This is a reprint from European Society for Medical Oncology Congress 2023 (ESMO 2023), which was originally presented in Madrid, Spain on October 20-24, 2023; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

First-line treatment patterns and overall survival of patients with advanced Merkel cell carcinoma in England from 2013-2022: results of a nationwide observational cohort study

C. S. Knott,[1] S. H. Mahmoudpour,[2] M. Kearney,[2] E. Boutmy,[2] P. Verpillat[2]*

[1]Health Data Insight CIC; NHS England, London, UK; [2]Merck Healthcare KGaA, Darmstadt, Germany

*Affiliation at the time the study was conducted.

SCOPE

- **This noninterventional study reports patient demographics, treatment characteristics, and overall survival (OS) in patients with locally advanced (stage III) or metastatic (stage IV) Merkel cell carcinoma (MCC) who were diagnosed between 2013 and 2020 in England**

CONCLUSIONS

- **Approximately 70% of patients with advanced MCC were untreated**
 - **This is a substantial proportion given the availability of effective treatments**
- **Results from this nationwide cohort study provide further real-world evidence for the clinical effectiveness of avelumab as a first-line (1L) treatment for patients with locally advanced (stage III) or metastatic (stage IV) MCC**
- **These findings validate results from part B of the JAVELIN Merkel 200 clinical trial,[1] as well as findings from observational studies outside of England[2,3]**

GET POSTER PDF

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

Correspondence: **Craig Knott, craig.knott1@nhs.net**

BACKGROUND

- MCC is a rare and aggressive cutaneous neoplasm[4] with no established treatment guideline in England[5]
- Overall prognosis is poor, with a 5-year disease-specific survival rate in England of 65.6% (95% CI, 63.8%-64.7%)
- The risk of death is 8 times higher among patients with stage IV disease vs patients with stage III disease (HR, 8.24 [95% CI, 5.84-11.61])[6]
- This poor survival rate is likely to have been exacerbated by a historic lack of effective treatment options, with <10% of all patients with MCC receiving systemic anticancer therapy in England[4]
- Both the European Medicines Agency[7,8] and the National Institute for Health and Care Excellence (NICE)[9] approved avelumab for the treatment of metastatic MCC (stage IV), marking the advent of immune checkpoint inhibitor therapy for MCC across the EU and England
- With avelumab having entered routine commissioning for metastatic MCC in England, initially for patients with prior chemotherapy in March 2018, followed by patients with untreated metastatic MCC in April 2021, this study reports the uptake of avelumab among patients in England who were diagnosed with stage IV disease, as well as differences in OS according to whether avelumab was prescribed
- Results are also presented for patients with stage III disease at diagnosis

METHODS

- Using the National Cancer Registration Dataset (NCRD),[10] a cohort was created that selected patients aged ≥18 years who were newly diagnosed with locally advanced or metastatic MCC between 2013 and 2020 in England (**Figure 1**)
- Patient records were linked to the Systemic Anti-Cancer Therapy (SACT) dataset,[11] which captures treatments prescribed by the National Health Service (NHS) in secondary care settings. Additionally, information from the Hospital Episode Statistics datasets was used to classify patients as immunocompromised if they had evidence of relevant healthcare events at diagnosis (eg, HIV or organ transplant)
- Patients were followed from diagnosis until death or end of study (May 2022), whichever occurred first
- Descriptive statistics were calculated for categorical and continuous variables of interest, and the Kaplan-Meier method was used to estimate OS
- OS is reported from initiation of 1L therapy and stratified by stage at diagnosis and treatment type

