Core Signs Associated with the Subtypes of Cutaneous Lupus Erythematosus: Concept Elicitation Interviews with Dermatologists and Rheumatologists

Victoria P Werth[1], Annegret Kuhn[2], Joseph F Merola[3], Joerg Wenzel[4], Cristina Vazquez-Mateo[5], Sanjeev Roy[6], Erik Thomas[5], Oliver Guenther[7], Ying Sun[7], Alexandra Lauer[7], Almary Guerra Rodriguez[8], Patricia Koochaki[9], Paul Kamudoni[7]

[1]University of Pennsylvania, Philadelphia, PA, USA; [2]Amsterdam University Medical Center, Amsterdam, Netherlands, and University of Münster, Münster, Germany; [3]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [4]University Hospital of Bonn, Bonn, Germany; [5]EMD Serono, Billerica, MA USA; [6]Global Clinical Development, Ares Trading SA, Eysins, Switzerland, an affiliate of Merck KGaA, Darmstadt, Germany; [7]the healthcare business of Merck KGaA, Darmstadt, Germany; [8]Laife Reply GmbH, Frankfurt am Main, Germany; [9]ICON Clinical Research LLC, Raleigh, NC, USA

GET POSTER PDF

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission

CONCLUSIONS

- **Signs of CLE associated with disease activity may vary across subtypes and must be considered in the definition and measurement of disease activity**
- **Erythema and scaling were the most frequently reported signs related to disease activity and considered relevant across all CLE subtypes**
- **Infiltration/edema was the second most frequently evaluated sign by dermatologists, and therefore, it needs to be considered a highly relevant symptom of CLE disease**
- **While descriptions of CLE activity and damage were consistent with existing measures, the definition of remission varied slightly among clinicians**
- **Overall, this study enhances our understanding of the signs and symptoms of CLE as experienced by patients from the clinicians' perspective**

INTRODUCTION

- Cutaneous lupus erythematosus (CLE) is an autoimmune disorder that can appear as an isolated skin condition or as a manifestation of SLE[1]
- The skin manifestations and symptoms of CLE are highly variable and include photosensitivity, alopecia, scarring, pain, itching, and burning[2]
- Greater understanding of cutaneous signs and symptoms is needed to comprehensively define and measure CLE disease activity

OBJECTIVE

We interviewed dermatologic and rheumatologic experts:

- **To explore key signs and symptoms relevant for defining disease activity and damage in CLE subtypes**
- **To explore potential differences and similarities in clinical signs and symptoms across CLE subtypes**

METHODS

- This was a non-interventional, mixed-methods research study, integrating qualitative interviews and latent consensus assessments
- Board-certified dermatologists (n=11) and rheumatologists (n=10) based in the United States and Europe with ≥5 years' experience in treating CLE were interviewed remotely using a semi-structured interview guide (**Figure 1**)

Figure 1. Data collection and analysis

Question	Data collection	Analysis
Understand known / novel concepts of interest relevant for disease activity and damage	**Clinician interviews** • Semi-structured interviews lasting 60 minutes • interviews were conducted by trained researchers	**Thematic analysis** • Summarise and organize data in rich detail through a thematic analysis approach
Evaluate the degree of professional consensus about CLE core signs, symptoms, and subtype identification	**Pile sort and ranking of symptoms** • Rank the representativeness and diagnostic importance of CLE symptoms • Classify signs/symptoms of CLE according to one of the CLE subtypes: CCLE/DLE, SCLE, or ACLE	**Consensus analysis** • Determine consensus on signs and symptoms by subtype • Identify most salient symptoms

RESULTS

Clinicians' profile

- In total, 11 dermatologists (8 from the United States and 3 from Europe) and 10 rheumatologists (5 from the United States and 5 from Europe) were interviewed one-on-one; the experience of clinicians in treating patients with CLE is presented in **Figure 2**
- Most dermatologists (91%) had treated up to 100 patients with CLE in the past 12 months whereas 50% of the rheumatologists had treated more than 100 patients with CLE during the same period (**Figure 2**)

