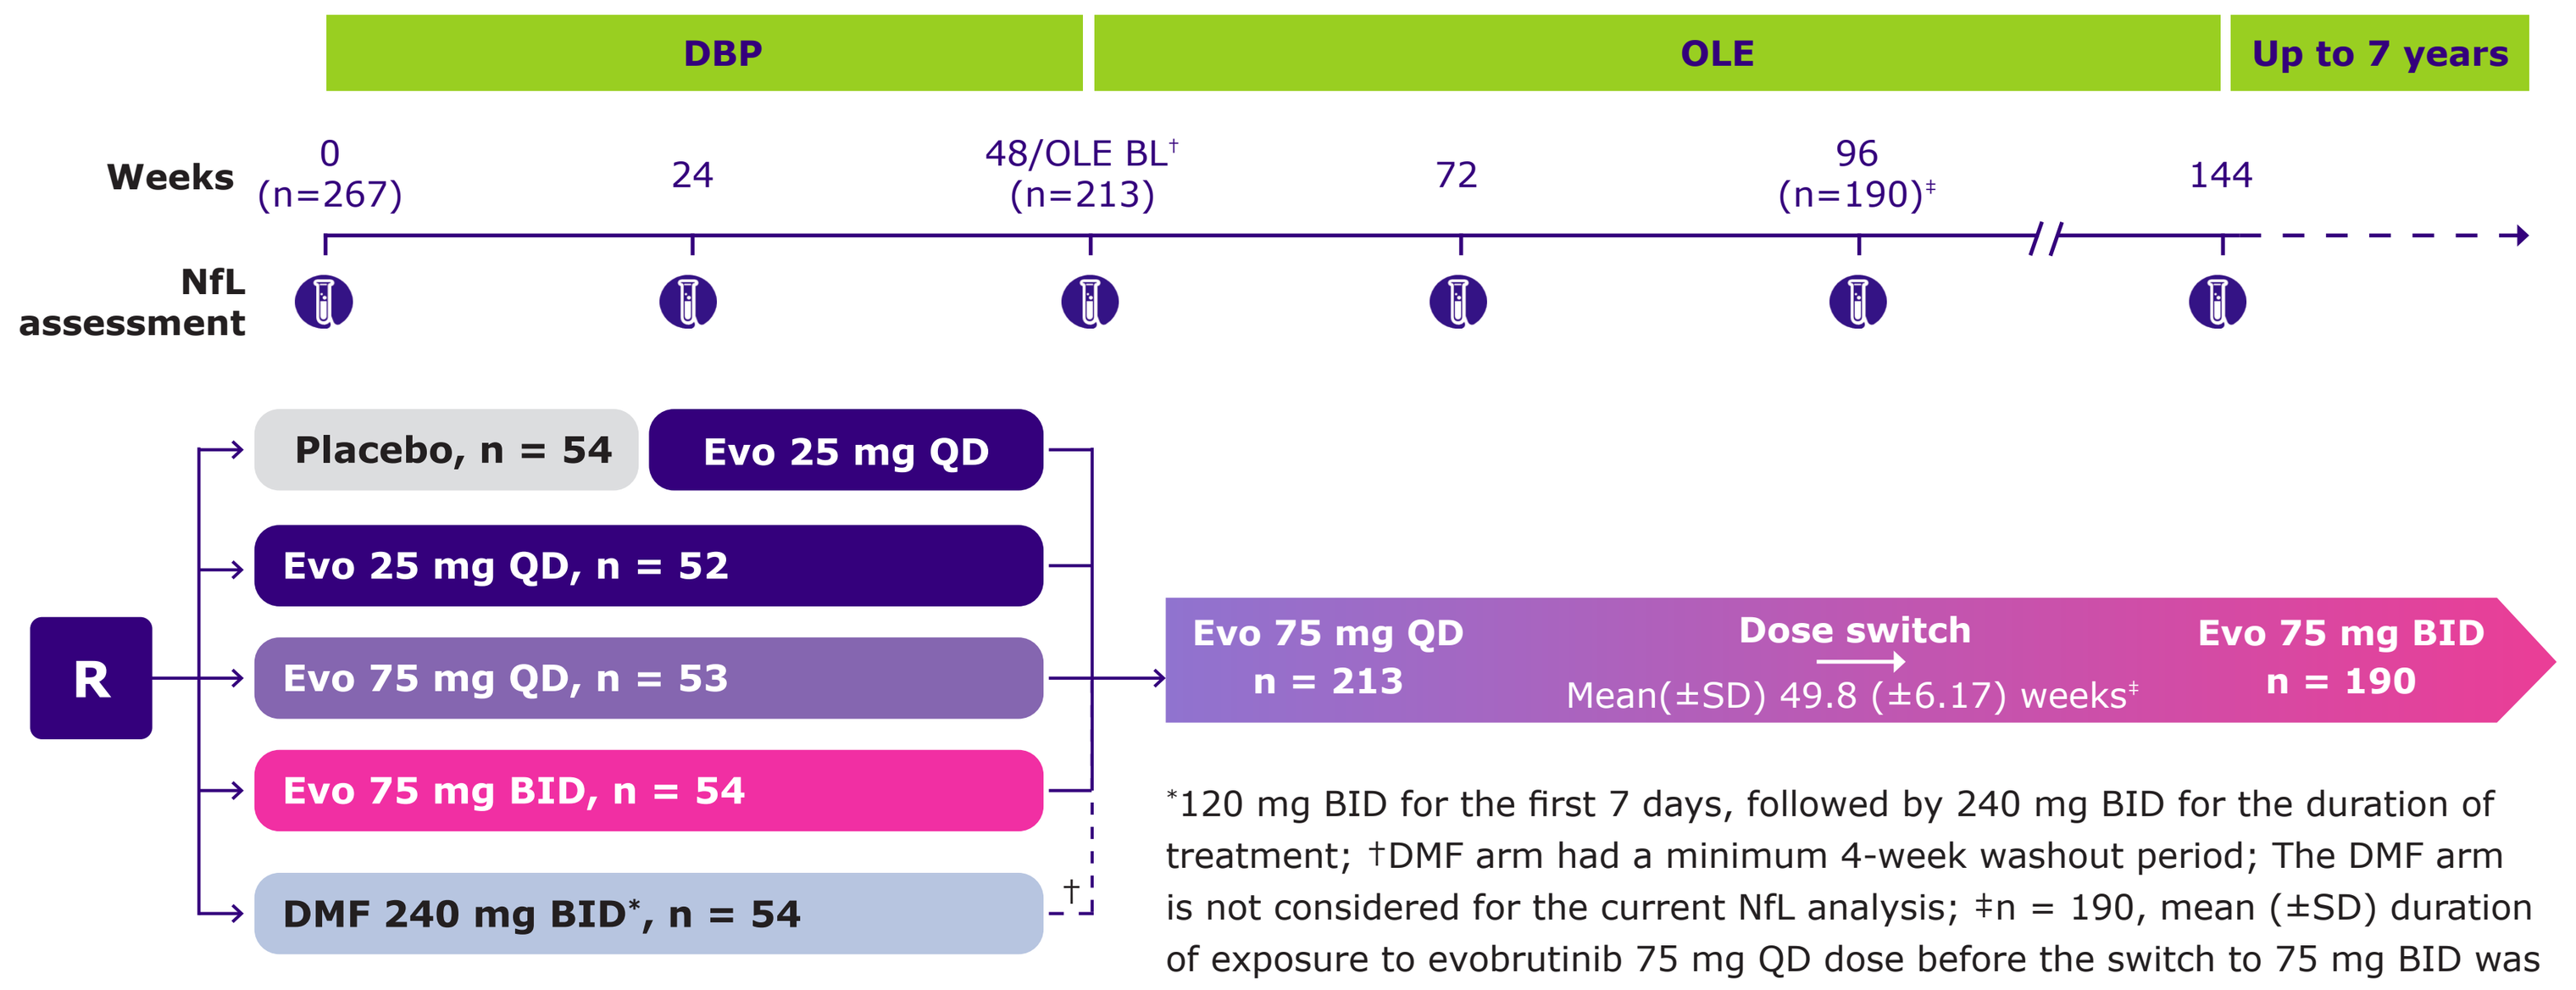
DBP
OLE
Up to 7 years
Weeks
0 (n=267)
24
48/OLE BL† (n=213)
72
96 (n=190)‡
144
NfL assessment
R
Placebo, n = 54
Evo 25 mg QD
Evo 25 mg QD, n = 52
Evo 75 mg QD, n = 53
Evo 75 mg BID, n = 54
DMF 240 mg BID*, n = 54
†
Evo 75 mg QD n = 213
Dose switch
Mean(±SD) 49.8 (±6.17) weeks‡
Evo 75 mg BID n = 190

