This is a reprint from the European Society for Medical Oncology Congress 2022 (ESMO 2022), which was originally presented in Paris, France on September 9-13, 2022; the references to "Merck" or "Merck KGaA" within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name "Merck". Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name "Merck KGaA, Darmstadt, Germany" and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark "Merck" in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the "Merck" trademark in all other countries of the world.

Undertreatment rates, associated factors, and survival among patients with locally advanced or metastatic urothelial cancer (la/mUC): a systematic literature review

T. Wilke,[1] L. Zhang,[2] E. Hubscher,[2] M. Musat,[2] S. Harricharan,[2] M. Kearney[3]

[1]Faculty of Economics, University of Wismar, Wismar, Germany; [2]Health Economics and Outcomes Research, Cytel, Waltham, MA, USA; [3]Global Evidence and Value Development, Merck Healthcare KGaA, Darmstadt, Germany

SCOPE

- **The objectives of this systematic literature review (SLR) were to comprehensively characterize treatment patterns in la/mUC, focusing on rates of patients who did not receive systemic treatment (NST), clinical and sociodemographic factors, and resulting overall survival (OS) in the real world**

CONCLUSIONS

- **No significant heterogeneity was observed in populations of patients with la/mUC and reported NST rates across study types, population sizes, and geographic location**
 - **A substantial proportion of patients were reported as NST and had extremely high mortality**
- **Reasons for undertreatment included older age, poor performance status, poor renal function, comorbidities, and other clinical and nonclinical factors**
- **Our results suggest that there is a group of patients who are eligible for first-line (1L) systemic treatment (ST) but do not receive it, which limits the opportunity for potential OS benefits**
 - **Given the availability of promising treatments that offer improved OS with manageable safety profiles, patient-focused, evidenced-based selection of 1L ST is needed**
- **Future research addressing treatment decisions in the evolving treatment landscape of immunotherapy (IO) and other novel agents is warranted to expand real-world patient eligibility and optimize OS benefits**

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster

Correspondence: **Thomas Wilke**, thomas.wilke@cytel.com

BACKGROUND

- Current treatment guidelines for la/mUC recommend platinum-based chemotherapy as 1L treatment for all eligible patients,[1-3] followed by 1L maintenance IO with avelumab in patients who are progression-free after platinum-based chemotherapy[1]
- In addition to maintenance, IO may be given in patients with la/mUC as 1L treatment for PD-L1–positive and for platinum-ineligible patients, or as second-line (2L) treatment following platinum-based 1L chemotherapy
- For end-stage disease, supportive care is recommended (eg, analgesia, antibiotics), as well as palliative surgery or radiotherapy
- Previous studies have suggested that some patients with la/mUC do not receive ST.[4-6] While median OS with ST is 9-24 months, NST prognosis is poor and OS is short[4,6-9]
- Underutilization of ST in patients with la/mUC has not been comprehensively investigated, and questions remain regarding the drivers behind treatment decisions and the high mortality rates despite the newly available agents in this setting

METHODS

- We conducted a Cochrane guideline–based SLR of real-world evidence of NST in patients with la/mUC published from 2017-2022 (including a study period of 2015 or later). Studies published in English, Spanish, French, and German languages from any country were included
- The Cochrane Library, EconLit, Embase, MEDLINE, and MEDLINE In-Process databases were searched on 25 February 2022. The following conferences were hand-searched for abstracts published in 2017 and later: American Society of Clinical Oncology (ASCO) Annual Meeting, ASCO Genitourinary Cancers Symposium, European Society for Medical Oncology Congress, International Society for Pharmacoeconomics and Outcomes Research annual conference, and European Association of Urology Congress
- In the absence of statistics describing associations of patient characteristics with NST within the publications of interests, we performed Fisher's exact tests (statistical test used to analyze contingency tables) using MedCalc software for categorical variables as post hoc statistical analyses

RESULTS

- Data were extracted from 29 selected records representing several regions (**Figure 1**), including the USA, the EU, the UK, Asia, Russia, and the Middle East. The 29 identified studies were either retrospective observational studies (n=10) or analyses of large databases or registries (n=19). Overall, the SLR represented 87,784 patients, with a range of individual study population sizes from 14 to18,888 participants
- Among the 29 studies (**Table 1**), 3 types of NST groups were identified (groups A, B, C)
 - A total of 33 subgroups were identified because 4 studies reported patients from 2 types of NST
- Patient characteristics differed slightly between the studies to include special populations of mUC, metastatic upper tract UC, or node-positive bladder cancer
- Among 13 studies (14 subgroups) from North America, NST rates were 14%-60% (**Figure 2**)
 - Yearly NST rates from 2015-2019 ranged from 48%-58%,[10] largely consistent with 3 large database analyses in the USA that reported NST rates ranging from 40%-60%[11-13]
 - The outliers of the recent group A studies were 2 USA studies with NST rates of 23% and 14%[14,15]
- In 8 studies (10 subgroups) from European countries:
 - NST rates of 40%-74% were reported within group A. NST rates were highest in the UK studies (70%-74%),[16,17] followed by Denmark (64%),[18] the Netherlands (48%),[19] and Spain (40%)[20]
 - NST rates within group B ranged from 8%-12%, except for a Dutch multicenter study[9] with a small study population (<100), which reported a rate of 53%
 - NST rates within group C were 22% (Denmark[21]) and 57% (the Netherlands[8])
- In 8 studies (9 subgroups) from the rest of the world, NST rates were reported to be on the lower end of the spectrum (7.1%-36%),[6,22-28] with only 2 studies reporting NST rates >40%[25,26]
- Seven studies[4,6,7,9,12,19,26] reported OS data for both treated and undertreated patients (**Figure 3**)
 - Median OS in patients with NST ranged from 2.0-6.9 months compared with 9.2-34.5 months in those who received ST
 - The difference in OS between patients with NST and those who received ST was statistically significant in all studies except one
- One study[7] reported a longer OS in patients treated with 1L chemotherapy followed by 2L IO than in those treated with 1L chemotherapy or 1L IO only; another study[6] reported longer OS in patients receiving IO than in those receiving chemotherapy (line of therapy not reported)
- Six studies[4,9,10,13,19,26] recorded characteristics of patients with NST (**Table 2**), but only 2 of these studies[13,26] performed statistical analyses comparing with patients who received ST; post hoc Fisher's exact test was performed to analyze the association of patient characteristics with NST for the remaining 4 studies[4,9,10,19]
- Age was the most commonly reported patient characteristic and was significantly associated with NST in 5 studies
- Other factors that were associated with NST included poor ECOG performance status (2/3), poor renal function (2/3), metastases spread outside the lymph nodes (2/2), comorbidities (2/2), non-White race or ethnicity (2/3), female sex (1/6), and the primary organ being the bladder (1/1)

