This is a reprint from European Society for Medical Oncology Congress 2023 (ESMO 2023), which was originally presented in Madrid, Spain on October 20-24, 2023; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

Clinical characteristics and survival of patients with advanced Merkel cell carcinoma (MCC) treated with avelumab: analysis of a prospective German MCC registry (MCC TRIM)

J. C. Becker,[1,2] S. H. Mahmoudpour,[3] D. Schadendorf,[4] U. Leiter,[5] F. Meier,[6,7] R. Herbst,[8] M. M. Sachse,[9] S. Grabbe,[10] C. Gebhardt,[11] F. Ziller,[12] C. Pföhler,[13] P. Mohr,[14] L. Zimmer,[15] E. Boutmy,[3] S. Hoff,[3] P. Verpillat,[3]* A. F. Milano,[16] M. Shlaen,[16] C. Fu,[17] S. Ugurel[18]

[1]Departments of Translational Skin Cancer Research and Dermatology, University Hospital Essen, Essen, Germany; [2]German Cancer Consortium (DKTK), Partner Site Essen/Düsseldorf, German Cancer Research Center (DKFZ), Heidelberg, Germany; [3]Merck Healthcare KGaA, Darmstadt, Germany; [4]Department of Dermatology, University Hospital Essen; German Cancer Consortium, Partner Site Essen; National Center for Tumor Diseases (NCT), NCT-West, Campus Essen; Research Alliance Ruhr – Research Center One Health, University Duisburg-Essen, Essen, Germany; [5]Department of Dermatology, Center for Dermatooncology, Eberhard Karls University of Tübingen, Tübingen, Germany; [6]Department of Dermatology, Faculty of Medicine and University Hospital Carl Gustav Carus, Technische Universität Dresden, Dresden, Germany; [7]Skin Cancer Center at the University Cancer Centre Dresden and National Center for Tumor Diseases, Dresden, Germany; [8]Department of Dermatology, Helios Klinikum Erfurt, Erfurt, Germany; [9]Department of Dermatology, Allergology and Phlebology, Bremerhaven-Reinkenheide Hospital, Bremerhaven, Germany; [10]Department of Dermatology, University Medical Center of the Johannes Gutenberg University, Mainz, Germany; [11]Department of Dermatology and Venereology, University Medical Center Hamburg-Eppendorf (UKE), Hamburg, Germany; [12]Department of Dermatology, DRK Krankenhaus Rabenstein, Chemnitz, Germany; [13]Department of Dermatology, Saarland University Medical School, Homburg/Saar, Germany; [14]Department of Dermatology, Elbe-Kliniken, Buxtehude, Germany; [15]Department of Dermatology, University Hospital Essen; German Cancer Consortium, Partner Site Essen; National Center for Tumor Diseases (NCT), NCT-West, Campus Essen, Germany; [16]IQVIA Commercial GmbH & Co. OHG, Frankfurt am Main, Germany; [17]IQVIA Solutions Enterprise Management Consulting (Shanghai) Co., Ltd., Beijing, China; [18]Department of Dermatology, University Hospital Essen, University of Duisburg-Essen & German Cancer Consortium, Heidelberg, Germany

*Affiliation at the time the study was conducted.

SCOPE

- **This registry-based analysis provides real-world evidence on patient characteristics and the effectiveness of avelumab treatment in patients with advanced MCC in Germany**

CONCLUSIONS

- **Demographics observed in MCC TRIM reflect those of the typical advanced MCC population**
- **This nationwide study indicates that avelumab is commonly prescribed and effective in patients with stage III and IV MCC in routine clinical practice in Germany**
- **In addition to showing that patients not considered immunocompromised and patients without diabetes had improved outcomes with avelumab, our real-world study also demonstrated the effectiveness of avelumab in those considered immunocompromised and those with diabetes**
- **Our findings are in line with the results from the JAVELIN Merkel 200 clinical trial and observational studies of advanced MCC populations performed outside Germany[1,2]**

GET POSTER PDF

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

Correspondence:
Jürgen C. Becker, j.becker@dkfz-heidelberg.de

GET PLAIN LANGUAGE SUMMARY

Please scan this QR code with your smartphone app to view a plain language summary of the accepted scientific abstract.

BACKGROUND

- MCC is a rare, neuroendocrine, cutaneous malignancy characterized by firm, painless, rapidly enlarging, dome-shaped nodules mostly on sun-exposed areas. The incidence of MCC is low (0.13/100,000 population in Europe), and it occurs more commonly in men than women. The proportion of patients with MCC diagnosed with distant metastatic disease has been reported to be between 4% and 12%[3]
- In September 2017, the European Commission approved avelumab for the treatment of metastatic MCC; avelumab was the first immune checkpoint inhibitor immunotherapy approved for patients with MCC[4]
- With avelumab having been integrated into routine clinical practice in Germany, it is necessary to understand the use and clinical benefits of avelumab from real-world data
- Accordingly, this study reports the use of avelumab in patients with advanced MCC in Germany as well as patient characteristics and associated clinical outcomes

METHODS

- Data from patients enrolled in a prospective, noninterventional, multicenter, dynamic cohort of the MCC TRIM study after March 2019 were analyzed
- The Independent Ethics Committee of University Duisburg-Essen approved the MCC TRIM protocol on 28 September 2018
- Primary data from a study-specific electronic case report form and secondary data from the German national skin cancer registry ADOReg were combined
- At data cutoff (30 September 2022), patients diagnosed with unresectable stage III or IV MCC prior to avelumab treatment were included in this analysis
- Descriptive statistics were calculated for categorical and continuous baseline variables at the start of avelumab treatment, and the Kaplan-Meier method was used to estimate overall survival (OS), progression-free survival (PFS), and median follow-up*

*The median follow-up has been calculated based on the Kaplan-Meier method as the smallest survival time for which the survivor function is ≤0.5. A CI for the median survival time is constructed using the log cumulative hazard transformation. End date of study period or date of the last contact before loss to follow-up has been considered. Patients were censored in case of death.

- Survival outcomes were reported from the date of avelumab initiation, stratified by stage of disease at avelumab initiation
- In the subanalyses, survival outcomes were reported stratified by immunosuppressive condition and diabetes status at avelumab initiation
- Patients were considered immunocompromised when specific comorbidities or concomitant medications were ongoing at the time of initiation of the systemic treatment
- All analyses were conducted descriptively with no adjustment for any comparison

RESULTS

Patients

- A total of 684 patients were included in the MCC TRIM study at data cutoff. The majority were male (53.8%), and the mean age was 73.5 (SD, 10.4) years. **Table 1** presents baseline characteristics of the overall MCC TRIM population, and **Table 2** presents comorbidities
- In total, 116 patients with advanced MCC received avelumab, of whom 62.1% were male. The mean age at diagnosis was 74.5 (SD, 10.4) years, and 64.7% had stage IV disease. Most patients had an ECOG performance status of ≤1 (76.7%; **Table 1**)
- 21.6% had an immunosuppressive condition or took immunosuppressive medication. Diabetes was the most common comorbidity (23.3%; **Table 2**)

Table 1. Baseline characteristics of the overall MCC TRIM population and patients with aMCC treated with avelumab

