Avelumab first-line maintenance therapy for locally advanced/metastatic urothelial carcinoma: results from the real-world US PATRIOT-II study

P. Grivas,[1] P. Barata,[2] H. Moon,[3] S. Gupta,[4] T. Hutson,[5] C. N. Sternberg,[6] J. Brown,[7] V. Dave,[8] C. Downey,[8] A. C. Shillington,[9] H. M. Katzenstein,[10]* M. Kirker,[11] S. Hanson,[11] F. X. Liu,[10]* V. Morris,[10] A. Bhanegaonkar,[10] G. P. Sonpavde[12]

[1]University of Washington; Fred Hutchinson Cancer Center, Seattle, WA, USA; [2]University Hospitals Seidman Cancer Center, Cleveland, OH, USA; [3]Kaiser Permanente Southern California, Riverside, CA, USA; [4]Taussig Cancer Institute, Cleveland Clinic Foundation, Cleveland, OH, USA; [5]Texas Oncology, Dallas, TX, USA; [6]Weill Cornell Medicine, Hematology/Oncology, New York, NY, USA; [7]Division of Oncology, University Hospitals Seidman Cancer Center, Cleveland, OH, USA; [8]RTI Health Solutions, Research Triangle Park, NC, USA; [9]EPI-Q, Inc, Oak Brook, IL, USA; [10]EMD Serono, Rockland, MA, USA; [11]Pfizer, New York, NY, USA; [12]AdventHealth Medical Group, Orlando, FL, USA

*Affiliation at the time the study was conducted.

CONCLUSIONS

- **The PATRIOT-II study examined real-world outcomes, treatment patterns, and healthcare resource utilization (HCRU) prior to and during avelumab first-line maintenance (1LM) in patients with locally advanced or metastatic urothelial carcinoma (la/mUC) in routine clinical practice in the United States**
- **Real-world overall survival (OS) and progression-free survival (PFS) results presented here are in line with the findings from the JAVELIN Bladder 100 phase 3 trial of avelumab 1LM, and no new safety concerns were identified**
 - **In this patient cohort without disease progression after 1L PBC, real-world OS measured from the start of 1L PBC was 30.5 months (95% CI, 23.4-37.6); this was similar to the OS of 29.7 months (95% CI, 25.2-34.0) in JAVELIN Bladder 100**
- **Limitations of studies using real-world data include lack of central imaging review, missing or unknown data, potential selection bias, and confounding factors due to the lack of randomization**
- **These real-world results further support the use of avelumab 1LM in patients with la/mUC who are progression free following 1L PBC**

PLAIN LANGUAGE SUMMARY

- **Based on clinical trial results, avelumab switch maintenance is the recommended treatment for people with advanced urothelial cancer whose cancer disappeared, shrank, or stopped growing with first-line chemotherapy**
- **The PATRIOT-II study examined outcomes in people with advanced urothelial cancer in the US who received avelumab maintenance treatment after chemotherapy outside of a clinical trial**
 - **Results were similar to clinical trial results, including how long people lived, how long they lived without their cancer getting worse, and safety findings (how well people tolerated therapy)**
- **The positive patient outcomes from this study provide support for using avelumab maintenance as a standard treatment for people with advanced urothelial cancer whose cancer disappeared, shrank, or stopped growing with first-line chemotherapy**

BACKGROUND

- UC (cancer arising in any part of the urinary tract) accounts for approximately 90% of all bladder cancers; the 5-year survival rate is 38% for locally advanced UC and 5% for metastatic UC[1,2]
 - The incidence rate in the US is approximately 18.2 cases per 100,000 per year[3]
- PBC is a standard-of-care 1L treatment for eligible patients with la/mUC; however, despite high initial response rates, durable responses are uncommon, and most patients ultimately experience disease progression in a median of approximately 9 months after the start of 1L PBC[4-6]
- Avelumab is an anti–PD-L1 antibody that was approved by the FDA in June 2020 for use as 1LM treatment for patients with la/mUC that has not progressed with 1L PBC[7,8]
- Avelumab was approved based the results of the JAVELIN Bladder 100 phase 3 trial (NCT02603432)[7,9]
 - After ≥2 years of follow-up, median OS (23.8 vs 15.0 months) and PFS (5.5 vs 2.1 months) were significantly improved with avelumab 1LM + best supportive care (BSC) vs BSC alone[9]
 - Any-grade treatment-emergent AEs (TEAEs) occurred in 338 patients (98%), including grade ≥3 TEAEs in 185 patients (54%)[9]
 - Any-grade treatment-related AEs (TRAEs) occurred in 269 patients (78%), including grade ≥3 TRAEs in 67 patients (19%)[9]

METHODS

- PATRIOT-II was a US-based, retrospective chart review study conducted in 37 geographically dispersed sites consisting of both community oncology practices and centers affiliated with academic institutions (**Figure 1**)
- The medical records of 160 patients were reviewed and extracted
- Patients were aged ≥18 years at diagnosis, had histologically confirmed la/mUC without progression following 1L PBC, and had started avelumab 1LM treatment
- Patients were followed from the start of PBC until 52 weeks after the start of avelumab, death, or end of study, whichever occurred first
- Data collected included baseline demographics, disease and cancer characteristics, PBC treatment response, real-world OS and PFS, AEs related to avelumab, and HCRU, including inpatient and emergency department (ED) visits before and after the start of avelumab 1LM
- All analyses were descriptive
 - Time-to-event outcomes were examined using Kaplan-Meier analysis and life tables, censoring for the end of study or loss to follow-up in patients not meeting a terminal endpoint
 - HCRU was analyzed using an incidence-based approach as observed events per person-year of follow-up

