This reprint might contain references to “Merck” or “Merck KGaA”, which refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

Avelumab in patients with previously treated Merkel cell carcinoma (JAVELIN Merkel 200): updated overall survival data after >5 years of follow-up

P. T. Nghiem,[1] S. Bhatia,[2] A. S. Brohl,[3] O. Hamid,[4] J. M. Mehnert,[5] P. Terheyden,[6] K. C. Shih,[7,8] I. Brownell,[9] C. Lebbé,[10,11] K. D. Lewis,[12] G. P. Linette,[13] M. Milella,[14] H. Xiong,[15] G. Guezel,[16] S. P. D'Angelo,[17,18]

[1]Division of Dermatology, Department of Medicine, University of Washington Medical Center at South Lake Union, Seattle, WA, USA; [2]Department of Medicine, University of Washington Medical Center, Seattle, WA, USA; [3]Sarcoma Department and Cutaneous Oncology, Moffitt Cancer Center, Tampa, FL, USA; [4]Department of Medical Oncology, The Angeles Clinic and Research Institute, a Cedars-Sinai Affiliate, Los Angeles, CA, USA; [5]Division of Medical Oncology, Rutgers Cancer Institute of New Jersey, New Brunswick, NJ, USA; [6]Department of Dermatology, University of Lübeck, Lübeck, Germany; [7]Department of Medical Oncology, Sarah Cannon Research Institute, Nashville, TN, USA; [8]Department of Medical Oncology, Tennessee Oncology, Nashville, TN, USA; [9]Dermatology Branch, National Institutes of Health, Bethesda, MD, USA; [10]Dermatologie, Université de Paris, INSERM U976 Paris, France; [11] Dermatology and CIC, AP-HP, Saint Louis Hospital, Paris, France; [12]Department of Medicine, University of Colorado Denver School of Medicine, Aurora, CO, USA; [13]Center for Cellular Immunotherapies, University of Pennsylvania, Philadelphia, PA, USA; [14]Section of Oncology, Department of Medicine, University of Verona School of Medicine and Verona University Hospital Trust (AOUI Verona), Verona, Italy; [15]Biostatistics, EMD Serono Research & Development Institute, Inc., Billerica, MA, USA; an affiliate of Merck KGaA, Darmstadt, Germany; [16]Clinical Development, Merck KGaA, Darmstadt, Germany; [17]Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [18]Department of Medicine, Weill Cornell Medical College, New York, NY, USA

JAVELIN Merkel 200

SCOPE

- **We report long-term overall survival (OS) data, after >5 years of follow-up, from part A of the JAVELIN Merkel 200 study that evaluated avelumab monotherapy in a cohort of patients with metastatic Merkel cell carcinoma (mMCC) whose disease had progressed following ≥1 prior line of chemotherapy**

CONCLUSIONS

- **To our knowledge, this is the longest follow-up for a cohort of patients with mMCC treated with an immune checkpoint inhibitor reported to date**
- **Avelumab monotherapy led to meaningful long-term OS in patients with mMCC whose disease had progressed following chemotherapy**
- **Longer OS was reported in patients with PD-L1+ vs PD-L1– tumors, but, as previously reported, responses to avelumab occurred in patients regardless of PD-L1 status[1]**
 - **The OS benefit observed in both subgroups greatly exceeds that seen in retrospective analyses of second-line or later chemotherapy in patients with mMCC[2,3]**
- **These results further support the role of avelumab as a standard of care for patients with mMCC**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ASCO and the author of this poster

Correspondence: Paul Nghiem, **pnghiem@uw.edu**

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app or **click here** to view a plain language summary of the accepted scientific abstract

BACKGROUND

- Patients with mMCC have a poor prognosis, with a historical 5-year OS rate from diagnosis of approximately 14%[4]
- Although mMCC is considered sensitive to chemotherapy, duration of response tends to be limited[2,3,5]
 - Historically, in patients with chemotherapy-refractory mMCC, the 1-year OS rate following further chemotherapy treatment was 0%[2,3]
- Avelumab (anti–PD-L1) became the first approved treatment for patients with mMCC based on results from the phase 2 JAVELIN Merkel 200 trial[6]
 - The trial investigated avelumab monotherapy in 2 cohorts of patients with mMCC: as second-line or later treatment in patients with disease progression after ≥1 line of chemotherapy (part A) and as first-line treatment (part B)
- A summary of previously reported efficacy outcomes from part A, after ≥3 years of follow-up, are shown in **Table 1**
- Here we report 5-year OS data from part A of JAVELIN Merkel 200

Table 1. Efficacy outcomes after ≥3 years of follow-up[1]

	Overall population (N=88)	PD-L1+ (n=57)	PD-L1– (n=16)
ORR (95% CI), %	33.0 (23.3-43.8)	36.8 (24.4-50.7)	18.8 (4.0-45.6)
PFS rate (95% CI), %			
2 years	26 (17-36)	NA	NA
3 years	21 (12-32)	NA	NA
OS, median (95% CI), months	12.6 (7.5-17.1)	12.9 (8.7-29.6)	7.3 (3.4-14.0)
OS rate (95% CI), %			
3 years	32 (23-42)	NA	NA
3.5 years	31 (22-41)	NA	NA

NA, not available; **ORR**, objective response rate; **OS**, overall survival; **PFS**, progression-free survival.

