Evaluation of novel anti-TIGIT antibody M6223 as a single agent and in combination with avelumab on human natural killer cell cytotoxicity

Anton Titov*[1], Nancy Sun[1], Christie Kelton[1], Mirek Jurzak[1], Hong Ma[1], Chunxiao Xu[1]

*Presenting and corresponding author (anton.titov@emdserono.com)

[1]EMD Serono, Billerica, MA, USA

GET POSTER PDF

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without written permission of the authors

CONCLUSIONS

Blockade of T cell immunoreceptor with Ig and ITIM domains (TIGIT) increases the cytotoxicity of CD56+ natural killer (NK) cells

M6223 plus avelumab elicits additive responses in NK cells, indicating that CD16-mediated antibody-dependent cell mediated cytotoxicity (ADCC) likely augments TIGIT blockade

NK cells may play an important role in the clinical efficacy of anti-TIGIT blockade but further *in vivo* studies are required

M6223 plus avelumab is being studied as first-line maintenance therapy for advanced urothelial carcinoma (UC) in the phase 2 JAVELIN Bladder Medley umbrella trial (NCT05327530)

INTRODUCTION

- M6223 is a fully human antagonistic anti-TIGIT immunoglobulin G1 (IgG1) antibody with fragment crystallizable (Fc)-mediated effector function[1,2]
- Preclinical studies demonstrated that M6223 could induce an antitumor immune response through several mechanisms, including direct blockade of the TIGIT pathway, stimulation of CD226 dimerization/activation and depletion of TIGIT+ immune subsets by Fc-mediated effector function[3]
 - NK cells have a wide range of TIGIT expression among healthy subjects[4]
- Avelumab is a human IgG1 anti–PD-L1 antibody with a wild-type Fc region that induces antitumor activity *in vitro* via both adaptive effector cells (T cells) and innate immune effector cells (ADCC via NK cells). It is approved for UC, renal cell carcinoma, and Merkel cell carcinoma[5,6]
- We report an evaluation of M6223 as a single agent and in combination with avelumab on human NK cell cytotoxicity

METHODS

- NK cell antitumor cytotoxicity was measured against the tumor cell line MDA-MB-231 with beta-2 microglobulin (B2M) knockout using fluorescent live cell imaging
 - Primary NK cells were treated with interleukin-15 for cryopreservation recovery
 - MDA-MB-231 wild-type cells were engineered to express green fluorescent protein and various gene knockouts including B2M, CD155 and CD112 to assess the dependence of NK cell antitumor cytotoxicity on the expression of these ligands
- MDA-MB-231 cell death was monitored using Incucyte® software to assess cytotoxicity relative to inactive IgG1 control
- Flow cytometry data were obtained on an LSRFortessa™ using single stain compensation and the following antibodies: anti-CD107a, viability dye efluor780 ebioscience 65-0865-14, anti-TIGIT C01 AF647 clone, anti-CD56, anti-DNAX accessory molecule-1 (DNAM1), and anti-CD16
- NK cells were treated with M6223 or its Fc effector null mutant version
- Avelumab was also tested alone or as a single-dose combination with M6223 to evaluate the additive potential of these antibodies

RESULTS

Effect of M6223 on NK cells

- M6223 induced significant NK cell antitumor cytotoxicity in 4 different NK donors at a dose level of 0.3 µg/mL versus Fc-mutant M6223 (*$p<0.05$; unpaired t-test with Welch's correction) with an EC_{50} value of approximately 100 ng/mL (**Table 1** and **Figure 1**)

Table 1 and Figure 1. Overall potency of M6223 or Fc-mutant M6223 in cytotoxicity assay using primary NK cells and MDA-MB-231 B2M-/- target

Non-linear fit EC_{50} (ng/mL)	Donor				Average (ng/mL)
	1	2	3	4	
M6223	73.0	188.7	38.8	139.2	**109.9**
Fc-mutant M6223	345.6	733.7	817.5	1280.0	**794.2**

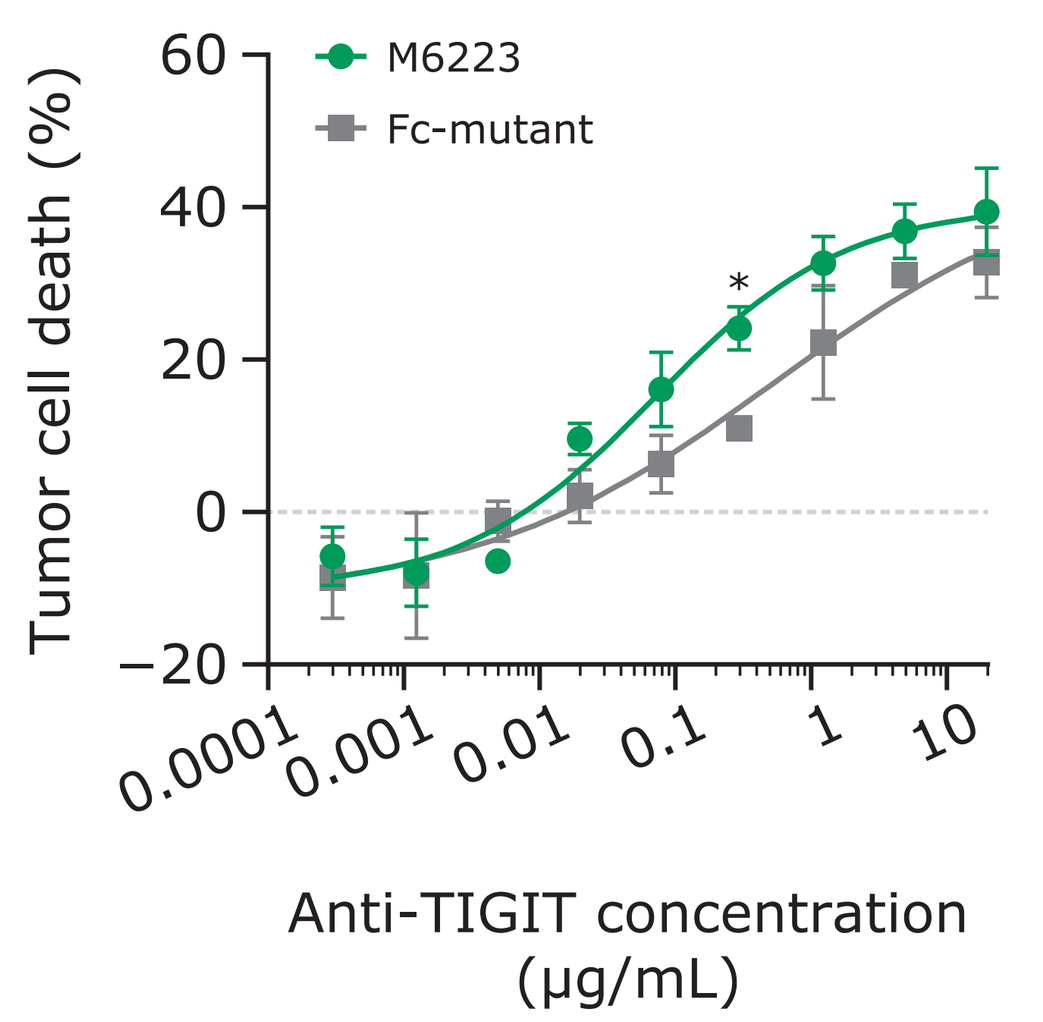
M6223
Fc-mutant
Tumor cell death (%)
Anti-TIGIT concentration (µg/mL)

- Flow cytometry profiling data is shown in **Figure 2** for primary CD56+ NK donor cells after treatment with 20 µg/mL M6223 or Fc-mutant M6223 (representative donor cells shown at time 0 hours before co-culture or treatment)