Figure 1. PATRIOT-II observational study design

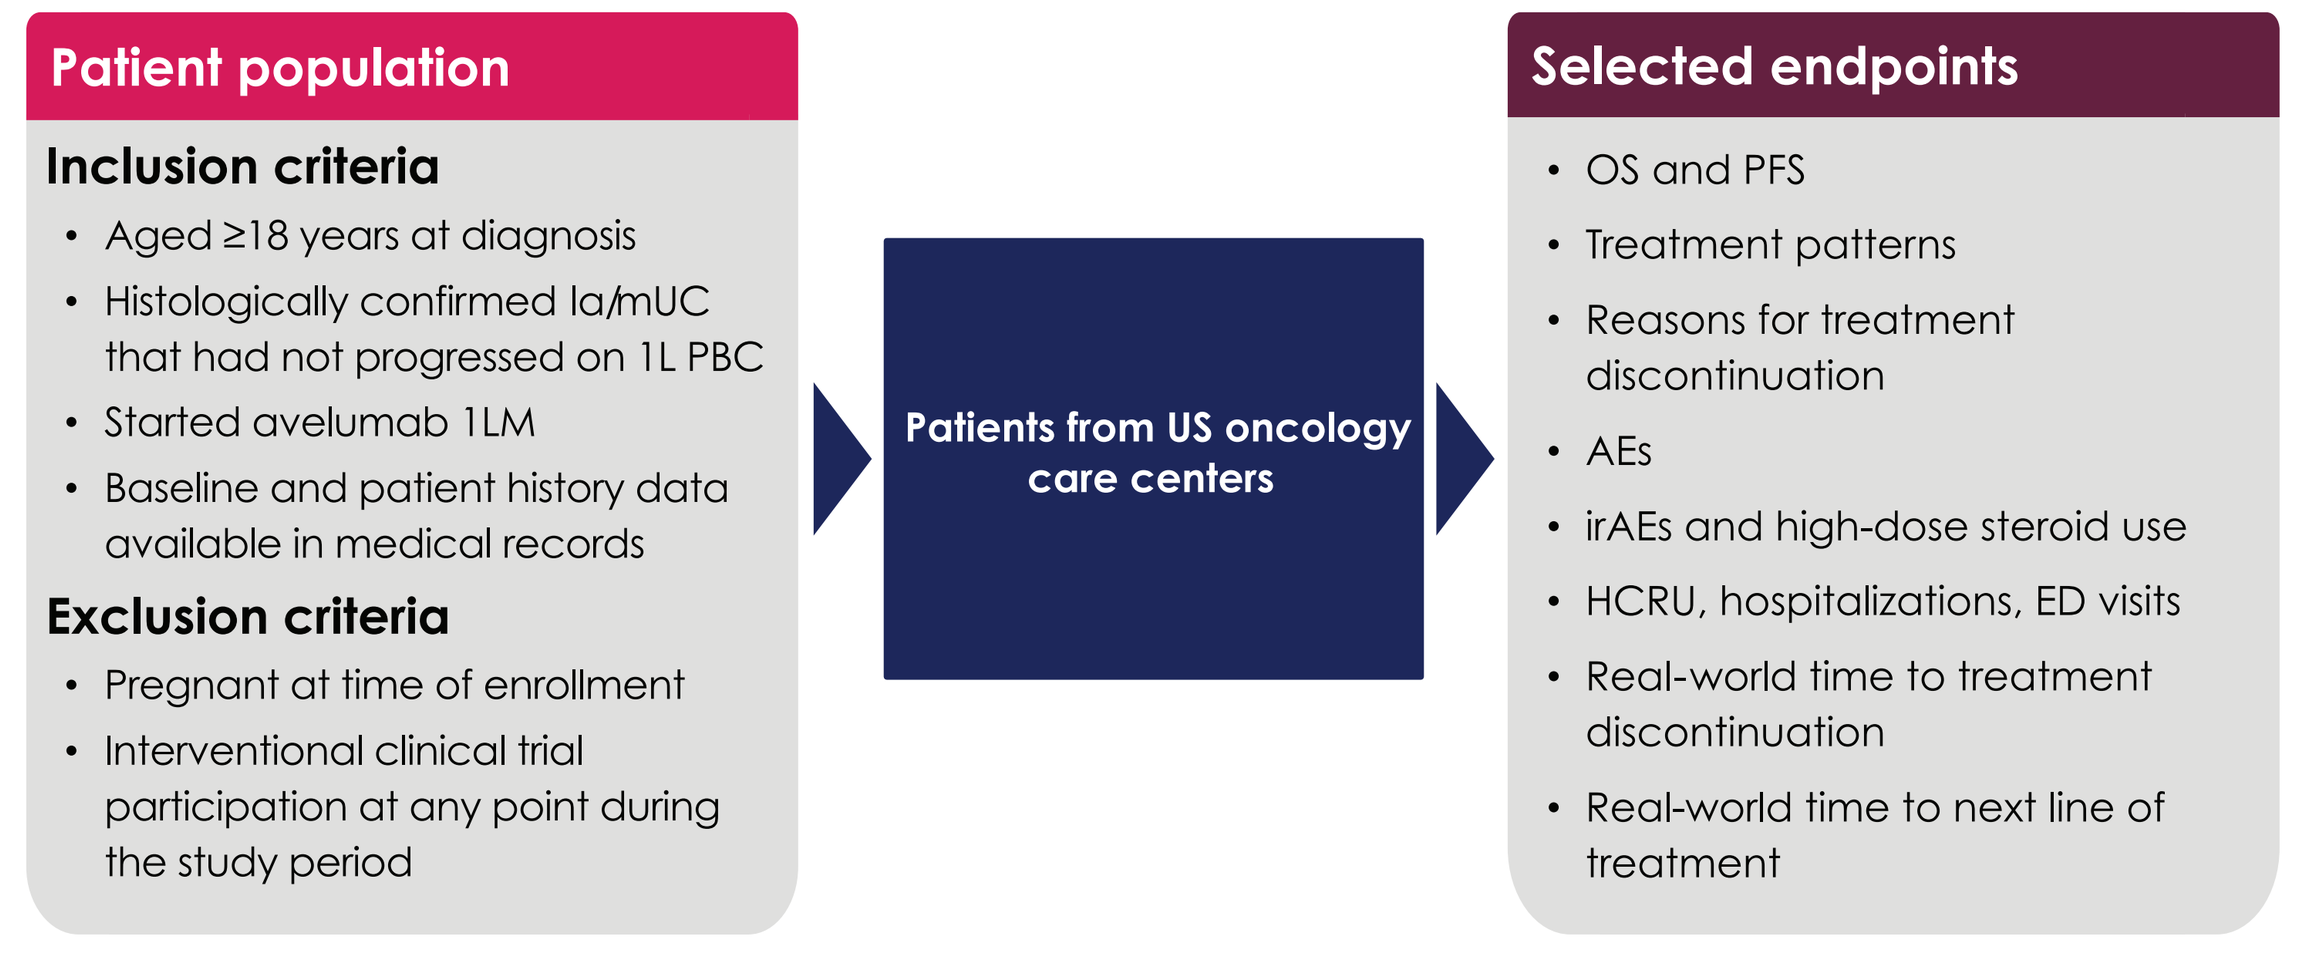

Patient population
Inclusion criteria
Aged ≥18 years at diagnosis
Histologically confirmed la/mUC that had not progressed on 1L PBC
Started avelumab 1LM
Baseline and patient history data available in medical records
Exclusion criteria
Pregnant at time of enrollment
Interventional clinical trial participation at any point during the study period
Patients from US oncology care centers
Selected endpoints
OS and PFS
Treatment patterns
Reasons for treatment discontinuation
AEs
irAEs and high-dose steroid use
HCRU, hospitalizations, ED visits
Real-world time to treatment discontinuation
Real-world time to next line of treatment

1L, first line; **1LM**, first-line maintenance; **AE**, adverse event; **ED**, emergency department; **HCRU**, healthcare resource utilization; **irAE**, immune-related adverse event; **la/mUC**, locally advanced/metastatic urothelial carcinoma; **OS**, overall survival; **PBC**, platinum-based chemotherapy; **PFS**, progression-free survival.

RESULTS

- 160 patients were enrolled following completion of 1L PBC
 - 100 patients (62%) received 1L cisplatin, and 60 patients (38%) received 1L carboplatin
 - 130 patients (81%) had an investigator-assessed response to 1L PBC (complete response, 21 [13%]; partial response, 109 [68%]; **Table 1**)
 - Patients started avelumab 1LM a median of 4 weeks after completing 1L PBC
 - Median follow-up from start of avelumab 1LM was 16 months (interquartile range, 11-21 months)
- At the end of follow-up, 31 patients (19%) were continuing to receive avelumab treatment, 118 patients (74%) had discontinued (due to progression, AEs, and/or ECOG performance status deterioration), and the remainder were lost to follow-up
