Clinical outcomes with split-dose cisplatin-based regimens in patients with locally advanced or metastatic urothelial carcinoma: results of a systematic literature review and network meta-analysis

R. O'Dwyer,[1] M. Kearney,[2] M. Musat,[3] I. Gulas,[3] S. Guenther,[2]* E. Hubscher,[3]* H. Moradian,[3] S. S. Sridhar[1]

[1]Princess Margaret Cancer Centre, Toronto, ON, Canada; [2]the healthcare business of Merck KGaA, Darmstadt, Germany; [3]Cytel, Waltham, MA, USA
*Affiliation at the time the study was conducted.

CONCLUSIONS

- **This systematic literature review (SLR) aimed to characterize the efficacy and safety of split-dose cisplatin regimens and treatment criteria in locally advanced or metastatic urothelial carcinoma (la/mUC)**
- **The comparative effectiveness of split-dose gemcitabine + cisplatin (GC) was examined in a network meta-analysis (NMA) vs other standard chemotherapy regimens in la/mUC**
- **In this NMA, split-dose GC, which is often used in cisplatin-ineligible patients (per Galsky criteria), provided survival and response rates comparable to standard-dose GC and methotrexate + vinblastine + doxorubicin + cisplatin (MVAC), and notably showed a trend toward improved outcomes compared with gemcitabine + carboplatin (GCa)**
- **Split-dose GC may be a viable frontline treatment alternative to standard GC and may allow more patients with la/mUC to receive cisplatin-based chemotherapy**
- **Further investigations and prospective studies are required to determine the optimal use of split-dose GC and the characteristics of patients who may derive the most benefit**

PLAIN LANGUAGE SUMMARY

- **Platinum-based combination chemotherapy that includes cisplatin or carboplatin is a standard treatment for people with advanced urothelial cancer**
- **Cisplatin is more effective than carboplatin but it causes more side effects, so many people cannot receive cisplatin, such as those with poor clinical status, reduced kidney function, or some other health problems**
- **Some studies suggest that instead of giving cisplatin on a single day, dividing the dose over 2 days, which is called split-dose cisplatin, might cause fewer side effects**
- **In this analysis, researchers reviewed all studies reporting on the use of split-dose cisplatin in patients with advanced urothelial cancer**
- **Most people treated with split-dose cisplatin in these studies had reduced kidney function, had poor clinical status, or were older**
- **Chemotherapy including split-dose cisplatin had similar effectiveness to standard cisplatin-based chemotherapy and seemed to be slightly more effective than carboplatin-based chemotherapy**
- **Overall, these findings suggest that chemotherapy including split-dose cisplatin might allow more people with advanced urothelial cancer to benefit from treatment**
- **More studies comparing split-dose cisplatin and standard cisplatin are needed to determine which patients might benefit the most from split-dose cisplatin**

BACKGROUND

- Platinum-based chemotherapy is crucial in the management of la/mUC, and it can be followed by avelumab maintenance in the first-line (1L) setting in patients achieving disease control[1]
- Because of greater efficacy, cisplatin-based regimens such as GC are preferred over GCa, but up to 50% of patients are deemed cisplatin ineligible due to renal impairment, poor performance status, or other comorbidities[2,3]
- Prior studies have shown that a split-dose cisplatin schedule, in which the cisplatin dose of 70 mg/m² is administered over 2 days (ie, 35 mg/m²/day), improved tolerability and had similar efficacy to that of conventional cisplatin regimens in la/mUC[4-7]

METHODS

- An SLR was conducted following the Cochrane guidelines to identify observational studies in patients with la/mUC reporting treatment patterns, effectiveness, and safety of chemotherapies, immunotherapies, and other approved pharmacological therapies. Randomized and nonrandomized clinical and observational studies in patients with mUC receiving split-dose cisplatin regimens were also identified
- Medline, Embase, and Cochrane Library databases were searched on March 8, 2023. No limits were placed on publication year or geography, and studies reported in English, Spanish, French, and German were included
- An NMA was conducted for objective response rate (ORR), progression-free survival (PFS), and overall survival (OS) to compare split-dose GC vs standard GC, GCa, and MVAC
- Results were reported as either hazard ratios (HRs) for PFS and OS or as odds ratios for ORR with 95% credible intervals (CrIs). HRs for OS and PFS that were adjusted for baseline characteristics from the published studies were used in the NMA if available. Since not all studies provided adjusted HRs, sensitivity analyses were conducted using only unadjusted HRs