	Overall population (N=684)		Avelumab-treated patients (n=116)	
	n (%)*	95% CI, %	n (%)*	95% CI, %
Sex, n (%)				
Male	368 (53.8)	50.1-57.5	72 (62.1)	53.2-70.9
Female	316 (46.2)	42.5-49.9	44 (37.9)	29.1-46.8
Age at initial diagnosis, years				
Mean (SD)	73.5 (10.4)	–	74.5 (10.4)	–
Median	75	–	77	–
Tumor stage, n (%)				
0	7 (1.0)	0.3-1.8	0	0.0-0.0
I	237 (34.6)	31.1-38.2	0	0.0-0.0
II	118 (17.3)	14.4-20.1	0	0.0-0.0
III	218 (31.9)	28.4-35.4	41 (35.3)	26.6-44.0
IV	95 (13.9)	11.3-16.5	75 (64.7)	56.0-73.4
ECOG performance status, n (%)				
Not recorded	106 (15.5)	12.8-18.2	22 (19.0)	11.8-26.1
0	424 (62.0)	58.4-65.6	58 (50.0)	40.9-59.1
1	121 (17.7)	14.8-20.5	31 (26.7)	18.7-34.8
2	26 (3.8)	2.4-5.2	4 (3.4)	0.1-6.8
3	5 (0.7)	0.1-1.4	0	0.0-0.0
4	2 (0.3)	0.0-0.7	1 (0.9)	0.0-2.5
Year of index/enrollment, n (%)				
2019	116 (17.0)	14.1-19.8	29 (25.0)	17.1-32.9
2020	266 (38.9)	35.2-42.5	54 (46.6)	37.5-55.6
2021	210 (30.7)	27.2-34.2	27 (23.3)	15.6-31.0
2022	92 (13.5)	10.9-16.0	6 (5.2)	1.1-9.2
Description of metastasis, n (%)				
Patient count	313 (45.8)	42.0-49.5	116 (100)	100-100
Information unavailable at data cutoff†	41 (13.1)	9.4-16.8	4 (3.4)	0.1-6.8
Satellite/in transit†	46 (14.7)	10.8-18.6	19 (16.4)	9.6-23.1
Lymph node†	139 (44.4)	38.9-49.9	18 (15.5)	8.9-22.1
Distant†	95 (30.4)	25.3-35.4	75 (64.7)	56.0-73.4

aMCC, advanced Merkel cell carcinoma.
*Excluding age category. †Percentage calculation based on patient count.

Table 2. Comorbidities in the overall MCC TRIM population and patients with aMCC treated with avelumab

	Overall population (N=684)		Avelumab-treated patients (n=116)	
	n (%)	95% CI, %	n (%)	95% CI, %
Other skin cancers, n (%)				
Melanoma	28 (4.1)	2.6-5.6	1 (0.9)	0.0-2.5
Squamous cell carcinoma	50 (7.3)	5.4-9.3	13 (11.2)	5.5-16.9
Actinic keratosis	43 (6.3)	4.5-8.1	11 (9.5)	4.2-14.8
Basal cell carcinoma	80 (11.7)	9.3-14.1	11 (9.5)	4.2-14.8
Other malignancies, n (%)				
Solid tumors	73 (10.7)	8.4-13.0	12 (10.3)	4.8-15.9
Hematologic malignancy	25 (3.7)	2.2-5.1	12 (10.3)	4.8-15.9
Immunosuppression, n (%)				
Patients considered immunocompromised	54 (7.9)	5.9-9.9	25 (21.6)	14.1-29.0
Systemic/connective tissue diseases, n (%)				
Rheumatoid arthritis	15 (2.2)	1.1-3.3	4 (3.4)	0.1-6.8
Systemic sclerosis	1 (0.1)	0.0-0.4	0	0.0-0.0
Gastrointestinal inflammatory diseases, n (%)				
Inflammatory bowel disease	9 (1.3)	0.5-2.2	2 (1.7)	0.0-4.1
Other comorbidities, n (%)				
Diabetes	127 (18.6)	15.7-21.5	27 (23.3)	15.6-31.0
COPD	21 (3.1)	1.8-4.4	3 (2.6)	0.0-5.5
Cerebrovascular disease/stroke	19 (2.8)	1.5-4.0	4 (3.4)	0.1-6.8
Moderate or severe renal disease	42 (6.1)	4.3-7.9	13 (11.2)	5.5-16.9
Ischemic heart disease/myocardial infarction	58 (8.5)	6.4-10.6	14 (12.1)	6.1-18.0
Moderate or severe liver disease	10 (1.5)	0.6-2.4	5 (4.3)	0.6-8.0
Thyroid disorders	67 (9.8)	7.6-12.0	13 (11.2)	5.5-16.9

aMCC, advanced Merkel cell carcinoma; **COPD**, chronic obstructive pulmonary disease.

Survival outcomes

- After a median follow-up of 28.75 months (95% CI, 20.5-32.5 months), median PFS was 11.8 months (95% CI, 5.6 months-not estimable [NE]) in patients with stage III disease and 9.3 months (95% CI, 3.5-13.7 months) in patients with stage IV disease (**Figure 1A**)
- Median OS was not reached in patients with stage III disease and 52.0 months (95% CI, 15.4 months-NE) in patients with stage IV disease (**Figure 1B**)
- Most patients (86.2%) received avelumab as first-line (1L) therapy; of these, 37.0% had stage III disease, and 63.0% had stage IV disease
- Median PFS with 1L avelumab in patients with stage III and IV disease was 15.6 months (95% CI, 5.6 months-NE) and 7.3 months (95% CI, 3.0-13.7 months), respectively (**Figure 2A**)
- Median OS with 1L avelumab was not reached in the stage III or IV subgroup, but fewer events were observed in patients with stage III vs IV disease (16.2% vs 36.5%; **Figure 2B**)

Figure 1. Survival analysis in all patients who received avelumab stratified by stage of disease