- Median real-world PFS from the start of avelumab was 5.4 months (95% CI, 3.8-6.9 months) (**Figure 2**), and median real-world OS was 24.4 months (95% CI, 20.4-28.4 months) (**Figure 3**)
- In this patient cohort without disease progression after 1L PBC, median real-world OS from the start of 1L PBC was 30.5 months (95% CI, 23.4-37.6 months) (**Figure 4**)

Table 1. Baseline characteristics at the start of 1L PBC

	All patients (N=160)		All patients (N=160)
Age, median (range), years	70 (40-90)	**PD-L1 status, n (%)**	
Sex, n (%)		Positive	44 (27.5)
Male	123 (76.9)	Negative	33 (20.6)
Female	37 (23.1)	Unknown	83 (51.9)
Race, n (%)		**Site of metastasis at the start of 1L PBC, n (%)**	
White	143 (89.4)	Visceral*	70 (44)
Black	5 (3.1)	Nonvisceral	51 (32)
Asian	4 (2.5)	None	23 (14)
American Indian/ Alaskan native	1 (0.6)	Unknown	16 (10)
Other/unknown	7 (4.4)	**1L PBC regimen**	
Ethnicity, n (%)		Cisplatin†, n (%)	100 (62.5)
Hispanic or Latino	7 (4.4)	No. of cycles, median (IQR)	4 (3-6)
Non-Hispanic or Latino	125 (78.1)	Carboplatin + gemcitabine, n (%)	60 (37.5)
Unknown	28 (17.5)	No. of cycles, median (IQR)	5 (4-6)
ECOG performance status, n (%)		**Best response to 1L PBC, n (%)**	
0	68 (42.5)	Complete response or partial response	130 (81.3)
1	53 (33.1)	Stable disease	17 (10.6)
≥2	12 (7.5)	Unknown	13 (8.1)
Unknown	27 (16.9)		
Creatinine clearance, n (%)			
≥60 mL/min	64 (40.0)		
<60 mL/min	66 (41.3)		
Unknown	30 (18.8)		

Percentages may not add up to 100 because of rounding.
1L, first line; **IQR**, interquartile range; **PBC**, platinum-based chemotherapy.
*Does not include bone. †Encompasses any cisplatin-based regimen.

