Real-world response rates and outcomes of patients treated with first-line platinum-based chemotherapy in advanced urothelial cancer

H. H. Moon,[1] J. B. Aragon-Ching,[2] A. Thompson,[3] A. Abraham,[4] A. Vlahiotis,[3] C. Ike,[5] D. Benjumea,[4] A. Shao,[4] H. Sun,[4] M. Kearney,[6] N. Gharibian,[3] S. Hanson,[3] B. Li,[3] M. Kirker,[3] P. Grivas[7]

[1]Kaiser Permanente, Riverside, CA, USA; [2]Inova Schar Cancer Institute, Fairfax, VA, USA; [3]Pfizer, New York, NY, USA; [4]Genesis Research, LLC, Hoboken, NJ, USA; [5]EMD Serono, Rockland, MA, USA; [6]The healthcare business of Merck KGaA, Darmstadt, Germany; [7]University of Washington; Fred Hutchinson Cancer Center, Seattle, WA, USA

SCOPE

- **This study aimed to assess current real-world response (rwR) rates and real-world outcomes in patients with advanced urothelial carcinoma (aUC) treated with first-line (1L) platinum-based chemotherapy (PBC) in the United States (US)**
- **Patient eligibility to receive avelumab 1L maintenance (1LM) after 1L PBC and early utilization of avelumab 1LM since US Food and Drug Administration (FDA) approval on June 30, 2020, were also assessed**

CONCLUSIONS

- **In this real-world study, 80% of patients with aUC treated with 1L PBC had a real-world complete response (rwCR)/partial response (rwPR) or stable disease (rwSD) and may have been eligible to receive avelumab 1LM**
- **rwR rates to 1L PBC appeared slightly higher than historical data and may reflect better general best supportive care, as also shown in recent large phase 3 trials (NCT02853305, NCT02516241, NCT02807636)[1-3]**
- **Early uptake of avelumab 1LM was observed only in 29% of maintenance-eligible patients; future studies with longer follow-up may demonstrate increased utilization and allow assessment of potential impact on outcomes**
- **Follow-up time for patients treated with avelumab 1LM was short, and outcomes were immature at the end of the study period**

BACKGROUND

- Urothelial carcinoma is one of the most common cancers worldwide, with poor survival in patients with advanced/metastatic disease[4-6]
- A standard of care for aUC is 1L PBC with avelumab 1LM in eligible patients based on the phase 3 JAVELIN Bladder 100 trial (NCT02603432), which showed that avelumab 1LM significantly prolonged overall survival (OS) and progression-free survival (PFS) in patients without progression after 1L PBC[7]
- Contemporary and robust real-world data are limited in patients treated with 1L PBC; however, because response status to PBC determines eligibility for avelumab 1LM, understanding rwR rates is crucial
- This study aimed to assess current rwR rates and outcomes in patients treated with 1L PBC, and to understand patient eligibility for avelumab 1LM and early utilization following the FDA approval on June 30, 2020[8]

METHODS

- A noninterventional, retrospective cohort study of patients with aUC in the US was conducted using Flatiron Health's electronic health record database; the dataset comprises demographically and geographically diverse data from approximately 280 cancer clinics[9]
- The study period was January 1, 2017, to December 31, 2021, and the identification period was January 1, 2017, to September 30, 2021, to ensure ≥3 months of follow-up unless a patient died
 - The index date was the start date for 1L treatment in the identification period (**Figure 1**)
- Patients with aUC were included if they had received 1L PBC during the identification period, did not have a progression event, and had an observed rwR (rwCR/rwPR or rwSD) during 1L PBC
- Patients were classified as having received avelumab 1LM if they had received 1L PBC, had initiated avelumab within 180 days of 1L PBC discontinuation, and had no documented progression before initiating avelumab
- Patient characteristics and outcomes were described by response type (rwCR/rwPR vs rwSD)
- rwPFS and rwOS from the time of 1L PBC initiation were estimated using the Kaplan-Meier method
- rwPFS analyses to demonstrate disease progression after completing 1L PBC, as a progression-free cohort, was used to evaluate the avelumab 1LM-eligible population

