Effect of extended treatment with IAP inhibitor xevinapant post radiotherapy on efficacy and the tumor microenvironment in preclinical models

T.-L. Yeung,[1] F. Jiang,[1] J. Qi,[1*] H. Yu,[1] L.-Y. Chiu,[1] B. Marelli,[1] H. Wang,[1*] M. Bonnemaison,[1] M. Jenkins,[1*] R. Ferretti,[1] R. Lindemann,[2] C. Esdar,[2] Y. Lan[1*]

[1]EMD Serono, Billerica, MA, USA; [2]The healthcare business of Merck KGaA, Darmstadt, Germany.
*Affiliation at the time of the study.

SCOPE

- We evaluated preclinically the effect of extended xevinapant treatment post concurrent radiotherapy (RT) on antitumor efficacy and tumor microenvironment modulation in MC38 syngeneic tumor and in vitro cell culture models

CONCLUSIONS

- The combination of xevinapant and RT demonstrated improved antitumor efficacy in the MC38 tumor model compared with RT alone
- MC38 syngeneic tumor-bearing mice that received extended dosing of xevinapant post concurrent RT, compared with treatment arms with shorter xevinapant dosing durations, showed improved antitumor efficacy and prolonged survival
- Our data suggests that xevinapant + RT mediates tumor microenvironment responses, including promotion of RT-induced cancer cell apoptosis, enhanced antitumor immunity, and suppressed pro-tumorigenic phenotype of cancer-associated fibroblasts
- Additional studies are warranted to further delineate the mechanisms that underscore the therapeutic benefit offered by extended dosing of xevinapant

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ASCO and the authors of this poster.

Correspondence: **Christina Esdar, christina.esdar@emdserono.com**

SUPPLEMENTARY DATA

Please scan this QR code with your smartphone app to view supplementary data associated with this poster.

BACKGROUND

- Xevinapant is a first-in-class, oral IAP (inhibitor of apoptosis protein) inhibitor designed to restore cancer cell sensitivity to apoptosis, induced by RT and chemotherapy, and potentially enhance antitumor immunity
- In a randomized phase 2 study of patients with unresected locally advanced squamous cell carcinoma of the head and neck, xevinapant + chemoradiotherapy (CRT) significantly improved locoregional control at 18 months[1] and improved 3-year progression-free survival (HR=0.33 [95% CI, 0.17-0.67], p=0.0019) and 5-year overall survival vs placebo + CRT (HR=0.47 [95% CI, 0.27-0.84], p=0.0101)[2]
- A phase 3 confirmatory study (TrilynX, NCT04459715) and a phase 3 study to evaluate xevinapant + RT in postoperative, high-risk, cisplatin-ineligible patients (XRay Vision, NCT05386550) are ongoing. In these studies, patients receive 3 additional cycles of xevinapant or placebo monotherapy after completing combination treatment[3]
- Based on the role of IAPs in apoptosis,[4] antitumor immunity,[5,6] and stromal activation,[7,8] continual dosing of xevinapant post RT may deliver additional therapeutic benefit through modulation of multiple tumor microenvironment compartments

METHODS

- The impact of xevinapant dosing duration on antitumor efficacy and animal survival was evaluated with the intramuscular MC38 syngeneic tumor model
 - Tumor-bearing mice were treated with vehicle control, RT alone (3.6 Gy every day [QD] for 5 days), or xevinapant (100 mg/kg QD 5 days on, 2 days off for 1, 2, or 4 weeks) + RT (**Figure 1**)
- To assess treatment-mediated tumor microenvironment modulation of immune compartments by combination of xevinapant + RT, an in vivo pharmacodynamic study with MC38 tumors was performed
 - The immune compartments of tumors were evaluated by fluorescence-activated cell sorting (FACS)–based immune profiling
 - Tumor-specific T-cell response was investigated by ex vivo enzyme-linked immunosorbent spot (ELISpot) assays
- The effect of xevinapant + RT on other tumor microenvironment cell types, including cancer cells and fibroblasts, was evaluated by in vitro cell culture models
 - Proliferation assays and expression level measurement of cleaved-caspase 3 in MC38 cells treated with xevinapant + RT was performed to assess radiosensitization and induction of apoptosis
 - Cancer-associated fibroblast markers and gene expression were profiled on fibroblast cell lines to determine impact on radiation-induced cancer-associated fibroblast activation and extracellular matrix protein production by xevinapant

Figure 1. Efficacy evaluation in MC38 model

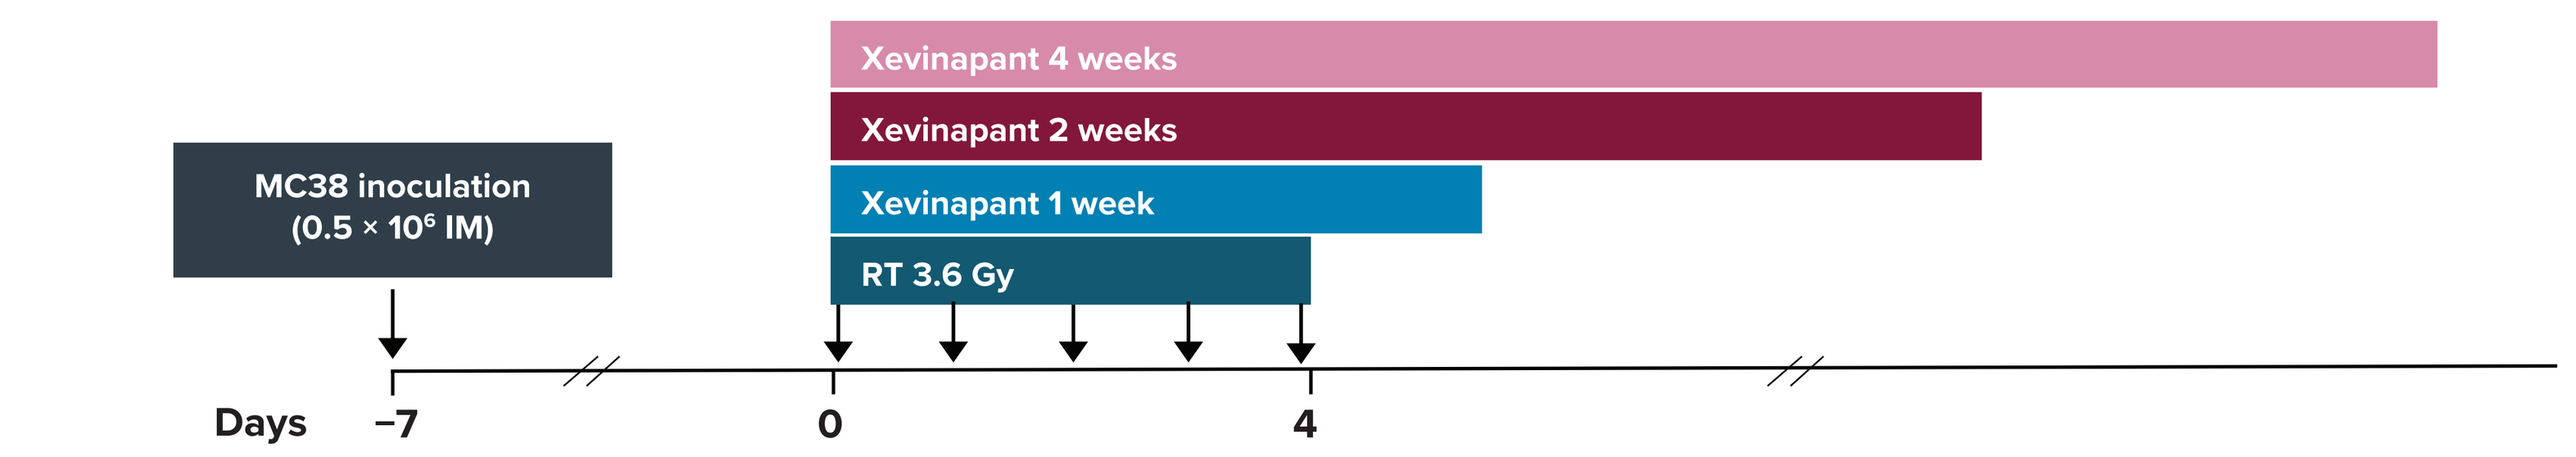
Xevinapant 4 weeks
Xevinapant 2 weeks
Xevinapant 1 week
RT 3.6 Gy
MC38 inoculation (0.5 × 10⁶ IM)
Days
−7
0
4

Treatment groups: N=10 per group

1. Vehicle
2. RT (3.6 Gy QD for 5 days) + vehicle
3. Xevinapant (100mg/kg QD 5 days on, 2 days off for 1 week) + RT
4. Xevinapant (100mg/kg QD 5 days on, 2 days off for 2 weeks) + RT
5. Xevinapant (100mg/kg QD 5 days on, 2 days off for 4 weeks) + RT

Study endpoints:

1. Tumor growth
2. Survival

IM, intramuscularly; **QD,** every day; **RT,** radiotherapy.

