First-line avelumab treatment in patients with metastatic Merkel cell carcinoma: 4-year follow-up from part B of the JAVELIN Merkel 200 trial

S. P. D'Angelo,[1] C. Lebbé,[2] L. Mortier,[3] A. S. Brohl,[4] N. Fazio,[5] J-J. Grob,[6] N. Prinzi,[7] G. J. Hanna,[8] J. C. Hassel,[9] F. Kiecker,[10] A. von Heydebreck,[11] G. Güzel,[12] P. Nghiem[13]

[1]Department of Medicine, Memorial Sloan Kettering Cancer Center; and Department of Medicine, Weill Cornell Medical College, New York, NY, USA; [2]Université Paris Cite, Dermato-Oncology, CIC AP-HP Hôpital Saint Louis, F-75010 Paris, France; INSERM U976; [3]Dermatology Clinic, CARADERM; and University of Lille, INSERM U1189, Lille Hospital-Claude Huriez Hospital, Lille Cedex, France; [4]Sarcoma Department and Cutaneous Oncology, Moffitt Cancer Center, Tampa, FL, USA; [5]Division of Gastrointestinal Medical Oncology and Neuroendocrine Tumors, European Institute of Oncology, IEO, IRCCS, Milan, Italy; [6]AP-HM Hospital, Aix-Marseille University, Marseille, France; [7]Medical Oncology Unit, Fondazione IRCCS Istituto Nazionale dei Tumori, Milano, Italy; [8]Head and Neck Oncology, Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, USA; [9]Department of Dermatology and National Center for Tumor Diseases, University Hospital Heidelberg, Heidelberg, Germany; [10]Charité Universitätsmedizin Berlin, Campus Charité Mitte, Berlin, Germany; [11]Global Biostatistics, the healthcare business of Merck KGaA, Darmstadt, Germany; [12]Global Clinical Development, the healthcare business of Merck KGaA, Darmstadt, Germany; [13]Division of Dermatology, Department of Medicine, University of Washington Medical Center at South Lake Union, Seattle, WA, USA

SCOPE

- We report long-term overall survival (OS) data after 4 years of follow-up from part B of the JAVELIN Merkel 200 study, which evaluated first-line (1L) avelumab monotherapy in a cohort of patients with metastatic Merkel cell carcinoma (mMCC)

CONCLUSIONS

- 1L avelumab monotherapy in patients with mMCC resulted in a 4-year OS rate of 38%
 - Long-term follow-up continues to show favorable OS with 1L avelumab compared with previous studies of 1L chemotherapy[1-4]
- Responses to avelumab occurred irrespective of PD-L1 status; although a numerical trend for longer OS was observed in patients with PD-L1+ vs PD-L1− tumors, data do not support patient selection based on PD-L1 status
- After discontinuing avelumab, some patients received subsequent chemotherapy, immunotherapy, or experimental agents
- These updated data further support the use of avelumab 1L monotherapy as standard-of-care treatment for patients with mMCC

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from SITC and the author of this poster

Correspondence: **Sandra P. D'Angelo**, dangelos@mskcc.org

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app to view a plain language summary of the accepted scientific abstract

BACKGROUND

- Prior to the approval of immune checkpoint inhibitors, patients with mMCC, a rare and aggressive neuroendocrine skin cancer, had a poor prognosis (5-year OS rate of 13.5-20%)[2,5-8]
- MCC is considered chemosensitive; however, responses to chemotherapy are generally not durable[5,7]
- In part A of the pivotal phase 2 JAVELIN Merkel 200 trial, treatment with avelumab (anti–PD-L1 antibody) resulted in meaningful long-term OS in patients with mMCC that had progressed after prior chemotherapy[9]
 - After >5 years of follow-up, median OS was 12.6 months, and the 5-year OS rate was 26%
- Part B of the JAVELIN Merkel 200 trial investigated avelumab as 1L treatment for mMCC[10]
 - In the primary analysis (reported after ≥15 months of follow-up), the objective response rate (ORR) was 39.7%, the durable (≥6 months) response rate was 30.2%, and median OS was 20.3 months
- Results from both parts of the JAVELIN Merkel 200 trial led to the approval of avelumab for the treatment of mMCC in multiple countries and its inclusion in treatment guidelines as a preferred or recommended treatment for disseminated or unresectable disease[11-13]
- Here, we report findings after 4 years of follow-up from part B of JAVELIN Merkel 200, including updated data for OS, patient disposition, and subsequent treatment

METHODS

- The design of the phase 2, prospective, single-arm, open-label JAVELIN Merkel 200 trial (NCT02155647) has been reported previously[10]
- Briefly, in part B, eligible patients were aged ≥18 years, had histologically confirmed stage IV mMCC, had not received prior systemic therapy for metastatic disease, and had an ECOG PS of 0-1
 - Patients were unselected for tumor PD-L1 expression and MCPyV (Merkel cell polyomavirus) status
- Patients received avelumab 10 mg/kg intravenous infusion every 2 weeks as monotherapy until confirmed disease progression, unacceptable toxicity, or withdrawal of consent
- OS was analyzed using the Kaplan-Meier method; 95% CIs for the median were calculated using the Brookmeyer-Crowley method
 - For illustrative purposes, OS in this population was compared with that in several recent retrospective real-world studies that reported outcomes in patients who received 1L chemotherapy[1-4]
- PD-L1+ status was defined as PD-L1 expression in ≥1% of tumor cells using the investigational PD-L1 IHC 73-10 pharmDx assay (Agilent Technologies, Carpinteria, CA)

RESULTS

Patient disposition

- 116 patients were enrolled and had received ≥1 dose of avelumab (**Table 1**)
- As of February 2, 2022 (data cutoff), median follow-up was 54.3 months (range, 48.0-69.7 months)
- Patient disposition is shown in **Table 2**