Figure 1. Cohort selection criteria

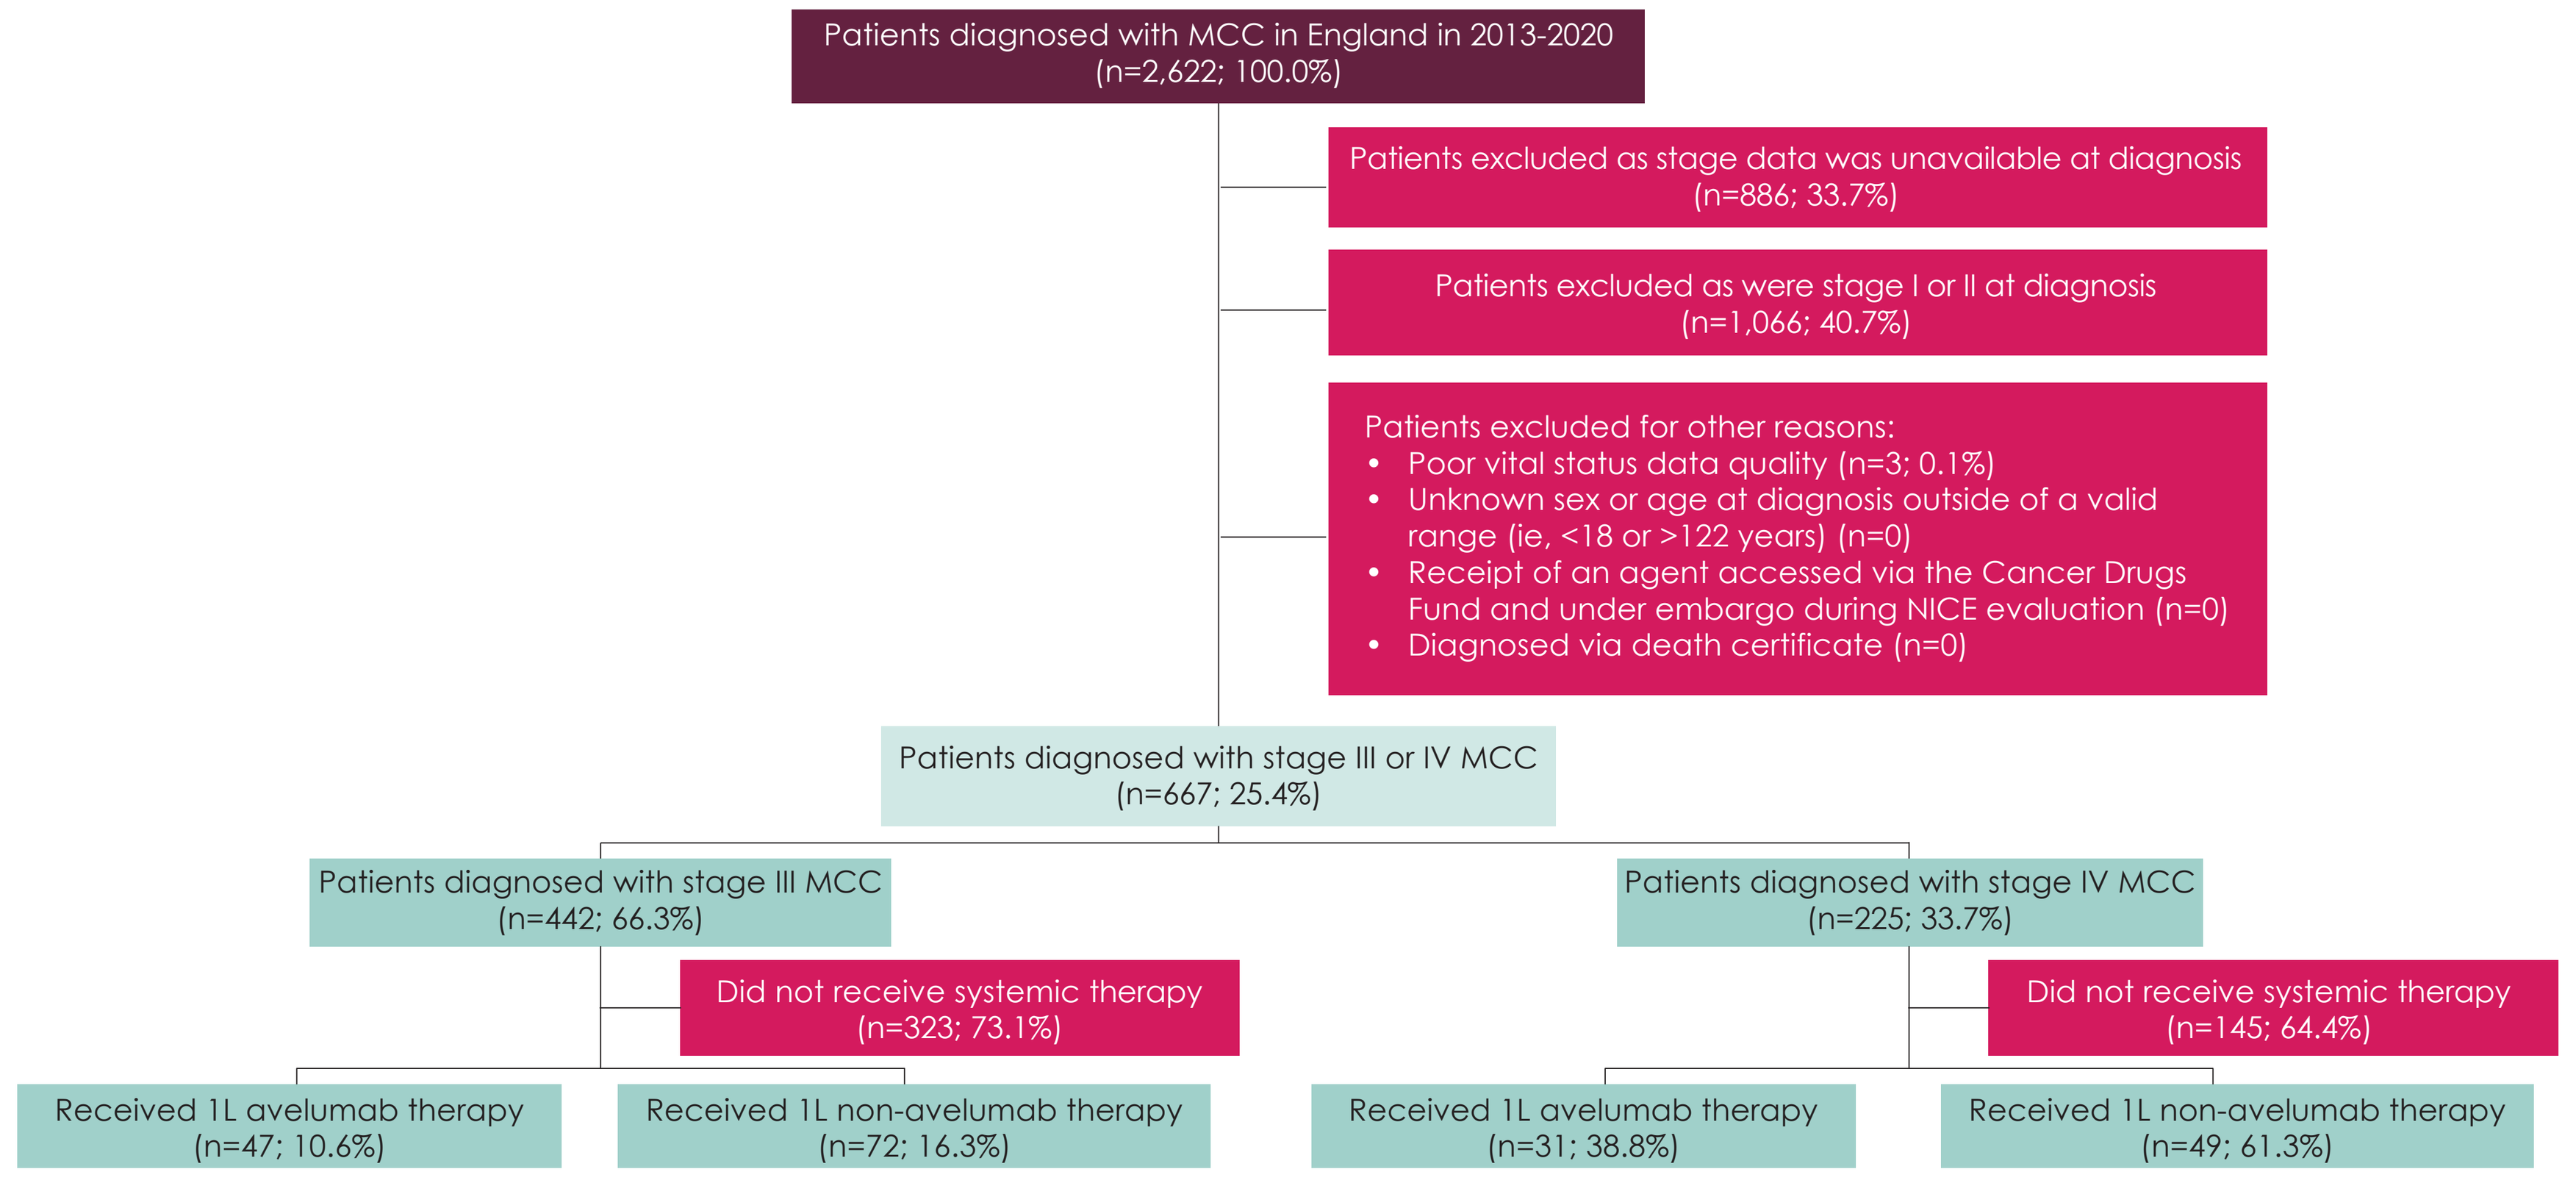
Patients diagnosed with MCC in England in 2013-2020 (n=2,622; 100.0%)
Patients excluded as stage data was unavailable at diagnosis (n=886; 33.7%)
Patients excluded as were stage I or II at diagnosis (n=1,066; 40.7%)
Patients excluded for other reasons:
• Poor vital status data quality (n=3; 0.1%)
• Unknown sex or age at diagnosis outside of a valid range (ie, <18 or >122 years) (n=0)
• Receipt of an agent accessed via the Cancer Drugs Fund and under embargo during NICE evaluation (n=0)
• Diagnosed via death certificate (n=0)
Patients diagnosed with stage III or IV MCC (n=667; 25.4%)
Patients diagnosed with stage III MCC (n=442; 66.3%)
Patients diagnosed with stage IV MCC (n=225; 33.7%)
Did not receive systemic therapy (n=323; 73.1%)
Did not receive systemic therapy (n=145; 64.4%)
Received 1L avelumab therapy (n=47; 10.6%)
Received 1L non-avelumab therapy (n=72; 16.3%)
Received 1L avelumab therapy (n=31; 38.8%)
Received 1L non-avelumab therapy (n=49; 61.3%)