RESULTS

In vivo efficacy

- Xevinapant + RT combination treatment showed increased antitumor efficacy compared with vehicle control and RT alone (**Figure 2A** and **Supplemental Figure 1**)
- Extended dosing of xevinapant for 4 weeks further improved antitumor efficacy and animal survival vs treatment arms with shorter xevinapant dosing duration (**Figure 2A** and **2B**)

Figure 2. Antitumor efficacy

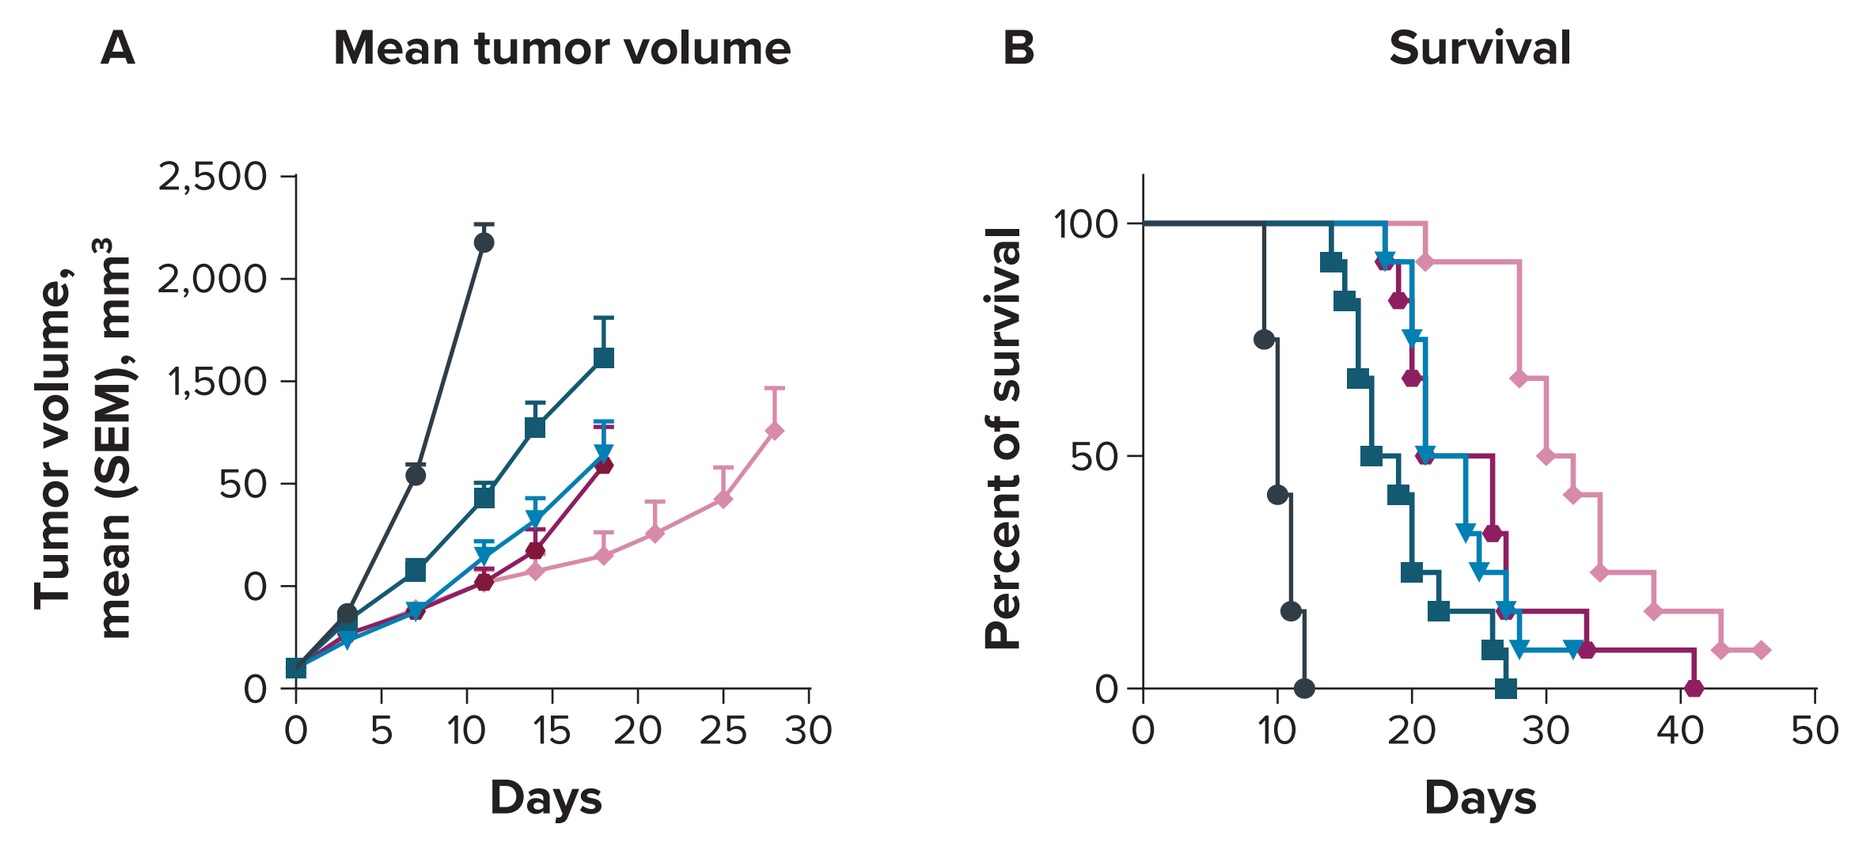
A
Mean tumor volume
Tumor volume, mean (SEM), mm³
2,500
2,000
1,500
50
0
Days
B
Survival
Percent of survival
100
50
Days

	Median survival time, days
Vehicle	10 Days
RT 3.6 Gy QD for 5 days	18 Days
RT + xevinapant 100 mg/kg 1 week	22.5 Days
RT + xevinapant 100 mg/kg 2 weeks	23.5 Days
RT + xevinapant 100 mg/kg 4 weeks	31 Days

Log-rank (Cox-Mantel) test: p<0.0001

RT, radiotherapy.

Reprogramming of immune compartments

- In a pharmacodynamic study with immune phenotyping by FACS (**Supplemental Figure 2**), the combination of xevinapant + RT modulated infiltration and phenotype of CD8+ T cells. Overall, a trend of increased number of CD8+ TILs and antigen-specific TILs was observed in the combination arm compared with baseline and RT alone, respectively (**Figure 3A**)
- Trends towards increased numbers of natural killer cells and dendritic cells as well as fewer regulatory T cells in the combination arm, compared with their corresponding baselines, were observed (**Figure 3B**)