Figure 1. Literature flow diagram

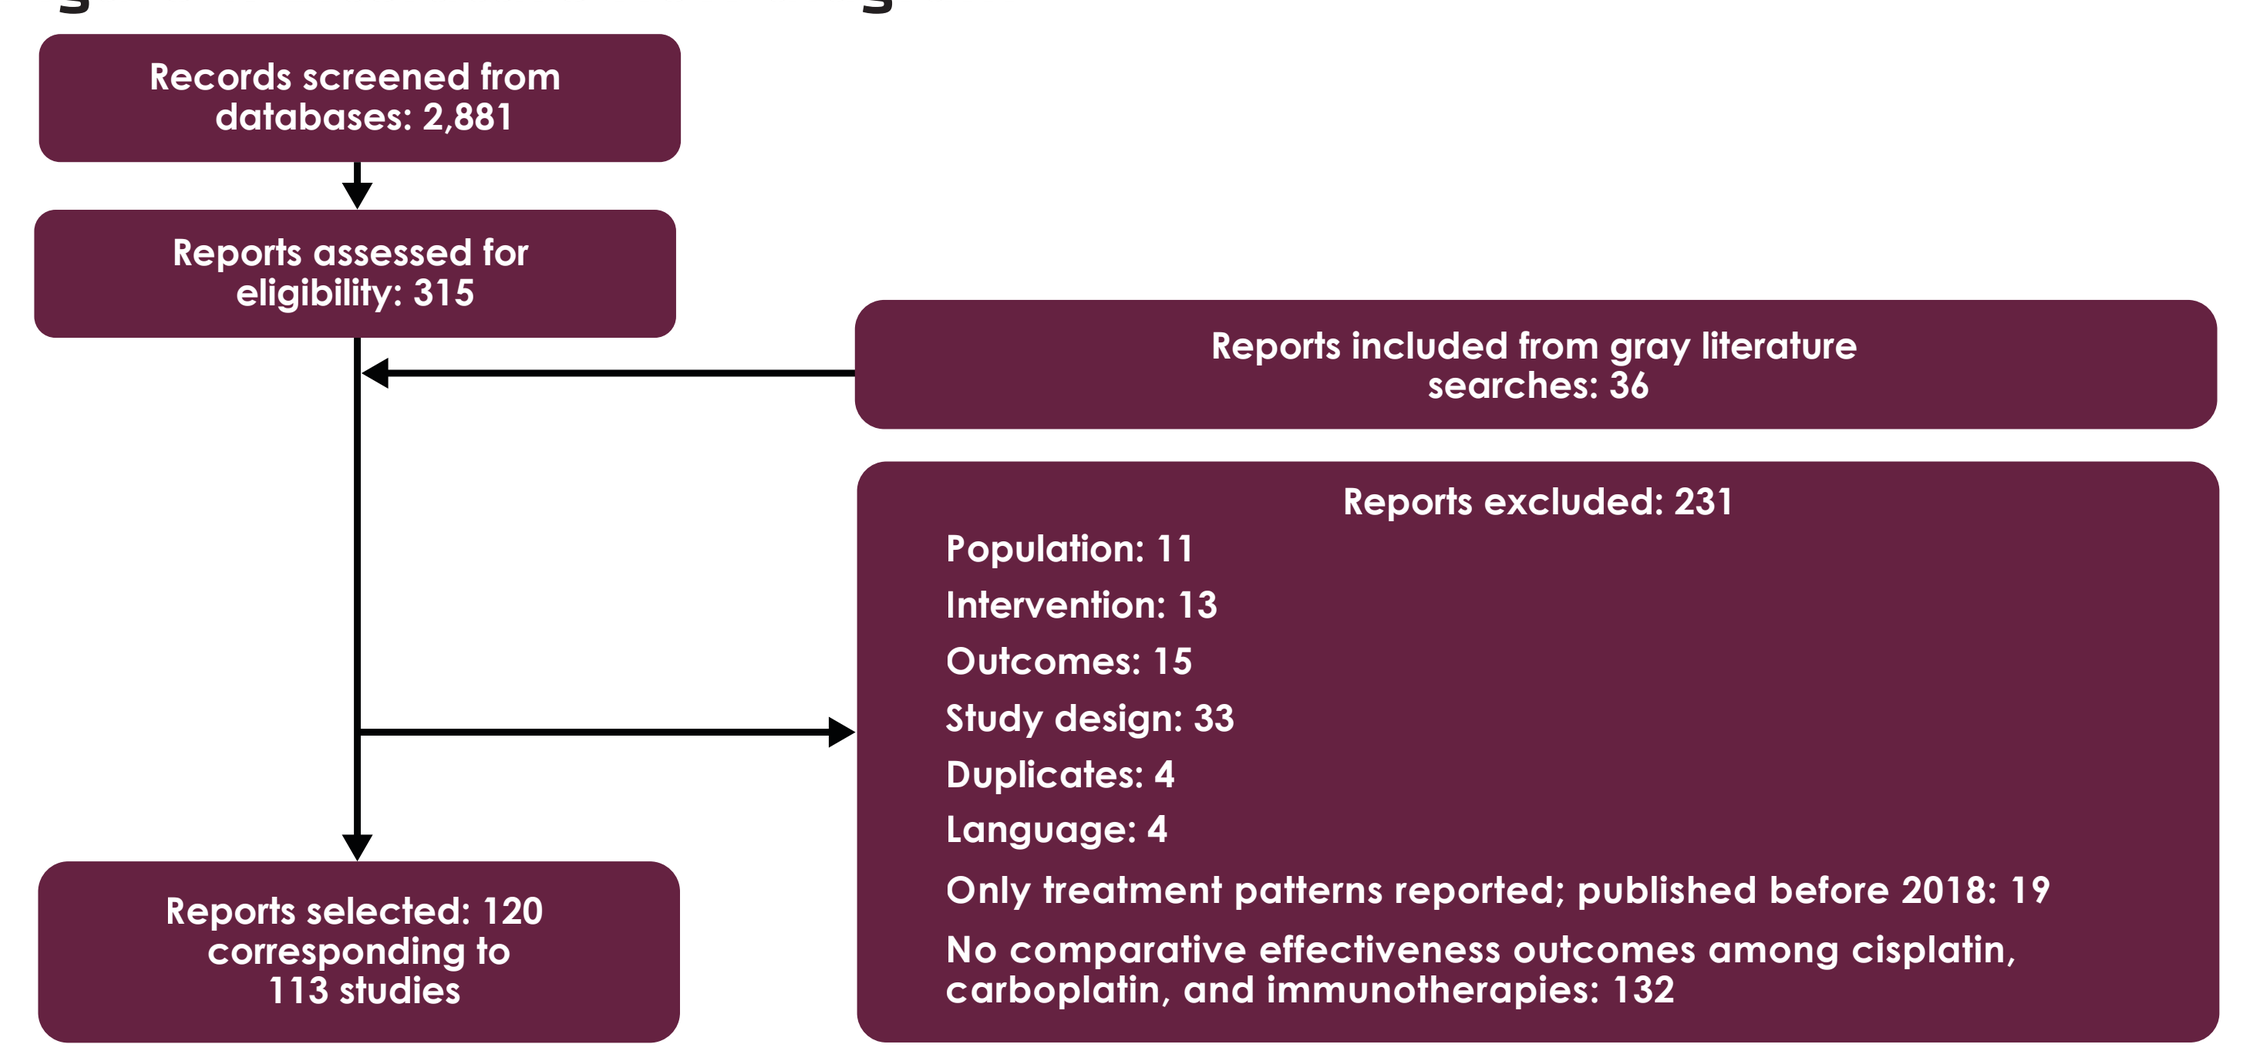
Records screened from databases: 2,881
Reports assessed for eligibility: 315
Reports included from gray literature searches: 36
Reports excluded: 231
Population: 11
Intervention: 13
Outcomes: 15
Study design: 33
Duplicates: 4
Language: 4
Only treatment patterns reported; published before 2018: 19
No comparative effectiveness outcomes among cisplatin, carboplatin, and immunotherapies: 132
Reports selected: 120 corresponding to 113 studies

