Real-world treatment patterns and clinical outcomes in patients with locally advanced or metastatic urothelial carcinoma in Germany: results of a retrospective observational study (CONVINCE)

S. Machtens,[1] K. Schlack,[2] T. Kubin,[3] M. Ruhnke,[4] C. Schulte,[5] A. Eisen,[6] U. Osowski,[7] S Guenther,[8*] M. Kearney,[8] R. Lipp,[6] S. Schmitz[9]

[1]Department of Urology, GFO Hospitals Rhein-Berg, Marien-Hospital, Bergisch-Gladbach, Germany; [2]Department of Urology, University Hospital Muenster, Muenster, Germany; [3]Department für Hematology, Oncology and Palliative Care, Clinics Südostbayern AG, Clinic Traunstein, Cuno-Niggl-Str. 3, 83278, Traunstein, Germany; [4]Clinic for Hematology, Oncology and Palliative Medicine, Helios Clinic Aue, Gartenstr. 6, 08280 Aue, Germany; [5]Practice for Hematology and Oncology, Dortmund, Germany; [6]GermanOncology GmbH, Überseeallee 1,20457 Hamburg, Germany; [7]Merck Healthcare Germany GmbH, Weiterstadt, Germany, an affiliate of Merck KGaA, Darmstadt, Germany; [8]the healthcare business of Merck KGaA, Darmstadt, Germany; [9]Medical Care Center for Oncology and Hematology, Am Sachsenring 59; 50677 Cologne, Germany

*Affiliation at the time the study was conducted.

CONCLUSIONS

- **CONVINCE is a retrospective, observational, multicenter study examining treatment patterns and associated outcomes in patients with locally advanced or metastatic urothelial cancer (la/mUC) treated in routine clinical practice in Germany between 2019 and 2021**
 - **This analysis compares demographics, treatment patterns, and associated clinical outcomes in patients with la/mUC who received first-line (1L) platinum-based chemotherapy (PBC), immuno-oncology (IO) monotherapy, or non-PBC**
- **Our results provide valuable insights about 1L systemic treatment in patients with la/mUC in routine clinical practice in Germany following the introduction of IOs**
- **Consistent with treatment guidelines, most patients (76.1%) received PBC as 1L systemic treatment, of whom approximately half received cisplatin + gemcitabine**
- **High attrition occurred across lines of treatment, with approximately 1 in 2 patients with la/mUC receiving a second-line (2L) therapy and 1 in 3 receiving a third-line or later-line therapy**
- **New IO approvals, including avelumab 1L maintenance and other novel agents, may improve outcomes in patients with la/mUC**
- **Given the rapidly evolving treatment landscape in la/mUC, further real-world studies should be performed to evaluate the uptake of these options in routine care**

PLAIN LANGUAGE SUMMARY

- **In the CONVINCE study, researchers looked at medical records of people receiving treatment for advanced urothelial cancer in Germany between January 2019 and June 2021**
- **First-line treatment (the first treatment that people received after developing advanced urothelial cancer) was chemotherapy including a platinum drug in 76% of people, immunotherapy in 19% of people, and other types of chemotherapy in 5% of people**
 - **51% of people received a second treatment when their first-line treatment stopped working, and 30% received a third treatment**
- **On average, people treated with first-line platinum-based chemotherapy lived for 18 months, and people treated with first-line immunotherapy lived for 16 months**
- **Overall, these findings from the CONVINCE study show that treatment for advanced urothelial cancer in Germany mostly follows guidelines for treatment that were developed by experts**

BACKGROUND

- Bladder cancer, 90% of which is UC,[1] is the fifth most common cancer in Germany, with >30,000 new cases diagnosed each year[2,3]
- In 2020, 3.4% of all cancer-related deaths in Germany were due to bladder cancer[2]
- In patients with unresectable la/mUC, current guidelines from the German Cancer Society, the European Association of Urology, and the European Society for Medical Oncology recommend 1L cisplatin-based chemotherapy in eligible patients[4-6]
- 1L PBC followed by avelumab 1L maintenance in patients without disease progression is a standard-of-care treatment for patients with la/mUC since the European Commission's (EC) approval of avelumab maintenance in January 2021[7]
- The use of PBC and non-PBC therapies in the 1L treatment of patients with la/mUC has not been widely examined in clinical routine care in Germany, especially since approvals of IO monotherapy and other novel agents