*120 mg BID for the first 7 days, followed by 240 mg BID for the duration of treatment; †DMF arm had a minimum 4-week washout period; The DMF arm is not considered for the current NfL analysis; ‡n = 190, mean (±SD) duration of exposure to evobrutinib 75 mg QD dose before the switch to 75 mg BID was 49.8 (±6.17) weeks

Note: All evobrutinib doses were given while fasted. Evobrutinib 75 mg BID fasted dose is predicted to be comparable, with respect to exposure and BTK occupancy, to the 45 mg BID fed dose used in Phase III[7] ***(Please refer to supplementary data via the QR code for further information)***

RESULTS

mITT population

- Of 213 patients who participated in the OLE, 166 were included in the mITT population (OLE safety population with baseline and ≥1 postbaseline NfL measurement)

Effect of evobrutinib on NfL over time

- Evobrutinib reduced NfL levels in a dose-dependent manner during the DBP, and these reduced levels were maintained up to Week 144 **(Figure 2)**
- After switching to evobrutinib 75 mg BID in the OLE (mean [±SD] duration of 75 mg QD dosing in OLE: 49.8 [±6.17]) weeks, NfL levels overall and within the original DBP treatment groups were reduced to similar levels

Figure 2: The effect of evobrutinib on NfL levels* over time

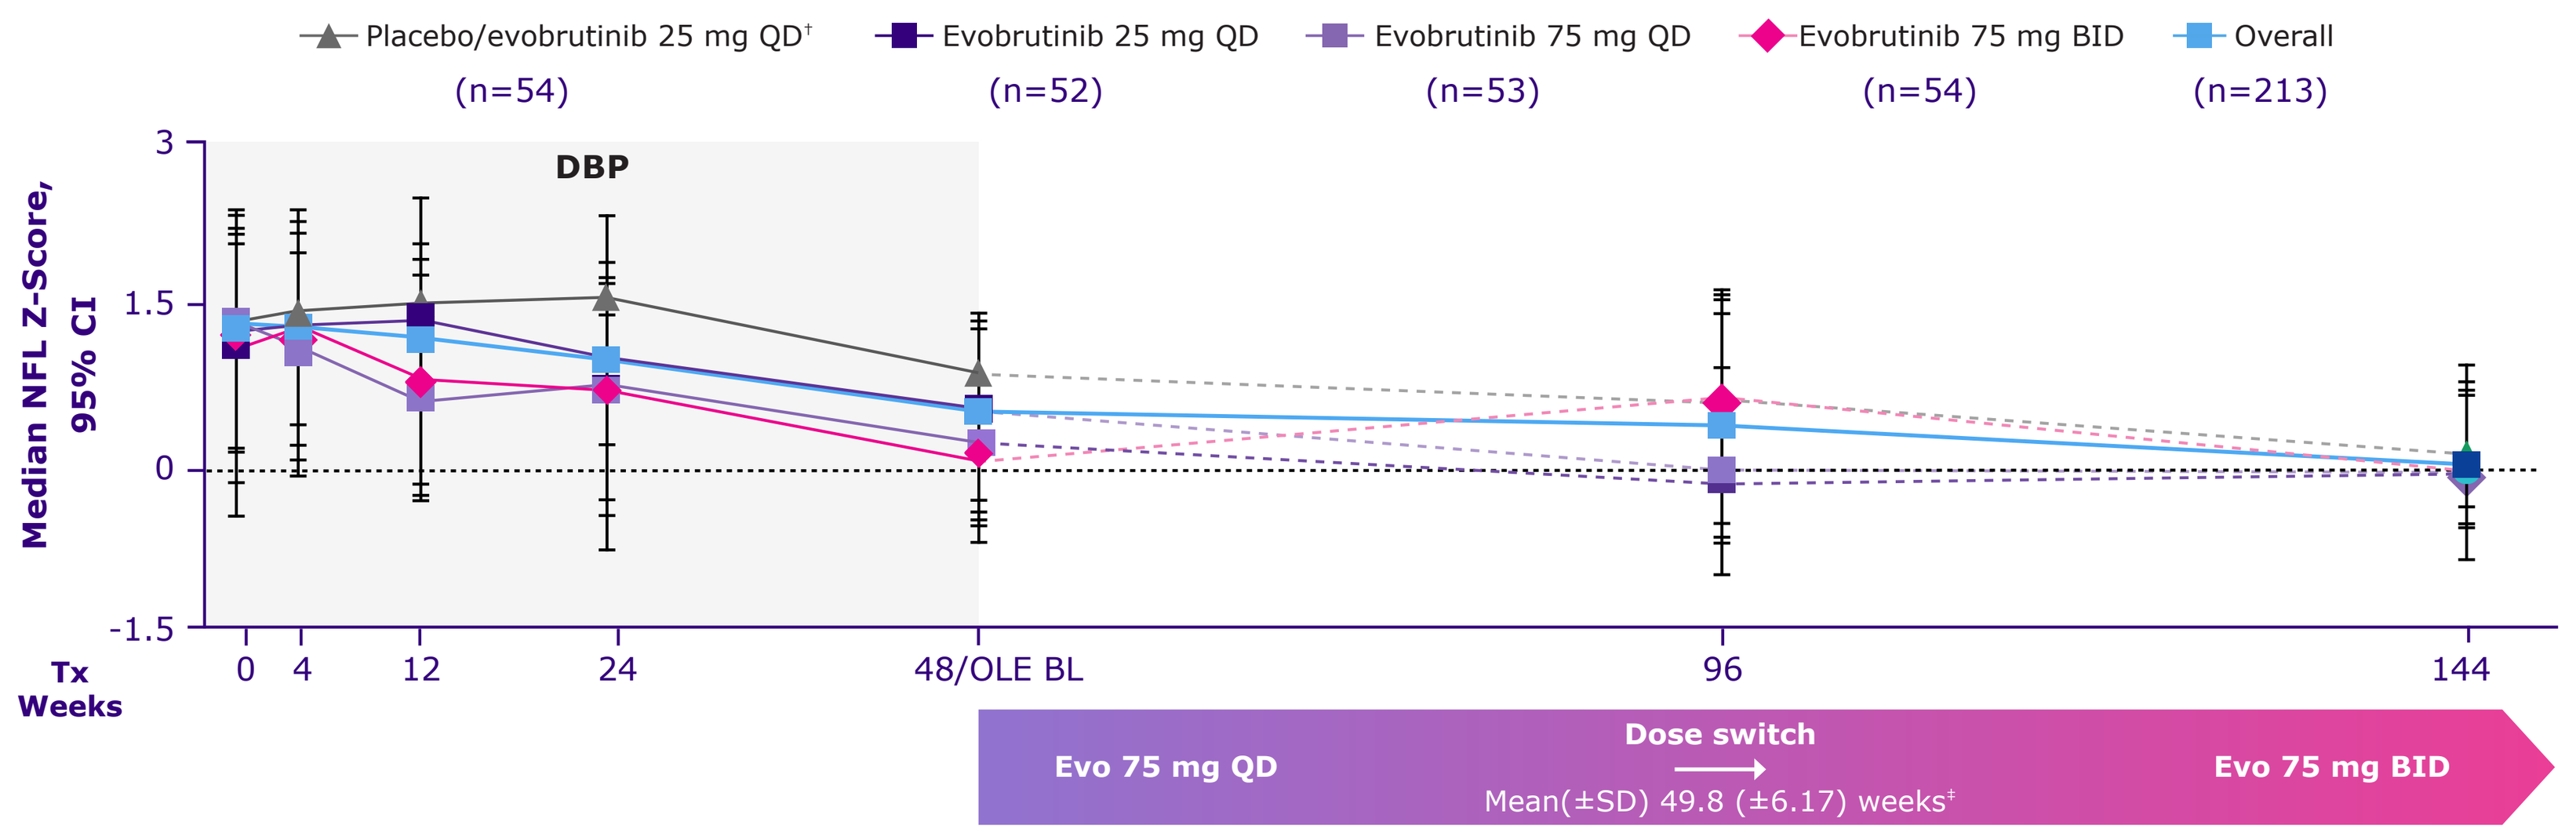
Placebo/evobrutinib 25 mg QD† (n=54)
Evobrutinib 25 mg QD (n=52)
Evobrutinib 75 mg QD (n=53)
Evobrutinib 75 mg BID (n=54)
Overall (n=213)
DBP
Median NFL Z-Score, 95% CI
3
1.5
0
-1.5
Tx Weeks
0
4
12
24
48/OLE BL