RESULTS

- Among 2,881 records identified in the database search, 120 publications of 113 unique studies met the prespecified inclusion criteria (**Figure 1**)
- 63 studies reported effectiveness and safety outcomes, including 16 studies in patients with la/mUC treated with split-dose GC
- Only 1 randomized trial compared the efficacy and safety of split-dose GC and GCa (**Figure 2A**)
- Among the 16 studies, split-dose GC was administered in 1L in 10 studies and in 1L or 2L in 3 studies (**Figure 2B**)
- Split-dose GC was most often administered in 2 equal doses of 35 mg/m² on days 1 and 8 or days 1 and 2 of the cycle (in 50% and 25% of studies, respectively) (**Figure 2C**)
- Renal impairment, poor performance status, older age, and comorbidities were associated with administration of split-dose GC (**Figure 2D**)

Figure 2. Characteristics of studies investigating split-dose cisplatin regimens in the SLR

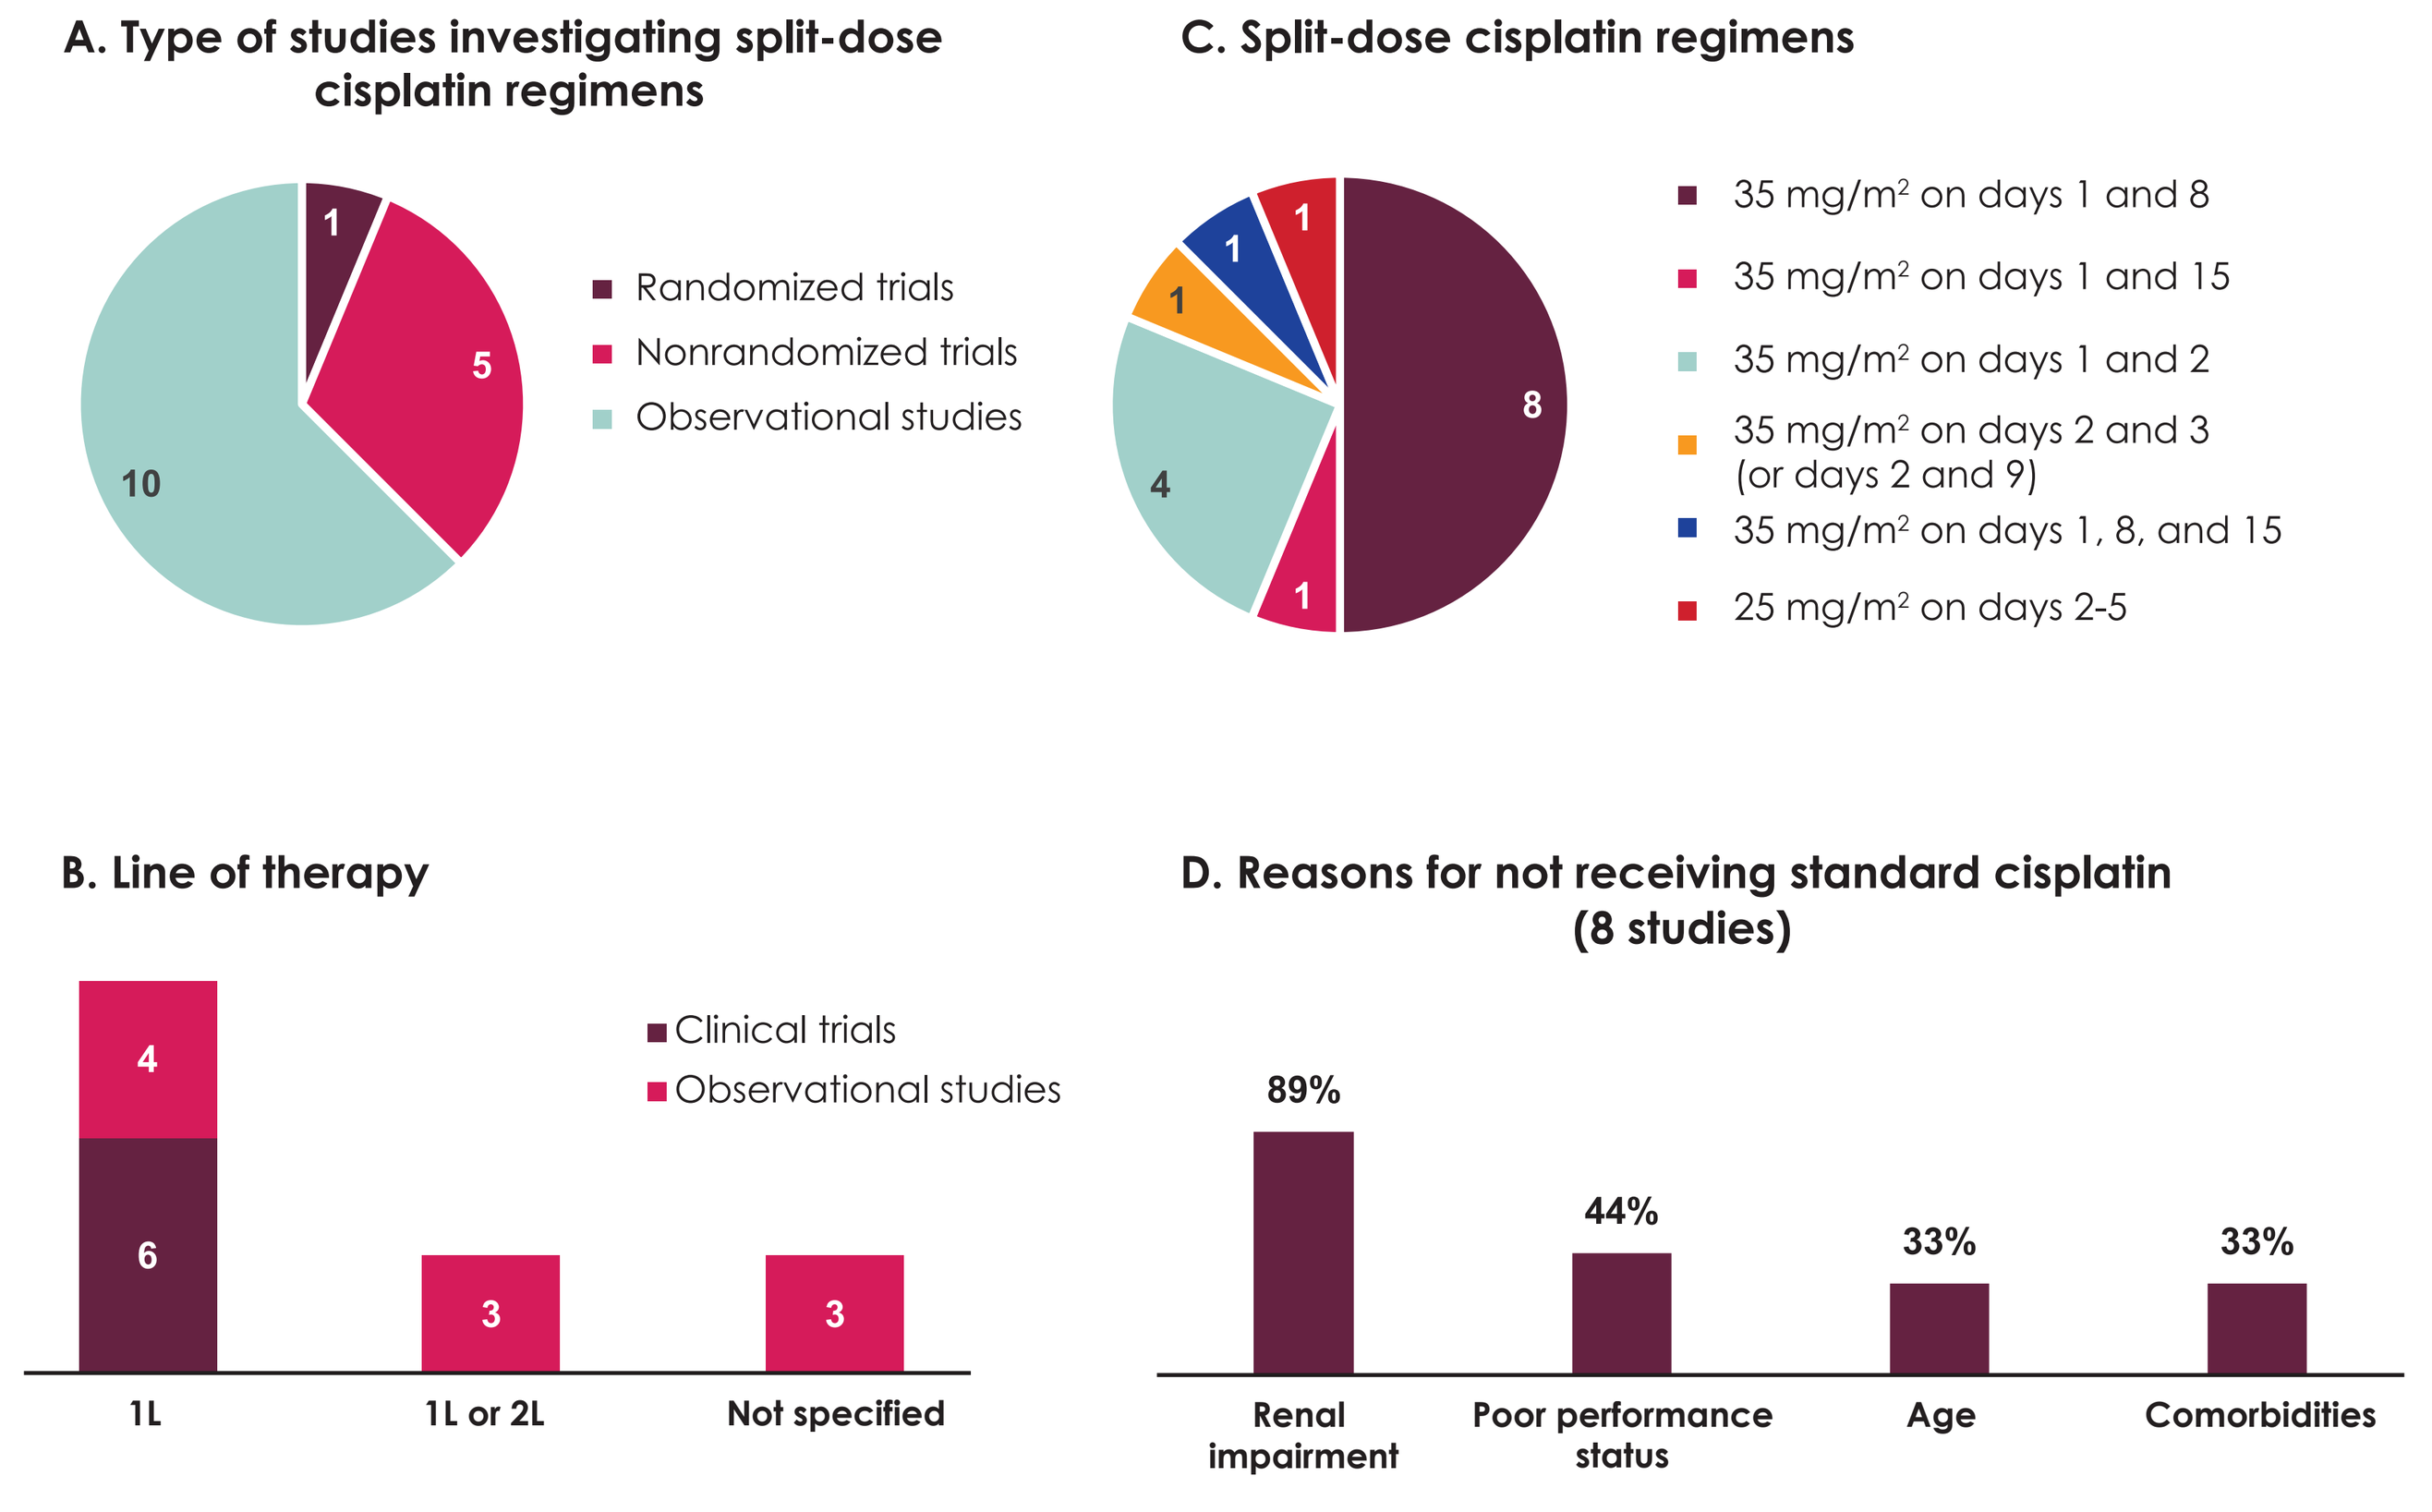
A. Type of studies investigating split-dose cisplatin regimens
Randomized trials 1
Nonrandomized trials 5
Observational studies 10
C. Split-dose cisplatin regimens
35 mg/m² on days 1 and 8: 8
35 mg/m² on days 1 and 15: 1
35 mg/m² on days 1 and 2: 4
35 mg/m² on days 2 and 3 (or days 2 and 9): 1
35 mg/m² on days 1, 8, and 15: 1
25 mg/m² on days 2-5: 1
B. Line of therapy
Clinical trials
Observational studies
1L: 6, 4
1L or 2L: 3
Not specified: 3
D. Reasons for not receiving standard cisplatin (8 studies)
Renal impairment 89%
Poor performance status 44%
Age 33%
Comorbidities 33%

1L, first line; 2L, second line; SLR, systematic literature review.

