XRay Vision: a phase 3 study of xevinapant plus radiotherapy (RT) for high-risk, cisplatin-ineligible patients with resected, locally advanced squamous cell carcinoma of the head and neck (LA SCCHN)

R. L. Ferris,[1] **H. Mehanna,**[2] **J. D. Schoenfeld,**[3] **M. Tahara,**[4] **S. S. Yom,**[5] **R. Haddad,**[3] **A. König,**[6] **S. Salmio,**[6] **M. Bajars,**[6] **C. Le Tourneau**[7]

[1]University of Pittsburgh Medical Center, Pittsburgh, PA, USA; [2]University of Birmingham, Birmingham, West Midlands, UK; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]National Cancer Center Hospital East, Kashiwa, Chiba Prefecture, Japan; [5]University of California San Francisco, San Francisco, CA, USA; [6]the healthcare business of Merck KGaA, Darmstadt, Germany; [7]Institut Curie, Paris, France

SCOPE

- XRay Vision (NCT05386550) is an international, randomized, double-blind, placebo-controlled, phase 3 study evaluating xevinapant in patients with resected LA SCCHN who have a high risk of relapse and are deemed ineligible for cisplatin
- XRay Vision aims to demonstrate improvement in disease-free survival (DFS) with xevinapant + RT vs placebo + RT, irrespective of subsequent anticancer therapy

STUDY STATUS

- Study recruitment began in October 2022 with patient enrollment ongoing
- The estimated primary completion date is October 2027
- For more information, please visit ClinicalTrials.gov at this QR code

GET POSTER PDF

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO and the authors of this poster

Correspondence: **Robert L. Ferris, ferrrl@upmc.edu**

GET PLAIN LANGUAGE SUMMARY

Please scan this QR code with your smartphone app to view a plain language summary

BACKGROUND

- Head and neck cancer is the eighth most commonly diagnosed cancer globally, with 878,348 new cases and 444,347 deaths recorded in 2020[1]
- Most head and neck cancers (≈90%) are squamous cell carcinomas,[2] and the majority of patients (≈60%) are diagnosed with LA disease[3]
- The current standard of care for patients with resected LA SCCHN who are at high risk of recurrence postoperatively is adjuvant chemoradiotherapy (CRT; cisplatin + concomitant fractionated RT [66 Gy in 33 daily fractions of 2 Gy over 6.5 weeks])[3,4]
- However, many patients are deemed ineligible to receive cisplatin-based chemotherapy,[5,6] and there is a lack of treatment options and consensus in international guidelines regarding the recommended adjuvant treatment for these patients
- RT alone remains the standard-of-care treatment for patients with resected, high-risk LA SCCHN who are deemed ineligible to receive cisplatin
- A high unmet need remains for this population of patients and novel treatment options are urgently required

- Xevinapant is a first-in-class, potent, oral, small-molecule inhibitor of apoptosis protein (IAP) inhibitor that is thought to restore cancer cell sensitivity to apoptosis and thereby enhance the efficacy of chemotherapy and RT[7-9] (**Figure 1**):
 - Xevinapant inhibits X-linked IAP and cellular IAP 1 and 2 (cIAP1/2), releasing the blockade on downstream caspase activity, which is crucial for apoptosis and anticancer activity of chemotherapy and RT[7]
 - Inhibition of cIAP1/2 may also amplify immune cell activation by activating noncanonical nuclear factor–κB signaling, which induces the production of inflammatory cytokines in response to tumor necrosis factor receptor signaling[7,10-12]
- In a randomized phase 2 study of patients with unresected LA SCCHN, xevinapant + CRT significantly improved locoregional control at 18 months after the end of chemoradiotherapy (54% vs 33%; odds ratio, 2.74; 95% CI, 1.15-6.53, *P*=0.0232) and halved the risk of death after 5 years of follow-up vs placebo + CRT (HR, 0.47; 95% CI, 0.27-0.84; *P*=0.0101; median OS, not reached vs 36.1 months)[14,15]
- Xevinapant has also shown synergistic activity with RT in preclinical models of SCCHN[7,13]
- The promising clinical activity of xevinapant in combination with CRT, as well as its preclinical antitumor activity in combination with RT alone provide the rationale for evaluating xevinapant in combination with RT in the phase 3 XRay Vision study

