Platinum eligibility criteria for patients with metastatic urothelial carcinoma: results of a physician survey in 5 European countries

S. Gupta,[1] **T. Powles**,[2] **M. Kearney**,[3] **L. Panattoni**,[4] **N. Land**,[4] **T. Flottemesch**,[4] **P. Sullivan**,[4*] **M. Kirker**,[5] **M. Bharmal**,[6*] **S. Guenther**,[3*] **N. Costa**,[7] **E. Grande**[8]

[1]Taussig Cancer Institute, Cleveland Clinic Foundation, Cleveland, OH, USA; [2]Barts Cancer Institute, Experimental Cancer Medicine Centre, Queen Mary University of London, St Bartholomew's Hospital, London, UK; [3]the healthcare business of Merck KGaA, Darmstadt, Germany; [4]Precision Value and Health, New York, NY, USA; [5]Pfizer, New York, NY, USA; [6]EMD Serono, Billerica, MA, USA; [7]Pfizer, Porto Salvo, Portugal; [8]MD Anderson Cancer Center Madrid, Madrid, Spain

*Affiliation at the time the study was conducted.

CONCLUSIONS

- **This study assessed the clinical thresholds used by European physicians to determine platinum eligibility (PE) in patients with metastatic urothelial carcinoma (mUC) and their alignment with published US physician thresholds**
- **Overall, physicians in Europe reported that 76.8% of patients were platinum eligible, with some differences across countries (range, 74.6% [Italy] to 79.5% [Spain])**
- **Criteria to determine PE were broadly consistent with those of previous US-based studies, except for age**
 - **The low age threshold observed is at odds with the older age of the mUC patient population and available evidence that older patients benefit from and can tolerate platinum-based chemotherapy (PBC)[1,2]**
 - **There were systematic differences in the type of physicians who reported using an age threshold compared with those who did not**
- **Standardization and wider use of these criteria could support clinical decision-making, reduce variations in care, and improve patient outcomes**

PLAIN LANGUAGE SUMMARY

- **The standard first main treatment chosen for people with metastatic urothelial cancer depends on whether they can receive platinum-based chemotherapy**
 - **Platinum-based chemotherapy includes a drug that contains platinum (cisplatin or carboplatin)**
- **In this study, we asked doctors from 5 European countries (France, Germany, Italy, Spain, and the UK) about the characteristics they use to decide if someone can receive platinum-based chemotherapy, and compared them with the characteristics used by US doctors that were reported in a previous study**
- **Most European doctors (69%) in the study had at least 10 years of experience treating people with metastatic urothelial cancer**
- **This study found that across the 5 European countries, most people with metastatic urothelial cancer (77%) should be able to receive platinum-based chemotherapy**
- **Characteristics considered by European doctors were similar to those reported by US doctors, except for age**
 - **Most European doctors (67%) would not treat someone aged >80 years with platinum-based chemotherapy, whereas US doctors did not use age to decide whether or not to use this treatment**
- **To ensure people receive optimal treatment, doctors need a standard set of characteristics to consider when deciding which people with metastatic urothelial cancer can receive platinum-based chemotherapy**

BACKGROUND

- Bladder cancer is the tenth most common cancer globally, with UC accounting for ≈90% of cases[3,4]
- Current US and European guidelines for mUC recommend first-line (1L) treatment based on eligibility for PBC (cisplatin or carboplatin)[5-7]
- Among patients eligible for PBC, the standard of care for mUC is PBC followed by avelumab 1L maintenance in patients without disease progression[5-8]
- A systematic review concluded that many patients with mUC may not be receiving guideline-concordant 1L care,[9] negatively impacting clinical outcomes[10]
- Underuse of guideline-recommended 1L therapy may be partly driven by physicians' uncertainty about the definition of PE
- Little is known about the criteria European physicians use in real-world practice to define PE or how they align with US criteria[11,12]

METHODS

- Physicians from France, Germany, Italy, Spain, and the UK completed a cross-sectional, quantitative online survey in August/September 2022
- Physicians eligible for inclusion:
 - Specialized in oncology or urology
 - Practiced medicine for ≥3 years
 - Spent ≥50% of time providing direct patient care
 - Treated ≥2 patients with mUC per month
- Physicians reported the percentage of patients they perceived as cisplatin eligible, carboplatin eligible but cisplatin ineligible, and platinum ineligible
 - Differences in the distribution of points for each category across countries were assessed using a Kruskal-Wallis test
- Clinical criteria to define PE were adapted from Gupta et al's US-based surveys presented at ASCO-GU 2019 and ASCO 2022[11,12]
 - The criteria included age, creatinine clearance (CrCl), ECOG performance status (PS), New York Heart Association (NYHA) heart failure class, and peripheral neuropathy grade
- The cumulative percentage of physicians who would not treat a patient with PBC at each threshold were compared with the consensus thresholds (cumulative percentage ≥50%) reported in the 2022 US survey[12]
- Logistic regression was used to analyze the physician characteristics associated with reporting no age threshold for PE vs any age threshold

