This is a reprint from the European Society for Medical Oncology Congress 2022 (ESMO 2022), which was originally presented in Paris, France on September 9-13, 2022; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.
There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

Factors associated with receipt of systemic treatment for metastatic urothelial carcinoma (mUC) in England

C. Knott,[1] M. Kearney,[2] S. H. Mahmoudpour,[3] P. Verpillat[3]

[1]Epidemiology, Health Data Insight CIC; NHS Digital, Leeds, UK; [2]Global Evidence & Value Development, Merck Healthcare KGaA, Darmstadt, Germany; [3]Global Epidemiology, Merck Healthcare KGaA, Darmstadt, Germany

SCOPE

- **This cohort study examined patient characteristics associated with receipt or nonreceipt of systemic treatment for mUC in England**

CONCLUSIONS

- **In this national retrospective cohort study, approximately 70% of this mainly geriatric patient population with mUC was untreated, which is a substantial proportion given the availability of effective treatments**
- **Patients who received systemic anticancer treatment were younger, healthier and less socioeconomically disadvantaged**
- **Specific measures are needed to address the possible multifactorial reasons for undertreatment and improve patient management, especially for patients with significant comorbidities and poor outcomes when untreated**
- **Newer therapies such as avelumab first-line (1L) maintenance, which has demonstrated an overall survival (OS) benefit after 1L chemotherapy, as well as novel antibody-drug conjugates, targeted therapies, and immuno-oncology therapies in the second line (2L), offer hope to an underserved patient population. Given the substantial survival benefits and variety of options becoming available, it is important for patients to receive systemic treatments where eligible**

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster

Correspondence: **Craig Knott**, craig.knott@phe.gov.uk

BACKGROUND

- Bladder cancer accounts for approximately 1 in every 30 new cancer diagnoses each year in the UK and is the 10th most common cancer in the UK[1]
- In 2019, 10,557 new patients were diagnosed with bladder cancer in England[2]
- It is a life-threatening condition with a 1-year age-standardized survival rate of 35.7% in patients with stage IV disease in England[3]
- Platinum-containing combination chemotherapy remains the standard 1L treatment for patients with locally advanced or metastatic UC[4,5]
 - Although disease control rates with platinum-containing combination chemotherapy regimens are high (70%), durable responses are uncommon, and most patients will ultimately experience disease progression
- A large proportion of patients with mUC do not receive systemic anticancer treatment

METHODS

Data source

- A retrospective cohort study was conducted using multiple linked data sources available through the National Cancer Registration and Analysis Service (NCRAS) at National Health Service (NHS) Digital (NHSD)
- Systemic treatment data were obtained from the Systemic Anti-Cancer Therapy (SACT) dataset,[6] which covers NHS funded therapies only
- Patient-level linkage was used to extract comprehensive national data on the disease and patient mortality (National Cancer Registration Dataset); inpatient and outpatient treatments, diagnostics, and diagnoses (Hospital Episode Statistics); anticancer radiotherapy (National Radiotherapy Dataset); systemic anticancer treatments (SACT dataset); and mortality (Office for National Statistics)

Study population

- The cohort included adults diagnosed with primary stage IV UC between 2013 and 2019 in the National Cancer Registration Dataset[7] with follow-up until 31 March 2021 (**Figure 1**)
- Inclusion criteria:
 - Resident in England at the date of diagnosis
 - At least 1 incident primary diagnosis of stage IV UC between 2013 and 2019
 - Age ≥18 years at the date of diagnosis
- Exclusion criteria:
 - No recorded disease stage, age, sex, or date of the diagnosis

Statistical analysis

- Patient demographics and tumor characteristics were compared between treated and untreated patients using χ^2 and Wilcoxon-Mann-Whitney tests for categorical and continuous variables, respectively
- Multivariable logistic regression was performed to identify factors associated with receipt of systemic treatment
- The Kaplan-Meier method was used to estimate median OS from diagnosis and, within the treated patient cohort, from the initiation of systemic treatment to death, censoring at the time of loss to follow-up or the end of the study period (March 2021)