Figure 1. Xevinapant mode of action

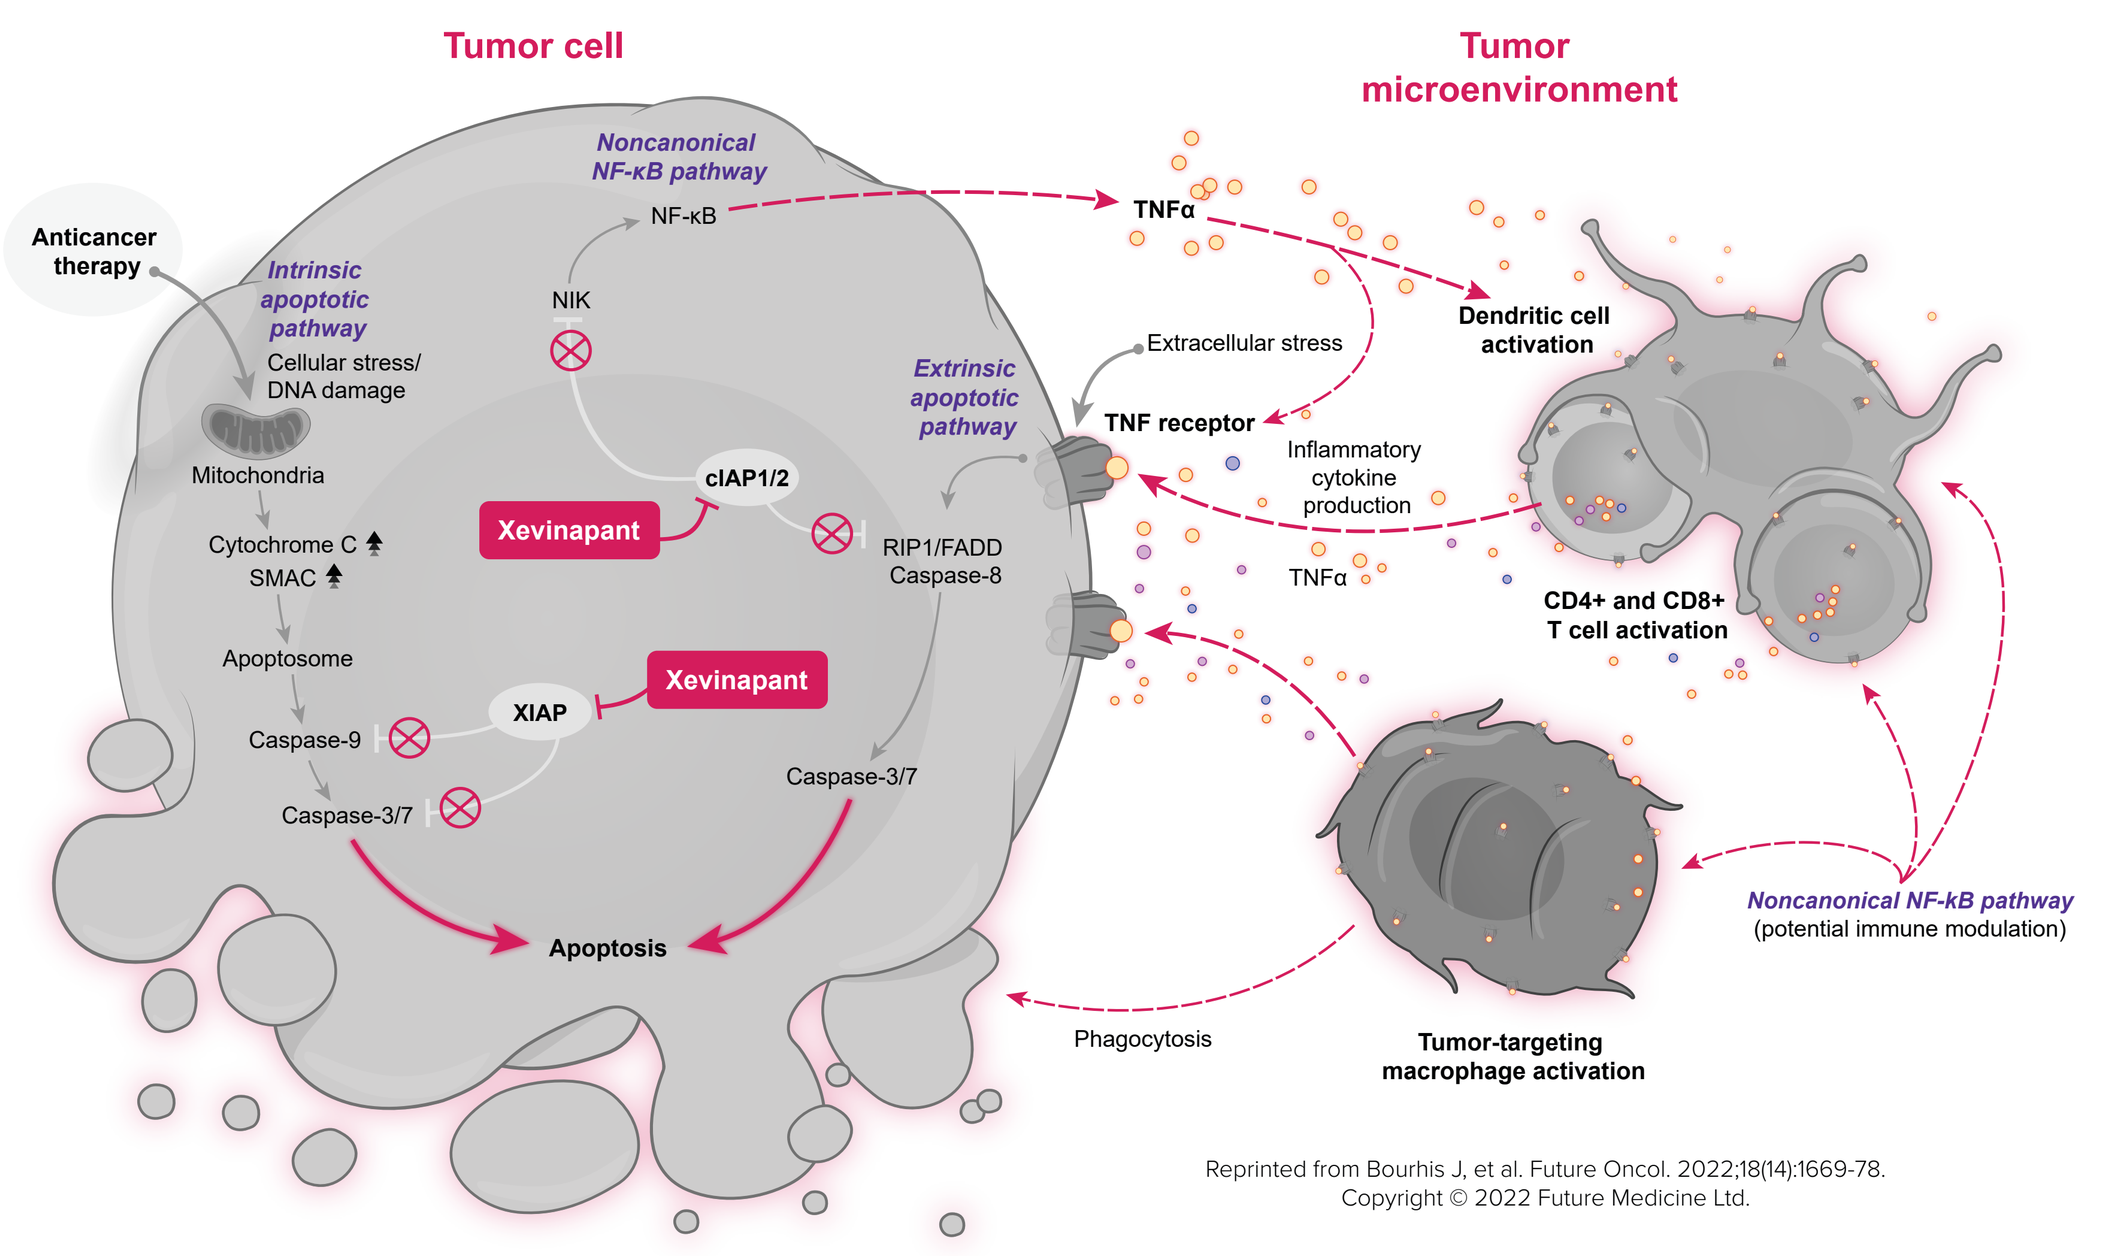
Tumor cell
Tumor microenvironment
Anticancer therapy
Noncanonical NF-κB pathway
NF-κB
Intrinsic apoptotic pathway
Cellular stress/ DNA damage
NIK
Mitochondria
Cytochrome C
SMAC
Apoptosome
Caspase-9
Caspase-3/7
Xevinapant
cIAP1/2
XIAP
Extrinsic apoptotic pathway
RIP1/FADD Caspase-8
Apoptosis
TNFα
Extracellular stress
TNF receptor
Inflammatory cytokine production
Dendritic cell activation
CD4+ and CD8+ T cell activation
Noncanonical NF-kB pathway (potential immune modulation)
Phagocytosis
Tumor-targeting macrophage activation
Reprinted from Bourhis J, et al. Future Oncol. 2022;18(14):1669-78. Copyright © 2022 Future Medicine Ltd.

