Real-world treatment patterns, sequencing, and outcomes in patients with locally advanced or metastatic urothelial carcinoma receiving avelumab first-line maintenance in the United States

H. H. Moon,[1] M. Kearney,[2] S. H. Mahmoudpour,[2] C. Ike,[3] V. Morris,[3] A. Rava,[4] S. Kim,[4] H. Sun,[4] M. Boyd,[4] G. Gomez Rey[4]

[1]Kaiser Permanente, Riverside, CA, USA; [2]the healthcare business of Merck KGaA, Darmstadt, Germany; [3]EMD Serono, Rockland, MA, USA; [4]Genesis Research Group, Hoboken, NJ, USA

CONCLUSIONS

- This study aimed to describe baseline demographic and clinical characteristics, real-world (rw) treatment patterns and sequencing following progression, and clinical outcomes in patients with locally advanced or metastatic urothelial carcinoma (la/mUC) in the US who received avelumab first-line (1L) maintenance (1LM) following treatment with 1L platinum-based chemotherapy (PBC)
- Clinical outcomes in this study are aligned with those of the JAVELIN Bladder 100 trial, further supporting avelumab 1LM after 1L PBC as a standard of care in la/mUC
 - Median rw progression-free survival (rwPFS) and rw overall survival (rwOS) from start of avelumab 1LM were 5.1 months (95% CI, 4.1-7.0 months) and 23.8 months (95% CI, 18.2 months-not estimable [NE]), respectively
- This study provides insights into the sequencing and effectiveness of second-line (2L) enfortumab vedotin (EV) in patients with la/mUC who are treated with avelumab 1LM and have disease control after 1L PBC
 - In patients who received 2L EV after avelumab 1LM, median rwPFS and rwOS from 2L initiation were 4.9 months (95% CI, 3.9-8.8 months) and 11.2 months (95% CI, 6.8 months-NE), respectively
- As novel treatments for la/mUC are approved and incorporated into clinical practice, further research is needed to understand the optimal treatment sequencing to maximize clinical outcomes in the rw setting

PLAIN LANGUAGE SUMMARY

- Based on clinical trial results, avelumab maintenance is considered a standard treatment for people with advanced urothelial cancer that disappeared, shrank, or stopped growing with platinum chemotherapy given as initial treatment
- In this study, researchers looked at how long people with advanced urothelial cancer lived after starting avelumab maintenance in the US outside of a clinical trial, and how long they lived depending on the type of treatment they received when avelumab was stopped
- On average, people treated with avelumab maintenance lived for 5 months without their cancer getting worse and for 24 months overall, which is similar to results from a previous clinical trial of avelumab maintenance
- 45% of people received a second treatment when avelumab was stopped, and the most common was a drug called enfortumab vedotin
 - After starting treatment with enfortumab vedotin, people lived on average for 5 months without their cancer getting worse and for 11 months overall
- Overall, these results provide more support for using avelumab maintenance as a standard treatment for advanced urothelial cancer

BACKGROUND

- UC, the most common type of bladder cancer, is among the most prevalent and deadly cancers worldwide, with poor survival rates in patients diagnosed with la/mUC[1,2]
- In platinum-eligible patients, a current standard of care is 1L PBC followed by avelumab 1LM, based on the results of the JAVELIN Bladder 100 trial (NCT02603432)[3-5]
- The therapeutic landscape for la/mUC is rapidly evolving following the approval of various immune checkpoint inhibitors (ICI), fibroblast growth factor receptor inhibitors, antibody-drug conjugates (ADC),[6] and drug combinations including pembrolizumab plus EV,[7] which has led to updates in clinical guidelines[8]
- This study describes the treatment landscape after US Food and Drug Administration (FDA) approval of avelumab for 1LM treatment in June 2020 by examining patient characteristics, treatment patterns and sequencing, and rw outcomes in patients treated with avelumab 1LM

METHODS

Data sources

- This noninterventional, retrospective cohort study of patients with la/mUC in the US used the nationwide Flatiron Health electronic health record–derived database, which is comprised of deidentified patient-level structured and unstructured data, curated via technology-enabled abstraction (**Figure 1**)[9,10]

Patient population

- During the study period (January 1, 2011-December 31, 2022), the deidentified data originated from approximately 280 cancer clinics (≈800 sites of care)
- The study included patients aged ≥18 years with ≥2 visits on/after January 1, 2011, diagnosed with la/mUC from January 1, 2019, to December 31, 2022
- Patients were required to have been treated with 1L PBC followed by initiation of avelumab 1LM on/after July 1, 2020, and within 90 days after 1L PBC discontinuation and to have had no disease progression for 8-14 weeks after the last administration of 1L PBC, based on oncologist-defined, rule-based lines of therapy
- The index date was defined as the date of avelumab 1LM initiation in the identification period

Statistical analysis

- Treatment patterns were assessed descriptively
- Clinical outcomes, including rwPFS, rwOS, time to treatment discontinuation (TTD), and time to next treatment (TTNT) from avelumab 1LM or 2L EV initiation, were assessed using Kaplan-Meier methods