METHODS

- The design of the phase 2, single-arm, open-label JAVELIN Merkel 200 trial (NCT02155647) has been reported previously[1,7]
- In part A, eligible patients had measurable (per RECIST 1.1) and histologically confirmed stage IV MCC that had progressed following ≥1 prior line of chemotherapy
 - Eligible patients also had an ECOG PS of 0-1 and were unselected for tumor PD-L1 expression
 - Patients were excluded if they had received previous therapy with immune checkpoint inhibitors
- Patients received avelumab 10 mg/kg by 1-hour intravenous infusion every 2 weeks until confirmed disease progression, unacceptable toxicity, or withdrawal
- OS was analyzed using the Kaplan-Meier method; 95% CIs for the median were calculated using the Brookmeyer-Crowley method
- PD-L1 expression was measured using the PD-L1 73-10 immunohistochemistry assay
 - PD-L1+ status was defined as ≥1% expression in tumor cells

RESULTS

Patient disposition

- A total of 88 patients were enrolled and treated with avelumab (**Table 2**)
 - Patient disposition is shown in **Table 3**
- As of September 25, 2020 (data cutoff), median follow-up was 65.1 months (range, 60.8-74.1 months)

Table 2. Baseline characteristics[1]

	N=88
Median age (range), years	72.5 (33-88)
Sex, n (%)	
Male	65 (73.9)
Female	23 (26.1)
ECOG PS, n (%)	
0	49 (55.7)
1	39 (44.3)
Site of primary tumor, n (%)	
Skin	67 (76.1)
Nonskin*	14 (15.9)
Missing	7 (8.0)
Visceral disease at study entry, n (%)	
Present	47 (53.4)
Absent	41 (46.6)
No. of prior systemic anticancer treatments, n (%)	
1	52 (59.1)
2	25 (28.4)
≥3	11 (12.5)
Tumor PD-L1 status, n (%)†	
Positive	57 (64.8)
Negative	16 (18.2)
Not evaluable	15 (17.0)
Tumor MCPyV status, n (%)	
Positive	46 (52.3)
Negative	31 (35.2)
Not evaluable	11 (12.5)

MCPyV, Merkel cell polyomavirus.
*Nonskin sites include lymph node (n=12 [13.6%]) and other sites (cheek mucosa and rectosigmoid junction; n=2 [2.3%]).
†PD-L1+ status was defined as expression on ≥1% of tumor cells, assessed using a Dako PD-L1 73-10 IHC assay.
From D'Angelo SP, et al. Avelumab in patients with previously treated metastatic Merkel cell carcinoma: long-term data and biomarker analyses from the single-arm phase 2 JAVELIN Merkel 200 trial. J Immunother Cancer. 2020;8:e000674. Creative Commons Attribution 4.0 International License (https://creativecommons.org/licenses/by/4.0/).

Table 3. Patient disposition

n (%)	N=88
Received ≥1 dose of study treatment	88 (100.0)
Treatment ongoing	1 (1.1)
Off treatment	87 (98.9)
Reason for discontinuation of treatment	87 (98.9)
AE	11 (12.5)
Lost to follow-up	1 (1.1)
Protocol noncompliance	1 (1.1)
Death	10 (11.4)
Disease progression	45 (51.1)
Withdrew consent	9 (10.2)
Other*	10 (11.4)
Discontinued treatment but still in follow-up	19 (21.6)
Reinitiated treatment with avelumab	1 (1.1)
Discontinued from the trial	68 (77.3)
Lost to follow-up	3 (3.4)
Death	58 (65.9)
Withdrew consent	7 (8.0)

AE, adverse event.
*Including complete response for ≥6 months on treatment (per protocol; n=5 [5.7%]) and switch to commercial avelumab for patient convenience (n=2 [2.3%]).

Overall survival

- Median OS was 12.6 months (95% CI, 7.5-17.1 months) in the overall population
 - OS rates at 4 and 5 years were 30% (95% CI, 20%-40%) and 26% (95% CI, 17%-36%), respectively
 - **Figure 1** shows OS for avelumab in comparison with retrospective analyses of second-line or later chemotherapy in patients with mMCC
- Longer median OS was observed in patients with PD-L1+ vs PD-L1– tumors (12.9 months [95% CI, 8.7-29.6 months] vs 7.3 months [95% CI, 3.4-14.0 months], respectively) and a higher 5-year OS rate (28% [95% CI, 17%-40%] vs 19% [95% CI, 5%-40%]) (**Figure 2**)

Deaths

- At data cutoff, 63 patients (71.6%) had died
 - The most common cause of death was disease progression (n=49 [55.7%])
 - Other causes were unknown reason (n=9 [10.2%]), adverse event (AE) not related to study treatment (n=3 [3.4%]), and other reason (n=2 [2.3%])
- No deaths due to treatment-related AEs were reported

Subsequent treatment

- In total, 26 patients (29.5%) received subsequent anticancer therapy (**Table 4**)
- The most common subsequent therapies were avelumab (n=4 [4.5%]), carboplatin + etoposide (n=4 [4.5%]), and pembrolizumab (n=4 [4.5%])

