This is a reprint from the European Society for Medical Oncology Congress 2022 (ESMO 2022), which was originally presented in Paris, France on September 9-13, 2022; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.
There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

Preliminary results from AVENANCE, an ongoing, noninterventional real-world, ambispective study of avelumab first-line maintenance treatment in patients with locally advanced or metastatic urothelial carcinoma

P. Barthélémy,[1] C. Thibault,[2] E. Voog,[3] J. C. Eymard,[4] A. Ravaud,[5] A. Fléchon,[6] C. Abraham,[7] W. Hilgers,[8] S. Le Moulec,[9] M. Chasseray,[10] D. Pouessel,[11] E. Amela,[12] V. Lorgis,[13] E. Nicolas,[14,15] E. Kazan,[16] G. Denechere,[17] M.-N. Solbes,[18] P. Lambert,[17] Y. Loriot[19]

[1]Medical Oncology, Institut de Cancérologie Strasbourg Europe, Strasbourg, France; [2]Department of Medical Oncology, Hôpital Européen Georges Pompidou, Institut du Cancer Paris CARPEM, AP-HP Centre, Université de Paris Cité, Paris, France; [3]ILC Groupe/Clinique Victor Hugo, Le Mans, France; [4]Department of Medical Oncology, Institut de Cancérologie Jean-Godinot, Reims, France; [5]Department of Medical Oncology, Bordeaux University Hospital, Bordeaux University, Bordeaux, France; [6]Department of Medical Oncology, Centre Léon Bérard, Lyon, France; [7]Medical Oncology, Foch Hospital, Suresnes, France; [8]Department of Medical Oncology, Avignon-Provence Cancer Institute, Avignon, France; [9]Medical Oncology, Groupe de Radiothérapie et d'Oncologie des Pyrénées, Clinique Marzet, Pau, France; [10]Centre Finistérien de Radiothérapie et d'Oncologie–Clinique Pasteur, Brest, France; [11]Medical Oncology Department, Institut Claudius Régaud–IUCT Oncopole, Toulouse, France; [12]Centre de Cancérologie Bourgogne, Hôpital Privé Le Bois, Lille, France; [13]Medical Oncology, Institut de Cancérologie de Bourgogne, Dijon, France; [14]Department of Medical Oncology, University Hospital of Nimes, Nimes, France; [15]Institute Desbrest of Epidemiology and Public Health, Research Unit INSERM, University of Montpellier, Montpellier, France; [16]Department of Medical Oncology, Ramsay Health Group–Private Hospital Villeneuve d'Ascq, Villeneuve-d'Ascq, France; [17]Pfizer Oncology, Paris, France; [18]Merck Santé S.A.S., Lyon, France, an affiliate of Merck KGaA; [19]Department of Cancer Medicine, DITEP, Gustave Roussy, Université Paris-Saclay, Villejuif, France

SCOPE

- **This preliminary analysis reports efficacy and safety outcomes from AVENANCE, an ongoing, real-world, ambispective (retrospective and prospective) study evaluating avelumab first-line (1L) maintenance treatment in patients with locally advanced or metastatic urothelial carcinoma (la/mUC) in France**

CONCLUSIONS

- **These first real-world data for avelumab 1L maintenance in patients with la/mUC from the ongoing AVENANCE study support the findings of the JAVELIN Bladder 100 trial[1,2]**
- **These early results (median follow-up, 13.5 months) confirm the clinical activity and acceptable safety profile of avelumab in a heterogeneous population outside of a clinical trial setting**
 - **The 12-month overall survival (OS) rate was 66.9%**
 - **Median progression-free survival (PFS) from the start of avelumab treatment was 5.7 months (95% CI, 5.0-7.9 months) comparable to results from the JAVELIN Bladder 100 trial[2]**
 - **The safety profile was consistent with that observed in other studies of avelumab monotherapy, and no new safety concerns were identified[3]**
- **These results further support the recommendation of avelumab 1L maintenance as standard of care for patients with la/mUC that has not progressed with 1L platinum-based chemotherapy**

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster.

Correspondence: **Philippe Barthélémy**, p.barthelemy@icans.eu

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app to view a plain language summary of the accepted scientific abstract.

BACKGROUND

- In the phase 3 JAVELIN Bladder 100 trial (NCT02603432), avelumab 1L maintenance therapy + best supportive care (BSC) significantly prolonged OS vs BSC alone in patients with la/mUC that had not progressed after 1L platinum-based chemotherapy[1,2]
 - Results from this trial led to the approval of avelumab 1L maintenance in various countries worldwide, and it is now recommended as standard of care in international treatment guidelines, based on level 1 evidence[4-7]
- Longer-term follow-up from JAVELIN Bladder 100 (≥2 years in all patients) continued to show prolonged OS and investigator-assessed PFS in patients treated with avelumab 1L maintenance + BSC vs BSC alone[2]
 - Median OS was 23.8 vs 15.0 months, respectively (hazard ratio [HR], 0.76 [95% CI, 0.631-0.915]; 2-sided p=0.0036), and 2-year OS rates were 49.8% vs 38.4%
 - Median PFS was 5.5 vs 2.1 months, respectively (HR, 0.54 [95% CI, 0.457-0.645]; 2-sided p<0.0001), and 2-year PFS rates were 23.4% vs 7.1%
 - Avelumab 1L maintenance also demonstrated an acceptable long-term safety profile
- AVENANCE is an ongoing, real-world study investigating efficacy and safety in patients with la/mUC treated with avelumab 1L maintenance in France

METHODS

- AVENANCE (NCT04822350) is a multicenter, ambispective, noninterventional study of patients with la/mUC treated with avelumab 1L maintenance in France
- In this ongoing study, eligible patients have previous, ongoing, or planned avelumab 1L maintenance treatment for la/mUC that did not progress after 1L platinum-based chemotherapy (ie, ongoing complete response, partial response, or stable disease)
 - Data collection started on 13 July 2021
- The primary endpoint is OS from start of avelumab treatment
- Secondary endpoints include OS from start of 1L chemotherapy, PFS, duration of treatment, and safety
- In this preliminary analysis, patients who started avelumab ≥6 months prior to data cutoff (31 January 2022) were analyzed
 - Efficacy and safety were analyzed in patients who had received ≥1 dose of avelumab