Figure 2. Real-world PFS from the start of avelumab 1LM

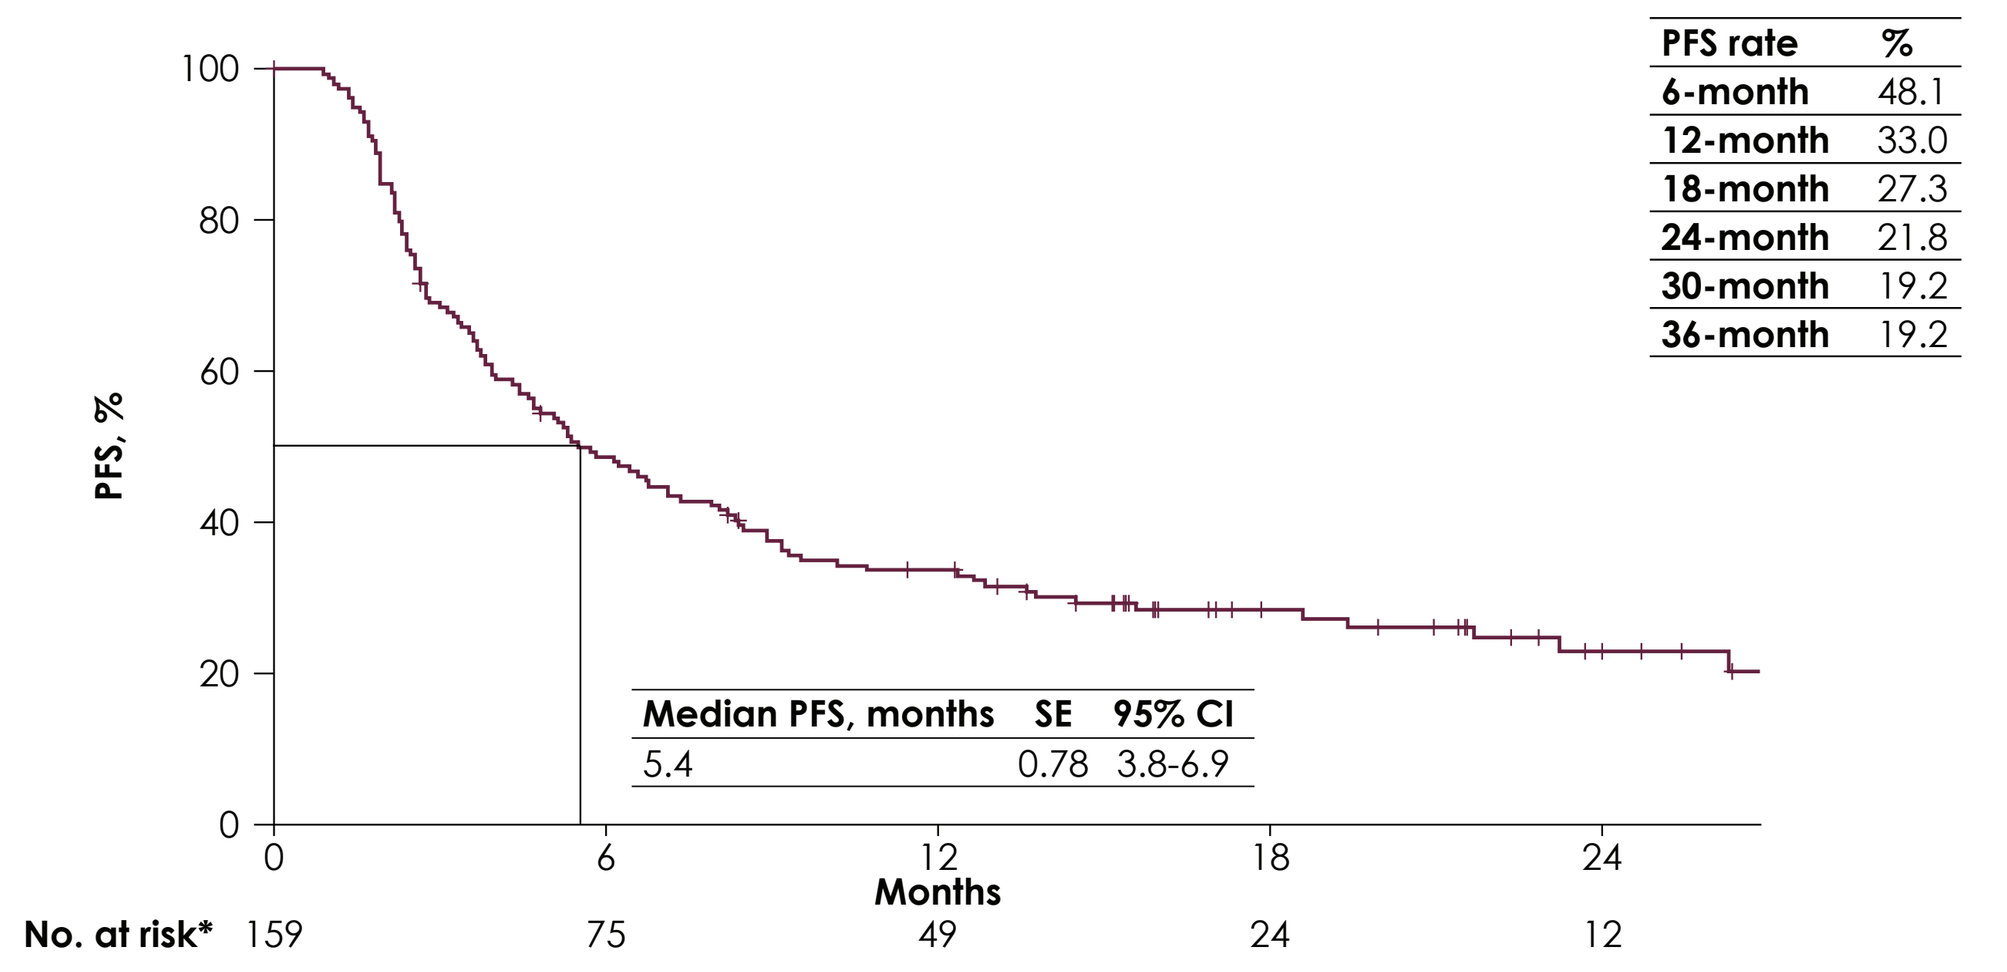

PFS rate %
6-month 48.1
12-month 33.0
18-month 27.3
24-month 21.8
30-month 19.2
36-month 19.2
Median PFS, months SE 95% CI
5.4 0.78 3.8-6.9
PFS, %
Months
No. at risk* 159 75 49 24 12

1LM, first-line maintenance; **PFS**, progression-free survival.
*Date of progression was not available for 1 patient. Progression status was unknown for 3 patients; these patients were censored at the most recent follow-up date.