Figure 2. Clinicians' profile

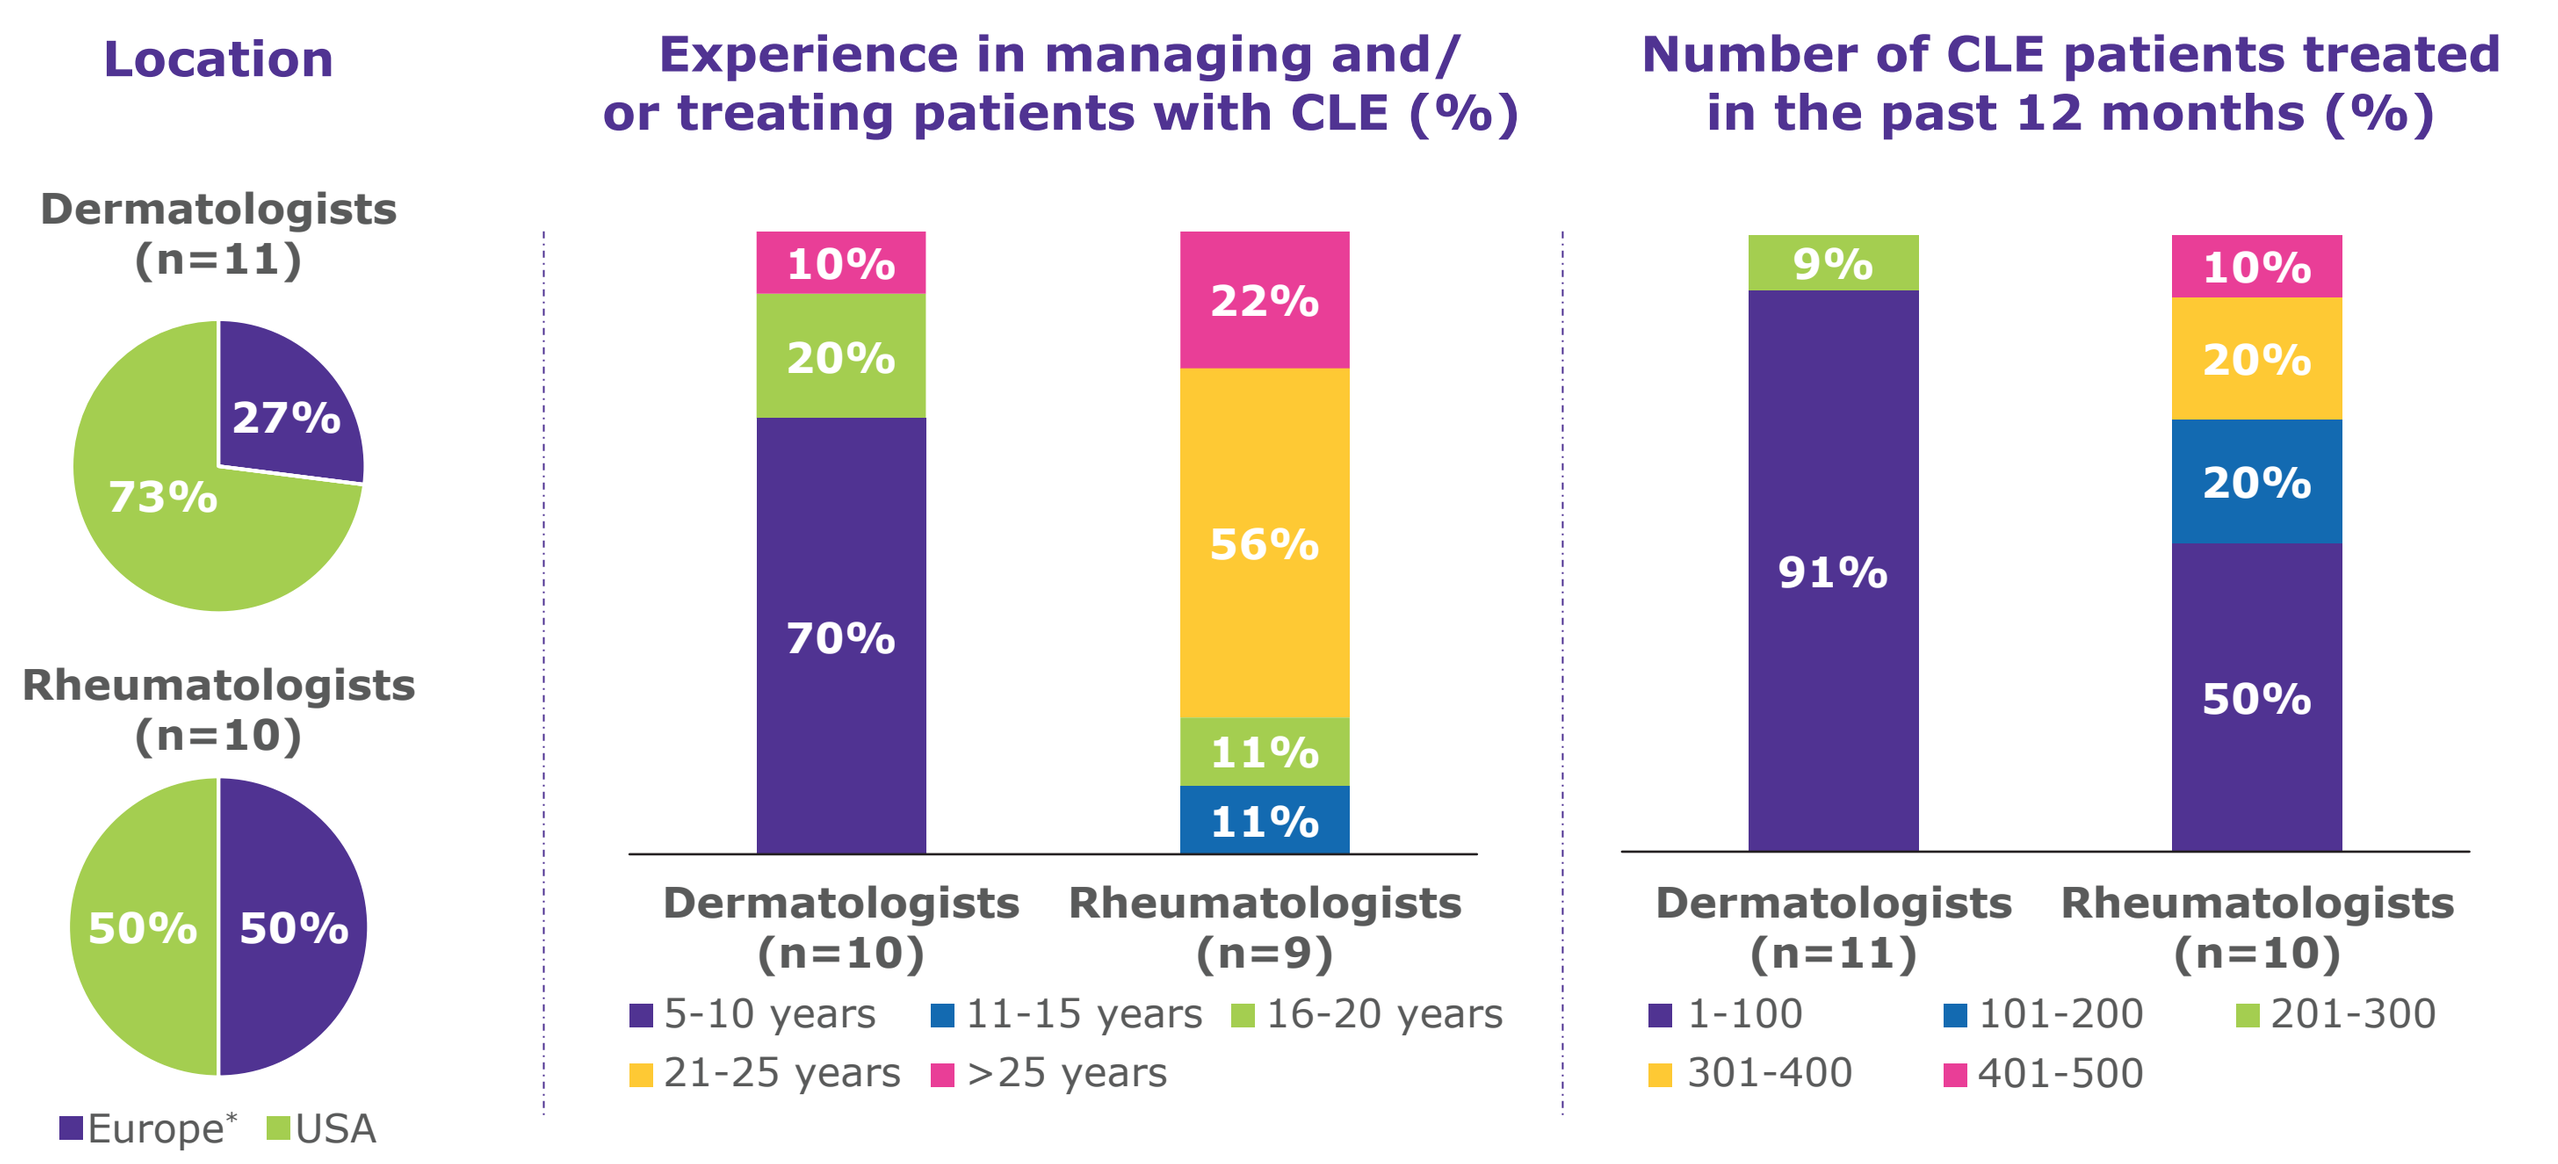
Location
Dermatologists (n=11)
27%
73%
Rheumatologists (n=10)
50%
50%
Europe* USA
Experience in managing and/or treating patients with CLE (%)
Dermatologists (n=10): 70%, 20%, 10%
Rheumatologists (n=9): 11%, 11%, 56%, 22%
5-10 years 11-15 years 16-20 years 21-25 years >25 years
Number of CLE patients treated in the past 12 months (%)
Dermatologists (n=11): 91%, 9%
Rheumatologists (n=10): 50%, 20%, 20%, 10%
1-100 101-200 201-300 301-400 401-500