Figure 1. PRISMA diagram

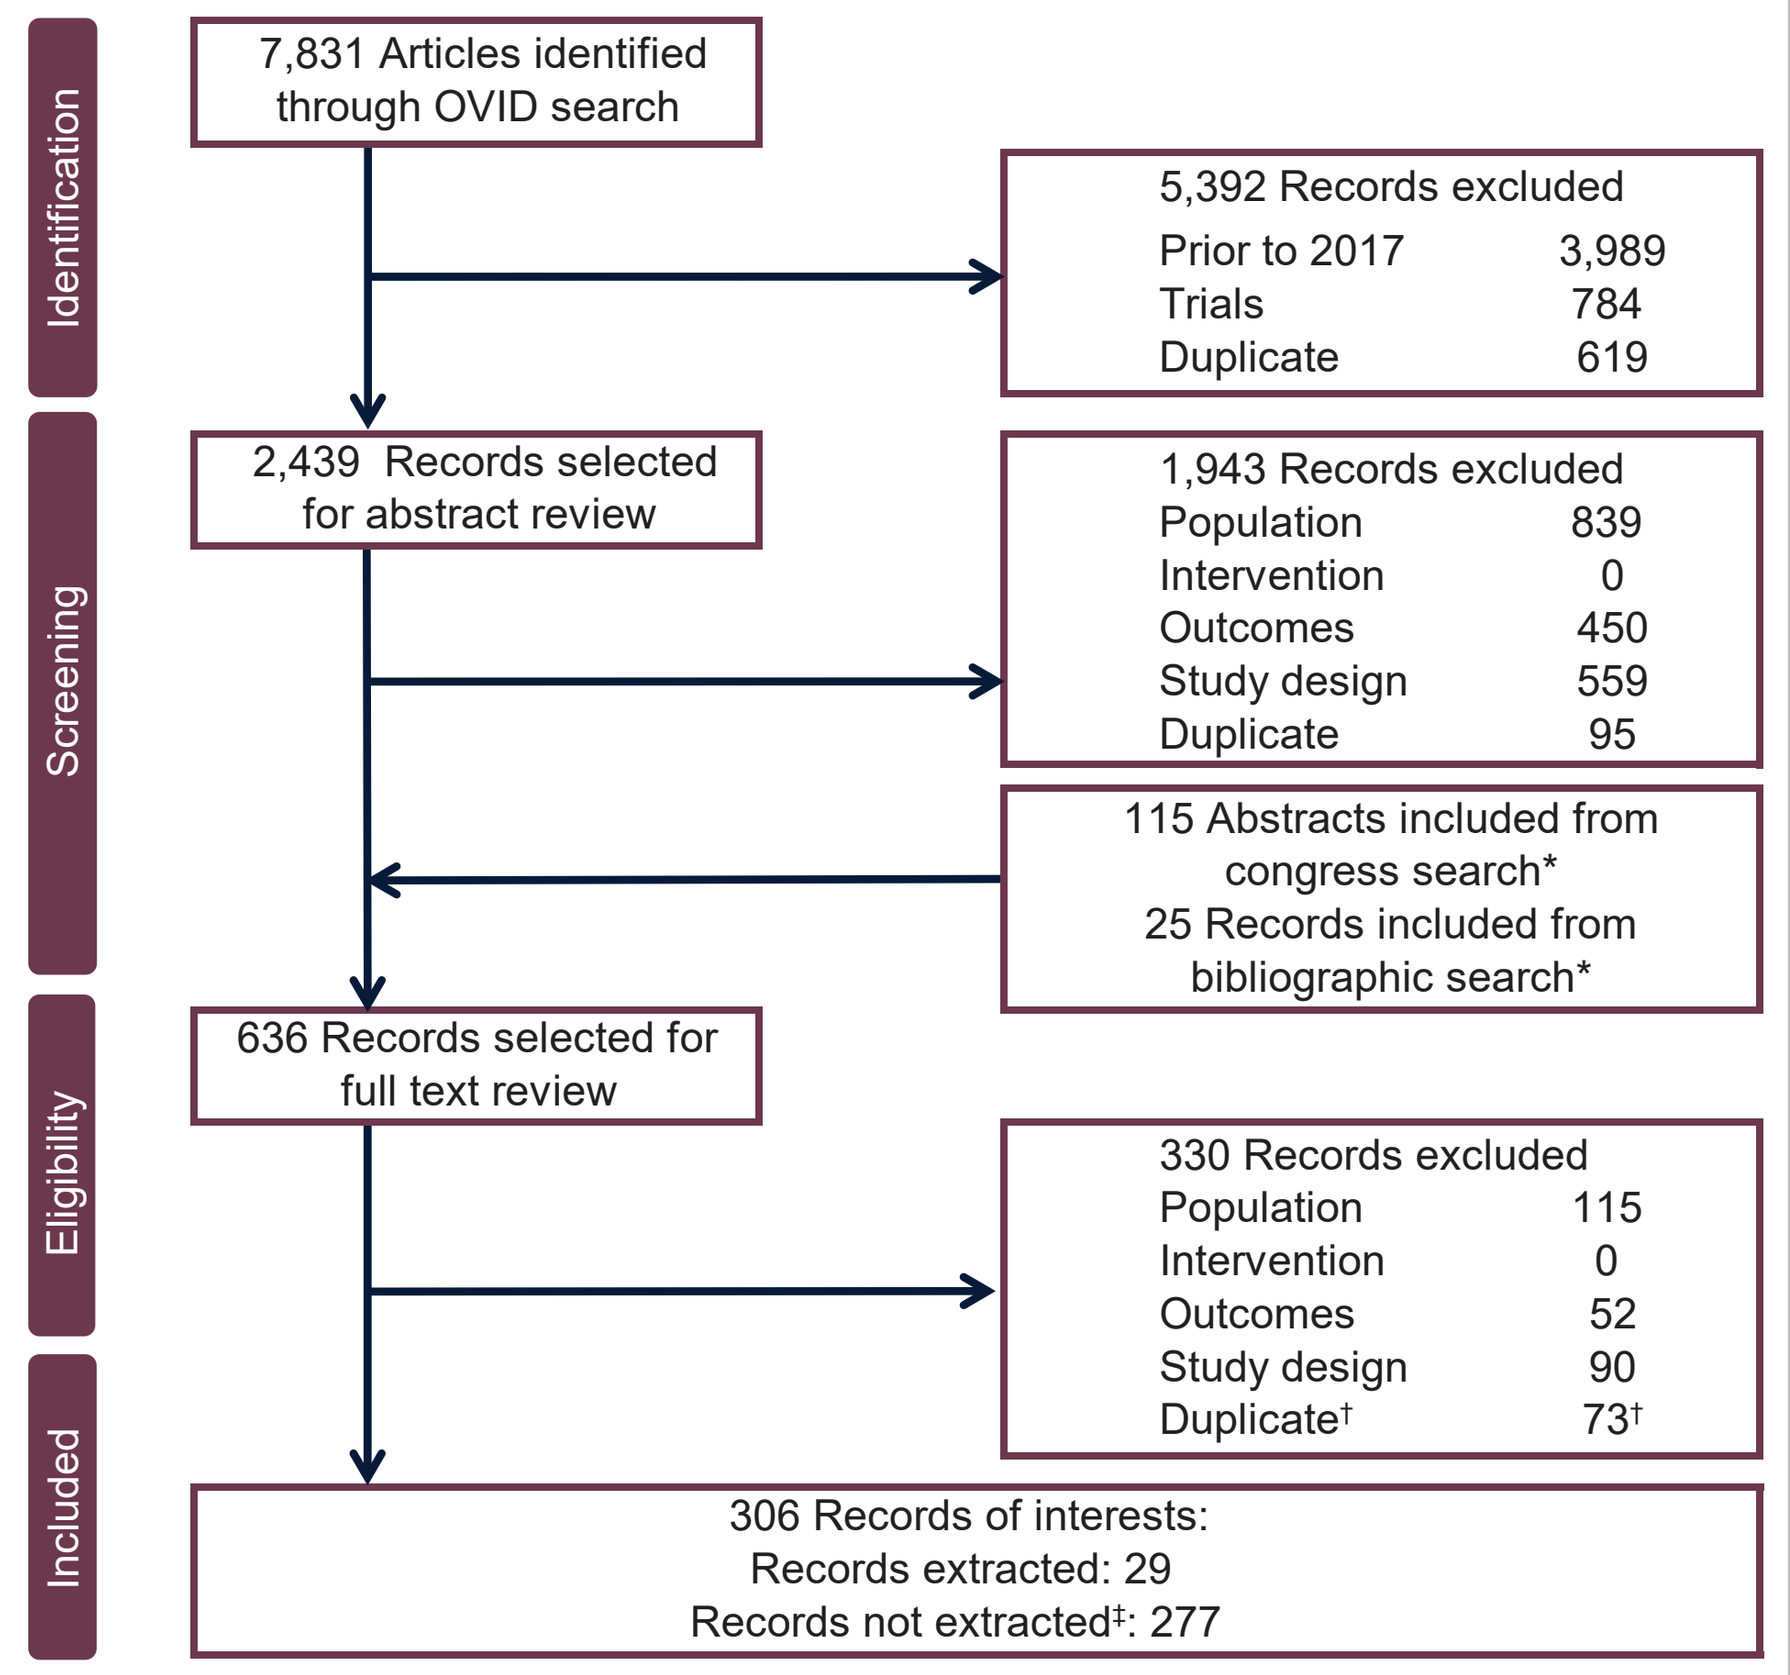
Identification
Screening
Eligibility
Included
7,831 Articles identified through OVID search
5,392 Records excluded
Prior to 2017 3,989
Trials 784
Duplicate 619
2,439 Records selected for abstract review
1,943 Records excluded
Population 839
Intervention 0
Outcomes 450
Study design 559
Duplicate 95
115 Abstracts included from congress search*
25 Records included from bibliographic search*
636 Records selected for full text review
330 Records excluded
Population 115
Intervention 0
Outcomes 52
Study design 90
Duplicate† 73†
306 Records of interests:
Records extracted: 29
Records not extracted‡: 277