RESULTS

- This analysis included 267 patients (of 500 planned); baseline characteristics are detailed in **Table 1**
- At data cutoff (31 January 2022), median follow-up since avelumab initiation (by reverse Kaplan-Meier estimation) was 13.5 months (95% CI, 12.8-14.7 months)
- Treatment was ongoing in 92 patients (34.5%)
 - Among the 174/175 patients for whom the reason for discontinuing avelumab was reported, the most common reason was disease progression (n=138 [79.3%])

Table 1. Baseline characteristics

	N=267
Age, median (interquartile range), years	73.1 (66.7-77.9)
Sex, n (%)	
Male	217 (81.3)
Female	50 (18.7)
Location of primary tumor, n (%)	
Bladder	197 (74.1)
Upper urinary tract	52 (19.5)
Urethra	17 (6.4)
Missing data	1
Tumor histology, n (%)	
Pure urothelial carcinoma	243 (93.1)
Urothelial carcinoma with variant (eg, squamous cell, adenocarcinoma, neuroendocrine)	12 (4.6)
Squamous cell carcinoma	5 (1.9)
Other	1 (0.4)
Missing data	6
Tumor status at start of 1L chemotherapy, n (%)	
Locally advanced	26 (9.8)
Metastatic	238 (90.2)
Missing data	3
ECOG PS at start of 1L chemotherapy, n (%)	
0	74 (35.9)
1	103 (50.0)
2	26 (12.6)
3	3 (1.5)
Missing data	61
Type of 1L chemotherapy, n (%)	
Carboplatin + gemcitabine	152 (57.6)
Cisplatin + gemcitabine	84 (31.8)
Other (including MVAC)	28 (10.6)
Missing data	3
No. of 1L chemotherapy cycles, median (range)	5 (1-10)
Response to last chemotherapy, n (%)	
Complete response	57 (21.8)
Partial response	146 (55.9)
Stable disease	53 (20.3)
Disease progression	1 (0.4)
Non-evaluable	4 (1.5)
Missing data	6
Presence of visceral metastasis at start of 1L chemotherapy, n (%)	**n=238**
No	41 (17.3)
Yes	196 (82.7)
Missing data	1
Metastasis sites at start of 1L chemotherapy, n (%)	**n=196**
Lymph nodes	114 (58.2)
Liver	35 (17.9)
Lung	57 (29.1)
Bone	71 (36.2)
Brain	1 (0.5)
Other	38 (19.4)

Percentages reported were calculated using the denominator of patients with available data for each characteristic.
1L, first line; **MVAC**, methotrexate, vinblastine, doxorubicin, and cisplatin.

- The 12-month OS rate from start of avelumab was 66.9% (95% CI, 60.5%-72.5%) (**Figure 1**) and from start of 1L chemotherapy was 79.1% (95% CI, 73.5%-83.6%; n=262 evaluable)
- Median PFS from start of avelumab was 5.7 months (95% CI, 5.0-7.9 months), and the 12-month PFS rate was 36.9% (95% CI, 30.8%-43.1%) (**Figure 2**)
- Median duration of avelumab treatment was 5.8 months (95% CI, 4.9-7.4 months)