Figure 3. Immune modulation by xevinapant + RT*

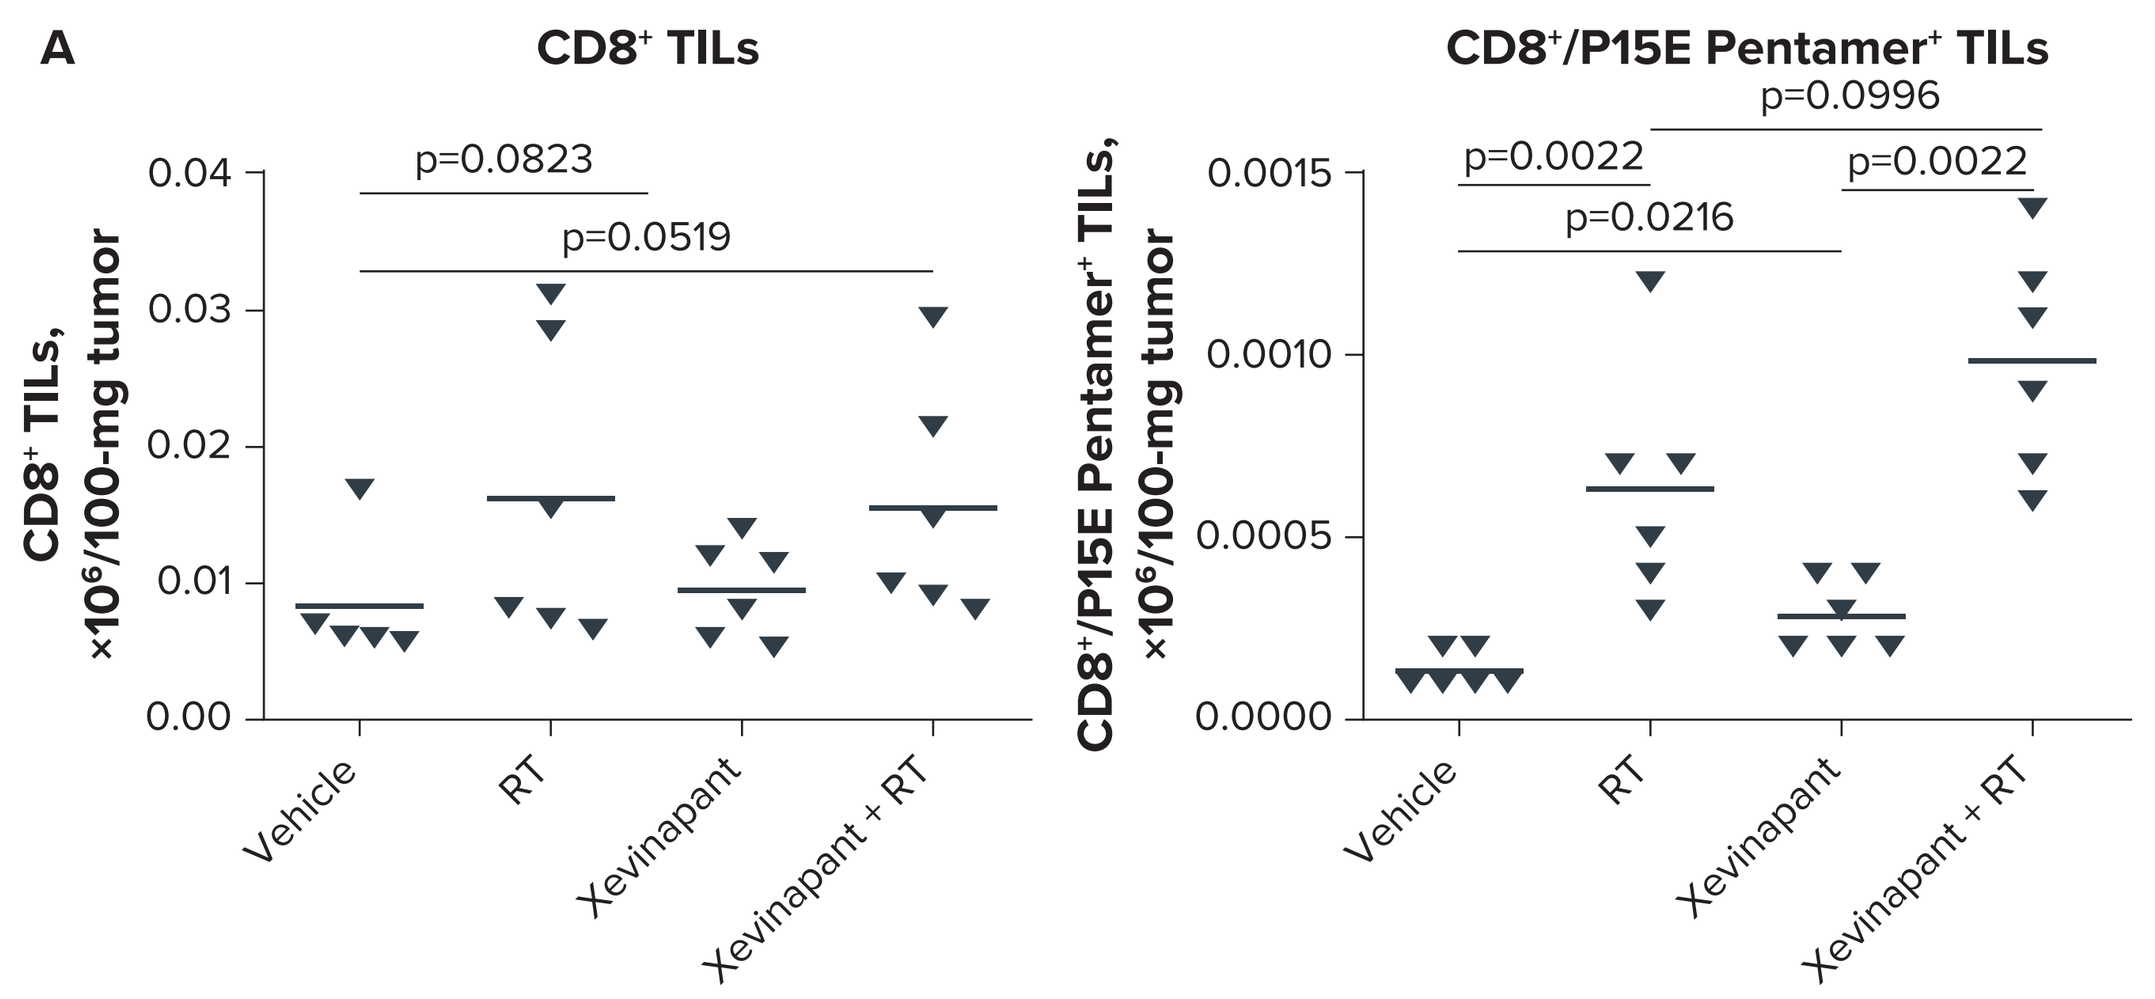
A
CD8⁺ TILs
CD8⁺ TILs, ×10⁶/100-mg tumor
p=0.0823
p=0.0519
CD8⁺/P15E Pentamer⁺ TILs
CD8⁺/P15E Pentamer⁺ TILs, ×10⁶/100-mg tumor
p=0.0022
p=0.0996
p=0.0216
p=0.0022
Vehicle
RT
Xevinapant
Xevinapant + RT

B

Immune cell type	Treatment: RT	Treatment: Xevinapant	Treatment: Xevinapant + RT
CD8⁺ cell	↑	—	↑
CD8⁺/P15E Pentamer⁺ T cell	↑	↑	↑↑
CD4⁺ T cell	↓	↓	↓
Treg	—	↓	↓
NK cell	—	↑	↑
NKT cell	—	↓	—
TAM	↓ M1 TAM	—	—
Dendritic cell	↑	↑	↑
Neutrophil	↑	↑	↑
MDSC	↑	↑	↑

Note: size and number of arrows indicate magnitude of change vs vehicle control.

*p values from Mann-Whitney tests.
CD, cluster of differentiation; **IM,** intramuscularly; **MDSC,** myeloid-derived suppressor cell; **NK,** natural killer; **NKT,** natural killer T; **RT,** radiotherapy; **TAM,** tumor associated macrophages; **TIL,** tumor-infiltrating lymphocyte; **Treg,** regulatory T cell.

Tumor-specific immune response

- Evaluation of splenic immune cells by ELISpot assays confirmed that combination therapy further enhanced tumor-specific T-cell activation compared with vehicle control and monotherapies (**Figure 4A**)
- In a study comparing tumor-specific immune response between animals treated with RT with or without extended xevinapant dosing, a larger number of interferon-γ–positive spots were detected in the extended dosing arm, suggesting an enhanced immunologic memory (**Figure 4B**)