RESULTS

- 503 physicians (69% oncologists, 31% urologists) completed the quota-based survey
- Most had been in practice for >10 years (69%), treated 5-19 patients with mUC per month (58%), and practiced in a public teaching hospital (40%) (**Table 1**)
- Physicians reported that approximately one-quarter (23.2%) of patients with mUC are platinum ineligible, ranging from 20.5% (Spain) to 25.4% (Italy) (**Figure 1**)
- Thresholds at which the full sample reached consensus were broadly consistent with thresholds from the 2022 US survey, except for age (**Figure 2**)
- European physicians reached consensus of platinum ineligibility at the US thresholds for ECOG PS (≥3) and NYHA heart failure (class III)
- European physicians reached consensus at a CrCl level of <25 mL/min and peripheral neuropathy grade ≥3, while US physicians recommended thresholds of <30 mL/min and grade ≥2
- The majority of European physicians (n=336 [66.8%]) would not treat patients aged >80 years with PBC, whereas US physicians did not recommend using an age threshold to define PE
- Physicians were more likely to use an age threshold if they were from Italy, if they practiced in a public nonteaching hospital or public or private office setting, and if they treated <20 patients with mUC per month (**Table 2**)

Table 1. Physician practice characteristics

	All countries (N=503)	France (n=100)	Germany (n=101)	Italy (n=100)	Spain (n=102)	UK (n=100)
Years in practice, %						
3-6 years	4.6***	9.0	3.0	3.0	2.9	5.0
7-10 years	26.6	31.0	21.8	39.0	23.5	18.0
11-14 years	33.2	27.0	34.7	28.0	38.2	38.0
15-18 years	18.7	16.0	19.8	14.0	18.6	25.0
>18 years	16.9	17.0	20.8	16.0	16.7	14.0
Primary specialty, %						
Oncology	69.0**	74.0	54.5	77.0	70.6	69.0
Urology	31.0	26.0	45.5	23.0	29.4	31.0
Practice type, %						
Public teaching hospital	40.4***	32.0	32.7	24.0	57.8	55.0
Public nonteaching hospital	24.1	19.0	24.8	32.0	20.6	24.0
Private hospital	20.3	37.0	6.9	24.0	19.6	14.0
Public office	7.2	3.0	23.8	5.0	0	4.0
Private office	3.6	4.0	5.0	9.0	0	0
Specialist cancer center	4.6	5.0	6.9	6.0	2.0	3.0
Average number of patients with mUC treated per month, %						
2-4 patients	15.1***	11.0	11.9	12.0	18.6	22.0
5-10 patients	30.4	20.0	43.6	29.0	29.4	30.0
11-19 patients	28.0	30.0	24.8	49.0	16.7	20.0
20-50 patients	19.7	30.0	18.8	8.0	26.5	15.0
>50 patients	6.8	9.0	1.0	2.0	8.8	13.0

mUC, metastatic urothelial carcinoma.
*p<0.05, **p<0.01, ***p<0.001; differences across countries.

Figure 1. Percentage of patients with mUC reported as platinum eligible or ineligible