Total number of clinicians=21; *Number of dermatologists and rheumatologists: Spain (0, 2), Germany (1, 0), Italy (1, 0), France (1,1), UK (0, 2)

Most frequently reported signs and symptoms of CLE

- The most frequently reported CLE-related signs by dermatologists included erythema (11/11) and infiltration/edema (10/11) followed by alopecia (scarring and non-scarring), malar rash, scaling and dyspigmentation (8/11 for each) (**Figure 3A**)
- Rheumatologists reported erythema (10/10) as the most frequently mentioned CLE-related sign, followed by alopecia (scarring and non-scarring), malar rash, scaling, dyspigmentation and infiltration/edema (**Figure 3A**)
- Based on patient history, itching was the most frequently reported symptom by dermatologists whereas photosensitivity was the most frequently reported symptom by rheumatologists (**Figure 3B**)
- Erythema and scaling were associated with all CLE subtypes, but infiltration/edema, alopecia and non-scarring alopecia varied across subtypes (**Figure 4**)

Figure 3. Most frequently reported signs and symptoms of CLE

A) Signs

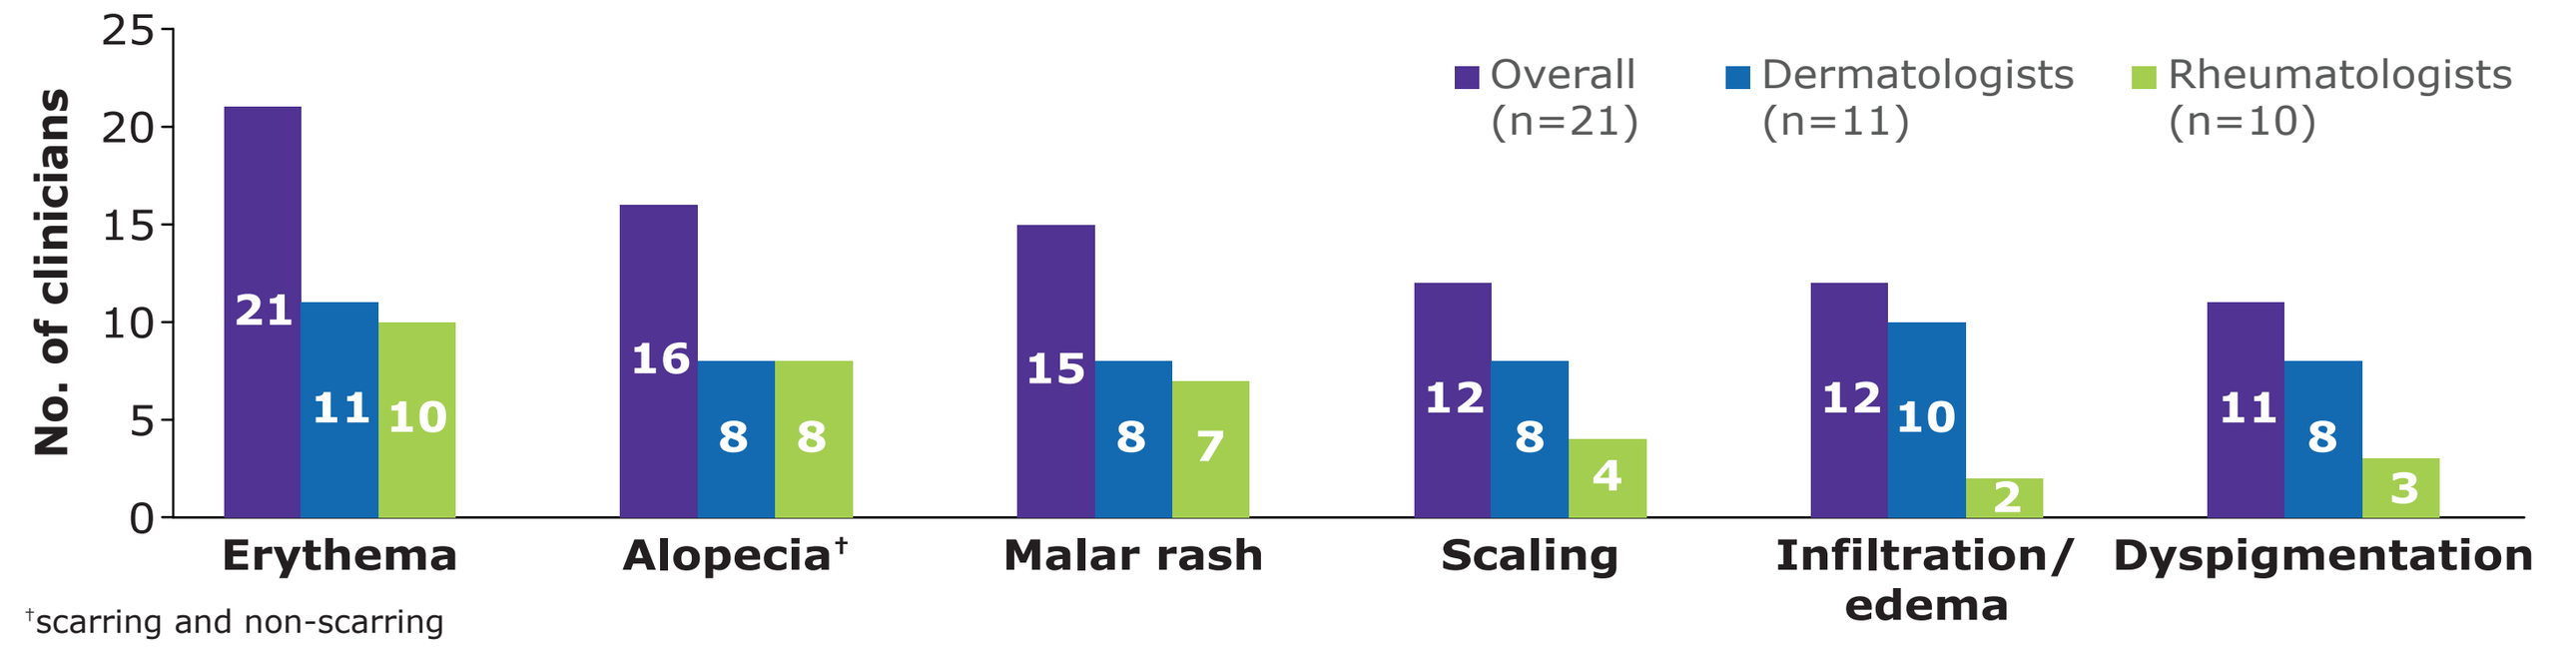
No. of clinicians
Overall (n=21) Dermatologists (n=11) Rheumatologists (n=10)
Erythema 21 11 10
Alopecia† 16 8 8
Malar rash 15 8 7
Scaling 12 8 4
Infiltration/ edema 12 10 2
Dyspigmentation 11 8 3