- In 12 studies reporting outcomes with split-dose GC, ORRs were 39-80% in 11 studies (**Figure 3A**), median PFS was 3.5-9.9 months in 7 studies (**Figure 3B**), and median OS was 8.5-18.1 months in 11 studies (**Figure 3C**)
- Only 3 studies provided comparative efficacy analyses for split-dose GC:
 - Split-dose GC vs GCa: significantly longer median PFS and OS in patients with estimated glomerular filtration rate (eGFR) of 40-60 mL/min[7]
 - Split-dose GC vs GC: higher mortality and progressive disease (adjusted for age, baseline eGFR, performance status, hypertension and diabetes)[8]
 - Split-dose GC vs GC: no significant difference in OS in patients with eGFR 50-60 mL/min[9]

Figure 3. Efficacy of split-dose cisplatin vs standard chemotherapy regimens

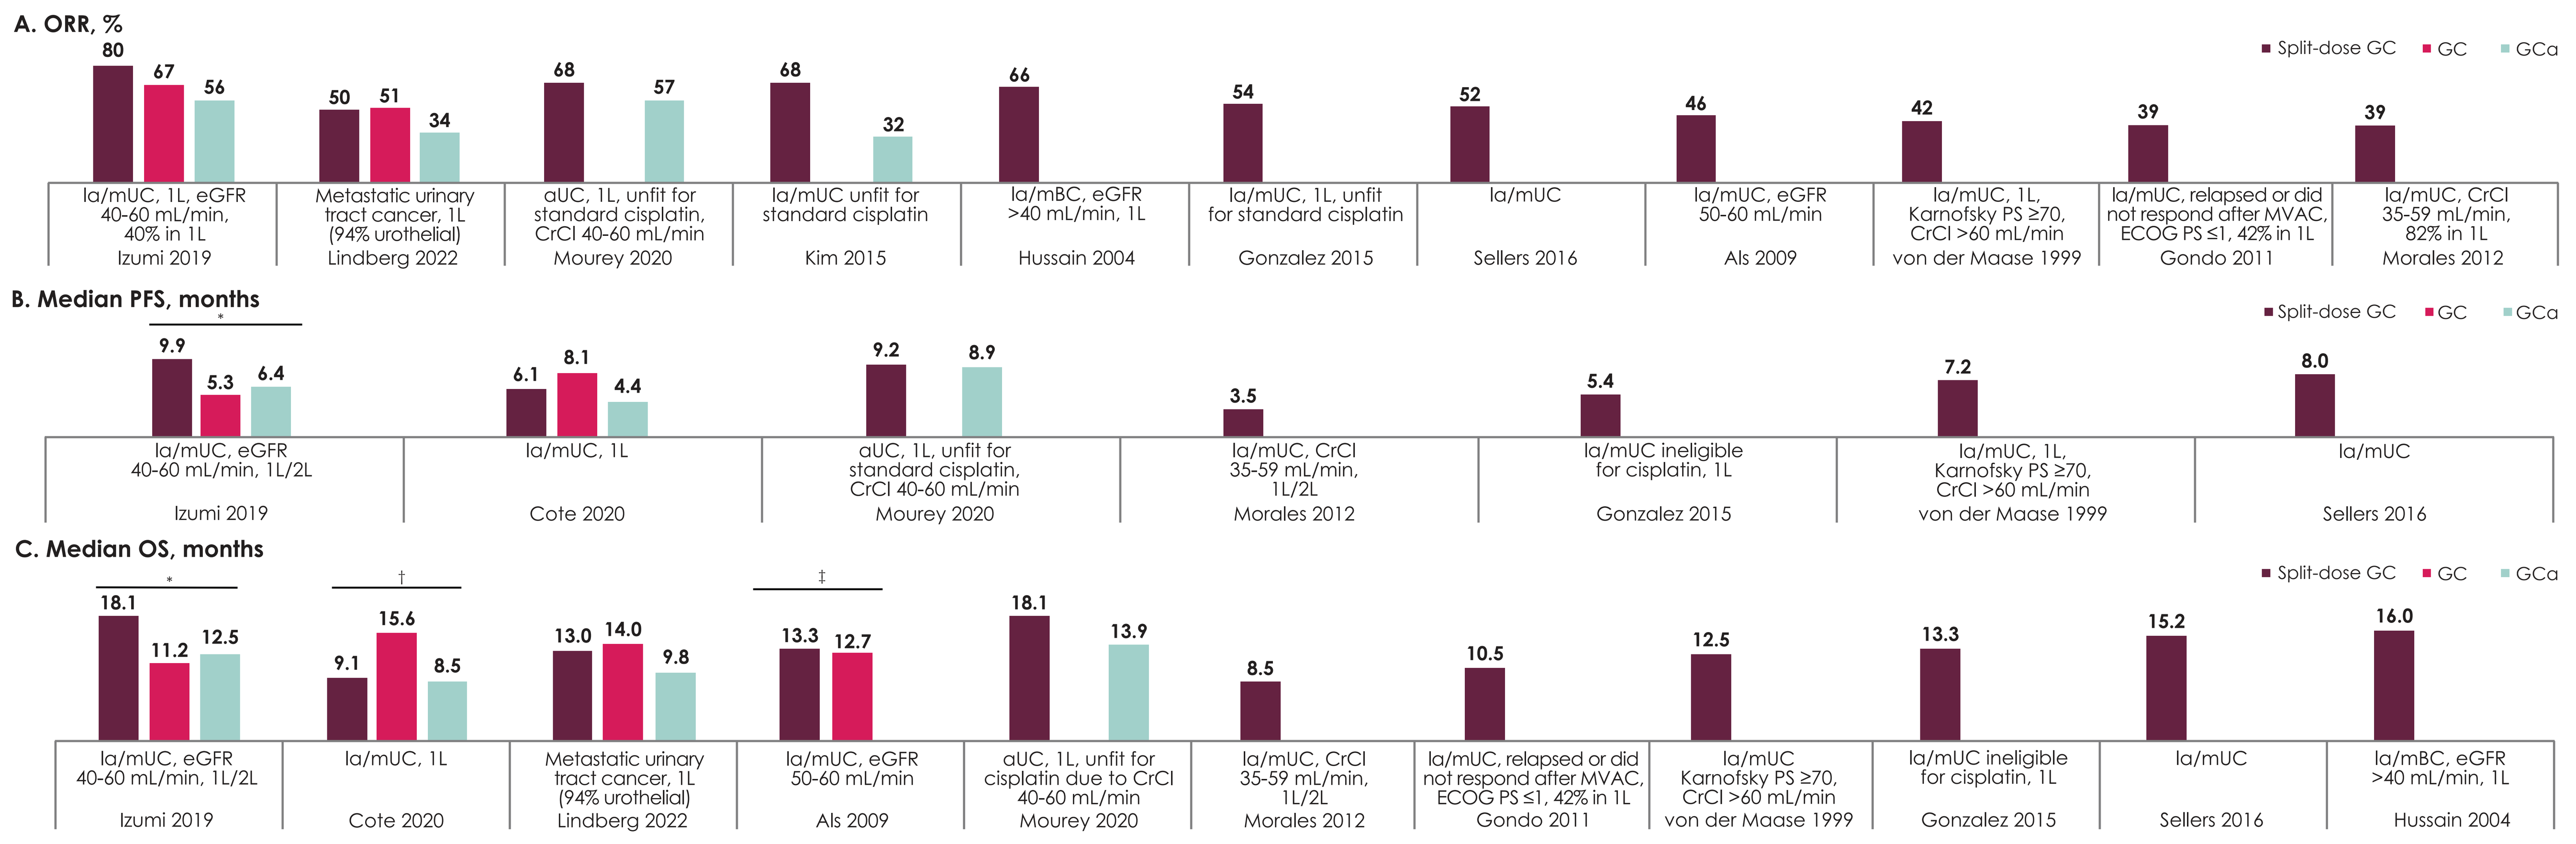
A. ORR, %
Split-dose GC
GC
GCa
la/mUC, 1L, eGFR 40-60 mL/min, 40% in 1L Izumi 2019: 80, 67, 56
Metastatic urinary tract cancer, 1L (94% urothelial) Lindberg 2022: 50, 51, 34
aUC, 1L, unfit for standard cisplatin, CrCl 40-60 mL/min Mourey 2020: 68, 57
la/mUC unfit for standard cisplatin Kim 2015: 68, 32
la/mBC, eGFR >40 mL/min, 1L Hussain 2004: 66
la/mUC, 1L, unfit for standard cisplatin Gonzalez 2015: 54
la/mUC Sellers 2016: 52
la/mUC, eGFR 50-60 mL/min Als 2009: 46
la/mUC, 1L, Karnofsky PS ≥70, CrCl >60 mL/min von der Maase 1999: 42
la/mUC, relapsed or did not respond after MVAC, ECOG PS ≤1, 42% in 1L Gondo 2011: 39
la/mUC, CrCl 35-59 mL/min, 82% in 1L Morales 2012: 39
B. Median PFS, months
la/mUC, eGFR 40-60 mL/min, 1L/2L Izumi 2019: 9.9, 5.3, 6.4 *
la/mUC, 1L Cote 2020: 6.1, 8.1, 4.4
aUC, 1L, unfit for standard cisplatin, CrCl 40-60 mL/min Mourey 2020: 9.2, 8.9
la/mUC, CrCl 35-59 mL/min, 1L/2L Morales 2012: 3.5
la/mUC ineligible for cisplatin, 1L Gonzalez 2015: 5.4
la/mUC, 1L, Karnofsky PS ≥70, CrCl >60 mL/min von der Maase 1999: 7.2
la/mUC Sellers 2016: 8.0
C. Median OS, months
la/mUC, eGFR 40-60 mL/min, 1L/2L Izumi 2019: 18.1, 11.2, 12.5 *
la/mUC, 1L Cote 2020: 9.1, 15.6, 8.5 †
Metastatic urinary tract cancer, 1L (94% urothelial) Lindberg 2022: 13.0, 14.0, 9.8
la/mUC, eGFR 50-60 mL/min Als 2009: 13.3, 12.7 ‡
aUC, 1L, unfit for cisplatin due to CrCl 40-60 mL/min Mourey 2020: 18.1, 13.9
la/mUC, CrCl 35-59 mL/min, 1L/2L Morales 2012: 8.5
la/mUC, relapsed or did not respond after MVAC, ECOG PS ≤1, 42% in 1L Gondo 2011: 10.5
la/mUC Karnofsky PS ≥70, CrCl >60 mL/min von der Maase 1999: 12.5
la/mUC ineligible for cisplatin, 1L Gonzalez 2015: 13.3
la/mUC Sellers 2016: 15.2
la/mBC, eGFR >40 mL/min, 1L Hussain 2004: 16.0