Figure 1. Study population

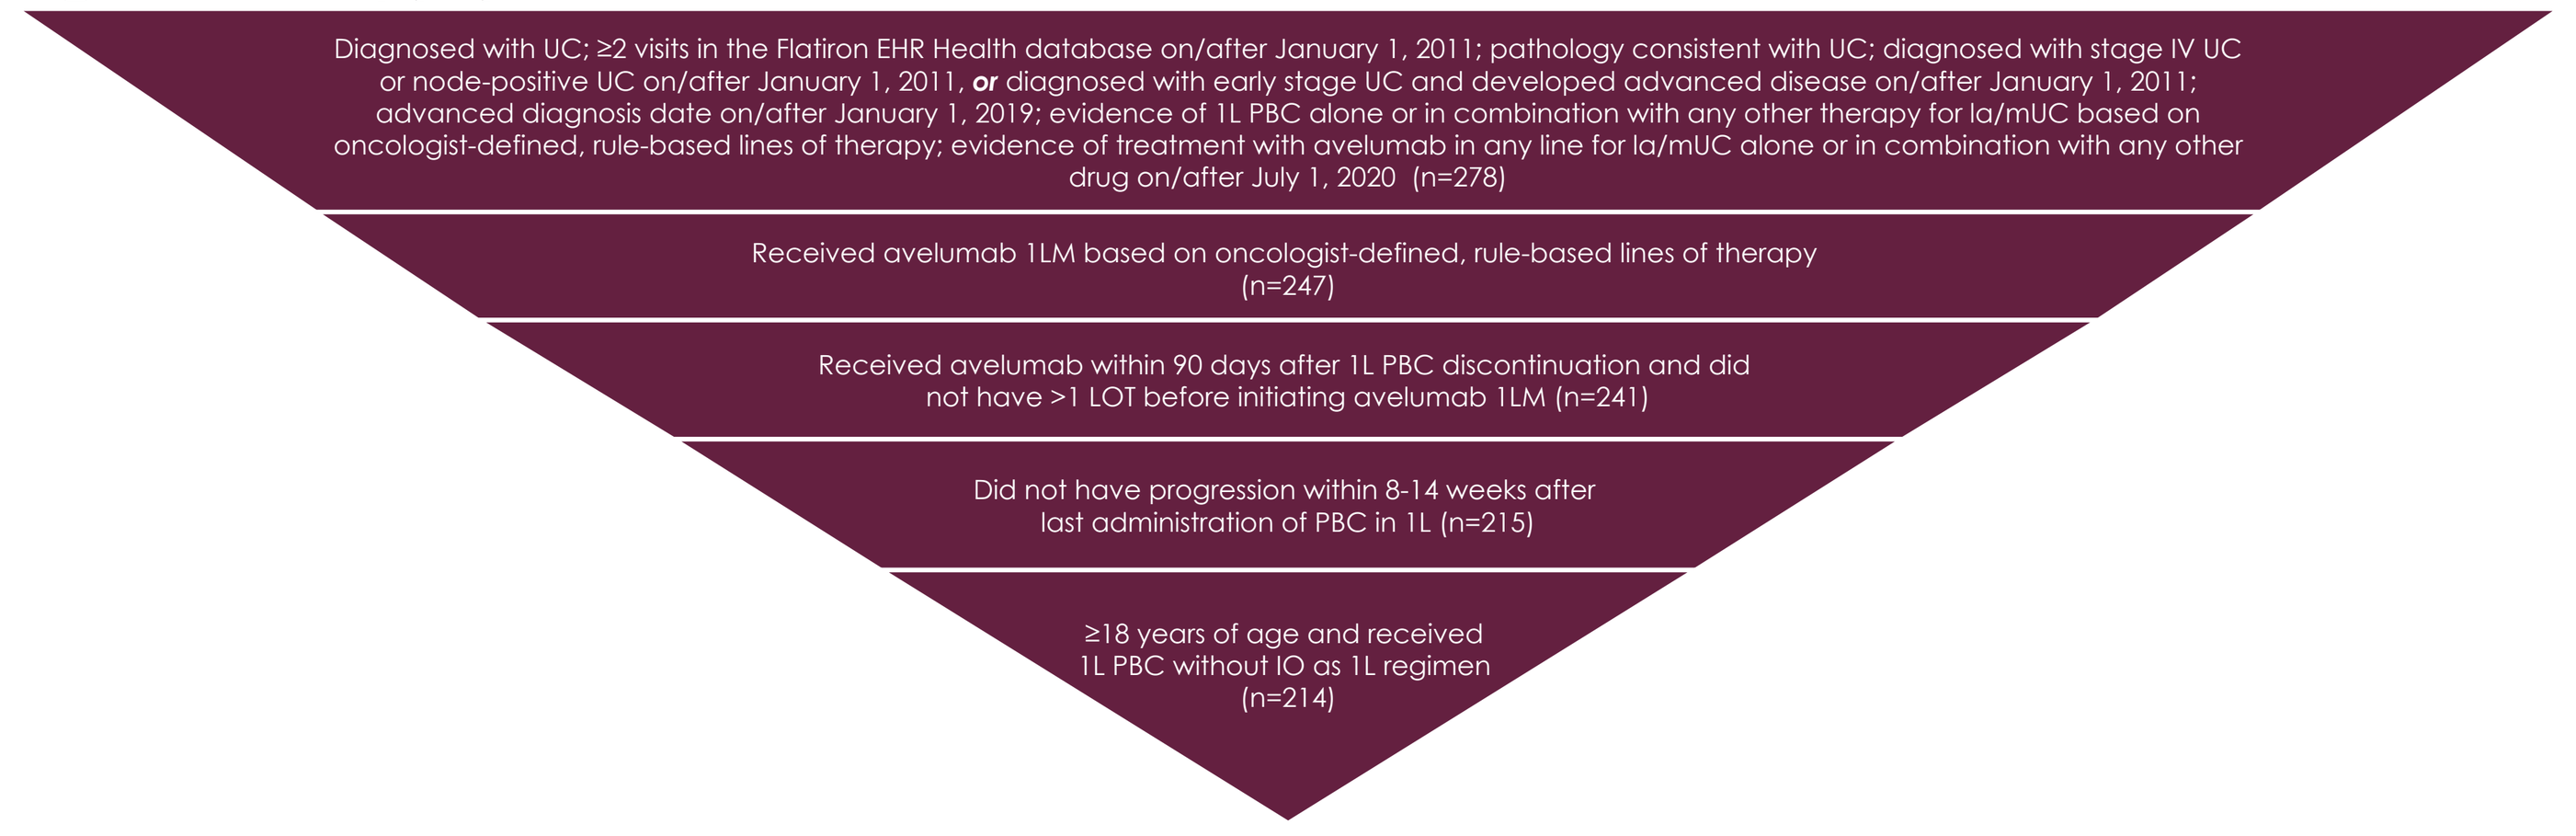

Diagnosed with UC; ≥2 visits in the Flatiron EHR Health database on/after January 1, 2011; pathology consistent with UC; diagnosed with stage IV UC or node-positive UC on/after January 1, 2011, or diagnosed with early stage UC and developed advanced disease on/after January 1, 2011; advanced diagnosis date on/after January 1, 2019; evidence of 1L PBC alone or in combination with any other therapy for la/mUC based on oncologist-defined, rule-based lines of therapy; evidence of treatment with avelumab in any line for la/mUC alone or in combination with any other drug on/after July 1, 2020 (n=278)
Received avelumab 1LM based on oncologist-defined, rule-based lines of therapy (n=247)
Received avelumab within 90 days after 1L PBC discontinuation and did not have >1 LOT before initiating avelumab 1LM (n=241)
Did not have progression within 8-14 weeks after last administration of PBC in 1L (n=215)
≥18 years of age and received 1L PBC without IO as 1L regimen (n=214)

1L, first line; **1LM**, first-line maintenance; **EHR**, electronic health record; **IO**, immuno-oncology; **la/mUC**, locally advanced/metastatic urothelial carcinoma; **LOT**, line of therapy; **PBC**, platinum-based chemotherapy; **UC**, urothelial carcinoma.

RESULTS

Patient population

- A total of 214 patients treated with avelumab 1LM were included
- Mean age was 69 years (SD, 9.2 years); most patients were male (77%) and White (66%) (**Table 1**)
- The majority of primary tumors occurred in the bladder (74%). The most common sites of metastases were distant lymph nodes (57%), bone (29%), and lungs (27%). Similar proportions were seen for the body mass index categories of normal (32%), overweight (28%), and obese (28%) (**Table 1**)
- Cisplatin-based treatment was the most common (54%) 1L treatment followed by carboplatin-based treatment (46%) (**Table 1**)

Table 1. Baseline demographics and clinical characteristics of patients who received avelumab 1LM treatment

Characteristic	Avelumab 1LM (N=214)
Age at diagnosis, years	
Mean (SD)	69.0 (9.2)
Year of index date, n (%)	
2020	43 (20.1)