96
144
Evo 75 mg QD
Dose switch
Mean(±SD) 49.8 (±6.17) weeks‡
Evo 75 mg BID

*Reported as NfL Z-scores (age- and BMI-adjusted based on control population). The control population was defined as participants without presence of CNS disease. To derive a reference database of NfL values, a control group was developed, comprising participants with no evidence of CNS disease participating in four cohort studies in Europe and North America.[8]
†Patients switched from placebo to evobrutinib 25 mg QD after 24 weeks in the DBP; ‡n=190, mean (±SD) duration of exposure to evobrutinib 75 mg QD dose before the switch to 75 mg BID.
The dotted lines post Week 48 represent the original treatment groups during the DBP. All patients from the DBP were switched to receive the same dose of evobrutinib after Week 48 (OLE phase).

NfL Z-scores and MRI activity

- Stratified according to Week 96 NfL Z-scores (<1 [n = 66]; ≥1 [n = 34]), the proportion of patients with no T1 Gd+ lesions and no new/enlarging T2 lesions (both at Week 96 and Week 144) were higher among patients with an NfL Z-score <1 than among those with an NfL Z-score ≥1 **(Figures 3A and 3B)**
- A similar trend was observed in MRI outcomes at Week 48 and Week 96 in patients stratified according to Week 48 NfL Z-scores (<1 [n = 84]; ≥1 [n = 40]) ***(Please refer to supplementary Figures 2A and 2B via the QR code)***

Figure 3: Association between NfL Z-score and MRI activity: Stratification according to NfL Z-score category (<1 and ≥1) at Week 96