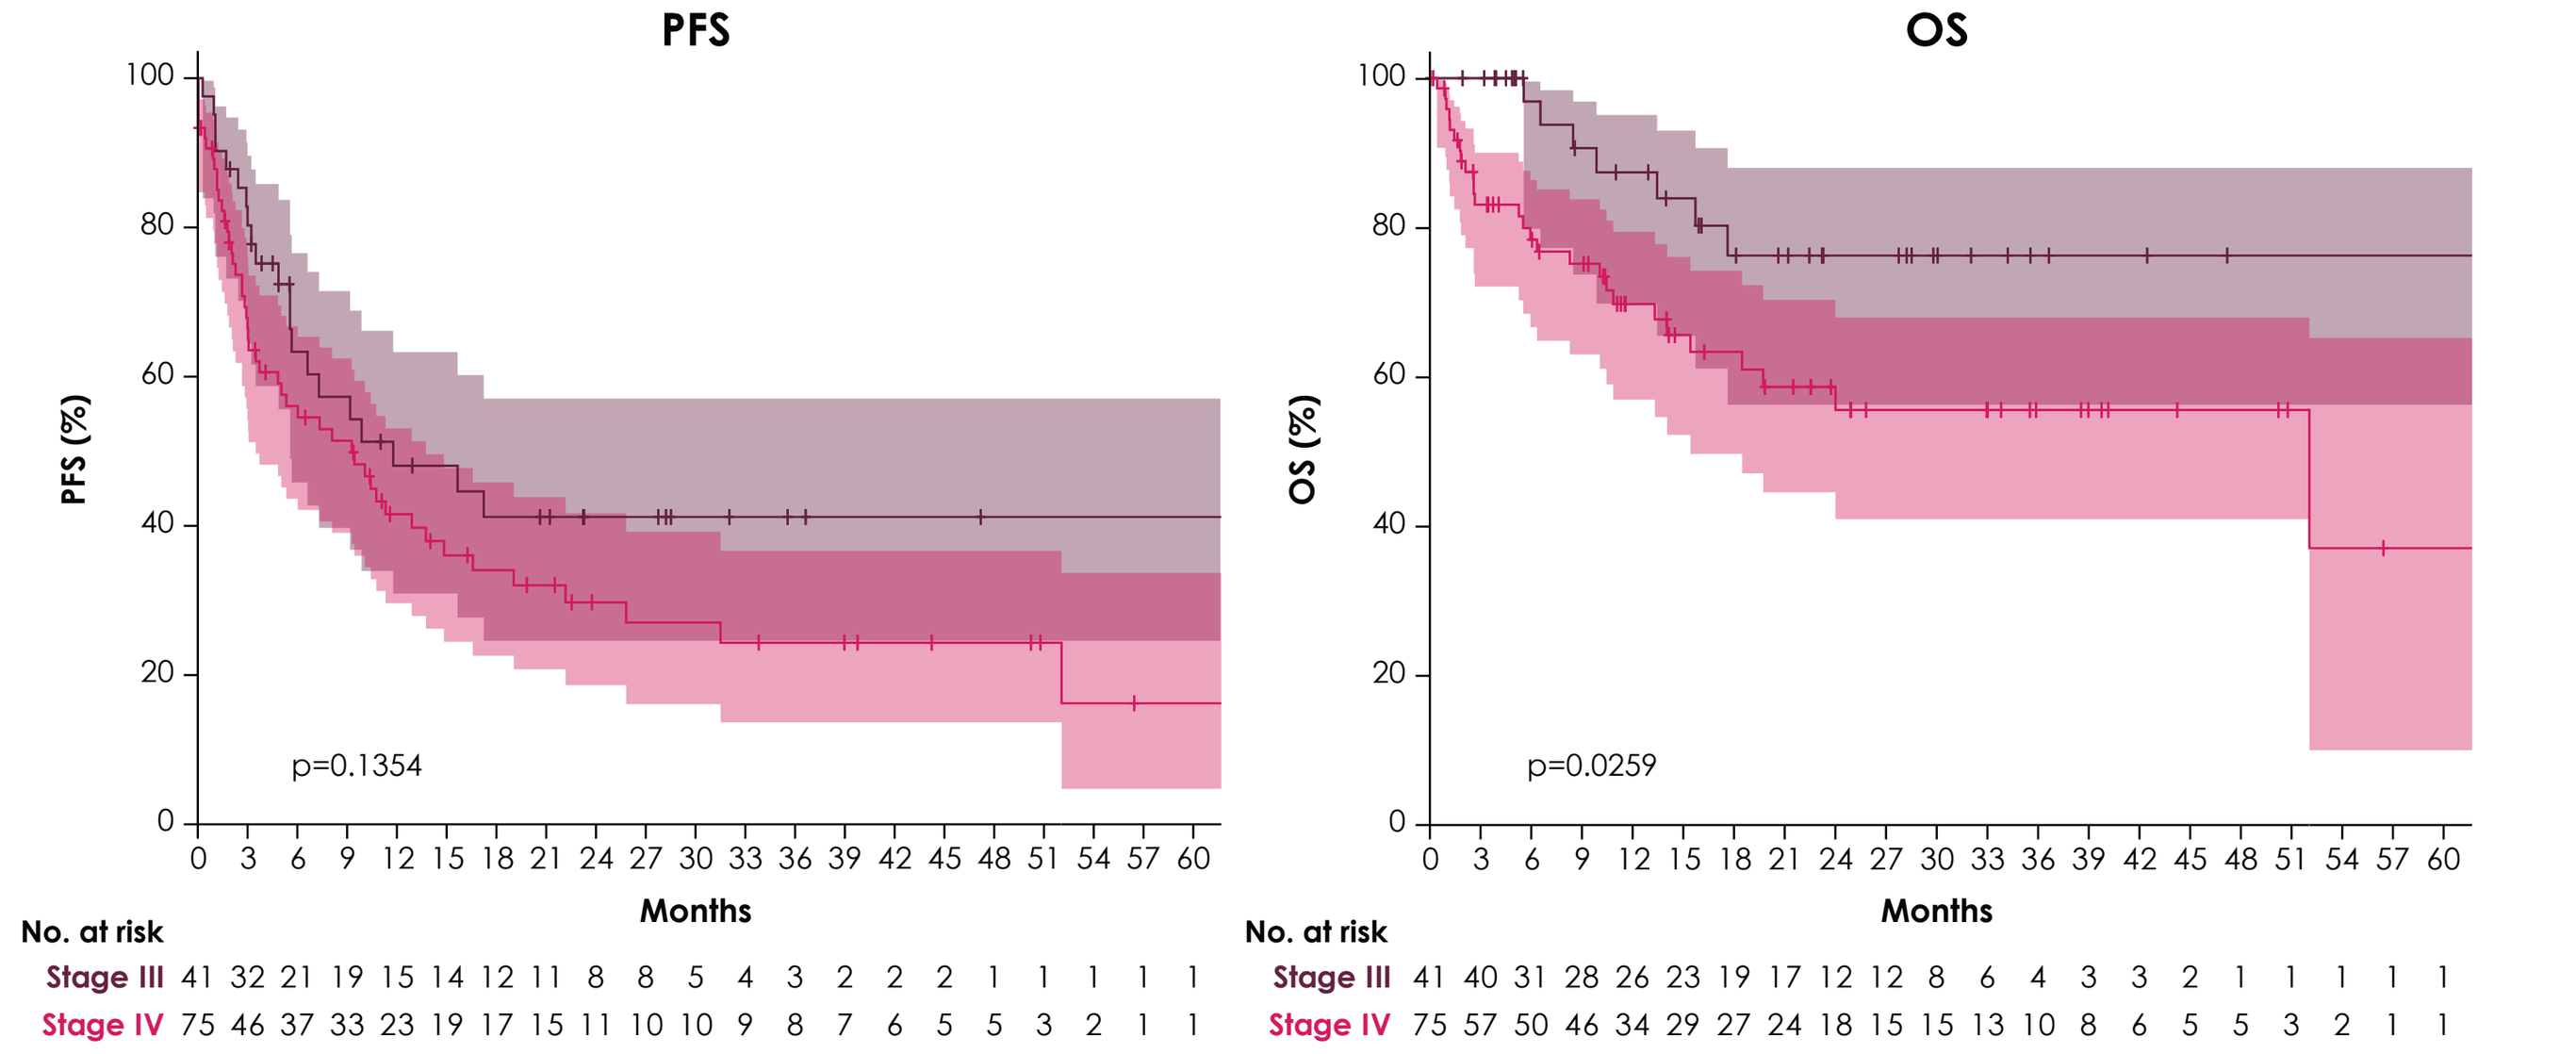

PFS
OS
PFS (%)
OS (%)
p=0.1354
p=0.0259
Months
No. at risk
Stage III 41 32 21 19 15 14 12 11 8 8 5 4 3 2 2 2 1 1 1 1 1
Stage IV 75 46 37 33 23 19 17 15 11 10 10 9 8 7 6 5 5 3 2 1 1
Stage III 41 40 31 28 26 23 19 17 12 12 8 6 4 3 3 2 1 1 1 1 1
Stage IV 75 57 50 46 34 29 27 24 18 15 15 13 10 8 6 5 5 3 2 1 1

	Stage III (n=41)	Stage IV (n=75)
Events, n	21	49
Censored, n	20	26
PFS, median (95% CI), months	11.8 (5.6-NE)	9.3 (3.5-13.7)

	Stage III (n=41)	Stage IV (n=75)
Events, n	7	27
Censored, n	34	48
OS, median (95% CI), months	NR (NE-NE)	52 (15.4-NE)

NE, not estimable; **NR**, not reached; **OS**, overall survival; **PFS**, progression-free survival.