Figure 3. Real-world OS from the start of avelumab 1LM

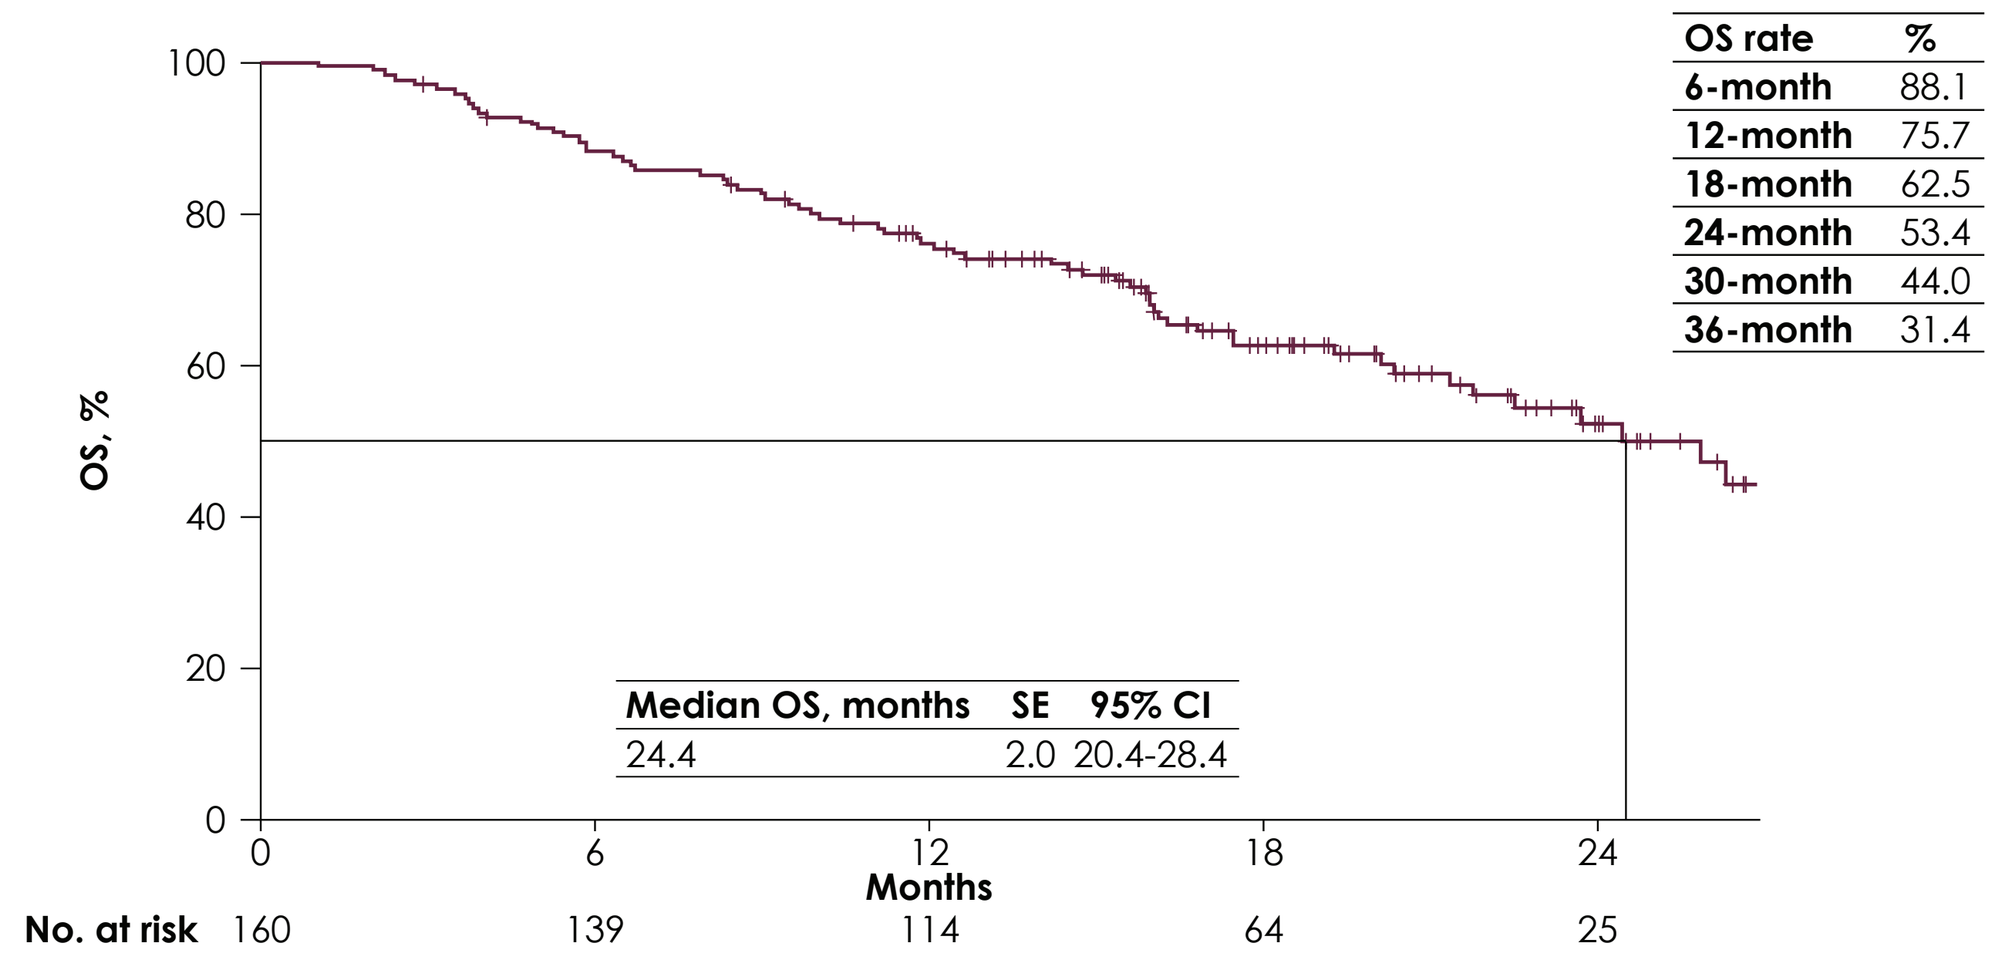

OS rate %
6-month 88.1
12-month 75.7
18-month 62.5
24-month 53.4
30-month 44.0
36-month 31.4
Median OS, months SE 95% CI
24.4 2.0 20.4-28.4
OS, %
Months
No. at risk 160 139 114 64 25

1LM, first-line maintenance; **OS**, overall survival.