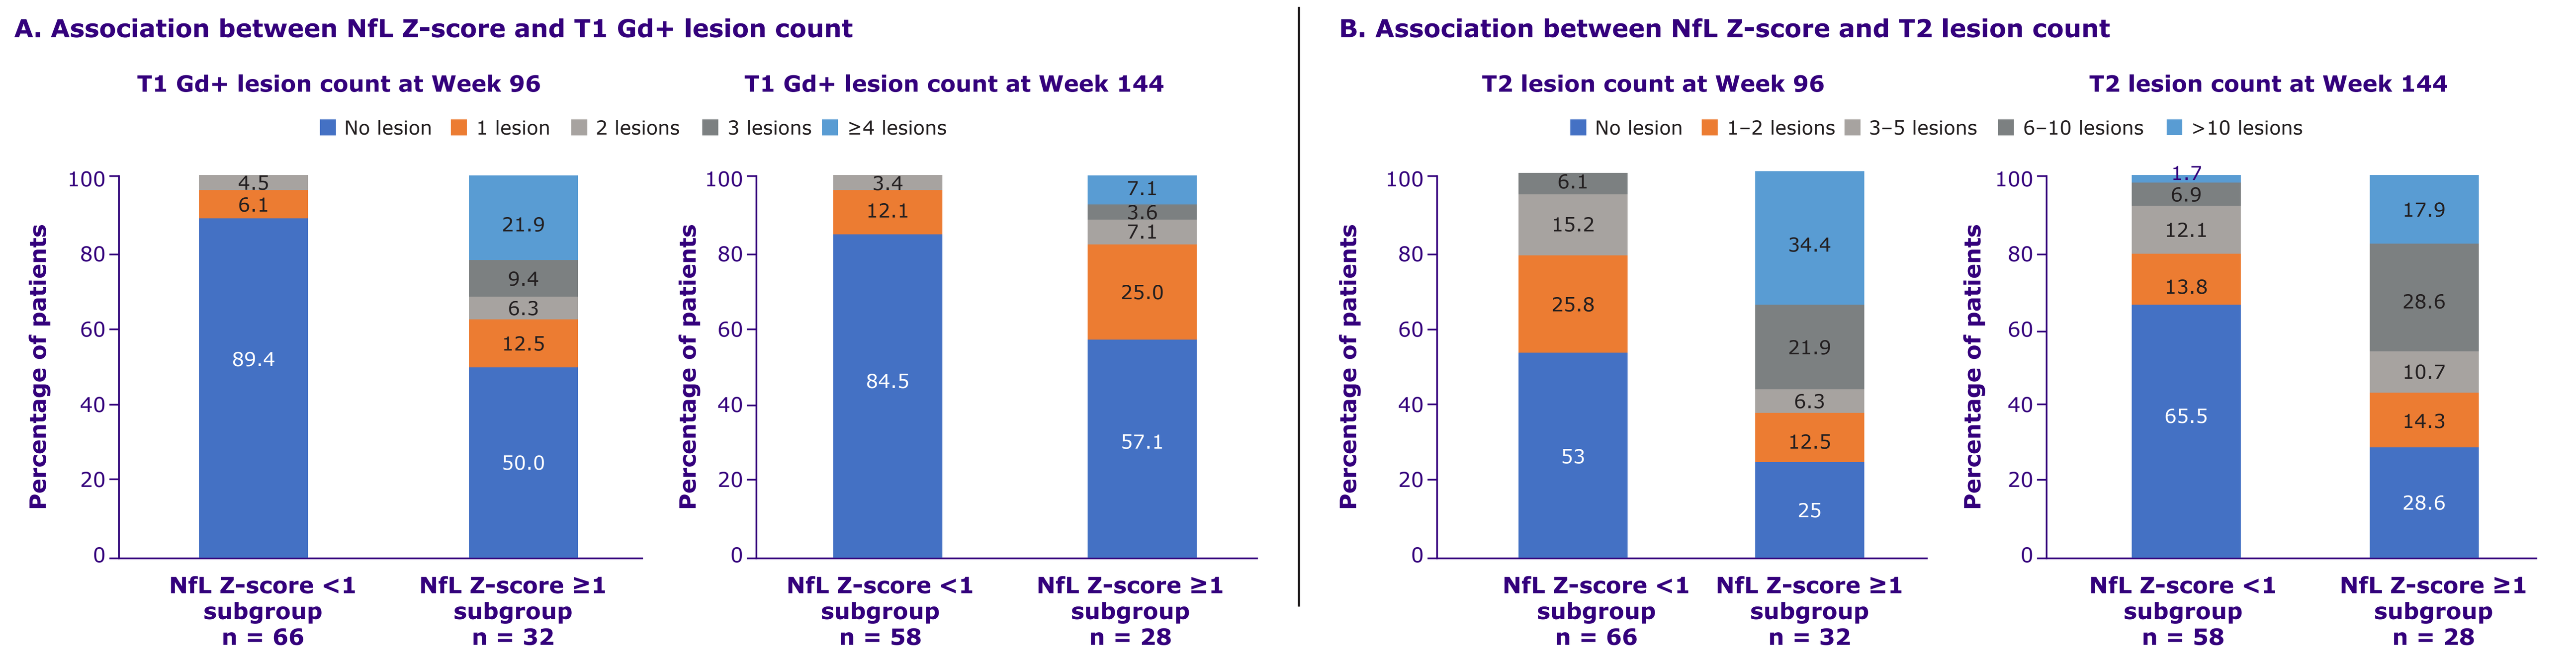
A. Association between NfL Z-score and T1 Gd+ lesion count
T1 Gd+ lesion count at Week 96
T1 Gd+ lesion count at Week 144
No lesion
1 lesion
2 lesions
3 lesions
≥4 lesions
Percentage of patients
100
80
60
40
20
0
89.4
6.1
4.5
50.0
12.5
6.3
9.4
21.9
84.5
12.1
3.4
57.1
25.0
7.1
3.6
7.1
NfL Z-score <1 subgroup n = 66
NfL Z-score ≥1 subgroup n = 32
NfL Z-score <1 subgroup n = 58
NfL Z-score ≥1 subgroup n = 28
B. Association between NfL Z-score and T2 lesion count
T2 lesion count at Week 96
T2 lesion count at Week 144
No lesion
1–2 lesions
3–5 lesions
6–10 lesions
>10 lesions
53
25.8
15.2
6.1
25
12.5
6.3
21.9
34.4
65.5
13.8
12.1
6.9
1.7
28.6
14.3
10.7
28.6
17.9
NfL Z-score <1 subgroup n = 66
NfL Z-score ≥1 subgroup n = 32
NfL Z-score <1 subgroup n = 58
NfL Z-score ≥1 subgroup n = 28

Data are presented for the overall group* (pooled population of placebo, evobrutinib 25 mg QD, evobrutinib 75 mg QD, and evobrutinib 75 mg BID treatment arms) stratified according to NfL Z-scores at Week 96. T1 Gd+ lesions and new/enlarging T2 lesions were evaluated at Week 96 and 144. *OLE safety patient population