†scarring and non-scarring

B) Symptoms

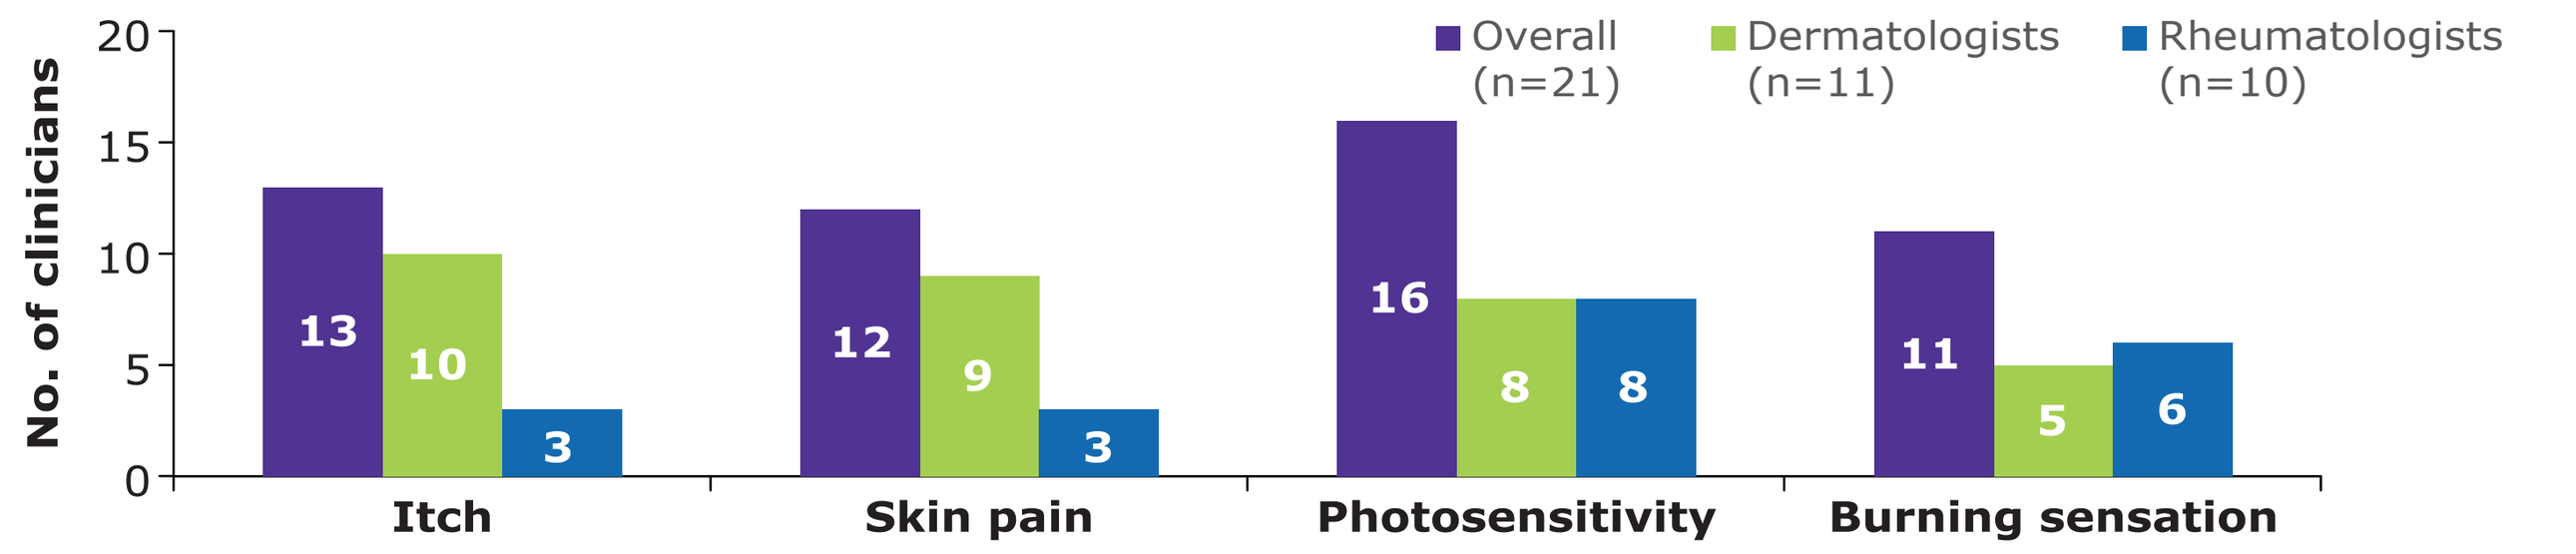
No. of clinicians
Overall (n=21) Dermatologists (n=11) Rheumatologists (n=10)
Itch 13 10 3
Skin pain 12 9 3
Photosensitivity 16 8 8
Burning sensation 11 5 6