cIAP1/2, cellular IAPs 1 and 2; **FADD,** fas-associated protein with death domain; **IAP,** inhibitors of apoptosis protein; **NFκB**, nuclear factor kappa-light-chain-enhancer of activated B cells; **NIK**, NF-κB-inducing kinase; **RIP1**, receptor interacting serine/threonine kinase; **SMAC,** second mitochondria-derived activator of caspase; **TNFα,** tumor necrosis factor-α; **TRAIL,** TNF-related apoptosis-inducing ligand; **XIAP,** X-linked apoptosis protein.

METHODS

Study design and patients

- XRay Vision is a randomized, double-blind, placebo-controlled, international, phase 3 study evaluating the efficacy of xevinapant + intensity-modulated RT (IMRT) vs placebo + IMRT in ≈700 patients with resected LA SCCHN who have a high risk of relapse and are deemed ineligible to receive cisplatin (**Figure 2**)
- The primary endpoint of the study is DFS, defined as the time from randomization to objective disease recurrence or death from any cause (a summary of all endpoints is given in **Table 1**)
- Eligible patients have histologically confirmed cancer of the oral cavity, oropharynx, hypopharynx, or larynx, prior surgery with curative intent 4-8 weeks before the start of treatment, and no residual disease (eligibility criteria are summarized in **Table 2**)

Figure 2. XRay Vision study design

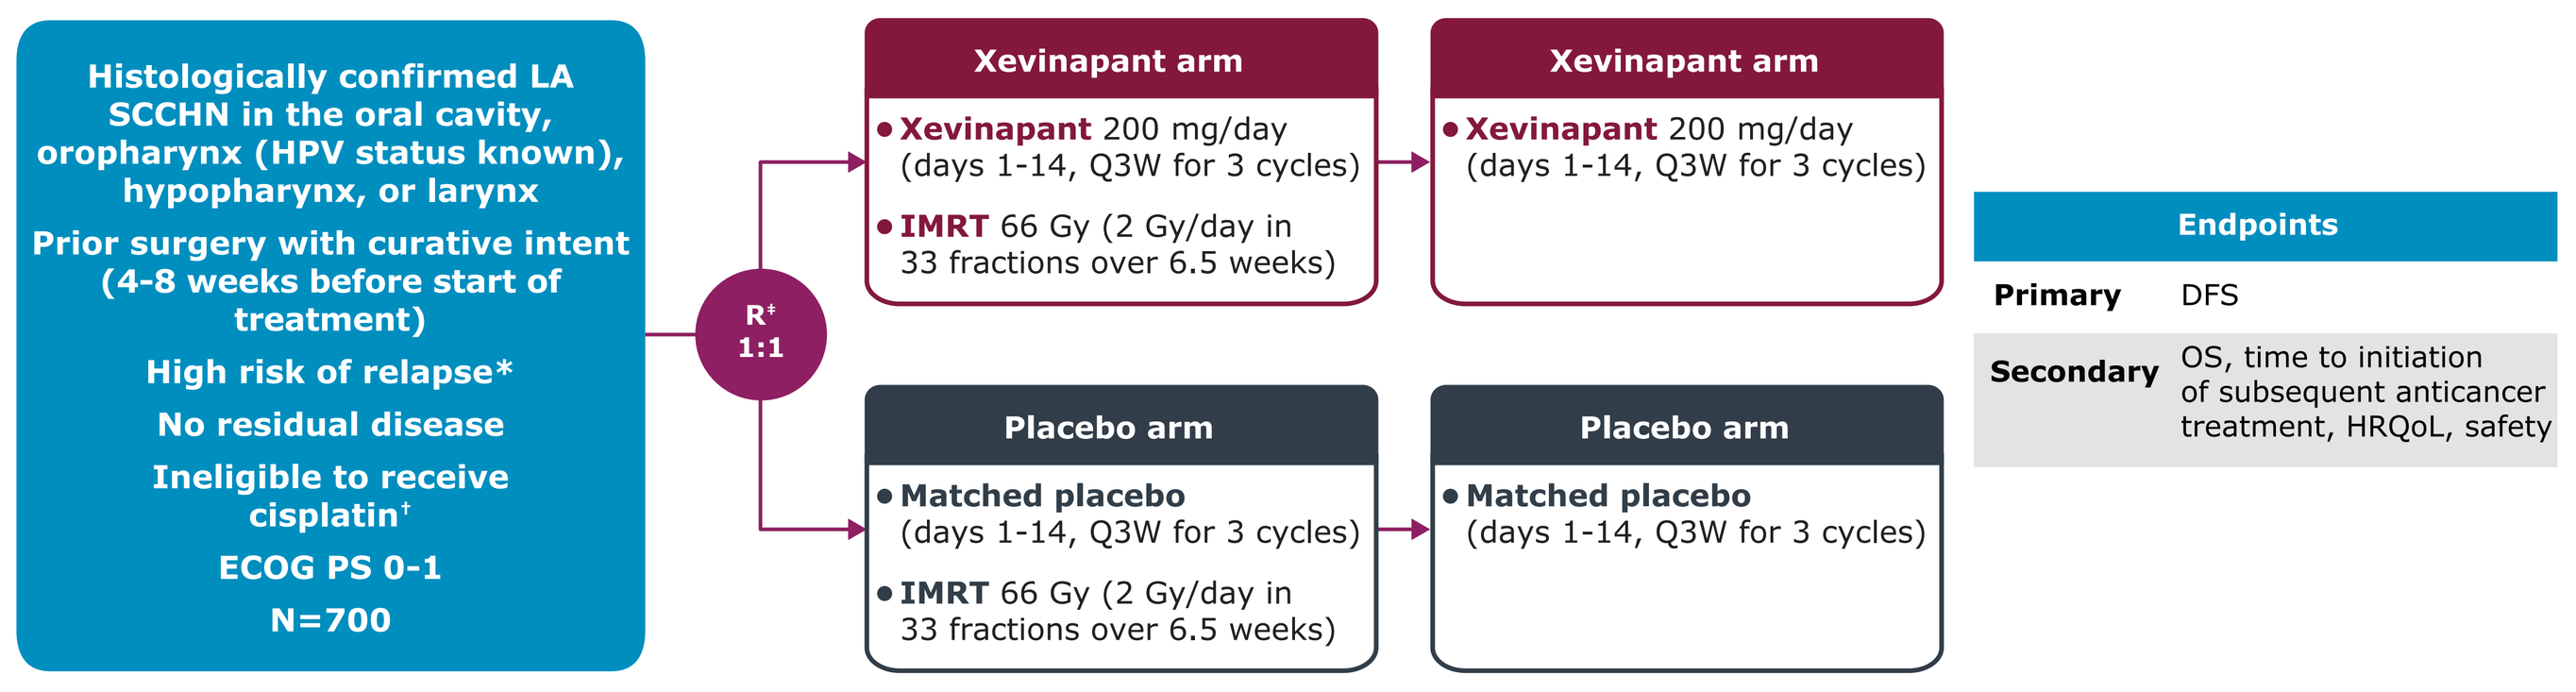
Histologically confirmed LA SCCHN in the oral cavity, oropharynx (HPV status known), hypopharynx, or larynx
Prior surgery with curative intent (4-8 weeks before start of treatment)
High risk of relapse*
No residual disease
Ineligible to receive cisplatin†
ECOG PS 0-1
N=700
R‡ 1:1
Xevinapant arm
Xevinapant 200 mg/day (days 1-14, Q3W for 3 cycles)
IMRT 66 Gy (2 Gy/day in 33 fractions over 6.5 weeks)
Xevinapant arm
Xevinapant 200 mg/day (days 1-14, Q3W for 3 cycles)
Placebo arm
Matched placebo (days 1-14, Q3W for 3 cycles)
IMRT 66 Gy (2 Gy/day in 33 fractions over 6.5 weeks)
Placebo arm
Matched placebo (days 1-14, Q3W for 3 cycles)
Endpoints
Primary DFS
Secondary OS, time to initiation of subsequent anticancer treatment, HRQoL, safety