NfL Z-scores and relapse activity

- Similar to the MRI outcomes, there was a clear segregation in the relapse activity between Week 48–96 (stratified at Week 48) where a higher proportion of patients with an NfL Z-score ≥1 experienced relapses versus those with an NfL Z-score <1 **(Figure 4A)**
- A similar albeit less pronounced trend, was observed in the relapse activity between Week 96–144 (stratified at Week 96; **Figure 4B**)

Figure 4: Association between NfL Z-score and relapse activity

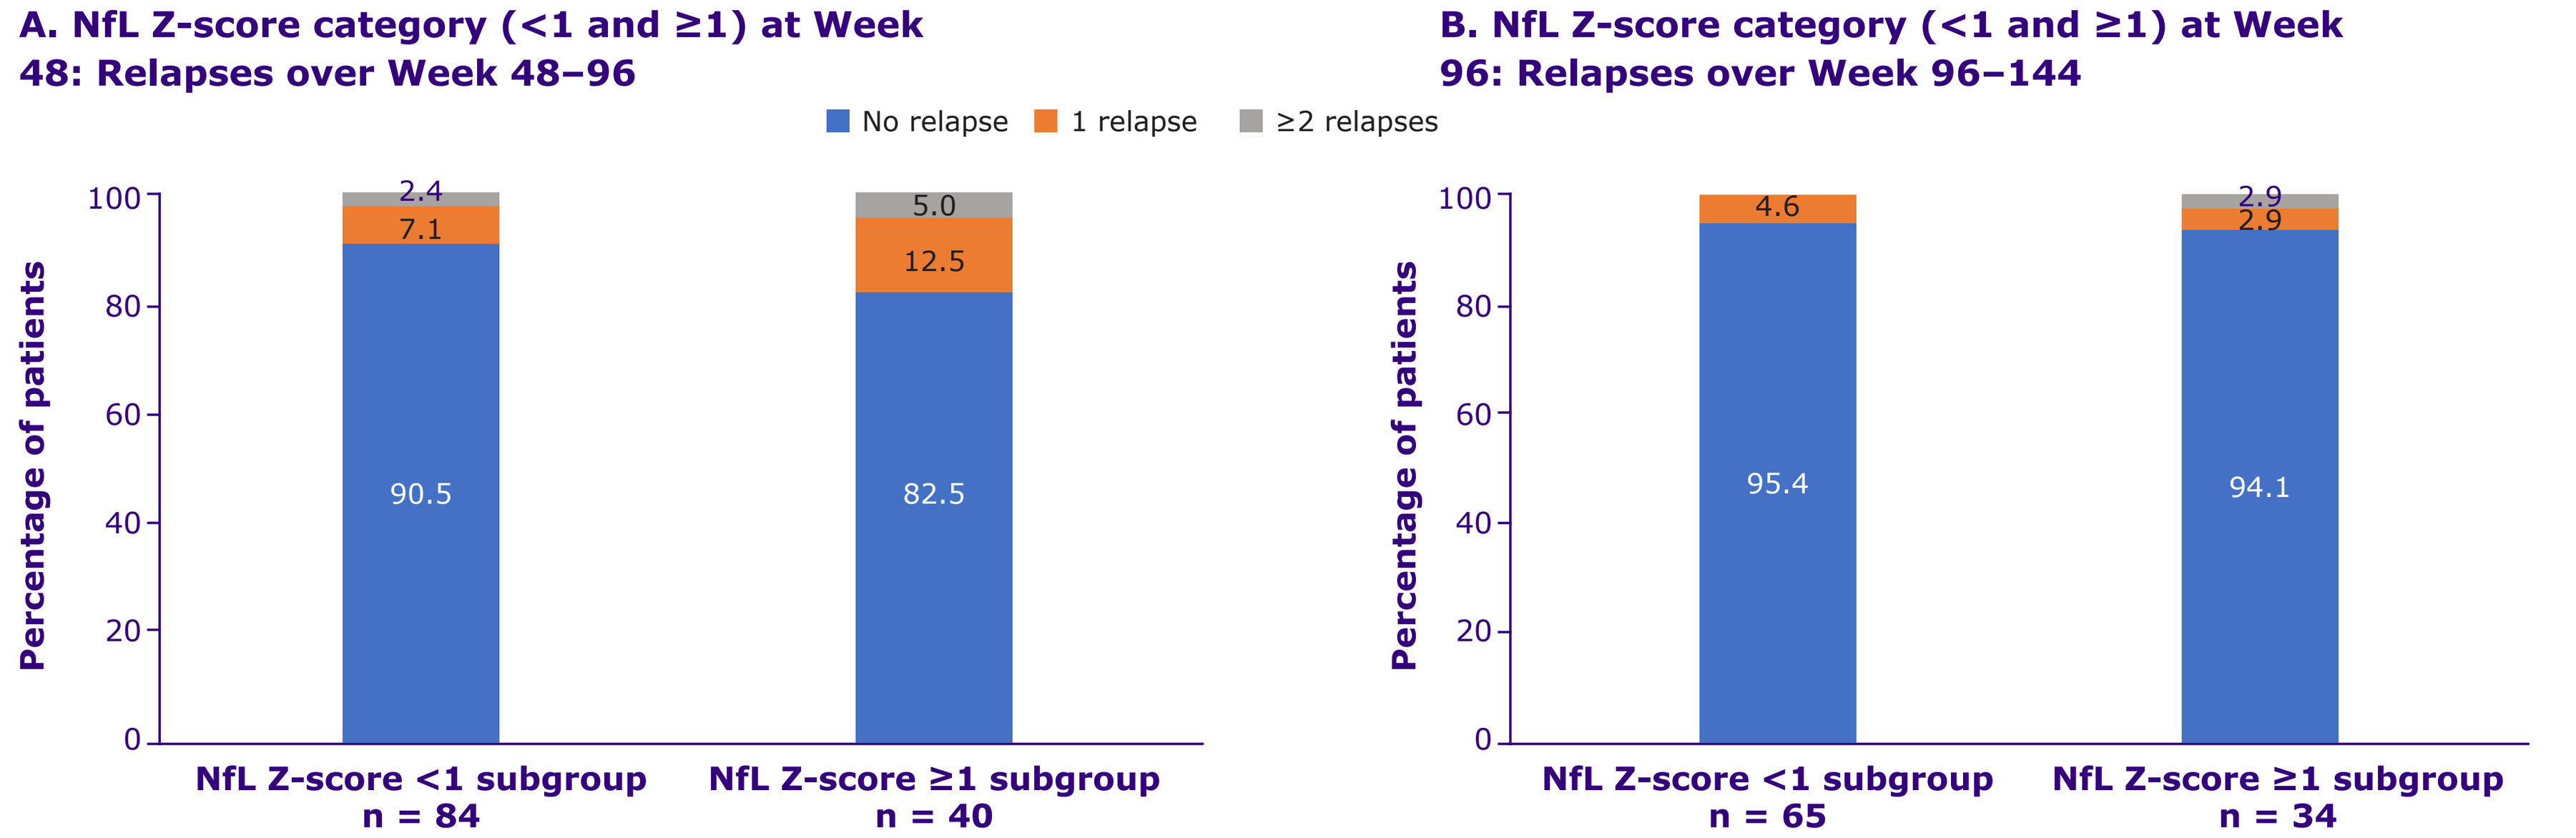
A. NfL Z-score category (<1 and ≥1) at Week 48: Relapses over Week 48–96
B. NfL Z-score category (<1 and ≥1) at Week 96: Relapses over Week 96–144
No relapse
1 relapse
≥2 relapses
Percentage of patients
100
80
60
40
20
0
90.5
7.1
2.4
82.5
12.5
5.0
95.4
4.6
94.1
2.9
2.9
NfL Z-score <1 subgroup n = 84
NfL Z-score ≥1 subgroup n = 40
NfL Z-score <1 subgroup n = 65
NfL Z-score ≥1 subgroup n = 34