Figure 1. OS from start of avelumab treatment

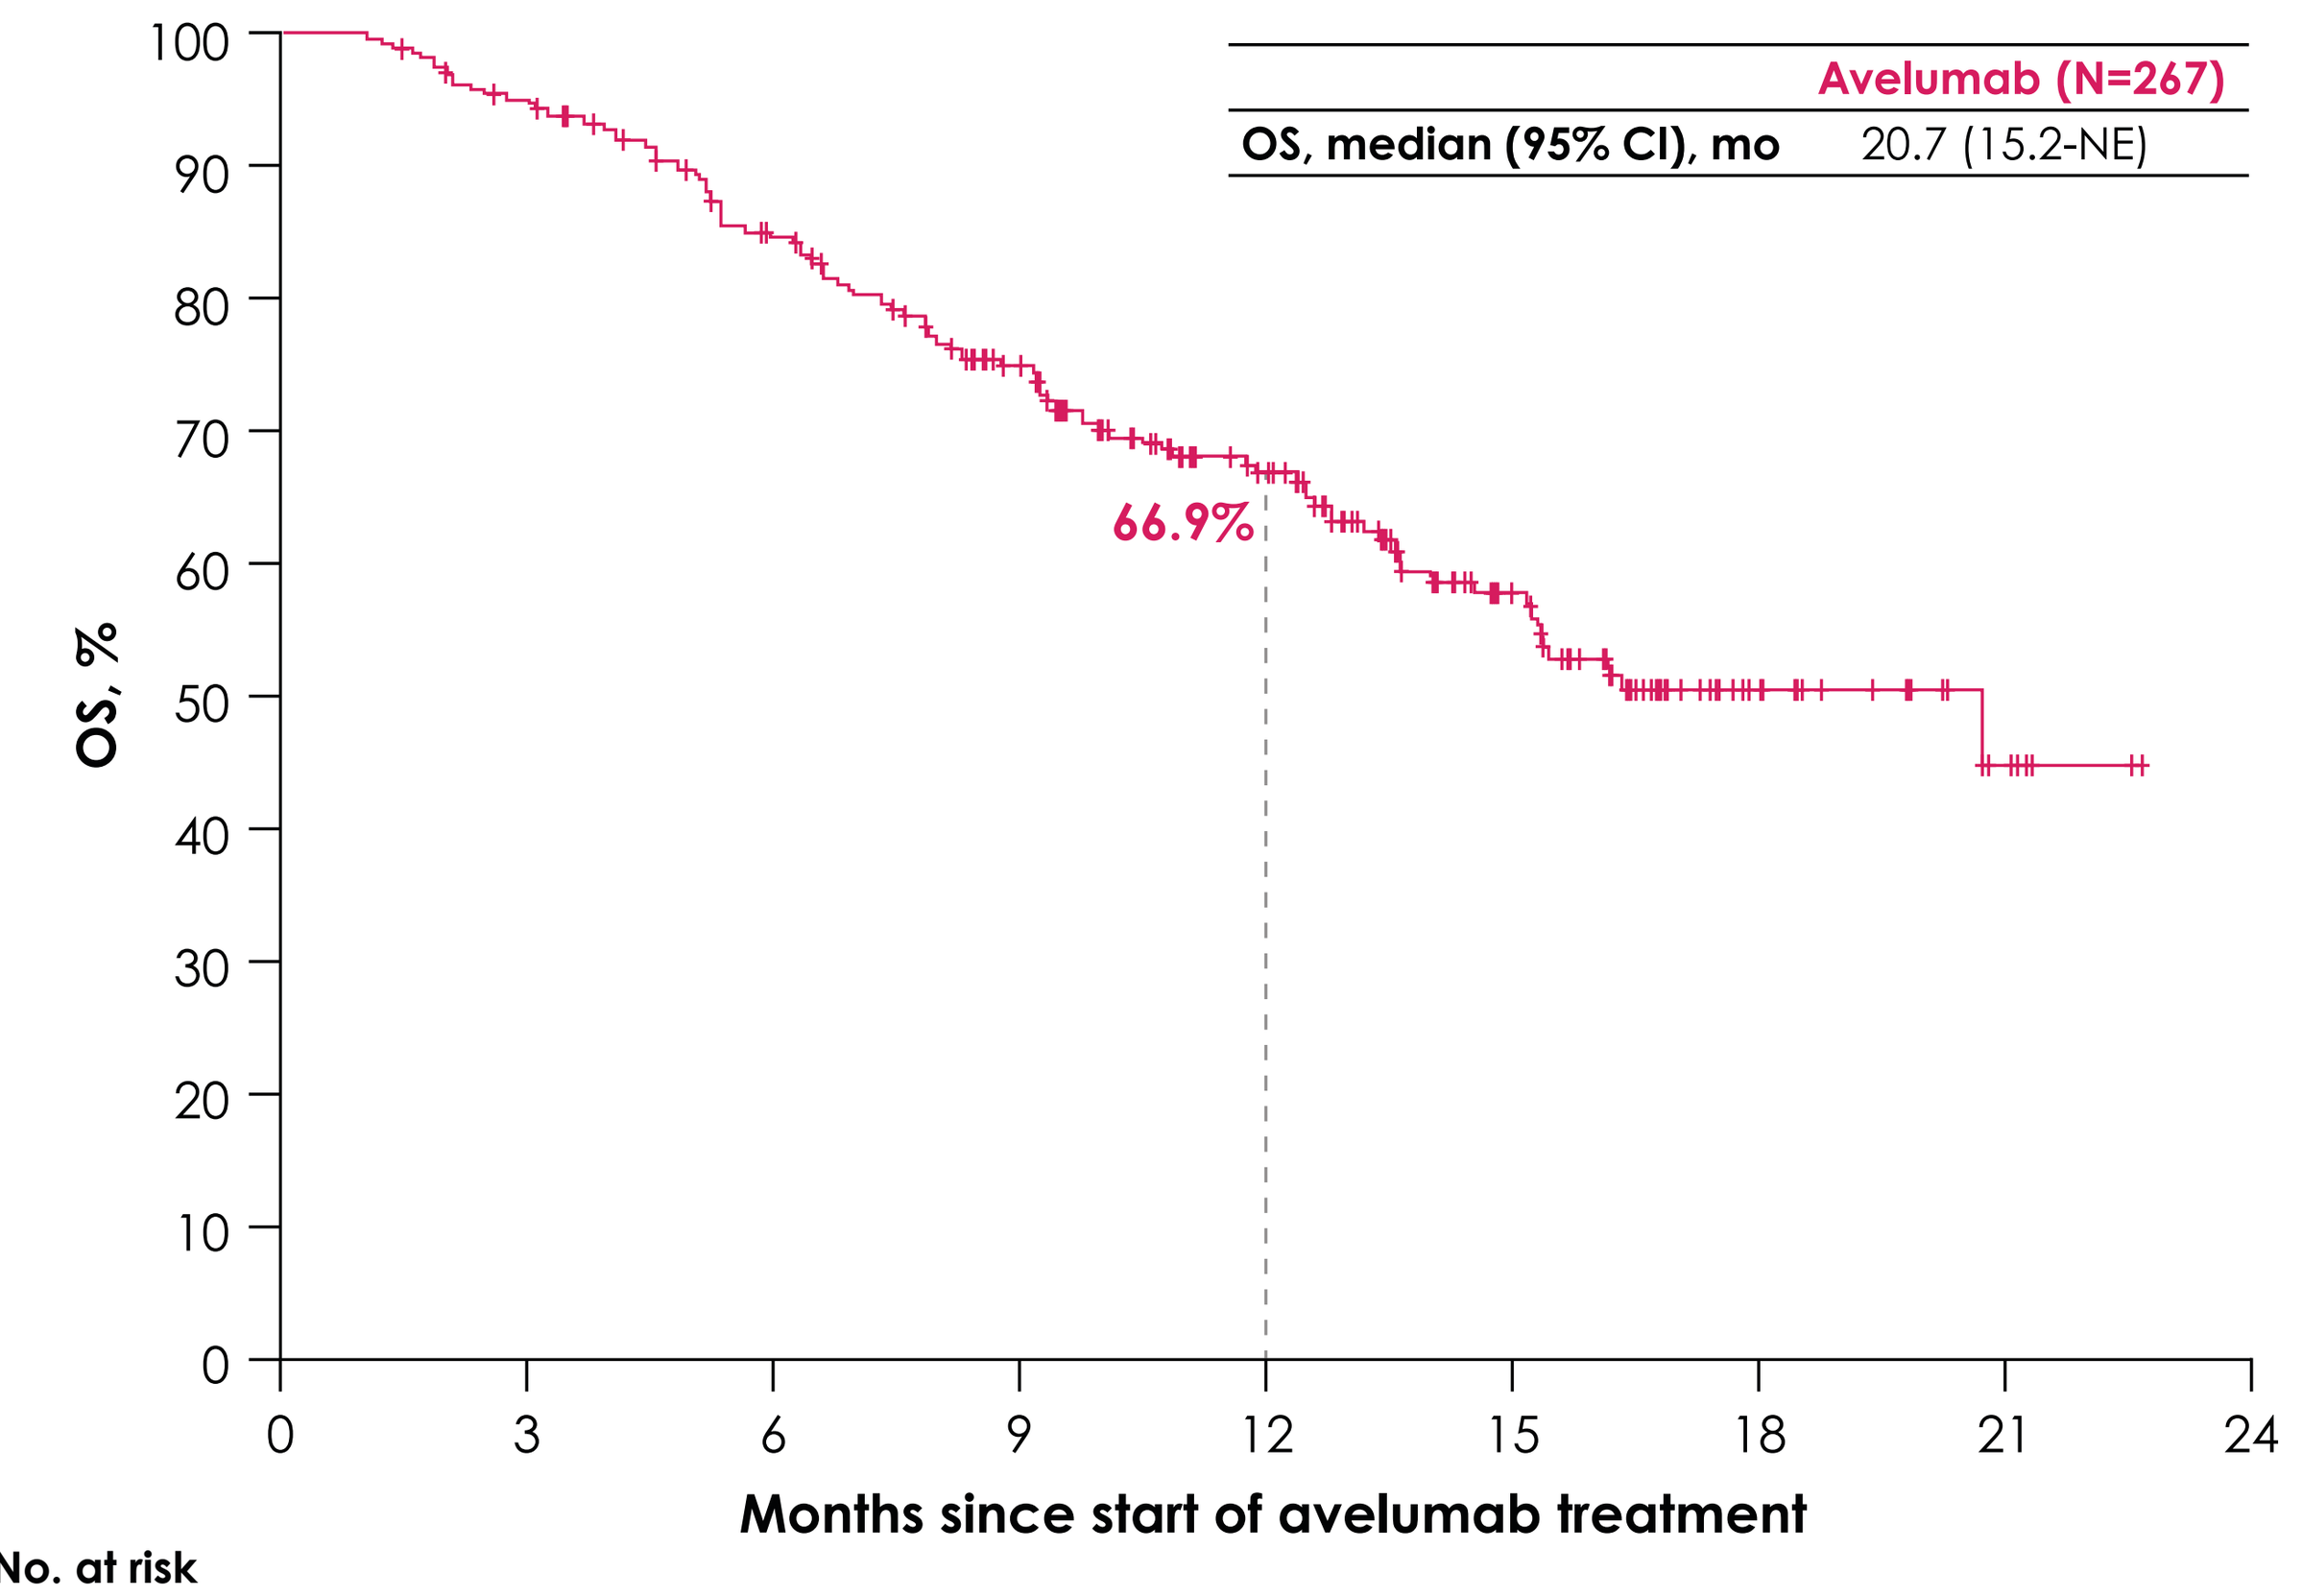

Avelumab (N=267)
OS, median (95% CI), mo
20.7 (15.2-NE)
66.9%
OS, %
0
10
20
30
40
50
60
70
80
90
100
0
3
6
9
12
15
18
21
24
Months since start of avelumab treatment

No. at risk (No. of censored patients)									
Avelumab	267 (0)	250 (3)	214 (13)	175 (28)	116 (70)	60 (113)	21 (145)	6 (159)	0 (165)

NE, not estimable; **OS**, overall survival.

