This is a reprint from the European Society for Medical Oncology Congress 2022 (ESMO 2022), which was originally presented in Paris, France on September 9-13, 2022; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

Clinical potential of circulating tumor DNA (ctDNA)–based molecular response (MR) and baseline blood-based tumor mutational burden (bTMB) for monitoring response to first-line (1L) chemoimmunotherapy in advanced squamous non-small cell lung cancer (sqNSCLC)

F. Ciardiello,[1]* Z. Feng,[2]* N. R. Smith,[3] Z. Andric,[4] S. A. Shell,[5] A. Yablonovitch,[6] G. Guezel,[7] J. Scheuenpflug[3]

*Co-first authors

[1]Division of Medical Oncology, The University of Campania Luigi Vanvitelli, Napoli, Italy; [2]Global Clinical Biomarkers and Companion Diagnostics, EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, an affiliate of Merck KGaA; [3]Global Clinical Biomarkers and Companion Diagnostics, Merck Healthcare KGaA, Darmstadt, Germany; [4]Medical Oncology, Clinical Hospital Center Bezanijska Kosa, Belgrade, Serbia; [5]Medical Affairs, Guardant Health, Redwood City, CA, USA; [6]Bioinformatics, Guardant Health, Redwood City, CA, USA; [7]Global Clinical Development, Merck Healthcare KGaA, Darmstadt, Germany

SCOPE

- To monitor and predict response during immunotherapy-related combination therapy and evaluate the clinical utility of ctDNA-based approaches, such as bTMB and MR, using longitudinal cohort samples from a phase 2a study of patients with treatment-naive advanced sqNSCLC treated with 1L avelumab and cetuximab in combination with platinum-based doublet chemotherapy (NCT03717155)

CONCLUSIONS

- Plasma bTMB-high (≥20 mutations per megabase [mut/Mb]) as a predictor of immunotherapy, in combination with putative predictive biomarkers, has the potential to identify patients with advanced sqNSCLC that could benefit from 1L avelumab and cetuximab in combination with cisplatin and gemcitabine
- Plasma ctDNA analysis supports MR assessment in patients treated with immunotherapy-related combination therapy, indicating its potential clinical utility as an adjunct to RECIST in monitoring tumor response

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster

Correspondence: Fortunato Ciardiello, fortunato.ciardiello@unicampania.it
Zheng Feng, zheng.feng@emdserono.com

BACKGROUND

- The combination of avelumab, cetuximab, cisplatin, and gemcitabine showed clinical activity and a manageable safety profile with no additional safety signals for avelumab or cetuximab in a phase 2a study in 1L metastatic sqNSCLC[1]
- The availability of biomarkers for response to immunotherapy-related combination therapy could potentially predict survival benefit with 1L treatment for advanced sqNSCLC
- The patient-centric liquid biopsy (LBx) approach to detect changes in ctDNA could provide an early indication of treatment response to therapies and is an emerging tool to aid clinicians in treatment decision-making for various tumor types[2]

METHODS

- 52 longitudinal plasma samples (covering baseline, day 85 post treatment, and end of treatment) were collected from 22 consenting patients with advanced sqNSCLC who had been treated with 1L avelumab and cetuximab in combination with cisplatin and gemcitabine for four 3-week cycles (84 days) followed by a maintenance schedule using avelumab and cetuximab (0-451 days)[1] (**Figure 1**)
- Radiographic assessments per RECIST 1.1 were obtained at each evaluation visit for patients who remained on therapy. Confirmed best overall response (BOR) per RECIST 1.1 was available for 18 patients
- Circulating free DNA (cfDNA) was extracted and tested using the GuardantOMNI[3,4] (2.145 Mb) LBx panel to detect somatic alterations in 497 genes and generate bTMB from baseline samples and MR scores from baseline and day 85 samples
- bTMB-high was defined as ≥20 mut/Mb, and MR scores were calculated using a modification to the validated Guardant360 Response algorithm[5] by including qualifying somatic alterations across the GuardantOMNI panel. Patients were evaluable for MR by having somatic alterations at each time point that exceeded the threshold required for reliable MR score calculation

Figure 1. Plasma sample collection scheme for ctDNA analysis

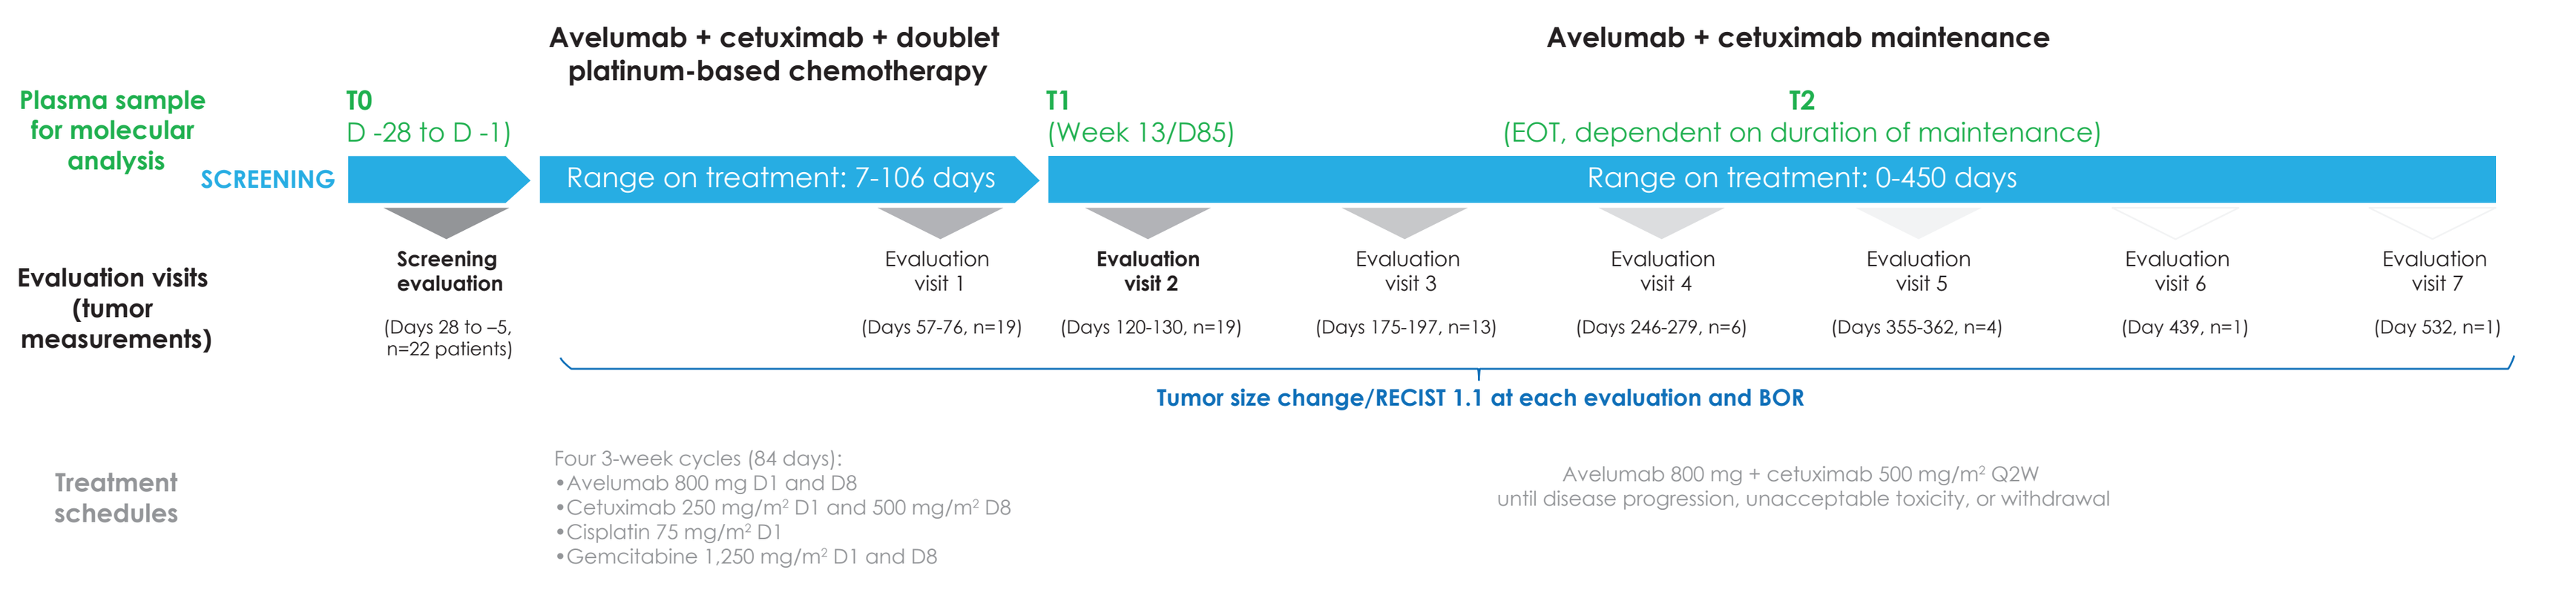
Avelumab + cetuximab + doublet platinum-based chemotherapy
Avelumab + cetuximab maintenance
Plasma sample for molecular analysis
SCREENING
T0
D -28 to D -1)
T1
(Week 13/D85)
T2
(EOT, dependent on duration of maintenance)
Range on treatment: 7-106 days
Range on treatment: 0-450 days
Evaluation visits (tumor measurements)
Screening evaluation
(Days 28 to –5, n=22 patients)
Evaluation visit 1
(Days 57-76, n=19)
Evaluation visit 2
(Days 120-130, n=19)
Evaluation visit 3
(Days 175-197, n=13)
Evaluation visit 4
(Days 246-279, n=6)
Evaluation visit 5
(Days 355-362, n=4)
Evaluation visit 6
(Day 439, n=1)
Evaluation visit 7
(Day 532, n=1)
Tumor size change/RECIST 1.1 at each evaluation and BOR
Treatment schedules
Four 3-week cycles (84 days):
•Avelumab 800 mg D1 and D8
•Cetuximab 250 mg/m² D1 and 500 mg/m² D8
•Cisplatin 75 mg/m² D1
•Gemcitabine 1,250 mg/m² D1 and D8
Avelumab 800 mg + cetuximab 500 mg/m² Q2W
until disease progression, unacceptable toxicity, or withdrawal