Figure 2. Flow cytometry profiling data

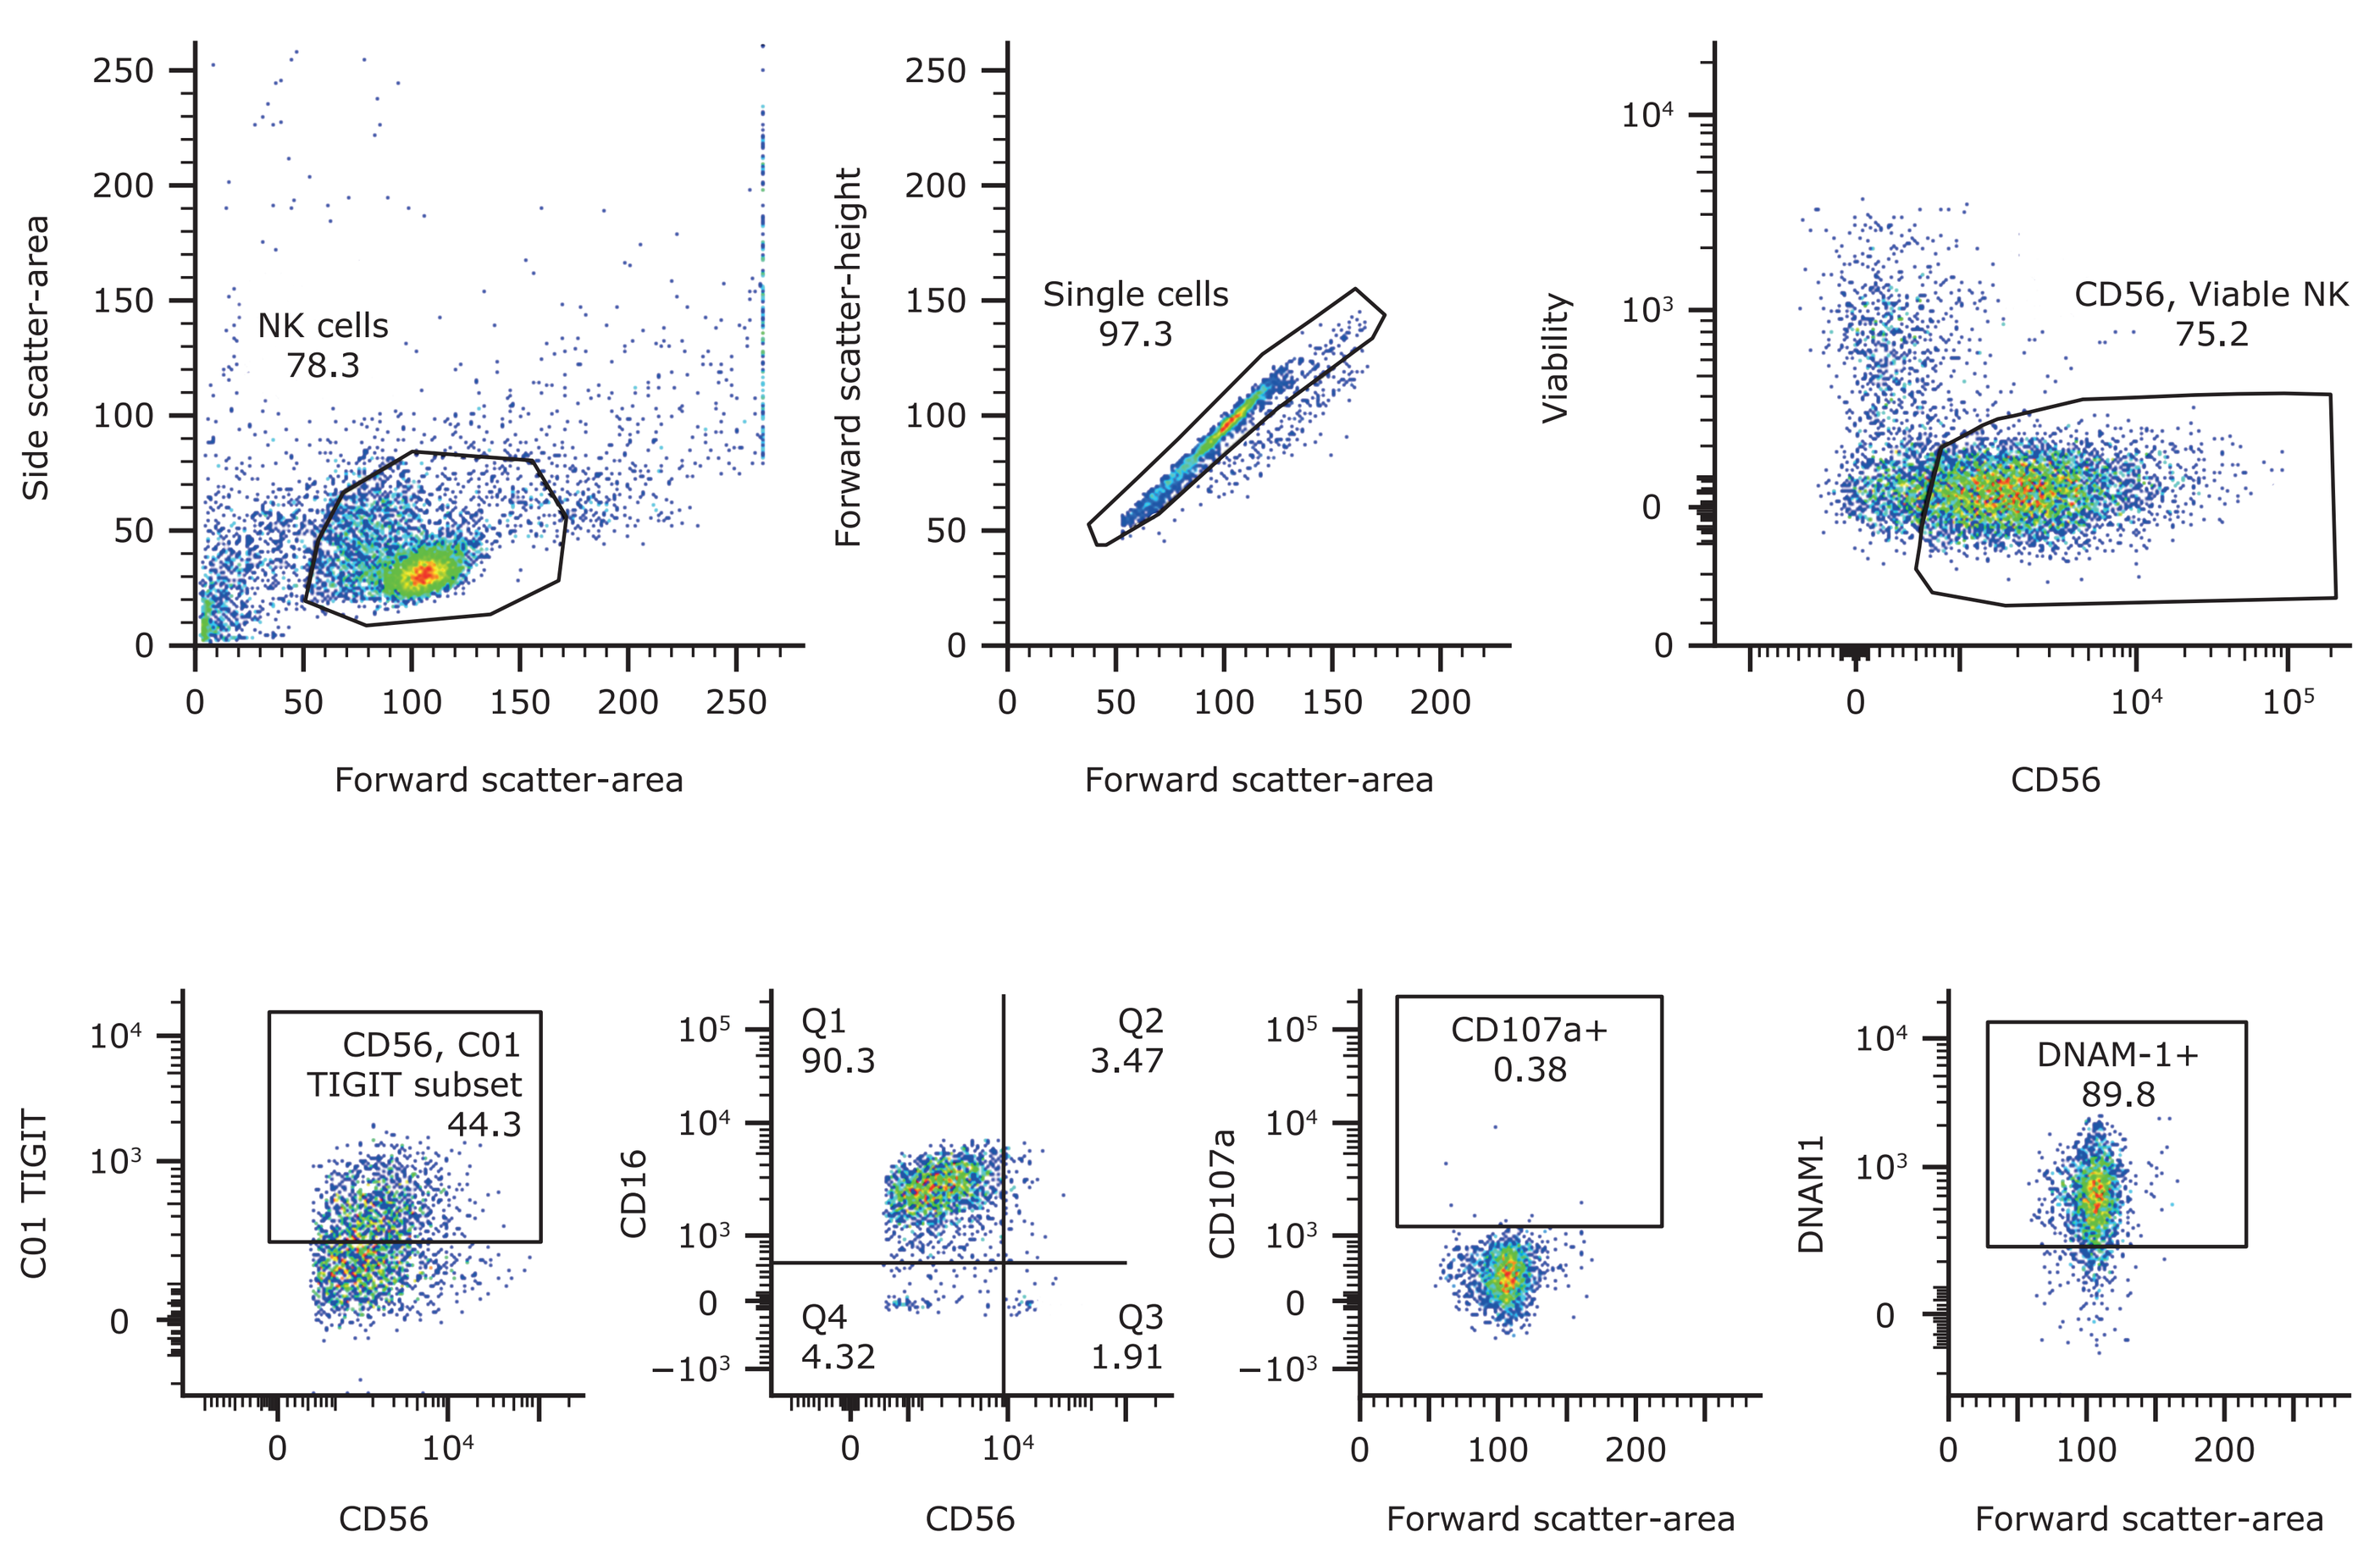
NK cells 78.3
Side scatter-area
Forward scatter-area
Single cells 97.3
Forward scatter-height
Forward scatter-area
CD56, Viable NK 75.2
Viability
CD56
CD56, C01 TIGIT subset 44.3
C01 TIGIT
CD56
Q1 90.3
Q2 3.47
Q4 4.32
Q3 1.91
CD16
CD56
CD107a+ 0.38
CD107a
Forward scatter-area
DNAM-1+ 89.8
DNAM1
Forward scatter-area