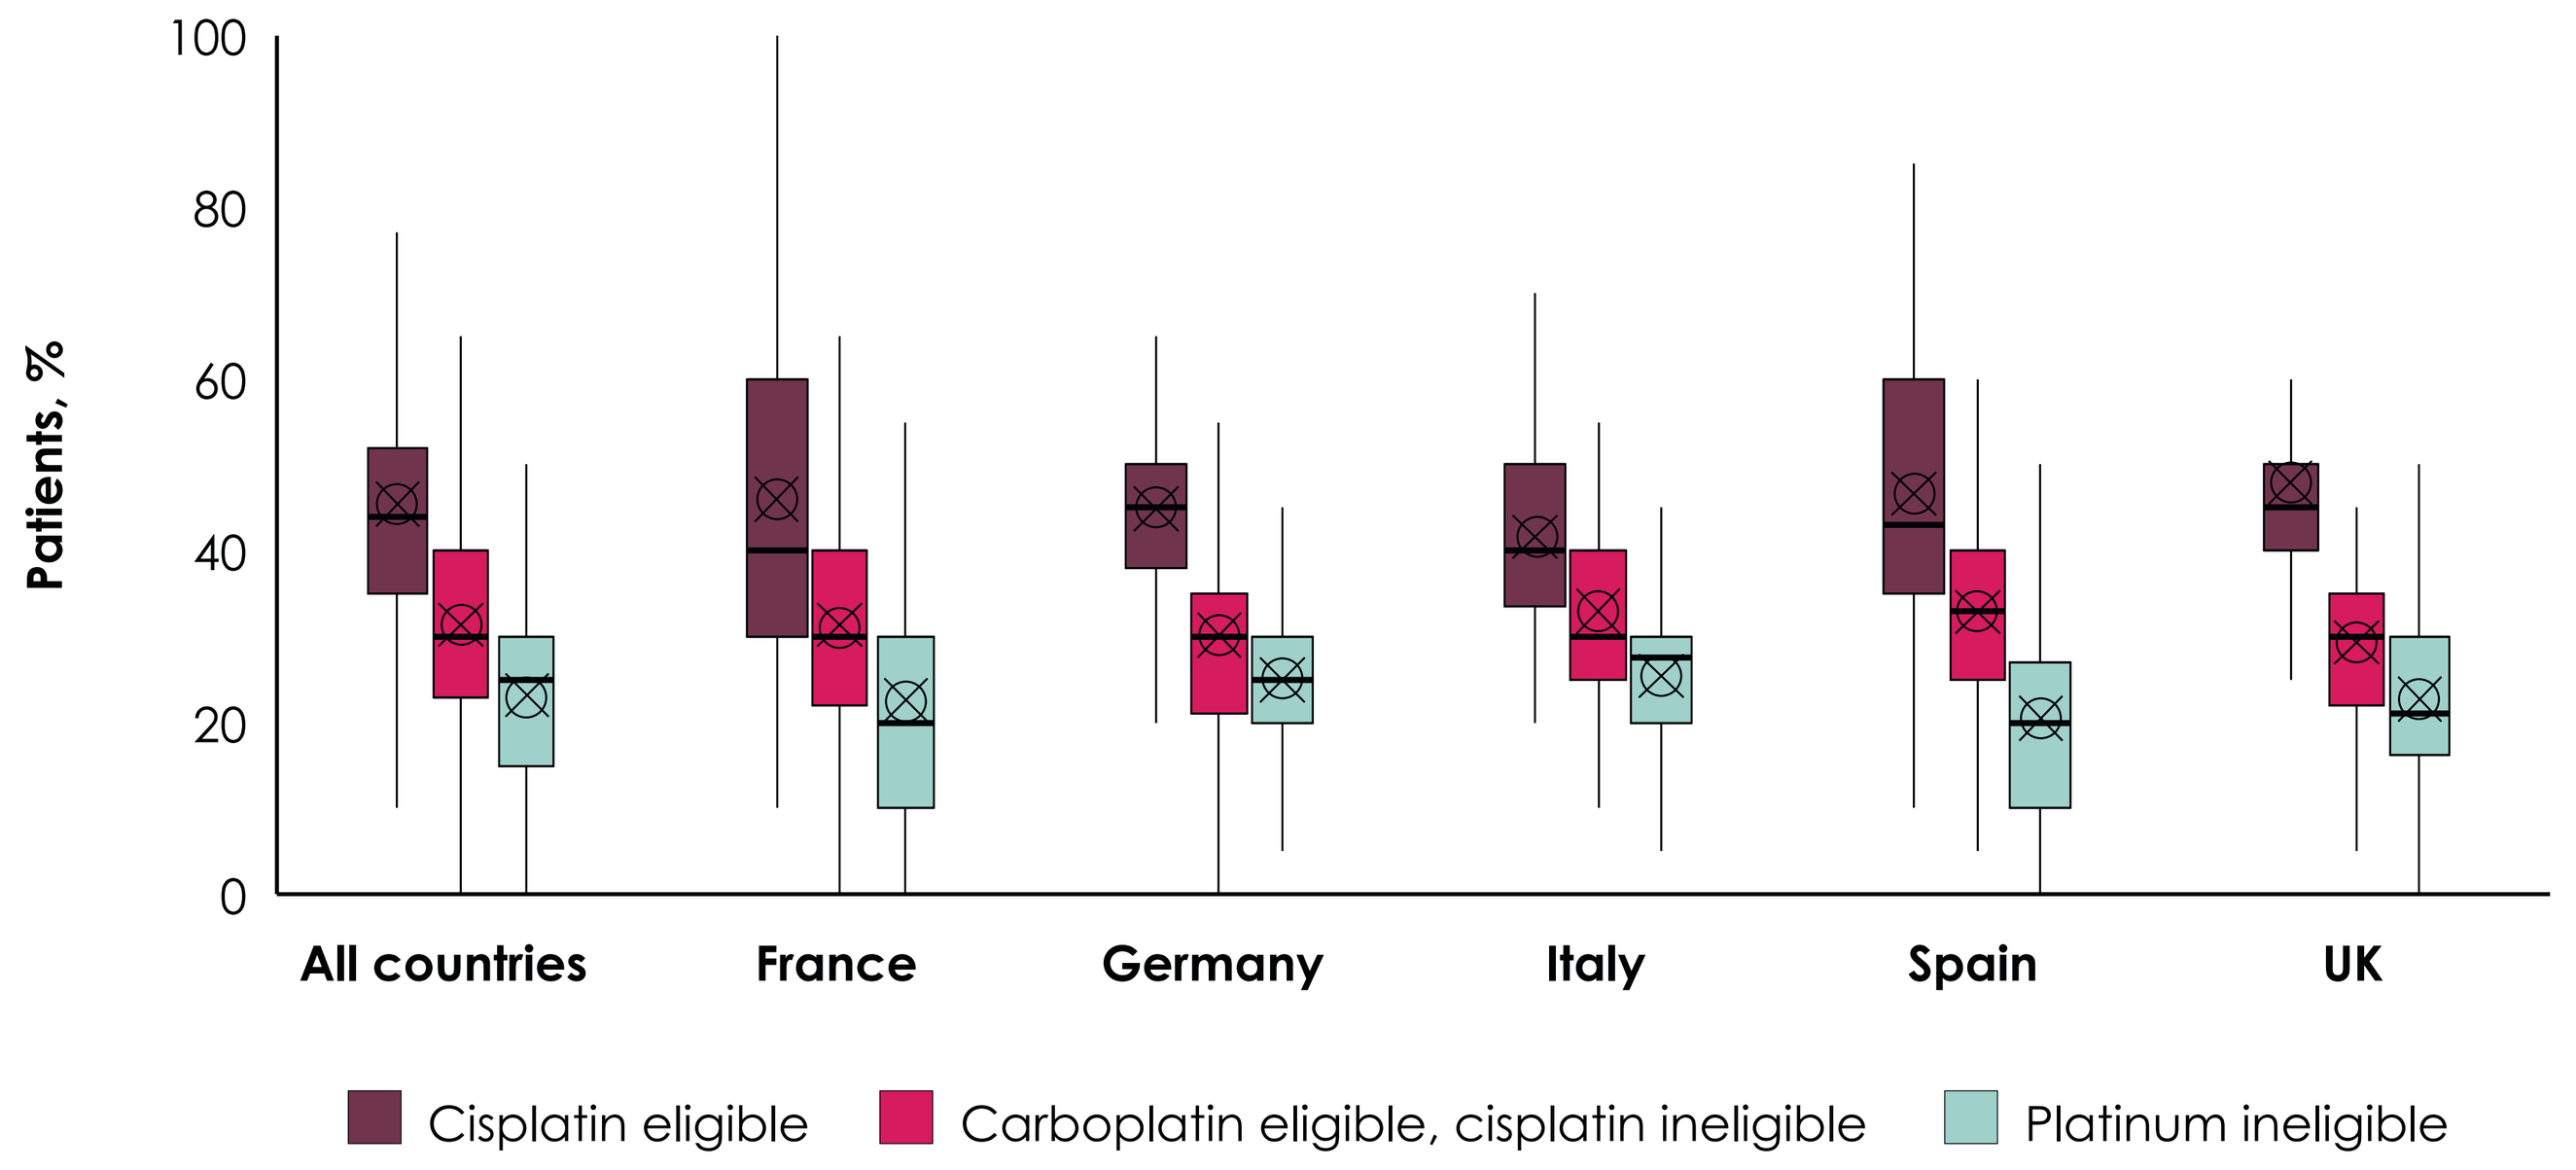
Patients, %
100
80
60
40
20
0
All countries
France
Germany
Italy
Spain
UK
Cisplatin eligible
Carboplatin eligible, cisplatin ineligible
Platinum ineligible