Figure 4. Real-world OS from the start of 1L PBC

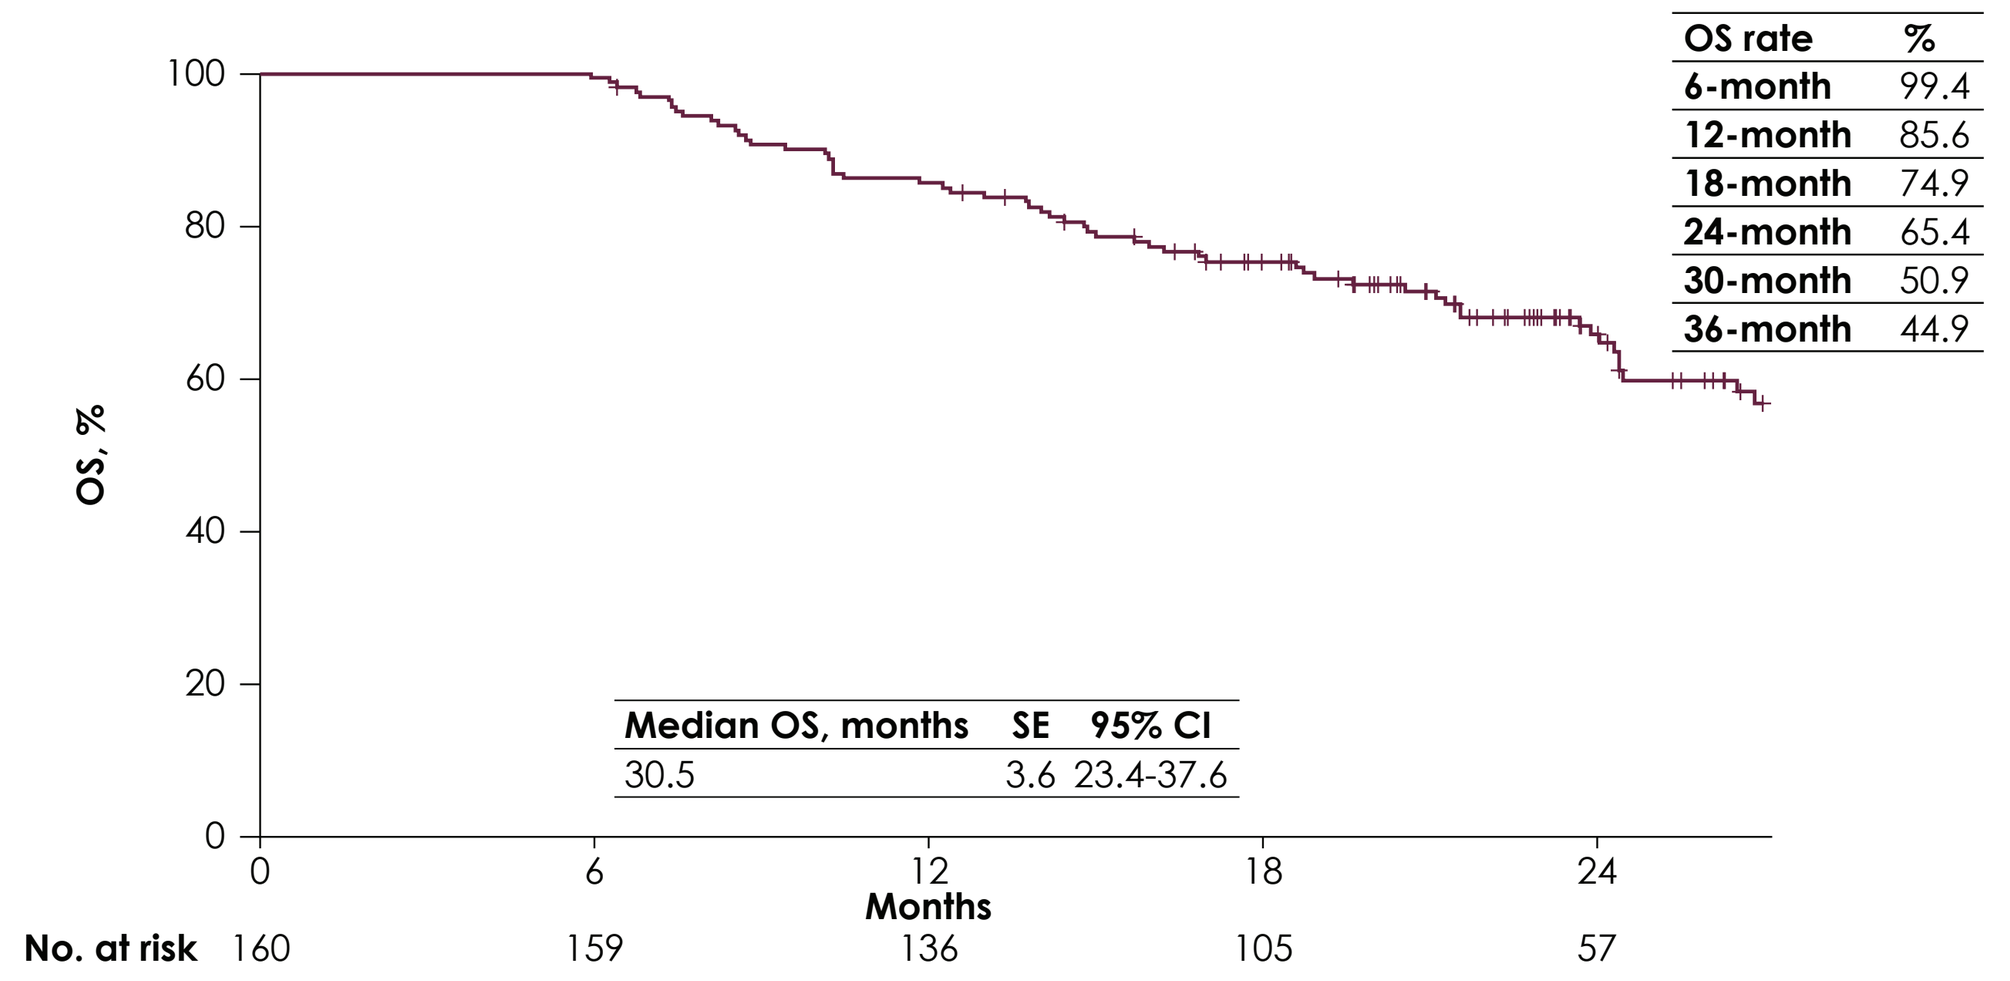

OS rate %
6-month 99.4
12-month 85.6
18-month 74.9
24-month 65.4
30-month 50.9
36-month 44.9
Median OS, months SE 95% CI
30.5 3.6 23.4-37.6
OS, %
Months
No. at risk 160 159 136 105 57

This analysis of OS measured from the start of 1L PBC in this selected population without progression on 1L PBC should be interpreted with caution.
1L, first line; **OS**, overall survival; **PBC**, platinum-based chemotherapy.