RESULTS CONTINUED

Effect of CD155 and CD112 expression on M6223 activity

- CD155 and CD112 wild-type expression was required for maximum efficacy of M6223 in MDA-MB-231 B2M-/- cells; reduced CD155 expression and CD112 knockout correlated with lower cytotoxicity (**Figures 3–6**)
- Antitumor cytotoxicity was retained with CD155 expression on target MDA-MB-231 cells at a wild-type level of 30–50% and was almost completely eliminated in CD155-/- cells (**Figures 3** and **4**)
- CD112 alone could not restore M6223 efficacy, consistent with a weaker role of DNAM-1-mediated recognition of this receptor (**Figures 5** and **6**)

Figure 3. 20 µg/mL M6223 (a) or Fc-mutant M6223 (b) in knock-down cell lines with varied CD155 expression

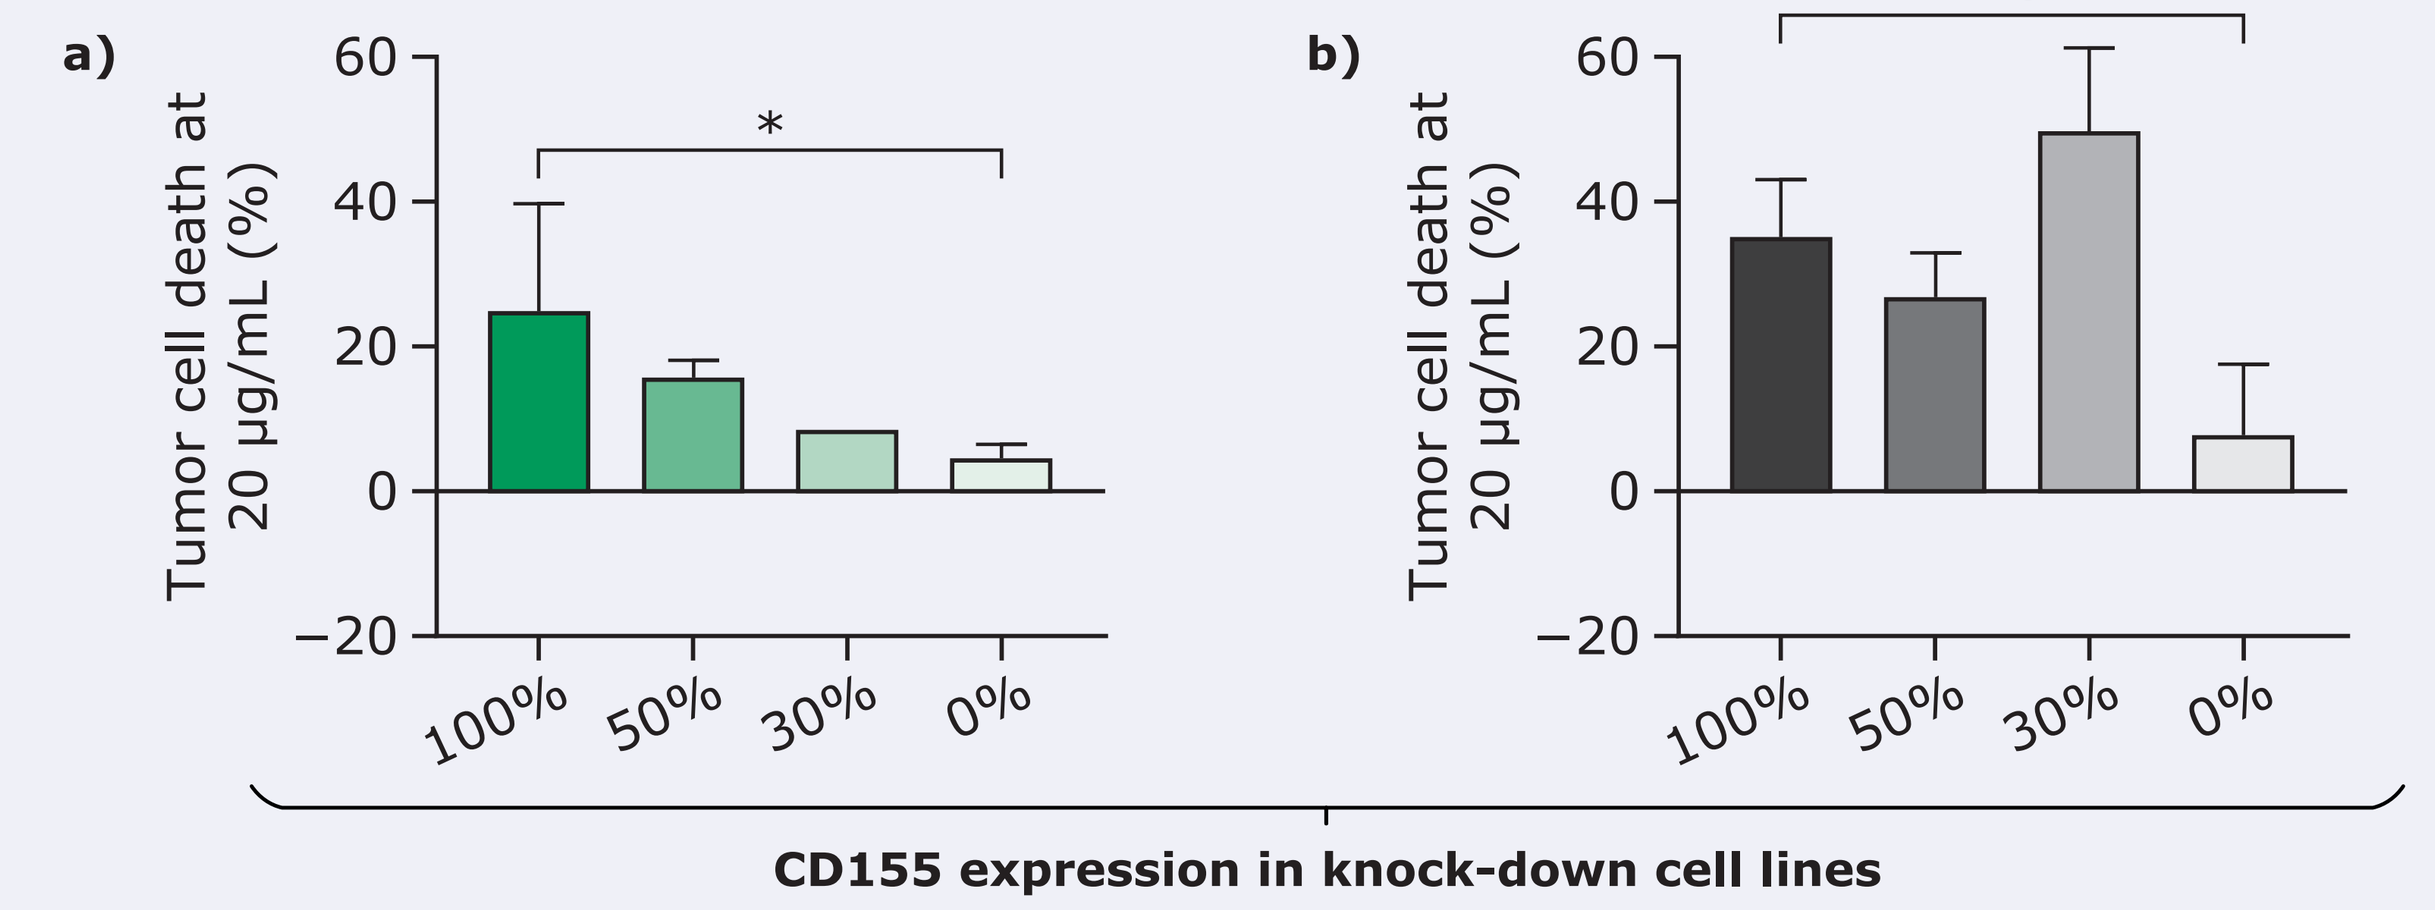
a)
b)
Tumor cell death at 20 µg/mL (%)
*
ns
100%
50%
30%
0%
CD155 expression in knock-down cell lines

*$p<0.05$ (unpaired t-test with Welch's correction); ns=nonsignificant

Figure 4. 0.1 µg/mL M6223 (a) or Fc-mutant M6223 (b) in knock-down cell lines with varied CD155 expression