*Meeting 1 or 2 of the following criteria, confirmed by local histopathology: nodal extracapsular extension or positive resection margins (R1 or close margin ≤1 mm). †Meeting ≥1 of the following criteria: estimated glomerular filtration rate <60 mL/min/1.73 m²; hearing loss (grade ≥2 audiometric hearing loss or grade ≥2 tinnitus); grade ≥2 peripheral neuropathy; and, if aged ≥70 years, unfit according to the G8 questionnaire (score ≤14). ‡Stratification factors: primary tumor site, oropharynx/oral cavity vs larynx vs hypopharynx; tumor stage, III vs IV; HPV p16 status for oropharynx primary tumor site, oropharynx p16 positive vs oropharynx p16 negative or larynx/hypopharynx/oral cavity.
DFS, disease-free survival; **ECOG PS,** Eastern Cooperative Oncology Group performance status; **HPV,** human papillomavirus; **HRQoL,** health-related quality of life; **IMRT,** intensity-modulated radiotherapy; **LA SCCHN,** locally advanced squamous cell carcinoma of the head and neck; **OS,** overall survival; **Q3W**, every 3 weeks.

Table 1. Endpoints

Primary
• DFS, defined as the time from randomization to objective disease recurrence or death from any cause
Secondary
• OS, defined as time from randomization to death from any cause
• Time to initiation of subsequent cancer treatment, defined as time from randomization to the start of the first subsequent cancer treatment
• HRQoL, assessed using the EORTC QLQ-H&N35 and QLQ-C30 questionnaires and the EuroQoL EQ-5D-5L VAS score
• Safety

DFS, disease-free survival; **EORTC**, European Organisation for Research and Treatment of Cancer; **HRQoL,** health-related quality of life; **OS,** overall survival.

Table 2. Summary of eligibility criteria

Key inclusion criteria
• Age ≥18 years with histologically confirmed LA SCCHN in ≥1 of the following sites: oral cavity, oropharynx, hypopharynx, or larynx
• Completed surgery with curative intent 4-8 weeks before the start of treatment
• High risk of relapse (meeting 1 or 2 of the following criteria, confirmed by local histopathology: nodal extracapsular extension or positive resection margins [R1 or close margin of ≤1 mm])
• Deemed ineligible to receive cisplatin (meeting ≥1 of the following criteria: eGFR <60 mL/min/1.73 m²; hearing loss [grade ≥2 audiometric hearing loss or grade ≥2 tinnitus*]; grade ≥2 peripheral neuropathy; and if aged ≥70 years, unfit according to G8 questionnaire [score ≤14])
• ECOG PS 0-1
• No residual disease by CT/MRI
• For patients with oropharynx tumors only, primary tumors must be HPV negative or, for heavy smokers (>25 pack-years), primary tumors can be HPV positive; HPV status must be determined by p16 expression using IHC
• Adequate renal, hematologic, and hepatic function
Key exclusion criteria
• Incomplete surgery
• Primary tumor site unknown or in the nasopharyngeal sinuses, paranasal sinuses, nasal cavity, salivary gland, thyroid gland, parathyroid gland, or skin
• Metastatic disease
• Prior definitive, neoadjuvant, concurrent, or adjuvant RT to the head and neck region that may jeopardize the primary tumor irradiation plan, or any other prior SCCHN systemic treatment

*In France, grade ≥3 audiometric hearing loss or grade ≥3 tinnitus.
CT, computed tomography; **ECOG PS,** Eastern Cooperative Oncology Group performance status; **eGFR,** estimated glomerular filtration rate; **HPV,** human papilloma virus; **IHC,** immunohistochemistry; **LA SCCHN**, locally advanced squamous cell carcinoma of the head and neck; **MRI,** magnetic resonance imaging; **RT,** radiotherapy.