Overall survival

- Median OS was 20.3 months (95% CI, 12.4-42.0 months), unchanged from the primary analysis
 - OS rates (95% CIs) at 2, 3, and 4 years were 49.3% (39.8%-58.0%), 43.6% (34.3%-52.5%), and 37.7% (28.7%-46.7%), respectively
 - For illustrative purposes, OS data from this study are shown alongside OS data from previous real-world studies of patients with mMCC treated with 1L chemotherapy (**Figure 1**)[1-4]
- In patients evaluable for PD-L1 status (n=109), median OS in patients with PD-L1+ tumors (n=21) was 38.7 months (95% CI, 11.3 to not estimable [NE]) and in patients with PD-L1− tumors (n=87) was 16.1 months (95% CI, 9.6-42.0 months) (**Figure 2**)

Table 1. Baseline characteristics[10]

Characteristics	N=116
Age	
<65 years, n (%)	22 (19.0)
≥65 years, n (%)	94 (81.0)
Median age (range), years	74.0 (41-93)
Sex, n (%)	
Male	81 (69.8)
Female	35 (30.2)
ECOG PS, n (%)	
0	72 (62.1)
1	44 (37.9)
Geographic region, n (%)	
North America	29 (25.0)
Western Europe	75 (64.7)
Australia	9 (7.8)
Asia	3 (2.6)
Median time since initial diagnosis (range), months	10.6 (0.7-120.9)
Median time since diagnosis of metastatic disease (range), months	2.2 (0.4-49.6)
Site of primary tumor, n (%)	
Skin	104 (89.7)
Lymph node	1 (0.9)
Not reported	11 (9.5)
Visceral metastases at baseline, n (%)	
Present	79 (68.1)
Absent	35 (30.2)
Not reported	2 (1.7)
PD-L1 status, n (%)	
Positive	21 (18.1)
Negative	87 (75.0)
Not reported	8 (6.9)
MCPyV status, n (%)	
Positive	70 (60.3)
Negative	37 (31.9)
Not reported	9 (7.8)

MCPyV, Merkel cell polyomavirus.

Figure 1. OS with 1L avelumab in all patients compared with historical real-world chemotherapy data