Figure 4. Assessment of tumor-specific T-cell response*

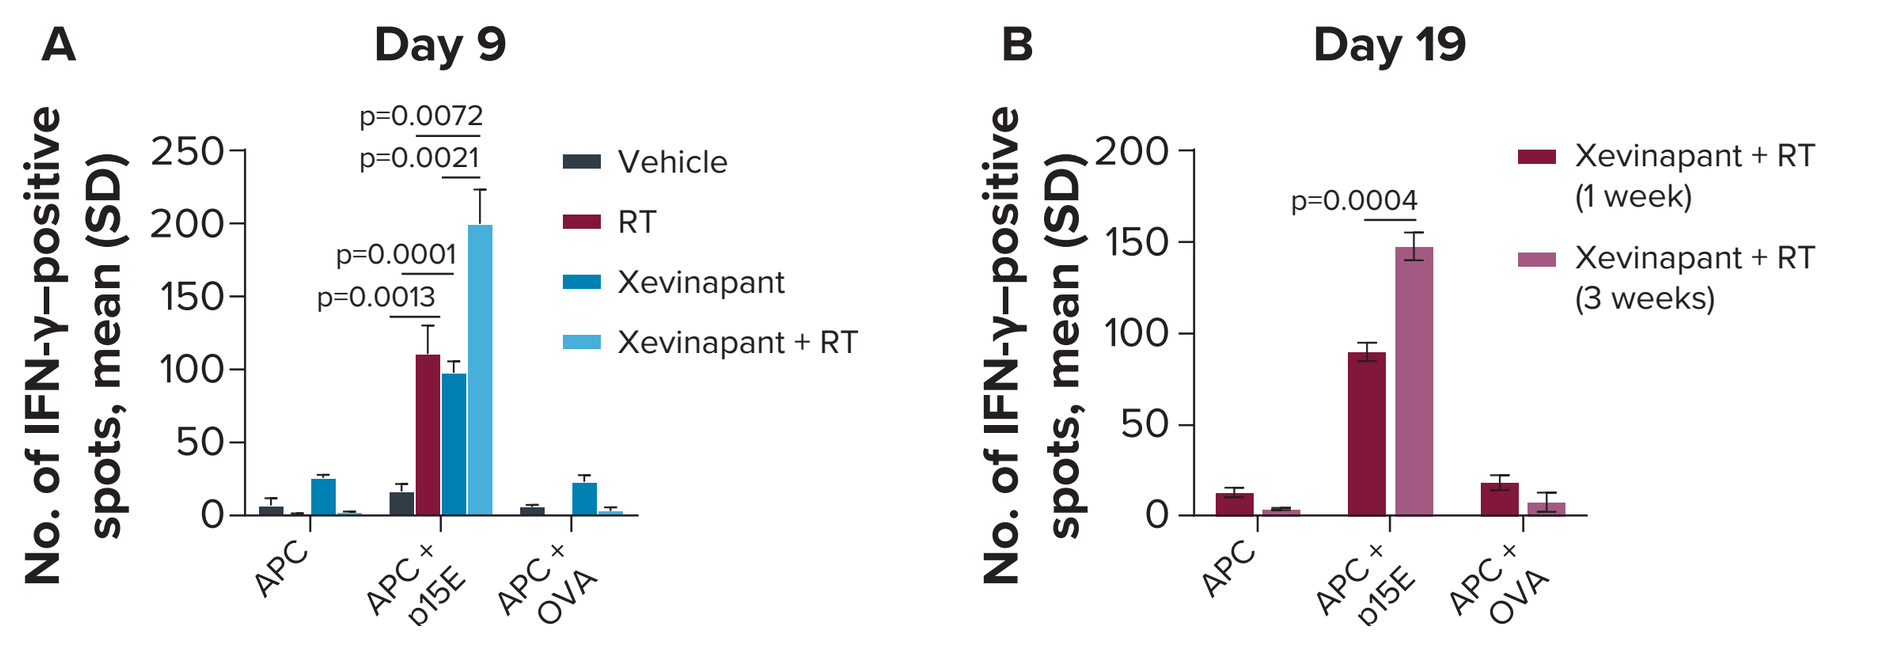
A
Day 9
No. of IFN-γ–positive spots, mean (SD)
p=0.0072
p=0.0021
p=0.0001
p=0.0013
Vehicle
RT
Xevinapant
Xevinapant + RT
APC
APC + p15E
APC + OVA
B
Day 19
p=0.0004
Xevinapant + RT (1 week)
Xevinapant + RT (3 weeks)

*p values from two-sample *t*-tests.
APC, antigen-presenting cell; **IFN-γ,** interferon gamma; **OVA,** ovalbumin; **RT,** radiotherapy.

Impact on other tumor microenvironment cell types

- Xevinapant single-agent treatment from 0.15 to 10 µM did not affect MC38 cell proliferation in vitro, while RT treatment suppressed MC38 cell growth (**Figure 5A**)
- Xevinapant + RT further suppressed cell proliferation compared with RT alone. In the combination setting with a fixed RT dosage, xevinapant suppressed tumor cell growth in a dose-dependent manner (**Figure 5A**)
- Western blot analysis of cellular IAP1 degradation and cleaved caspase-3 levels confirmed xevinapant target engagement and dose-dependent induction of apoptosis in MC38 cells by the combination therapy (**Figure 5B**)
- Xevinapant monotherapy from 0.15 to 10 µM did not affect fibroblast proliferation in vitro. However, unlike tumor cells, fibroblasts demonstrated higher resistance to radiation-induced cell death, and combination of xevinapant + RT did not further suppress cell proliferation compared with RT alone (**Supplemental Figure 3**)
- Despite the absence of cell growth modulation in fibroblasts, xevinapant suppressed RT-mediated fibroblast activation and extracellular matrix protein production, as indicated by downregulation of activation markers and cancer-associated fibroblast-derived secretory protein gene expression (**Figure 5C**)

Figure 5. Modulation of cancer cell and fibroblast phenotype by xevinapant and RT

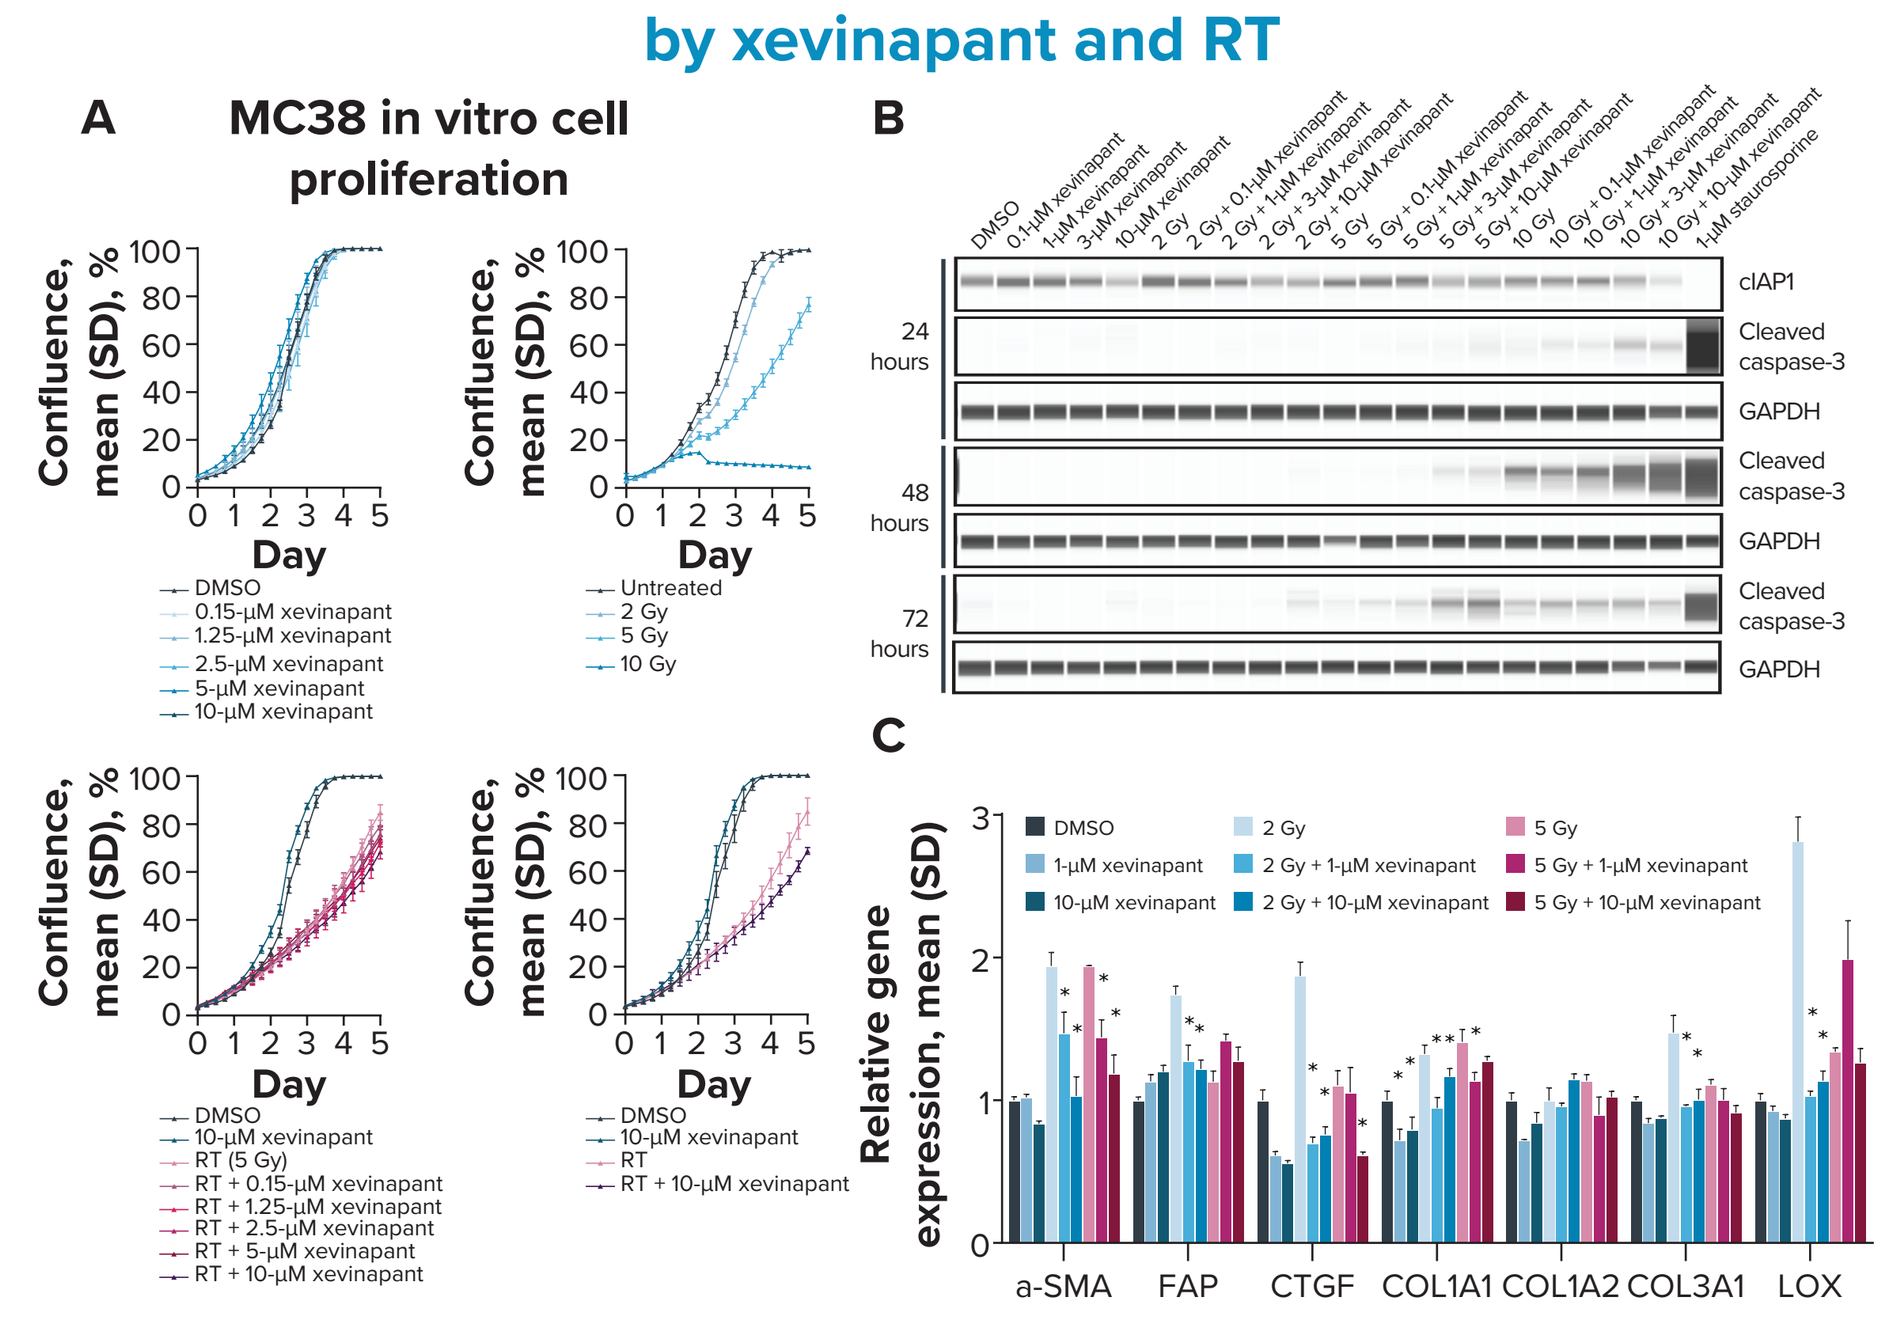
A
MC38 in vitro cell proliferation
Confluence, mean (SD), %
Day
DMSO
0.15-µM xevinapant
1.25-µM xevinapant
2.5-µM xevinapant
5-µM xevinapant
10-µM xevinapant
Untreated
2 Gy
5 Gy
10 Gy
10-µM xevinapant
RT (5 Gy)
RT + 0.15-µM xevinapant
RT + 1.25-µM xevinapant
RT + 2.5-µM xevinapant
RT + 5-µM xevinapant
RT + 10-µM xevinapant
RT
B
24 hours
48 hours
72 hours
cIAP1
Cleaved caspase-3
GAPDH
C
Relative gene expression, mean (SD)
DMSO
1-µM xevinapant
10-µM xevinapant
2 Gy
2 Gy + 1-µM xevinapant
2 Gy + 10-µM xevinapant
5 Gy
5 Gy + 1-µM xevinapant
5 Gy + 10-µM xevinapant
a-SMA
FAP
CTGF
COL1A1
COL1A2
COL3A1
LOX