Figure 2. Survival analysis in patients who received avelumab as 1L treatment stratified by stage of disease

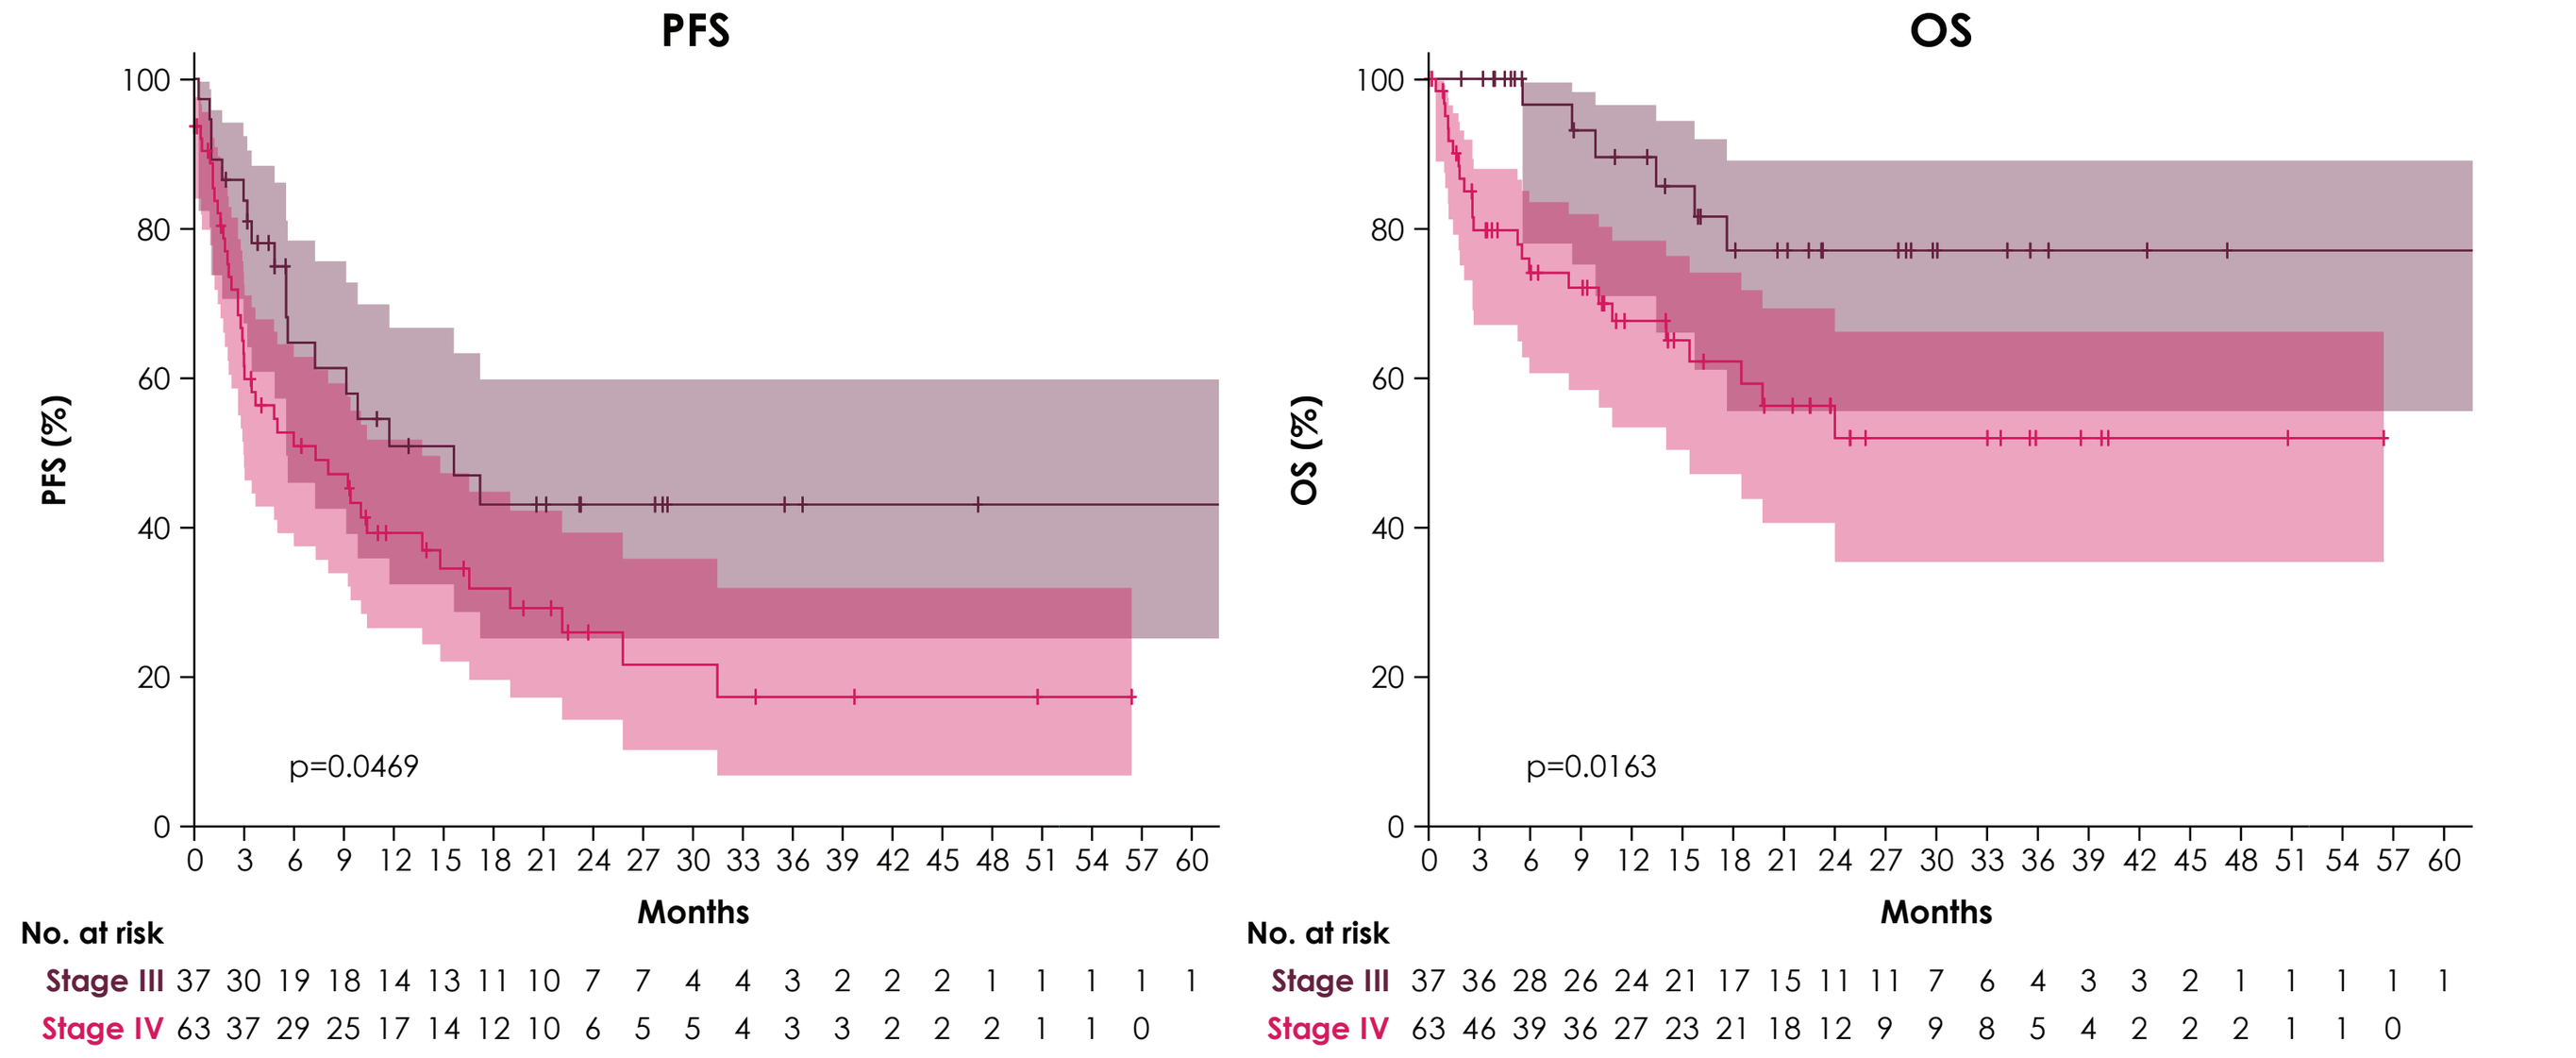

PFS
OS
PFS (%)
OS (%)
p=0.0469
p=0.0163
Months
No. at risk
Stage III 37 30 19 18 14 13 11 10 7 7 4 4 3 2 2 2 1 1 1 1 1
Stage IV 63 37 29 25 17 14 12 10 6 5 5 4 3 3 2 2 2 1 1 0
Stage III 37 36 28 26 24 21 17 15 11 11 7 6 4 3 3 2 1 1 1 1 1
Stage IV 63 46 39 36 27 23 21 18 12 9 9 8 5 4 2 2 2 1 1 0

	Stage III (n=37)	Stage IV (n=63)
Events, n	18	42
Censored, n	19	21
PFS, median (95% CI), months	15.6 (5.6-NE)	7.3 (3.0-13.7)

	Stage III (n=37)	Stage IV (n=63)
Events, n	6	23
Censored, n	31	40
OS, median (95% CI), months	NR (NE-NE)	NR (15.4-NE)

1L, first line; **NE**, not estimable; **NR**, not reached; **OS**, overall survival; **PFS**, progression-free survival.

Survival outcomes for patients with comorbidities

- When stratified based on immunosuppressive conditions, median PFS was 10.7 months (95% CI, 5.7-19.0 months) in patients not considered immunocompromised and 9.3 months (95% CI, 3.5-15.6 months) in patients considered immunocompromised (**Figure 3A**)
- Median OS was not reached in patients not considered immunocompromised and 18.5 months (95% CI, 10.4 months-NE) in patients considered immunocompromised (**Figure 3B**)
- Median PFS was 10.1 months (95% CI, 6.0-17.2 months) in patients without diabetes and 5.6 months (95% CI, 2.7-22.1 months) in patients with diabetes (**Figure 4A**)
- Median OS with avelumab was not reached in patients without diabetes and 52.0 months (95% CI, 6.3-52.0 months) in patients with diabetes (**Figure 4B**)

Figure 3. Survival analysis in all patients who received avelumab stratified by immunosuppressive condition