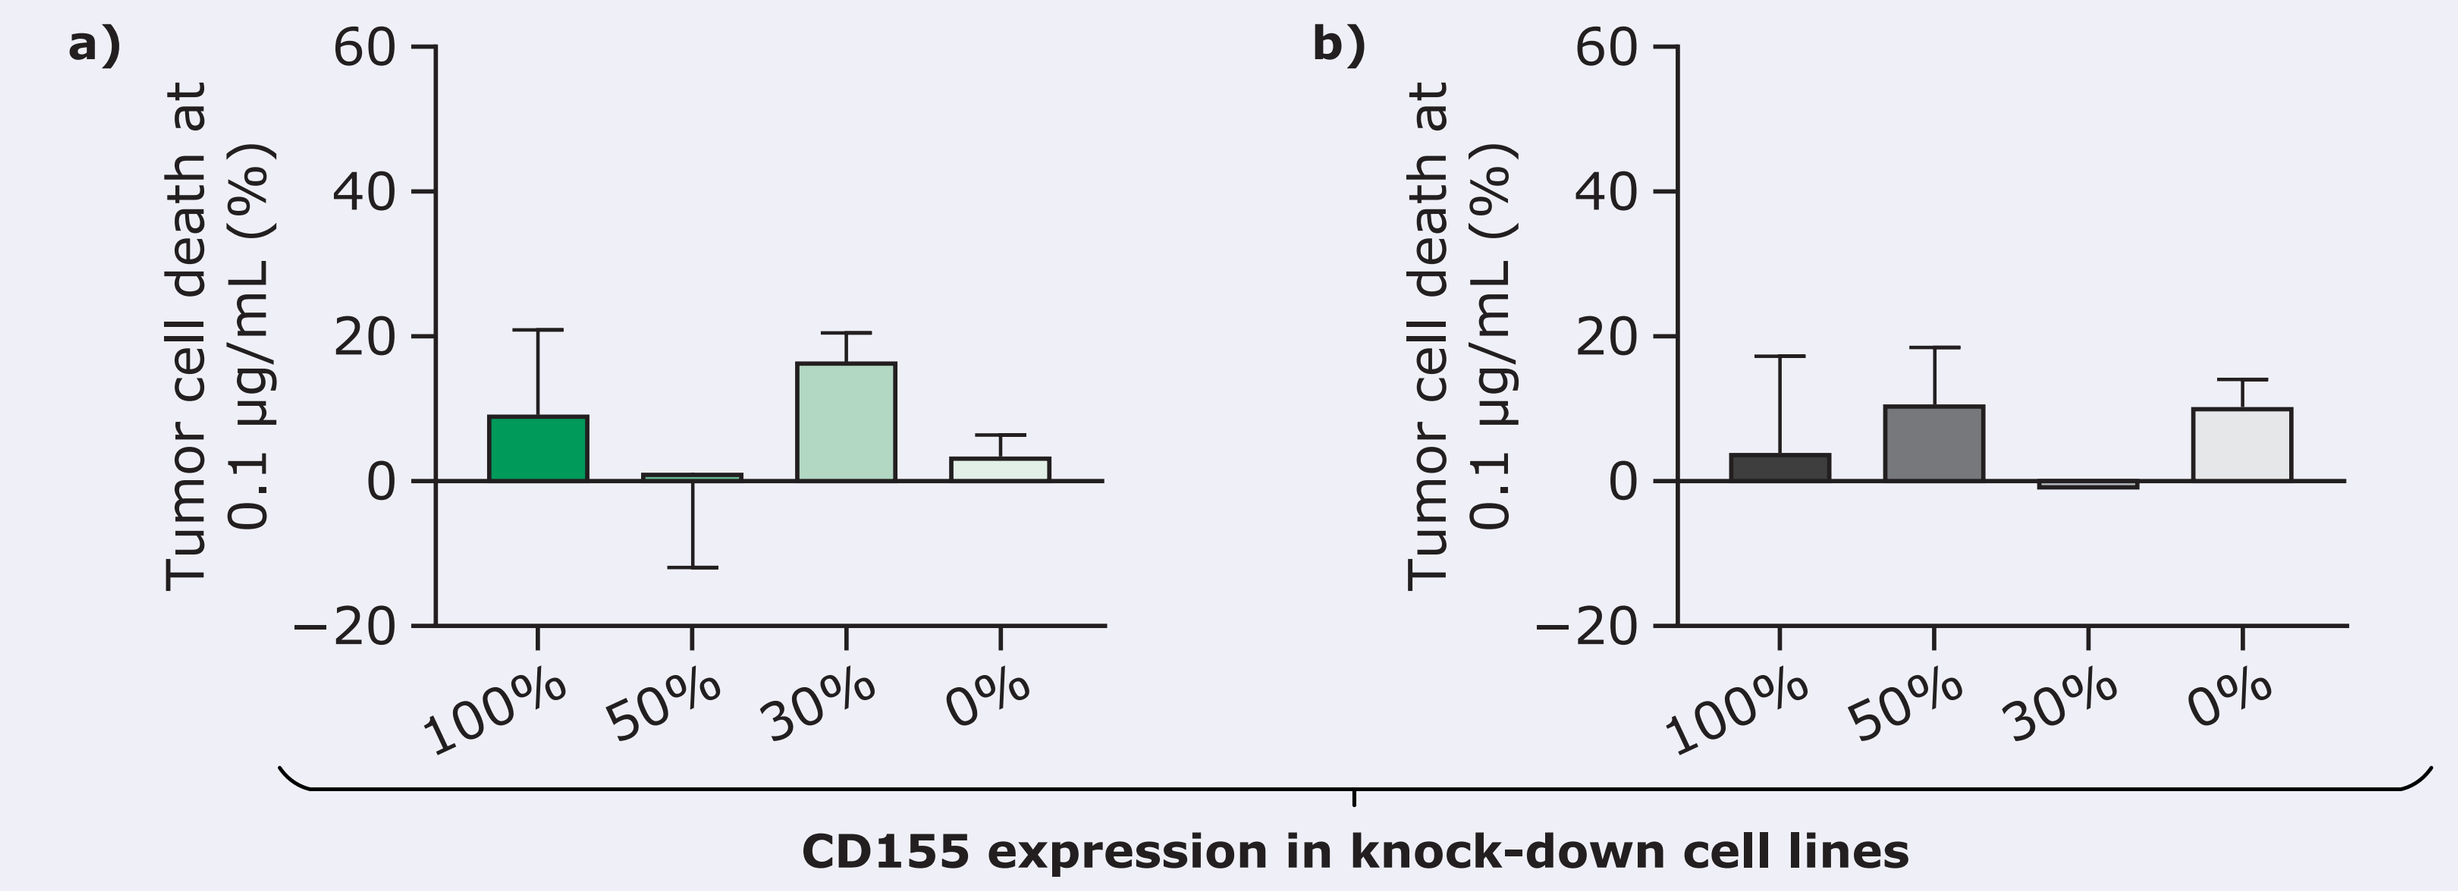
a)
b)
Tumor cell death at 0.1 µg/mL (%)
100%
50%
30%
0%
CD155 expression in knock-down cell lines

Figure 5. 20 µg/mL M6223 or Fc-mutant M6223 in cell lines with CD155 knockout and CD112 wild-type expression

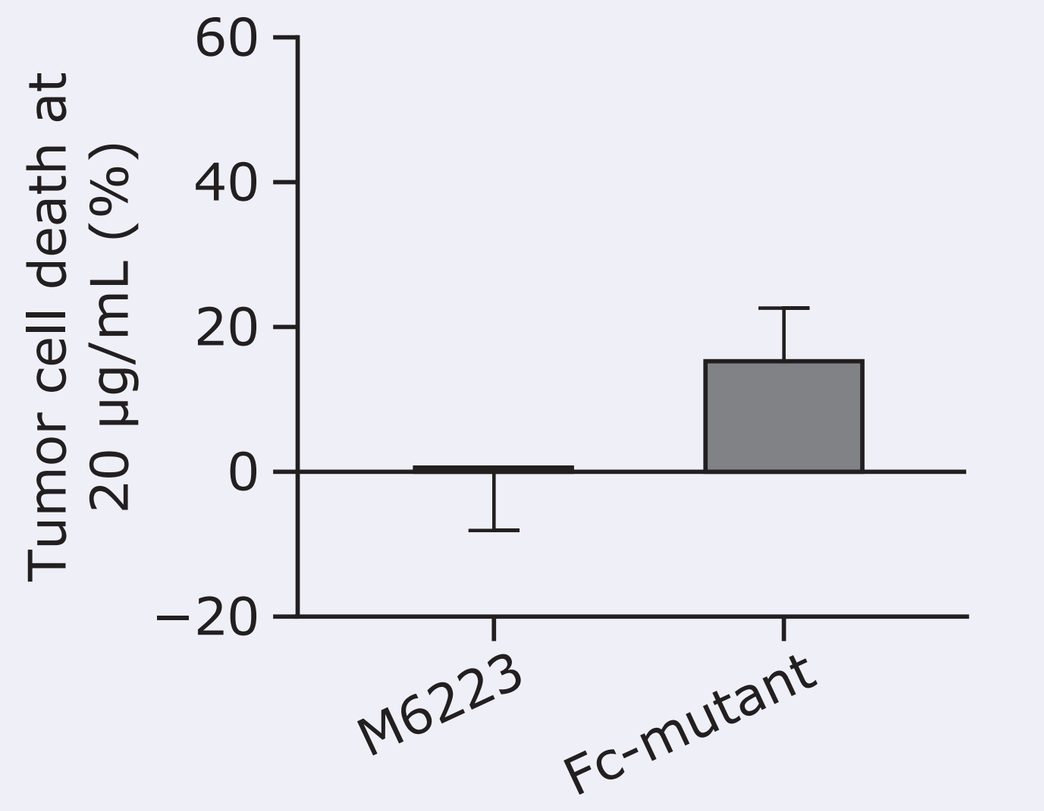
Tumor cell death at 20 µg/mL (%)
M6223
Fc-mutant

Figure 6. 0.1 µg/mL M6223 or Fc-mutant M6223 in cell lines with CD155 knockout and CD112 wild-type expression