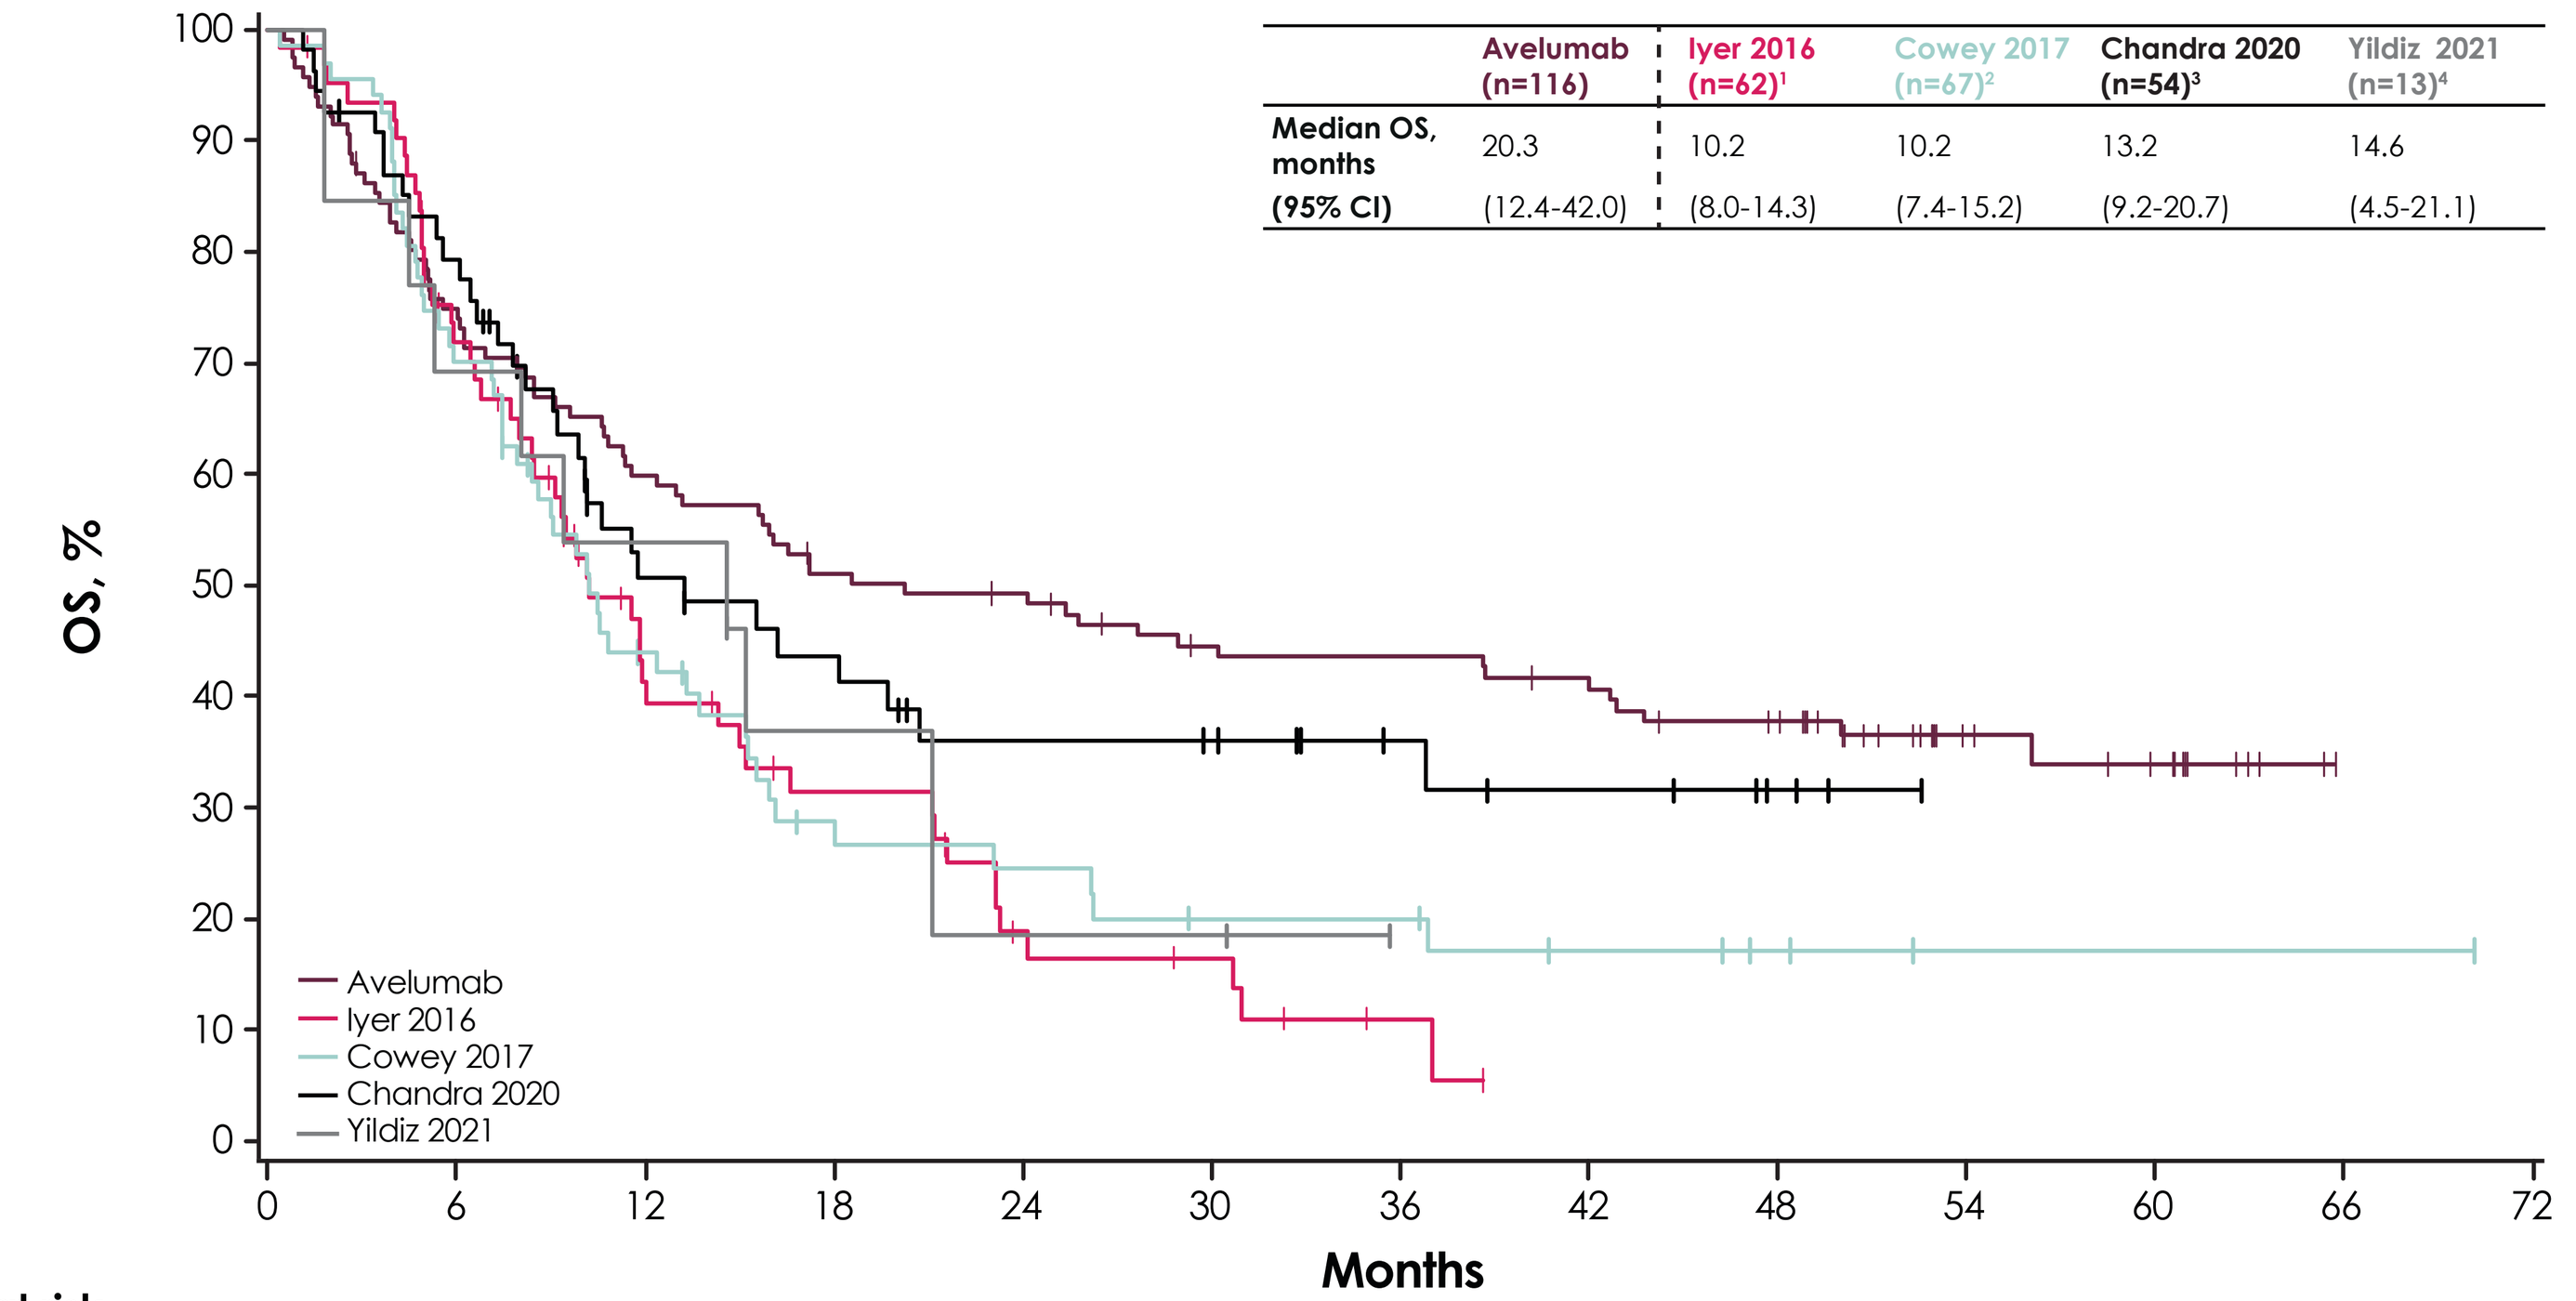
OS, %
Months
Avelumab
Iyer 2016
Cowey 2017
Chandra 2020
Yildiz 2021