Figure 1. Patient attrition

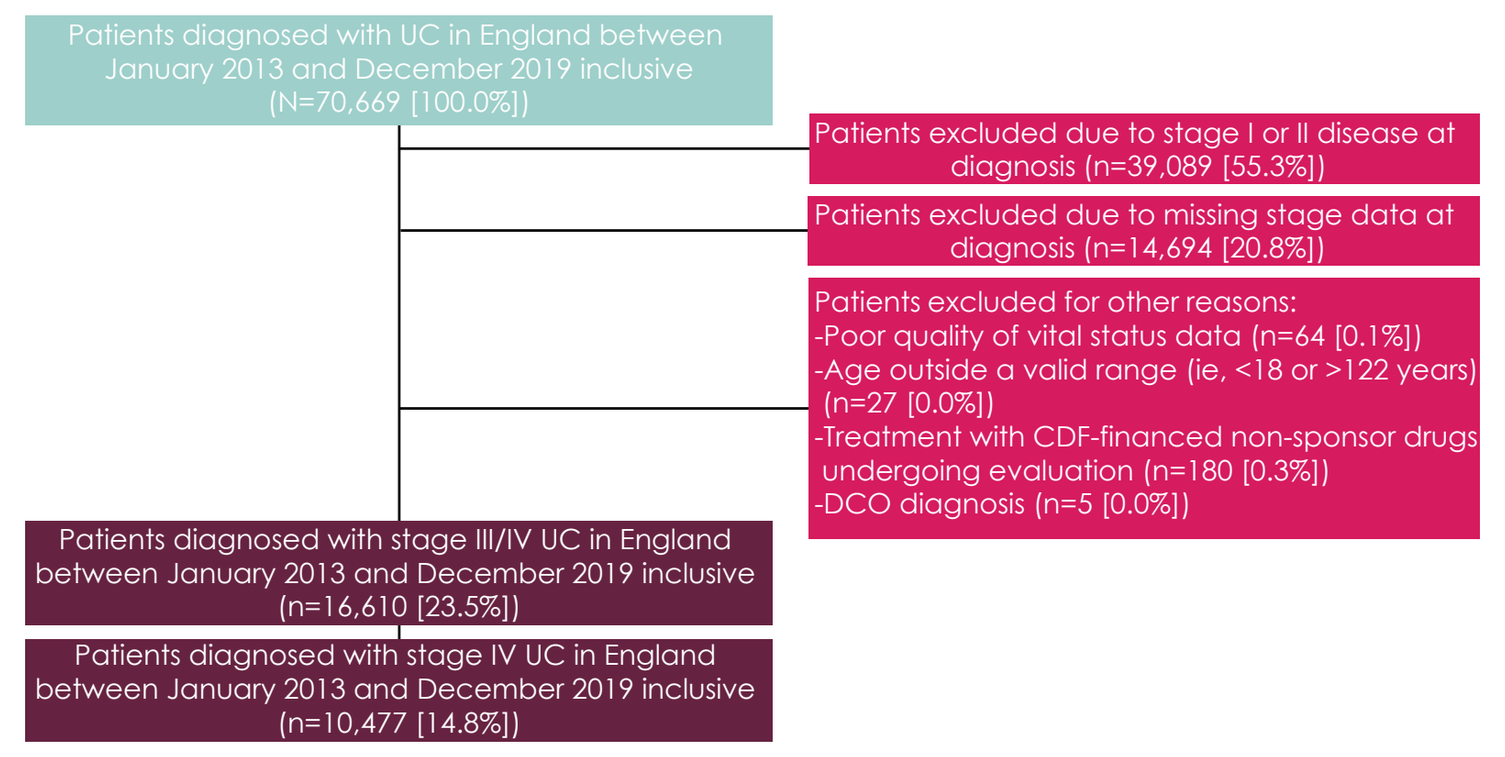

Patients diagnosed with UC in England between January 2013 and December 2019 inclusive (N=70,669 [100.0%])
Patients excluded due to stage I or II disease at diagnosis (n=39,089 [55.3%])
Patients excluded due to missing stage data at diagnosis (n=14,694 [20.8%])
Patients excluded for other reasons:
-Poor quality of vital status data (n=64 [0.1%])
-Age outside a valid range (ie, <18 or >122 years) (n=27 [0.0%])
-Treatment with CDF-financed non-sponsor drugs undergoing evaluation (n=180 [0.3%])
-DCO diagnosis (n=5 [0.0%])
Patients diagnosed with stage III/IV UC in England between January 2013 and December 2019 inclusive (n=16,610 [23.5%])
Patients diagnosed with stage IV UC in England between January 2013 and December 2019 inclusive (n=10,477 [14.8%])

All the proportions/percentages calculated are from the total patients diagnosed with UC in England between January 2013 and December 2019 inclusive.
CDF, Cancer Drugs Fund; **DCO,** death certificate only; **UC,** urothelial carcinoma.