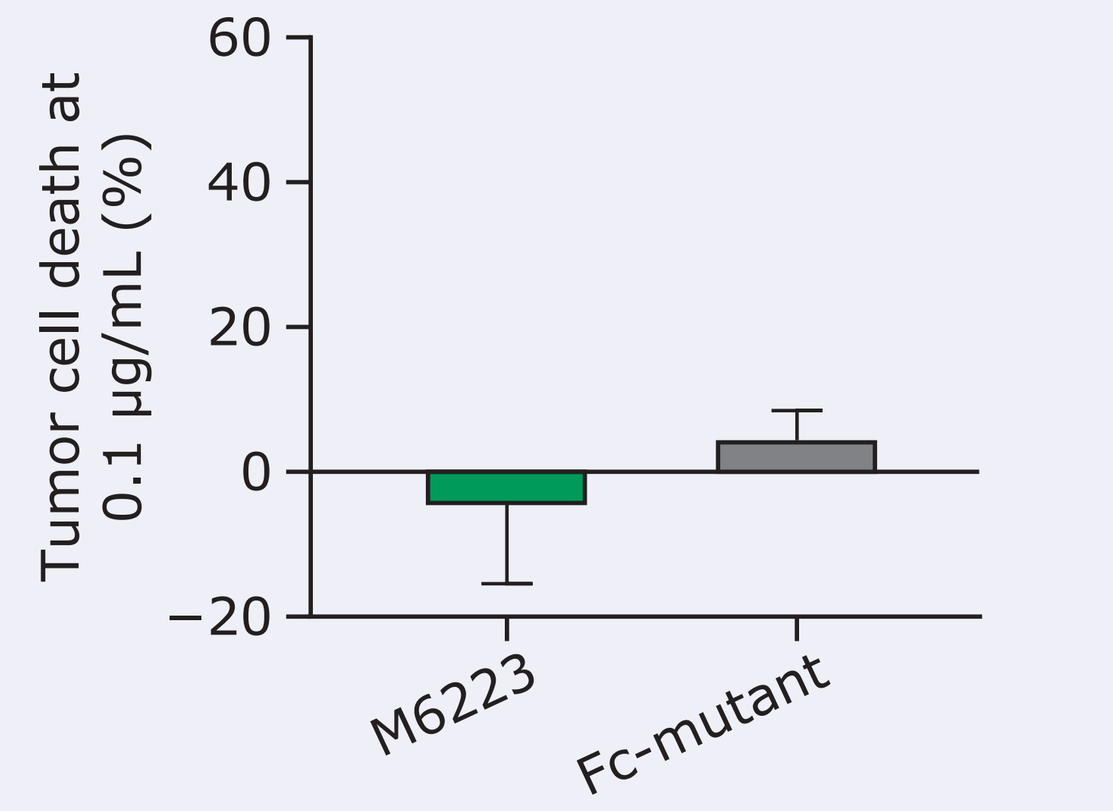
Tumor cell death at 0.1 µg/mL (%)
M6223
Fc-mutant

Effect of M6223 and avelumab alone or in combination on NK cell activity

- Avelumab facilitates CD16-mediated ADCC against PDL-1+ MDA-MB-231 cells (**Figure 7**)
- M6223 in combination with avelumab was more effective in killing MDA-MB-231 cells than M6223 alone (**Figure 8**)

Figure 7. Effect of avelumab on NK cell cytotoxicity

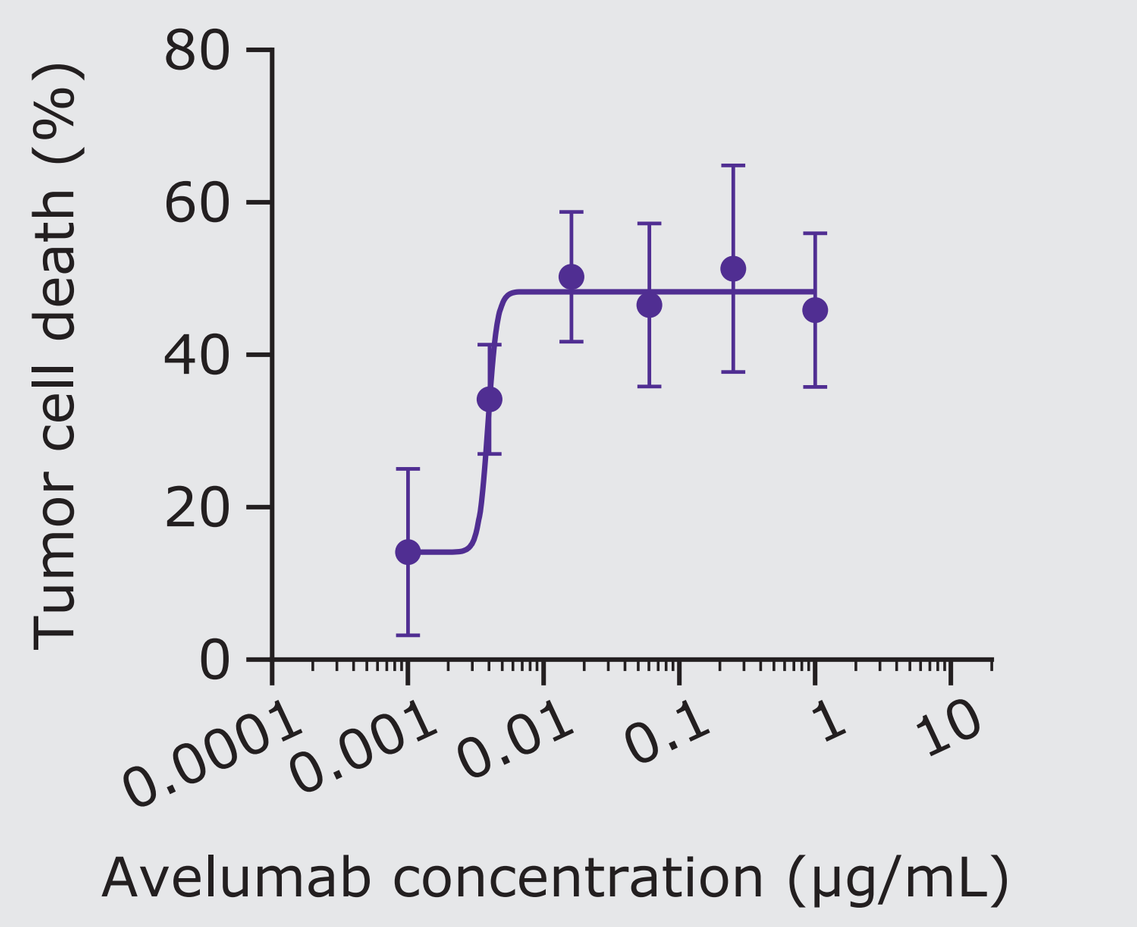
Tumor cell death (%)
Avelumab concentration (µg/mL)

Figure 8. Cytotoxicity of NK cells against MDA-MB-231 tumor cells following treatment with M6223 ± 1 µg/mL (a) or 4 ng/mL (b) avelumab