Assessments

- Clinical outcomes will be evaluated using a combination of clinical, radiological, and, if appropriate, fiberoptic endoscopy and pathological assessments
- Imaging and clinical tumor assessments will be performed at the end of treatment (20 weeks), at 9 and 12 months in the first year, every 4 months in years 2 and 3, and every 6 months thereafter
- Safety will be assessed at each visit and graded based on National Cancer Institute Common Terminology Criteria for Adverse Events v5
 - Treatment-related adverse events will be monitored until the end-of-therapy visit
- Patient-reported health-related quality of life (HRQoL) will be assessed by EuroQoL EQ-5D-5L visual analogue scale scores and the European Organisation for Research and Treatment of Cancer QLQ-H&N35 and QLQ-C30 questionnaires

Statistical analysis

- The primary hypothesis is that xevinapant will prolong DFS in patients with resected LA SCCHN who have a high risk of relapse and are deemed ineligible for cisplatin
- All randomized patients will be included in efficacy and HRQoL analyses
- Safety will be evaluated in all patients who received ≥1 dose of study treatment
- The sequential testing procedure is DFS per investigator assessment, then DFS per independent review committee assessment, and then OS
 - DFS and OS between treatment arms will be compared using a 1-sided stratified log-rank test that preserves the type 1 error rate at 2.5%
- DFS, OS, and time to initiation of subsequent cancer treatment will be summarized using Kaplan-Meier estimates
- The primary analysis will occur when 100% of DFS events have occurred