Figure 1. Study design

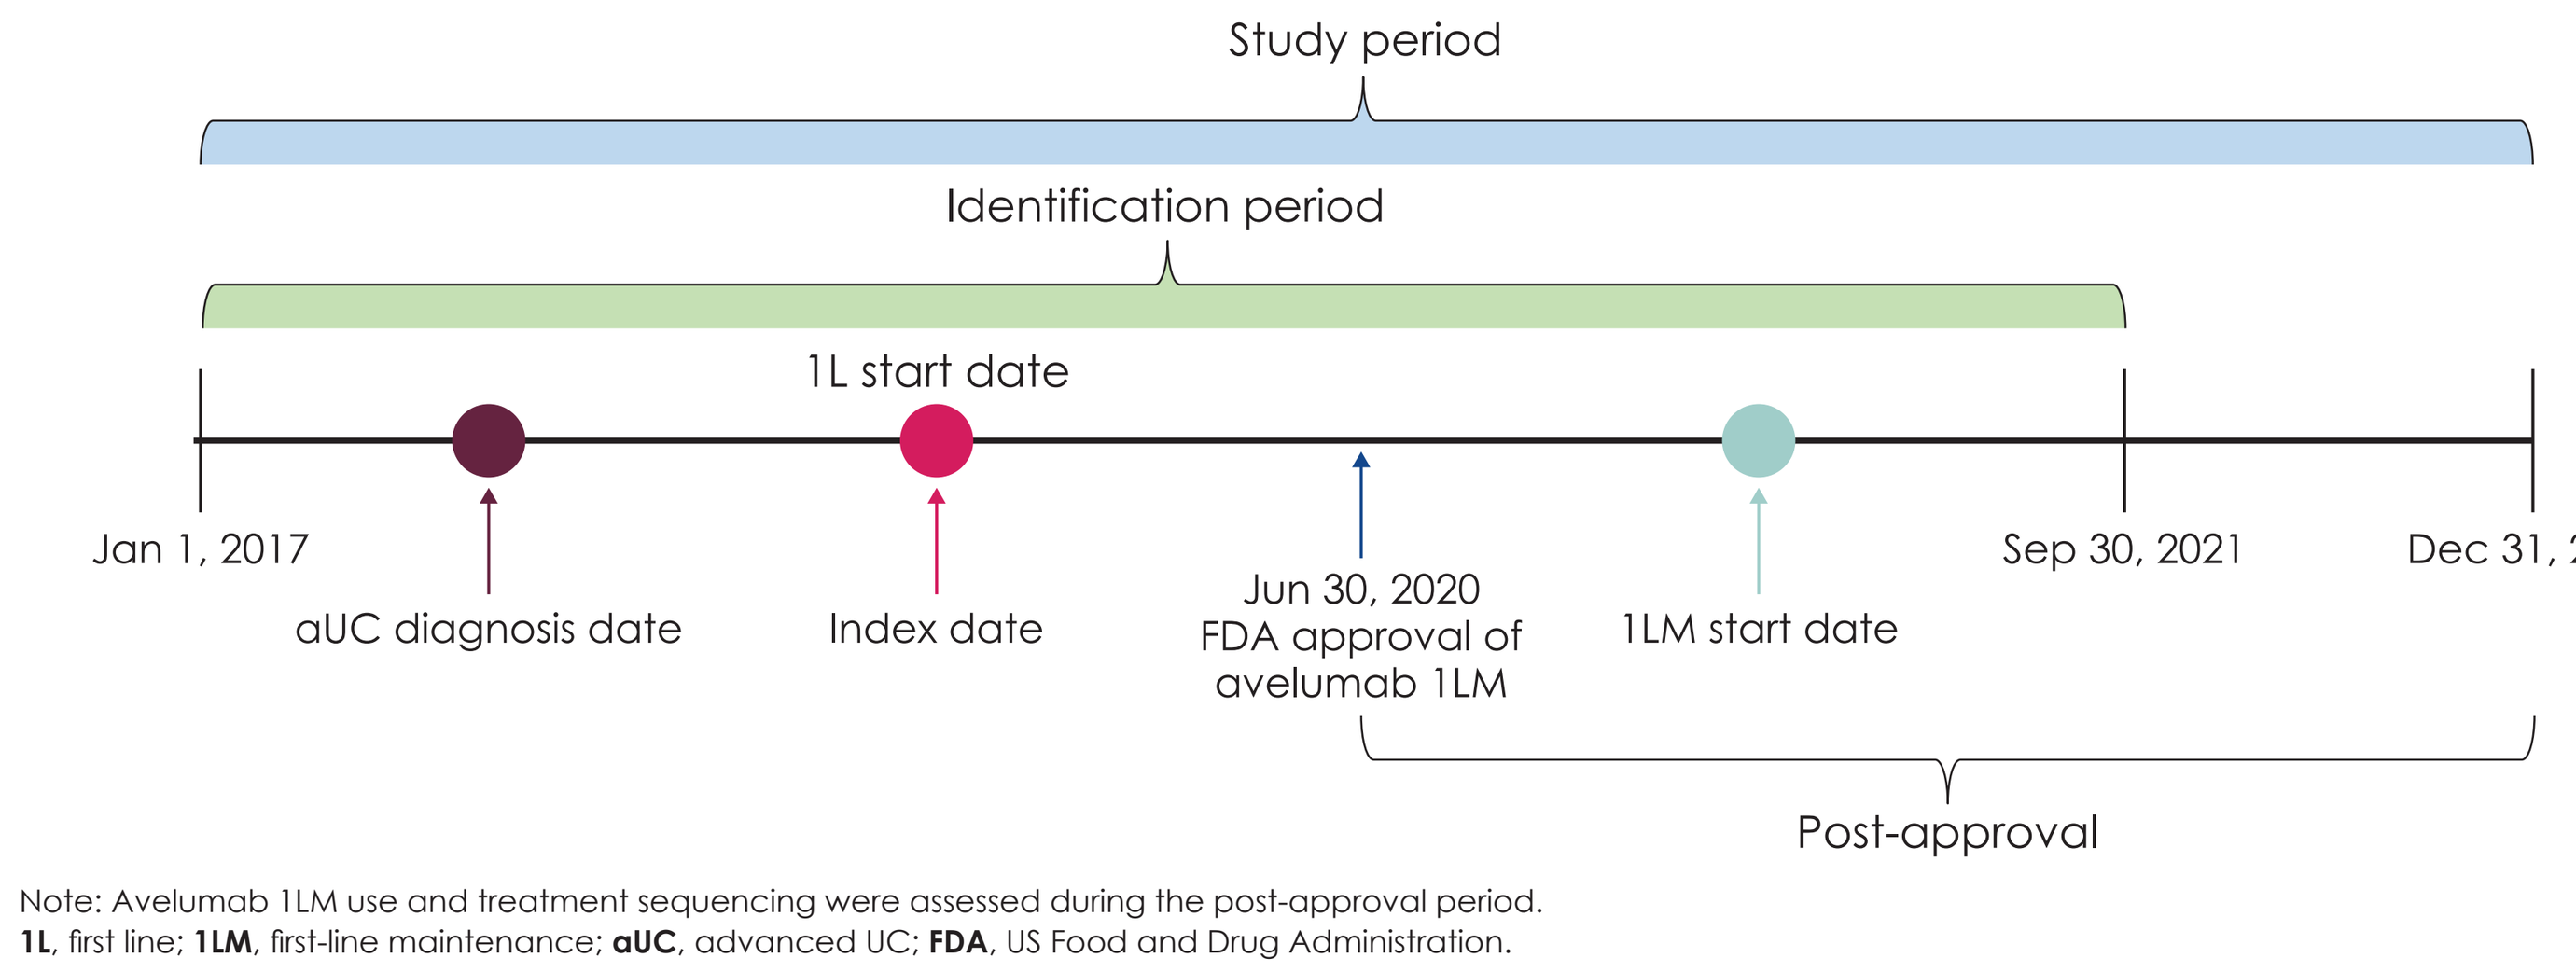

Study period
Identification period
1L start date
Jan 1, 2017
aUC diagnosis date
Index date
Jun 30, 2020
FDA approval of avelumab 1LM
1LM start date
Sep 30, 2021
Dec 31, 2021
Post-approval

Note: Avelumab 1LM use and treatment sequencing were assessed during the post-approval period.
1L, first line; **1LM**, first-line maintenance; **aUC**, advanced UC; **FDA**, US Food and Drug Administration.

RESULTS

- Of 1,703 patients with aUC treated with 1L PBC, 1,245 (73%) had rwR data available during 1L PBC; of these, 80% (n=998) may have been eligible to receive avelumab 1LM (rwCR/rwPR, 60%; rwSD, 20%)
- Patient demographics and clinical characteristics were similar between patients with rwCR/rwPR and rwSD (**Table 1** and **Table 2**)
- Patients with rwCR/rwPR had a longer median rwPFS after 1L PBC vs patients with rwSD (9.1 months [95% CI, 8.5-9.9] vs 6.5 months [95% CI, 6.0-8.1], respectively; **Figure 2**)
- Median rwOS was longer in patients with rwCR/rwPR vs patients with rwSD (23.2 months [95% CI, 20.8-26.6] vs 18.7 months [95% CI, 14.9-24.4], respectively; **Figure 2**)
- During the post-approval period (**Figure 1**), 339 of 998 patients who may have been eligible for avelumab 1LM discontinued 1L PBC (**Table 3**); of these patients, 29% (n=97) received avelumab 1LM (**Table 3**)
- 40 patients who received avelumab 1LM received subsequent second-line (2L) treatment, of which enfortumab vedotin was most common (60%; **Figure 3**)
- Among 57 patients who did not receive/report 2L treatment, 70% received avelumab 1LM within 4 weeks of the end of the study period; these patients may have still been receiving treatment, thus subsequent therapies may not have been available
- The median follow-up time for patients treated with avelumab 1LM was 7.5 months from initiation of 1LM