	Avelumab (n=116)	Iyer 2016 (n=62)[1]	Cowey 2017 (n=67)[2]	Chandra 2020 (n=54)[3]	Yildiz 2021 (n=13)[4]
Median OS, months	20.3	10.2	10.2	13.2	14.6
(95% CI)	(12.4-42.0)	(8.0-14.3)	(7.4-15.2)	(9.2-20.7)	(4.5-21.1)

No. at risk																			
Avelumab	116	100	85	76	68	65	57	55	54	49	46	45	45	42	36	18	11	0	0
Iyer 2016	62	57	42	33	22	18	15	15	8	7	6	3	2	0	0	0	0	0	0
Cowey 2017	67	64	46	36	24	20	14	13	11	9	8	8	8	5	3	1	1	1	0
Chandra 2020	54	49	42	33	23	20	18	13	13	13	12	9	8	6	3	0	0	0	0
Yildiz 2021	13	11	9	8	7	5	4	4	2	2	2	1	0	0	0	0	0	0	0

OS, overall survival.
This figure is for illustrative purposes only and is not a head-to-head comparison. 1L chemotherapy regimens in the study by Cowey et al were carboplatin + etoposide (n=44), cisplatin + etoposide (n=11), topotecan (n=6), and other regimens (n=6)[2]; data for chemotherapy regimens were not reported in the other studies shown.[1,3,4]