Enrollment status

- The study is ongoing and is recruiting in patients worldwide

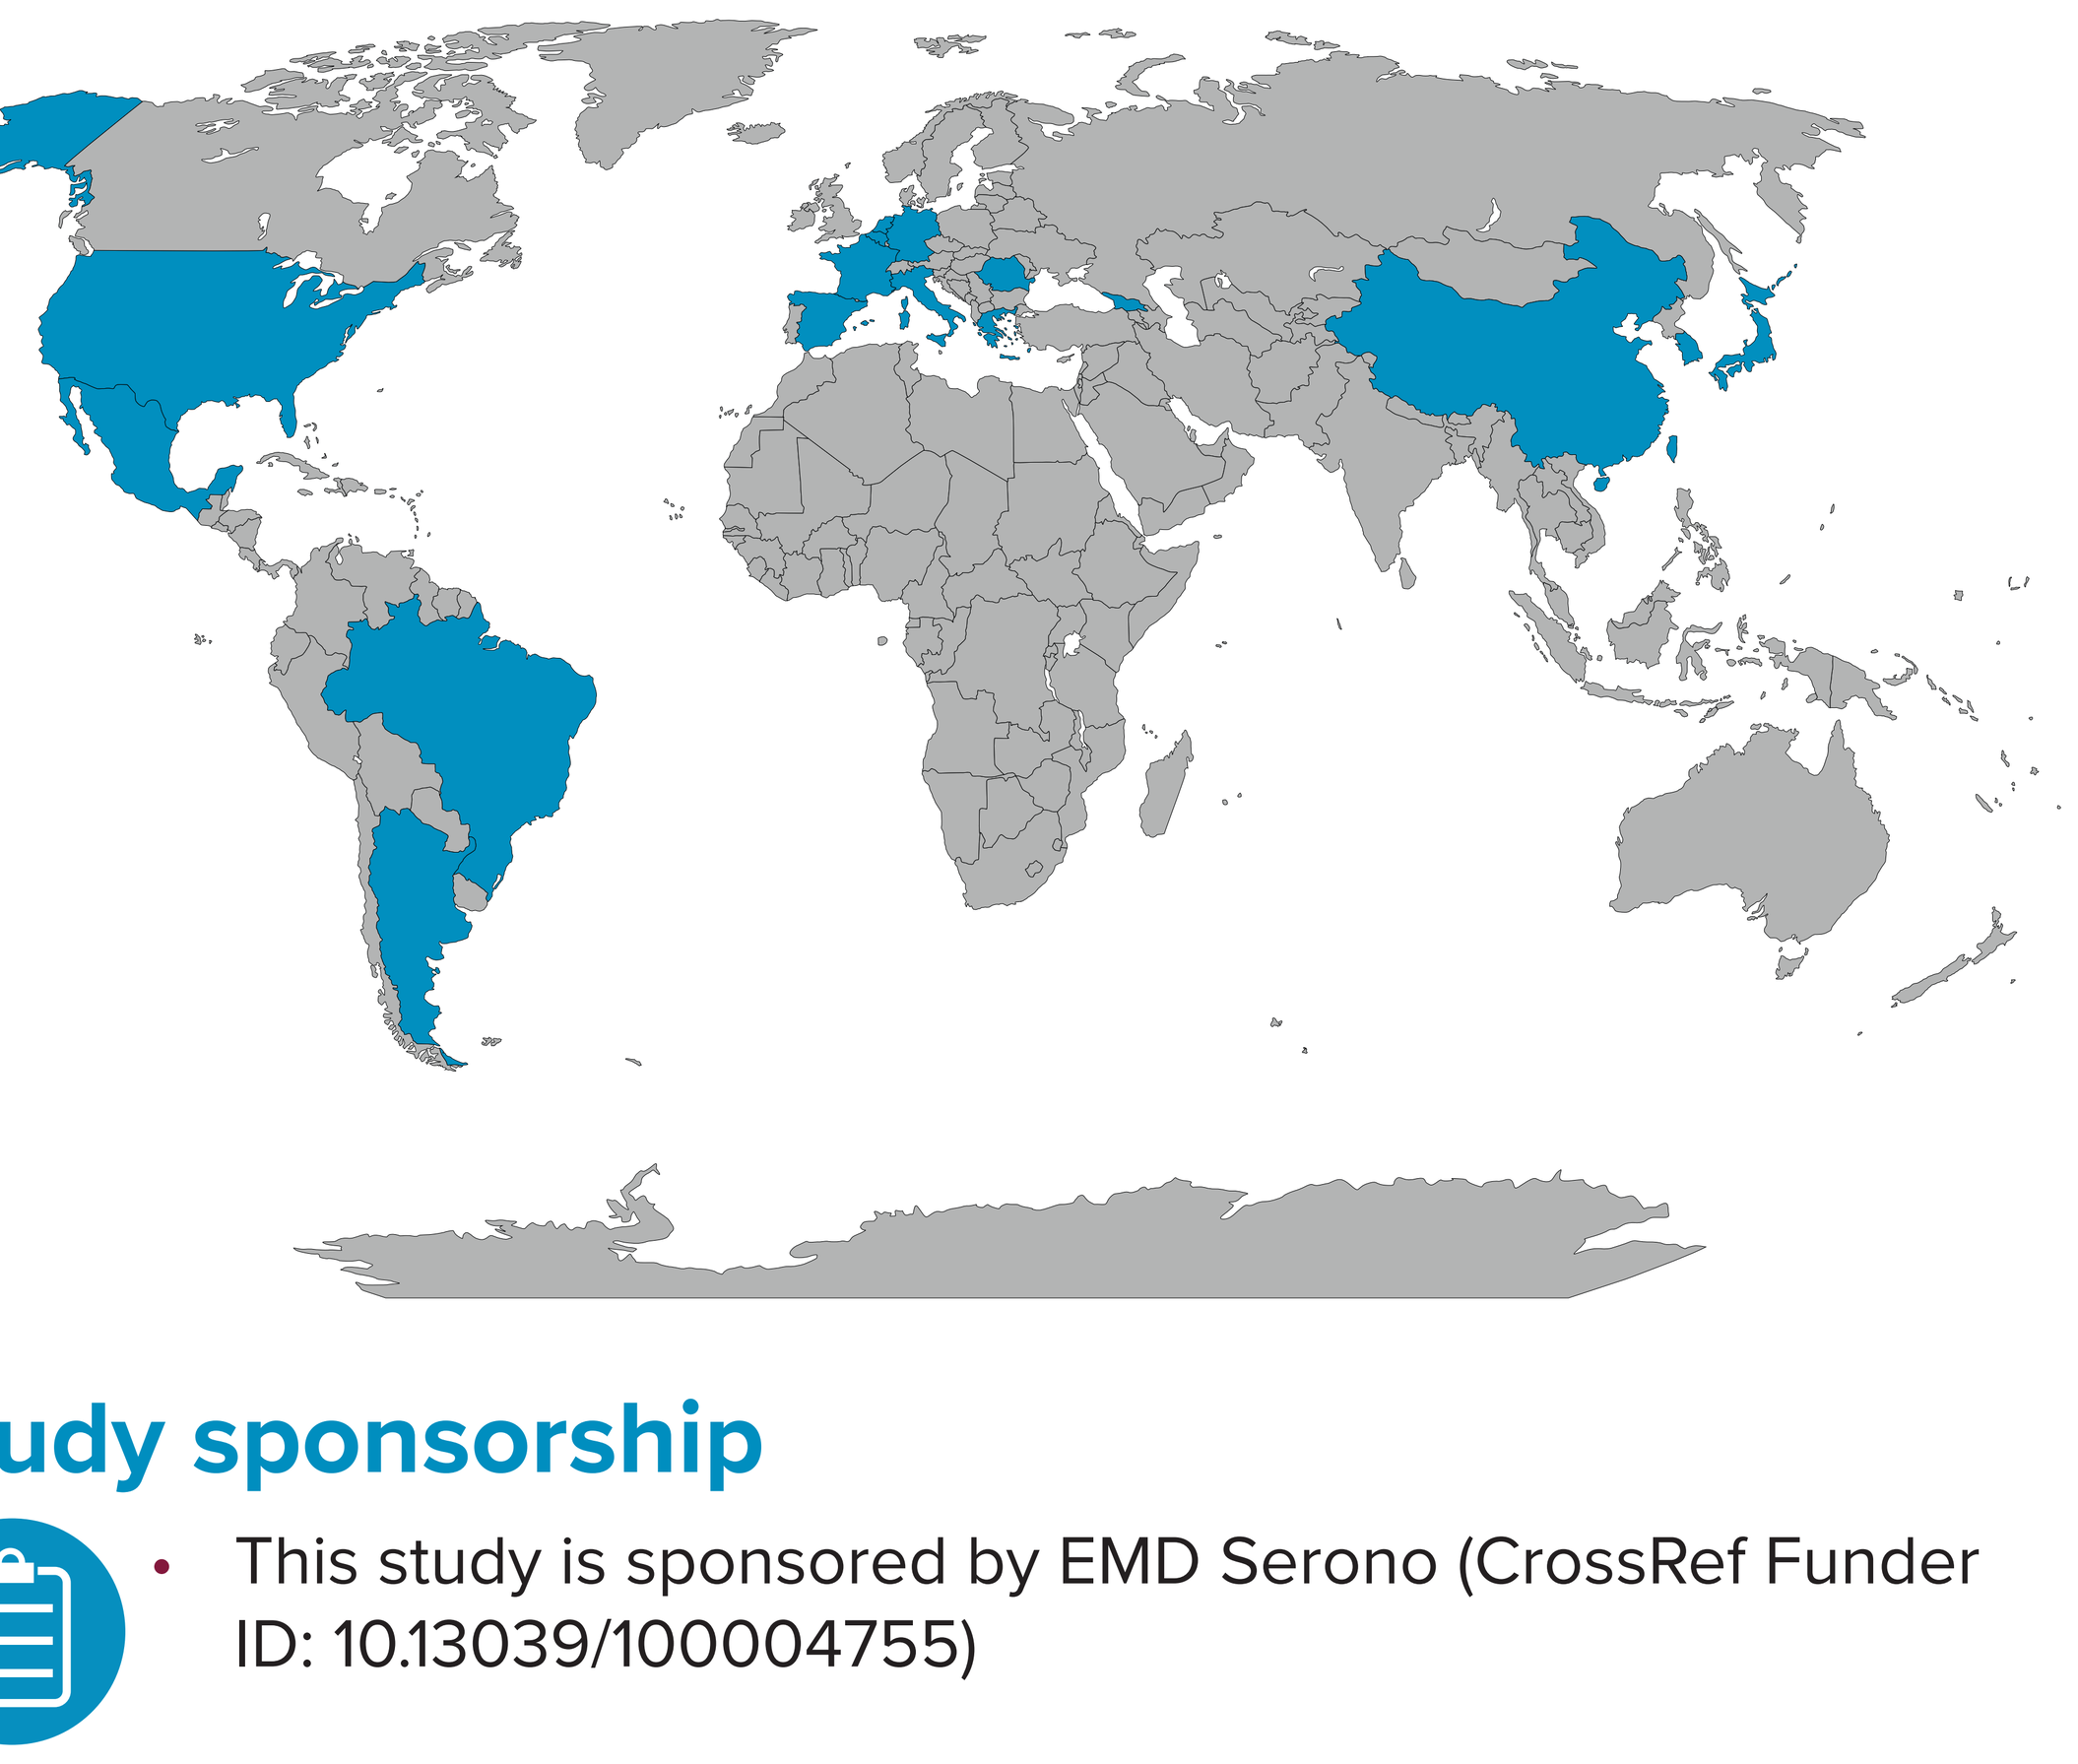

Study sponsorship

- This study is sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755)