RESULTS

Patient population

- 10,477 patients with mUC (mean [SD] age, 73.5 [11.2] years) were included; 65.2% were male (**Table 1**)
- Mean (SD) follow-up was 13.0 (18.0) months
- A total of 3,212 patients (30.7%) received systemic treatment (ie, initiated 1L), of whom 1,020 (31.7%) received 2L treatment
- Treated patients were younger (67.4 vs 76.1 years), had a lower comorbidity burden (baseline mean modified Deyo-Charlson Comorbidity Index (DCCI) score, 2.6 vs 3.8), and were less likely to have comorbid diagnoses consistent with probable cisplatin ineligibility (8.5% vs 23.6%) (**Table 1**)
- Within the overall study population, the most frequently captured comorbidities at baseline were hypertension (23.8%) and type 2 diabetes (8.1%)

Treated patients

- The most frequently prescribed anticancer agents in 1L were platinum agents (92.3%) and antimetabolites (89.1%)
- Immune checkpoint inhibitors (ICIs) were received by 40 patients (1.2%) in 1L and 359 patients (35.2%) in 2L of the total 1,020 patients who received 2L treatment

Table 1. mUC cohort and demographic characteristics stratified by receipt of systemic therapy

	All patients	Received systemic therapy	No systemic therapy	p value¶
Patients with incident mUC diagnosis, n (%)	10,477 (100)	3,212 (30.7)	7,265 (69.3)	
Cisplatin ineligible at baseline, n (%)*				
Yes	1,983 (18.9)	272 (8.5)	1,711 (23.6)	<0.001
Age at diagnosis, mean (SD), years	73.5 (11.2)	67.4 (10.0)	76.1 (10.6)	<0.001
Sex, n (%)				
Male	6,836 (65.2)	2,223 (69.2)	4,613 (63.5)	
Female	3,641 (34.8)	989 (30.8)	2,652 (36.5)	<0.001
England Government Office Region, n (%)				
East Midlands	796 (7.6)	242 (7.5)	554 (7.6)	<0.001
East of England	1,224 (11.7)	343 (10.7)	881 (12.1)	
London	1,020 (9.7)	402 (12.5)	618 (8.5)	
North East	708 (6.8)	197 (6.1)	511 (7.0)	
North West	1,445 (13.8)	508 (15.8)	937 (12.9)	
South East	1,625 (15.5)	484 (15.1)	1,141 (15.7)	
South West	1,178 (11.2)	395 (12.3)	783 (10.8)	
West Midlands	1,199 (11.4)	293 (9.1)	906 (12.5)	
Yorkshire and the Humber	1,282 (12.2)	348 (10.8)	934 (12.9)	
English Index of Multiple Deprivation 2015, income component, n (%)†				
Quintile 1 (least deprived)	1,966 (18.8)	649 (20.2)	1,317 (18.1)	0.001
Quintile 2	2,243 (21.4)	725 (22.6)	1,518 (20.9)	
Quintile 3	2,264 (21.6)	699 (21.8)	1,565 (21.5)	
Quintile 4	2,006 (19.1)	580 (18.1)	1,426 (19.6)	
Quintile 5 (most deprived)	1,998 (19.1)	559 (17.4)	1,439 (19.8)	
Year of diagnosis, n (%)				
2013	1,480 (14.1)	438 (13.6)	1,042 (14.3)	0.672
2014	1,482 (14.1)	477 (14.9)	1,005 (13.8)	
2015	1,684 (16.1)	498 (15.5)	1,186 (16.3)	
2016	1,678 (16.0)	508 (15.8)	1,170 (16.1)	
2017	1,930 (18.4)	597 (18.6)	1,333 (18.3)	
2018	1,222 (11.7)	377 (11.7)	845 (11.6)	
2019	1,001 (9.6)	317 (9.9)	684 (9.4)	
ECOG performance status at diagnosis, n (%)‡				
0	1,123 (10.7)	632 (19.7)	491 (6.8)	<0.001
1	893 (8.5)	381 (11.9)	512 (7.0)	
2	431 (4.1)	77 (2.4)	354 (4.9)	
3	289 (2.8)	16 (0.5)	273 (3.8)	
4	75 (0.7)	3 (0.1)	72 (1.0)	
Not recorded	7,666 (73.2)	2,103 (65.5)	5,563 (76.6)	
Duration of follow-up from diagnosis, mean (SD), months	13.0 (18.0)	22.4 (20.3)	8.9 (15.1)	<0.001
Modified DCCI score at baseline§,‖				
Mean, SD	3.4 (1.7)	2.6 (1.4)	3.8 (1.7)	<0.001