- 62 patients (39%) had an AE related to avelumab; 35 (22%) had an immune-related AE; 23 (14%) received high-dose steroids due to any TRAE; 13 (8%) were hospitalized due to a TEAE; and 16 (10%) stopped therapy due to an AE (**Table 2**)
 - The rate of AEs related to avelumab was lower than the rate reported in the JAVELIN Bladder 100 trial (78%); this is likely because AEs were not prospectively solicited in this real-world study
- Most common TEAEs were fatigue (n=7 [4%]), hypothyroidism (n=7 [4%]), anemia (n=6 [4%]), infusion-related reaction (n=6 [4%]), nausea (n=6 [4%]), elevated creatinine (n=5 [3%]), diarrhea (n=4 [3%]), and rash (n=4 [3%]) (**Table 3**)

Table 2. Summary of safety for avelumab 1LM

	All patients (N=160)
Any TRAE, n (%)	62 (38.8)
Time to onset, mean (SD), days	95 (127)
Median (range)	56 (0-793)
Any immune-related AE, n (%)	35 (21.9)
Time to onset, mean (SD), days	146 (173)
Median (range)	91 (0-793)
Therapy stopped due to any TRAE, n (%)	16 (10.0)
Therapy delayed due to any TRAE, n (%)	20 (12.5)
Received steroid (including topical) due to any TRAE, n (%)	36 (32.1)
Received high-dose systemic steroid due to any TRAE, n (%)	23 (14.3)
TRAE(s) outcome, n (%)	**N=165***
Resolved	105 (63.6)
Unresolved	32 (19.4)
Resolved with sequelae	2 (1.2)
Unknown	26 (15.8)
Duration of TRAE(s), days†	**N=165***
Mean (SD)	97 (151)
Median (range)	31 (0-657)
Hospitalized due to TRAE, n (%)	
Yes	13 (8.1)
No	147 (91.9)

1LM, first-line maintenance; **AE**, adverse event; **TRAE**, treatment-related adverse event.
*N represents number of AEs, duplicated in data file. †Most recent follow-up date used if unresolved.

Table 3. Most commonly observed AEs related to avelumab

Patients, n (%)	All patients (N=160)
Fatigue	7 (4.4)
Hypothyroidism	7 (4.4)
Anemia	6 (3.8)
Infusion-related reaction	6 (3.8)
Nausea	6 (3.8)
Blood creatinine increased	5 (3.1)
Diarrhea	4 (2.5)
Rash	4 (2.5)
Hypocalcemia	3 (1.9)
Hyponatremia	3 (1.9)
Muscular weakness	3 (1.9)
Neuropathy peripheral	3 (1.9)
Pneumonitis	3 (1.9)
Psoriasis	3 (1.9)
Vomiting	3 (1.9)

AE, adverse event.

- During the avelumab treatment period, 44 patients (28%) were hospitalized for a mean of 11.5 days per person-year. Avelumab-related AEs were the reason for admission in 13 (8%), with the remainder of admissions being related to disease progression, surgical procedures, and comorbidities (**Table 4**)
- 25 patients (16%) were hospitalized for a mean of 3 days per person-year during the PBC treatment period (**Table 4**)
- During the avelumab treatment period, 12 patients (8%) required an ED visit, and 11 patients (7%) had an ED visit during the PBC treatment period (**Table 5**)
- HCRU was observed to be higher during and 90 days after the avelumab treatment period than during the 1L platinum-based chemotherapy (PBC) treatment period, most likely due to the impact of several potential confounders, including cancer progression

Table 4. Inpatient hospitalizations during PBC and avelumab 1LM treatment periods

	PBC treatment period (N=160)*	Avelumab 1LM treatment period (N=160)*
Mean duration (SD), weeks	22.9 (20.6)	38.2 (30.7)
Median duration (range), weeks	19.4 (8.1-174.1)	25.1 (2.1-121.4)
Any hospitalization, n (%)†	25 (15.6)	44 (27.5)
Frequency of hospitalization, n (%)		
0	135 (84.4)	116 (72.5)
1	18 (11.3)	22 (13.8)
≥2	7 (4.4)	22 (13.8)
Inpatient days per patient-year‡		
Mean (SD)	2.7 (8.4)	11.5 (37.4)
Median (range)	0 (0-65)	0 (0-274)
Any ICU stay during hospitalization, n (%)	**n=25**	**n=44**
	0	4 (9.1)

1LM, first-line maintenance; **AE**, adverse event; **ED**, emergency department; **ICU**, intensive care unit; **PBC**, platinum-based chemotherapy; **SD**, standard deviation.
*The PBC treatment period was defined as the time between the start of 1L PBC and the start of avelumab 1LM. The avelumab treatment period was defined as the time between the start of avelumab and its discontinuation + 90 days to allow for HCRU related to late-emerging AEs, death if within 90 days of discontinuation, or start of second-line therapy. †Hospitalization, including those who were admitted via ED. ‡Including all patients with no hospitalization. This is an annualized length of stay.

Table 5. ED visits without hospitalization during PBC and avelumab 1LM treatment periods

	PBC treatment period (N=160)*	Avelumab 1LM treatment period (N=160)*
Any ED visit, n (%)	11 (6.9)	12 (7.5)
Frequency of ED visits, n (%)		
0	149 (93.1)	148 (92.5)
1	11 (6.9)	8 (5.0)
≥2	0	4 (2.5)
ED visits per patient-year†		
Mean (SD)	0.2 (0.9)	0.4 (2.3)
Median (range)	0 (0-6)	0 (0-24)

1LM, first-line maintenance; **AE**, adverse event; **ED**, emergency department; **PBC**, platinum-based chemotherapy; **SD**, standard deviation
*The PBC treatment period was defined as the time between the start of 1L PBC and the start of avelumab 1LM. The avelumab treatment period was defined as the time between the start of avelumab and its discontinuation + 90 days to allow for HCRU related to late-emerging AEs, death if within 90 days of discontinuation, or the start of second-line therapy. †Including patients with no visits. These data show annualized visits.

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Petros Grivas**, pgrivas@uw.edu

REFERENCES 1. Cancer.net. Bladder cancer: introduction. Accessed December 6, 2023. https://www.cancer.net/cancer-types/bladder-cancer/introduction **2.** Cancer.net. Bladder cancer: statistics. Accessed December 6, 2023. https://www.cancer.net/cancer-types/bladder-cancer/statistics **3.** National Cancer Institute. Cancer Stat Facts: bladder cancer. Accessed December 6, 2023. https://seer.cancer.gov/statfacts/html/urinb.html **4.** von der Maase H, et al. J Clin Oncol. 2000;17:3068-77. **5.** De Santis M, et al. J Clin Oncol. 2012;30:191-9. **6.** Sternberg CN, et al. Eur J Cancer. 2006;42:50-4. **7.** Powles T, et al. N Engl J Med. 2020;383:1218-30. **8.** Bavencio (avelumab). Prescribing information. EMD Serono, Rockland, MA, USA; 2022. **9.** Powles T, et al. J Clin Oncol. 2023;41:3486-92.