Figure 2. PFS from start of avelumab treatment

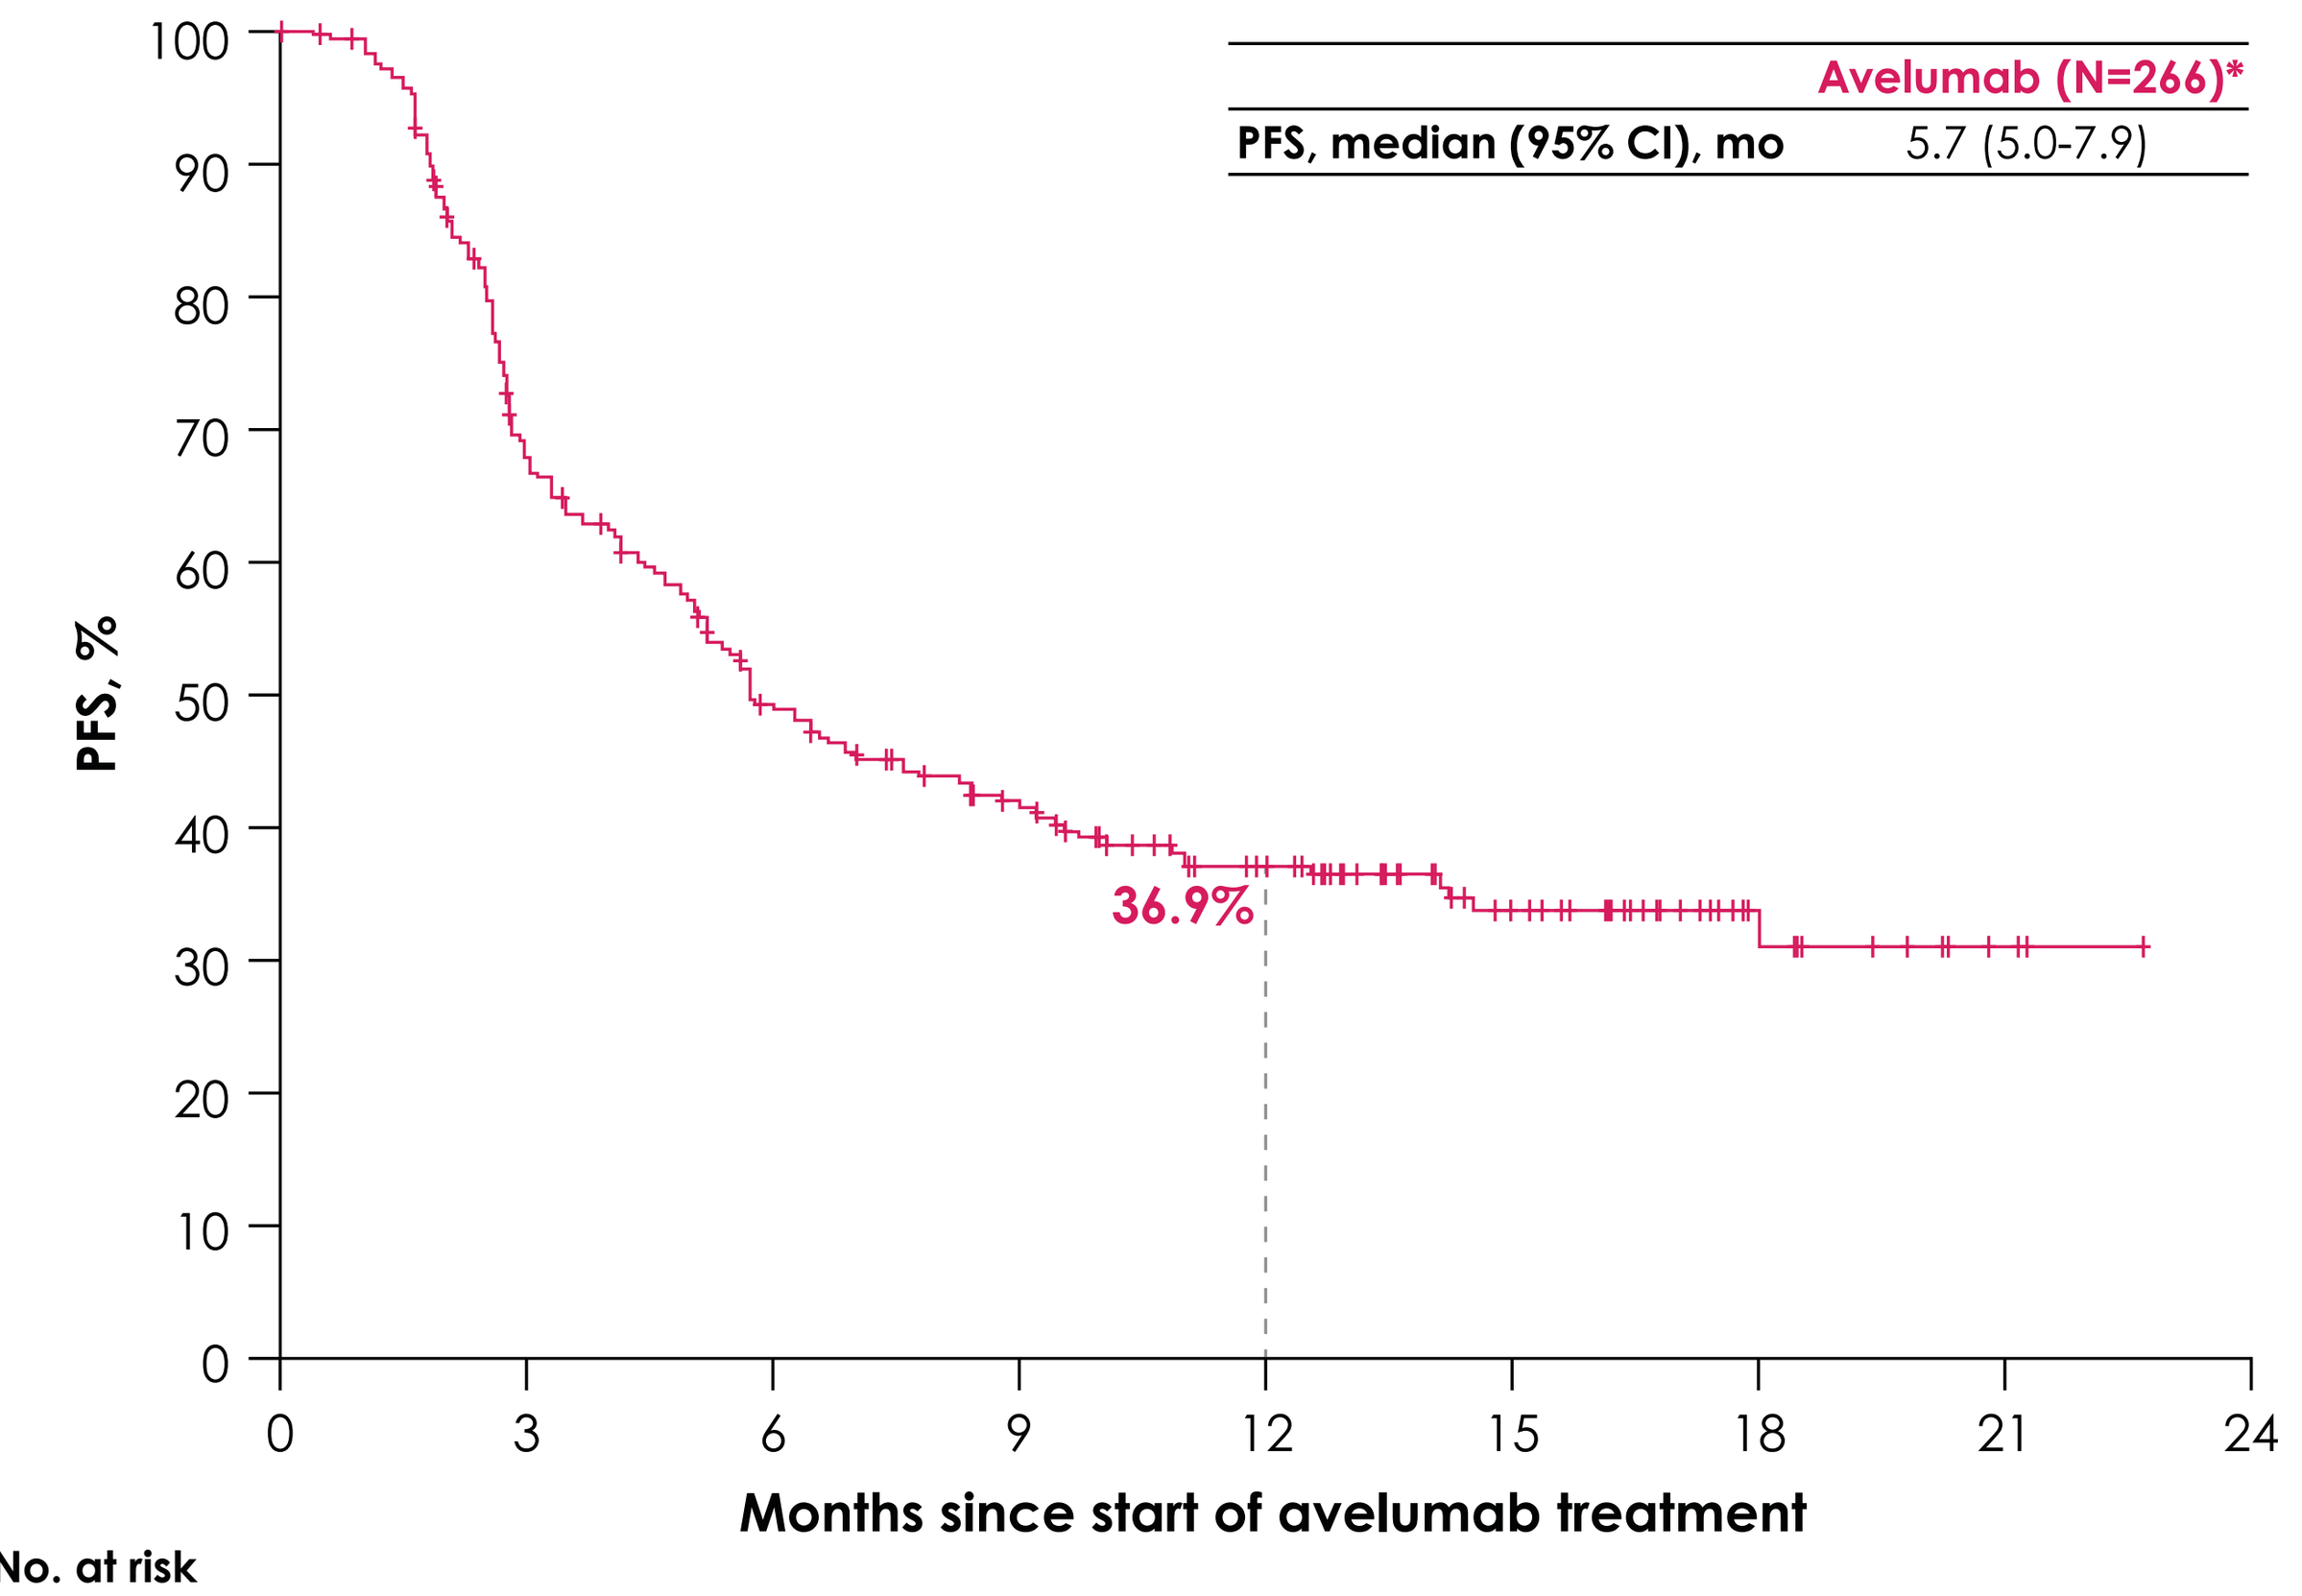

Avelumab (N=266)*
PFS, median (95% CI), mo
5.7 (5.0-7.9)
36.9%
PFS, %
0
10
20
30
40
50
60
70
80
90
100
0
3
6
9
12
15
18
21
24
Months since start of avelumab treatment

No. at risk (No. of censored patients)									
Avelumab	266 (0)	171 (12)	117 (20)	92 (28)	63 (47)	32 (74)	12 (94)	3 (102)	0 (105)

PFS, progression-free survival.
*1 patient has been excluded from this analysis because of an incorrect date of first injection.

- 112 patients (41.9%) reported receiving subsequent anticancer treatment (including second-line and later) (**Table 2**)
- Safety data are summarized in **Table 3**
 - Treatment-emergent adverse events (TEAEs) occurred in 170 patients (63.7%), with serious TEAEs in 75 patients (28.1%)
 - The most common TEAEs (in >5% of patients) were asthenia (n=35 [13.1%]) and pruritus (n=29 [10.9%])

Table 2. Subsequent treatment (second-line and later)

Subsequent treatment, n (%)	n=112
Non–platinum-based chemotherapy	71 (63.4)
Paclitaxel monotherapy	59 (52.7)
Paclitaxel + gemcitabine	4 (3.6)
Gemcitabine monotherapy	5 (4.5)
Docetaxel monotherapy	1 (0.9)
Vinflunine monotherapy	2 (1.8)
Platinum-based chemotherapy	24 (21.4)
Carboplatin + gemcitabine	12 (10.7)
Carboplatin monotherapy	4 (3.6)
Carboplatin + paclitaxel	4 (3.6)