BOR, best overall response; **ctDNA,** circulating tumor DNA; **D,** day; **EOT,** end of therapy; **Q2W,** every 2 weeks; **T,** sample collection time point.

RESULTS

bTMB and predictive biomarkers of immunotherapy response (**Figure 2**)

- 5 of 18 baseline samples were defined as bTMB-high, and all bTMB-high patients had a BOR of partial response (PR; n=3) or stable disease (SD; n=2). No bTMB-high patients had a BOR of progressive disease (PD)
- 13 of 18 baseline samples were bTMB-low, of which 10 had a BOR of PR (n=2) or SD (n=8) and 3 had a BOR of PD

Genetic predictive biomarkers of immunotherapy response

- **Positive predictive biomarkers of response:** Mutation (*ARID1A*) was present in 7 of 18 baseline samples, all of which had a BOR of PR or SD. *ARID1A* mutations were present in 4 of 13 bTMB-low and 2 of 5 bTMB-high cases
- **Negative predictive biomarkers of response:** Mutations (*STK11, KEAP1, PTEN*) were present in 7 of 18 baseline samples, 5 of which had a BOR of PR or SD and 2 had non-PD
- Although the sample number is small, the presence or absence of positive response mutations was deemed potentially more informative in defining the response to immunotherapy-related combination therapy than negative response mutations

bTMB combined with positive predictive biomarkers of immunotherapy response

- Biomarker-positive patients were defined as those with bTMB-high and/or mutations in the positive predictive biomarkers of immunotherapy response, *ARID1A* mutations
 - 9 of 18 baseline samples were biomarker positive; all biomarker-positive patients had a BOR of PR or SD, and none had PD
 - The other 9 baseline samples were biomarker negative; 6 had a BOR of PR or SD, and 3 had a BOR of PD
- All 3 patients who had PD were bTMB-low, and 1 patient had a dual *STK11/KRAS* alteration, which was previously shown to negatively affect the clinical benefit of chemotherapy + immunotherapy[6]
- In this small data set, the presence or absence of mutations in positive predictive biomarkers combined with bTMB was deemed potentially more informative in defining the response to immunotherapy-related combination therapy than combining negative predictive biomarkers with bTMB

Figure 2. Predictive value of bTMB-high combined with predictive biomarkers of immunotherapy response