METHODS

Study design

- CONVINCE, a retrospective, multicenter medical chart review study, was initiated in December 2021 and included patients who received 1L therapy for la/mUC between January 2019 and September 2021[8]
- Data were obtained from 27 oncology or urology institutions (8 hospitals and 19 office-based practices) across Germany (**Figure 1**); the institutions were geographically dispersed and of various sizes
- Fully anonymized data were extracted from patient medical charts and entered into electronic case report forms. Parameters included sex, age, UC history, ECOG performance status (PS) at diagnosis, treatments received, and clinical outcomes
- The study population was divided into 3 treatment groups: PBC, IO, or non-PBC (**Figure 2**)
- Clinical outcomes, including objective response rate (ORR), disease control rate (DCR), real-world progression-free survival (rwPFS), and real-world overall survival (rwOS), were described for the PBC and IO groups (the sample size for the non-PBC group was too small for calculations) and were defined from the start of 1L treatment
- Due to the one-point retrospective documentation and full anonymization with no option for longitudinal follow-up, rwPFS and rwOS were calculated from date of first drug administration up to date of progression or death; if none of these events occurred, then by last contact before documentation
 - Median values for rwPFS and rwOS represent "minimum values" and data in the Kaplan-Meier curves are descriptive; as the number of events at the time point of documentation was too small and longer follow-up was not possible due to the full anonymization
- The choice of regimen administered to each patient (PBC, IO, or non-PBC) was determined by the attending physician

Figure 1. Overview of participating sites and physician disciplines

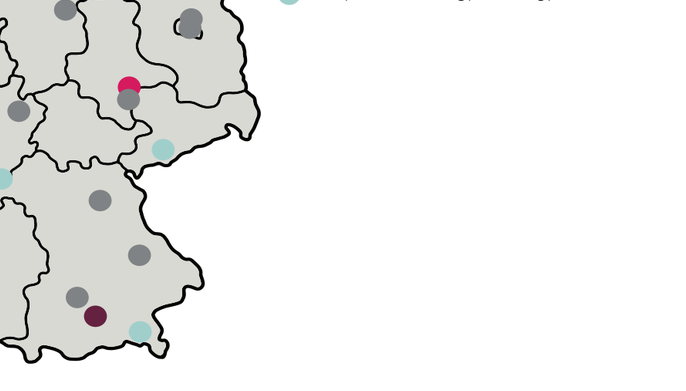

Office-based urology practice
Hospital urology
Office-based hematology/oncology practice
Hospital hematology/oncology

Participating sites, n (%)	N=27
Hospital urology	4 (15)
Hospital oncology	4 (15)
Office-based urology practice	3 (11)
Office-based oncology practice	16 (59)
Medical speciality of sites, n (%)	**N=27**
Urology	7 (26)
Medical oncology	20 (74)
Distribution of patients by medical specialty, n (%)	**N=188**
Hospital urology	61 (32.5)
Hospital oncology	17 (9.0)
Office-based urology practice	14 (7.4)
Office-based oncology practice	96 (51.1)