Figure 2. OS with 1L avelumab in subgroups defined by PD-L1 status

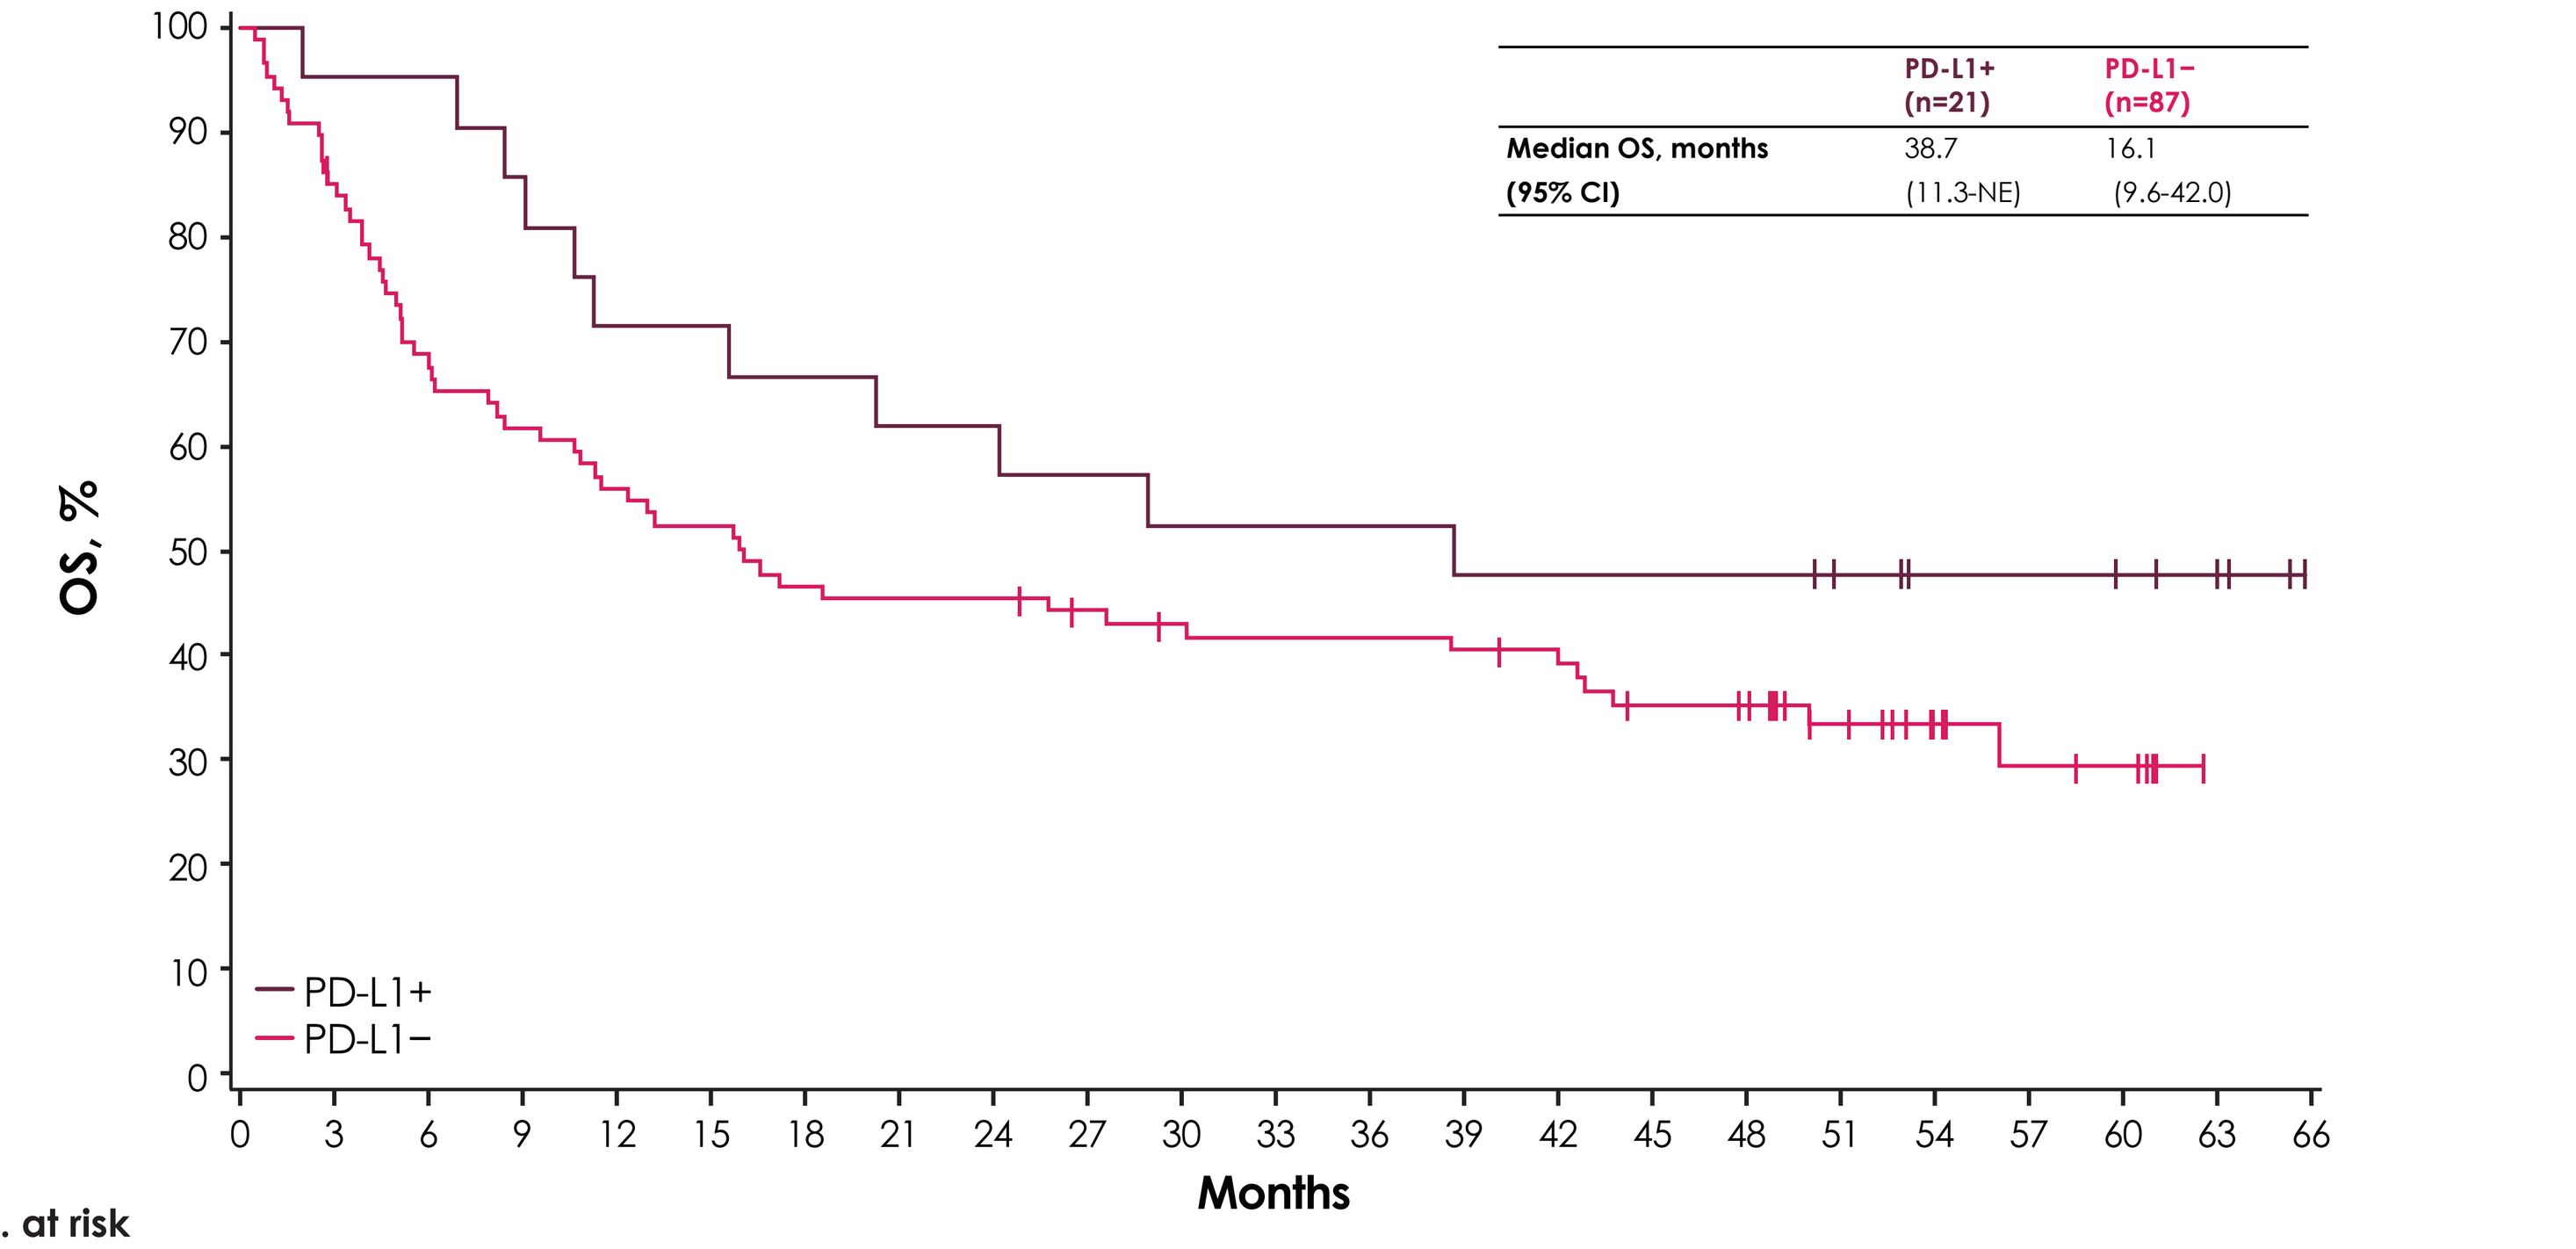
OS, %
Months
PD-L1+
PD-L1−

	PD-L1+ (n=21)	PD-L1− (n=87)
Median OS, months	38.7	16.1
(95% CI)	(11.3-NE)	(9.6-42.0)

No. at risk																							
PD-L1+	21	20	20	18	15	15	14	13	13	12	11	11	11	10	10	10	10	8	6	6	5	4	0
PD-L1−	87	73	59	53	48	45	40	39	39	36	34	33	33	32	31	26	25	18	12	7	6	0	0

OS, overall survival; **NE**, not estimable.

Table 2. Patient disposition

	N=116
Received ≥1 dose of avelumab, n (%)	116 (100)
Study treatment ongoing	7 (6.0)
Discontinued study treatment	109 (94.0)
Reason for discontinuation of study treatment, n (%)	
Disease progression	54 (46.6)
Adverse event	27 (23.3)
Withdrawal of consent	6 (5.2)
Death	5 (4.3)
Loss to follow-up	1 (0.9)
Other	16 (13.8)
Discontinued study treatment but remained in follow-up, n (%)	22 (19.0)
Discontinued from the trial, n (%)	87 (75.0)
Reason for discontinuation from the trial, n (%)	
Death	71 (61.2)
Loss to follow-up	4 (3.4)
Withdrawal of consent	4 (3.4)
Other	8 (6.9)
Re-initiated study treatment with avelumab, n (%)	2 (1.7)

Causes of death

- At data cutoff, 72 patients (62.1%) had died
 - The most common cause of death was disease progression (n=57 [49.1%])
 - Other causes were unknown reason (n=7 [6.0%]), adverse event (AE) not related to study treatment (n=3 [2.6%]), and other reason (n=5 [4.3%])
 - No deaths due to treatment-related AEs were reported

Subsequent treatment

- 48 patients (41.4%) received ≥1 subsequent anticancer drug therapy after discontinuing study treatment with avelumab (**Table 3**)
- The most common subsequent therapies (any line) were etoposide (20 [17.2%]), carboplatin (18 [15.5%]), and avelumab (14 [12.1%]; all 14 patients received single-agent avelumab outside of a clinical trial)
- In 14 patients who received further avelumab after discontinuing from the study:
 - Reasons for discontinuation from the study were progressive disease (PD) in 6 patients, AE in 3, and other reasons in 5 (physician and/or patient decision in 3, complete metabolic remission in 1, and not specified in 1)