DCCI, Deyo-Charlson Comorbidity Index; **ECOG,** Eastern Cooperative Oncology Group; **ICD-10,** International Classification of Diseases, 10th Revision; **mUC,** metastatic urothelial carcinoma.
*Patients were considered unfit for cisplatin therapy if they met any of the following criteria during the period between 6 years and 1 day prior to the first cohort-relevant diagnosis: an ECOG performance status >1 (defined at diagnosis as it is the earliest performance status data available); chronic kidney disease, stage 3 or above (N18.3, N18.4, and N18.5); hearing loss (H90-H91); neuropathy (G60-G64); or heart failure (I11.0, I13.0, I13.2, I50.1-I50.4, I97.1). †The population-weighted quintile of income-related deprivation as defined for small areas in England. Further information on the 2015 index can be found here: https://www.gov.uk/government/statistics/english-indices-of-deprivation-2015 ‡The end of follow-up was defined as the date of death, embarkation, or the end of the study period, whichever came first. §The modified DCCI score quantifies the burden of patient comorbidity from 17 prespecified medical conditions, including but not limited to myocardial infarction, dementia, liver disease, and metastatic disease other than primary mUC. ‖Diagnoses abstracted from Hospital Episode Statistics inpatient and outpatient data only. "Baseline" refers to the period between 3 and 27 months prior to diagnosis. *ICD-10* codes exclude Z codes. Counts refer to patients with ≥1 cormobid diagnosis at baseline. ¶P values indicate the statistical significance of bivariate differences in the distributions of variables of interest between patients who did and did not receive systemic therapy. Categorical variables were tested using a χ^2 test. Continuous variables were tested using the Wilcoxon-Mann-Whitney test. This is a nonparametric analogue to the 2-tailed *t* test and appropriate when handling continuous skewed distributions and interval data.

Predictors of receipt of systemic anticancer treatment

- In multivariable analyses, patients were less likely to receive treatment if they were female (odds ratio, 0.72 [95% CI, 0.66-0.80]), cisplatin ineligible (0.58 [0.48-0.69]), older (0.94 [0.94-0.95] per year of age at diagnosis), were living in the lowest income quintile (0.58 [0.50-0.68] in the least deprived vs the most deprived), had a poor performance status (0.07 [0.02-0.24] for performance status 4 vs 0), or had a high comorbidity burden (0.88 [0.84-0.92] per additional unit of the modified DCCI score) (**Table 2**)

Table 2. Putative predictors of receipt of systemic therapy following diagnosis in patients with stage IV mUC

Demographic characteristics	Patients, n	Odd ratio (95% CI)	p value
Immunocompromised at baseline (n, %)*			
No	10,230	Reference	
Yes	247	0.87 (0.64-1.19)	0.379
Cisplatin ineligible at baseline (n, %)†			
No	8,494	Reference	
Yes	1,983	0.58 (0.48-0.69)	<0.001
Age at diagnosis, years	10,477	0.94 (0.94-0.95)	<0.001
Sex (n, %)			
Male	6,836	Reference	
Female	3,641	0.72 (0.66-0.80)	<0.001
England Government Office Region (n, %)			
London	1,020	Reference	
East Midlands	796	0.71 (0.57-0.88)	<0.001
East of England	1,224	0.60 (0.49-0.74)	<0.001
North East	708	0.61 (0.49-0.77)	<0.001
North West	1,445	0.79 (0.65-0.95)	0.012
South East	1,625	0.62 (0.52-0.75)	<0.001
South West	1,178	0.82 (0.67-0.99)	0.044
West Midlands	1,199	0.52 (0.42-0.63)	<0.001
Yorkshire and the Humber	1,282	0.57 (0.47-0.69)	<0.001
English Index of Multiple Deprivation 2015, income component (n, %)‡			
Quintile 1 (least deprived)	1,966	Reference	
Quintile 2	2,243	0.92 (0.80-1.06)	0.241
Quintile 3	2,264	0.80 (0.70-0.93)	0.003
Quintile 4	2,006	0.65 (0.55-0.75)	<0.001
Quintile 5 (most deprived)	1,998	0.58 (0.50-0.68)	<0.001
Year of diagnosis (n, %)			
2013	2,150	Reference	
2014	2,209	1.12 (0.94-1.33)	0.191
2015	2,513	1.01 (0.85-1.19)	0.952
2016	2,465	1.02 (0.86-1.21)	0.793
2017	2,736	1.08 (0.91-1.27)	0.377
2018	2,499	1.16 (0.96-1.39)	0.116
2019	2,038	1.17 (0.97-1.42)	0.109
ECOG performance status at diagnosis			
0	1,123	Reference	
1	893	0.82 (0.68-0.99)	0.044
2	431	0.46 (0.33-0.64)	<0.001
3	289	0.12 (0.07-0.20)	<0.001
4	75	0.07 (0.02-0.24)	<0.001
Not recorded	7,666	0.42 (0.36-0.48)	<0.001
Modified DCCI score at baseline§‖	10,477	0.88 (0.84-0.92)	<0.001