1L, first line; **MCC**, Merkel cell carcinoma; **NICE**, National Institute for Health and Care Excellence.

RESULTS

- A total of 667 patients met the cohort selection criteria. Mean age at diagnosis was 77.4 years (standard deviation, 10.3), and the majority of patients were male (61.0%). A small proportion of patients were classified as immunocompromised (14.1%), and most had disease that was locally advanced at diagnosis (stage III, 66.3%; stage IV, 33.7%)
- Most patients (70.2%) received no systemic anticancer therapy. Of patients who received systemic therapy, 39.2% received 1L avelumab therapy. Among the 121 patients (60.8%) in the non-avelumab group, 95.5% received 1L chemotherapy
- The proportion of patients who received 1L therapy was higher among those diagnosed with stage IV disease (stage III, 26.9%; stage IV, 38.7%). Avelumab was received by a similar proportion of patients with stage III and stage IV disease at diagnosis (**Figure 2**)
- In the overall cohort, 478 patients (71.7%) died during follow-up, and median OS from diagnosis was 18.4 months (95% CI, 15.6-20.9)
- Median OS with 1L avelumab was 19.9 months (95% CI, 5.0-not estimable); this finding was consistent with results from part B of the JAVELIN Merkel 200 clinical trial (20.3 months [95% CI, 12.4-not estimable])[1]
- Median OS following the initiation of 1L therapy was more than twice as long among recipients of avelumab than recipients of non-avelumab therapy, regardless of stage at diagnosis (**Table 1** and **Figure 3**)