 - Response status when avelumab was discontinued (per investigator assessment) was complete response (CR) in 1 patient, partial response (PR) in 5, PD in 6, and NE in 2 patients
 - Best response to post-study avelumab (per investigator assessment) was CR in 5 patients, SD in 1, PD in 2, and not reported in 6 patients
- Responses to post-study avelumab based on responses when patients discontinued from the study were as follows:
 - In 6 patients who had CR or PR when avelumab was discontinued, best response to subsequent avelumab was CR in 3, SD in 1, and unknown in 2 (none of the 6 patients received any other therapy prior to post-study avelumab)
 - In 6 patients who had PD when avelumab was discontinued, best response to subsequent avelumab was CR in 1, PD in 2, and unknown in 3 (5/6 patients received ≥1 different drug therapy and/or radiotherapy prior to subsequent avelumab)
 - In 2 patients who had response NE when avelumab was discontinued, best response to subsequent avelumab was CR in 1 and unknown in 1 (neither patient received any other therapy prior to post-study avelumab)

Table 3. Subsequent treatment (in any line)

Subsequent treatment, n (%)	n=48
Etoposide	20 (17.2)
Carboplatin	18 (15.5)
Avelumab	14 (12.1)
Pembrolizumab	9 (7.8)
Paclitaxel	5 (4.3)
Ipilimumab	4 (3.4)
Cisplatin	3 (2.6)
Carboplatin + etoposide	2 (1.7)
Cisplatin + etoposide	2 (1.7)
Nivolumab	2 (1.7)
Temozolomide	2 (1.7)
AMG 232	1 (0.9)
Calcium folinate + fluorouracil + irinotecan hydrochloride	1 (0.9)
Capecitabine + temozolomide	1 (0.9)
Carboplatin + paclitaxel	1 (0.9)
Cetuximab	1 (0.9)
Combination of antineoplastic agents	1 (0.9)
Cyclophosphamide	1 (0.9)
Cyclophosphamide + doxorubicin + vincristine	1 (0.9)
Dacarbazine	1 (0.9)
Doxorubicin	1 (0.9)
Epirubicin	1 (0.9)
Fluorouracil + folinic acid	1 (0.9)
Fluorouracil + folinic acid + oxaliplatin	1 (0.9)
Olaparib	1 (0.9)
Pegylated liposomal doxorubicin hydrochloride	1 (0.9)
Sabatolimab	1 (0.9)

Some patients received >1 subsequent treatment and are subsequently included in more than one row of the table.