Carboplatin + etoposide	1 (0.9)
Cisplatin + paclitaxel	1 (0.9)
Cisplatin + gemcitabine	1 (0.9)
MVAC	1 (0.9)
Enfortumab vedotin*	12 (10.7)
Sacituzumab govitecan*	3 (2.7)
Pembrolizumab	2 (1.8)

MVAC, methotrexate, vinblastine, doxorubicin, and cisplatin.
*Not approved for use in the EU at the time of study.

Table 3. Summary of AEs

Events, n (%)	N=267
TEAE	170 (63.7)
Serious TEAE	75 (28.1)
TEAE leading to temporary/permanent discontinuation	72 (27.0)
TEAE leading to death	31 (11.6)
TRAE	102 (38.2)
Serious TRAE	14 (5.2)
TRAE leading to temporary/permanent discontinuation	26 (9.7)
TRAE leading to death	1 (0.4)

AE, adverse event; **TEAE**, treatment-emergent adverse event; **TRAE**, treatment-related adverse event.

REFERENCES 1. Powles T, et al. N Engl J Med. 2020;383(13):1218-30. **2.** Powles T, et al. J Clin Oncol. 2022;40(6 Suppl). Abstract 487. **3.** Kelly K, et al. Cancer. 2018;124(9):2010-17. **4.** NCCN Clinical Practice Guidelines. Bladder Cancer. V2.2022. **5.** Powles T, et al. Ann Oncol. 2022;33(3):244-58. **6.** Cathomas R, et al. Eur Urol. 2022;81(1):95-103. **7.** Japanese Urological Association. Supplemental to Clinical Practice Guidelines for Bladder Cancer (2019). Accessed August 15, 2022. https://www.urol.or.jp/lib/files/other/guideline/39_bladder_cancer_2019_rev2021_info.pdf.