2021	75 (35.0)
2022	96 (44.9)
Sex, n (%)	
Female	50 (23.4)
Male	164 (76.6)
Race, n (%)	
White	142 (66.4)
Unknown	33 (15.4)
Other	31 (14.5)
Black or African American	6 (2.8)
Asian	2 (0.9)
Region of residence, n (%)	
South	99 (46.3)
Northeast	37 (17.3)
Midwest	30 (14.0)
Unknown	24 (11.2)
West	23 (10.7)
Other	1 (0.5)
Setting, n (%)	
Community	191 (89.3)
Academic	18 (8.4)
Both	5 (2.3)
Follow-up, median (IQR), months	8.7 (4.5-15.7)
Site of disease, n (%)	
Bladder	158 (73.8)
Renal pelvis	30 (14.0)
Ureter	24 (11.2)
Urethra	2 (0.9)
Disease grade, n (%)	
High (G2/G3/G4)	174 (81.3)
Low (G1)	12 (5.6)
Unknown/not documented	28 (13.1)

Characteristic	Avelumab 1LM (N=214)
Stage at initial diagnosis, n (%)	
0	2 (0.9)
I	3 (1.4)
II	9 (4.2)
III	7 (3.3)
IV	99 (46.3)
Unknown/not documented	94 (43.9)
ECOG performance status at diagnosis, n (%)	
0	69 (32.2)
1	96 (44.9)
2+	21 (9.8)
Unknown/not documented	28 (13.1)
Treatment group in 1L, n (%)	
Cisplatin based	115 (53.7)
Carboplatin based	99 (46.3)
Body mass index category, n (%)	
Underweight (<18.5 kg/m²)	9 (4.2)
Normal (18.5-24.9 kg/m²)	69 (32.2)
Overweight (25-29.9 kg/m²)	59 (27.6)
Obese (≥30 kg/m²)	60 (28.0)
Unknown	17 (7.9)
Sites of metastases, n (%)	
Distant lymph node	122 (57.0)
Bone	63 (29.4)
Lung	58 (27.1)
Liver	37 (17.3)
Soft tissue	21 (9.8)
Peritoneum	8 (3.7)
Other	6 (2.8)
Pleura	5 (2.3)
Adrenal	5 (2.3)
Brain	2 (0.9)
Skin	1 (0.5)
Kidney	1 (0.5)

1L, first line; **1LM**, first-line maintenance; **G**, grade; **la/mUC**, locally advanced or metastatic urothelial carcinoma.

Treatment patterns

- Median follow-up from start of avelumab 1LM was 8.7 months (**Table 1**)
- At time of analysis, 83 patients (39%) were still receiving avelumab 1LM and 32 (33%) were receiving ongoing 2L treatment (**Figure 2**)
 - 96 patients (45%) received 2L treatment post avelumab 1LM, most commonly EV monotherapy (55%) followed by carboplatin-based therapy (11%)
- 40 patients (19%) received 3L treatment, most commonly EV monotherapy (25%) followed by sacituzumab govitecan (20%) (**Figure 2**)

Figure 2. Treatment sequencing from avelumab 1LM to 2L and 3L treatment regimens during the observation period