Figure 2. CONVINCE study overview

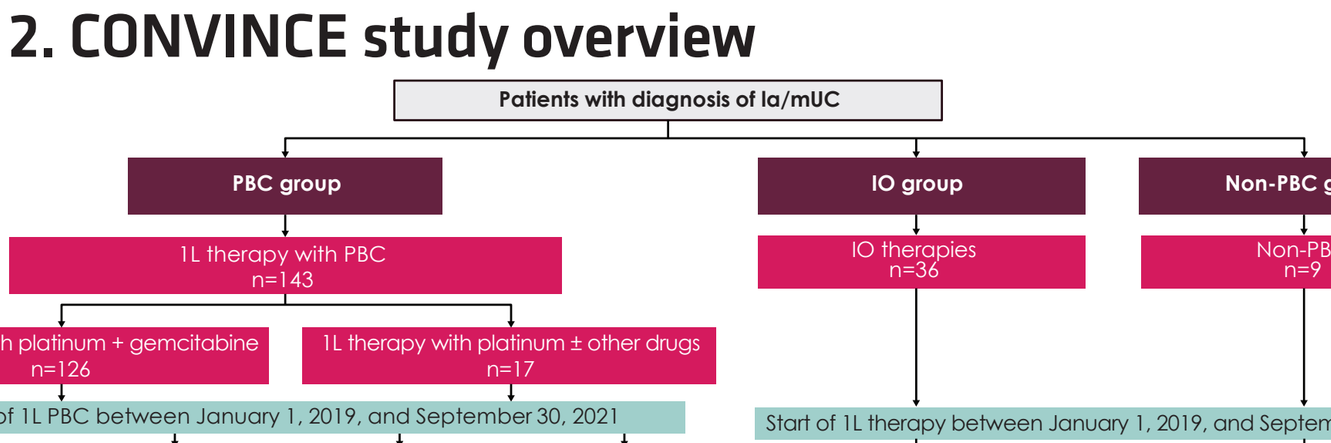

Patients with diagnosis of la/mUC
PBC group
IO group
Non-PBC group
1L therapy with PBC n=143
IO therapies n=36
Non-PBC n=9
1L therapy with platinum + gemcitabine n=126
1L therapy with platinum ± other drugs n=17
End of 1L PBC between January 1, 2019, and September 30, 2021
Start of 1L therapy between January 1, 2019, and September 30, 2021
Platinum + gemcitabine without avelumab n=120
Platinum + gemcitabine with avelumab n=6
Platinum ±other drugs without avelumab n=17
Platinum + other drugs with avelumab n=0
Subsequent therapies (2L-6L)

Inclusion of patients in study and documentation by sites between February 2022 and February 2023.
1L, first line; **2L,** second line; **6L,** sixth line; **IO,** immuno-oncology; **la/mUC,** locally advanced or metastatic urothelial cancer; **PBC,** platinum-based chemotherapy.

Eligibility criteria

- Patients aged ≥18 years with la/mUC diagnosis
- 1L PBC had to be completed between January 1, 2019, and June 30, 2021, with a follow-up of ≥6 months after last administration
- Patients were excluded if they participated in an interventional clinical trial within 30 days before the start of 1L PBC

Ethics approval

- This study was performed in accordance with the Declaration of Helsinki
- Because of the retrospective nature of the study and the anonymized data collection, the need for patient informed consent and advice according to the German doctors' professional code of conduct was waived by the independent ethics board of North Rhine-Westphalia

Statistical analysis

- Patient characteristics, treatment patterns, and best response were analyzed descriptively
- Time-to-event outcomes, including rwPFS and rwOS, were analyzed using the Kaplan-Meier method

RESULTS

Patient characteristics

- The median age of patients at the start of 1L treatment was 70 years (range, 41-87 years) in the overall population (N=188) and 69 years (range, 48-84 years) in the PBC group (**Table 1**). Patients in the IO group were the oldest, with a median age of 77 years (range, 41-87 years)
- Most patients (92.5%) had an ECOG PS of 0/1 at first diagnosis, and 6.9% had an ECOG PS of 2. The highest percentage of patients with an ECOG PS of 2 was in the IO group (13.9%) and the lowest percentage was in the PBC group (4.9%)
- Most patients (72.3%) were male; the percentage of male patients was highest in the non-PBC group (88.9%) and lowest in the IO group (66.7%)

Treatment patterns

- Of the total patient population enrolled (N=188), 143 (76.1%) received PBC, 36 (19.1%) received IO, and 9 (4.8%) received non-PBC as 1L treatment (**Table 2**)
- The rate of prior neoadjuvant/adjuvant therapies was higher in the IO group (13.9%/16.7%) than in the PBC group (2.0%/5.6%) (**Table 1**)

Table 1. Baseline characteristics of the total population and by treatment groups