Figure 2. Receipt of anticancer therapy by stage at diagnosis

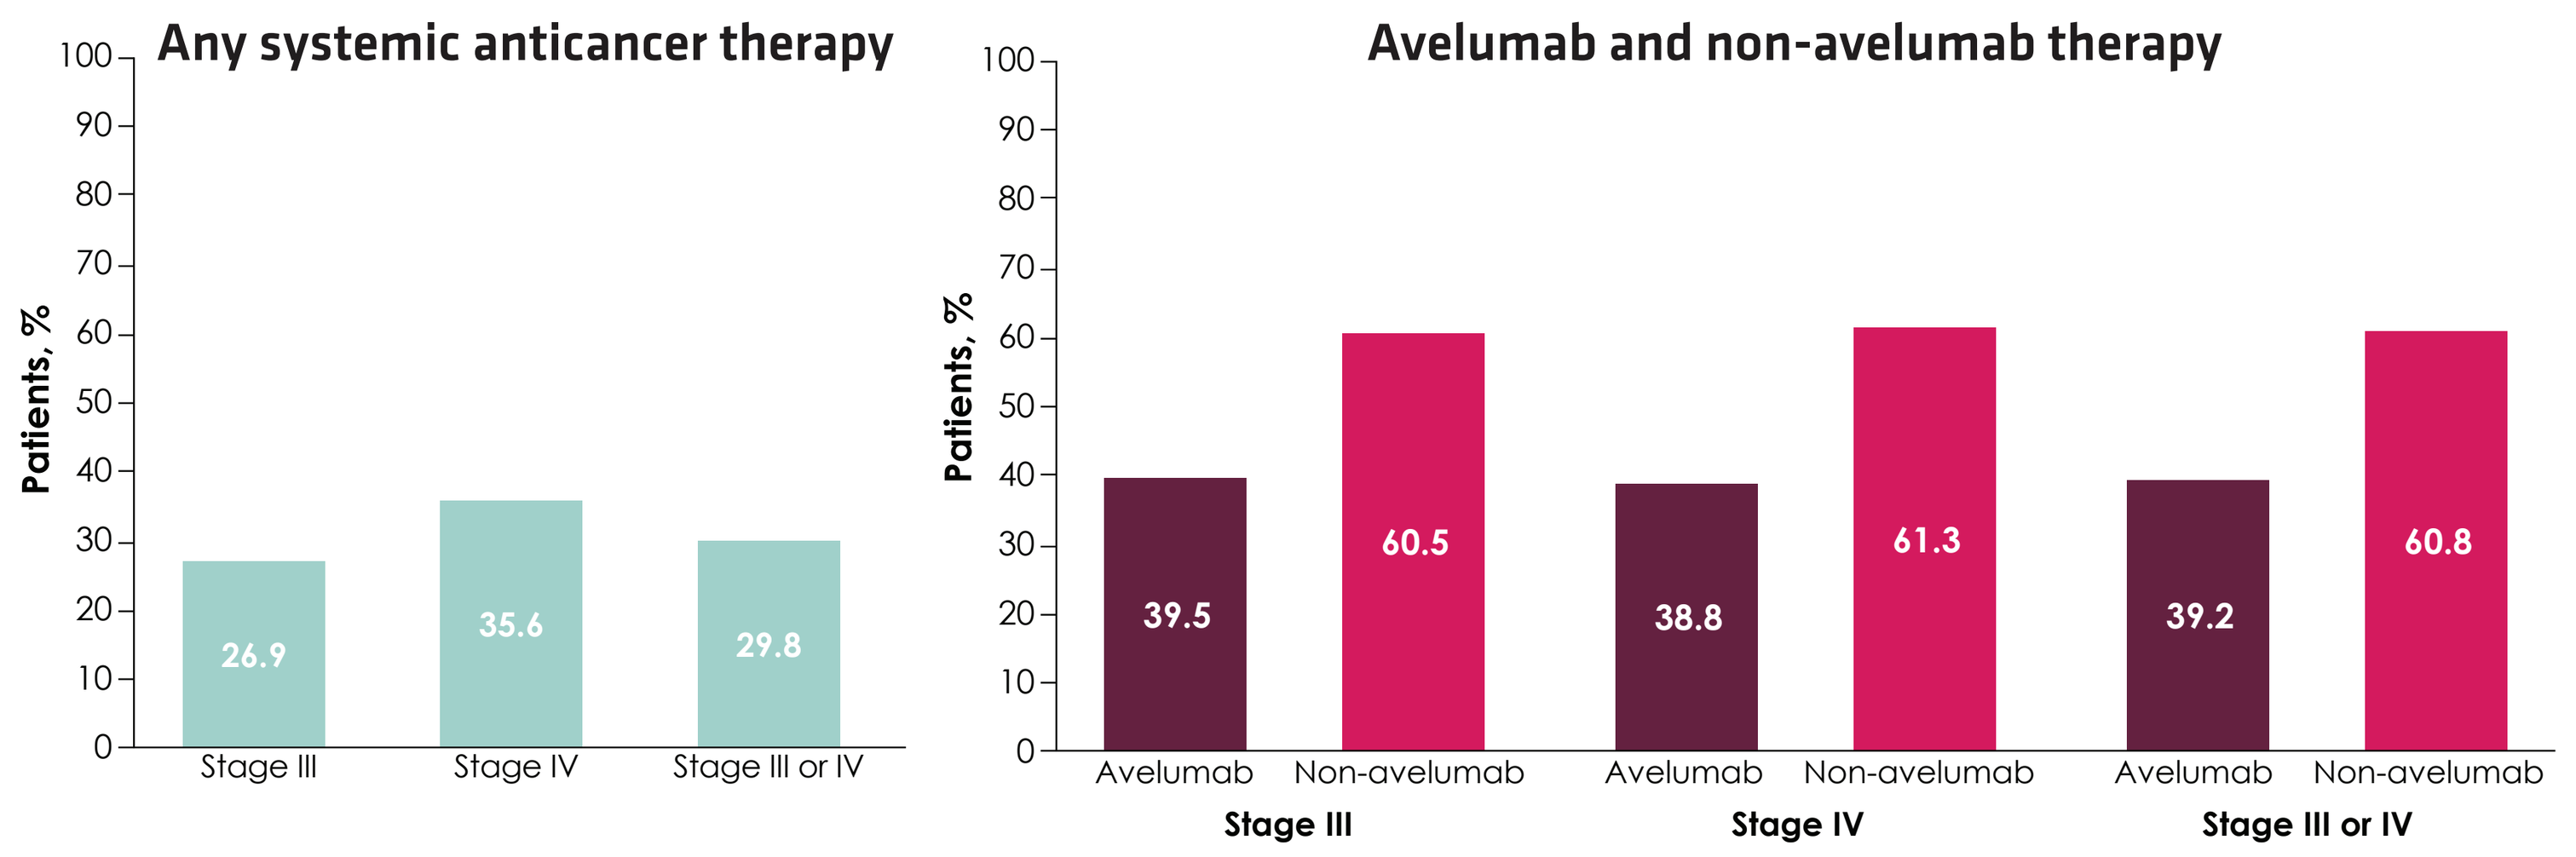
Any systemic anticancer therapy
Patients, %
0
10
20
30
40
50
60
70
80
90
100
26.9
35.6
29.8
Stage III
Stage IV
Stage III or IV
Avelumab and non-avelumab therapy
39.5
60.5
38.8
61.3
39.2
60.8
Avelumab
Non-avelumab
Stage III
Stage IV
Stage III or IV