*p<0.05 vs corresponding control (two-sample *t*-test).
cIAP1, cellular inhibitor of apoptosis protein 1; **DMSO,** dimethyl sulfoxide; **GAPDH,** glyceraldehyde 3-phosphate dehydrogenase; **RT,** radiotherapy.

Proposed mechanism of action (MoA)

- Based on the current findings, we believe that the combination of xevinapant + RT may modulate several key tumor microenvironment components (**Figure 6**)

Figure 6. Overall mechanism of action of xevinapant + RT

Working hypothesis of xevinapant + RT combination MoA

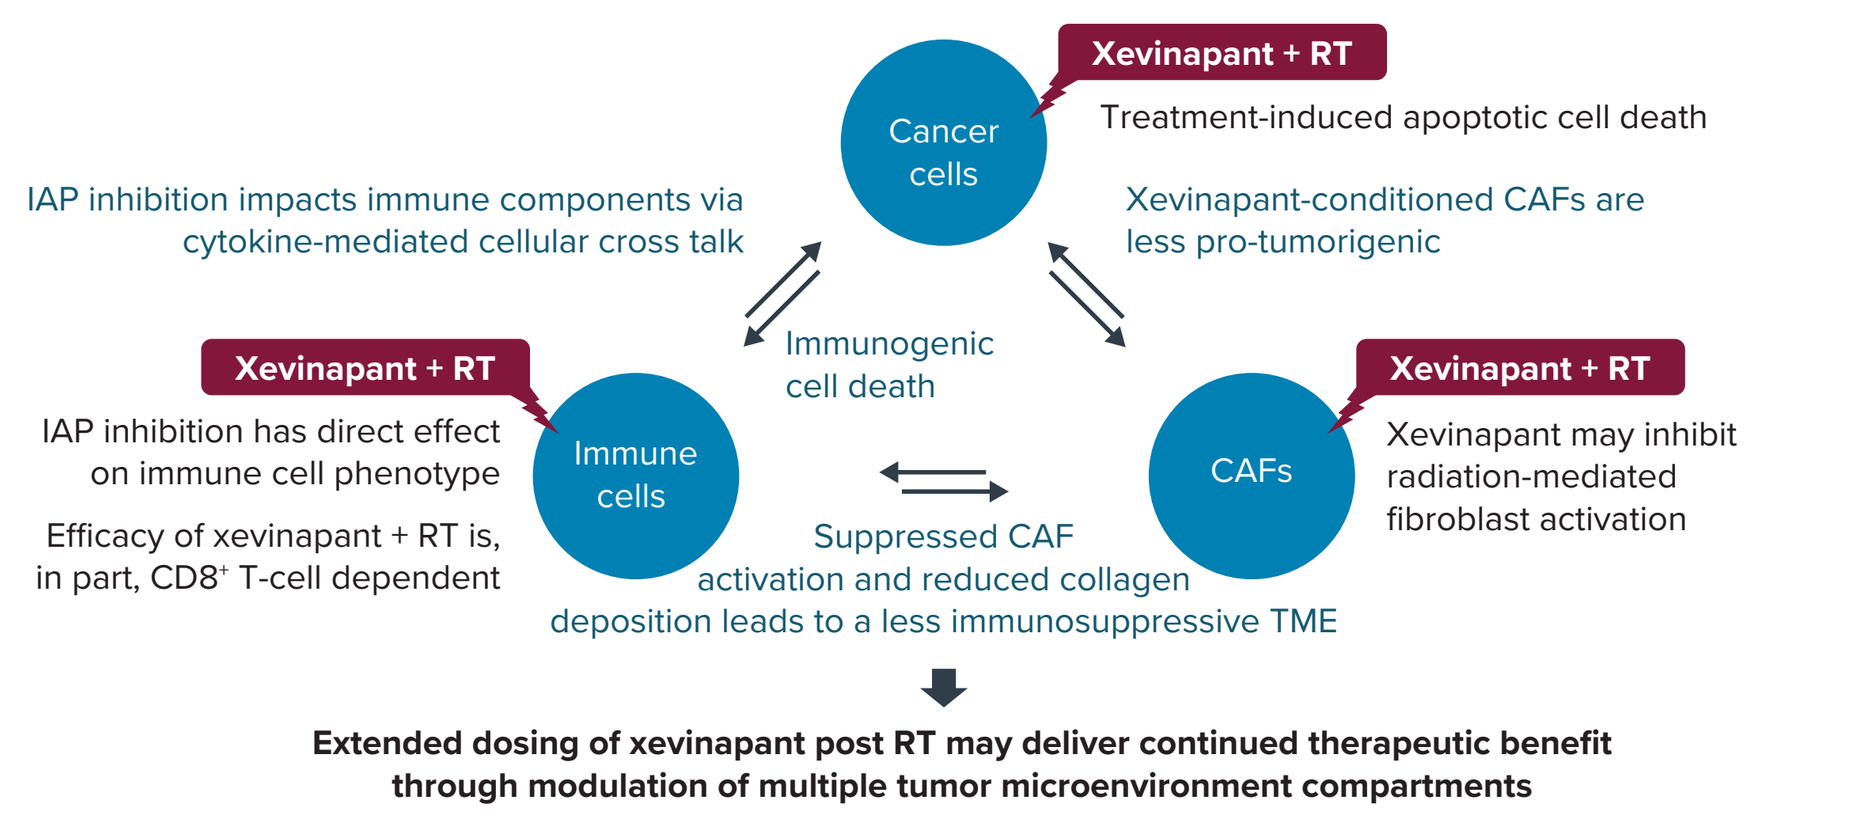
Xevinapant + RT
Cancer cells
Treatment-induced apoptotic cell death
IAP inhibition impacts immune components via cytokine-mediated cellular cross talk
Xevinapant-conditioned CAFs are less pro-tumorigenic
Immunogenic cell death
Xevinapant + RT
IAP inhibition has direct effect on immune cell phenotype
Efficacy of xevinapant + RT is, in part, CD8⁺ T-cell dependent
Immune cells
Xevinapant + RT
CAFs
Xevinapant may inhibit radiation-mediated fibroblast activation
Suppressed CAF activation and reduced collagen deposition leads to a less immunosuppressive TME
Extended dosing of xevinapant post RT may deliver continued therapeutic benefit through modulation of multiple tumor microenvironment compartments