Figure 2. Kaplan-Meier estimates of rwPFS and rwOS

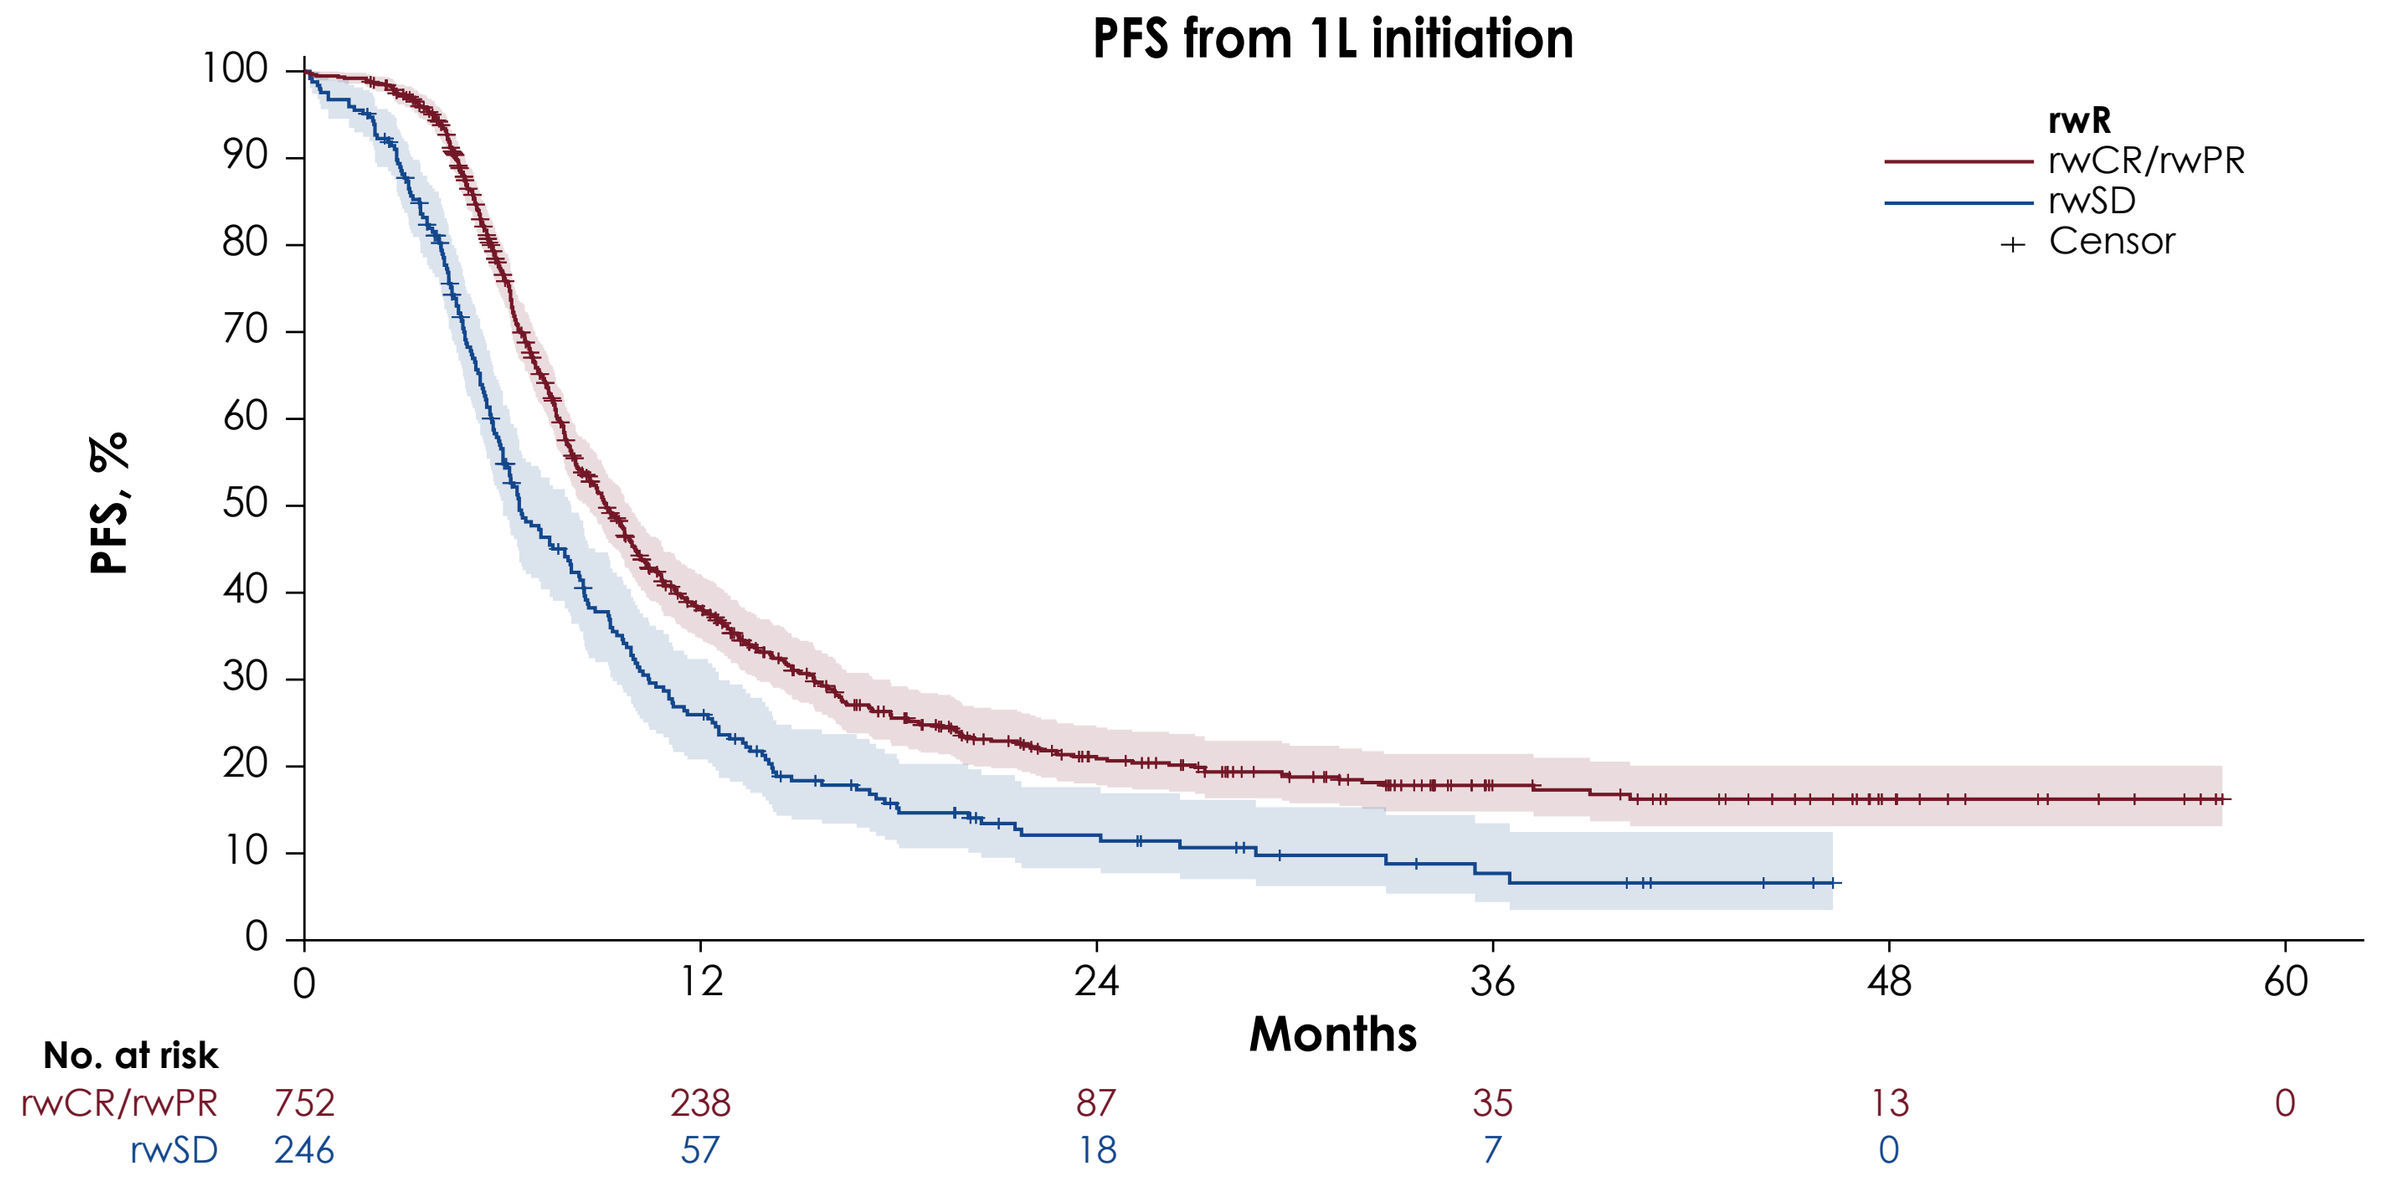

PFS from 1L initiation
rwR
rwCR/rwPR
rwSD
Censor
PFS, %
Months
No. at risk
rwCR/rwPR 752 238 87 35 13 0
rwSD 246 57 18 7 0

rwR	Events/patients	Median PFS (95% CI), months
rwCR/rwPR	535/752	9.1 (8.5-9.9)
rwSD	204/246	6.5 (6.0-8.1)