Data are presented for the overall group* (pooled population of placebo, evobrutinib 25 mg QD, evobrutinib 75 mg QD, and evobrutinib 75 mg BID treatment arms) stratified according to NfL Z-scores at (A) Week 48; Number of relapses over Week 48–96 was assessed and (B) at Week 96; number of relapses over Week 96–144 was assessed. *OLE safety patient population

Abbreviations: ARR, annualized relapse rate; **BID**, twice daily; **BTK**, Bruton's tyrosine kinase; **CI**, confidence interval; **CNS**, central nervous system; **DBP**, double-blind period; **DMF**, dimethyl fumarate; **Evo**, evobrutinib; **Gd+**, gadolinium-enhancing; **mITT**, modified intention-to-treat; **MRI**, magnetic resonance imaging; **NfL**, neurofilament light chain; **OLE**, open-label extension; **QD**, once daily; **RMS**, relapsing multiple sclerosis; **SD**, standard deviation

REFERENCES: 1. Haselmayer P, et al. *J Immunol*. 2019;202:2888–2906; 2. Caldwell RD, et al. *J Med Chem*. 2019;62:7643–7655; 3. Boschert U, et al. *Mult Scler*. 2017;23(Suppl. 3):327 (P678); 4. Kuhle J, et al. *Neurology*. 2021;96:e2783–e2788; 5. Vermersch P, et al. Presented at ECTRIMS 2022 [P 731]; 6. Kuhle J, et al. Presented at ECTRIMS 2022 [EP1021]; 7. Papasouliotis O, et al. *Clin Transl Sci*. 2022;15(12):2888–2898; 8. Benkert P, et al. *Lancet Neurol*. 2022;21:246–257.

DISCLOSURES: JK's institution has received speaker fees, research support, travel support, and/or honoraria for participation in advisory boards from Swiss MS Society, Swiss National Research Foundation (320030_189140/1), University of Basel, Progressive MS Alliance, Bayer, Biogen, Celgene, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Octave Bioscience, Roche and Sanofi. **LK's** institution (University Hospital Basel) has received the following exclusively for research support: Steering committee, advisory board, and consultancy fees (Actelion, Bayer HealthCare, Biogen, BMS, Genzyme, GSK, Janssen, Japan Tobacco, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, Santhera, Shionogi, TG Therapeutics); speaker fees (Bayer HealthCare, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche and Sanofi); support of educational activities (Allergan, Bayer HealthCare, Biogen, CSL Behring, Desitin, Genzyme, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Pfizer, Sanofi, Shire and Teva); license fees for Neurostatus products; and grants (Bayer HealthCare, Biogen, European Union, InnoSwiss, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Swiss MS Society and Swiss National Research Foundation). **XM** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with Abbvie, Actelion, Alexion, Bayer, Biogen, Bristol-Myers Squibb/Celgene, EMD Serono, Genzyme, Hoffmann-La Roche, Immunic, Janssen Pharmaceuticals, Medday, the healthcare business of Merck KGaA, Darmstadt, Germany, Mylan, Nervgen, Novartis, Sandoz, Sanofi-Genzyme, Teva Pharmaceutical, TG Therapeutics, Excemed, MSIF and NMSS. **PB** has nothing to disclose. **YL and KT** are employees of EMD Serono. **YH** is/was an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **DT** is an employee of Ares Trading SA, Eysins, Switzerland, an affiliate of Merck KGaA, Darmstadt, Germany and received stock or an ownership interest from Novartis.

Medical writing assistance was provided by Nidhi Hans of Merck Specialties Pvt. Ltd., Bangalore, India, an affiliate of Merck KGaA, Darmstadt, Germany.

Evobrutinib is currently in Phase III trials for relapsing multiple sclerosis and has not yet been approved by any regulatory authority.

Presented at ACTRIMS Forum | February 23–25 | San Diego, California, USA

This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100004755).

For reactive Medical use only

February 2023

Supplementary Figure 1 : Evobrutinib dosing for the Phase III trials was optimized for exposure and BTKO levels correlating with maximal efficacy