CAF, cancer associated fibroblast; **IAP,** inhibitor of apoptosis protein; **MoA,** mechanism of action; **RT,** radiotherapy; **TME,** tumor microenvironment.

References: 1. Sun XS, et al. *Lancet Oncol*. 2020;21(9):1173-87. **2.** Tao Y, et al. *Eur J Cancer*. 2023;183:24-37. **3.** Bourhis J, et al. *Future Oncol*. 2022;18(14):1669-78. **4.** Ferris RL, et al. *Cancer Treat Rev*. 2022;113:102492. **5.** Roehle K, et al. *Sci Transl Med*. 2021;13(594):eabf5058. **6.** Hoefsmit EP, et al. *Cancer Immunol Res*. 2023;11(4):450-65. **7.** Ashley SL, et al. *Am J Respir Cell Mol Biol*. 2016;54(4):482-92. **8.** Bergmann C, et al. *Ann Rheum Dis*. 2021;80(8):1048-56.

Acknowledgments: Medical writing support was provided by Clinical Thinking, which was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) in accordance with Good Publication Practice guidelines (https://www.ismpp.org/gpp-2022).

Disclosures: T.-L. Yeung, F. Jiang, H. Yu, L.-Y. Chiu, B. Marelli, M. Bonnemaison, and **R. Ferretti** are employees of EMD Serono, Billerica, MA, USA. **J. Qi, H. Wang, M. Jenkins,** and **Y. Lan** were employees of EMD Serono, Billerica, MA, USA, at the time of the research. **R. Lindemann** and **C. Esdar** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany.

Poster No. 6027. Presented at the 2023 ASCO Annual Meeting, June 2-6, 2023; Chicago, IL, USA.

Supplemental Figure 1. Antitumor efficacy

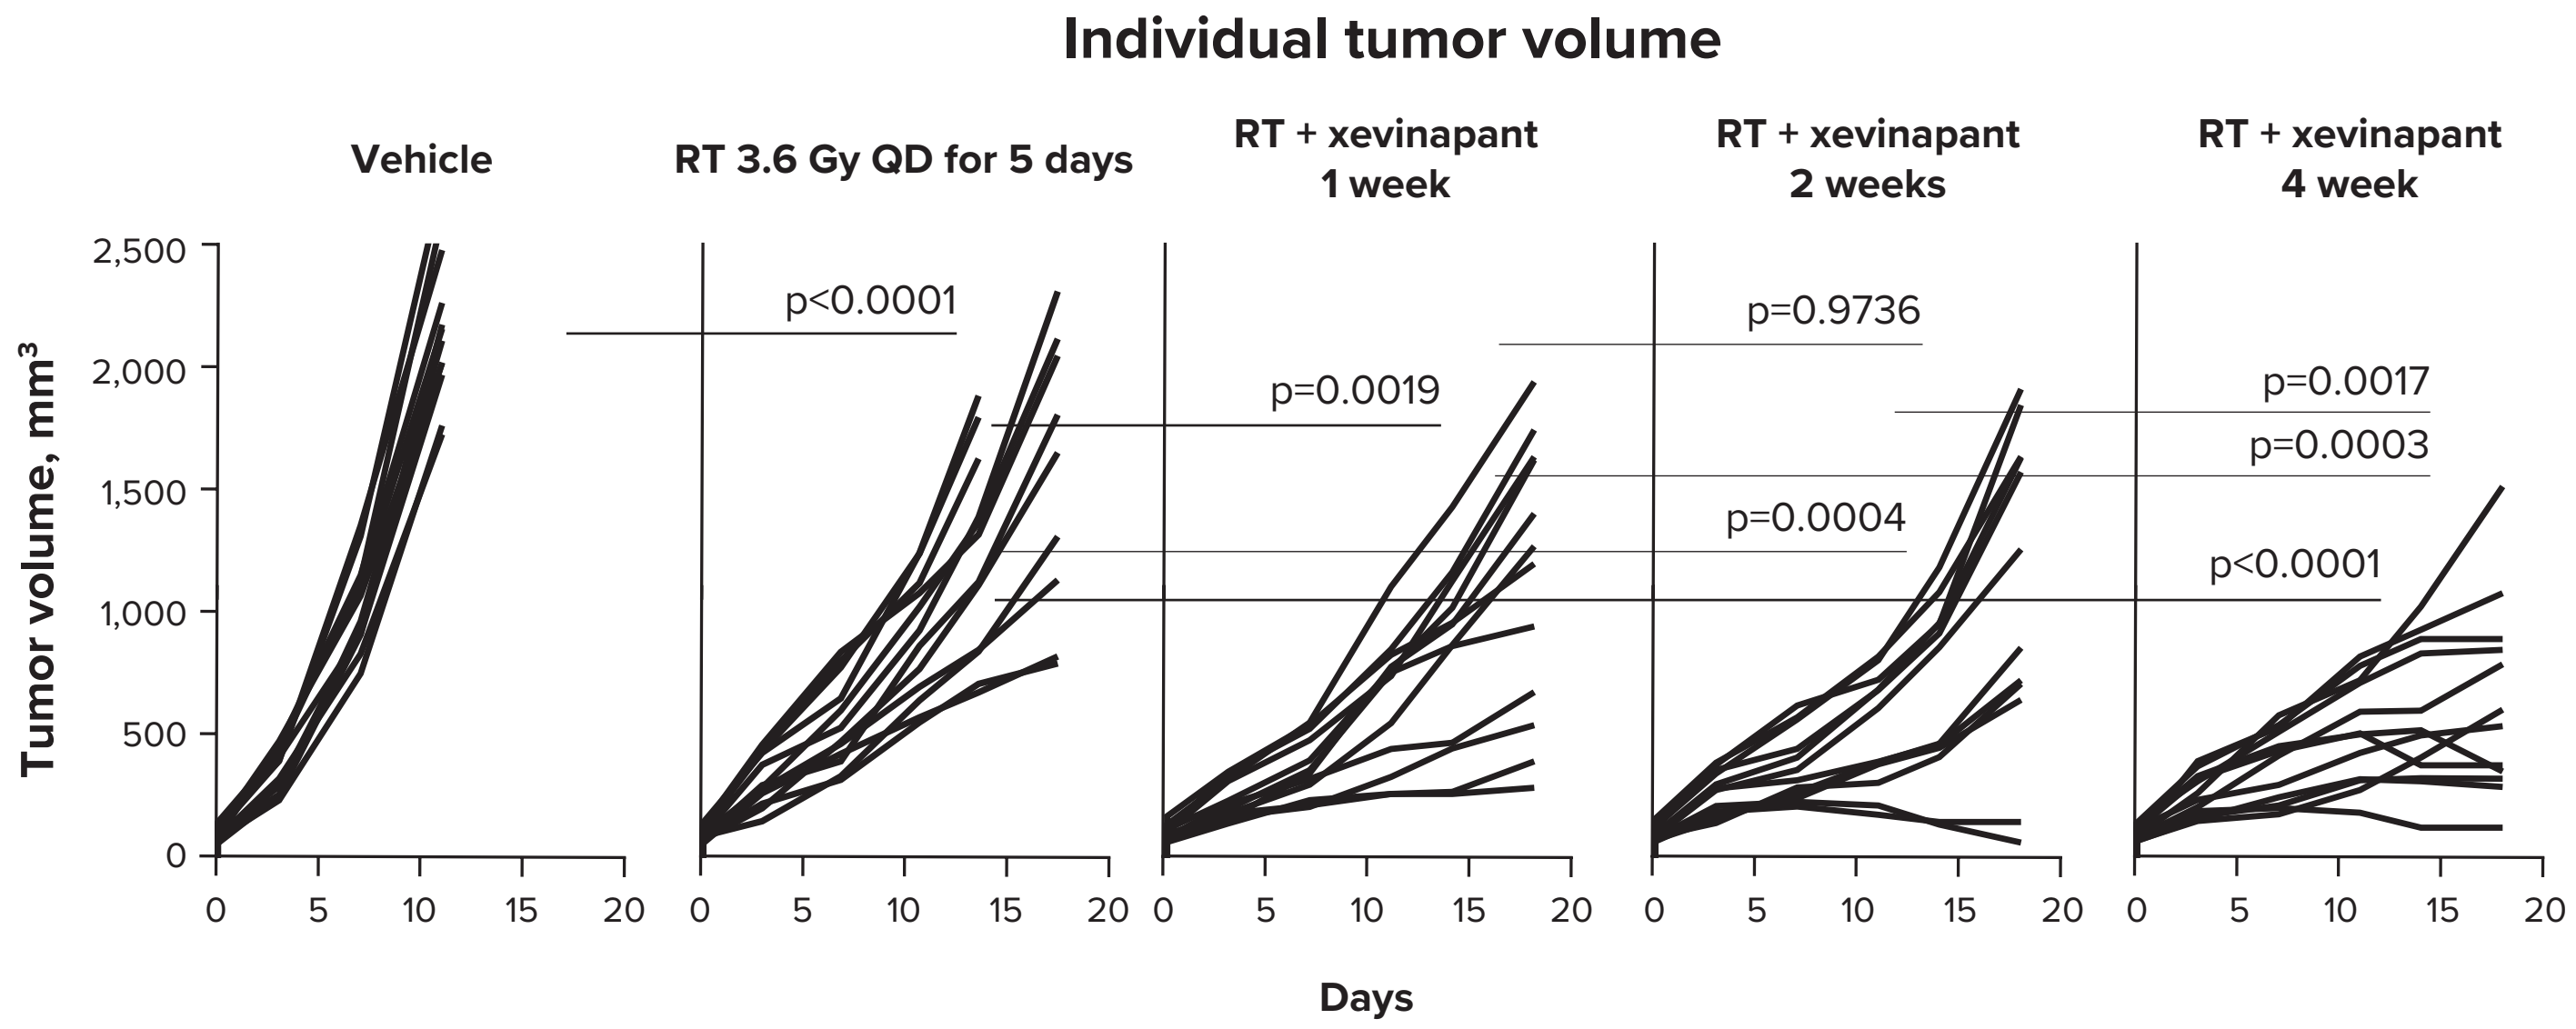
Individual tumor volume
Vehicle
RT 3.6 Gy QD for 5 days
RT + xevinapant 1 week
RT + xevinapant 2 weeks
RT + xevinapant 4 week
Tumor volume, mm³
2,500
2,000
1,500
1,000
500
0
0 5 10 15 20
p<0.0001
p=0.0019
p=0.9736
p=0.0004
p=0.0017
p=0.0003
p<0.0001
Days