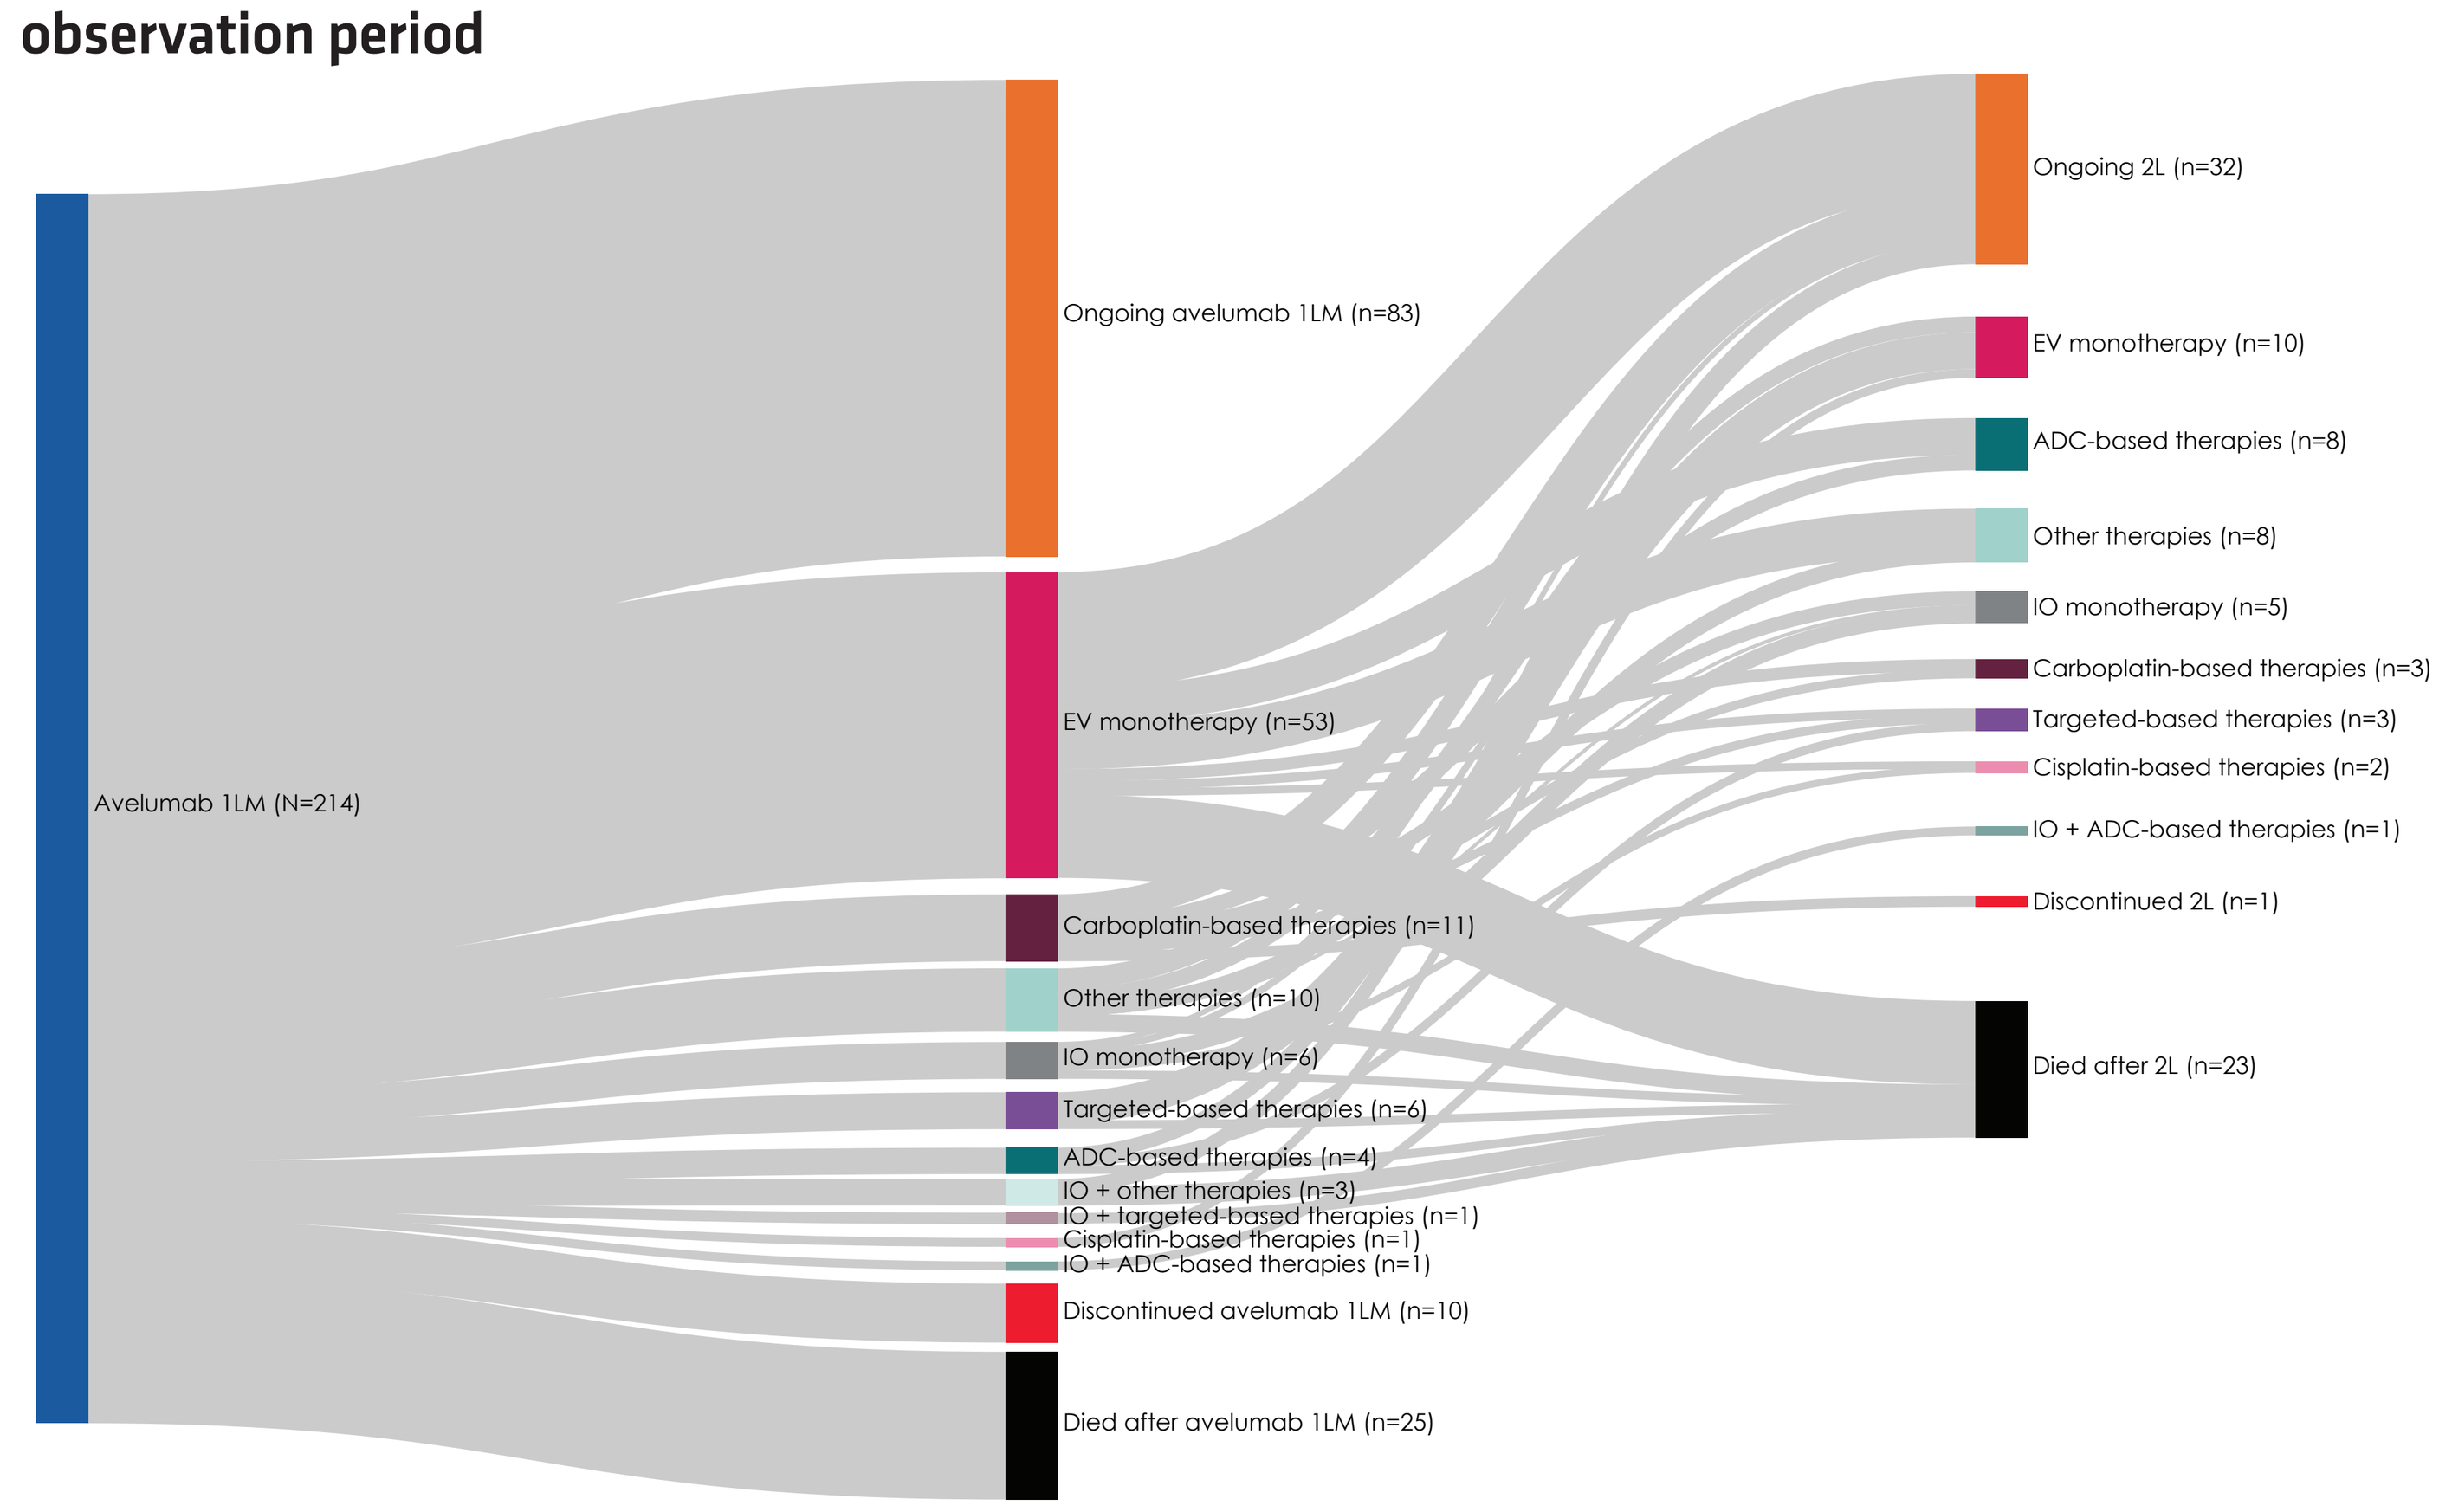

Avelumab 1LM (N=214)
Ongoing avelumab 1LM (n=83)
EV monotherapy (n=53)
Carboplatin-based therapies (n=11)
Other therapies (n=10)
IO monotherapy (n=6)
Targeted-based therapies (n=6)
ADC-based therapies (n=4)
IO + other therapies (n=3)
IO + targeted-based therapies (n=1)
Cisplatin-based therapies (n=1)
IO + ADC-based therapies (n=1)
Discontinued avelumab 1LM (n=10)
Died after avelumab 1LM (n=25)
Ongoing 2L (n=32)
EV monotherapy (n=10)
ADC-based therapies (n=8)
Other therapies (n=8)
IO monotherapy (n=5)
Carboplatin-based therapies (n=3)
Targeted-based therapies (n=3)
Cisplatin-based therapies (n=2)
IO + ADC-based therapies (n=1)
Discontinued 2L (n=1)
Died after 2L (n=23)

1L, first line; **1LM**, first-line maintenance; **2L**, second line; **3L**, third line; **ADC**, antibody-drug conjugate; **EV**, enfortumab vedotin; **IO**, immuno-oncology.