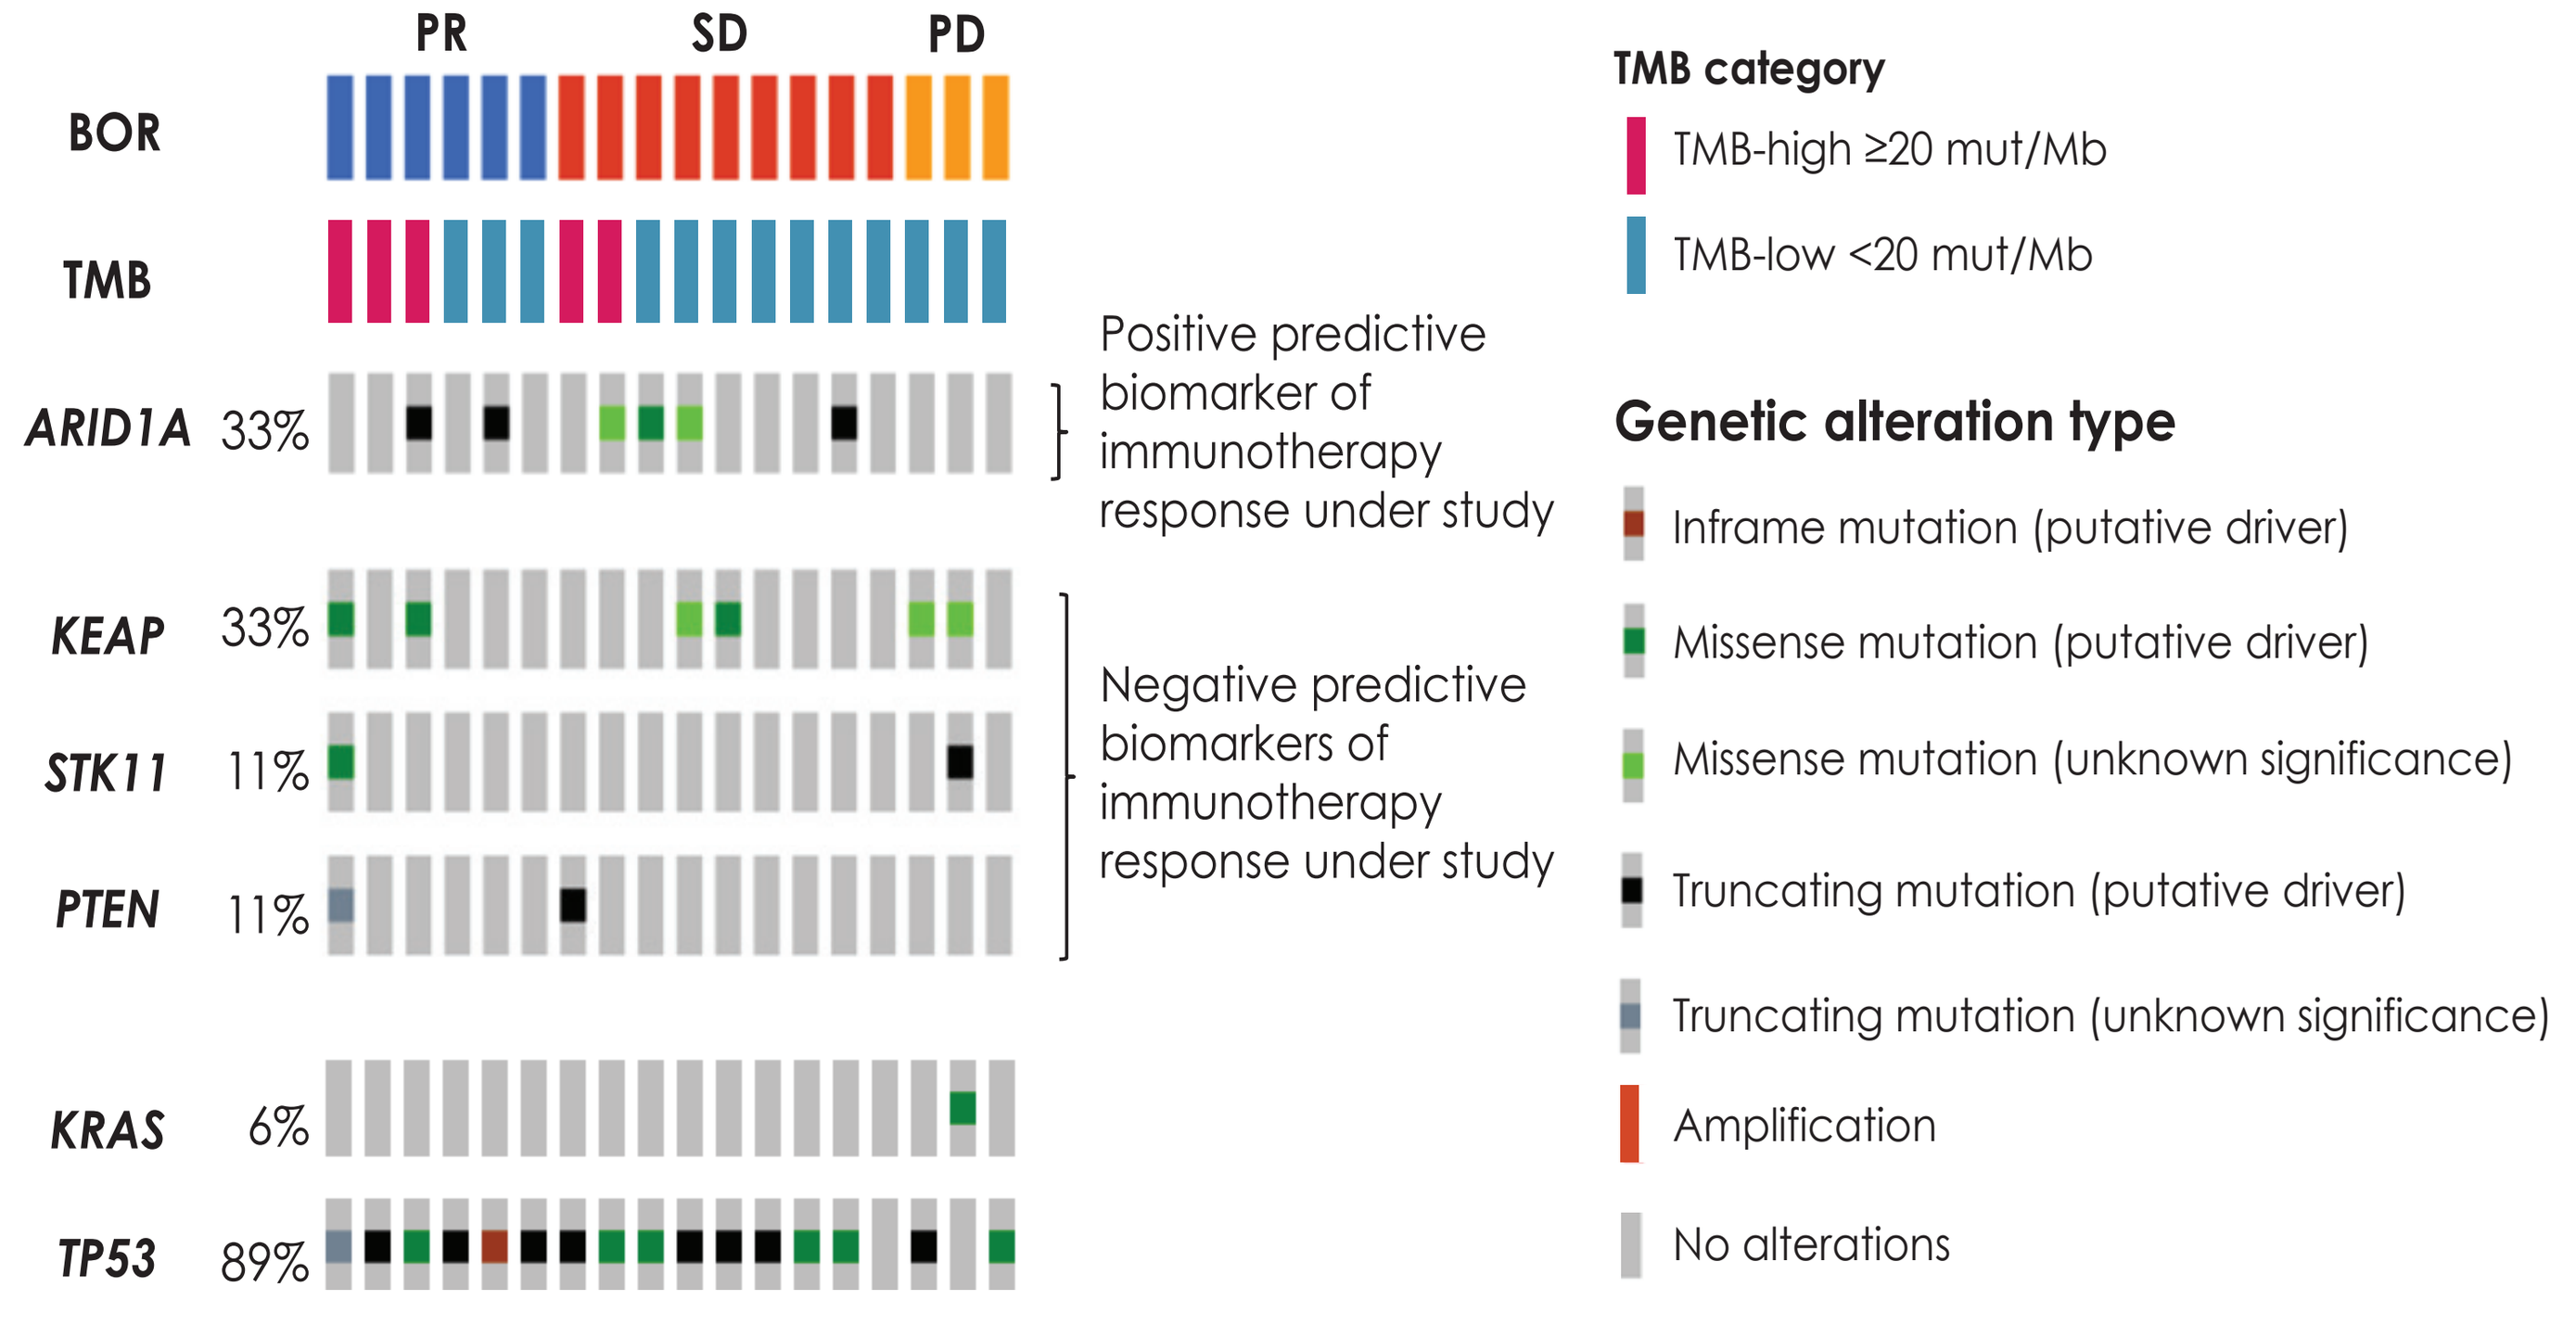
PR
SD
PD
BOR
TMB
ARID1A 33%
KEAP 33%
STK11 11%
PTEN 11%
KRAS 6%
TP53 89%
Positive predictive biomarker of immunotherapy response under study
Negative predictive biomarkers of immunotherapy response under study
TMB category
TMB-high ≥20 mut/Mb
TMB-low <20 mut/Mb
Genetic alteration type
Inframe mutation (putative driver)
Missense mutation (putative driver)
Missense mutation (unknown significance)
Truncating mutation (putative driver)
Truncating mutation (unknown significance)
Amplification
No alterations

BOR, best overall response; **bTMB,** blood-based tumor mutational burden; **mut/Mb,** mutation per megabase; **PD,** progressive disease; **PR,** partial response; **SD,** stable disease; **TMB,** tumor mutational burden.

Time on treatment with chemoimmunotherapy (**Figure 3**)

- For the 5 of 18 patients with bTMB-high, average time on treatment was 297 days (SD ± 148)
 - 3 patients had a mutation in *ARID1A, ARID1B,* or *ARID2*, 2 had *KEAP1* mutations, and 1 had a *PTEN* loss of function (LOF) mutation
- For the 13 patients with bTMB-low, average time on treatment was 226 days (SD ± 107)
 - 1 patient, who had a BOR of PR, had *STK11* LOF, *KEAP1* mutation, and *KRAS* activation alterations, which have been associated with diminished efficacy of immunotherapy in lung adenocarcinoma[6]