NST, no systemic treatment.
Includes duplicates with title/abstract review. †Includes duplicates between title/abstract review and manually added records from congress search and bibliographic search.* ‡Records were not extracted unless fulfilling the additional criteria for data extraction: reporting of NST rates and specification of period when data were acquired.

Figure 2. NST rates by geography

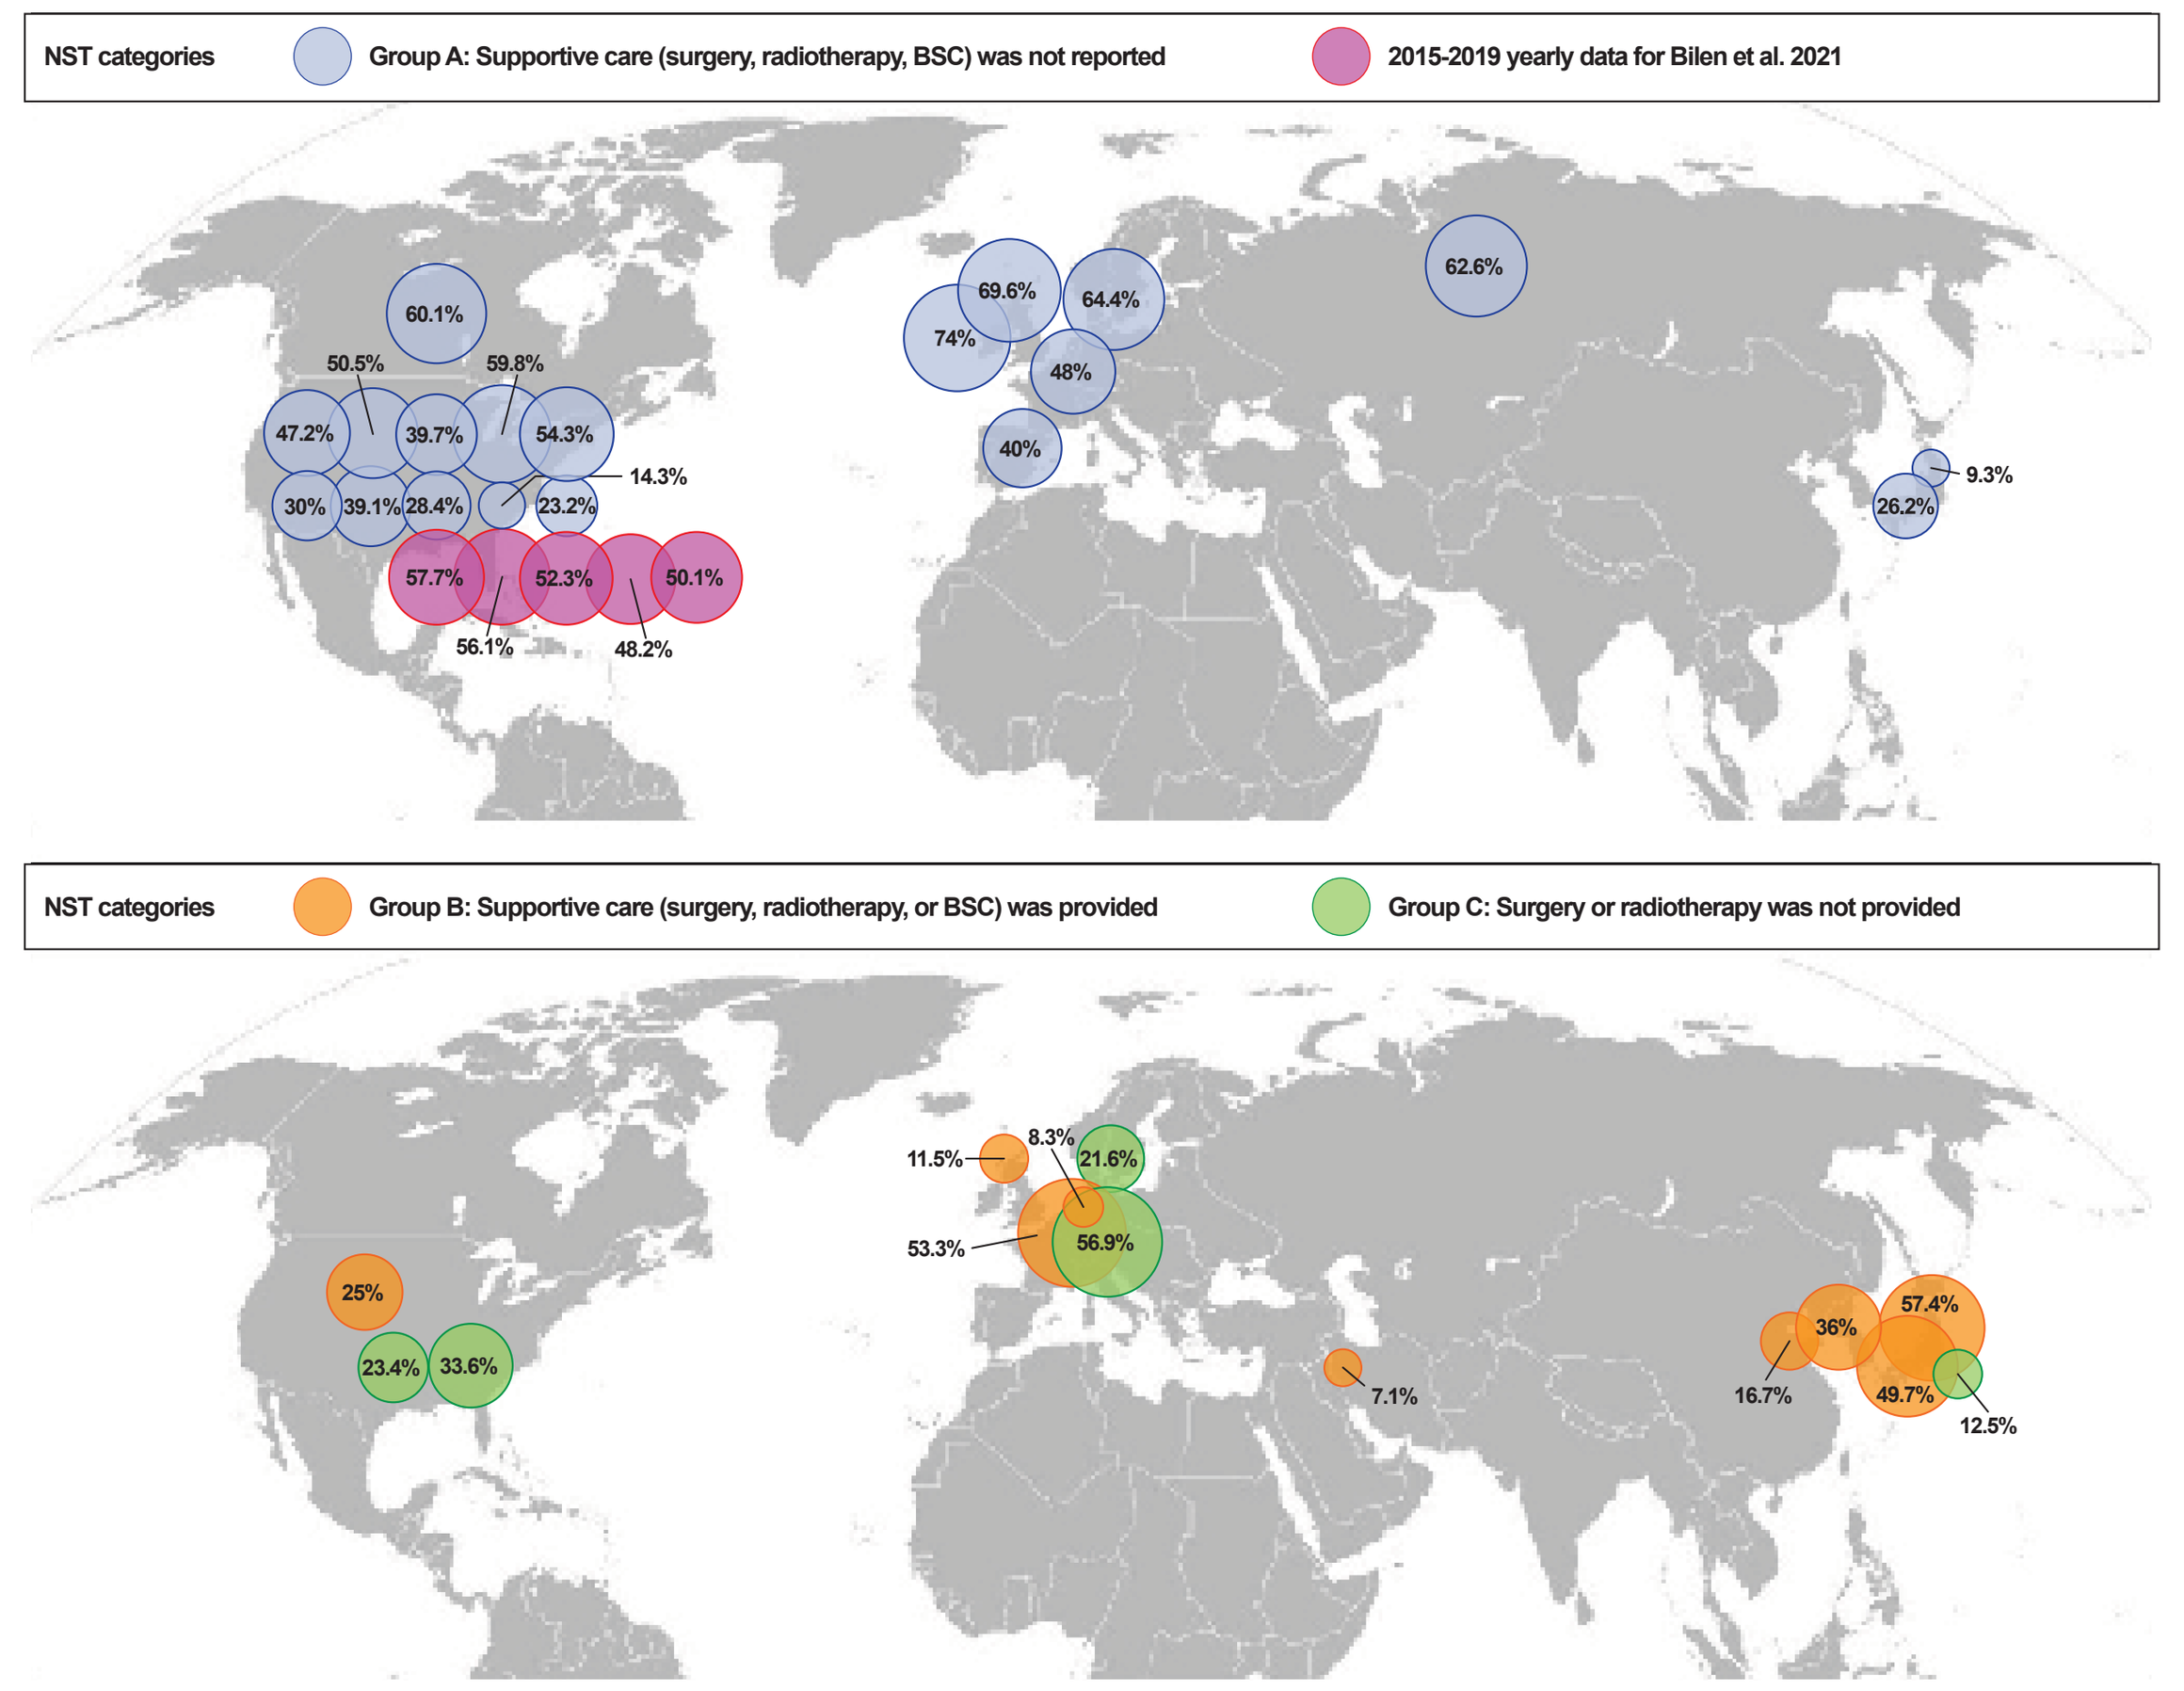
NST categories
Group A: Supportive care (surgery, radiotherapy, BSC) was not reported
2015-2019 yearly data for Bilen et al. 2021
60.1%
50.5%
59.8%
47.2%
39.7%
54.3%
14.3%
30%
39.1%
28.4%
23.2%
57.7%
52.3%
50.1%
56.1%
48.2%
69.6%
64.4%
74%
48%
40%
62.6%
9.3%
26.2%
NST categories
Group B: Supportive care (surgery, radiotherapy, or BSC) was provided
Group C: Surgery or radiotherapy was not provided
11.5%
8.3%
21.6%
53.3%
56.9%
25%
23.4%
33.6%
7.1%
16.7%
36%
57.4%
49.7%
12.5%