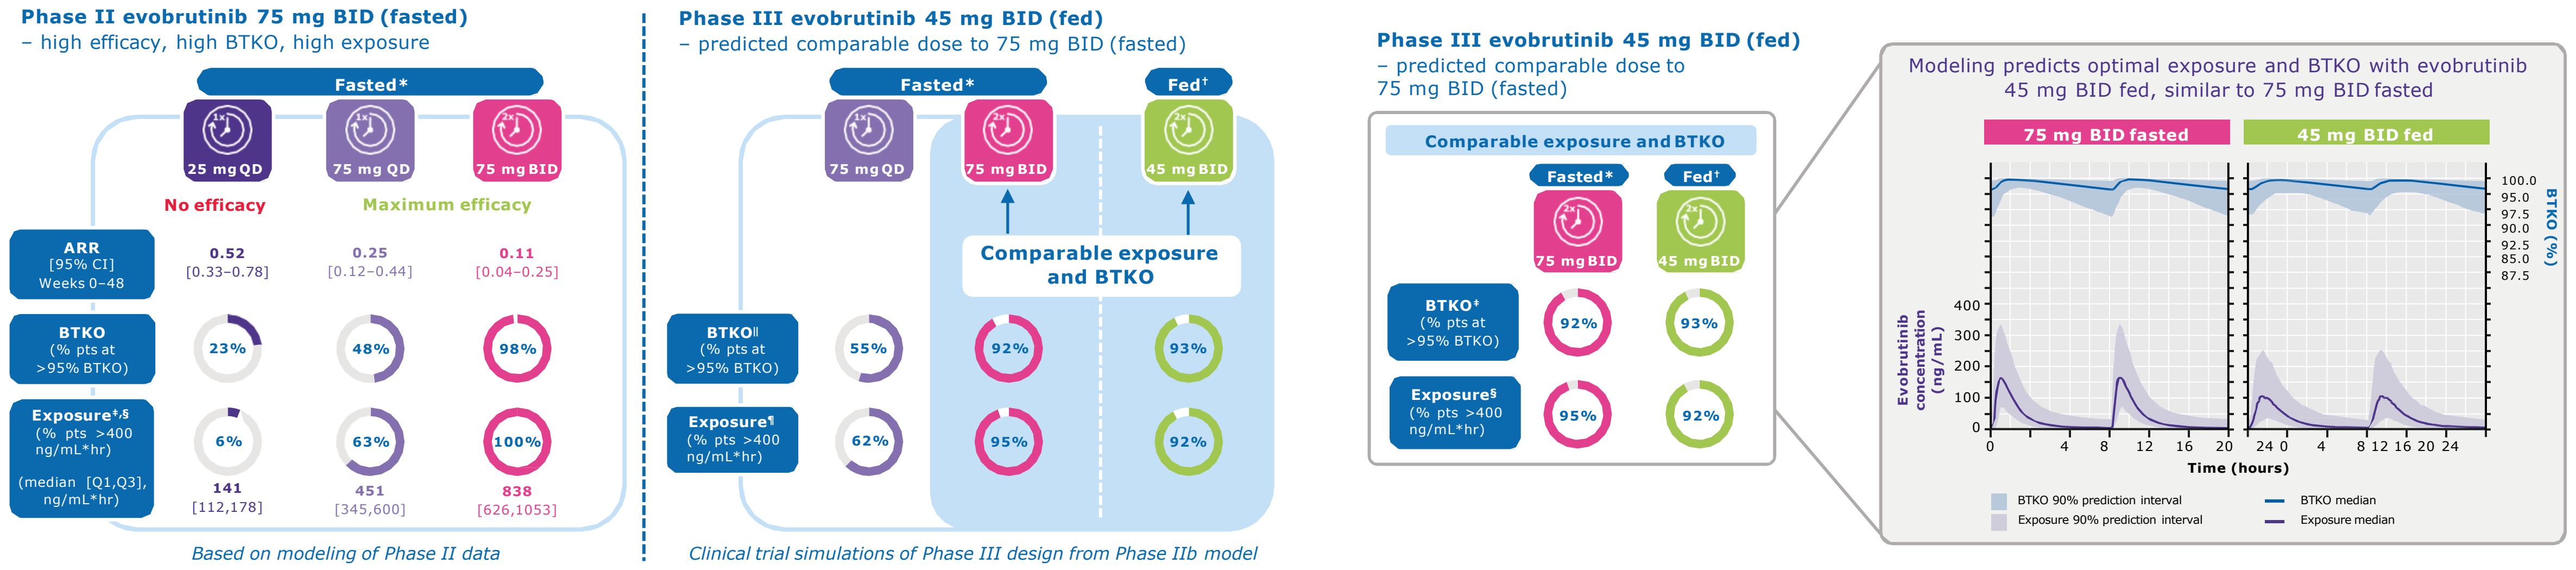

Phase II evobrutinib 75 mg BID (fasted)
– high efficacy, high BTKO, high exposure
Fasted*
25 mg QD
75 mg QD
75 mg BID
No efficacy
Maximum efficacy
ARR [95% CI] Weeks 0–48
0.52 [0.33–0.78]
0.25 [0.12–0.44]
0.11 [0.04–0.25]
BTKO (% pts at >95% BTKO)
23%
48%
98%
Exposure‡,§ (% pts >400 ng/mL*hr) (median [Q1,Q3], ng/mL*hr)
6%
63%
100%
141 [112,178]
451 [345,600]
838 [626,1053]
Based on modeling of Phase II data
Phase III evobrutinib 45 mg BID (fed)
– predicted comparable dose to 75 mg BID (fasted)
Fasted*
Fed†
75 mg QD
75 mg BID
45 mg BID
Comparable exposure and BTKO
BTKO‖ (% pts at >95% BTKO)
55%
92%
93%
Exposure¶ (% pts >400 ng/mL*hr)
62%
95%
92%
Clinical trial simulations of Phase III design from Phase IIb model
Phase III evobrutinib 45 mg BID (fed)
– predicted comparable dose to 75 mg BID (fasted)
Comparable exposure and BTKO
Fasted*
Fed†
75 mg BID
45 mg BID
BTKO‡ (% pts at >95% BTKO)
92%
93%
Exposure§ (% pts >400 ng/mL*hr)
95%
92%
Modeling predicts optimal exposure and BTKO with evobrutinib 45 mg BID fed, similar to 75 mg BID fasted
75 mg BID fasted
45 mg BID fed
Evobrutinib concentration (ng/mL)
BTKO (%)
Time (hours)
BTKO 90% prediction interval
Exposure 90% prediction interval
BTKO median
Exposure median

Measurements of evobrutinib concentration and BTKO were made in blood and PBMCs, respectively, from patients with MS in the Phase II trial
*Fasted state: >1 hour pre-meal or >2 hours post-meal; †Fed state: with food (high-fat or moderate-fat meal);
‡percentage of patients with AUC >400 ng/ml*hr over 24 hours; §Modeled maximal efficacy AUC >400 ng/ml*hr and no efficacy AUC <355 ng/ml*hr; ‖Simulated percentage of patients with BTKO >95% over 24 hours at steady state;
¶Simulated percentage of patients with AUC >400 ng/ml*hr over 24 hours
ARR is a measure of clinical efficacy, BTK occupancy indicates the fraction of evobrutinib occupying BTK (in PBMCs), **exposure** is the AUC: the area under concentration time profile of evobrutinib in the blood over 24 hours at steady state

Supplementary Figure 2 : Association between NfL Z-score and MRI activity: Stratification according to NfL Z-score category [<1 and ≥1] at Week 48