DCCI, Deyo-Charlson Comorbidity Index; **ECOG,** Eastern Cooperative Oncology Group; **ICD-10,** International Classification of Diseases, 10th Revision.
*Patients were considered immunocompromised if they had a history of multiple myeloma, chronic lymphocytic leukemia, hypogammaglobulinemia, a positive HIV or AIDS test, or had received an organ transplant (including allogeneic stem cell transplant). Diagnoses and procedures were captured from the cancer registry and the Hospital Episode Statistics inpatient and outpatient tables. The "baseline" period for this indicator is defined as the period between 6 years and 1 day prior to diagnosis. †Patients were considered unfit for cisplatin therapy if they met any of the following criteria during the period between 6 years and 1 day prior to the first cohort-relevant diagnosis: an ECOG performance status >1 (defined at diagnosis as it is the earliest performance status data available); chronic kidney disease, stage 3 or above (N18.3, N18.4, and N18.5); hearing loss (H90-H91); neuropathy (G60-G64); or heart failure (I11.0, I13.0, I13.2, I50.1-I50.4, I97.1). ‡The population-weighted quintile of income-related deprivation as defined for small areas in England. Further information on the 2015 index can be found here: https://www.gov.uk/government/statistics/english-indices-of-deprivation-2015. §The modified DCCI score quantifies the burden of patient comorbidity from 17 prespecified medical conditions, including but not limited to myocardial infarction, dementia, liver disease, and metastatic disease other than primary mUC. ‖Diagnoses abstracted from Hospital Episode Statistics (HES) inpatient and outpatient data only. "Baseline" refers to the period between 3 and 27 months prior to diagnosis. *ICD-10* codes exclude Z codes. Counts refer to patients with ≥1 cormobid diagnosis at baseline.

Clinical outcomes

- Median OS from diagnosis of stage IV disease was 15.1 (95% CI, 14.5-15.8) and 3.4 (3.3-3.6) months in treated and untreated patients, respectively (**Figure 2**)

Figure 2. Unadjusted OS from diagnosis in patients with stage IV UC, stratified by treatment status