DISCLOSURES P. Barthélémy has served in advisory roles for Amgen, Astellas, Bayer, Bristol Myers Squibb, Ipsen, Janssen Cilag, Merck, MSD, Novartis, and Pfizer and reports speaker services for AstraZeneca and Seagen. **C. Thibault** has received institutional research funding from AstraZeneca and Sanofi; has served in advisory roles for AstraZeneca, Bristol Myers Squibb, Ipsen, Merck, MSD, and Pfizer; and reports speaker services for Astellas, AstraZeneca, Bristol Myers Squibb, Ipsen, Janssen, MSD, and Sanofi. **E. Voog** has nothing to declare. **J. C. Eymard** has received personal and institutional research funding from Bristol Myers Squibb, Ipsen, and Pfizer and has served in advisory roles for Astellas, Ipsen, Janssen, and Sanofi. **A. Ravaud** has received institutional research funding from Ipsen, Merck, and Pfizer; has served in advisory roles for AstraZeneca, Bristol Myers Squibb, Eisai, Ipsen, Merck, and Pfizer; and has received travel and accommodation expenses from AstraZeneca, Bristol Myers Squibb, Eisai, Ipsen, Merck, and Pfizer. **A. Fléchon** has received honoraria from Gilead Sciences, Janssen, Merck, MSD, and Pfizer. **C. Abraham** has nothing to declare. **W. Hilgers** has nothing to declare. **S. Le Moulec** has nothing to declare. **M. Chasseray** has nothing to declare. **D. Pouessel** has received institutional research funding from MSD; has served in advisory roles for Astellas, AstraZeneca, Merck, MSD, and Pfizer; reports speaker services for Astellas, AstraZeneca, Bristol Myers Squibb, Ipsen, Janssen, MSD, and Pfizer; and has received travel and accommodation expenses from MSD and Pfizer. **E. Amela** has nothing to declare. **V. Lorgis** has nothing to declare. **E. Nicolas** has served in an advisory role for Merck. **E. Kazan** has nothing to declare. **G. Denechere** reports employment with Pfizer. **M.-N. Solbes** reports employment with Merck Santé S.A.S., Lyon, France, an affiliate of Merck KGaA. **P. Lambert** reports employment with Pfizer. **Y. Loriot** has served in consulting or advisory roles for Astellas, AstraZeneca, Bristol Myers Squibb, Gilead Sciences, Janssen, Merck, MSD, Pfizer, Roche, Seagen, and Taiho; has served as a steering committee member for Astellas, Basilea, Gilead Sciences/Immunomedics, Janssen, MSD, and Taiho; has served as a local principal investigator for Astellas, AstraZeneca, Bristol Myers Squibb, Exelixis, Gilead Sciences, Incyte, Janssen, Merck, MSD, and Pfizer; has served as a coordinating principal investigator for Janssen; and has received institutional research funding from Celsius, Janssen, MSD, Roche, and Sanofi. **ACKNOWLEDGMENTS** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by Pfizer as part of an alliance between Pfizer and Merck (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Eleanor Bishop of ClinicalThinking and was funded by Pfizer and Merck.

Poster No. 1757P. Presented at the ESMO Congress 2022, 9-13 September 2022; Paris, France; Hybrid.

Please note that this summary only contains information from the full scientific abstract

Avelumab first-line maintenance treatment in people with advanced urothelial cancer: early results from the real-world AVENANCE study in France

View scientific abstract

The full title of this abstract is: Preliminary results from AVENANCE, an ongoing, noninterventional real-world, ambispective study of avelumab first-line (1L) maintenance treatment in patients (pts) with locally advanced or metastatic urothelial carcinoma (la/mUC)

Please note this summary only contains information from the scientific abstract

View scientific abstract

Date of summary: September 2022

For more information on this study, go to: https://clinicaltrials.gov/ct2/show/NCT04822350

Pronunciations

Medical terms pronunciations

Avelumab <a-VEL-yoo-mab> **Urothelial** <YOOR-oh-THEE-lee-ul>

Key takeaways

What are the key takeaways from this summary?

- Avelumab first-line maintenance treatment, which is given after first-line chemotherapy, helped people with advanced urothelial cancer live longer than people who were not treated with avelumab in the JAVELIN Bladder 100 clinical trial
 - A plain language summary of the overall results from the JAVELIN Bladder 100 clinical trial is available at this link
- In the real-world AVENANCE study, researchers looked at how effective avelumab first-line maintenance treatment is in people with advanced urothelial cancer in France in a day-to-day setting. They also looked at side effects related to the treatment
 - These people were treated in hospitals or clinics in the community and were not part of a clinical trial setting
- They found that on average, people in the AVENANCE study lived for 6 months without their cancer getting worse and that 64% of people were still alive 1 year after starting avelumab first-line maintenance treatment
- Side effects reported in this study were similar to those reported in the JAVELIN Bladder 100 clinical trial
- AVENANCE is the first study of avelumab first-line maintenance treatment outside of a clinical trial and confirms the findings of the JAVELIN Bladder 100 clinical trial
- Overall, the results from this study provide more support for using avelumab first-line maintenance treatment as a standard treatment for people with advanced urothelial cancer that does not get worse after first-line chemotherapy

Background

What did this study look at?

What is advanced urothelial cancer?

- Urothelial cancer is a cancer that develops in the urinary tract
- The urinary tract contains the parts of the body that move urine from the kidneys to the outside of the body. It includes:
 - The bladder
 - The inner part of the kidneys
 - Tubes that connect the kidneys to the bladder and the bladder to the outside of the body
- Urothelial cancer is called advanced when it has spread outside of the urinary tract

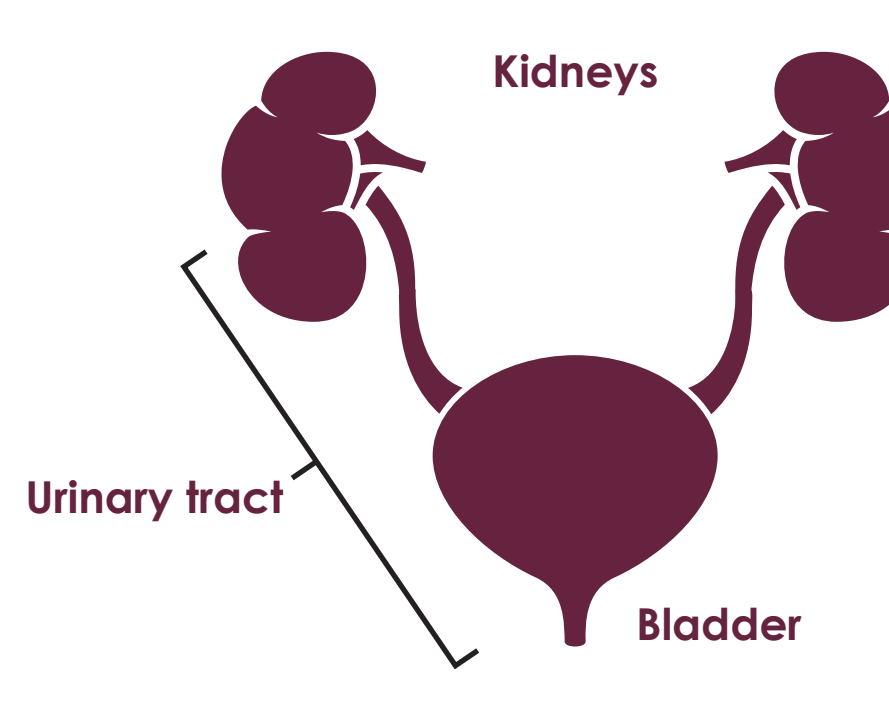
Kidneys
Urinary tract
Bladder

How is advanced urothelial cancer usually treated?