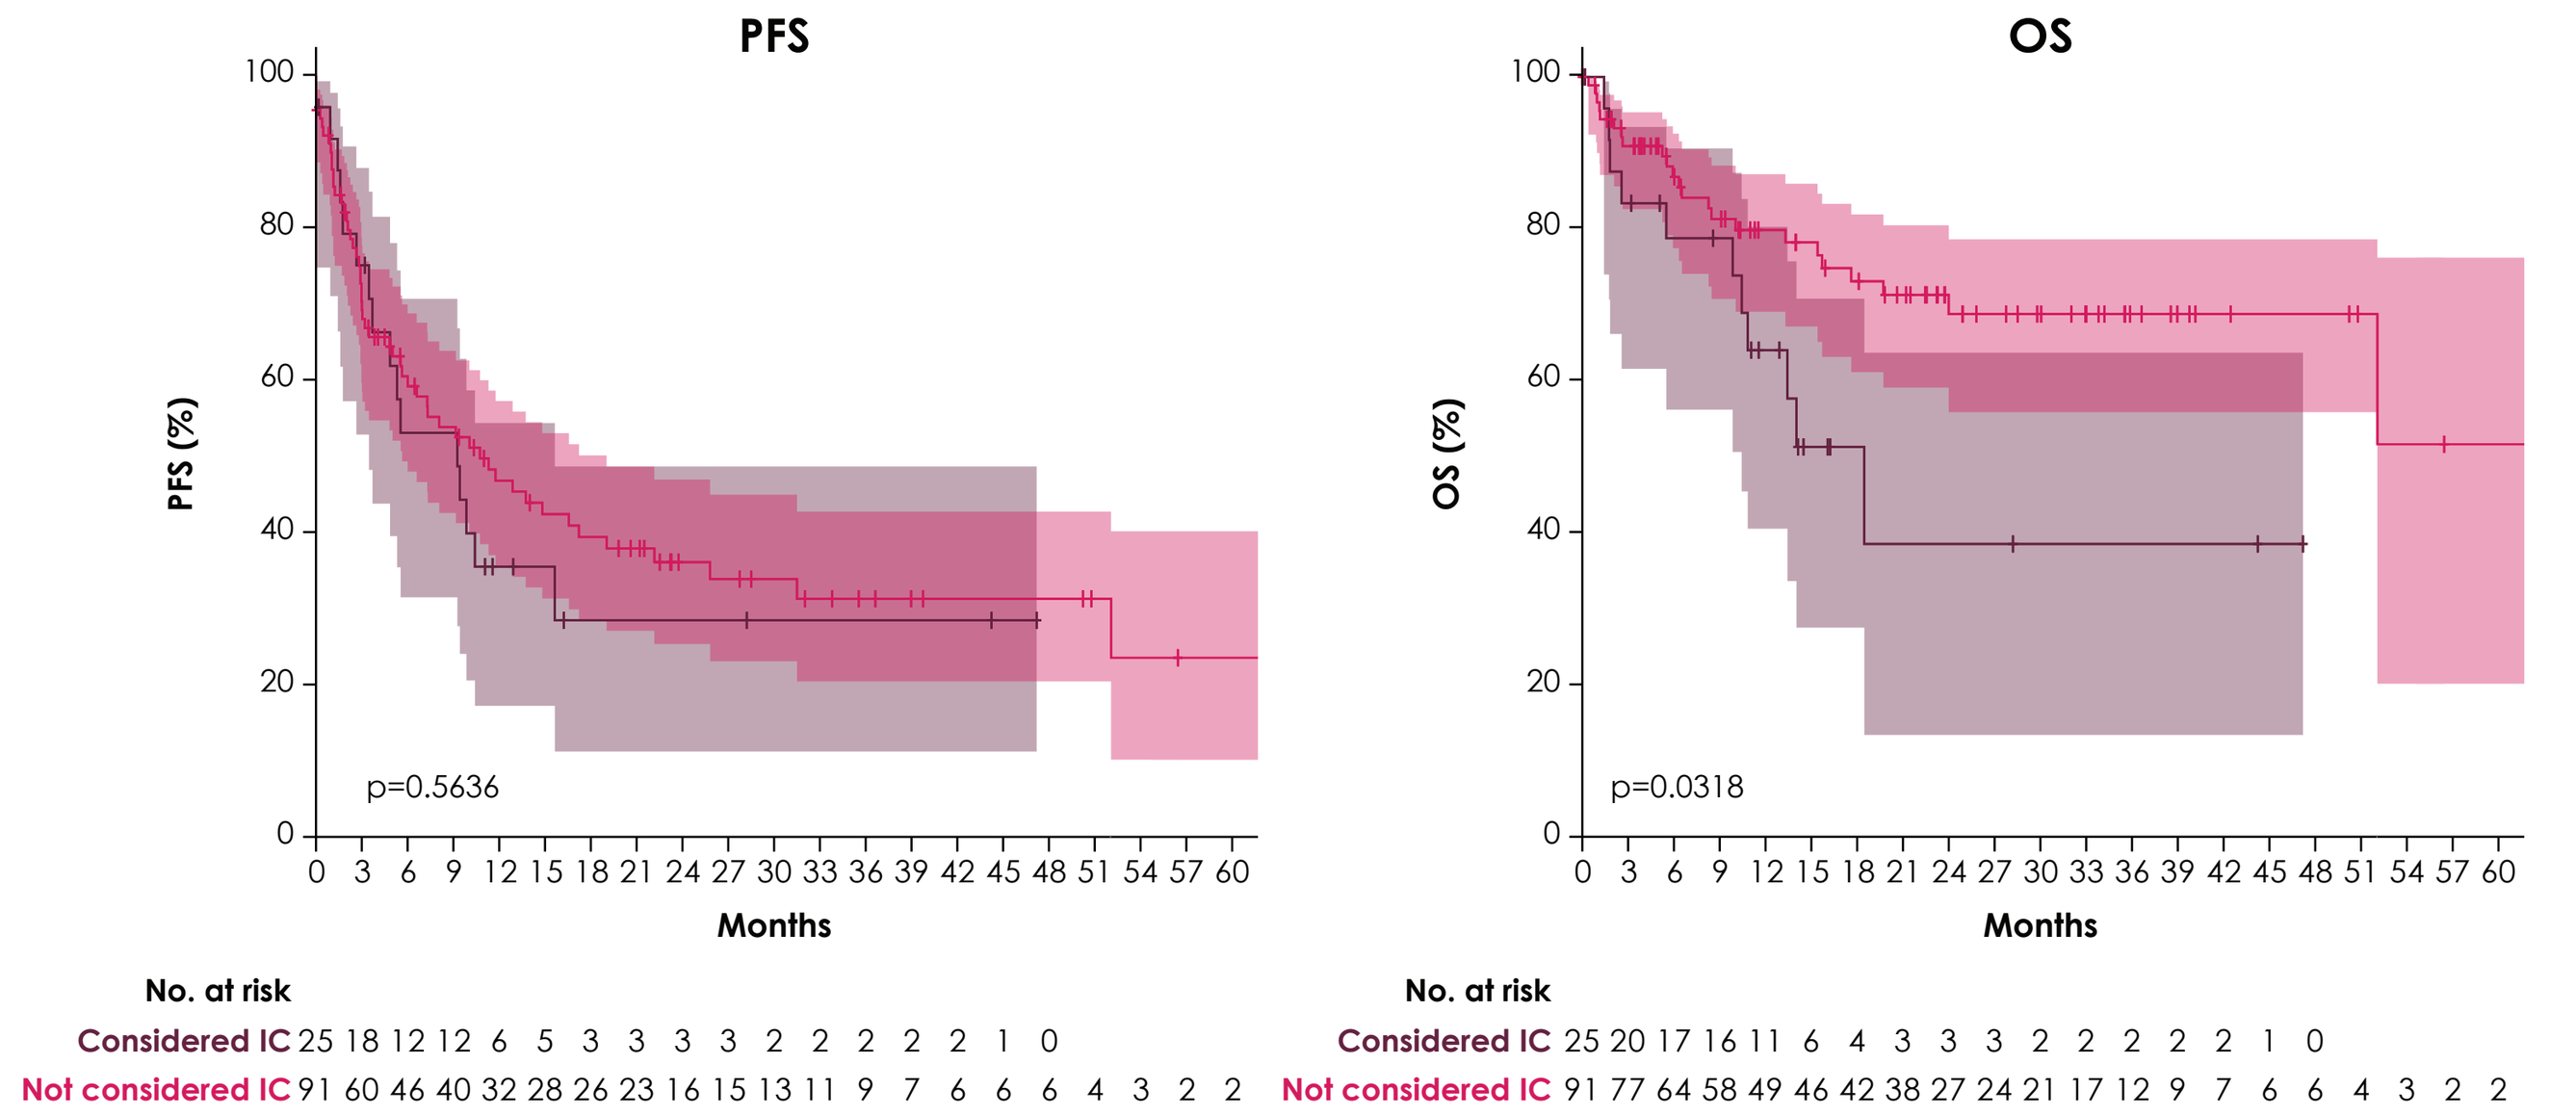

PFS
OS
PFS (%)
OS (%)
p=0.5636
p=0.0318
Months
No. at risk
Considered IC 25 18 12 12 6 5 3 3 3 3 2 2 2 2 2 1 0
Not considered IC 91 60 46 40 32 28 26 23 16 15 13 11 9 7 6 6 6 4 3 2 2
Considered IC 25 20 17 16 11 6 4 3 3 3 2 2 2 2 2 1 0
Not considered IC 91 77 64 58 49 46 42 38 27 24 21 17 12 9 7 6 6 4 3 2 2

	Considered IC (n=25)	Not considered IC (n=91)
Events, n	16	54
Censored, n	9	37
PFS, median (95% CI), months	9.3 (3.5-15.6)	10.7 (5.7-19.0)

	Considered IC (n=25)	Not considered IC (n=91)
Events, n	11	23
Censored, n	14	68
OS, median (95% CI), months	18.5 (10.4-NE)	NR (52.0-NE)

IC, immunocompromised; **NE**, not estimable; **OS**, overall survival; **PFS**, progression-free survival.

Figure 4. Survival analysis in all patients who received avelumab stratified by diabetes status