1L, first line; **BSC,** best supportive care; **ST,** systemic treatment.

Figure 3. OS with NST vs ST

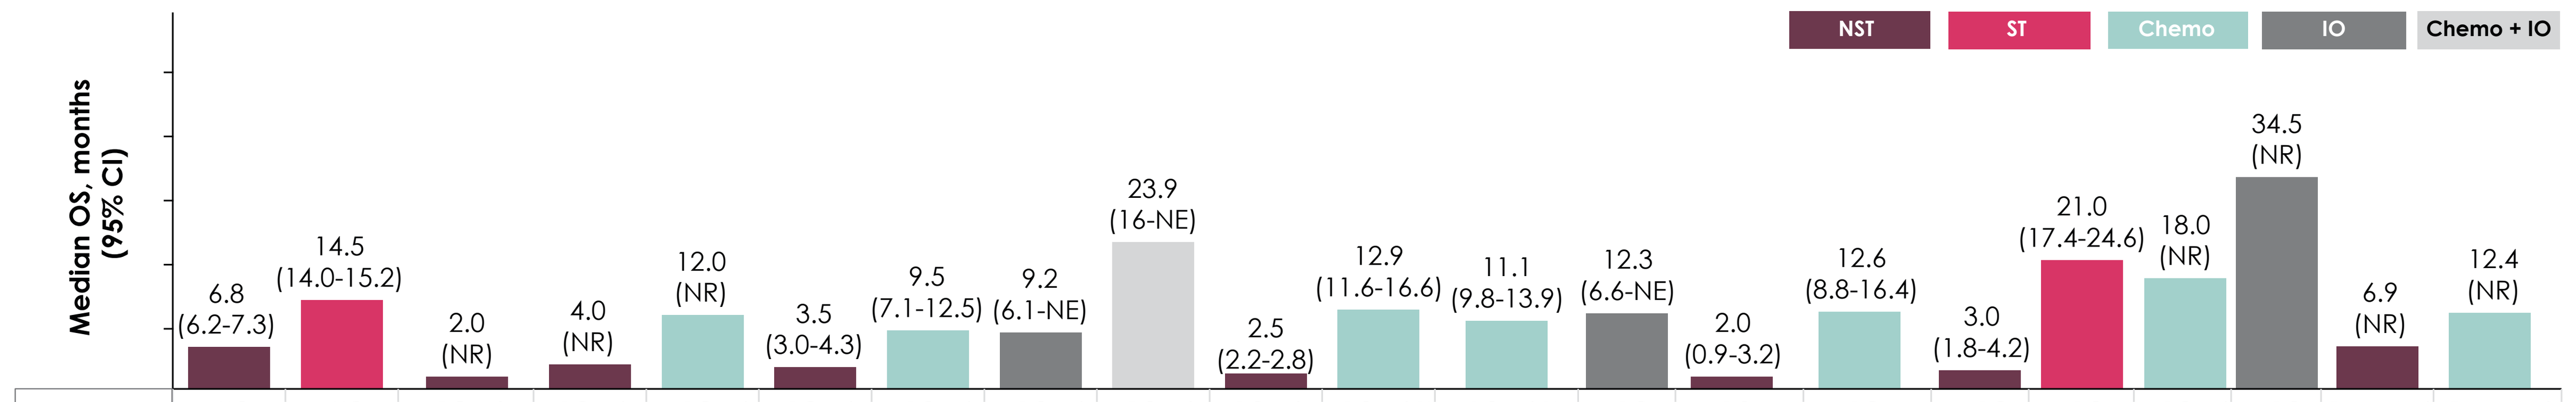
NST
ST
Chemo
IO
Chemo + IO
Median OS, months (95% CI)
6.8 (6.2-7.3)
14.5 (14.0-15.2)
2.0 (NR)
4.0 (NR)
12.0 (NR)
3.5 (3.0-4.3)
9.5 (7.1-12.5)
9.2 (6.1-NE)
23.9 (16-NE)
2.5 (2.2-2.8)
12.9 (11.6-16.6)
11.1 (9.8-13.9)
12.3 (6.6-NE)
2.0 (0.9-3.2)
12.6 (8.8-16.4)
3.0 (1.8-4.2)
21.0 (17.4-24.6)
18.0 (NR)
34.5 (NR)
6.9 (NR)
12.4 (NR)