RT, radiotherapy.

Supplemental Figure 2. Immune modulation by xevinapant + RT ELISpot design

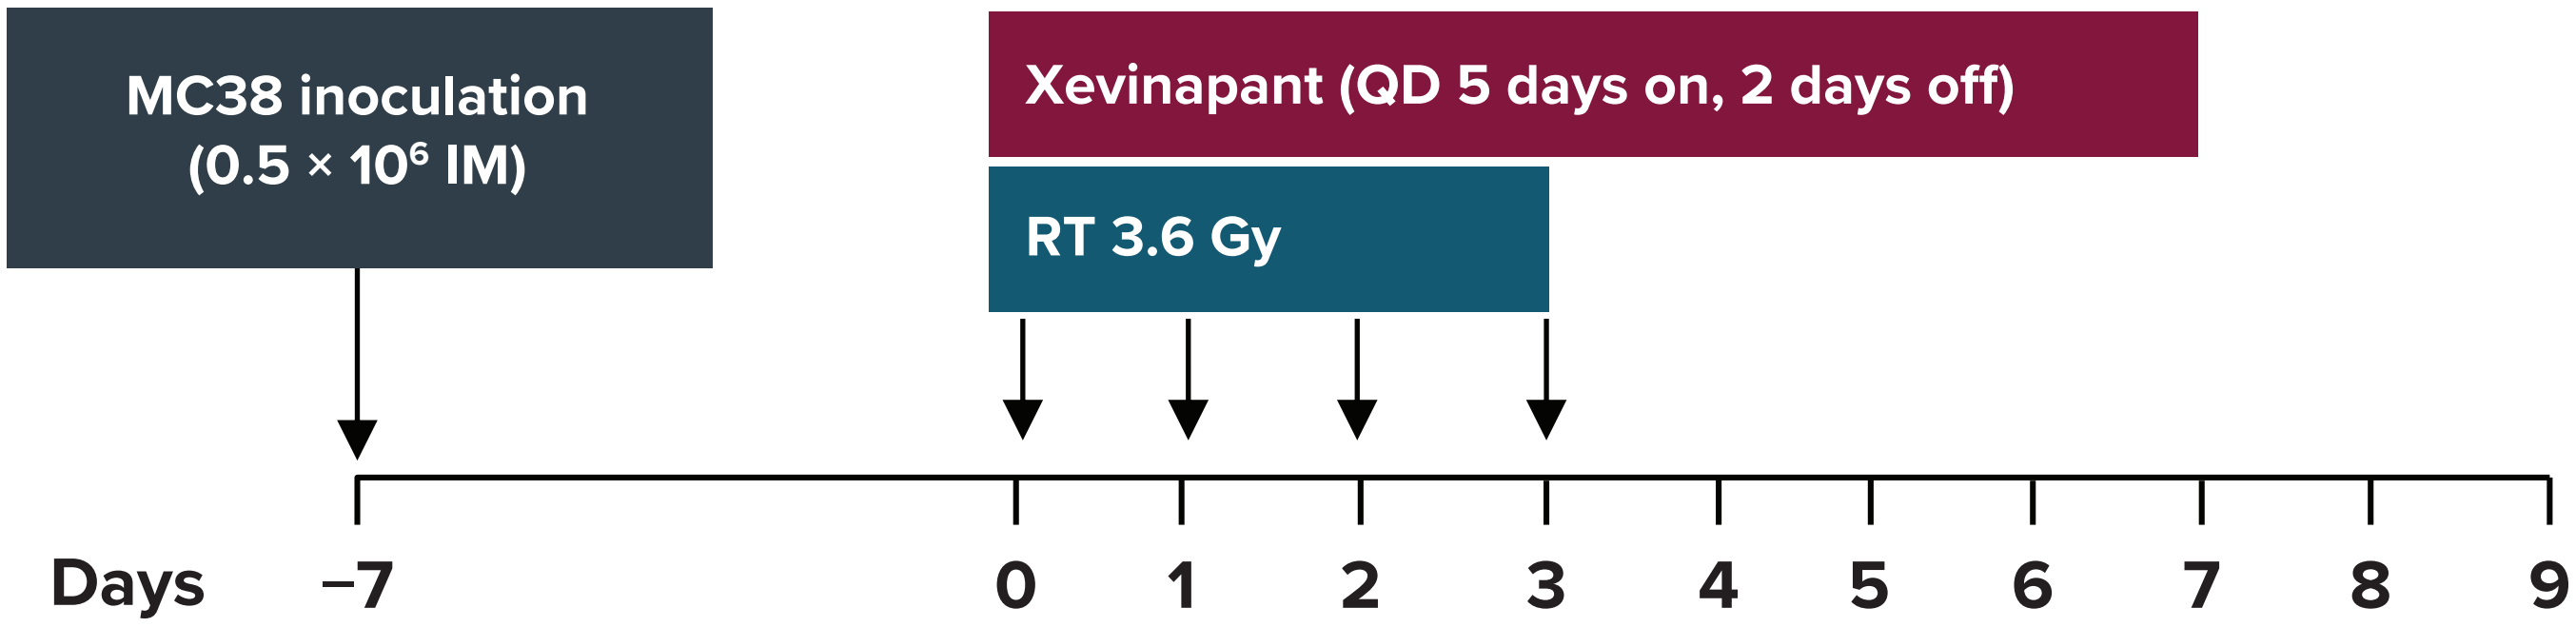
MC38 inoculation
(0.5 × 10⁶ IM)
Xevinapant (QD 5 days on, 2 days off)
RT 3.6 Gy
Days
−7
0
1
2
3
4
5
6
7
8
9

Treatment Groups: N=6 per group

1. Vehicle
2. RT (3.6 Gy QD for 4 days) + vehicle
3. Xevinapant (100 mg/kg QD 5 days on, 2 days off)
4. Xevinapant (100 mg/kg QD 5 days on, 2 days off) + RT

ELISpot, enzyme-linked immunosorbent spot; **IM,** intramuscularly; **QD,** every day; **RT,** radiotherapy.