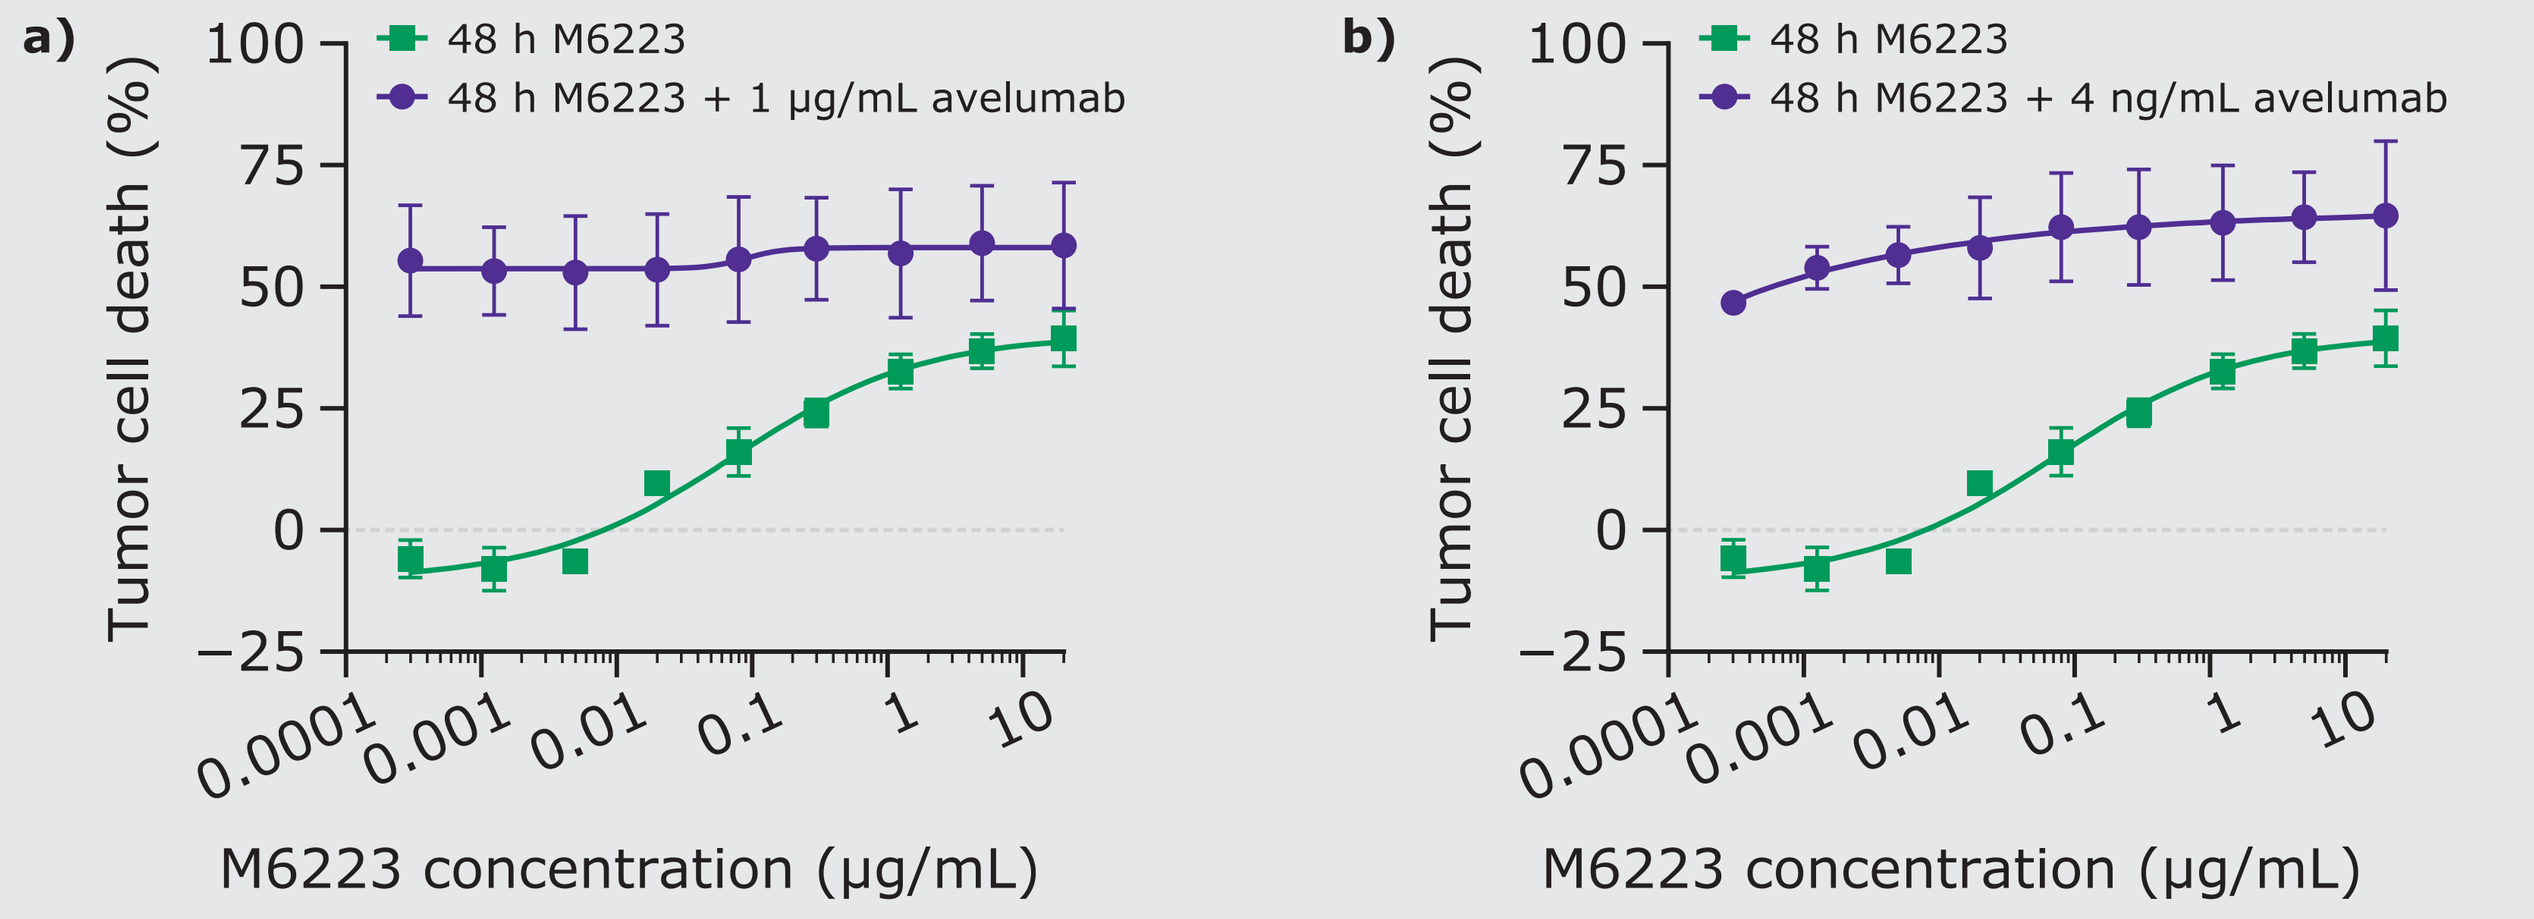
a)
48 h M6223
48 h M6223 + 1 µg/mL avelumab
b)
48 h M6223
48 h M6223 + 4 ng/mL avelumab
Tumor cell death (%)
M6223 concentration (µg/mL)

Impact of M6223 effector component on NK cell activity

- At 4 hours post-treatment, CD56+CD107a+ NK cells had numerically increased with M6223 treatment and had significantly increased with Fc-mutant M6223 treatment (*$p<0.05$; unpaired t-test with Welch's correction) (**Figure 9**)

Figure 9. CD56+CD107a+ NK cells at 4 (a) and 24 (b) hours post-M6223 or Fc-mutant M6223 antibody treatment

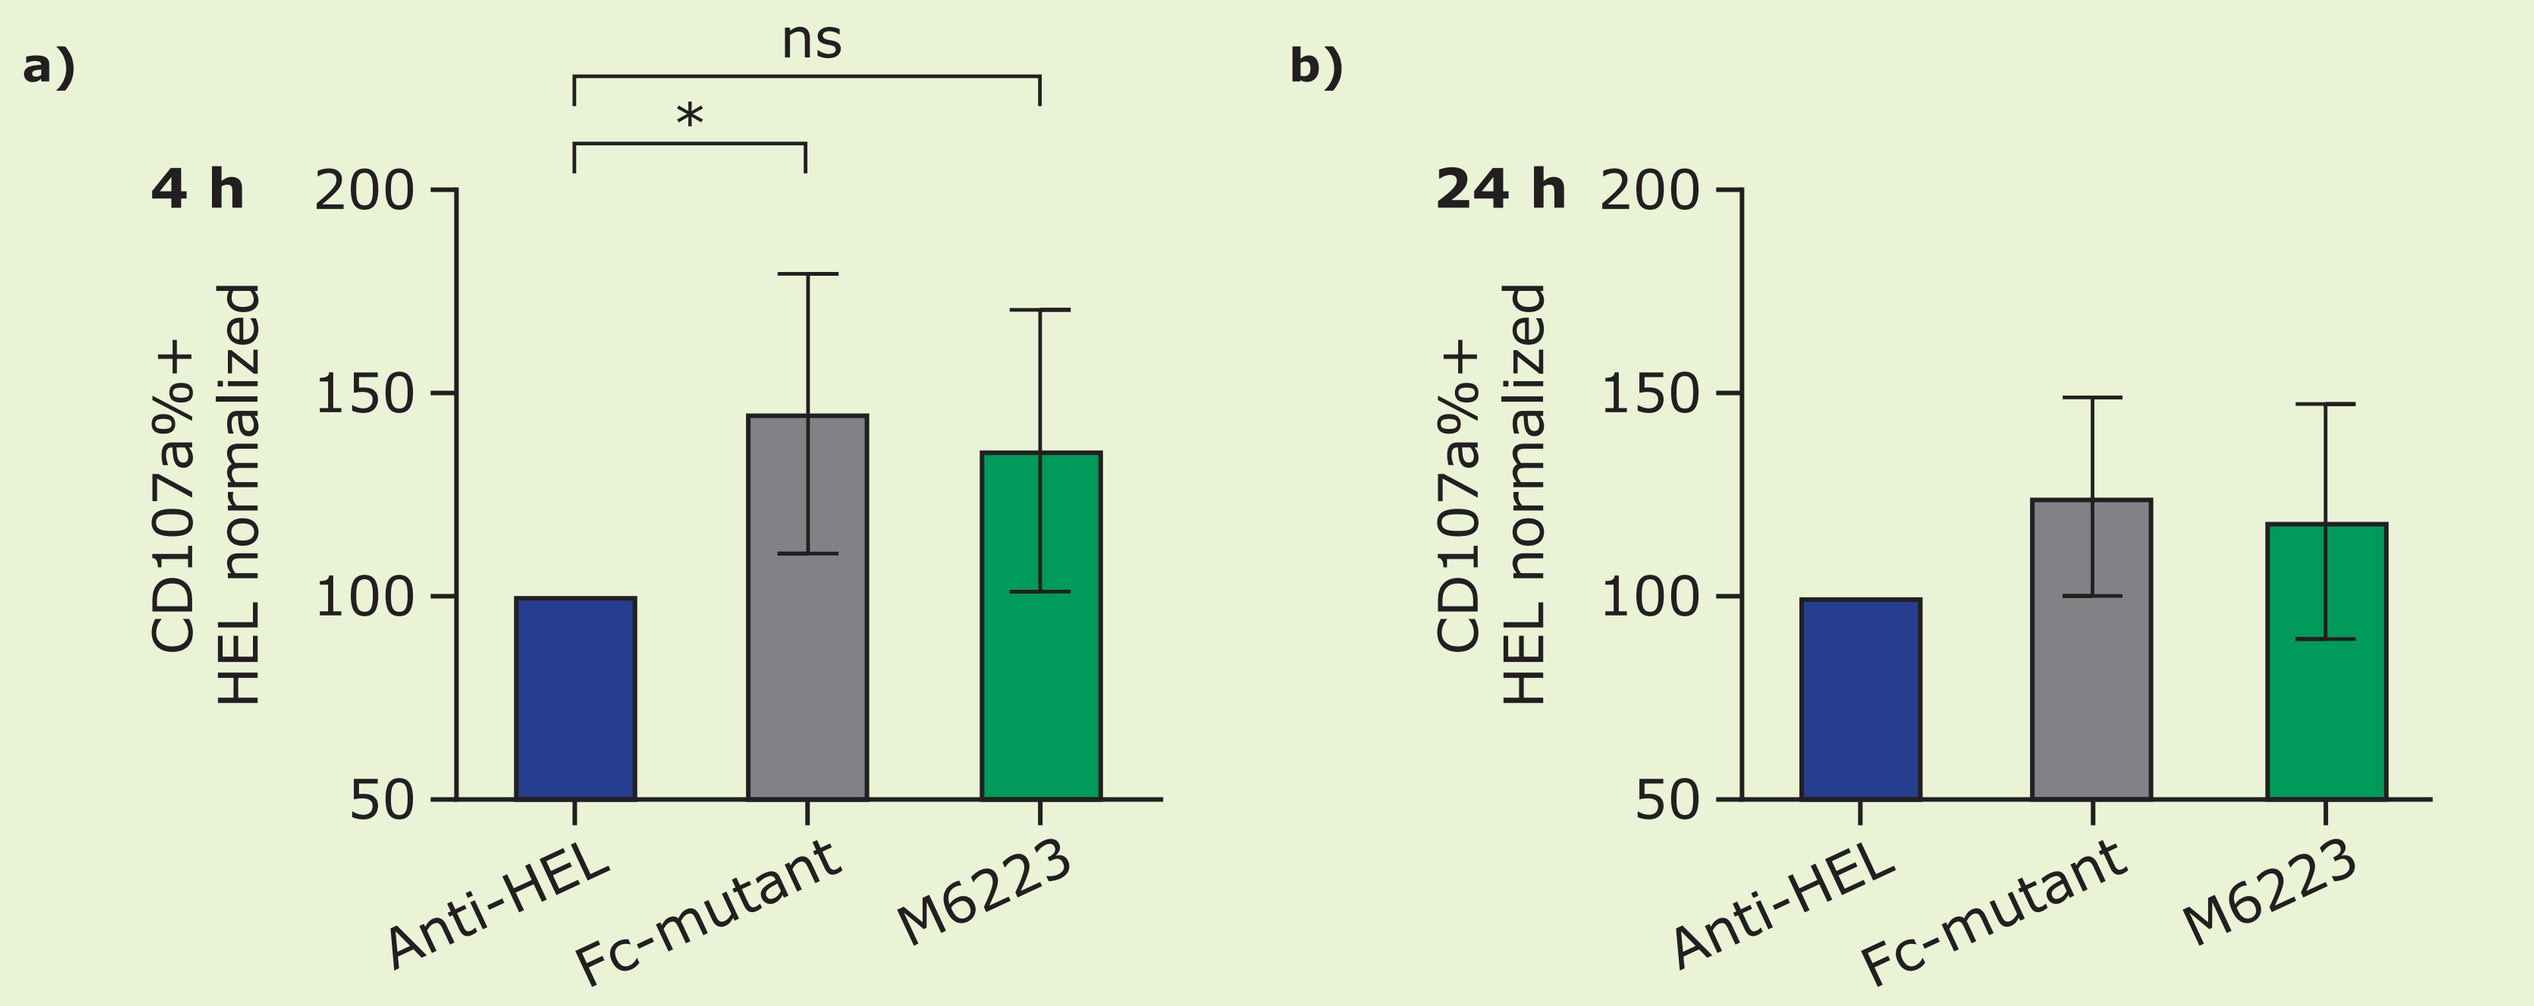
a)
4 h
ns
*
b)
24 h
CD107a%+ HEL normalized
Anti-HEL
Fc-mutant
M6223