	Total population	PBC group	IO group	Non-PBC group
Patients, n (%)	188 (100)	143 (76.1)	36 (19.1)	9 (4.8)
Sex, %				
Male	72.3	72.7	66.7	88.9
Female	27.7	27.3	33.3	11.1
Age at first diagnosis, median (range), years	69 (39-87)	68 (48-84)	75 (39-87)	70 (68-79)
Age at start of 1L therapy, median (range), years	70 (41-87)	69 (48-84)	77 (41-87)	70 (68-80)
ECOG PS, %				
0	50.5	51.7	52.8	22.2
1	42.0	43.4	33.3	55.6
2	6.9	4.9	13.9	11.1
Unknown	0.5	0	0	11.1
Neoadjuvant therapies, n (%)	8 (4.3)	3 (2.0)	5 (13.9)	0
Adjuvant therapies, n (%)	17 (9.0)	8 (5.6)	6 (16.7)	3 (33.3)
No. of cycles in 1L, median (range)	5 (1-42)	4 (1-7)	13 (2-42)	3 (1-8)
Subsequent therapies, n (%)				
2L	96 (51.1)	76 (53.1)	13 (36.1)	6 (66.7)
≥3L	64 (34.0)	56 (39.2)	4 (11.1)	4 (44.4)
Survival status of patients, n (%)				
Alive at time of inclusion	109 (58.0)	82 (57.3)	19 (52.8)	8 (88.9)

1L, first line; **2L,** second line; **3L,** third line; **IO,** immuno-oncology; **PBC,** platinum-based chemotherapy; **PS,** performance status.

Table 2. Distribution of 1L therapies by treatment group

PBC group			IO group			Non-PBC group		
n/N, %	**143/188**	**76.1%**	n/N, %	**36/188**	**19.1%**	n/N, %	**9/188**	**4.8%**
Therapy	**n**	**%**	**Therapy**	**n**	**%**	**Therapy**	**n**	**%**
Cisplatin + gemcitabine	98	68.5	Atezolizumab	16	44.4	Gemcitabine	6	66.7
Cisplatin + gemcitabine → avelumab 1LM	6	4.2	Pembrolizumab	15	41.7	Vinflunine	2	22.2
Carboplatin + gemcitabine	22	15.4	Nivolumab	5	13.9	Mitomycin	1	11.1
Cisplatin monotherapy	5	3.5	**Total**	**36**	**100**	**Total**	**9**	**100**
Cisplatin + doxorubicin + methotrexate + vinblastine	4	2.8						
Cisplatin + etoposide	4	2.8						
Carboplatin + etoposide	2	1.4						
Carboplatin + paclitaxel	2	1.4						
Total	**143**	**100**						

1L, first line; **1LM,** first-line maintenance; **IO,** immuno-oncology; **PBC,** platinum-based chemotherapy.

- Distribution of 1L therapies is described in **Table 2**
- Of 143 patients treated with 1L PBC, 126 (88.1%) received platinum + gemcitabine, 12 received platinum with non-gemcitabine agents, and 5 (3.5%) received cisplatin monotherapy
 - Of 126 patients who received platinum + gemcitabine, 104 (82.5%) received cisplatin and 22 (17.5%) received carboplatin (**Table 1**)
- IO agents were used only as 1L monotherapies; 16 of 36 patients (44.4%) received atezolizumab and 15 of 36 (41.7%) received pembrolizumab
- Avelumab 1L maintenance therapy following 1L PBC was used in 6 of 143 patients (4.2%). This low rate is explained by the eligibility criteria (end of 1L PBC therapy in 2019 to September 2021 with ≥6 months of follow-up) and avelumab's EC approval date occurring late in the study period[7]
- Median follow-up was 16.5 months. At the time of documentation by sites, 58.0% of patients were alive (no longitudinal follow-up was possible due to full anonymization)
- The median number of cycles administered was 5 (range, 1-42) in the total population, 4 (range, 1-7) in the PBC group, 13 (range, 2-42) in the IO group, and 3 (range, 1-8) in the non-PBC group (**Table 1**)
- The percentage of patients receiving 2L treatment was 51.1%; this percentage was highest in the non-PBC group (66.7%) and lowest in the IO group (36.1%) (**Table 1**)
- Reasons for nonplatinum use in 1L included decision of physicians (28.9%), impaired renal function (20.0%), other preexisting diseases (15.6%), and cisplatin use as adjuvant therapy (8.9%) (**Table 3**)