Table 1. OS from initiation of 1L therapy by stage at diagnosis and treatment type

	Stage III		Stage IV	
	Avelumab	**Non-avelumab**	**Avelumab**	**Non-avelumab**
Total patients, n	47	72	31	49
Deaths during follow-up, n (%)	20 (42.6)	54 (75.0)	16 (51.6)	40 (81.6)
Median OS (95% CI), months	37.8 (12.9-NE)	13.0 (8.3-19.6)	19.9 (5.0-NE)	7.2 (5.9-9.1)
12-month OS rate (95% CI), %	67.0 (51.3-78.7)	52.1 (40.0-63.0)	54.8 (36.0-70.3)	30.6 (18.5-43.6)
18-month OS rate (95% CI), %	55.8 (39.4-69.4)	40.9 (29.4-52.0)	51.6 (33.0-67.4)	24.5 (13.6-37.1)
24-month OS rate (95% CI), %	55.8 (39.4-69.4)	35.1 (24.2-46.1)	47.6 (29.2-64.0)	NE

1L, first line; **NE**, not estimable; **OS**, overall survival.

Figure 3. OS from initiation of 1L therapy by stage at diagnosis

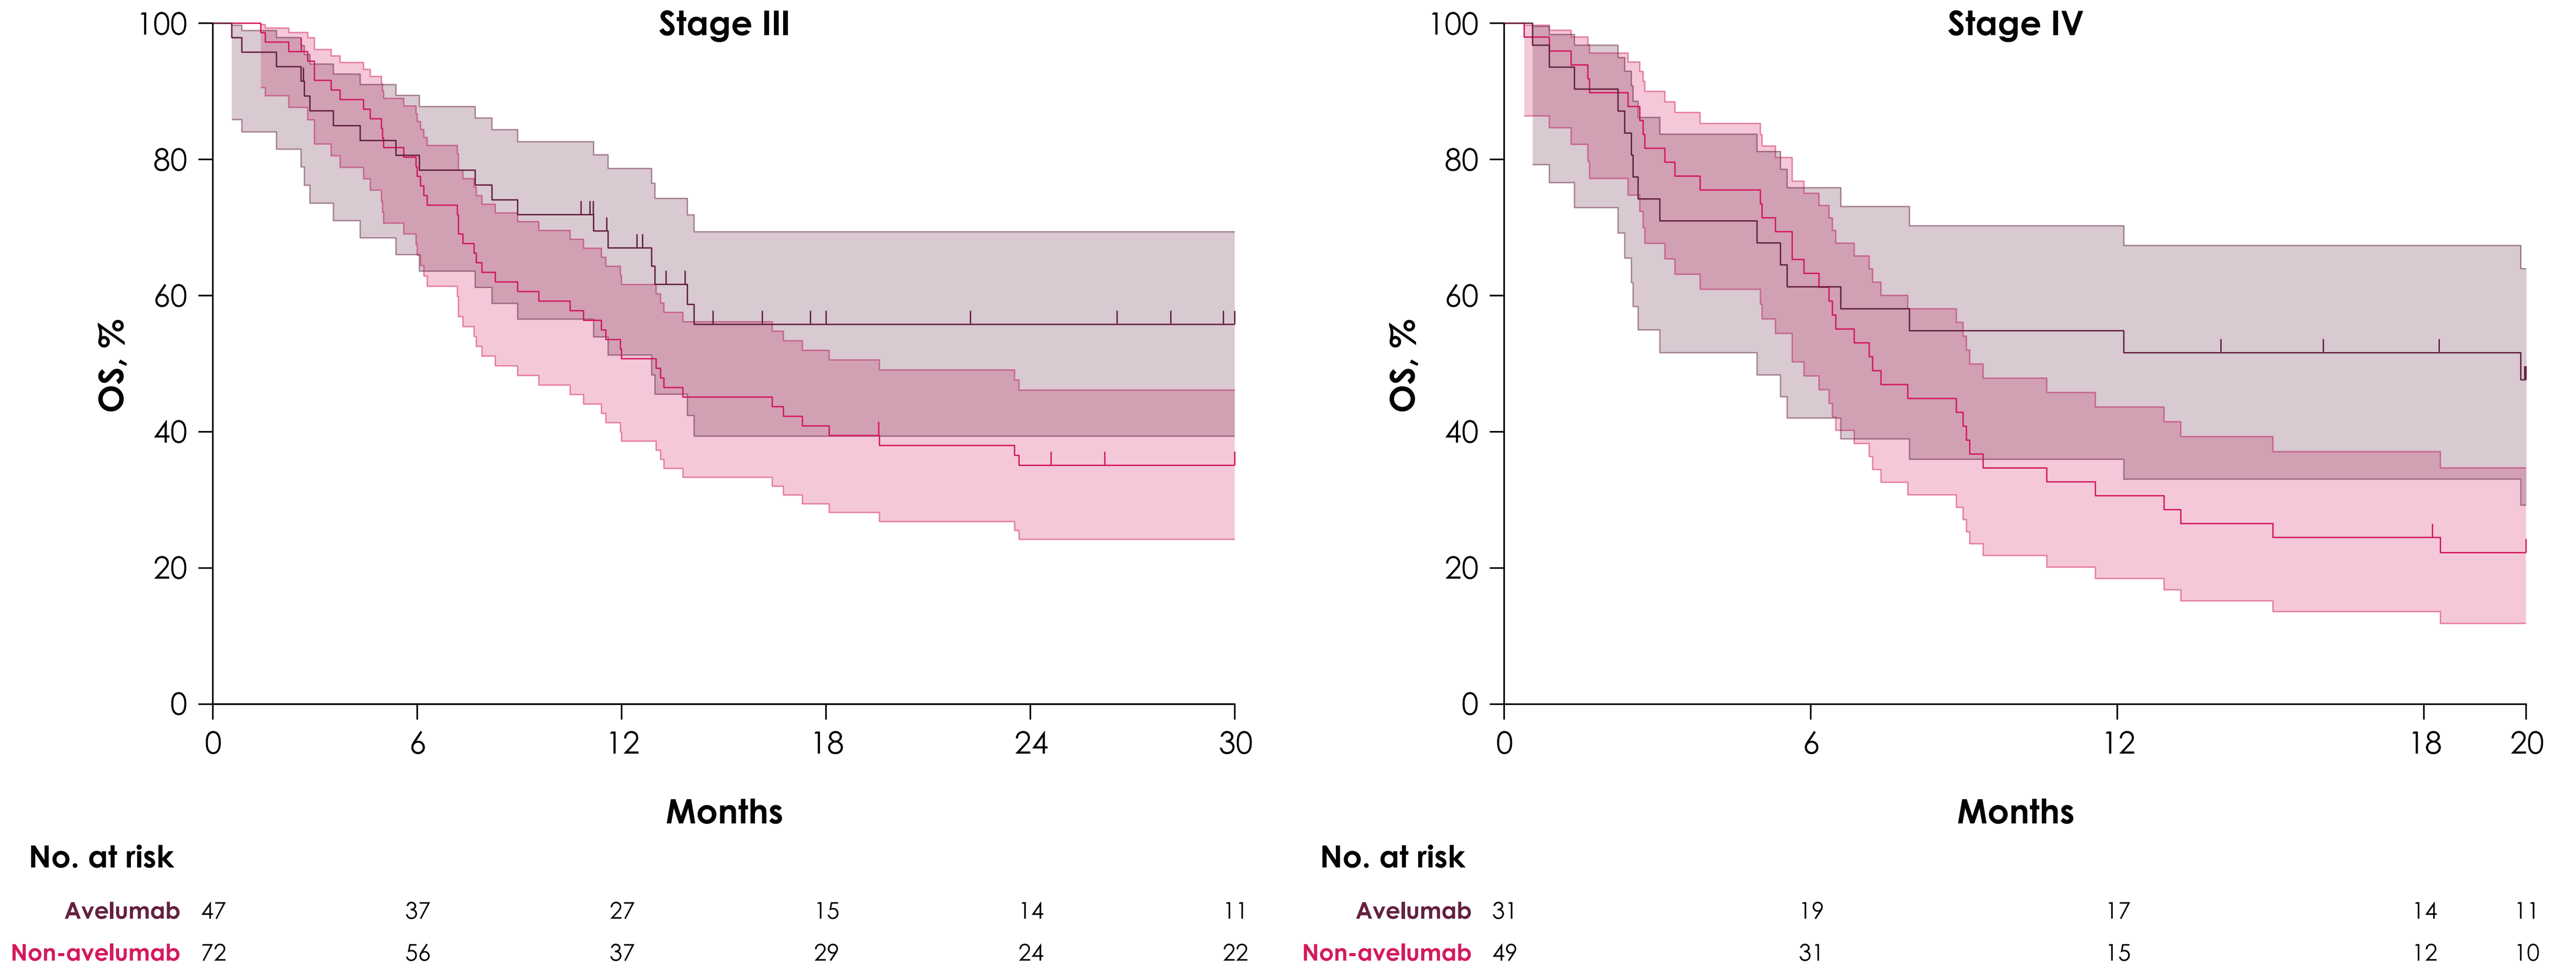
Stage III
OS, %
0
20
40
60
80
100
0
6
12
18
24
30
Months
No. at risk
Avelumab 47 37 27 15 14 11
Non-avelumab 72 56 37 29 24 22
Stage IV
OS, %
0
20
40
60
80
100
0
6
12
18
20
Months
No. at risk
Avelumab 31 19 17 14 11
Non-avelumab 49 31 15 12 10