1L, first line; 2L, second line, aUC, advanced urothelial carcinoma; CrCl, creatinine clearance; eGFR, estimated glomerular filtration rate; GCa, gemcitabine + carboplatin; GC, gemcitabine + cisplatin; la/mBC, locally advanced/metastatic bladder cancer; la/mUC, locally advanced or metastatic urothelial carcinoma; MVAC, methotrexate + vinblastine + doxorubicin + cisplatin; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PS, performance status.
*Split-dose GC was significantly better than GCa. †GC was significantly better than split-dose GC. ‡No significant difference was found between interventions.

- In the NMA, a total of 9, 5, and 11 studies reporting ORR, HR for PFS, and HR for OS, respectively, formed connected networks that allowed the comparison of split-dose GC with other chemotherapy regimens (GC, GCa, or MVAC) (**Figure 4**)
- In the base case analyses, the best fit according to the deviance information criteria was obtained with the fixed-effects model for ORR and OS and the random-effects model for PFS
- PFS and OS with split-dose GC were comparable vs GC, GCa, and MVAC (**Figure 4A** and **4B**)
- Split-dose GC was associated with a significantly higher probability of achieving an objective response than GCa (odds ratio, 1.97; 95% CrI, 1.29-3.02). The odds ratio for objective response was not statistically significant between split-dose GC compared with GC and MVAC (**Figure 4C**)
- In sensitivity analyses, split-dose GC was associated with significantly longer median PFS than standard GC, GCa, and MVAC in the fixed-effects model but not in the random-effects model, and with significantly better OS vs all 3 comparators in both the fixed- and random-effects models
- Reported rates of discontinuation due to adverse events with split-dose GC were heterogeneous, but generally consistent with those observed with other chemotherapy regimens, and ranged from 5%-36%

Figure 4. NMA results: comparative efficacy of split-dose GC vs standard chemotherapy regimens

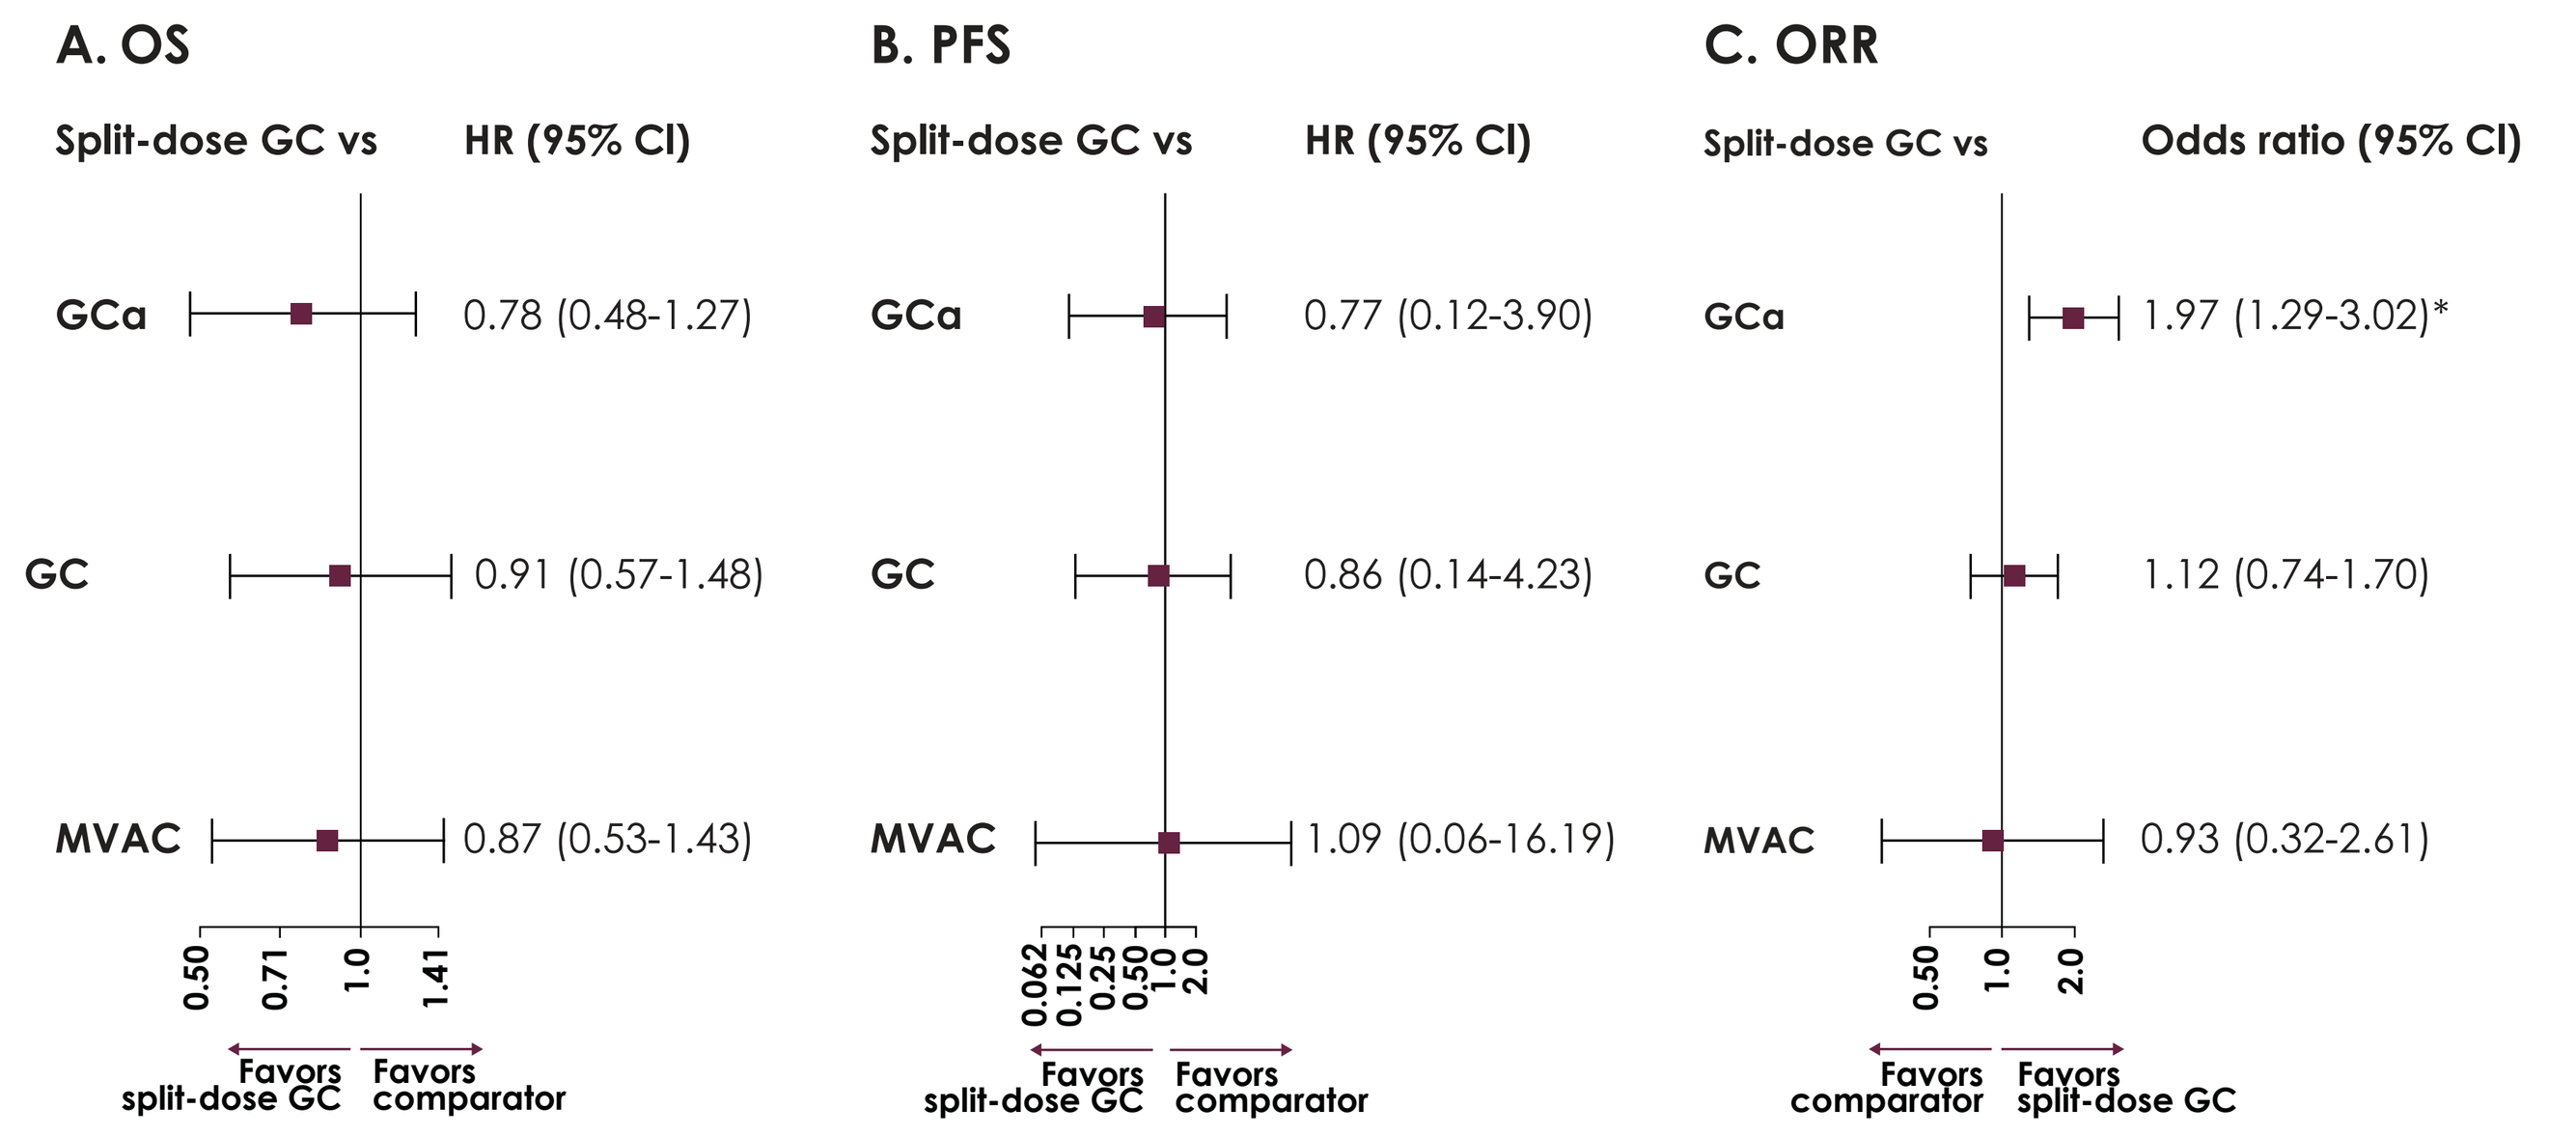
A. OS
Split-dose GC vs
HR (95% CI)
GCa 0.78 (0.48-1.27)
GC 0.91 (0.57-1.48)
MVAC 0.87 (0.53-1.43)
0.50 0.71 1.0 1.41
Favors split-dose GC
Favors comparator
B. PFS
Split-dose GC vs
HR (95% CI)
GCa 0.77 (0.12-3.90)
GC 0.86 (0.14-4.23)
MVAC 1.09 (0.06-16.19)
0.062 0.125 0.25 0.50 1.0 2.0
Favors split-dose GC
Favors comparator
C. ORR
Split-dose GC vs
Odds ratio (95% CI)
GCa 1.97 (1.29-3.02)*
GC 1.12 (0.74-1.70)
MVAC 0.93 (0.32-2.61)
0.50 1.0 2.0
Favors comparator
Favors split-dose GC