Figure 1. Overall survival in all patients compared with historical chemotherapy data

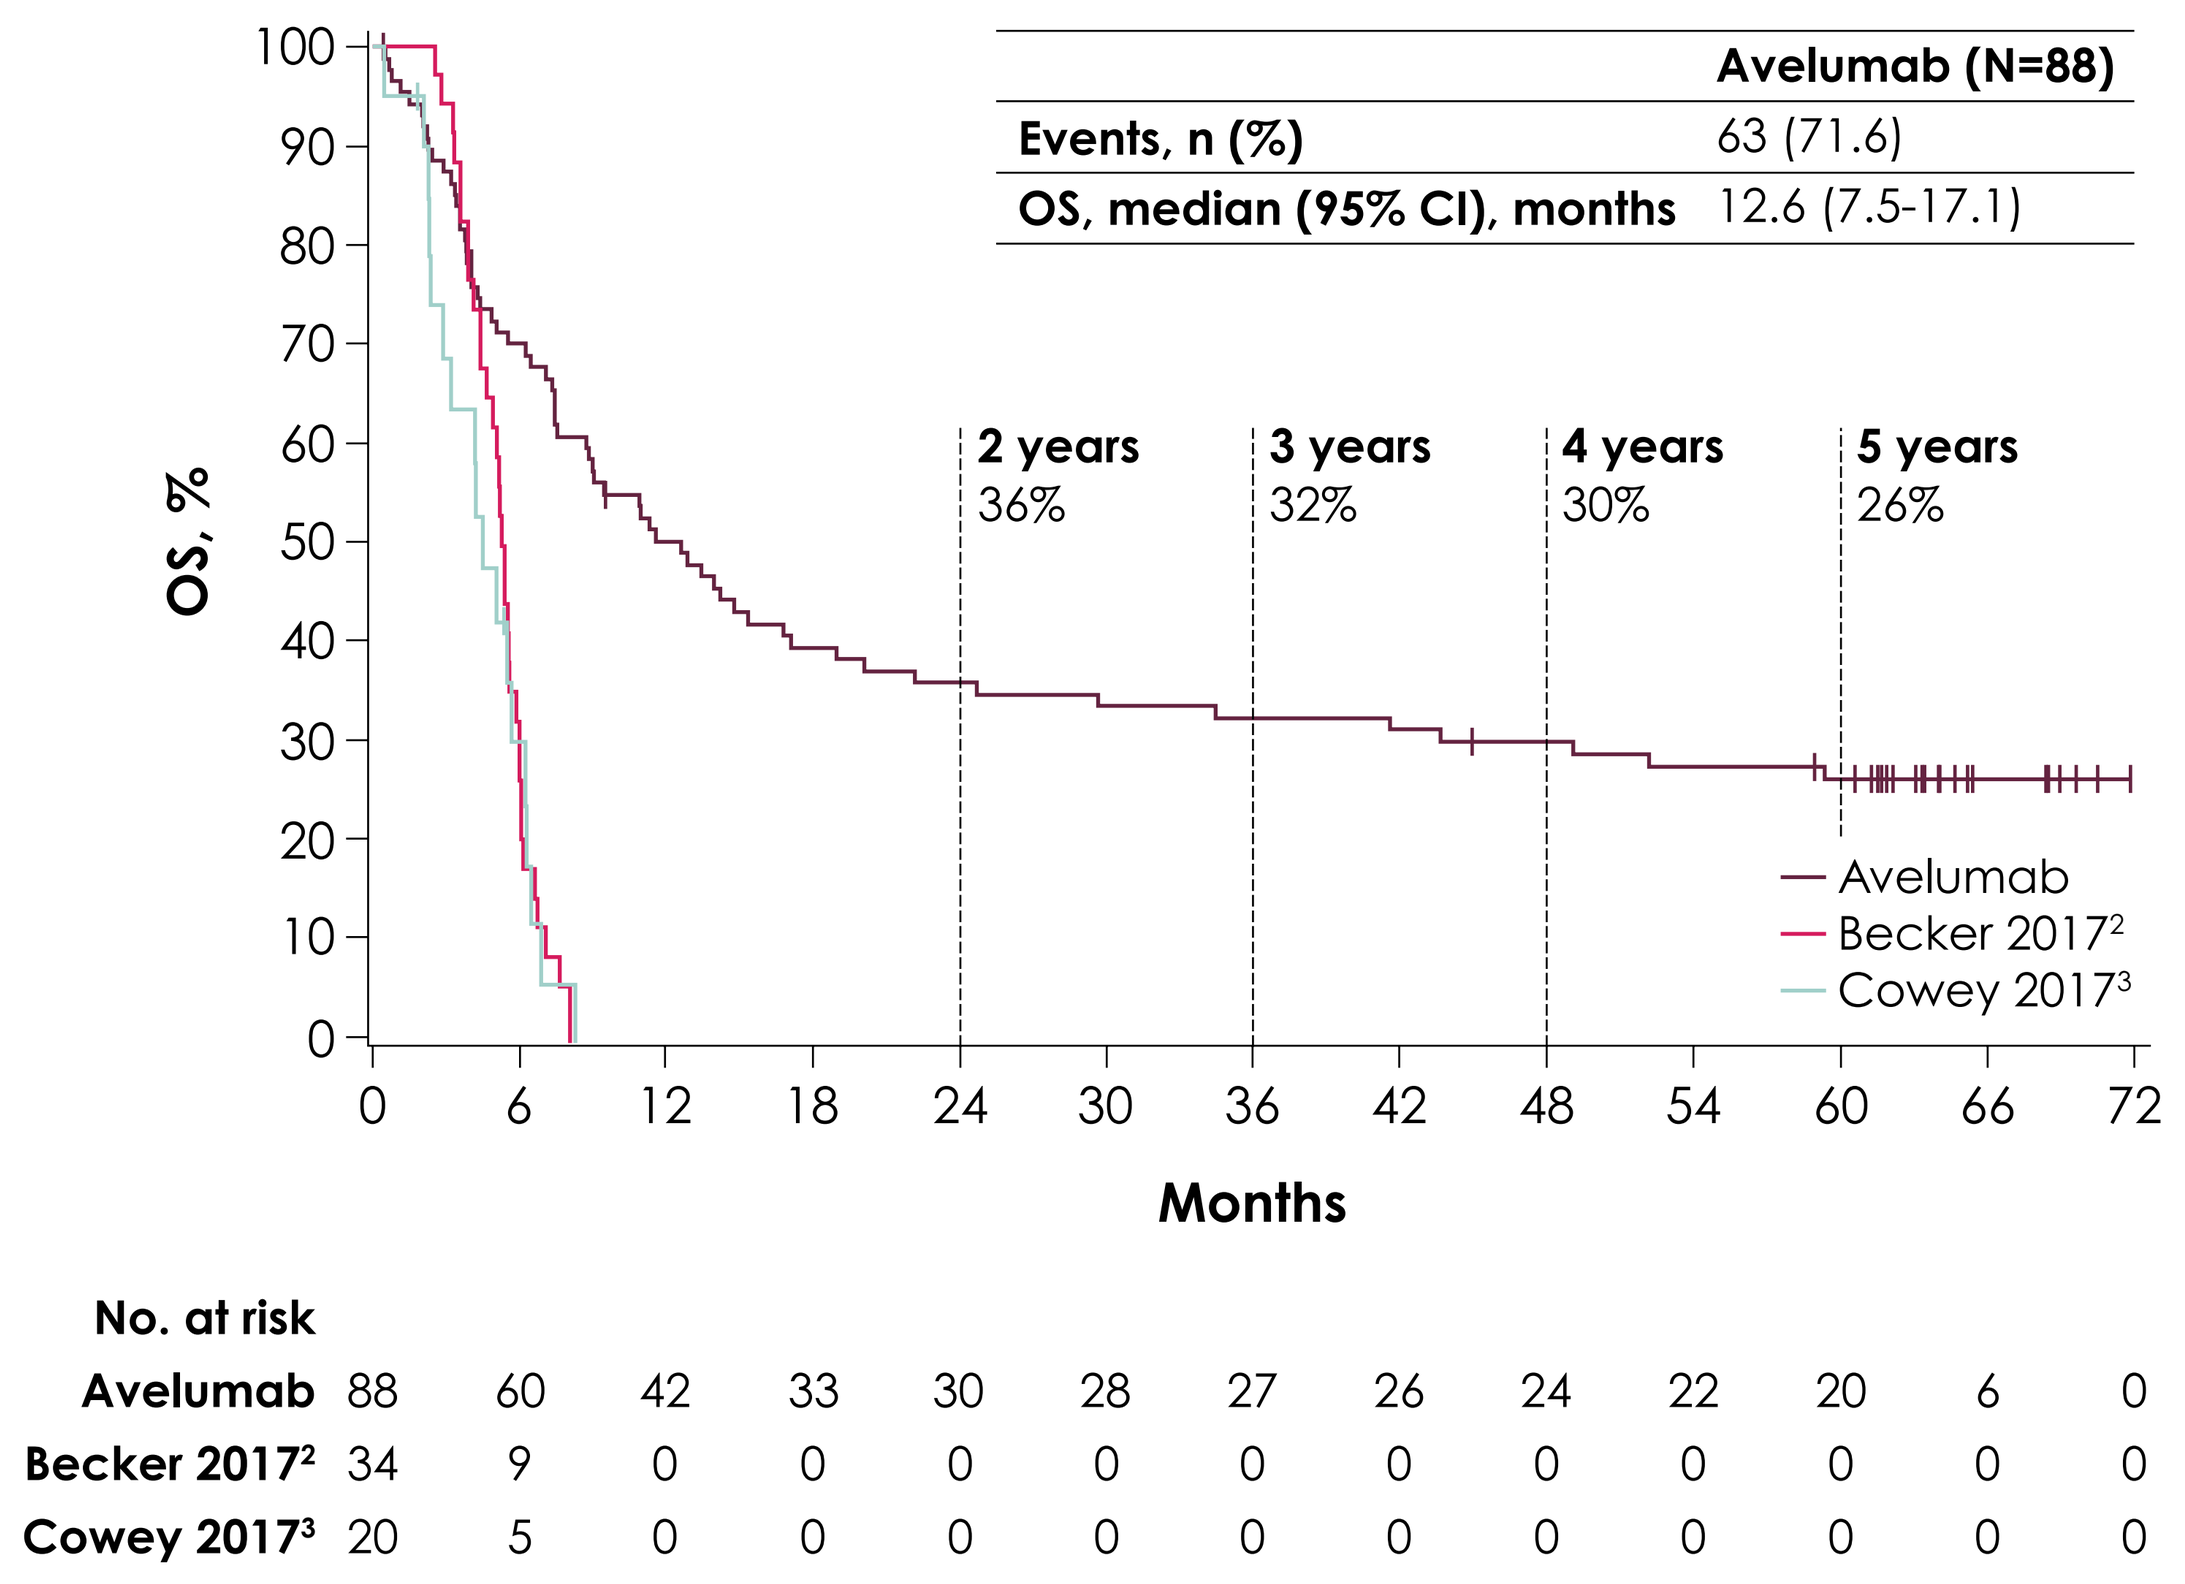

Avelumab (N=88)
Events, n (%) 63 (71.6)
OS, median (95% CI), months 12.6 (7.5-17.1)
OS, %
100 90 80 70 60 50 40 30 20 10 0
2 years 36%
3 years 32%
4 years 30%
5 years 26%
Avelumab
Becker 2017[2]
Cowey 2017[3]
0 6 12 18 24 30 36 42 48 54 60 66 72
Months
No. at risk
Avelumab 88 60 42 33 30 28 27 26 24 22 20 6 0
Becker 2017[2] 34 9 0 0 0 0 0 0 0 0 0 0 0
Cowey 2017[3] 20 5 0 0 0 0 0 0 0 0 0 0 0

OS, overall survival.
This figure is for illustrative purposes only and is not a head-to-head comparison.