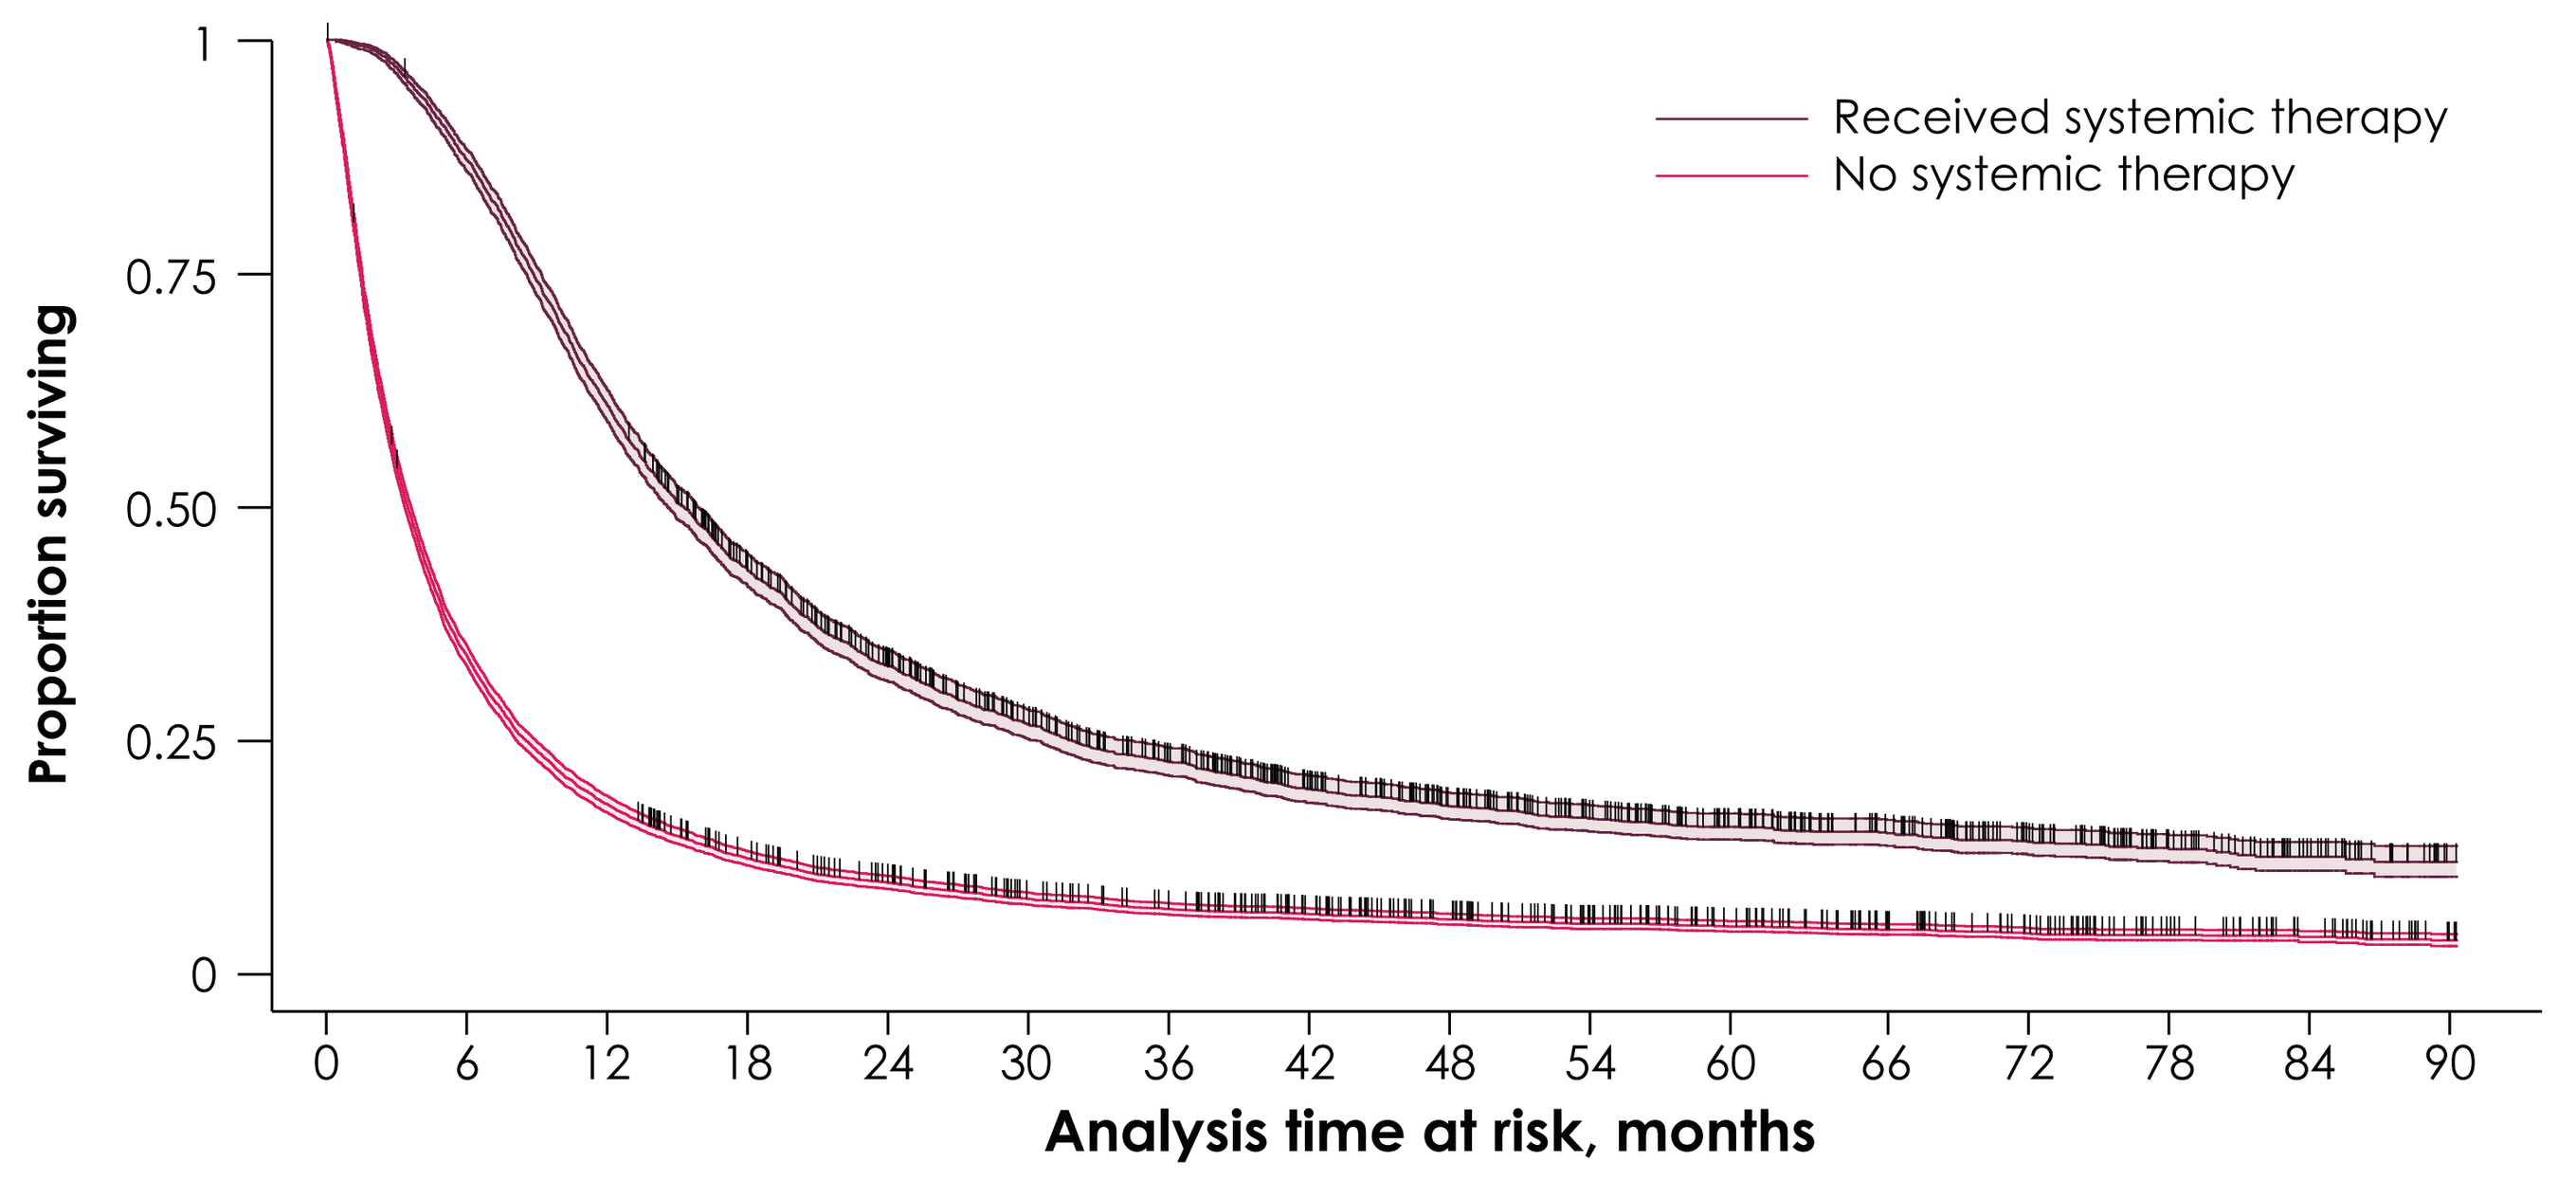

Received systemic therapy
No systemic therapy
Proportion surviving
1
0.75
0.50
0.25
0
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90
Analysis time at risk, months

No. at risk																
Received systemic therapy	3,212	2,792	1,940	1,338	983	757	612	478	393	318	256	200	151	99	58	28
No systemic therapy	7,265	2,458	1,320	884	681	530	450	376	299	244	197	149	119	82	58	28

All the proportions/percentages calculated are from the total patients diagnosed with UC in England between January 2013 and December 2019 inclusive.
OS, overall survival; **UC,** urothelial carcinoma.