1L, first line; **OS**, overall survival.

STRENGTHS AND LIMITATIONS

- The NCRD is a population-based registry that contains all documented malignant and premalignant neoplasms diagnosed in England since 1971
- While providing a wealth of information about patients and their time of diagnosis, the NCRD lacks prospective patient data, such as recurrence or disease progression. Accordingly, it was not possible to include patients who were diagnosed with early-stage (stage I or II) disease that later progressed. Furthermore, some patients with stage III disease at diagnosis may have received additional treatment following disease progression that this study could not capture
- This study used patient-level linkage to the SACT dataset, through which it was possible to track patients through their systemic anticancer therapy pathways. It is important to note that the SACT dataset only captures prescriptions made within the secondary-care system of NHS; private- or primary-care prescriptions are not documented
- Higher rates of avelumab use may be observed for patients newly diagnosed with advanced MCC from 2021 onwards; the present cohort comprises patients diagnosed from 2013 to 2020, and while patients could access avelumab via the Cancer Drugs Fund from March 2018, NICE did not grant approval for routine commissioning for 1L treatment until April 2021
- Here, OS data are presented without adjustment for confounding by indication between avelumab and non-avelumab treatments. Data completeness for important confounding factors, such as patient performance status, is limited within the NCRD. Adjusted estimates would have required selection of a subset of an already rare patient population

REFERENCES 1. D'Angelo SP, et al. J Immunother Cancer. 2021;9:e002646. **2.** Bhatia S, et al. J Immunother Cancer. 2022;10:e004904. **3.** Cowey CL, et al. Future Oncol. 2021;17:2339-50. **4.** Zaggana E, et al. Cancers (Basel). 2023;15:103. **5.** Afzal B, et al. Br J Dermatol. 2022;187(suppl 1):60. **6.** Mistry K, et al. Br J Dermatol. 2023;188:228-36. **7.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA; 2022. **8.** National Institute for Health and Care Excellence. Accessed 22 August 2023. https://www.nice.org.uk/guidance/ta691. **9.** Henson KE, et al. Int J Epidemiol. 2020;49:16-16h. **10.** Bright CJ, et al. Int J Epidemiol. 2020;49:15-15l. **11.** Gauci ML, et al. Eur J Cancer. 2022;171:203-31.
DISCLOSURES C. S. Knott reports employment by Health Data Insight CIC and NHS England. **S. H. Mahmoudpour** reports employment by Merck. **M. Kearney** reports employment by and stock ownership in Merck. **E. Boutmy** reports employment by and stock ownership interest in Merck. **P. Verpillat** reports employment by Merck at the time of the study. **ACKNOWLEDGMENTS** This study was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945), and was previously conducted under an alliance between Merck and Pfizer. Editorial support was provided by Clinical Thinking and was funded by Merck. This study used patient-level information collected by the NHS as part of the care and support of patients with cancer. The data are collated, maintained, and quality assured by the National Cancer Registration and Analysis Service, which is part of NHS England.

Poster No. 1147P. Presented at the ESMO Congress 2023, 20-24 October 2023; Madrid, Spain.