Figure 3. Time on treatment

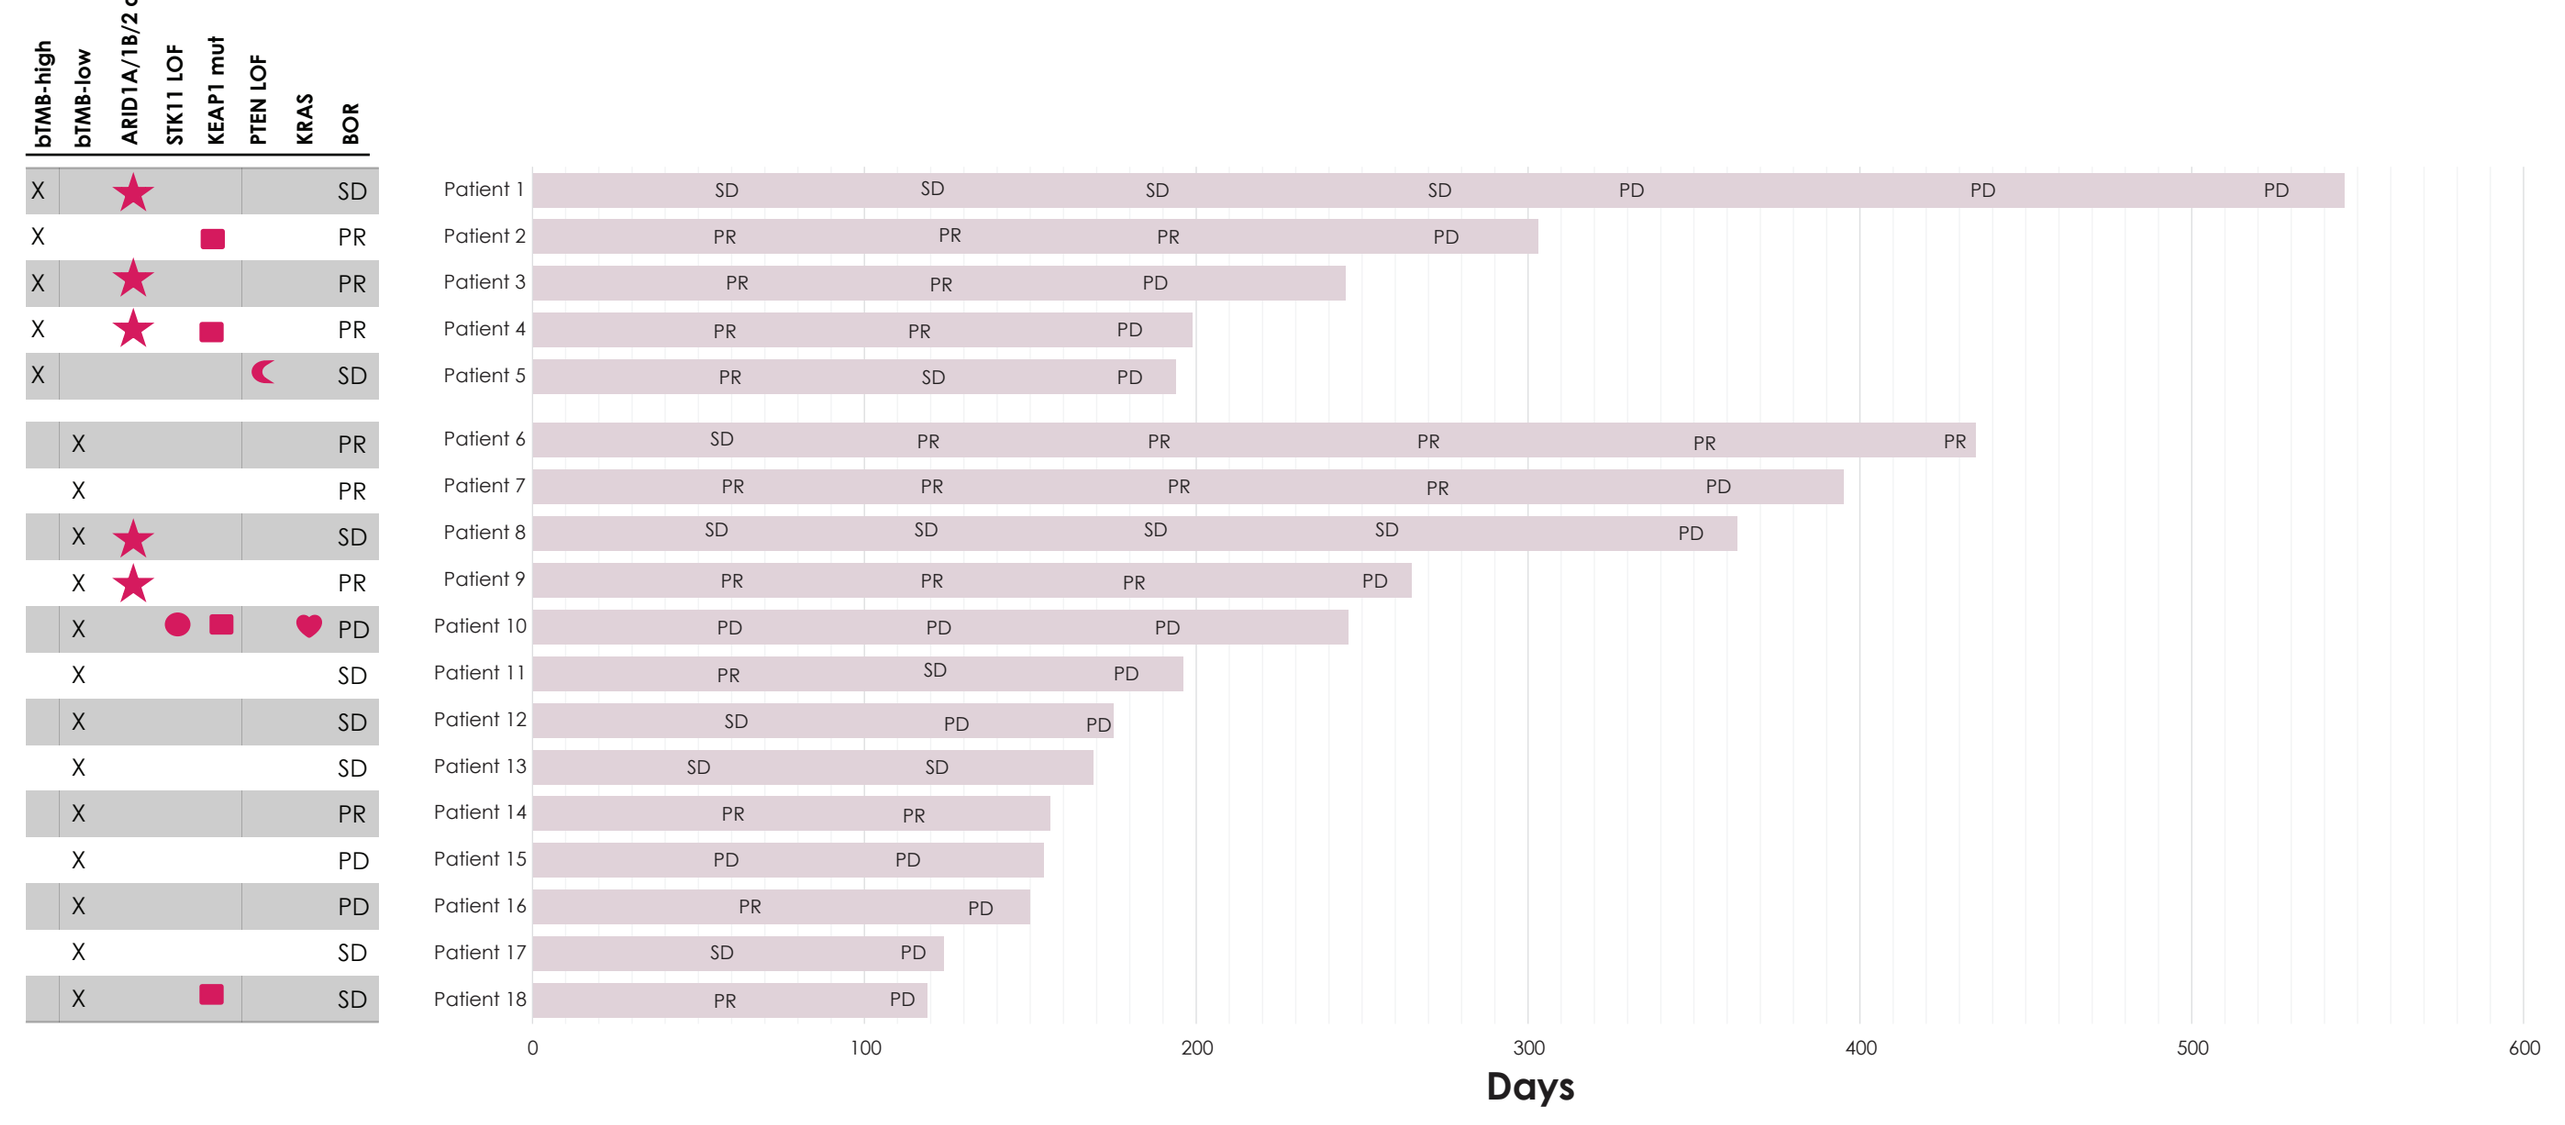
bTMB-high
bTMB-low
ARID1A/1B/2 del
STK11 LOF
KEAP1 mut
PTEN LOF
KRAS
BOR
Days

Not all patients had samples collected at each time point.
BOR, best overall response; **bTMB,** blood-based tumor mutational burden; **PD,** progressive disease; **PR,** partial response; **SD,** stable disease.