REFERENCES 1. Iyer JG, et al. Cancer Med. 2016;5:2294-301. **2.** Cowey C, et al. Future Oncol. 2017;13:1699-710. **3.** Chandra S, et al. Future Oncol. 2020;16:2521-536. **4.** Yildiz F, et al. J Cancer Res Ther. 2021;17:1525-529. **5.** Schadendorf D, et al. Eur J Cancer. 2017;71:53-69. **6.** Lebbe C, et al. Eur J Cancer. 2015;16:2396-403. **7.** Nghiem P, et al. Future Oncol. 2017;13:1263-79. **8.** Gauci ML, et al. Eur J Cancer. 2022;171:203-31. **9.** D'Angelo SP, et al. ESMO Open. 2021;6:1002905. **10.** D'Angelo S, et al. J Immunother Cancer. 2021;7:e002646. **11.** Bavencio (avelumab). Prescribing information. EMD Serono, Inc., Rockland, MA, USA, an affiliate of Merck KGaA, Darmstadt, Germany; 2022. **12.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA, Darmstadt, Germany; 2022. **13.** NCCN Clinical Practice Guidelines: Merkel cell carcinoma. V2. 2022. **DISCLOSURES SP D'Angelo** has served as a consultant or advisor for Amgen, the healthcare business of Merck KGaA, Darmstadt, Germany, GSK, Immune Design, Incyte, Merck & Co., Kenilworth, NJ, and Nektar; has received research grants from Amgen, Bristol Myers Squibb, Deciphera, the healthcare business of Merck KGaA, Darmstadt, Germany, Incyte, Merck & Co., Kenilworth, NJ, and Nektar; and received reimbursement for travel and accommodation expenses from Adaptimmune, the healthcare business of Merck KGaA, Darmstadt, Germany, and Nektar. **C Lebbé** has received honoraria from Amgen, Bristol Myers Squibb, Incyte, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Pierre Fabre, and Roche; has served as a consultant or advisor for Amgen, Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Novartis, and Roche; is a member of a speakers bureau for Amgen, Bristol Myers Squibb, Novartis, and Roche; has received research funding from Bristol Myers Squibb and Roche; has received reimbursement for travel and accommodation expenses from Bristol Myers Squibb; and has other relationships with Avantis Medical Systems. **L Mortier** has received reimbursement for travel and accommodation expenses from Bristol Myers Squibb, Novartis, and Roche/Genentech. **AS Brohl** has served as an advisor or consultant for Deciphera and Bayer. **N Fazio** has served as an invited speaker for Advanced Accelerator Applications and Ipsen; has served as an advisor or consultant for Advanced Accelerator Applications, Hutchinson MediPharma, the healthcare business of Merck KGaA, Darmstadt, Germany, Merck & Co., Kenilworth, NJ, Novartis, and Pfizer; is the local principle investigator of trials being conducted by 4SC, Astellas, BeiGene, FibroGen, Incyte, Ipsen, Nucana; has received institutional research grants from Advanced Accelerator Applications, Ipsen, Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has nonfinancial interests in ENETS (executive committee member), ESMO (GI and NET faculty member), and SPARC Europe (steering committee member). **J-J Grob** has received honoraria from Amgen, Bristol Myers Squibb, the healthcare business of Merck KGaA, Darmstadt, Germany, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Pierre Fabre, Roche, and Sanofi; has served as a consultant or advisor for Amgen, Bristol Myers Squibb, the healthcare business of Merck KGaA, Darmstadt, Germany, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Pierre Fabre, Roche, and Sanofi; is a member of a speakers bureau for Novartis; and has received reimbursement for travel and accommodation expenses from Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Novartis, and Pierre Fabre. **N Prinzi** has received reimbursement for travel and accommodation expenses from Novartis. **GJ Hanna** has received research funding from Bristol Myers Squibb, Exicure, GSK, Kite Pharma, NantKwest/Altor BioScience, Regeneron Pharmaceuticals, Sanofi Genzyme, and Kartos Therapeutics; and has received honoraria and served as an advisor or consultant for Bio-Rad Laboratories, Bristol Myers Squibb, Kura Oncology, Maverick Therapeutics, the healthcare business of Merck KGaA, Darmstadt, Germany, Prelude Therapeutics, and Regeneron Pharmaceuticals/Sanofi. **JC Hassel** has received honoraria from Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, and Roche; has served as a consultant or advisor for Merck & Co. Kenilworth, NJ, and Pierre Fabre; has received research funding from 4SC, Amgen, BioNTech, Bristol Myers Squibb, Immunocore, Novartis, Philogen, and Roche; and has received reimbursement for travel and accommodation expenses from Pierre Fabre. **F Kiecker** has received honoraria from Amgen, Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Novartis, Pierre Fabre, and Roche; has served as a consultant or advisor for Amgen, Bristol Myers Squibb, Incyte, Merck & Co., Kenilworth, NJ, Novartis, and Roche; has received research funding from Novartis; and has received reimbursement for travel and accommodation expenses from Bristol Myers Squibb and Novartis. **A von Heydebreck** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **G Güzel** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **P Nghiem** has served as a consultant or advisor for Almirall, Instil Bio, the healthcare business of Merck KGaA, Darmstadt, Germany, Pfizer, and Rain Therapeutics. **ACKNOWLEDGMENTS** This trial was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) as part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer. Medical writing support was provided by Eleanor Bishop of Clinical Thinking and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer.

Abstract No. 604. Society for Immunotherapy of Cancer's (SITC) 37th Annual Meeting & Pre-Conference Programs (SITC 2022), November 8-12, 2022; Boston, MA, USA.

Please note that this summary only contains information from the full scientific abstract

Long-term results from the JAVELIN Merkel 200 clinical trial in people with metastatic Merkel cell carcinoma who were treated with avelumab as their first treatment

JAVELIN

View scientific abstract

The full title of this abstract is: First-line avelumab treatment in patients with metastatic Merkel cell carcinoma: 4-year follow-up from the JAVELIN Merkel 200 trial

Please note this summary only contains information from the scientific abstract

View scientific abstract

Date of summary: November 2022

For more information on this study, go to: https://clinicaltrials.gov/ct2/show/NCT02155647

Pronunciations

Medical terms pronunciations

Avelumab <a-VEL-yoo-mab> **Carcinoma** <kar-sih-NO-mu> **Metastatic** <meh-tuh-STA-tik>

Key takeaways

What are the key takeaways from this summary?