- Chemotherapy is often the first main treatment given to people with advanced urothelial cancer. This is called first-line treatment
- Although the cancer may get better with chemotherapy at first, it is likely to start growing again
- If a person's cancer stops growing or shrinks after first-line chemotherapy, they may then receive a different treatment. This is called maintenance treatment. It aims to stop the cancer from getting worse or coming back

What is avelumab?

Avelumab is a type of immunotherapy. Immunotherapy can help the body's immune system find and destroy cancer cells. Avelumab is given as a drip (infusion) into a vein for about an hour once every 2 weeks

Results from the JAVELIN Bladder 100 clinical trial have shown that avelumab first-line maintenance treatment can help people with advanced urothelial cancer live longer. A plain language summary of the overall results is available at this link

Avelumab is the only approved maintenance treatment available for people with advanced urothelial cancer that has stopped growing or shrunk with first-line chemotherapy

What is the JAVELIN Bladder 100 clinical trial?

- The JAVELIN Bladder 100 clinical trial looked at avelumab first-line maintenance treatment in people with advanced urothelial cancer in various countries worldwide
- All people taking part in the clinical trial had received first-line chemotherapy, and their cancer had disappeared, shrunk, or stopped growing. They were put into 2 treatment groups:
 - Treatment group 1 received avelumab first-line maintenance treatment plus best supportive care. Best supportive care includes treatments that help to manage symptoms but do not affect the cancer
 - Treatment group 2 received only best supportive care
- Researchers found that, on average, people who were treated with avelumab plus best supportive care lived longer than people who received only best supportive care

What is the AVENANCE study?

- The AVENANCE study is looking at avelumab first-line maintenance treatment for people with advanced urothelial cancer in France
- AVENANCE is a real-world study. This means that information is collected from people receiving normal day-to-day treatment. Real-world studies help to show how a drug works in a day-to-day setting, outside of a clinical trial
- All people taking part in the study received first-line chemotherapy and their cancer had disappeared, shrunk, or stopped growing. They then received avelumab first-line maintenance treatment

What did the researchers want to find out?

Researchers wanted to look at early results from the real-world AVENANCE study. They wanted to see if avelumab first-line maintenance treatment helped people with advanced urothelial cancer in France to live longer in day-to-day use, outside of a clinical trial setting

Study design

What happened during the study?

Who took part in the study?

People with advanced urothelial cancer who received first-line chemotherapy in France → **About 500 people** whose cancer has not gotten worse will receive avelumab maintenance treatment → Results were reported from the **first 267 people who took part**

- Researchers collected early results from the AVENANCE study in January 2022
- At this time, 267 people with advanced urothelial cancer had started avelumab treatment at least 6 months before results were collected
- On average, people had taken part in the study for 11 months

Aims of this summary

What did the researchers look at?

- Researchers looked at the following:
 - How long people lived overall
 - How long people lived without their cancer getting worse
 - How long people received avelumab treatment for
 - How many people had side effects

Results

What were the results of the study?

What were the characteristics of people before they were treated with avelumab?

Before receiving chemotherapy

- **90%** had cancer that had spread to other parts of the body
- **10%** had cancer that had spread to nearby tissues only
- **86%** were in good physical shape
- **14%** had difficulty doing physical activities in their daily life

After receiving chemotherapy

- Average age was **73 years**
- **22%** had cancer that became undetectable
- **56%** had cancer that had shrunk
- **20%** had cancer that had stopped growing

What were the outcomes after people were treated with avelumab?

- **6 MONTHS** was how long people received avelumab treatment for, on average
- **38%** of people were still receiving avelumab treatment when results were collected
- **6 MONTHS** was how long people lived without their cancer getting worse, on average
- **64%** of people were still alive after 12 months

What were people's side effects after being treated with avelumab?

Any side effect	Serious* side effects	Side effects that caused avelumab treatment to be stopped
53%	22%	22%

*A side effect is called serious if it is life threatening, needs or lengthens hospital care, needs emergency treatment, causes lasting problems, or is fatal

Conclusions

What were the main conclusions reported by the researchers?

- The results from the real-world AVENANCE study are the first results for avelumab first-line maintenance treatment in normal day-to-day use, outside of a clinical trial setting
- Results from AVENANCE confirm the findings of the JAVELIN Bladder 100 clinical trial
- These early results show the effectiveness and safety of avelumab in a real-world group of people with advanced urothelial cancer
- The AVENANCE study is ongoing, and updated results will be reported in the future

Disclaimers

Avelumab is approved to treat the condition that is discussed in this summary. This summary reports the results of a single study. The results of this study may differ from those of other studies. Health professionals should make treatment decisions based on all available evidence, not on the results of a single study. This study described is still ongoing, therefore the final outcomes of this study may differ from the outcomes described in this summary

Study sponsors

Who sponsored this study?

Pfizer
235 East 42nd Street
New York, NY 10017, USA
Phone (United States): +1 212-733-2323

Merck Healthcare KGaA
Frankfurter Strasse 250
Darmstadt, 64293, Germany
Phone (Germany): +49 6151 720

The sponsors would like to thank all of the people who took part in this study

More information

Where can I find more information?

For more information on this study, please visit:
ESMO Congress 2022 Scientific Abstract
https://clinicaltrials.gov/ct2/show/NCT04822350

For more information on clinical studies in general, please visit:
https://www.clinicaltrials.gov/ct2/about-studies/learn
https://www.cancer.org/treatment/treatments-and-side-effects/clinical-trials.html

Writing support for this summary was provided by Eleanor Bishop of ClinicalThinking and was funded by Pfizer and Merck (CrossRef Funder ID: 10.13039/100009945)