Clinical outcomes

- Median duration of avelumab 1LM was 3.9 months (IQR, 1.9-7.2 months)
- Median rwPFS and rwOS from start of avelumab 1LM were 5.1 months (95% CI, 4.1-7.0 months) and 23.8 months (95% CI, 18.2 months-NE), respectively (**Table 2**, **Figure 3**)
- Median TTNT from avelumab 1LM initiation was 7.0 months (95% CI, 5.6-8.6 months) (**Table 2**)
- In patients who received 2L EV after avelumab 1LM, median rwPFS and rwOS from 2L initiation were 4.9 months (95% CI, 3.9-8.8 months) and 11.2 months (95% CI, 6.8 months-NE), respectively (**Table 2**, **Figure 4**)

Table 2. Clinical outcomes associated with patients treated with avelumab 1LM (N=214)

Patients treated with avelumab 1LM (N=214)	Median (95% CI), months
From 1LM (N=214)	
rwPFS	5.1 (4.1-7.0)
rwOS	23.8 (18.2-NE)
TTNT	7.0 (5.6-8.6)
TTD	4.9 (4.2-6.5)
From 2L EV (n=53)	
rwPFS	4.9 (3.9-8.8)
rwOS	11.2 (6.8-NE)
TTNT	5.8 (5.2-9.5)
TTD	4.7 (4.2-8.5)

1LM, first-line maintenance; **2L**, second line; **EV**, enfortumab vedotin; **NE**, not estimable; **rwOS**, real-world overall survival; **rwPFS**, real-world progression-free survival; **TTD**, time to treatment discontinuation; **TTNT**, time to next treatment.