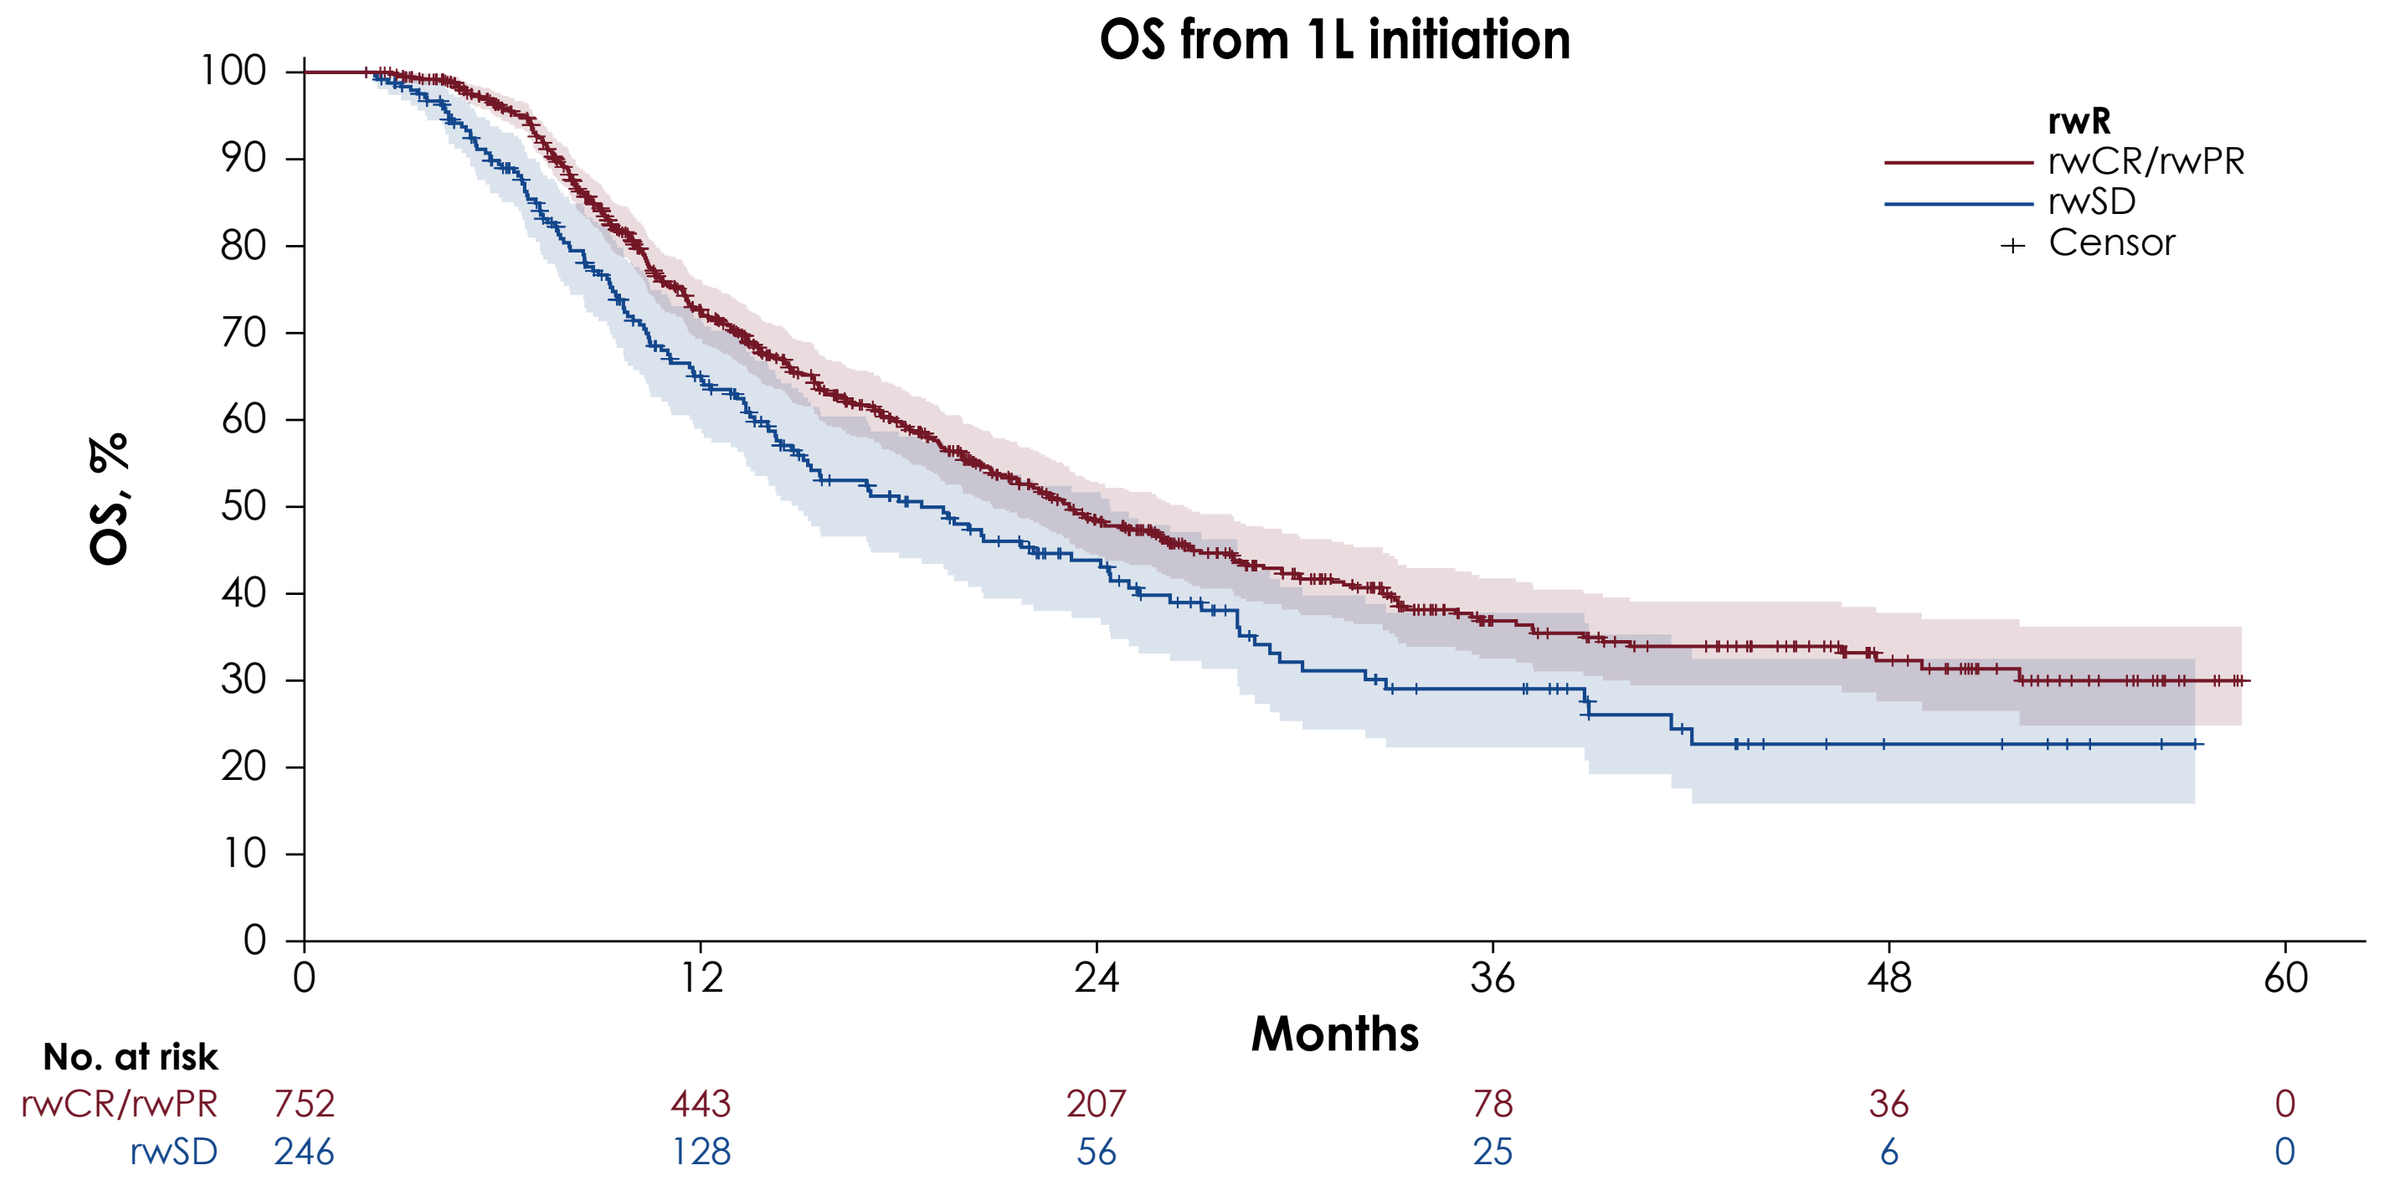

OS from 1L initiation
rwR
rwCR/rwPR
rwSD
Censor
OS, %
Months
No. at risk
rwCR/rwPR 752 443 207 78 36 0
rwSD 246 128 56 25 6 0

rwR	Events/patients	Median OS (95% CI), months
rwCR/rwPR	356/752	23.2 (20.8-26.6)
rwSD	133/246	18.7 (14.9-24.4)

CR, complete response; **OS**, overall survival; **PFS**, progression-free survival; **PR**, partial response; **rw**, real world; **rwR**, real-world response; **SD**, stable disease.

Table 1. Patient demographics at 1L PBC initiation

Characteristic	rwCR/rwPR	rwSD
Total, n (%)	752 (100)	246 (100)
Age at aUC diagnosis, years		
Mean (SD)	69.6 (9.0)	69.6 (10.1)
Sex, n (%)		
Female	209 (27.8)	54 (22.0)
Male	543 (72.2)	192 (78.0)
Race, n (%)		
Asian	12 (1.6)	2 (0.8)
Black or African American	40 (5.3)	11 (4.5)
Hispanic or Latino	1 (0.1)	1 (0.4)
White	493 (65.6)	169 (68.7)
Other race	127 (16.9)	41 (16.7)
Unknown	79 (10.5)	22 (8.9)
Region of residence, n (%)		
Northeast	88 (11.7)	39 (15.9)
Midwest	107 (14.2)	30 (12.2)
South	350 (46.5)	114 (46.3)
West	115 (15.3)	33 (13.4)
Other	3 (0.4)	3 (1.2)
Unknown	89 (11.8)	27 (11.0)

aUC, advanced urothelial carcinoma; **rwCR**, real-world complete response; **rwPR**, real-world partial response; **rwSD**; real-world stable disease.