- Viable CD56^dim^CD16+ NK cells slightly decreased by 24 hours of treatment with either M6223 or Fc-mutant antibody (*$p<0.05$ for both M6223 and Fc-mutant M6223 vs anti-HEL; unpaired t-test with Welch's correction) (**Figure 11**)

- There was limited *in vitro* NK fratricide and an Fc-mutant version of M6223 reduced antitumor cytotoxicity (**Figure 10**)

Figure 10. CD56+TIGIT+ NK cells stained with a non-competitive antibody at 4 (a) and 24 (b) hours post-M6223 or Fc-mutant M6223 treatment

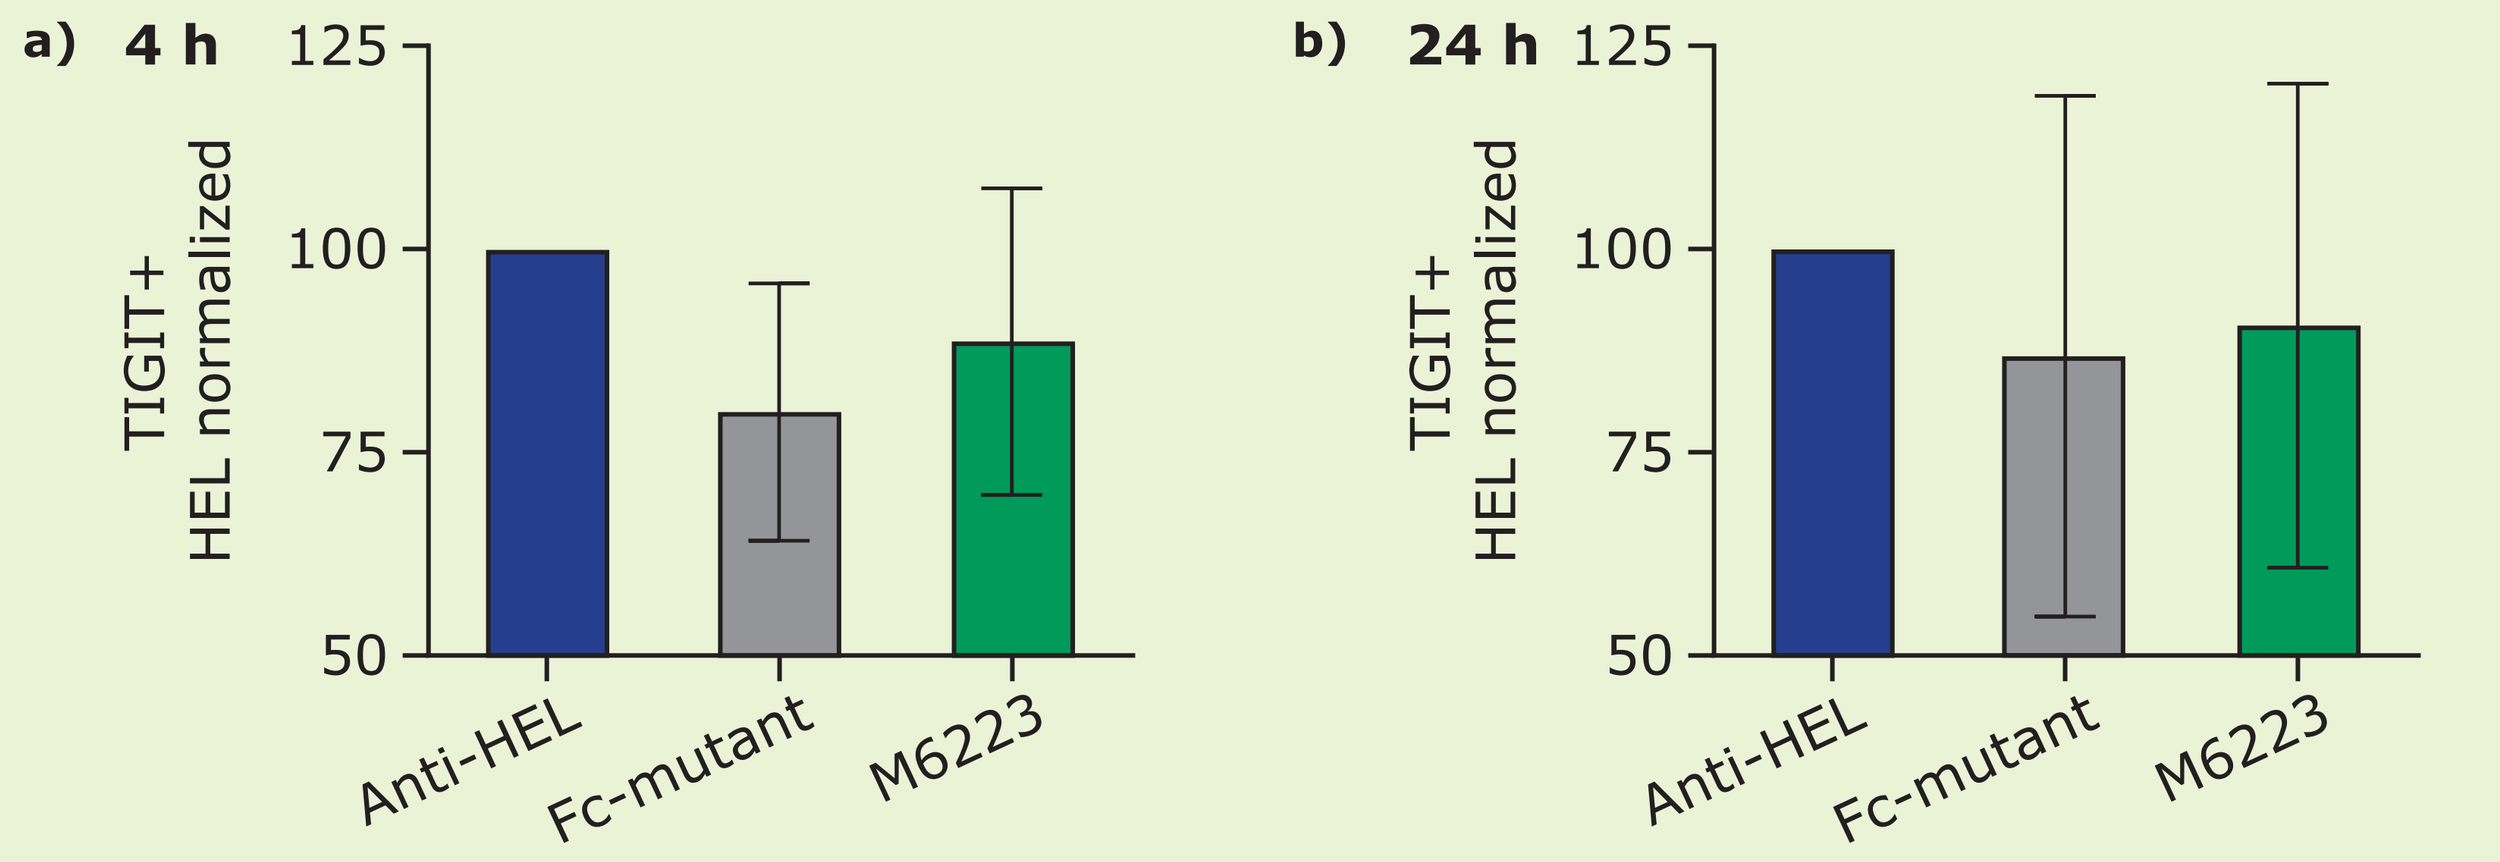
a) 4 h
b) 24 h
TIGIT+ HEL normalized
Anti-HEL
Fc-mutant
M6223