Longitudinal change of mean variant allele frequency (VAF)

- 21 of 22 patients were evaluable for MR by having somatic alterations at each time point that exceeded the threshold required for reliable MR score calculation (**Figure 4**)
- 1 patient with PR demonstrated mean VAF reduction from baseline to day 85 and EOT (**Figure 5, left panel**)
- 1 patient with PR demonstrated mean VAF reduction from baseline to day 85. Mean VAF increased at EOT (**Figure 5, right panel**)

Figure 4. Summary of mean VAF reductions from baseline for evaluable patients

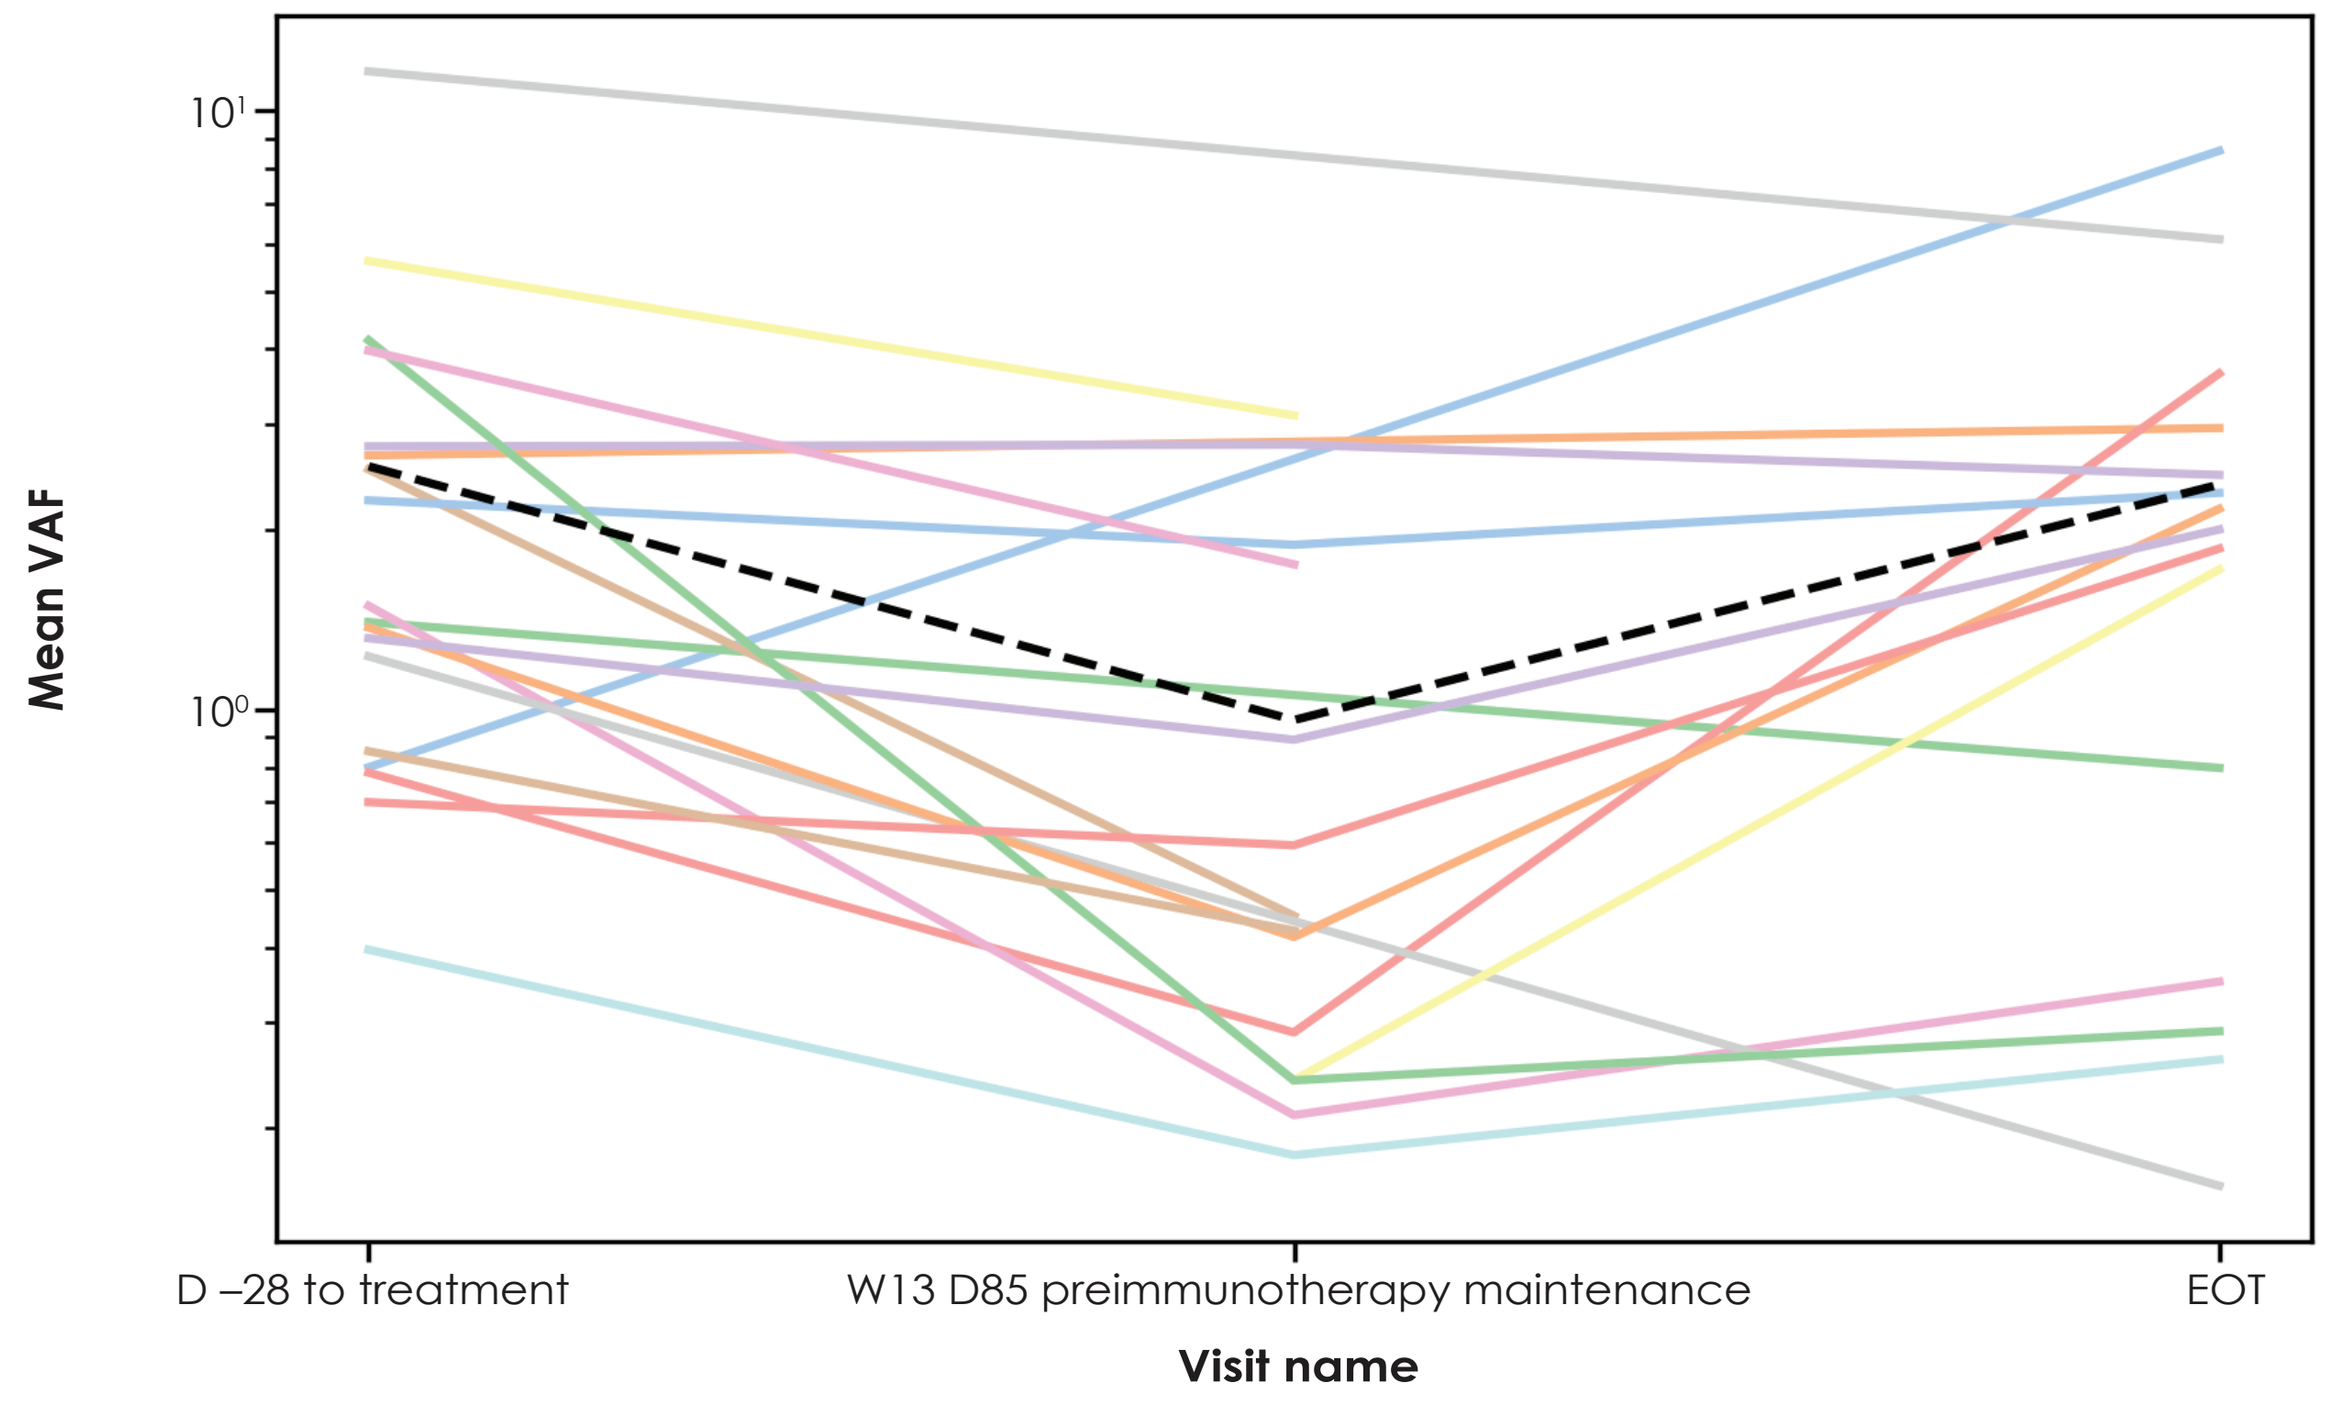
Mean VAF
Visit name
D –28 to treatment
W13 D85 preimmunotherapy maintenance
EOT