Figure 3. Treatment sequence from avelumab 1LM to 2L in the post-FDA approval period

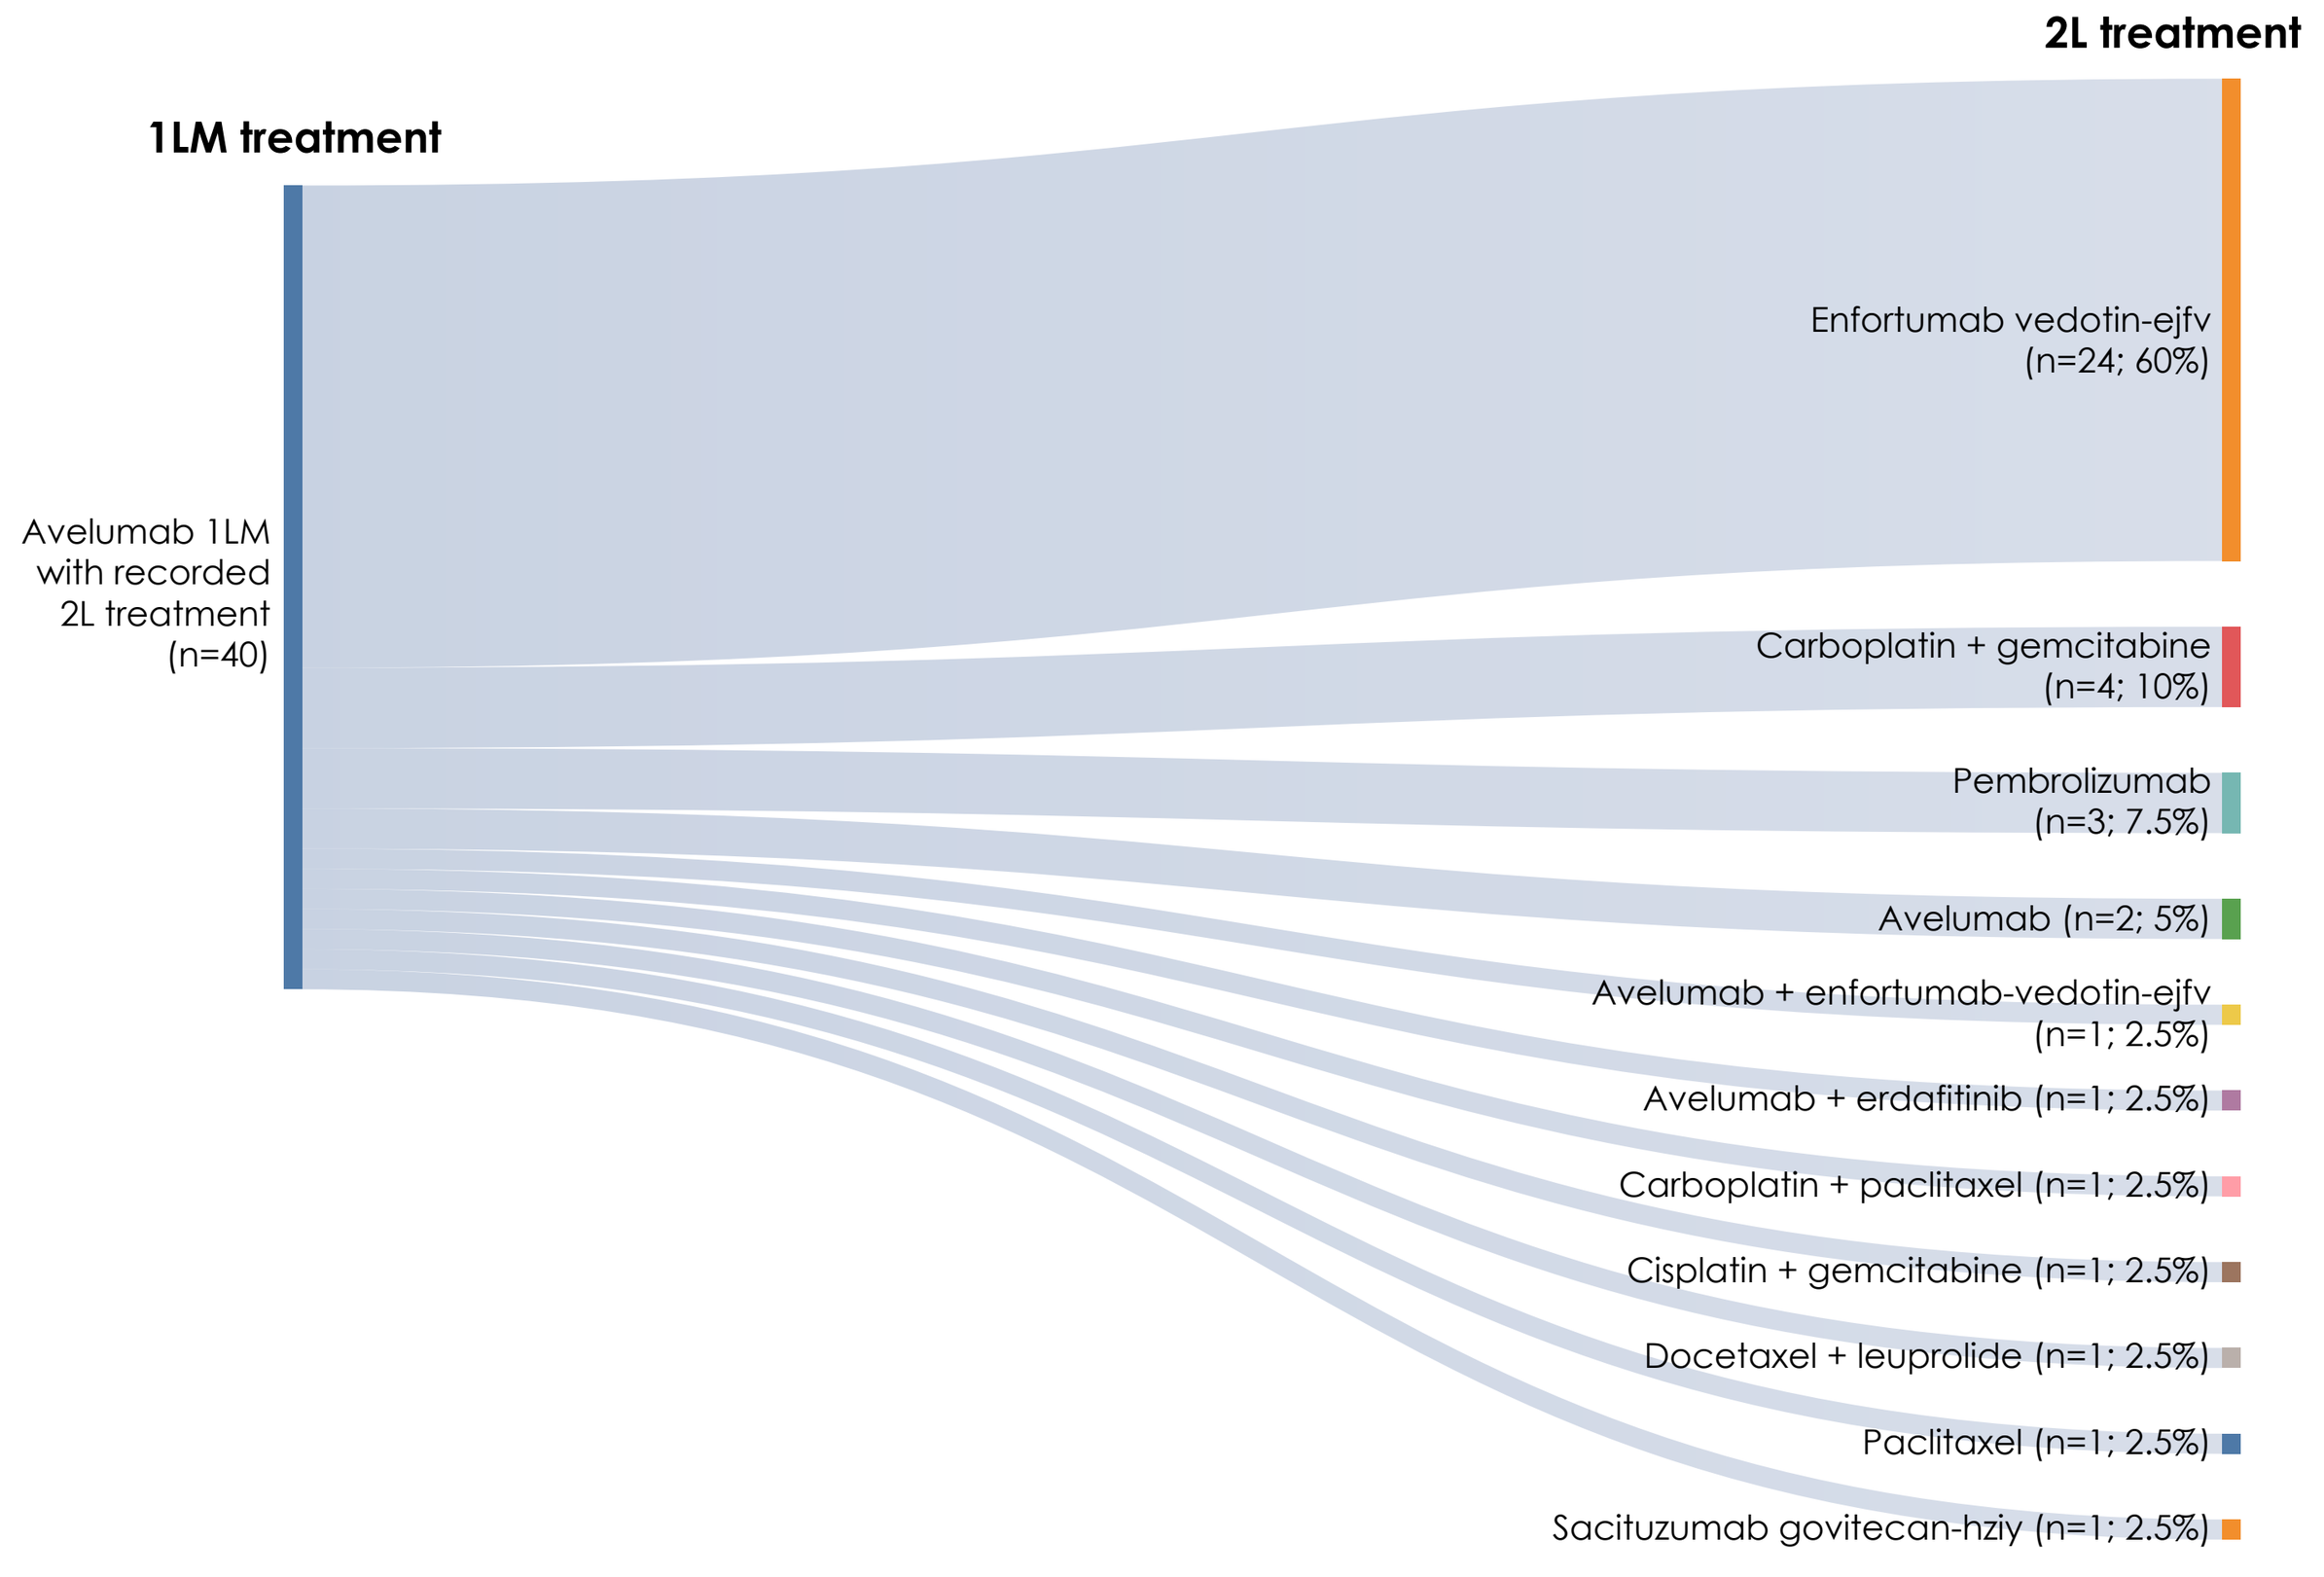

1LM treatment
2L treatment
Avelumab 1LM with recorded 2L treatment (n=40)
Enfortumab vedotin-ejfv (n=24; 60%)
Carboplatin + gemcitabine (n=4; 10%)
Pembrolizumab (n=3; 7.5%)
Avelumab (n=2; 5%)
Avelumab + enfortumab-vedotin-ejfv (n=1; 2.5%)
Avelumab + erdafitinib (n=1; 2.5%)
Carboplatin + paclitaxel (n=1; 2.5%)
Cisplatin + gemcitabine (n=1; 2.5%)
Docetaxel + leuprolide (n=1; 2.5%)
Paclitaxel (n=1; 2.5%)
Sacituzumab govitecan-hziy (n=1; 2.5%)

1LM, first-line maintenance; **2L**, second line.