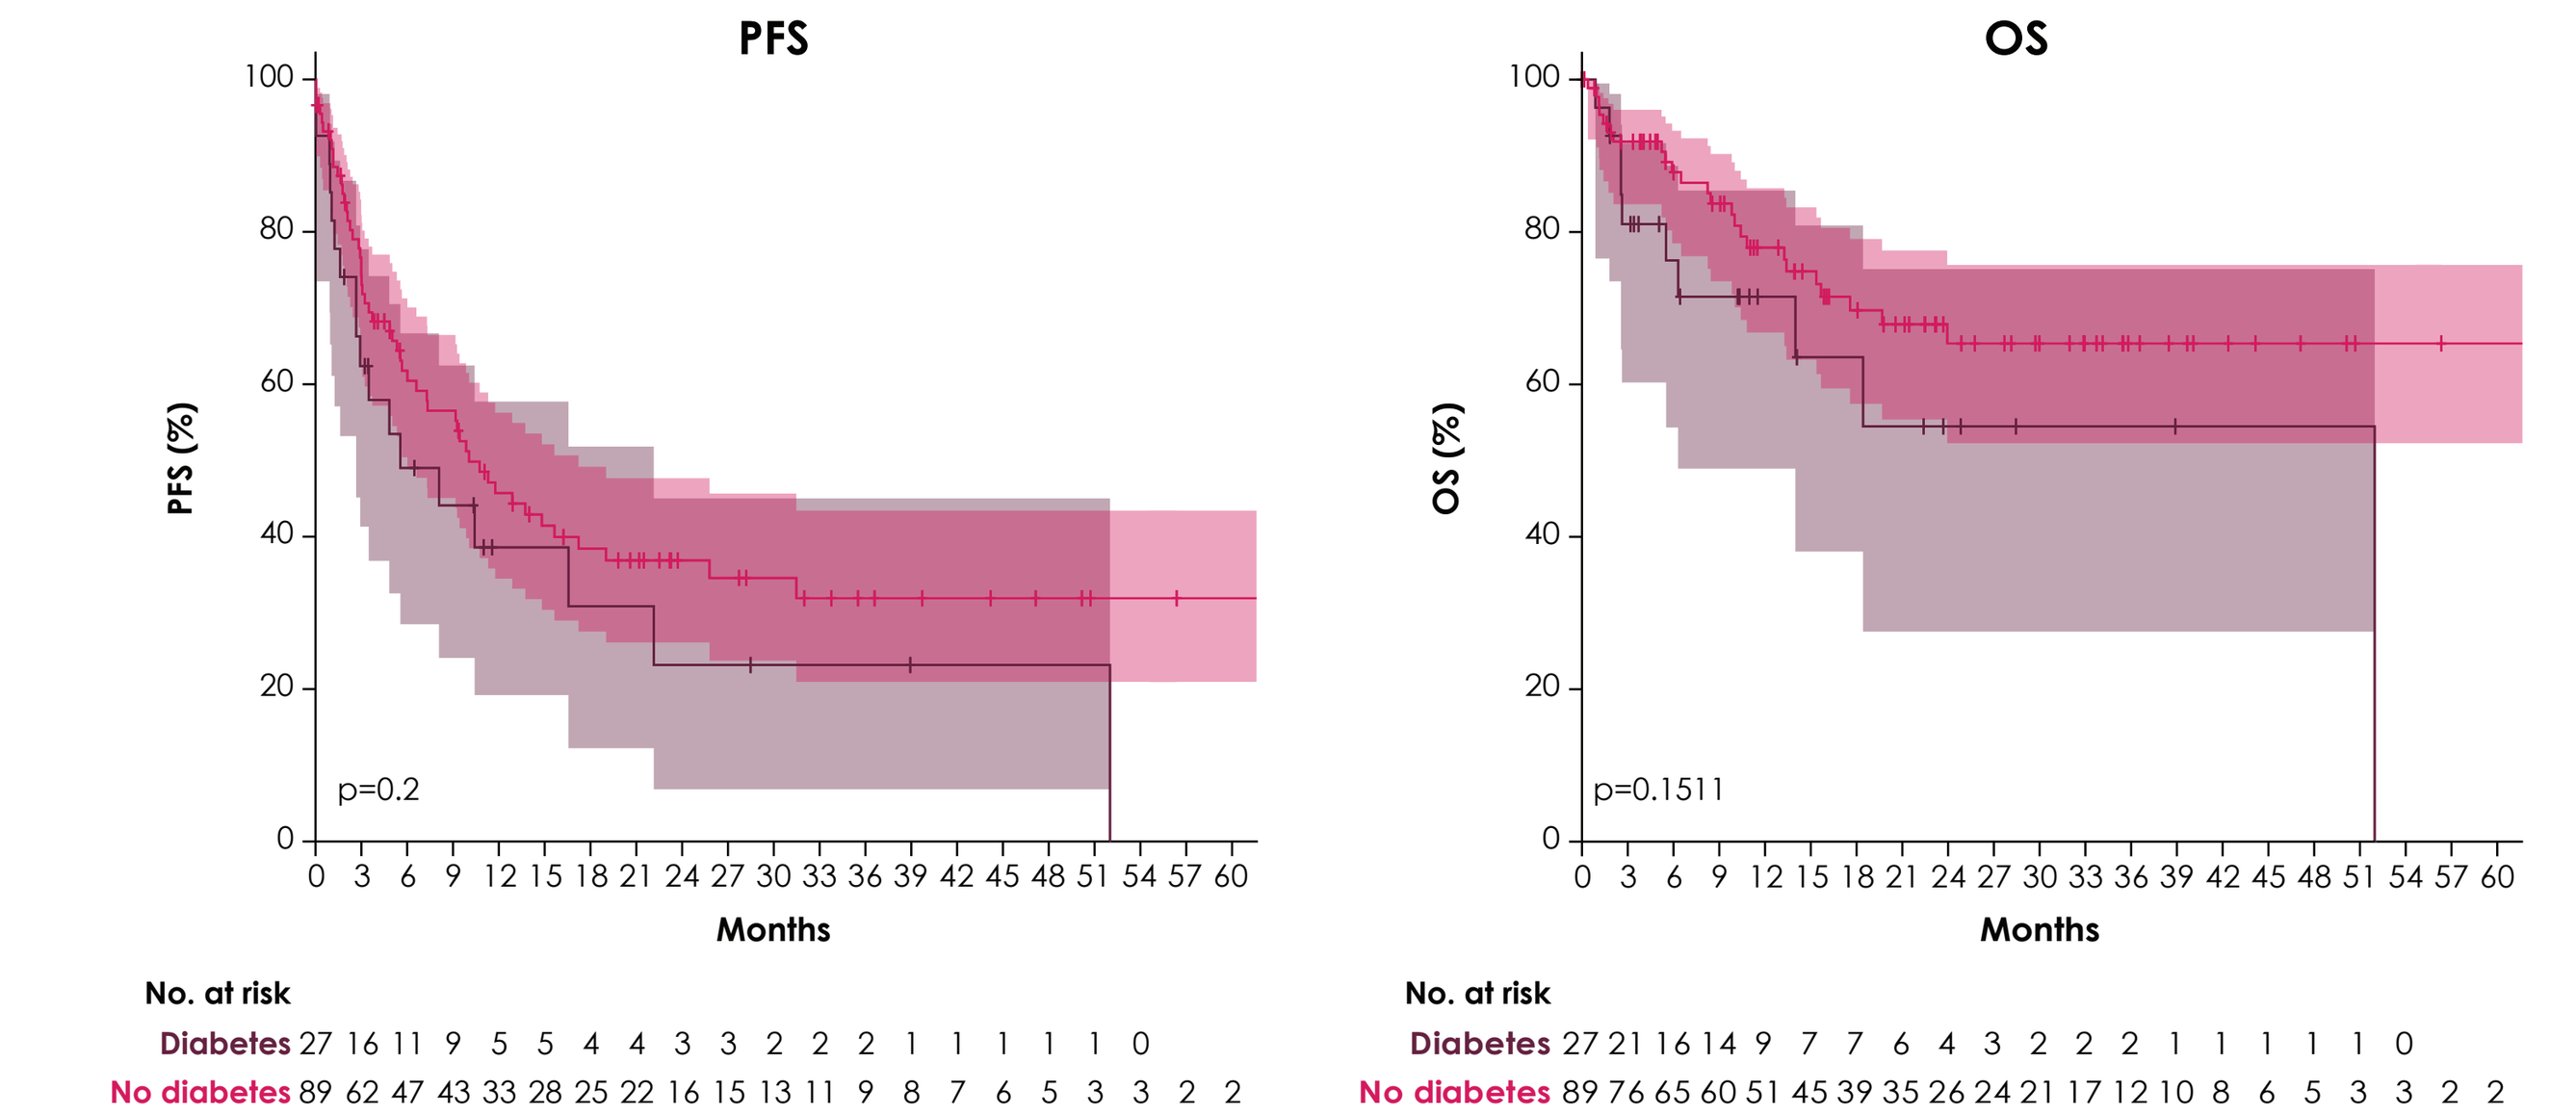

PFS
OS
PFS (%)
OS (%)
p=0.2
p=0.1511
Months
No. at risk
Diabetes 27 16 11 9 5 5 4 4 3 3 2 2 2 1 1 1 1 1 0
No diabetes 89 62 47 43 33 28 25 22 16 15 13 11 9 8 7 6 5 3 3 2 2
Diabetes 27 21 16 14 9 7 7 6 4 3 2 2 2 1 1 1 1 1 0
No diabetes 89 76 65 60 51 45 39 35 26 24 21 17 12 10 8 6 5 3 3 2 2