Figure 11. Levels of $CD56^{dim}$ CD16+ NK cells at 4 (a) and 24 (b) hours post-M6223 or Fc-mutant M6223 antibody treatment

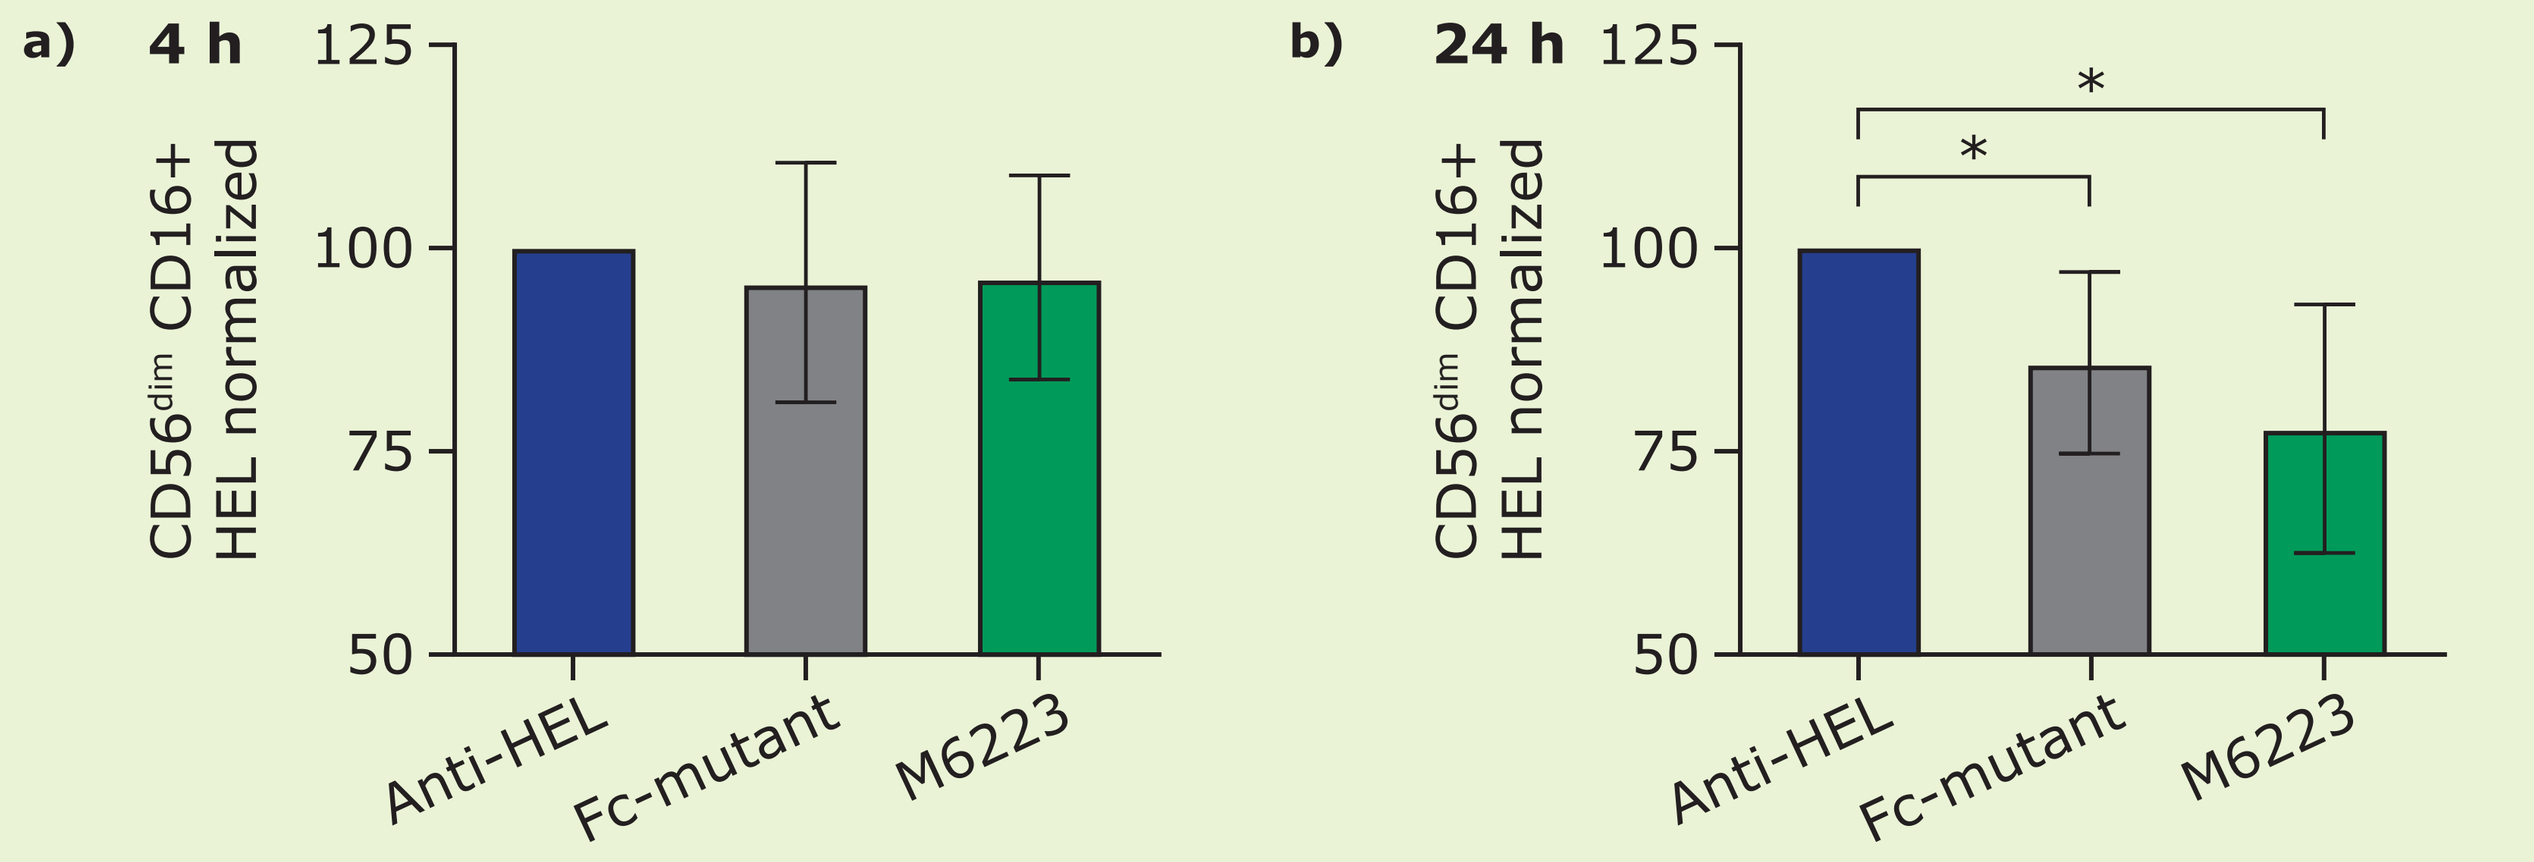
a) 4 h
b) 24 h
*
*
CD56dim CD16+ HEL normalized
Anti-HEL
Fc-mutant
M6223

References: 1. Harjunpää H & Guillerey C. Clin Exp Immunol 2019;200:108–119; **2.** Watson G, et al. J ImmunoTher Cancer 2021;9:A530; **3.** Xu C, et al. J Immunother Cancer 2021;8:doi: 10.1136/jitc-2021-SITC2021.326; **4.** Wang F, et al. Eur J Immunol. 2015;45:2886–2897; **5.** Hamilton G. Expert Opin Biol Ther 2021;21:311–322; **6.** Hamilton G and Rath B. Expert Opin Biol Ther. 2017;17:515–523

Acknowledgements: The trial was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Medical writing assistance was provided by Melody Watson of Bioscript Group, Macclesfield, UK, and funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)

Disclosures: All authors are employees of EMD Serono, Billerica, MA, USA

Poster 432. Presented at the Society for Immunotherapy of Cancer (SITC) 37th Annual Meeting | November 10–12 2022 | Boston, MA, USA