Dashed line represents the mean value of all mean VAFs measured at that particular time point across all patients.
D, day; **EOT,** end of treatment; **VAF,** variant allele frequency; **W,** week.

Figure 5. Longitudinal MAF plots for 2 patients who were bTMB-high at baseline

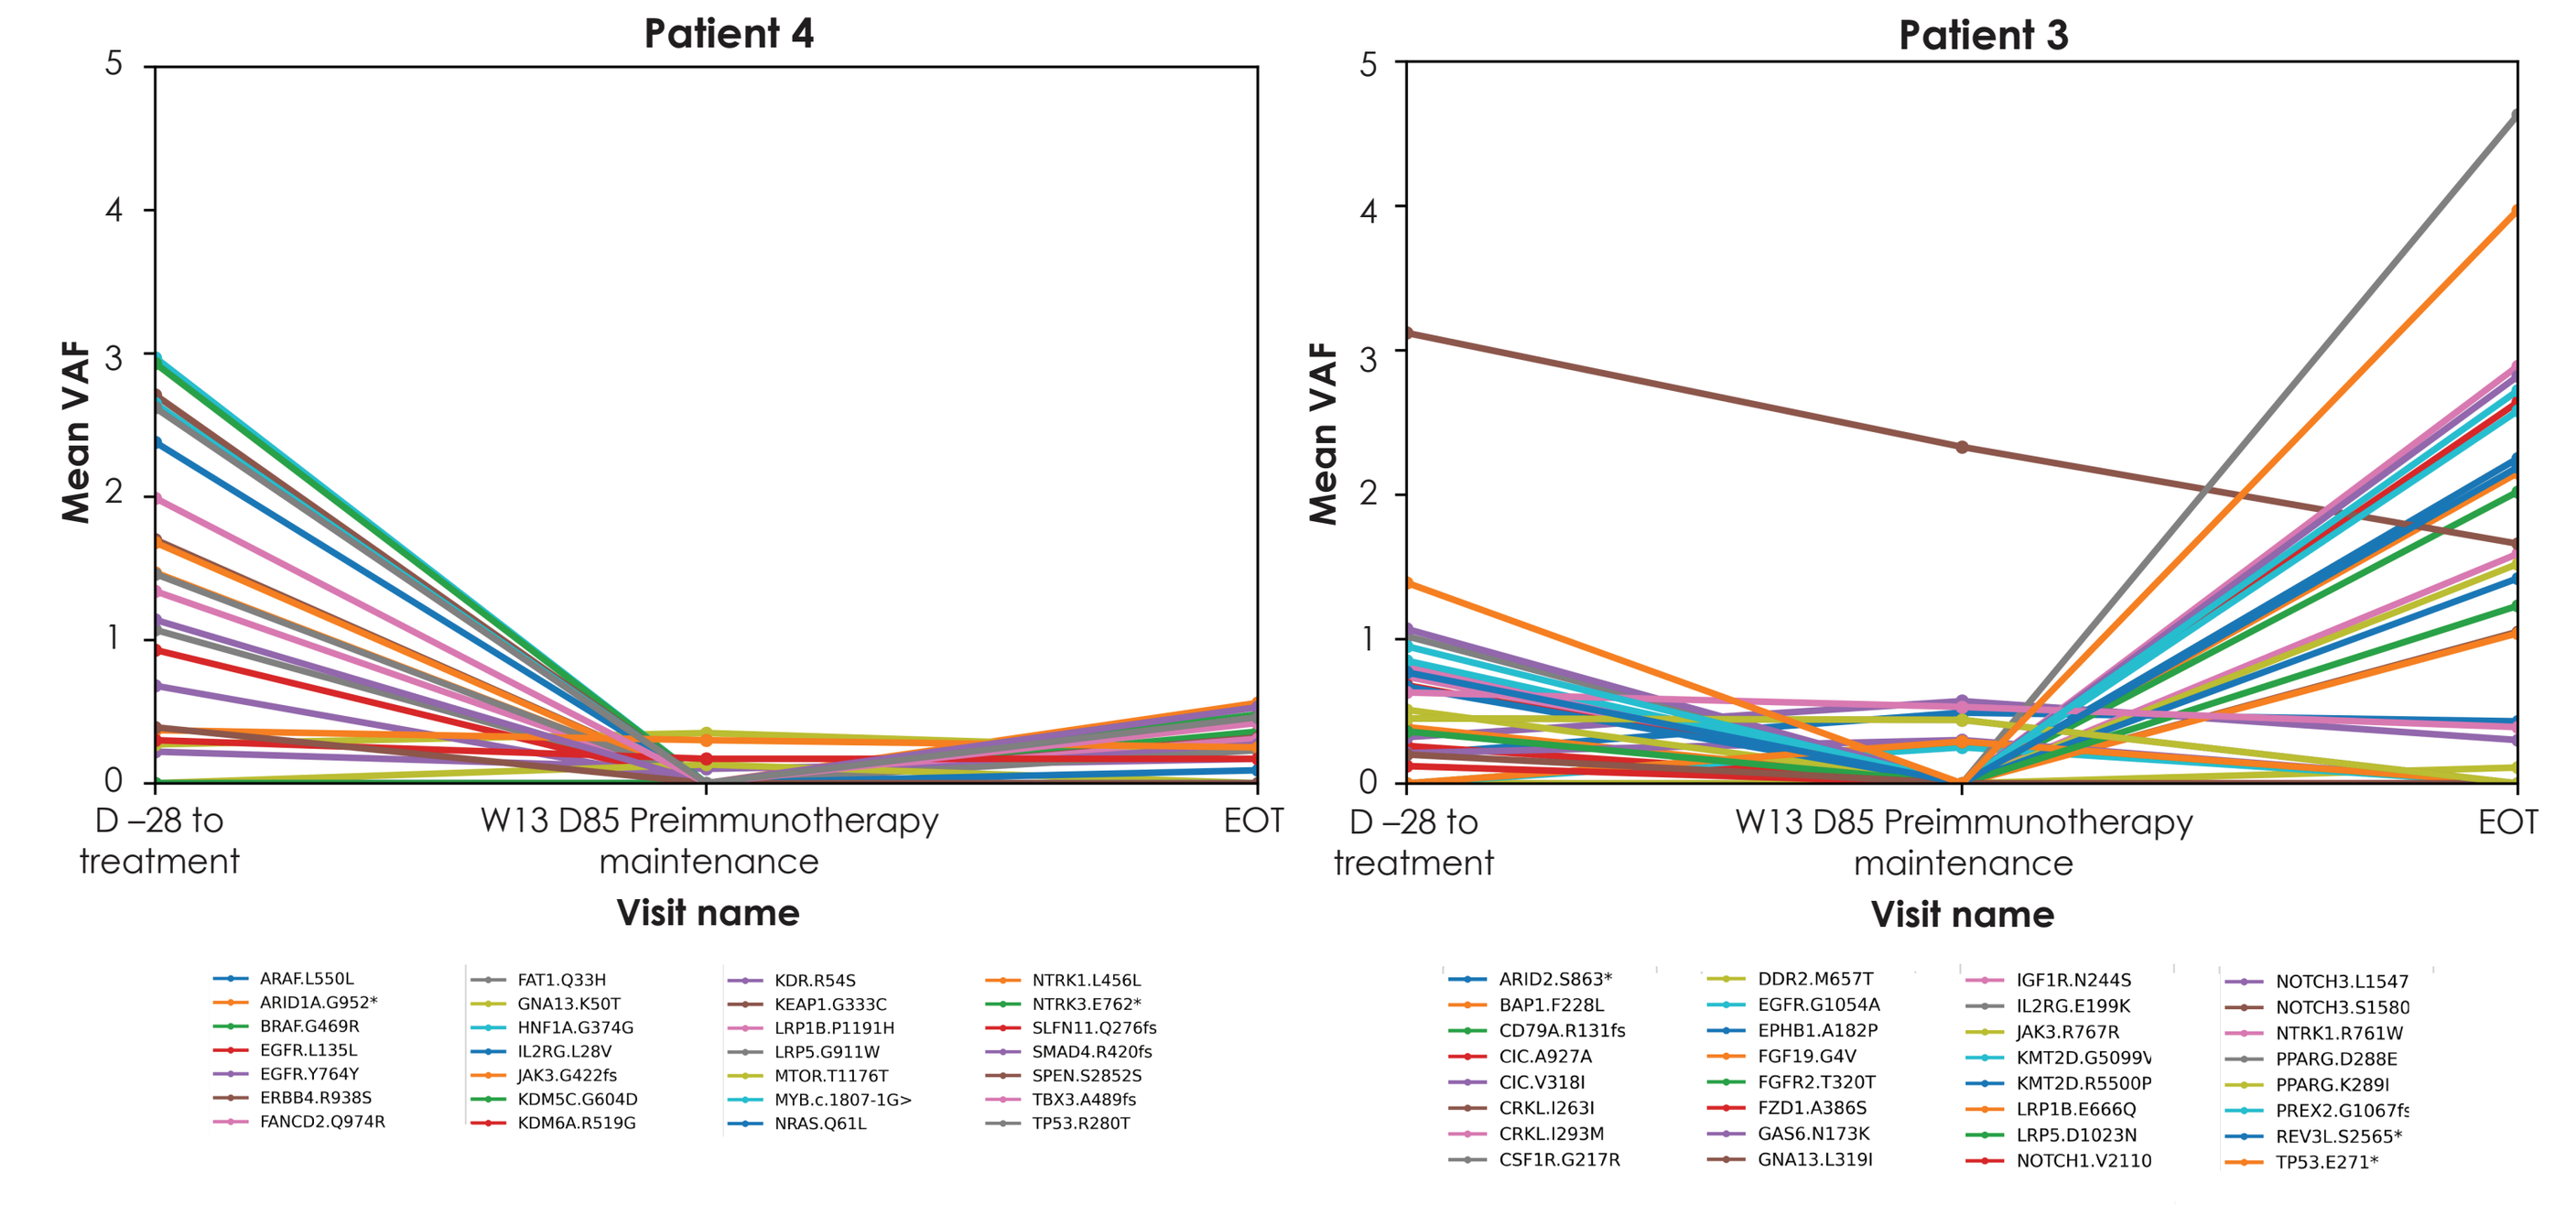
Patient 4
Patient 3
Mean VAF
Visit name
D –28 to treatment
W13 D85 Preimmunotherapy maintenance
EOT