Figure 2. Overall survival in subgroups defined by PD-L1 status

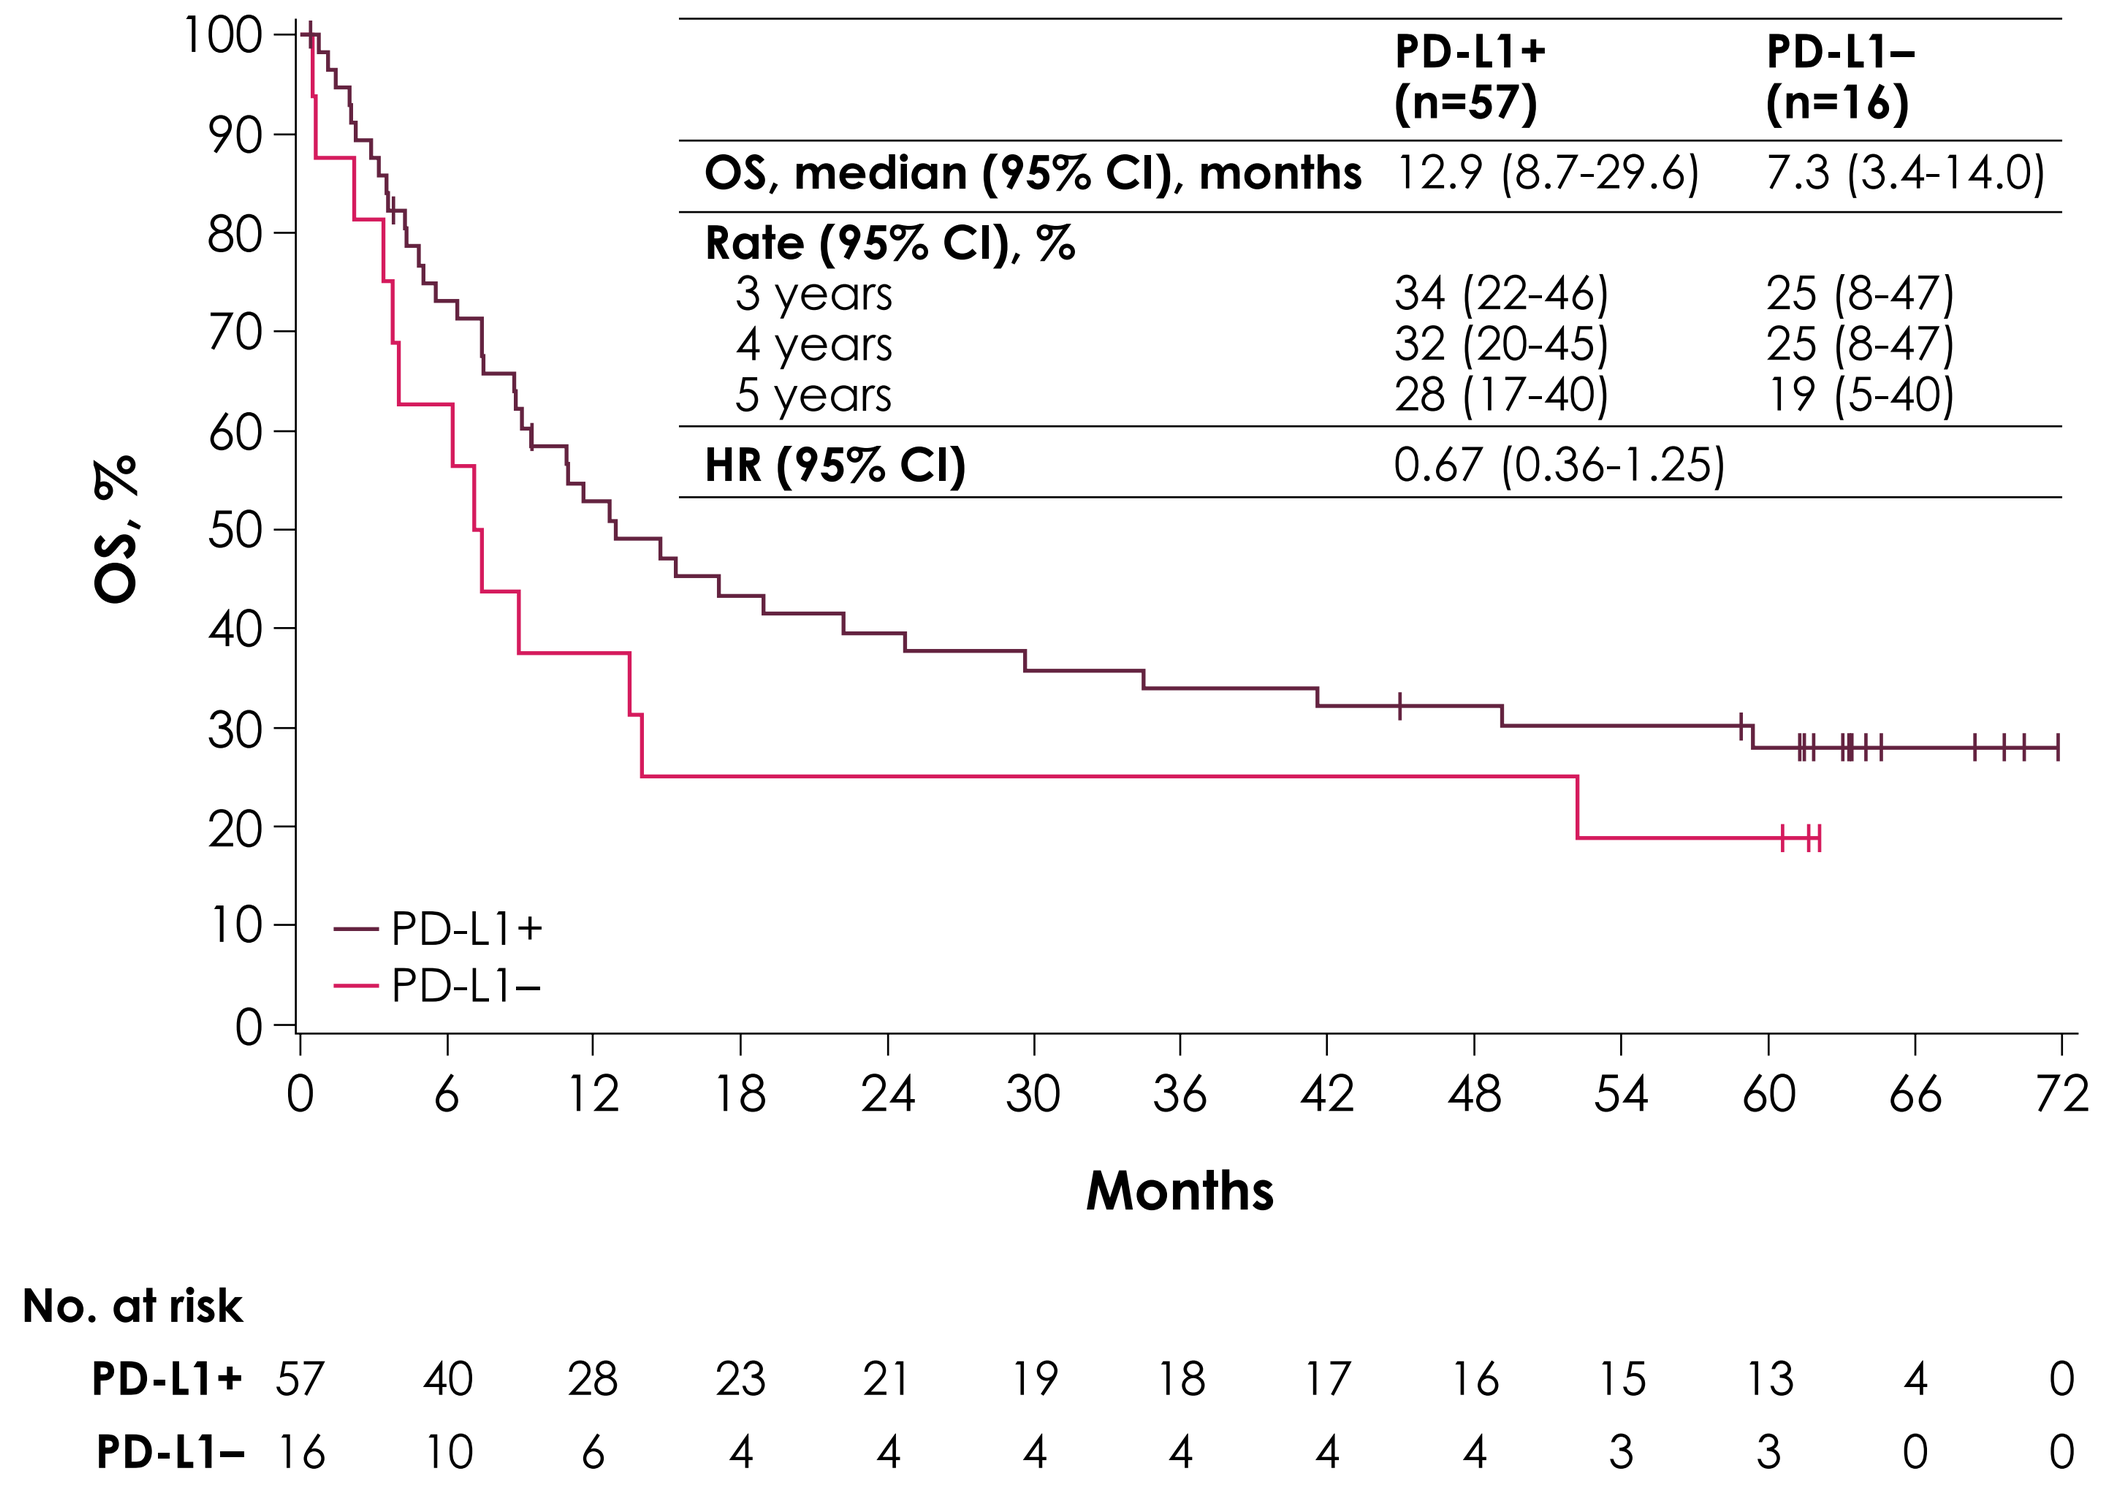

PD-L1+ (n=57)
PD-L1– (n=16)
OS, median (95% CI), months 12.9 (8.7-29.6) 7.3 (3.4-14.0)
Rate (95% CI), %
3 years 34 (22-46) 25 (8-47)
4 years 32 (20-45) 25 (8-47)
5 years 28 (17-40) 19 (5-40)
HR (95% CI) 0.67 (0.36-1.25)
OS, %
100 90 80 70 60 50 40 30 20 10 0
PD-L1+
PD-L1–
0 6 12 18 24 30 36 42 48 54 60 66 72
Months
No. at risk
PD-L1+ 57 40 28 23 21 19 18 17 16 15 13 4 0
PD-L1– 16 10 6 4 4 4 4 4 4 3 3 0 0

HR, hazard ratio; **OS**, overall survival.