- In the JAVELIN Merkel 200 study, researchers looked at people with metastatic Merkel cell carcinoma who received avelumab as their first treatment
- Long-term results showed that 38% of people who took part in the study were still alive after 4 years
- Survival rates seemed to be better than those seen in earlier studies where chemotherapy was given as the first treatment
- Results from this study provide more support for using avelumab as the first treatment for people with metastatic Merkel cell carcinoma

Background

What did this study look at?

What is metastatic Merkel cell carcinoma?

- Merkel cell carcinoma (also known as MCC) is a rare skin cancer that grows quickly
- In many people who have MCC, the cancer spreads to other parts of the body (known as metastatic MCC) and is hard to treat. This affects how long people live

How is metastatic Merkel cell carcinoma usually treated?

- In the past, people with metastatic MCC were given chemotherapy as the first treatment for their disease. This is called first-line treatment
- Although the cancer may get better with chemotherapy at first, it is likely to start growing again

What is avelumab?

Avelumab is a type of immunotherapy. Immunotherapy can help the body's immune system find and destroy cancer cells. Avelumab is given as a drip (infusion) into a vein for about an hour once every 2 weeks

Avelumab attaches to a protein called PD-L1. This protein can be found on the surface of cancer cells and other cells in the cancer. Some people have more PD-L1 on these cells, and this is known as PD-L1–positive cancer. When avelumab attaches to PD-L1, it helps the body's immune cells find and destroy cancer cells

Avelumab has been studied as a potential treatment for people with metastatic MCC. Researchers found that first-line avelumab treatment can help people with metastatic MCC live longer than people who received first-line chemotherapy in previous studies

Avelumab is now a recommended first-line treatment for people with metastatic MCC

What is the JAVELIN Merkel 200 study?

- The JAVELIN Merkel 200 study looked at avelumab treatment for people with metastatic MCC
- In one part of the trial, avelumab was given as first-line treatment
- The first results of the study were reported after people had been studied for 15 months
- Researchers found that avelumab treatment made MCC tumors shrink or disappear in about 40% of people with metastatic MCC. Researchers also found that half of all people treated in the study lived for at least 20 months
- These results led to avelumab being approved in many countries worldwide as a first-line treatment for people with metastatic MCC

What did the researchers want to find out?

Researchers wanted to look at long-term results from the JAVELIN Merkel 200 clinical study in people treated with first-line avelumab for at least 4 years. They wanted to find out how many people were still being treated and how long people lived.

Study design

What happened during the study?

Who took part in the study?

116 people from around the world with metastatic MCC who received first-line avelumab

- Researchers looked at everyone who had received first-line avelumab in the JAVELIN Merkel 200 study
- Results for this study were collected in February 2022
- On average, people had been monitored for 54 months when results were collected

Aims of this summary

What did the researchers look at?

- Researchers looked at the following:
 - How many people were still receiving avelumab
 - Reasons why avelumab was stopped
 - How long people lived overall
 - How long people lived depending on whether they had a marker called PD-L1 on their cancer cells
 - What treatments people received after they stopped receiving avelumab in the study

Results

What were the results of the study?

How many people with metastatic MCC were still receiving avelumab?

7 people were still receiving avelumab

109 people had stopped receiving avelumab

What were the reasons for stopping avelumab?

54 people's cancer had gotten worse

27 people had side effects

6 people no longer wanted to take part in the study

5 people died during treatment

1 person stopped attending appointments

16 people stopped for other reasons

How many people with metastatic MCC were still alive?

36 people were still alive

72 people had died

Current information was unavailable for 8 people

How long did they live for?

49% of people were still alive after **2 years**

44% of people were still alive after **3 years**

38% of people were still alive after **4 years**

20 MONTHS — How long people lived, on average, including everyone who took part in the study

39 MONTHS — How long people lived, on average, who had PD-L1 on at least 1% of their cancer cells

16 MONTHS — How long people lived, on average, who did not have PD-L1 on at least 1% of their cancer cells

How many people with metastatic MCC received a second treatment after they left the study?

41% of people received another drug(s) for their cancer after avelumab

What treatments were most commonly received after leaving the study?

17% of people received etoposide, a type of chemotherapy

16% of people received carboplatin, a type of chemotherapy

14% of people received more avelumab, outside of the study

Conclusions

What were the main conclusions reported by the researchers?

- Results from the JAVELIN Merkel 200 study showed that after 4 years, 38% of people with metastatic MCC treated with first-line avelumab were still alive
- These results showed how well avelumab worked and support the use of avelumab as a recommended treatment for people with metastatic MCC

Disclaimers

Avelumab is approved to treat the condition that is discussed in this summary. This summary reports the results of a single study. The results of this study may differ from those of other studies. Health professionals should make treatment decisions based on all available evidence, not on the results of a single study. This study described is still ongoing; therefore, the final outcomes of this study may differ from the outcomes described in this summary.

Study sponsors

Who is sponsoring this study?

the healthcare business of Merck KGaA, Darmstadt, Germany
Frankfurter Strasse 250
Darmstadt, 64293, Germany
Phone (Germany): +49 6151 720

Pfizer
235 East 42nd Street
New York, NY 10017, USA
Phone (United States): +1 212-733-2323

The sponsors would like to thank all of the people who took part in this study

More information

Where can I find more information?

For more information on this study, please visit:
SITC 2022 Scientific Abstract
https://clinicaltrials.gov/ct2/show/NCT02155647

For more information on clinical studies in general, please visit:
https://www.clinicaltrials.gov/ct2/about-studies/learn
https://www.cancer.org/treatment/treatments-and-side-effects/clinical-trials.html

Writing support for this summary was provided by Eleanor Bishop of Clinical Thinking and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and Pfizer.