GCa, gemcitabine + carboplatin; GC, gemcitabine + cisplatin; HR, hazard ratio; MVAC, methotrexate + vinblastine + doxorubicin + cisplatin; NMA, network meta-analysis; ORR, objective response rate; OS, overall survival; PFS, progression-free survival.
*Denotes a statistically significant difference.

REFERENCES 1. Cathomas R, et al. Eur Urol. 2022;81:95-103. 2. Bamias A, et al. Ann Oncol. 2019;30:1841. 3. Milloy N, et al. Ann Oncol. 2022;33:S1490. 4. Hussain SA, et al. Br J Cancer. 2004;91:844-9. 5. Morales-Barrera R, et al. Eur J Cancer. 2012;48:1816-21. 6. Hamada A, et al. In Vivo. 2021;35:2821-9. 7. Izumi K, et al. In Vivo. 2019;33:167-72. 8. Cote G, et al. Kidney360. 2023;4:e1203-11. 9. Als AB, et al. Acta Oncol. 2008;47:110-9.
DISCLOSURES **R. O'Dwyer** has received support for travel and attending meetings from Pfizer and research funding from EMD Serono. **M. Kearney** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **M. Musat, I. Gulas, E.,** and **H. Moradian** are employees of Cytel, which was contracted by the healthcare business of Merck KGaA, Darmstadt, Germany, for the work reported. **S. Guenther** was an employee of the healthcare business of Merck KGaA, Darmstadt, Germany, at the time the study was conducted. **E. Hubscher** was an employee of Cytel at the time the study was conducted, which was contracted by the healthcare business of Merck KGaA, Darmstadt, Germany, for the work reported. **S. S. Sridhar** has served in consulting or advisory roles for Astellas Pharma, AstraZeneca, Bayer, Bristol Myers Squibb, Janssen, Merck & Co., Kenilworth, NJ, Pfizer, Roche/Genentech, and Seagen; and has received institutional research funding from Bayer, Janssen, and Pfizer. **ACKNOWLEDGMENTS** The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Editorial support was provided by Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 589. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Richard O'Dwyer**, Richard.Odwyer@uhn.ca