STRENGTHS AND LIMITATIONS

Strengths

- Patients were selected from the National Cancer Registration Dataset, a nationally representative registry of all primary cancers diagnosed in England
- The registry benefits from mandatory data submission and robust validation procedures, maximizing completeness and standardizing recording to reduce the risk of misclassification
- The study uses patient-level linkage to the SACT dataset, which contains detailed information on all such therapies funded by the NHS and prescribed within a secondary care setting
- The secondary use of routine data ensures that the study did not affect the diagnostic process or the management of clinical symptoms and so did not introduce surveillance bias
- Reported findings are representative of real-world clinical practice in secondary care in England

Limitations

- Patients receiving non-sponsor agents delivered through the Cancer Drugs Fund were not included due to an embargo on treatment and health outcomes data. These patients may have differed in their treatment pathway and prognosis vs those selected into the cohorts
- The SACT dataset does not capture treatments provided in private or primary care settings, such that treatment may be proportionately underrepresented
- Lines of therapy are not readily available in the SACT dataset and so had to be inferred via algorithm according to observed changes in regimens over time, which may result in some degree of misclassification
- No information was available in the database regarding potential reasons why patients did not receive 1L systemic treatment
 - It is important to recognize that both physician and patient factors could have contributed to the choice of these treatments for older patients with mUC

REFERENCES 1. Cancer Research UK. Bladder cancer statistics. Accessed July 20, 2022. https://www.cancerresearchuk.org/health-professional/cancer-statistics/statistics-by-cancer-type/bladder-cancer. **2.** NHS Digital. Cancer registration statistics, England 2019. Accessed July 26, 2022. https://digital.nhs.uk/data-and-information/publications/statistical/cancer-registration-statistics/england-2019. **3.** Office for National Statistics. Cancer registration statistics, England: 2017. Accessed July 20, 2022. https://www.ons.gov.uk/peoplepopulationandcommunity/healthandsocialcare/conditionsanddiseases/bulletins/cancerregistrationstatisticsengland/2017. **4.** Witjes JA, et al; European Association of Urology Guidelines Panel for Muscle-invasive and Metastatic Bladder Cancer. EAU Guidelines on muscle-invasive and metastatic bladder cancer. Accessed July 14, 2022. https://uroweb.org/guideline/bladder-cancer-muscle-invasive-and-metastatic/. **5.** Powles T, et al. Bladder cancer: ESMO Clinical Practice Guideline for diagnosis, treatment and follow-up. Ann Oncol. 2022;33(3):244-58. **6.** Bright CJ, et al. Data resource profile: the Systemic Anti-Cancer Therapy (SACT) dataset. Int J Epidemiol. 2020;49(1):15-15l. **7.** Henson KE, et al. Data resource profile: National Cancer Registration Dataset in England. Int J Epidemiol. 2020;49(1):16-16h.

DISCLOSURES C. Knott has no conflicts to declare. **M. Kearney** is an employee of Merck Healthcare KGaA, Darmstadt, Germany, and holds stocks/shares in Merck, Novartis Pharma, and UCB Biopharma SPRL. **S. H. Mahmoudpour** is an employee of Merck Healthcare KGaA, Darmstadt, Germany. **P. Verpillat** is an employee of and holds stocks/shares in Merck. **ACKNOWLEDGMENTS** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This research was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945) as part of an alliance between Merck and Pfizer. Editorial support was provided by Hiba Al-Ashtal of Clinical Thinking and was funded by Merck and Pfizer. This work uses data that have been provided by patients and collected by the NHS as part of their care and support. The data are collated, maintained, and quality assured by the National Disease Registration Service, which is part of NHS Digital.

Poster No. 1750P. Presented at the ESMO Congress 2022, 9-13 September 2022; Paris, France; Hybrid.