Table 3. Reasons for nonplatinum use in 1L

Patients without platinum in 1L (IO + non-PBC) groups, n (%)	n=45
Decision of physician	13 (28.9)
Impaired renal function	9 (20.0)
Preexisting diseases	7 (15.6)
Older age	5 (11.1)
Adjuvant therapy with cisplatin	4 (8.9)
More convenient outpatient administration of other agents (eg, short infusion of IO only once every 3 weeks)	4 (8.9)
Patient preference	2 (4.4)
Unknown	1 (2.2)

1L, first line; **IO,** immuno-oncology; **PBC,** platinum-based chemotherapy.

Clinical outcomes

- Clinical outcomes are shown only for the PBC and IO groups due to the very small sample size of the non-PBC group (n=9)
- Best overall response to 1L systemic treatment was assessed by physicians. Data were available in the patient medical records for best response in 136 of 143 patients (95.1%) in the PBC group and 30 of 36 (83.3%) in the IO group
- The ORR was 56.6% in the PBC group and 60.0% in the IO group
- The DCR was 86.8% in the PBC group and 76.7% in the IO group
- No significant difference was observed in rwPFS and rwOS with PBC and IO at documentation, but there was a nonsignificant trend toward a longer rwPFS with IO and a longer rwOS with PBC, which was analysed descriptively by the Kaplan-Meier method (**Table 4** and **Figure 3**)
- All avelumab 1L maintenance–treated patients (n=6) received cisplatin + gemcitabine as induction therapy
- Median PFS from the start of avelumab was 5.9 months

Table 4. Clinical outcomes and best response to 1L therapy

	Total population	PBC group	IO group
n/N (%)	**188/188 (100)**	**143/188 (76.1)**	**36/188 (19.1)**
Clinical outcomes			
rwPFS			
12-month rate, %	64	60	78
18-month rate, %	54	51	64
Median (95% CI), months*	10.1 (9.2-11.6)	10.5 (9.2-11.6)	12.6 (8.9-22.9)
rwOS			
12-month rate, %	88	90	81
18-month rate, %	70	70	64
Median (95% CI), months†	18.0 (16.3-18.7)	18.1 (16.5-19.0)	15.9 (11.1-24.5)
Best response (physician assessment), n (%)			
Evaluable patients	**172 (91.5)**	**136 (95.1)**	**30 (83.3)**
CR	25 (14.5)	19 (14.0)	6 (20.0)
PR	75 (43.6)	58 (42.6)	12 (40.0)
SD	47 (27.3)	41 (30.1)	5 (16.7)
PD	25 (14.5)	18 (13.2)	7 (23.3)
ORR (CR + PR)	**100 (58.1)**	**77 (56.6)**	**18 (60.0)**
DCR (CR + PR + SD)	**147 (85.5)**	**118 (86.8)**	**23 (76.7)**

1L, first line; **CR,** complete response; **DCR,** disease control rate; **ORR,** objective response rate; **PD,** progressive disease; **PR,** partial response; **rwOS,** real-world overall survival; **rwPFS,** real-world progression-free survival; **SD,** stable disease.
*Calculated from date of first drug administration to date of tumor progression or last contact. †Calculated from date of first drug administration to date of death or last contact.