Cisplatin eligible: p=0.054. Carboplatin eligible, cisplatin ineligible: p=0.088. Platinum ineligible: p=0.004. Box and whisker plots show the median (horizontal black line), mean (circle with X), interquartile range (top to bottom of box), and 2.5th and 97.5th percentile data range (whiskers).
mUC, metastatic urothelial carcinoma.

Figure 2. Cumulative percentage of physicians who would not treat patients with mUC with 1L platinum-based chemotherapy at each threshold

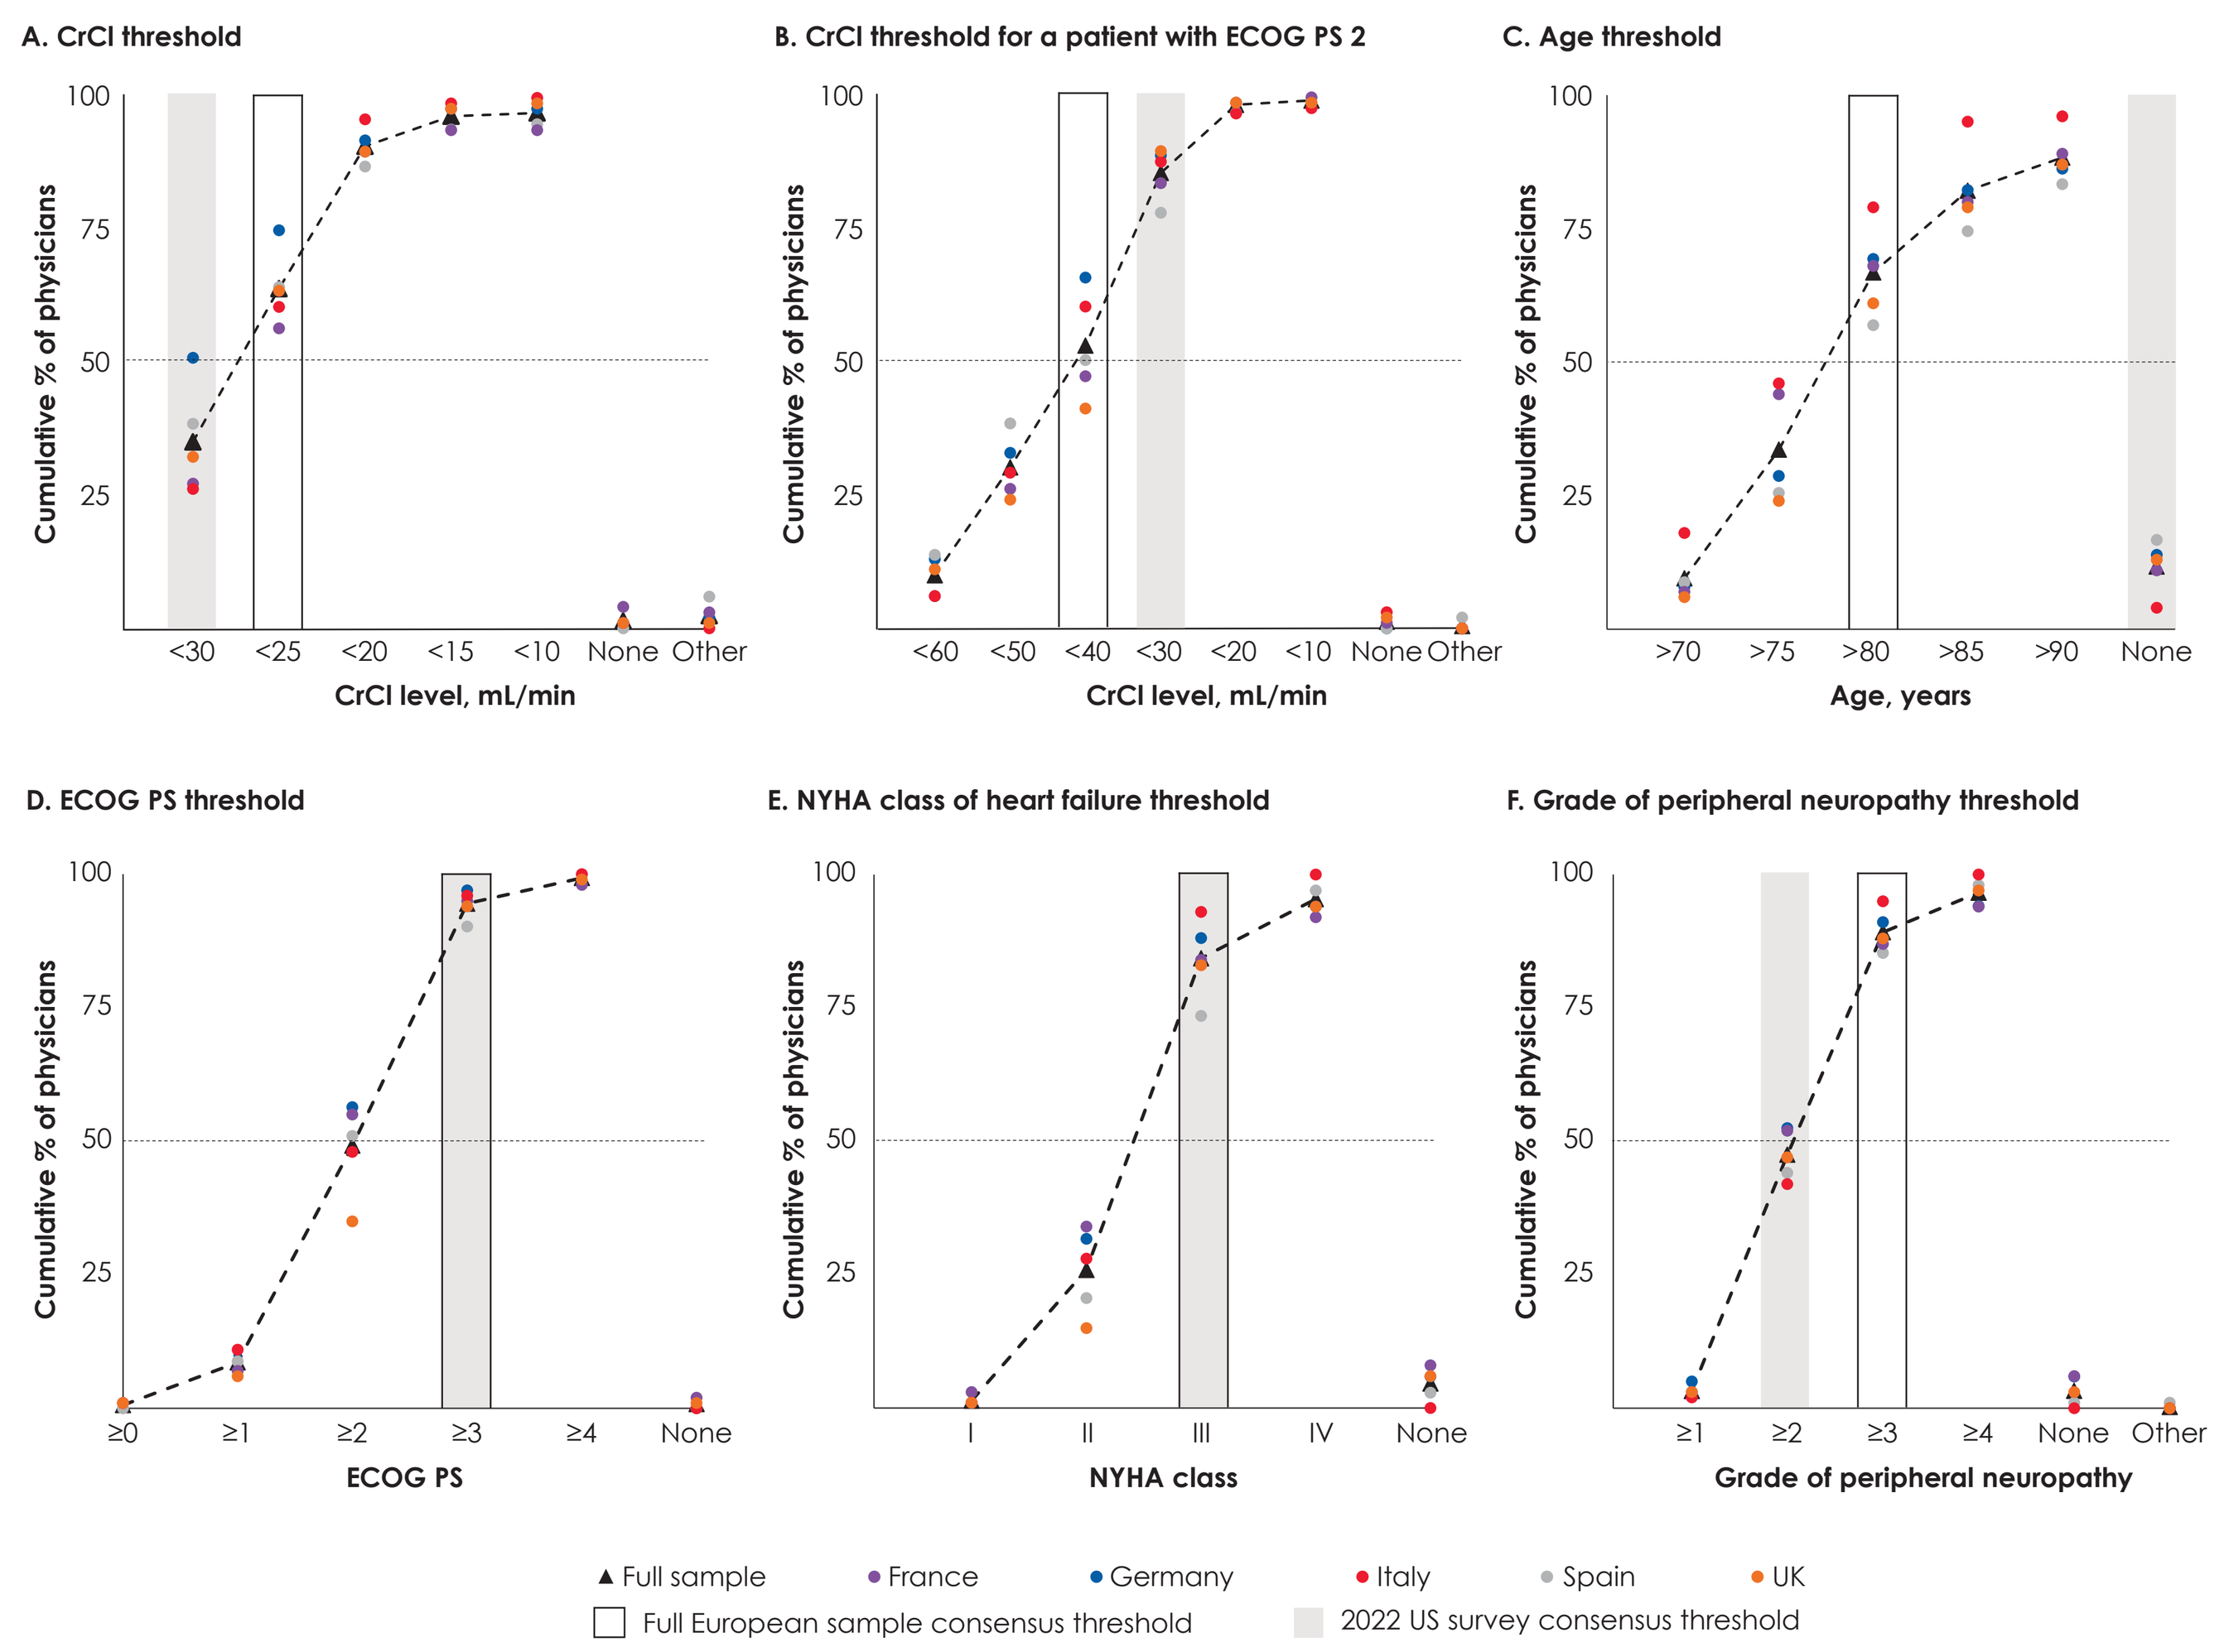
A. CrCl threshold
Cumulative % of physicians
<30 <25 <20 <15 <10 None Other
CrCl level, mL/min
B. CrCl threshold for a patient with ECOG PS 2
<60 <50 <40 <30 <20 <10 None Other
CrCl level, mL/min
C. Age threshold
>70 >75 >80 >85 >90 None
Age, years
D. ECOG PS threshold
≥0 ≥1 ≥2 ≥3 ≥4 None
ECOG PS
E. NYHA class of heart failure threshold
I II III IV None
NYHA class
F. Grade of peripheral neuropathy threshold
≥1 ≥2 ≥3 ≥4 None Other
Grade of peripheral neuropathy
Full sample
France
Germany
Italy
Spain
UK
Full European sample consensus threshold
2022 US survey consensus threshold