DISCLOSURES P. Grivas has served in consulting or advisory roles for the healthcare business of Merck KGaA, Darmstadt, Germany, Bristol Myers Squibb, AstraZeneca, Seagen, Pfizer, Janssen, Roche, Dyania Health, 4D Pharma, Astellas Pharma, Guardant Health, UroGen Pharma, Gilead Sciences, Silverback Therapeutics, BostonGene, Fresenius Kabi, Lucence, PureTech, G1 Therapeutics, Aadi, CG Oncology, Strata Oncology, ImmunityBio, and Asieris Pharmaceuticals; and has received institutional research funding from Pfizer, Clovis Oncology, Bavarian Nordic, Bristol Myers Squibb, Debiopharm Group, the healthcare business of Merck KGaA, Darmstadt, Germany, QED Therapeutics, GSK, Mirati Therapeutics, G1 Therapeutics, Gilead Sciences, Acrivon Therapeutics, and ALX Oncology. **P. Barata** has served in consulting or advisory roles for AVEO Oncology, Astellas Pharma, AstraZeneca, Bayer, Bristol Myers Squibb, Exelixis, Janssen, Pfizer, the healthcare business of Merck KGaA, Darmstadt, Germany, Eisai, Caris Life Sciences, Clovis Oncology, Dendreon, UroToday, and Seagen. **H. Moon** has served in consulting or advisory roles for Bayer, Seagen, the healthcare business of Merck KGaA, Darmstadt, Germany, and Pfizer and has received institutional research funding from Janssen, the healthcare business of Merck KGaA, Darmstadt, Germany, Seagen, Bristol Myers Squibb, Apollomics, and Nektar. **S. Gupta** has served in consulting or advisory roles for Bristol Myers Squibb, Gilead Sciences, Guardant Health, the healthcare business of Merck KGaA, Darmstadt, Germany, Pfizer, Merck & Co., Kenilworth, NJ, Bayer, and Seagen; has reported speakers services for Bristol Myers Squibb, Janssen Oncology, Seagen, and Gilead Sciences; and reports stock or other ownership in Nektar, Moderna, and BioNTech. **T. Hutson** is an employee of Texas Oncology; has served in consulting or advisory roles for Bayer/Onyx, Pfizer, Novartis, Astellas Pharma, Johnson & Johnson, Bristol Myers Squibb, Eisai, and Exelixis; has reported speakers services for Pfizer, Johnson & Johnson, Eisai, Exelixis, Astellas Pharma, and Bristol Myers Squibb; has received honoraria from Pfizer, Astellas Pharma, Bristol Myers Squibb, Exelixis, Eisai, Novartis, Johnson & Johnson, and Bayer/Onyx; and has received research funding from Pfizer, Johnson & Johnson, Exelixis, Eisai, and Bristol Myers Squibb. **C. N. Sternberg** has served in consulting or advisory roles for Bayer, Merck & Co., Kenilworth, NJ, Pfizer, Roche, Amgen, Incyte, Bristol Myers Squibb, AstraZeneca, the healthcare business of Merck KGaA, Darmstadt, Germany, Medscape, UroToday, Astellas Pharma, Genzyme, Gilead Sciences, and Foundation Medicine. **J. Brown** has reported speakers services for the healthcare business of Merck KGaA, Darmstadt, Germany; has received honoraria from AstraZeneca; and has received research funding from Bicycle Therapeutics, Jounce Therapeutics, Merck & Co., Kenilworth, NJ, Novita Pharmaceuticals, and Seagen. **V. Dave** reports no conflicts of interest. **C. Downey** reports no conflicts of interest. **A. C. Shillington** reports stock or other ownership in Johnson & Johnson, AbbVie, Lilly, and EPI-Q and has received research funding from Seagen, the healthcare business of Merck KGaA, Darmstadt, Germany, Sunovion, Janssen Scientific Affairs, Merck & Co., Kenilworth, NJ, Neurocrine Biosciences, Amolyt Pharma, ChemoCentryx, Pfizer, Bristol Myers Squibb, United Therapeutics, and Acceleron Pharma. **H. M. Katzenstein** reported employment by EMD Serono, Rockland, MA, USA at the time of the study. **M. Kirker** reports employment by, stock or other ownership in, and has received honoraria from Pfizer. **S. Hanson** reports employment by and stock or other ownership in Pfizer. **F. X. Liu** reports employment by EMD Serono, Rockland, MA, USA, at the time of the study and reports stock or other ownership in the healthcare business of Merck KGaA, Darmstadt, Germany. **V. Morris** reports employment by EMD Serono, Rockland, MA, USA. **A. Bhanegaonkar** reports employment by EMD Serono, Rockland, MA, USA, at the time of the study; reports stock or other ownership in the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel, accommodations, and expenses from the healthcare business of Merck KGaA, Darmstadt, Germany. **G. P. Sonpavde** has served in consulting or advisory roles for Genentech, Eisai, AstraZeneca, Janssen, Bristol Myers Squibb, Exelixis, the healthcare business of Merck KGaA, Darmstadt, Germany, Astellas Pharma, Bicycle Therapeutics, Pfizer, Seagen, Gilead Sciences, Scholar Rock, and G1 Therapeutics; has reported speakers services for Physicians' Education Resource, OncLive, Research to Practice, and Medscape; has received travel and accommodations expenses from Bristol Myers Squibb; has other relationships with AstraZeneca, Bristol Myers Squibb, Astellas Pharma, Bavarian Nordic, Debiopharm Group, QED Therapeutics, and Elsevier; has received honoraria from UpToDate; and has received research funding from Janssen, Sanofi, AstraZeneca, Gilead Sciences, and QED Therapeutics. **ACKNOWLEDGMENTS** This study was sponsored by EMD Serono (CrossRef Funder ID: 10.13039/100004755) and was previously conducted under an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany, and Pfizer. Editorial support was provided by Jamie Ratcliffe of Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 697. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.