Population	aUC w/o ST	aUC w/ ≥ 1L ST	mBC w/o chemo, radio, or cystectomy	mBC w/o chemo	mBC w/ chemo	mBC w/o 1L ST	mBC w/ 1L chemo only	mBC w/ 1L IO only	mBC w/ 1L chemo & 2L IO	mBC w/o 1L ST	mBC w/ 1L cisplatin-based chemo	mBC w/ 1L carboplatin-based chemo	mBC w/1L IO	mBC or inoperable disease w/BSC	mBC or inoperable disease w/ 1L+ chemo	mBC w/o ST	mBC w/ST	mBC w/ chemo	mBC w/IO	Recurrent or mUTUC w/ BSC	Recurrent or mUTUC w/ salvage chemo
N	2,328	5,855	2,772	5,581	6,243	164	63	17	29	415	90	108	18	40	24	154	92	70	22	73	74
OS measurement	Months since aUC diagnosis		NR			NR				Months since mBC diagnosis				Months since treatment start		NR				Months since diagnosis of recurrent/mUTUC	
Location	USA		USA			Canada				The Netherlands				The Netherlands		Russia				Japan	
Data period	2011-2020		2004-2015			2014-2016				2016-2017				2008-2016		2017-2018				1990-2015	

1L, first line; **2L,** second line; **aUC,** advanced urothelial carcinoma; **BSC,** best supportive care; **chemo,** chemotherapy; **IO,** immunotherapy; **mBC,** metastatic bladder cancer; **mUTUC,** metastatic upper tract urothelial carcinoma; **NE,** not estimable; **NR,** not reported; **NST,** no systemic treatment; **OS,** overall survival; **radio,** radiotherapy; **ST,** systemic treatment; **w/,** with; **w/o,** without.

Table 1. NST summaries by category

Group	Description of patients with NST	No. of studies	Patients undertreated: Median (range), %	North America, range, %	European countries, range, %	Rest of the world,† range, %
A	Supportive care (surgery, radiotherapy, BSC) was not reported	19	47.2 (9.3-74.0)	14.3-60.1	39.6-74.0	9.3-62.6
B	Supportive care (surgery, radiotherapy, or BSC) was provided instead of 1L ST	9	25.0 (7.1-57.4)	25.0‡	8.3-53.3	7.1-57.4
C	Surgery or radiotherapy was not provided	5	23.4 (12.5-56.9)	23.4-33.6	21.6-56.9	12.5‡
	All categories	29*	39.1 (7.1-74.0)	14.3-60.1	8.3-74.0	7.1-62.6

BSC, best supportive care; **NST,** no systemic treatment; **ST,** systemic treatment.
*Number does not add up because several studies report patients from several groups. †Rest of the world included Russia, Japan, South Korea, and Middle East countries. ‡No range was provided because only one study was available.