The consensus threshold was defined as the threshold at which the cumulative percentage reached ≥50%.
1L, first line; **CrCl**, creatinine clearance; **mUC**, metastatic urothelial carcinoma; **NYHA**, New York Heart Association; **PS**, performance status.

Table 2. Factors associated with using an age threshold to define PE

	Unadjusted		Adjusted	
	No age threshold (n=59)	Any age threshold (n=444)	No vs any threshold: Odds ratio	No vs any threshold: p value
Country, %				
Germany	24	20*	–	
France	19	20	0.44	0.112
Italy	7	22	**0.22**	**0.02**
Spain	29	19	0.63	0.323
UK	22	20	0.57	0.231
Sex, %				
Male	61	73	–	
Female	27	22	1.37	0.389
Prefer not to answer	12	5	4.23	0.01
Primary specialty, %				
Oncology	68	69	–	
Urology	32	31	1.16	0.676
Years in practice, %				
3-6 years	3	5*	–	
7-10 years	14	28	0.46	0.363
11-14 years	44	32	1.22	0.808
15-18 years	12	20	0.47	0.394
>18 years	27	16	1.82	0.471
Time spent providing direct patient care, %				
50%-74%	5	17*	–	
75%-100%	95	83	2.11	0.274
Practice type, %				
Private hospital	24	20	–	
Public nonteaching hospital	15	25	**0.28**	**0.01**
Public teaching hospital	49	39	0.48	0.097
Public/private office	5	11	**0.14**	**0.01**
Specialist cancer center	7	4	0.73	0.654
Average number of patients with mUC treated per month, %				
≥20 patients	49	23***	–	
11-19 patients	12	30	**0.25**	**0.004**
2-10 patients	39	46	**0.36**	**0.006**

mUC, metastatic urothelial carcinoma; **PE**, platinum eligibility.
*p<0.05, **p<0.01, ***p<0.001; bolded values are significant.

GET POSTER PDF AND VIEW VIDEO PRESENTATION

Copies of this poster and any associated video or audio files obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements, you will be redirected to the ASCO meeting site.

Correspondence: **Shilpa Gupta, guptas5@ccf.org**

REFERENCES 1. Bamias A, et al. Ann Oncol. 2005;16(2):307-13. **2.** Galsky MD, et al. Urol Oncol. 2014;32(1):30.e15-21. **3.** Sung H, et al. CA Cancer J Clin. 2021;71(3):209-49. **4.** Cancer.net. Accessed December 12, 2023. https://www.cancer.net/cancer-types/bladder-cancer **5.** NCCN Clinical Practice Guidelines in Oncology: Bladder Cancer. V3.2023. **6.** EAU Guidelines. Edn. presented at the EAU Annual Congress Milan 2023. ISBN 978-94-92671-19-6. **7.** Powles T, et al. Ann Oncol. 2022;33:244-58. **8.** Powles T, et al. N Engl J Med. 2020;383:1218-30. **9.** Kearney M, et al. Future Oncol. Published online August 1, 2023. **10.** Geynisman DM, et al. Urol Oncol. 2022;40(5):195.e1-e11. **11.** Gupta S, et al. J Clin Oncol. 2019;37:7(Suppl):Abstract 451. **12.** Gupta S, et al. J Clin Oncol. 2022;40:16(Suppl):Abstract 4577.

DISCLOSURES S Gupta has served in a consulting or advisory role for AVEO, Bayer, Bristol Myers Squibb, Gilead Sciences, Guardant Health, Lilly, Loxo, Pfizer, and the healthcare business of Merck KGaA, Darmstadt, Germany; has provided speaker services for Bristol Myers Squibb, Gilead Sciences, Janssen Oncology, and Seagen; and has stock and other ownership interests in BioNTech, Moderna, and Nektar. **T Powles** has served in a consulting or advisory role for Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Eisai, Exelixis, Incyte, Ipsen, Johnson & Johnson, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Roche, Seagen, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel and accommodation expenses from Pfizer. **M Kearney** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany and has stock and other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, and UCB. **L Panattoni, N Land,** and **T Flottemesch** are employees of Precision Value and Health. **P Sullivan** was an employee of Precision Value and Health at the time of this study. **M Kirker** is an employee of, owns stock in, and has received honoraria from Pfizer. **M Bharmal** was an employee of EMD Serono, Billerica, MA, USA at the time of the study and has stock or other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany. **S Guenther** was an employee of the healthcare business of Merck KGaA, Darmstadt, Germany at the time of this study and has stock or other ownership interests in the healthcare business of Merck KGaA, Darmstadt, Germany. **N Costa** is an employee of, owns stock in, and has received honoraria from Pfizer. **E Grande** has received honoraria from Advanced Accelerator Applications, Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Dr Reddy's Laboratories, Eisai, EUSA Pharma, GSK, Ipsen, Janssen-Cilag, Lilly, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Roche, and the healthcare business of Merck KGaA, Darmstadt, Germany; has served in a consulting or advisory role for Bristol Myers Squibb, Ipsen, Merck & Co., Kenilworth, NJ, Pfizer, and Roche; reports receiving research funding from Astellas Pharma, AstraZeneca, Ipsen, Lexicon, Molecular Templates, Pfizer, and Roche; and has received travel and accommodation expenses from Bristol Myers Squibb, Ipsen, Janssen-Cilag, Pfizer, and Roche/Genentech. **ACKNOWLEDGMENTS** This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and was previously conducted under an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer. Editorial support was provided by Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 696. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.