Figure 3. rwPFS and rwOS from avelumab 1LM initiation

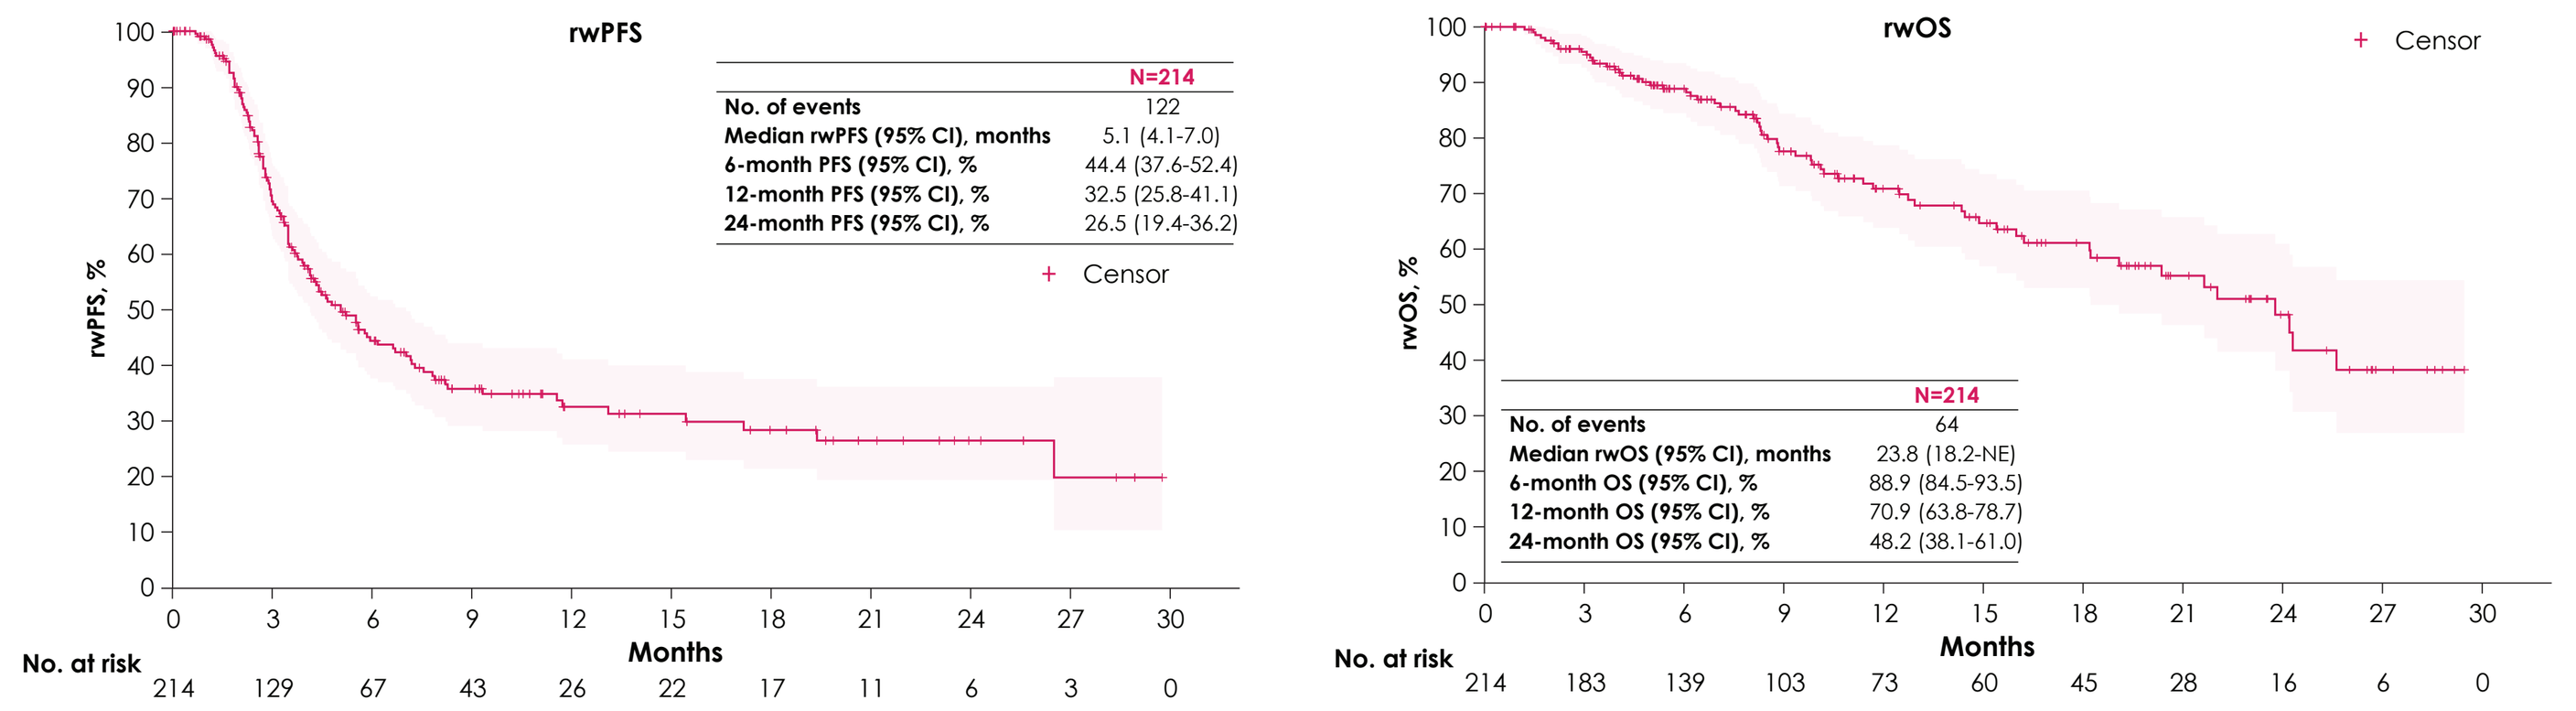

rwPFS
N=214
No. of events 122
Median rwPFS (95% CI), months 5.1 (4.1-7.0)
6-month PFS (95% CI), % 44.4 (37.6-52.4)
12-month PFS (95% CI), % 32.5 (25.8-41.1)
24-month PFS (95% CI), % 26.5 (19.4-36.2)
Censor
rwPFS, %
Months
No. at risk 214 129 67 43 26 22 17 11 6 3 0
rwOS
Censor
N=214
No. of events 64
Median rwOS (95% CI), months 23.8 (18.2-NE)
6-month OS (95% CI), % 88.9 (84.5-93.5)
12-month OS (95% CI), % 70.9 (63.8-78.7)
24-month OS (95% CI), % 48.2 (38.1-61.0)
rwOS, %
Months
No. at risk 214 183 139 103 73 60 45 28 16 6 0

1LM, first-line maintenance; **NE**, not estimable; **rwOS**, real-world overall survival; **rwPFS**, real-world progression-free survival.