*Contains a nonsense mutation that results in premature termination of the protein sequence.
bTMB, blood-based tumor mutational burden; **D,** day; **EOT,** end of treatment; **MAF,** mutation annotation format; **VAF,** variant allele frequency; **W,** week.

Correlation of change in ctDNA percentage with response at initiation of maintenance

- 6 of 7 (86%) patients with ctDNA change at a selected cutoff ≥70% had a visit 2 (V2) response of PR (n=3) or SD (n=3)
- At a selected cutoff <70% for ctDNA change, 4 of 6 (67%) patients had a V2 response of PD
- The average ctDNA reduction from baseline to day 85 was significantly greater for patients classified as PR or SD than for patients with PD (82% vs 57%; p=0.032)

Figure 6. ctDNA percentage change from baseline and response at initiation of immunotherapy maintenance

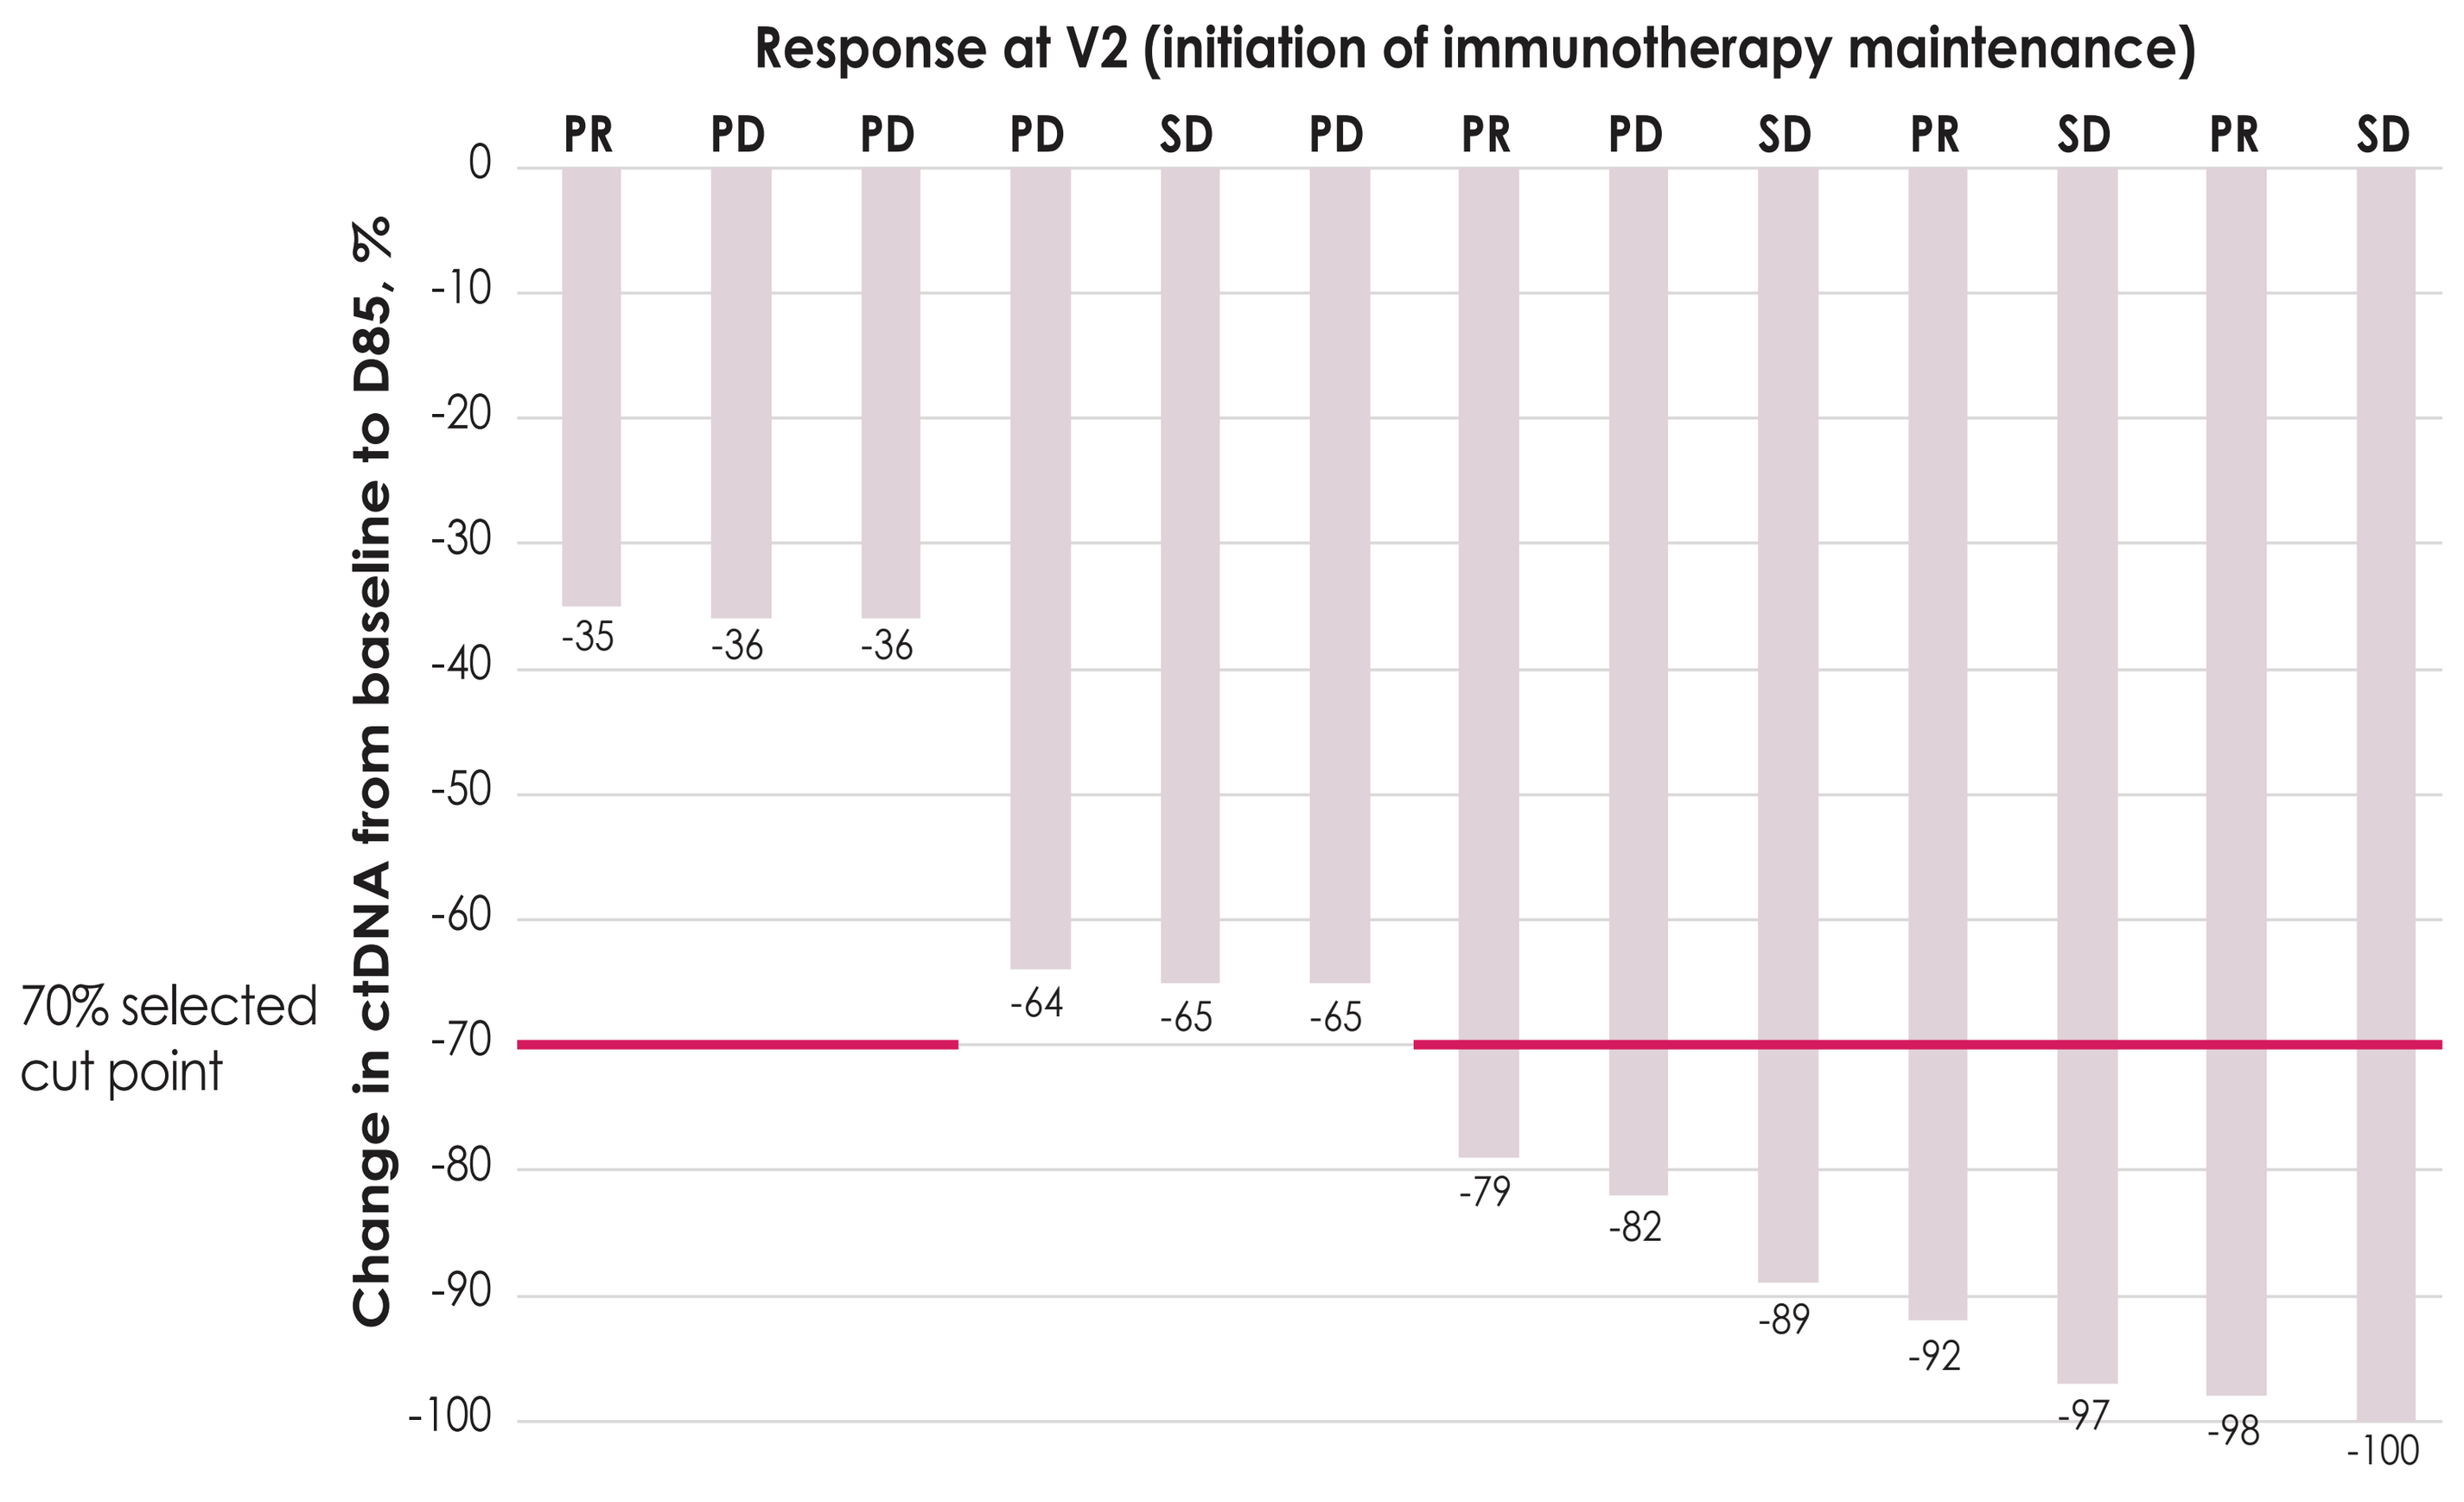
Response at V2 (initiation of immunotherapy maintenance)
PR PD PD PD SD PD PR PD SD PR SD PR SD
Change in ctDNA from baseline to D85, %
70% selected cut point
-35 -36 -36 -64 -65 -65 -79 -82 -89 -92 -97 -98 -100

ctDNA, circulating tumor DNA; **D,** day; **PD,** progressive disease; **PR,** partial response; **SD,** stable disease; **V2,** visit 2.