Table 4. Subsequent anticancer drug treatment

	N=88
Received subsequent therapy, n (%)	26 (29.5)
Avelumab	4 (4.5)
Carboplatin + etoposide	4 (4.5)
Pembrolizumab	4 (4.5)
Everolimus	3 (3.4)
Nivolumab	3 (3.4)
Pazopanib	3 (3.4)
Capecitabine	2 (2.3)
Cyclophosphamide + doxorubicin + vincristine	2 (2.3)
Paclitaxel	2 (2.3)
Pegylated liposomal doxorubicin hydrochloride	2 (2.3)
Temozolomide	2 (2.3)
Topotecan	2 (2.3)
Amrubicin	1 (1.1)
Carboplatin	1 (1.1)
Carboplatin + paclitaxel	1 (1.1)
Cisplatin	1 (1.1)
Combinations of antineoplastic agents	1 (1.1)
Cyclophosphamide	1 (1.1)
Ipilimumab + nivolumab	1 (1.1)
Octreotide	1 (1.1)
Sunitinib	1 (1.1)
Somatostatin	1 (1.1)
Other therapeutic product	1 (1.1)

REFERENCES **1.** D'Angelo SP, et al. J Immunother Cancer. 2020;8:e000674. **2.** Becker JC, et al. Oncotarget. 2017;8:79731–41. **3.** Cowey CL, et al. Future Oncol. 2017;13:1699–710. **4.** Harms KL, et al. Ann Surg Oncol. 2016;23:3564-71. **5.** Iyer JG, et al. Cancer Med. 2016;5(9):2294-2301. **6.** NCCN Clinical Practice Guidelines in Oncology. Merkel Cell Carcinoma. v1.2021. **7.** Kaufman HL, et al. Lancet Oncol. 2016;17(10):1374-85.