Supplemental Figure 3. Modulation of fibroblast phenotype by xevinapant and RT

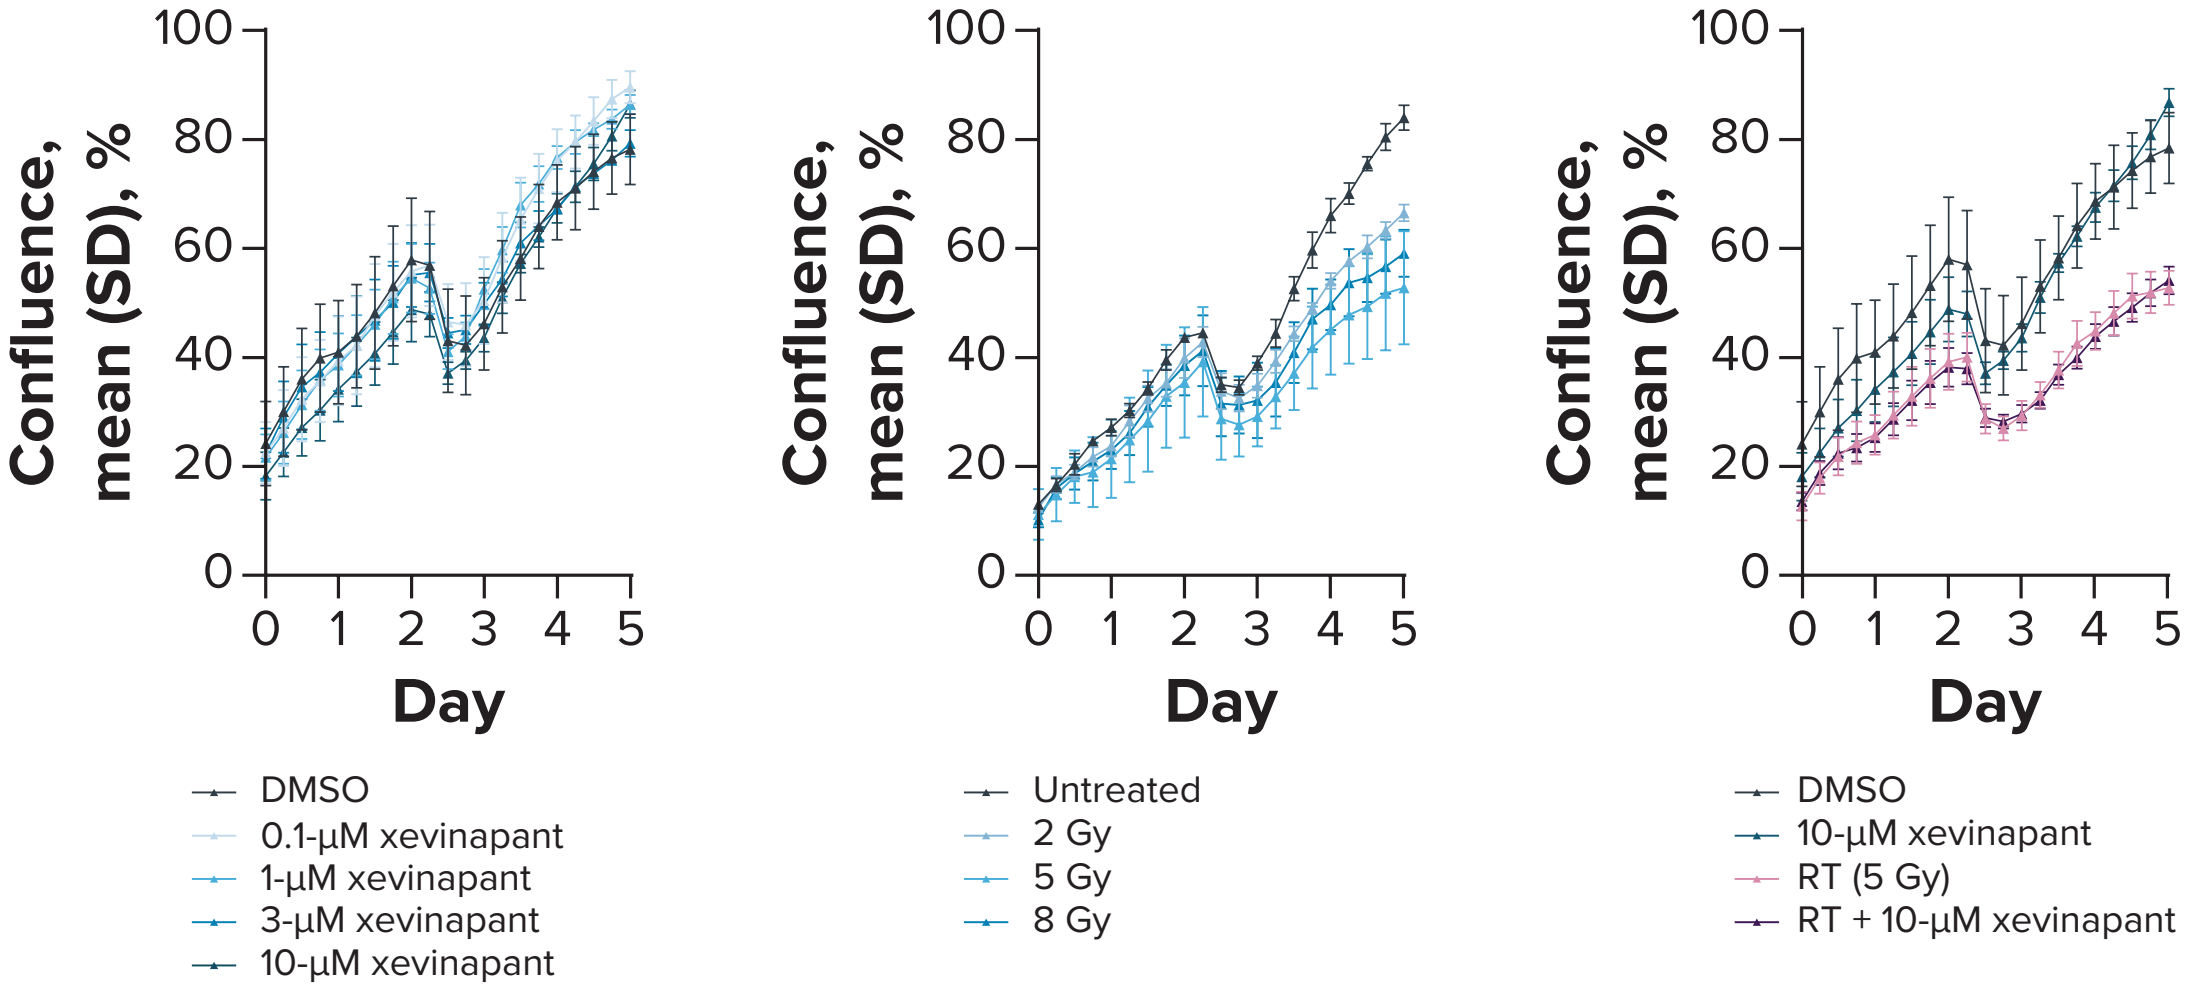

Confluence, mean (SD), %
100
80
60
40
20
0
0 1 2 3 4 5
Day
DMSO
0.1-µM xevinapant
1-µM xevinapant
3-µM xevinapant
10-µM xevinapant
Confluence, mean (SD), %
Day
Untreated
2 Gy
5 Gy
8 Gy
Confluence, mean (SD), %
Day
DMSO
10-µM xevinapant
RT (5 Gy)
RT + 10-µM xevinapant

DMSO, dimethyl sulfoxide; **RT,** radiotherapy.

Proposed mechanism of action (MoA)

- Evidence suggests that IAP inhibitors could enhance the efficacy of anti-cancer treatments by re-sensitizing cancer cells to apoptosis and modulate immune cell types; however, some mechanistic detail remains unexplored, including the tumor microenvironment impact by IAP inhibitors in combination with RT and how continuous administration of IAP inhibitors post RT may confer additional therapeutic benefit
- To address this gap in knowledge, we utilize preclinical models to delineate the MoA underlying the therapeutic benefit mediated by 1) combination therapy vs monotherapy and 2) extended xevinapant dosing post RT
- Based on the current findings, we believe that the combination of xevinapant + RT may modulate several key tumor microenvironment components (**Figure 6**):
 - Combination treatment directly promotes cell death via apoptotic pathways
 - For stromal fibroblasts, a highly relevant compartment in the radiation therapy setting, xevinapant may inhibit radiation-mediated fibroblast activation and reduced collagen production resulting in fibroblasts that are less tumor promoting and immunosuppressive
 - Xevinapant + RT promotes tumor-specific T-cell response and continual dosing of xevinapant post RT may encourage the development of a stronger immunological memory