Figure 4. Signs of CLE subtypes as per clinicians‡

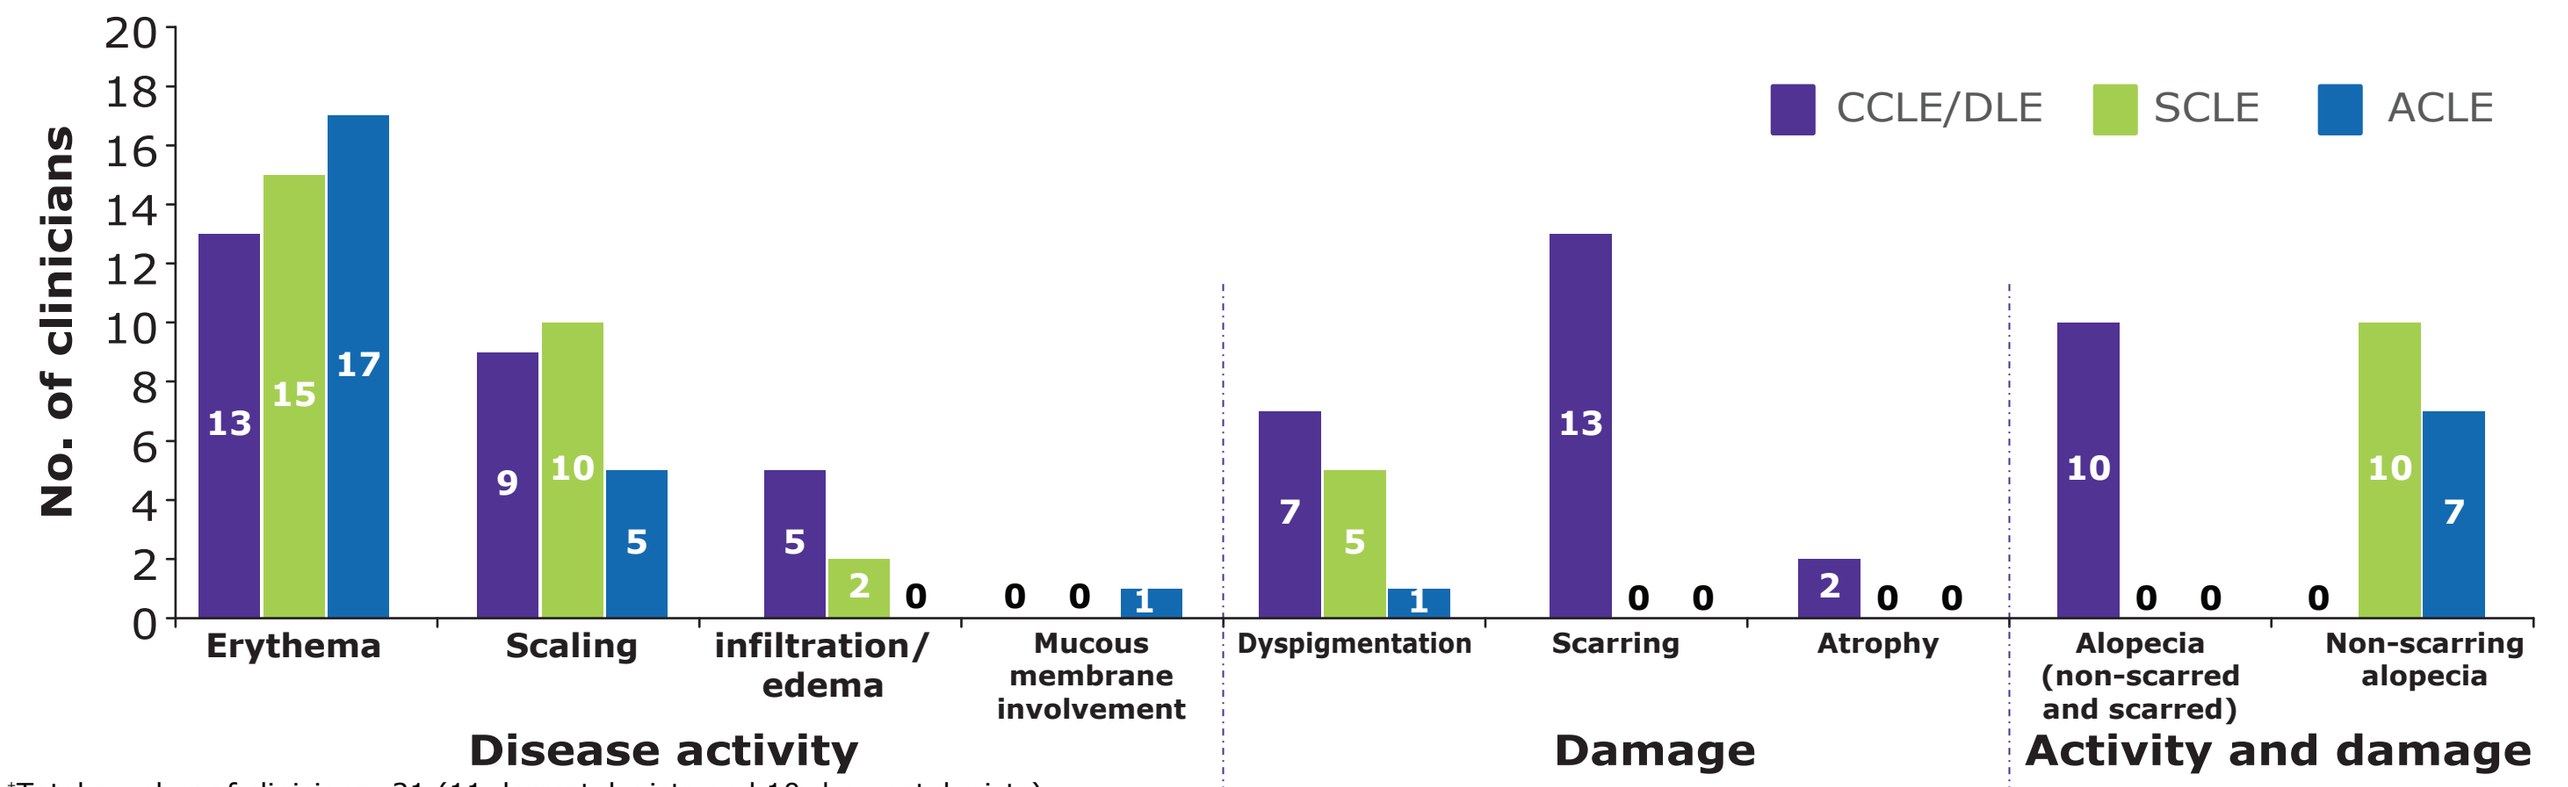
No. of clinicians
CCLE/DLE SCLE ACLE
Erythema 13 15 17
Scaling 9 10 5
infiltration/ edema 5 2 0
Mucous membrane involvement 0 0 1
Disease activity
Dyspigmentation 7 5 1
Scarring 13 0 0
Atrophy 2 0 0
Damage
Alopecia (non-scarred and scarred) 10 0 0
Non-scarring alopecia 0 10 7
Activity and damage