Table 2. Factors associated with NST

Reference	Parikh et al. 2019[13]	Ikeda et al. 2020[26]	Geynisman et al. 2022[4]	Bilen et al. 2021[10]	Reesink et al. 2020[9]	Richters et al. 2020[19]
Geographic location	USA	Japan	USA	USA	The Netherlands	The Netherlands
Study period	2015-2017	1990-2015	2011-2020	2015-2019	2008-2016	2016-2017
Statistics	Reported	Reported	Post hoc	Post hoc	Post hoc	Post hoc
Older age	<65 vs ≥65 years	Median age	<65 vs ≥65 years	—	<70 vs ≥70 years	<60 vs ≥60 years
	p<0.005	p<0.05	p<0.05		p<0.001	p<0.001
Poor ECOG PS	0 -1 vs ≥2	—	0 -1 vs ≥2	—	—	0 -1 vs ≥2
	p<0.05		NS			p<0.001
Poor renal function	—	eGFR change rate: normal vs moderate vs severe	—	—	eGFR ≤30 vs >30; ≤60 vs >60 mL/min/1.73 m²	eGFR ≤30 vs >30; ≤60 vs >60 mL/min/1.73 m²
		NS			p<0.05	p<0.001
Metastatic disease	—	—	—	—	M0 vs M1	Lymph nodes only vs spread outside lymph nodes
					p<0.05	p<0.001
Comorbidities	—	—	—	Comorbidities vs no comorbidities	—	No. of comorbidities 0-1 vs ≥2
				p<0.001		p<0.05
Race	White vs Non-White	—	White vs Non-White	White vs Non-White	—	—
	NS		p<0.05	p<0.001		
Female sex	Male vs female	Male vs female	Male vs female	Male vs female	Male vs female	Male vs female
	NS	NS	NS	p<0.001	NS	NS
Primary organ being bladder	—	—	Bladder vs nonbladder	—	—	—
			p<0.001			
Higher stage at initial diagnosis	—	T stage <pT3 vs ≥pT3	—	—	—	—
		NS				

BSC, best supportive care; **eGFR,** estimated glomerular filtration rate; **NS,** nonsignificant; **NST,** no systemic treatment; **ST,** systemic treatment.
*Number does not add up because several studies report patients from several groups.

REFERENCES **1.** Powles T, et al. Ann Oncol. 2022;33(3):244-58. **2.** Cathomas R, et al. Eur Urol. 2022;81(1):95-103. **3.** Flaig TW, et al. J Natl Compr Canc Netw. 2020;18(3):329-54. **4.** Geynisman DM, et al. Urol Oncol. 2022;40(5):195.e1-195.e11. **5.** Swami U, et al. Cancer Treat Res Commun. 2021;27:100325. **6.** Petkau V, et al. J Clin Oncol. 2021;39(Suppl 28). Abstract 88. **7.** Vafaei-Nodeh S, et al. J Clin Oncol. 2020;38(Suppl 6). Abstract 450. **8.** Richters A, et al. Eur Urol Focus. Published online 14 September 2021. **9.** Reesink DJ, et al. Sci Rep. 2020;10(1):15822. **10.** Bilen MA, et al. Presented online at: ESMO Congress 2021; 16-21 September 2021. Abstract 701P. **11.** Flannery K, et al. Future Oncol. 2019;15(12):1323-34. **12.** Mao W, et al. Int Urol Nephrol. 2019;51(7):1145-56. **13.** Parikh RB, et al. Oncologist. 2019;24(6):e397-9. **14.** Morgans AK, et al. Ann Oncol. 2021;32(Suppl 5):S714-5. Abstract 704P. **15.** Doshi G, et al. Value Health. 2018;21(Suppl 3):S72. Abstract PCN339. **16.** Davies FJ, et al. Value Health. 2020;23(Suppl 2):S480-1. Abstract PCN329. **17.** Kearney M, et al. Value Health. 2020;23(Suppl 2):S483. Abstract PCN341. **18.** Jensen JB, et al. Ann Oncol. 2021;32(Suppl 5):S716. Abstract 707P. **19.** Richters A, et al. Cancer Treat Res Commun. 2020;25:100266. **20.** Sevillano E, et al. J Clin Oncol. 2020;38(Suppl 15). Abstract e17042. **21.** Sørup S, et al. Value Health. 2021;24(Suppl 1):S65. Abstract PCN242. **22.** Fukushima H, et al. J Urol. 2017;197(4S):e433-4. Abstract MP34-17. **23.** Miyake M, et al. Clin Genitourin Cancer. 2022;20(2):196.e1-196.e9. **24.** Okuyama A, Higashi T. Jpn J Clin Oncol. 2018;48(5):417-25. **25.** Kohei N, et al. SAGE Open Med. 2018;6:2050312118783011. **26.** Ikeda M, et al. Chemotherapy. 2020;65(5-6):134-40. **27.** Park K, et al. Ewha Med J. 2021;44(3):63-9. **28.** Abu-Hijlih RA, et al. J Clin Oncol. 2021;39(Suppl 15). Abstract e16521.

DISCLOSURES T. Wilke reports employment with IPAM and Cytel, and has served in advisory roles for Novo Nordisk, AbbVie, Merck, GSK, BMS, Leo Pharma, Bayer, and Boehringer Ingelheim. **L. Zhang, E. Hubscher, M. Musat, and S. Harricharan** report employment with Cytel and report consulting services for Merck. **M. Kearney** reports employment with Merck Healthcare KGaA, Darmstadt, Germany, and holds stocks/shares at Merck, Novartis, and UCB Biopharma SPRL. **ACKNOWLEDGMENTS** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This research was sponsored by Merck (CrossRef Funder ID:10.13039/100009945) as part of an alliance between Merck and Pfizer. Editorial support was provided by Katherine Quiroz-Figueroa of ClinicalThinking and was funded by Merck and Pfizer.

Poster No. 1765P. Presented at the ESMO Congress 2022, 9-13 September 2022; Paris, France; Hybrid.