Figure 4. rwPFS and rwOS from 2L EV monotherapy initiation

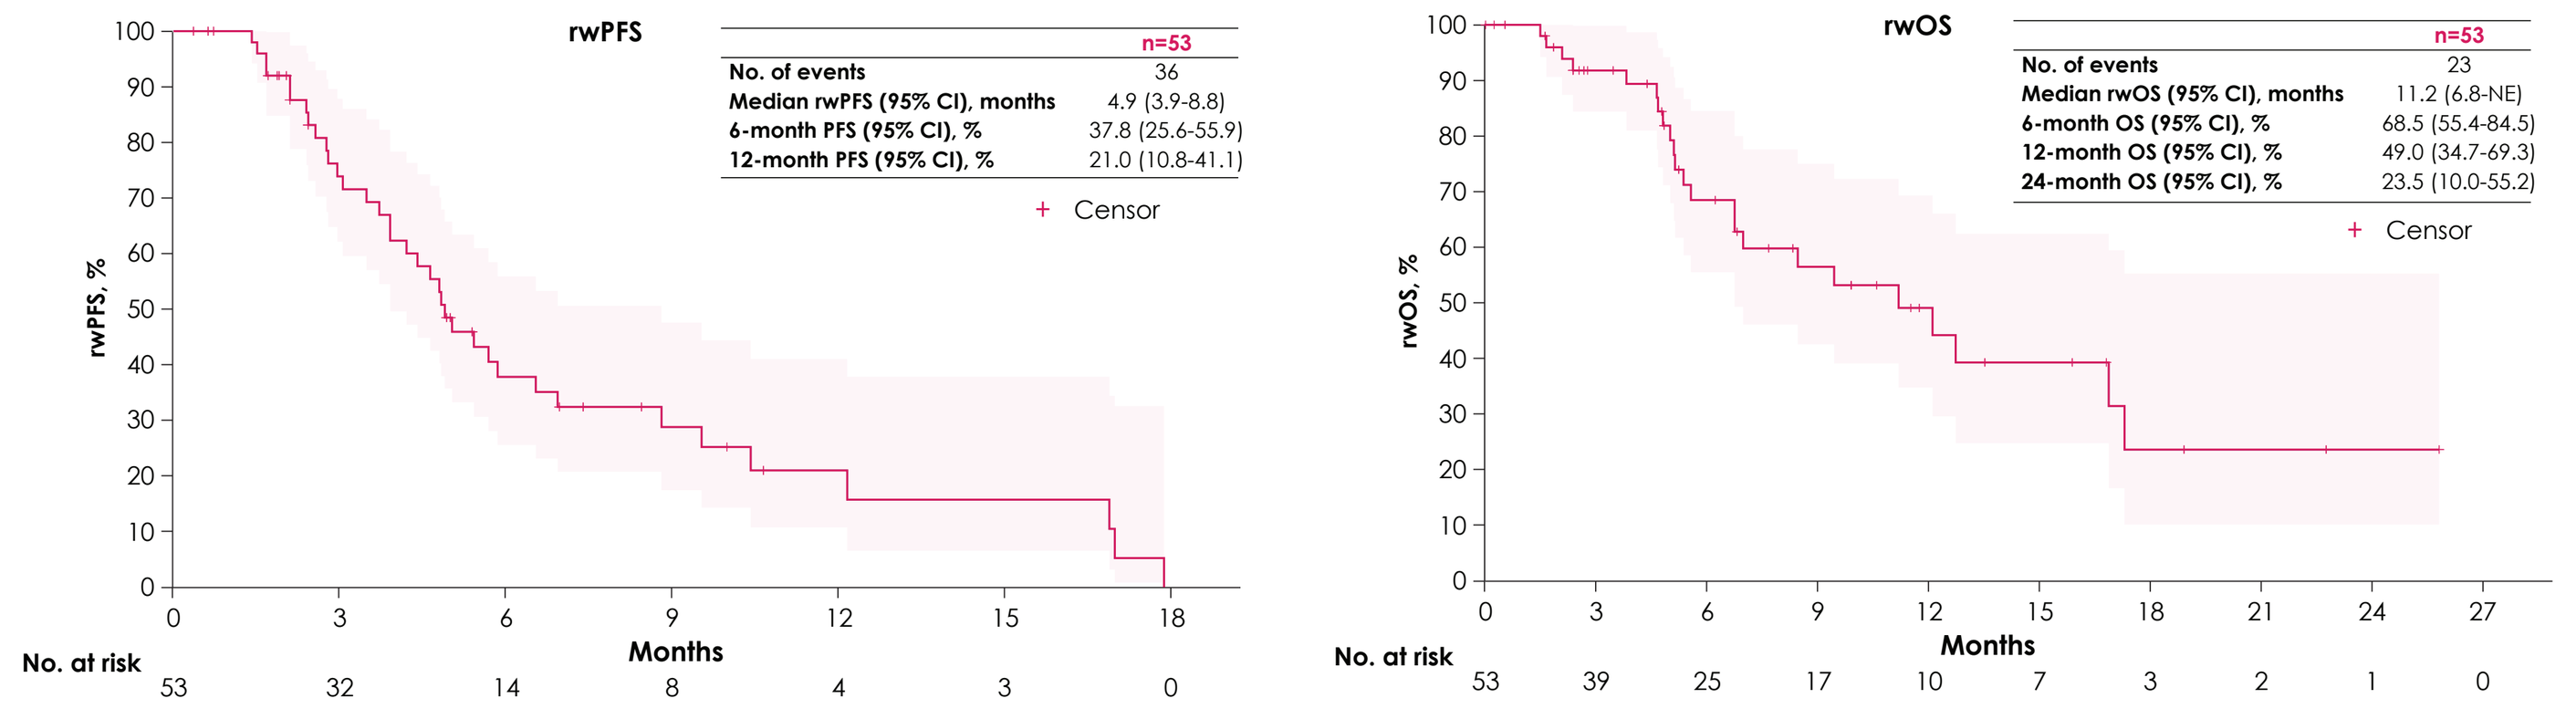

rwPFS
n=53
No. of events 36
Median rwPFS (95% CI), months 4.9 (3.9-8.8)
6-month PFS (95% CI), % 37.8 (25.6-55.9)
12-month PFS (95% CI), % 21.0 (10.8-41.1)
Censor
rwPFS, %
Months
No. at risk 53 32 14 8 4 3 0
rwOS
n=53
No. of events 23
Median rwOS (95% CI), months 11.2 (6.8-NE)
6-month OS (95% CI), % 68.5 (55.4-84.5)
12-month OS (95% CI), % 49.0 (34.7-69.3)
24-month OS (95% CI), % 23.5 (10.0-55.2)
Censor
rwOS, %
Months
No. at risk 53 39 25 17 10 7 3 2 1 0

2L, second line; **EV**, enfortumab vedotin; **NE**, not estimable; **rwOS**, real-world overall survival; **rwPFS**, real-world progression-free survival.

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Helen H. Moon**, Helen.h.moon@kp.org

REFERENCES **1.** Antoni S, et al. Eur Urol. 2017;71(1):96-108. **2.** Sung H, et al. CA Cancer J Clin. 2021;71(3):209-49. **3.** Powles T, et al. N Engl J Med. 2020;383(13):1218-30. **4.** Bavencio (avelumab). Prescribing information. EMD Serono, Rockland, MA, USA; 2020. **5.** Grivas P, et al. Cancer Treat Rev. 2021;97:102187. **6.** Bianchi A, et al. Expert Opin Pharmacother. 2023;24(2):177-95. **7.** US Food and Drug Administration. FDA grants accelerated approval to enfortumab vedotin-ejfv with pembrolizumab for locally advanced or metastatic urothelial carcinoma. Accessed January 9, 2024. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-grants-accelerated-approval-enfortumab-vedotin-ejfv-pembrolizumab-locally-advanced-or-metastatic. **8.** Powles T, et al. Ann Oncol. 2022;33(3):244-58. **9.** Ma X, et al. medRxiv. Preprint posted online June 7, 2023. **10.** Birnbaum B, et al. arXiv. Preprint posted online January 13, 2020.

DISCLOSURES H. H. Moon has participated in consulting or advisory roles for the healthcare business of Merck KGaA, Darmstadt, Germany, and Pfizer; has provided speaker services for Bristol Myers Squibb, Janssen, Seagen, CITN, Nektar, Apollomics, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel, accommodation and expenses from Bayer. **M. Kearney** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany, and has stock and other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, and UCB. **S. H. Mahmoudpour** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **C. Ike** is an employee of EMD Serono and has stock and other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany. **V. Morris** is an employee of EMD Serono and has stock and other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany. **A. Rava, S. Kim, H. Sun, M. Boyd,** and **G. Gomez Rey** are employees of Genesis Research Group. **ACKNOWLEDGMENTS** This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Editorial support was provided by Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany. Medical writing support was provided by Henry Blanton, an employee of Genesis Research Group.

Abstract No. 605. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.