‡Total number of clinicians=21 (11 dermatologists and 10 rheumatologists)

CLE disease progression

- Most clinicians mentioned that CLE progression is often unpredictable, varied across patients and may be influenced by multiple factors including CLE subtype, sun exposure, smoking and medication compliance
- The clinical experts characterised remission as the absence of signs and symptoms, with or without medication
- CLE remission is negatively influenced by sun exposure and smoking, whereas medication compliance positively impacts duration of remission

Abbreviations: ACLE, acute cutaneous lupus erythematosus; **CLE,** cutaneous lupus erythematosus; **CCLE,** chronic cutaneous lupus erythematosus; **DLE,** discoid lupus erythematosus; **SCLE,** subacute cutaneous lupus erythematosus; **SLE,** systemic lupus erythematosus.

References: 1. Wenzel J. *Nature Reviews Rheum.* 2019 15(9):519–532; 2. Ogunsanya ME, et al. *Lupus.* 2020;29(13):1691–1703

Author disclosures: VPW is a consultant in Celgene, MedImmune, Resolve, Genentech, Idera, Janssen, Lilly, Biogen, Bristol Myers Squibb, Gilead, Amgen, Medscape, Nektar, Incyte, EMD Serono, CSL Behring, Principia, Crisalis, Viela Bio, Argenx, Kirin, AstraZeneca, AbbVie, GlaxoSmithKline, Cugene, UCB, Corcept and Beacon Bioscience; and has received research support from Celgene, Janssen, Biogen, Gilead, AstraZeneca, Viela, Amgen and Lupus Research Alliance/Bristol Myers Squibb. **AK** has received consultancy fees from Biogen, GlaxoSmithKline (GSK), Grünenthal, and Lilly. She has received reimbursement of costs for conference attendance, travel, and accommodation as well as payments for the preparation of scientific meetings from Basilea, Biogen, GSK, La Roche Posay, Lilly, and Spirig Pharma GmbH. **JFM** is a consultant and/or investigator for EMD Serono, AbbVie, Dermavant, Eli Lilly, Novartis, Janssen, UCB, Celgene, Sanofi, Regeneron, Arena, Sun Pharmaceuticals, Biogen, Pfizer, EMD Serono, Avotres, and LEO Pharma. **JW** has received financial support from GSK, Incyte, and Spirig for investigator-initiated trials; GSK, Roche, Leo, EMD Serono, and ArrayBio for clinical studies; and GSK, Medac, Actelion, Celgene, Biogen, Roche, and Leo for serving on advisory boards. **CVM** and **EK** are the employees of EMD Serono. **SR** is an employee of Merck, Ares Trading SA, Eysins, Switzerland, an affiliate of Merck KgaA. **OG, YS, AL,** and **PK** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **AGR** is a consultant in Laife Reply GmbH, Frankfurt, Germany. **PKo** is an employee of ICON plc, Cincinnati, OH USA

This study was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Medical writing and editorial support were provided by Khasim Shaik of Merck Specialties Pvt. Ltd., Bangalore, India, the healthcare business of Merck KGaA, Darmstadt, Germany. This data was previously published by or presented at EADV 2023

Presented at the ACR 2023 congress | San Diego, California| 10–15 November 2023

Copyright © 2023 remains with the authors

Correspondence: victoria.werth@pennmedicine.upenn.edu