REFERENCES 1. Andric Z, et al. J Thorac Oncol. 2021;16(suppl 4):S748-S802. **2.** Hofman P, et al. Ann Oncol. 2019;30(9):1448-59. **3.** Feng Z, et al. Cancer Res. 2020;80(suppl 16). Abstract 1992. **4.** Smith N, et al. Cancer Res. 2022;82(suppl 12). Abstract 5748. **5.** Mak AJ, et al. Cancer Res. 2021;81(suppl 13). Abstract 401. **6.** Ricciuti B, et al. J Thorac Oncol. 2022;17(3):399-410.

DISCLOSURES F. Ciardiello has received research grants from Merck, Amgen, and Roche and has served in an advisory role for Merck, Amgen, Roche, MSD, Pierre Fabre, and Servier. **Z. Feng** is an employee of EMD Serono Research & Development Institute Inc., Billerica, MA, USA, an affiliate of Merck KGaA. **N. R. Smith** is a consultant for Merck Healthcare KGaA, Darmstadt, Germany. **Z. Andric** does not have any conflicts to declare. **S. A. Shell** is an employee of and has stocks/shares in Guardant Health. **A. Yablonovitch** is an employee of and has stocks/shares in Guardant Health. **G. Guezel** is an employee of Merck Healthcare KGaA, Darmstadt, Germany. **J. Scheuenpflug** is an employee of Merck Healthcare KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This research was sponsored by Merck (CrossRef Funder ID:10.13039/100009945) as part of an alliance between Merck and Pfizer. Medical writing support was provided by Abhijith Thippeswamy of ClinicalThinking and was funded by Merck and Pfizer.

Poster No. 1092P. Presented at the ESMO Congress 2022, 9-13 September 2022; Paris, France; Hybrid.