Table 2. Clinical characteristics at 1L PBC initiation

Characteristic	rwCR/rwPR	rwSD
Total, n (%)	752 (100)	246 (100)
Site of disease, n (%)		
Bladder	553 (73.5)	183 (74.4)
Renal pelvis	112 (14.9)	38 (15.4)
Ureter	84 (11.2)	21 (8.5)
Urethra	3 (0.4)	4 (1.6)
Disease grade, n (%)		
High grade (G2/G3/G4)	652 (86.7)	210 (85.4)
Low grade (G1)	35 (4.7)	12 (4.9)
Unknown/not documented	65 (8.6)	24 (9.8)
Stage at initial UC diagnosis, n (%)		
Stage 0	4 (0.5)	1 (0.4)
Stage I	17 (2.3)	4 (1.6)
Stage II	38 (5.1)	10 (4.1)
Stage III	82 (10.9)	27 (11.0)
Stage IV	340 (45.2)	94 (38.2)
Unknown/not documented	271 (36.0)	110 (44.7)
ECOG PS at aUC diagnosis date, n (%)		
0	264 (35.1)	82 (33.3)
1	254 (33.8)	85 (34.6)
2+	63 (8.4)	25 (10.2)
Unknown/not documented	171 (22.7)	54 (22.0)
PD-L1 at aUC diagnosis date, n (%)		
Negative	89 (11.8)	35 (14.2)
Positive	38 (5.1)	12 (4.9)
Unknown/not documented	625 (83.1)	199 (80.9)
GFR level at aUC diagnosis date, mL/min/1.73m²		
Mean (SD)	67.8 (23.8)	65.5 (26.9)
Treatment group, n (%)		
Carboplatin-based	311 (41.4)	110 (44.7)
Cisplatin-based	441 (58.6)	135 (54.9)
Oxaliplatin-based	0	1 (0.4%)

aUC, advanced urothelial carcinoma; **ECOG PS**, Eastern Cooperative Oncology Group performance status; **G**, grade; **GFR**, glomerular filtration rate; **rwCR**, real-world complete response; **rwPR**, real-world partial response; **rwSD**; real-world stable disease; **UC**, urothelial carcinoma.

Table 3. Summary of avelumab 1LM and 2L treatment in the post-FDA approval period

Treatment criteria	n (% of previous step)
Total study population	1,245 (100)
rwCR/PR or rwSD recorded during 1L PBC	998 (80.1)
Discontinued 1L PBC after avelumab 1LM approval (June 30, 2020)	339 (34.0)
Initiated avelumab 1LM	97 (28.6)
2L treatment received after avelumab 1LM	
Received	40 (41.2)
Not received/not reported*	57 (58.8)
2L treatment not received/reported but received avelumab within 4 weeks of study period end	40 (70.2)

1L, first line; **1LM**, first-line maintenance; **2L**, second line; **PBC**, platinum-based chemotherapy; **rwCR/PD**, real-world complete or partial response; **rwSD**; real-world stable disease.
*Patients who initiated avelumab 1LM may still be receiving treatment, but data were not available.

Limitations

- Immortal time and selection biases since patients were required to have recorded response or SD, thus resulting in a potential overestimation of rwOS
- This study could not estimate true rwPFS, given that a 1L progression-free cohort was used to evaluate the avelumab 1LM-eligible population; only PFS after completion of 1L PBC was estimated
- Retrospective study, lack of randomization, lack of granularity in specific data points (eg, site of metastasis, laboratory values), lack of central scan review, residual confounding, and short duration of study period post approval of avelumab 1LM by the FDA

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Helen H. Moon**, helen.h.moon@kp.org

REFERENCES 1. Powles T, et al. Lancet Oncol. 2021;22(7):931-5. **2.** Powles T, et al. Lancet Oncol. 2020;21(12):1574-88. **3.** Galsky MD, et al. Lancet. 2020;395(10236):1547-57. **4.** Antoni S, et al. Eur Urol. 2017;71(1):96-108. **5.** Bray F, et al. CA Cancer J Clin. 2018;68(6):394-424. **6.** World Health Organization. Accessed May 3, 2023. https://gco.iarc.fr/today/home. **7.** NCCN Clinical Practice Guidelines: Bladder Cancer. V2.2023. **8.** US Food and Drug Administration. 2020. Accessed May 3, 2023. https://www.fda.gov/drugs/drug-approvals-and-databases/fda-approves-avelumab-urothelial-carcinoma-maintenance-treatment. **9.** Flatiron. 2022. Accessed May 3, 2023. https://flatiron.com/real-world-evidence/.

DISCLOSURES H. H. Moon reports honoraria from EMD Serono, Pfizer; research funding from Bristol Myers Squibb, Amgen, Prometheus, Genentech, Seagen, Arcus Biosciences, Apollomics, Nektar, Revimmune, HUYA Bioscience International, and AVEO. **J. Aragon-Ching** has served in consulting or advisory roles for Algeta/Bayer, Amgen, AstraZeneca, AstraZeneca/MedImmune, AVEO, Bayer, Dendreon, Exelixis, Immunomedics, Janssen Biotech, Merck & Co., Kenilworth, NJ, Pfizer, Myovant, Sanofi, Seagen, and the healthcare business of Merck KGaA, Darmstadt, Germany; has reported speakers services for Astellas Pharma, Bristol Myers Squibb, Janssen-Ortho, and Seagen/Astellas; has received travel and accommodation expenses from Algeta/Bayer, Astellas Pharma, Bristol Myers Squibb, Dendreon, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received honoraria from Astellas Scientific and Medical Affairs Inc, Bristol Myers Squibb, Pfizer, and the healthcare business of Merck KGaA, Darmstadt, Germany. **A. Thompson** reports employment at Pfizer. **A. Abraham** reports employment at Genesis Research. **A. Vlahiotus** reports employment at Pfizer. **C. Ike** reports employment at EMD Serono and stocks and other ownership interests with the healthcare business of Merck, KGaA, Darmstadt, Germany. **D. Benjumea** reports employment at Genesis Research. **A. Shao** reports employment at Genesis Research. **H. Sun** reports employment at Genesis Research. **M. Kearney** reports employment at the healthcare business of Merck KGaA, Darmstadt, Germany, and stock and other ownership interests with the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, and UCB. **N. Gharibian** reports employment at Pfizer. **S. Hanson** reports employment at Pfizer and stock and other ownership interests with Pfizer. **B. Li** reports employment at Pfizer. **M. Kirker** reports employment at Pfizer. **P. Grivas** has served in consulting or advisory roles for 4D Pharma, Aadi Bioscience, Astellas Pharma, AstraZeneca, BostonGene, Bristol Myers Squibb, CG Oncology Inc., Dyania Health, Exelixis, Fresenius Kabi, G1 Therapeutics, Genentech, Gilead Sciences, Guardant Health, ImmunityBio, Infinity Pharmaceuticals, Janssen, Lucence, Mirati Therapeutics, Merck & Co., Kenilworth, NJ, Pfizer, PureTech, Regeneron, Roche, Seagen, Silverback Therapeutics, Strata Oncology, QED Therapeutics, the healthcare business of Merck KGaA, Darmstadt, Germany, and UroGen Pharma and has received institutional research funding from Bavarian Nordic, Bristol Myers Squibb, Clovis Oncology, Debiopharm Group, G1 Therapeutics, Gilead Sciences, GlaxoSmithKline, Mirati Therapeutics, Merck & Co., Kenilworth, NJ, Pfizer, QED Therapeutics, and the healthcare business of Merck KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS** This study was sponsored by Pfizer as part of an alliance between Pfizer and the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Editorial support was provided by Genesis Research and Clinical Thinking, and was funded by Pfizer and the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 4567. Presented at the 2023 ASCO Annual Meeting; June 2-6, 2023; Chicago, IL.