	Diabetes (n=27)	No diabetes (n=89)
Events, n	18	52
Censored, n	9	37
PFS, median (95% CI), months	5.6 (2.7-22.1)	10.1 (6.0-17.2)

	Diabetes (n=27)	No diabetes (n=89)
Events, n	10	24
Censored, n	17	65
OS, median (95% CI), months	52.0 (6.3-52.0)	NR (NE-NE)

NE, not estimable; **NR**, not reached; **OS**, overall survival; **PFS**, progression-free survival.

STRENGTHS

- MCC TRIM is a nationally representative registry of patients with MCC of any stage and with any treatment in Germany; therefore, the reported findings are representative of real-world clinical practice in Germany
- The diagnosis of MCC for patients enrolled in this study was confirmed uniformly and centrally via laboratories in the German Cancer Research Center (DKFZ)
- The MCC TRIM registry benefits from primary data collection and robust validation procedures combined with the use of secondary data from electronic medical records, which maximize completeness and standardize recording to reduce the risk of misclassification

LIMITATIONS

- Data presented in this study are purely descriptive and may not be compared statistically with other real-world data studies because of the various known and unknown confounding factors
- For some of the patients who were diagnosed and initiated treatment before enrollment to the registry, the time from the diagnosis to the enrollment could be considered immortal time

REFERENCES 1. D'Angelo SP, et al. J Immunother Cancer. 2021;9(7):e002646. **2.** Bhatia S, et al. J Immunother Cancer. 2022;10(8):e004904. **3.** Schadendorf D, et al. Eur J Cancer. 2017;71:53-69. **4.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA; 2022.

DISCLOSURES J. Becker has received speakers bureau honoraria from Amgen, Merck, Pfizer, Recordati, and Sanofi; has served in consulting or advisory roles for 4SC, Almirall, Boehringer Ingelheim, ICON, InProTher, Pfizer, and Sanofi/Regeneron; and has received research funding from Alcedis, Bristol Myers Squibb, HTG Molecular Diagnostics, IQVIA, and Merck. **S. Mahmoudpour** reports employment by Merck. **D. Schadendorf** reports speaker services for Bristol Myers Squibb, Merck, MSD, Novartis, Roche, and Sanofi; has served in consulting or advisory roles for Bristol Myers Squibb, Immunocore, MSD, NeraCare, Novartis, Pfizer, Philogen, Pierre Fabre, and Sanofi/ Regeneron; has received research funding from Bristol Myers Squibb and MSD; has served as a steering committee member for Bristol Myers Squibb, MSD, and Novartis; has served as a coordinating PO for Bristol Myers Squibb, MSD, Novartis, and Pierre Fabre; has served as a local PI for Philogen and Sanofi; and is a member of the board of directors for EORTC-MG. **U. Leiter** has received research support from MSD; has received fees for speaker services from MSD, Novartis, Roche, Sanofi, and Sun Pharma; and has served in consulting or advisory roles for Almiral Hermal, MSD, Novartis, Roche, Sanofi, and Sun Pharma. **F. Meier** has received honoraria for travel support, speaker services, and/or advisory roles from Bristol Myers Squibb, Immunocore, MSD, Novartis, Pierre Fabre, Roche, and Sanofi; and has received research funding from Novartis and Roche. **R. Herbst** reports employment by Helios Klinikum Erfurt, Erfurt, Germany. **M. Sachse** has received speaker honoraria from Novartis; and has received advisory board honoraria from Sanofi Genzyme. **S. Grabbe** has received grants or contracts from Novartis and Pierre Fabre; has received consulting fees from Bristol Myers Squibb, Klinge Pharma, Kyowa Kirin, Novartis, MSD, and Sun Pharma; and has received travel and accommodation expenses from UCB. **C. Gebhardt** reports speaker services for Almirall, Bristol Myers Squibb, Janssen, MSD, Novartis, Roche, Sanofi, Sun Pharma; and Sysmex; consulting or advisory roles for Almirall, BioNTech, Bristol Myers Squibb, Immunocore, MSD, Novartis, Pierre Fabre, Sanofi/Regeneron, and Sun Pharma; and research funding from Novartis and Sanofi/Regeneron; and is a co-founder of Dermagnostix. **F. Ziller** has received fees for speaker services, advisory board honoraria, and/or travel support from Bristol Myers Squibb, MSD, Novartis, Pierre Fabre, Roche, Sanofi, and Sun Pharma. **C. Pföhler** reports employment by the Department of Dermatology, Saarland University Medical School, Homburg/Saar, Germany. **P. Mohr** has received research funding from Almirall Hermal, Amgen Inc., Beiersdorf, Bristol Myers Squibb, Immunocore LLC, Merck, MSD, Novartis Pharmaceuticals, Pfizer, Pierre Fabre, Regeneron Pharmaceuticals, Sun Pharma, and Sanofi Genzyme; has received speaker and advisory board honoraria from Amgen, Bristol Myers Squibb, MSD, Novartis, Pierre Fabre, Sanofi Genzyme, and Sun Pharma; has received travel and accommodation expenses from Bristol Myers Squibb, MSD, Novartis, Pierre Fabre, and Sun Pharma; and has served in a leadership or fiduciary role in other board, society, committee, or advocacy groups for MID Melanom Info Deutschland. **L. Zimmer** has received honoraria from Bristol Myers Squibb, MSD, Pierre Fabre, and Novartis; has received travel and accommodation expenses from Bristol Myers Squibb, MSD, Novartis, Pierre Fabre, and Sun Pharma; has served on a data safety monitoring board or advisory board for Bristol Myers Squibb, MSD, Novartis, and Pierre Fabre; has served in a leadership role for cancer survivorship committee, DeCOG (unpaid); and has received equipment, materials, drugs, medical writing, gifts, or other services from Merck (medical writing). **E. Boutmy** reports employment by Merck. **S. Hoff** reports employment by Merck. **P. Verpillat** reports employment by Merck at time of study. **A. Milano** reports employment by IQVIA Commercial GmbH & Co. OHG, Frankfurt am Main, Germany. **M. Shlaen** reports employment by IQVIA Commercial GmbH & Co. OHG, Frankfurt am Main, Germany. **C. Fu** reports employment by IQVIA Solutions Enterprise Management Consulting (Shanghai) Co., Ltd., Beijing, China. **S. Ugurel** has received research support from Bristol Myers Squibb and Merck; has received speaker and advisory board honoraria from Bristol Myers Squibb, MSD, Merck, and Novartis; and has received travel and accommodation expenses from Bristol Myers Squibb, MSD, and Pierre Fabre. **ACKNOWLEDGMENTS** This study was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945) and was previously conducted under an alliance between Merck and Pfizer. Editorial support was provided by Clinical Thinking and was funded by Merck.

Poster No. 1145P. Presented at the ESMO Congress 2023, 20-24 October 2023; Madrid, Spain.