References: **1.** Sung H, et al. *CA Cancer J Clin*. 2021;71:209-49. **2.** Psyrri A, et al. *Ann Oncol*. 2014;25:2101-15. **3.** NCCN Clinical Practice Guidelines in Oncology. Head and Neck Cancers. V1, 2023. **4.** Machiels JP, et al. *Ann Oncol*. 2020;31:1462-75. **5.** Porceddu SV, et al. *Front Oncol*. 2019;9:1522. **6.** Ahn MJ, et al. *Oral Oncol*. 2016;53:10-6. **7.** Matzinger O, et al. *Radiother Oncol*. 2015;116:495-503. **8.** Cai Q, et al. J Med Chem. 2011;54:2714-26. **9.** Thibault B, et al. Sci Rep. 2018;8:17862. **10.** Dougan SK, et al. *Immunotherapy*. 2018;10:787-96. **11.** Gomez-Roca C, et al. *Clin Transl Sci*. 2022;15:55-62. **12.** Yu H, et al. *Signal Transduct Target Ther*. 2020;5:209. **13.** Viertl D, et al. *Cancer Res*. 2013;73:Abstract 2055. **14.** Sun XS, et al. *Lancet Oncol*. 2020;21:1173-87. **15.** TAO Y, et al. Eur J Cancer. 2023;183:24-37. **Acknowledgments:** The authors would like to thank the patients and their families, the investigators, the co-investigators, and the study teams at each of the participating centers. This study was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755). Medical writing support was provided by Jamie Ratcliffe of Clinical Thinking and funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). **Disclosures: R. L. Ferris** reports receiving grant support and personal fees from Merck & Co., Kenilworth, NJ, Bristol Myers Squibb, Pfizer, GSK, the healthcare business of Merck KGaA, Darmstadt, Germany, Eisai, Bayer, AstraZeneca, Kura Oncology, the National Comprehensive Cancer Network, Nanobiotix, ISA Therapeutics, and Mirati; and consulting fees from Coherus BioSciences. **H. Mehanna** reports receiving speakers fees from Merck & Co., Kenilworth, NJ, the healthcare business of Merck KGaA, Darmstadt, Germany, and Sanofi; honoraria from AstraZeneca; and research funding from AstraZeneca, GSK, Merck & Co., Kenilworth, NJ, and Sanofi. **J. D. Schoenfeld** reports serving in consulting or advisory roles for ACI Clinical, Catenion, Debiopharm, Immunitas, LEK, Tilos Therapeutics, Astellas, and STIMIT; receiving research funding from BMS, the healthcare business of Merck KGaA, Darmstadt, Germany, and Regeneron; and providing expert testimony for Heidell, Pittoni, Murphy & Bach, Kline & Specter P.C. Equity, and Doximity. **M. Tahara** reports serving in consulting or advisory roles for Ono Pharmaceutical, Merck & Co., Kenilworth, NJ, Bristol Myers Squibb, Eisai, Pfizer, LOXO, Nektar Therapeutics, Lilly, GSK, Boehringer Ingelheim, AstraZeneca, and Rakuten Medical; and receiving grants from Bayer and Ono Pharmaceutical. **S. S. Yom** reports patents, royalties or intellectual property from UpToDate and Springer and research funding from Genentech, the healthcare business of Merck KGaA, Darmstadt, Germany, EMD Serono, BMS, and BioMimetix. **R. I. Haddad** reports grant support and personal fees from Merck & Co., Kenilworth, NJ, Bristol Myers Squibb, Pfizer, GSK, the healthcare business of Merck KGaA, Darmstadt, Germany, Eisai, Bayer, AstraZeneca, Kura Oncology, NCCN, Nanobiotix, ISA Therapeutics, and Mirati; and consulting fees from Coherus BioSciences. **A. Konig, S. Salmio,** and **M. Bajars** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **C. Le Tourneau** reports serving in consulting or advisory roles and has received honoraria from Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, the healthcare business of Merck KGaA, Darmstadt, Germany, AstraZeneca, Nanobiotix, GSK, Rakuten Medical, Seagen, ALX Oncology, Celgene, and Roche.

Poster No. TPS6101. Presented at the 2023 ASCO Annual Meeting, June 2-6, 2023; Chicago, IL, USA.

The XRay Vision study is looking at xevinapant plus radiotherapy in people with locally advanced squamous cell carcinoma of the head and neck who had surgery and are unable to receive a type of chemotherapy called cisplatin

Study number: NCT05386550 **Study start date:** October 2022

The full title of this abstract is: XRay Vision: A phase 3 study of xevinapant plus radiotherapy (RT) for high-risk, cisplatin-ineligible patients with resected, locally advanced squamous cell carcinoma of the head and neck (LA SCCHN)

Xevinapant is not yet approved to treat any condition, anywhere in the world.
This study is ongoing and is estimated to be completed by October 2027.
More information can be found in the scientific abstract of this study, which you can find here:
2023 American Society of Clinical Oncology Annual Meeting Scientific Abstract

Medical terms pronunciations

Xevinapant <zeh-VIN-uh-pant> **Squamous** <SKWAY-mus> **Carcinoma** <KAR-sih-NOH-muh>

WHAT IS SQUAMOUS CELL CARCINOMA OF THE HEAD AND NECK (SCCHN)?

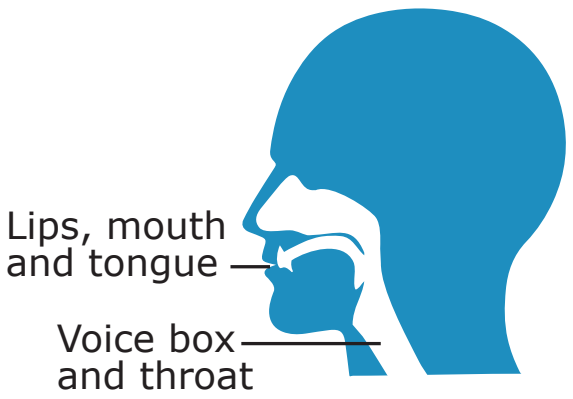
Lips, mouth and tongue
Voice box and throat

Head and neck cancer is the eighth most common type of cancer in the world. SCCHN is the most common form and includes cancers of the lips, mouth, throat, tongue, and voice box.