DISCLOSURES **P.T. Nghiem** has served in a consulting or advisory role for 4SC, EMD Serono, Merck & Co., Pfizer, and Sanofi/Regeneron; received travel and accommodations expenses from Merck & Co. and Sanofi/Regeneron; has a patent pending for high-affinity T-cell receptors that target the Merkel polyomavirus; and has received research funding from Bristol Myers Squibb and EMD Serono. **S. Bhatia** has served in a consulting or advisory role for Bristol Myers Squibb, EMD Serono, Exicure, Genentech/Roche, and Sanofi/Regeneron (self and institution); has received travel and accommodations expenses from NantWorks and Sanofi/Regeneron; has received honoraria from Bristol Myers Squibb, EMD Serono, Genentech/Roche, and Sanofi/Regeneron; and has received institutional research funding from Bristol Myers Squibb, EMD Serono, Exicure, Immune Design, Merck & Co., NantWorks, Nektar, Novartis,OncoSec, and Incyte. **A.S. Brohl** has served in a consulting or advisory role for Bayer, Decihpera, and EMD Serono; and their immediate family member has provided expert testimony for GlaxoSmithKline. **O. Hamid** has served in a consulting or advisory role for Aduro Biotech, Akeso Biopharma, Amgen, BeiGene, BioAtla, Bristol Myers Squibb, Merck & Co., Genentech, GlaxoSmithKline, Idera, Immunocore, Incyte, Janssen, NextCure, Novartis, Pfizer, Regeneron, Roche, Sanofi, Seattle Genetics, Tempus, and Zelluna; has provided speaker services for Bristol Myers Squibb, Novartis, Sanofi/Regeneron, and Pfizer; has received honoraria from Bristol Myers Squibb, Pfizer, Novartis, and Sanofi/Regeneron; and has received institutional research funding from Aduro Biotech, Akeso Biopharma, Amgen, Arcus Biosciences, BioAtla, Bristol Myers Squibb, CytomX Therapeutics, EMD Serono, Exelixis, Genentech, GlaxoSmithKline, Idera, Immunocore, Incyte, Iovance Biotherapeutics, Merck & Co., Moderna Therapeutics, NextCure, Novartis, Pfizer, Regeneron, Roche, Sanofi, Seattle Genetics, Torque, and Zelluna. **J.M. Mehnert** has served in a consulting or advisory role for Bristol Myers Squibb, Merck & Co., Sanofi/Regeneron, Seattle Genetics; has received travel and accommodations expenses from Array BioPharma, Bristol Myers Squibb, EMD Serono, and Merck & Co.; has stock and other ownership interests with Pfizer; has received honoraria from EMD Serono and Pfizer; and has received institutional research funding from Amgen, AstraZeneca, Bristol Myers Squibb, Incyte, Macrogenics, Merck & Co., Novartis, and Regeneron. **P. Terheyden** has served in a consulting or advisory role for Bristol Myers Squibb, Merck KGaA, Novartis, Pierre Fabre, Roche, and Sanofi; has received travel and accommodations expenses from Bristol Myers Squibb and Pierre Fabre; and has received honoraria from Bristol Myers Squibb, CureVac, EMD Serono, Novartis, Merck & Co., and Roche. **C. Lebbé** has served in a consulting or advisory role for Amgen, Bristol Myers Squibb, EMD Serono, Merck & Co., Novartis, Pierre Fabre, Roche, and Sanofi; has provided speaker services for Amgen, Bristol Myers Squibb, Novartis, Merck & Co., and Roche; has received travel and accommodations expenses from Bristol Myers Squibb, Merck & Co., Novartis, Pierre Fabre, and Sanofi; reports other relationships with Avantis Medical Systems; has received honoraria from Amgen, Bristol Myers Squibb, Incyte, Novartis, Merck & Co., Pfizer, Pierre Fabre, and Roche; and has received institutional research funding from Bristol Myers Squibb and Roche. **K.D. Lewis** has served in a consulting or advisory role for Array BioPharma, EMD Serono, Iovance Biotherapeutics, Regeneron, Roche, and Sanofi; has received travel and accommodations expenses from Alkermes, EMD Serono, Neon Therapeutics, Regeneron, and Roche/Genentech; has received honoraria from Array BioPharma and Iovance Biotherapeutics; has received institutional research funding from Alkermes, Array BioPharma, Bristol Myers Squibb, EMD Serono, Incyte, Iovance Biotherapeutics, Kartos Therapeutics, Nektar, Neon Therapeutics, OncoSec, Regeneron, Roche/Genentech, and Ultimovacs; and has uncompensated relationships with Regeneron and Roche/Genentech. **M. Milella** has served in a consulting or advisory role for Pfizer and Novartis and has received travel and accommodations expenses from Novartis. **H. Xiong** is employed by EMD Serono Research & Development Institute, Inc., Billerica, MA; an affiliate of Merck KGaA, Darmstadt, Germany. **G. Guezel** is employed by Merck KGaA, Darmstadt, Germany. **S.P. D'Angelo** has served in a consulting or advisory role for Adaptimmune, Amgen, EMD Serono, GlaxoSmithKline, Immune Design, Immunocore, Incyte, and Nektar; has received travel and accommodations expenses from Adaptimmune, EMD Serono, and Nektar; and has received institutional research funding from Amgen, Bristol Myers Squibb, Deciphera, EMD Serono, Incyte, Merck & Co., and Nektar. **K.C. Shih**, **I. Brownell**, and **G.P. Linette** have no conflicts to disclose. **ACKNOWLEDGMENTS** The authors would like to thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany. This trial was sponsored by Merck KGaA, Darmstadt, Germany and is part of an alliance between Merck KGaA and Pfizer. Medical writing support was provided by Felicia Barklund of ClinicalThinking and was funded by Merck KGaA and Pfizer.

Abstract No. 9517. Presented at 2021 ASCO Annual Meeting, June 4-8, 2021; Virtual.