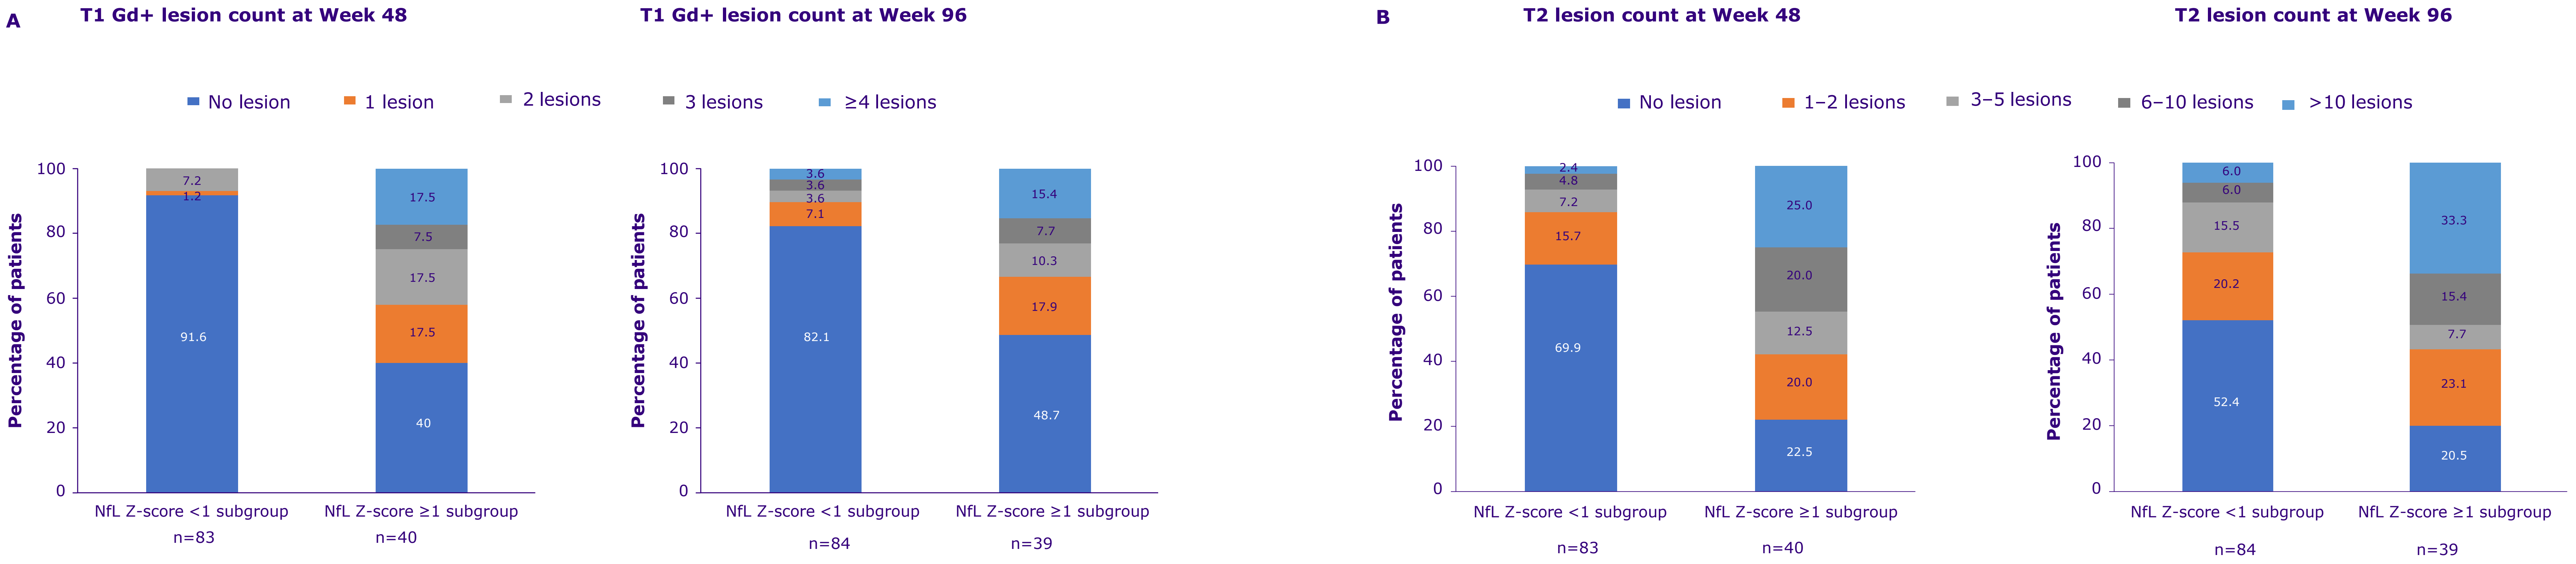

A
T1 Gd+ lesion count at Week 48
T1 Gd+ lesion count at Week 96
No lesion
1 lesion
2 lesions
3 lesions
≥4 lesions
Percentage of patients
91.6
1.2
7.2
40
17.5
17.5
7.5
17.5
NfL Z-score <1 subgroup n=83
NfL Z-score ≥1 subgroup n=40
82.1
7.1
3.6
3.6
3.6
48.7
17.9
10.3
7.7
15.4
NfL Z-score <1 subgroup n=84
NfL Z-score ≥1 subgroup n=39
B
T2 lesion count at Week 48
T2 lesion count at Week 96
No lesion
1–2 lesions
3–5 lesions
6–10 lesions
>10 lesions
69.9
15.7
7.2
4.8
2.4
22.5
20.0
12.5
20.0
25.0
NfL Z-score <1 subgroup n=83
NfL Z-score ≥1 subgroup n=40
52.4
20.2
15.5
6.0
6.0
20.5
23.1
7.7
15.4
33.3
NfL Z-score <1 subgroup n=84
NfL Z-score ≥1 subgroup n=39

Abbreviations: ARR, annualized relapse rate; **AUC**, area under the concentration–time curve; **BID**, twice daily; **BL**, baseline; **BTK**, Bruton's tyrosine kinase; **BTKO**, Bruton's tyrosine kinase occupancy; **CI,** confidence interval; **DBP**, double-blind period; **DMF**, dimethyl fumarate; **Evo**, evobrutinib; **NfL**, neurofilament light chain; **OLE**, open-label extension; **SD**, standard deviation; **QD**, once daily

For reactive Medical use only

February 2023