SCCHN is called “locally advanced” when the cancer has spread to nearby areas but not to other parts of the body. For some people, surgery to remove the cancer is the first recommended treatment, but the cancer may come back.

The standard treatment for locally advanced SCCHN following surgery is chemotherapy plus radiotherapy (chemoradiotherapy) when there is a high risk that the cancer could come back.

Chemotherapy and **radiotherapy** are two treatments used to kill the cancer or stop it from coming back after surgery. When given together, they are called **chemoradiotherapy**.

Many people are unable to receive the standard chemotherapy treatment—a medicine called cisplatin—due to old age, poor kidney function, or other medical conditions.

For people who have a high risk of their cancer coming back after surgery and who are unable to receive cisplatin, there are limited treatment options.

WHAT IS XEVINAPANT?

Xevinapant is being developed as a possible new type of cancer treatment. It is a liquid that is taken by mouth or given through a feeding tube.

Radiotherapy usually destroys or slows down cancer growth, but sometimes the treatment does not work. Adding xevinapant to the standard treatment aims to make the radiotherapy more effective against the cancer.

continued →

WHAT DID PREVIOUS STUDIES OF XEVINAPANT FIND?

In a previous study of 96 people with locally advanced SCCHN who had not had surgery, half of the people were randomly assigned to receive **xevinapant plus chemoradiotherapy**. The other half of the people were randomly assigned to receive **placebo plus chemoradiotherapy**. This placebo was a liquid that looked and tasted the same as xevinapant but did not contain any medicine.

The study found that people who were treated with **xevinapant plus chemoradiotherapy** lived longer on average than people treated with **placebo plus chemoradiotherapy**.

Xevinapant plus radiotherapy has shown promising results in early laboratory studies.

These results suggest that **xevinapant plus radiotherapy** could potentially help improve the lives of people with locally advanced SCCHN who have a high risk of their cancer coming back after surgery and who are unable to receive cisplatin.

WHAT IS THE XRAY VISION STUDY?

Researchers have started a large international study called XRay Vision. In this study, researchers will compare **xevinapant plus radiotherapy** versus **placebo plus radiotherapy** in people with locally advanced SCCHN who have a high risk of their cancer coming back after surgery and who are unable to receive cisplatin.

People will be randomly assigned to receive either **xevinapant plus radiotherapy** or **placebo plus radiotherapy** for 6.5 weeks. After that, they will receive either xevinapant or placebo alone. Xevinapant and placebo will be given for 18 weeks overall.

The study will show if adding xevinapant to radiotherapy can help stop the cancer from coming back after surgery and help people live longer.

WHO WILL TAKE PART IN THE XRAY VISION STUDY?

About 700 people will take part in this study

People who take part in the study:

- Aged 18 years or older
- Have locally advanced SCCHN and had surgery to cure their cancer 4-8 weeks before the start of treatment
- Have a high risk of their cancer coming back after surgery
- Are unable to receive cisplatin
- Are either fully active or are unable to do physically hard work but can do light physical work

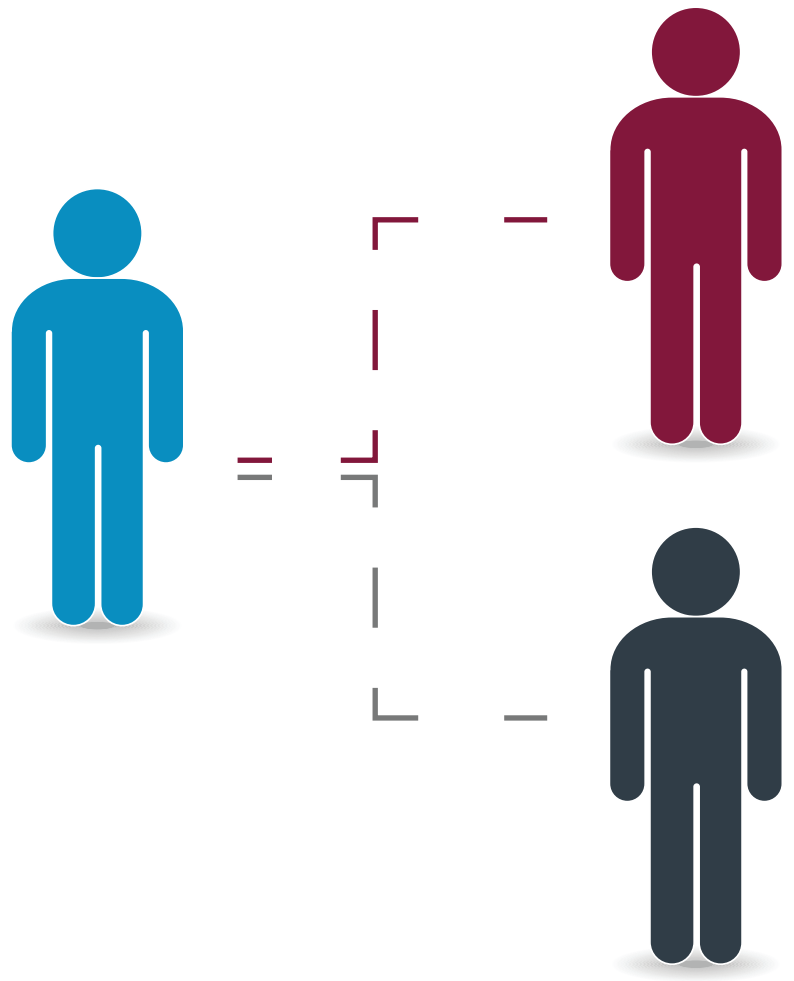

350 people will be treated with **xevinapant plus radiotherapy**

350 people will be treated with **placebo plus radiotherapy**

WHO IS SPONSORING THIS STUDY?

EMD Serono (CrossRef Funder ID: 10.13039/100004755): **http://www.emdgroup.com**

FURTHER INFORMATION

For more information on this study, please visit:
2023 American Society of Clinical Oncology Annual Meeting Scientific Abstract
https://clinicaltrials.gov/ct2/show/NCT05386550

For more information on clinical studies in general, please visit:
https://www.clinicaltrials.gov/ct2/about-studies/learn
https://www.cancer.org/cancer/managing-cancer/making-treatment-decisions/clinical-trials.html

Writing support for this summary was provided by Jamie Ratcliffe of Clinical Thinking and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.