Figure 3. Kaplan-Meier analysis of rwPFS (A) and rwOS (B) in the PBC and IO groups (n=179)

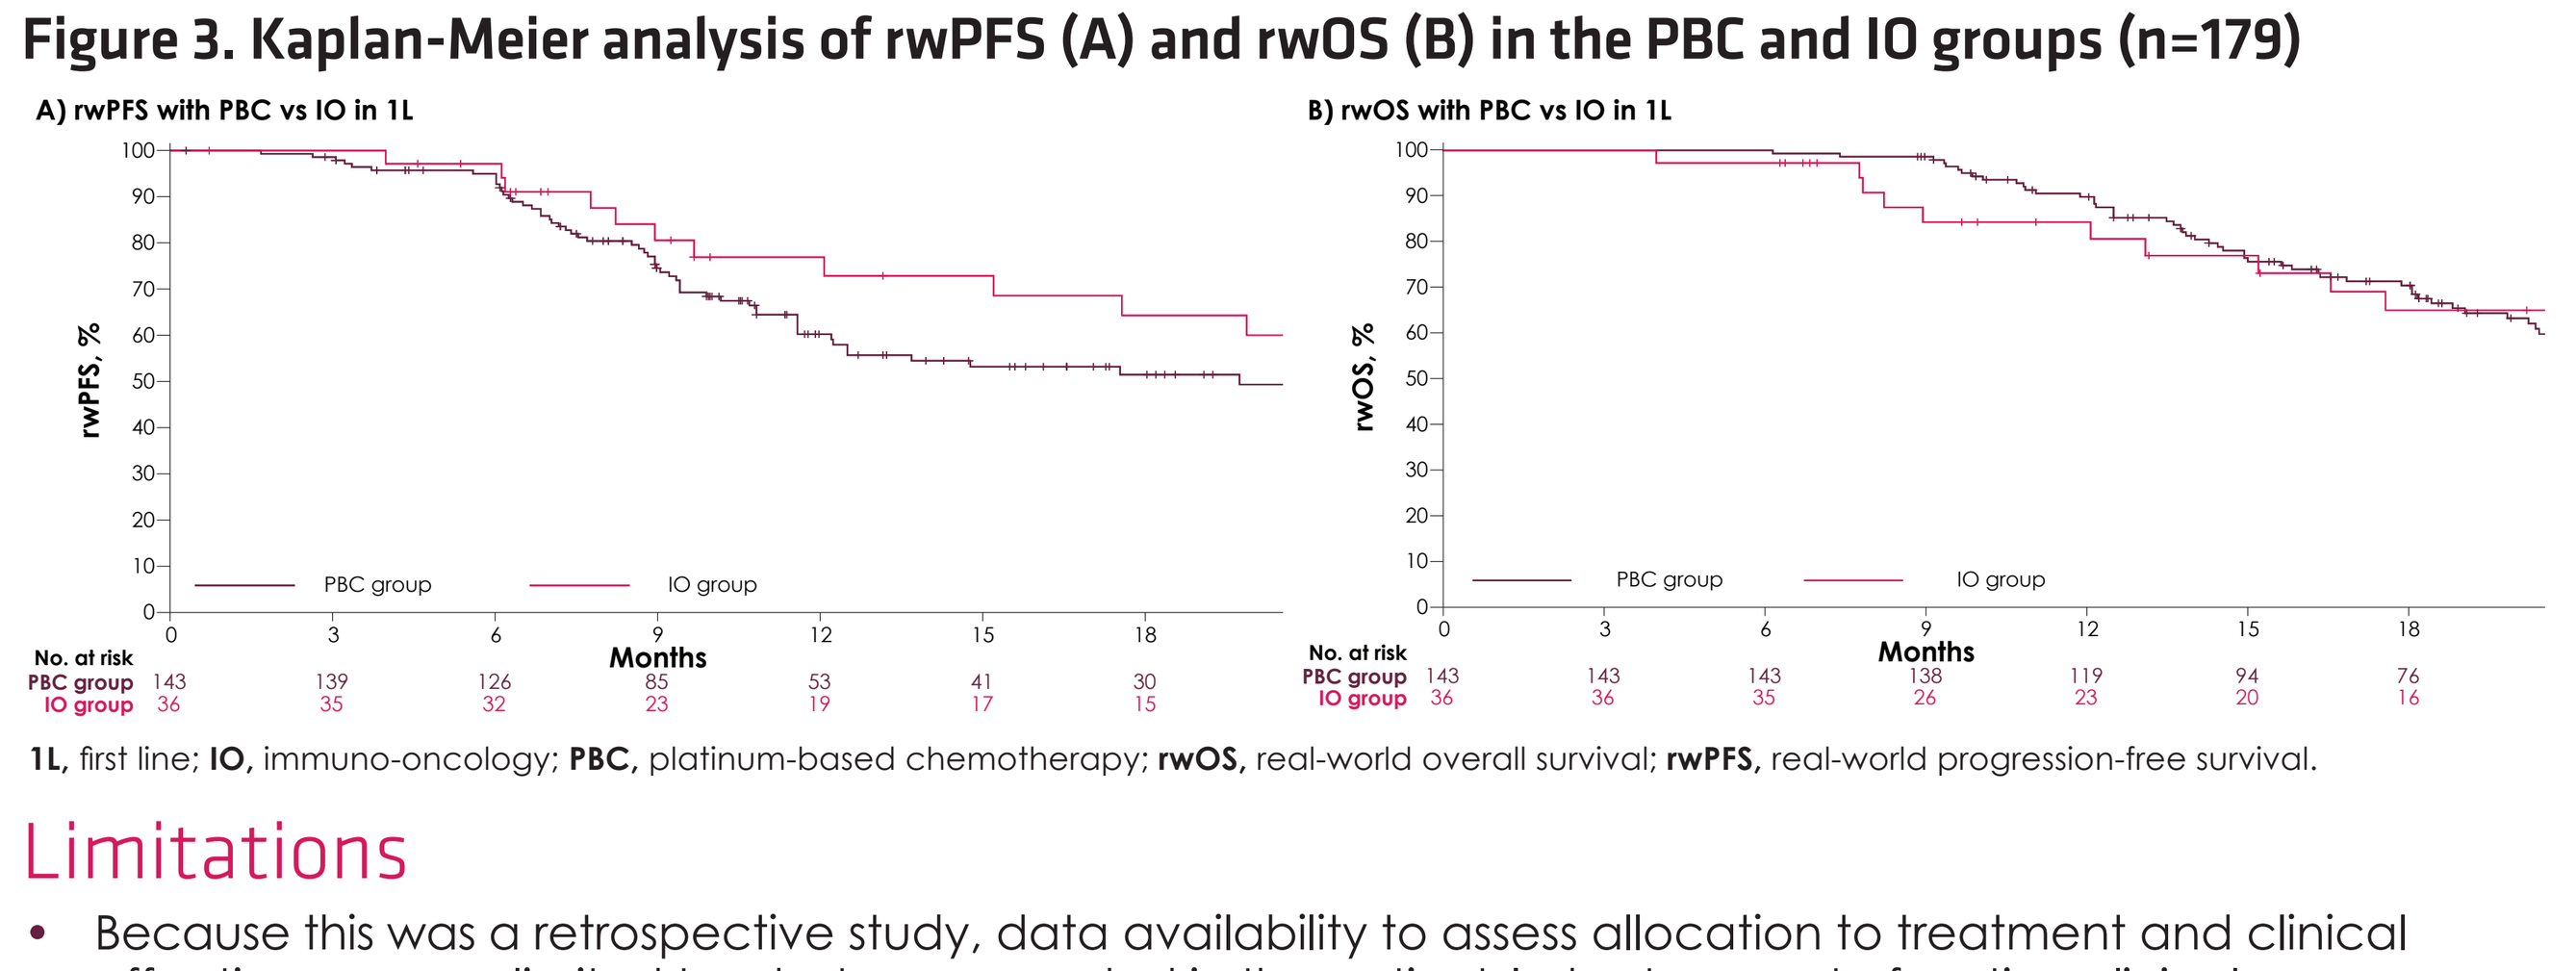

A) rwPFS with PBC vs IO in 1L
rwPFS, %
PBC group
IO group
Months
No. at risk
PBC group 143 139 126 85 53 41 30
IO group 36 35 32 23 19 17 15
B) rwOS with PBC vs IO in 1L
rwOS, %
PBC group
IO group
Months
No. at risk
PBC group 143 143 143 138 119 94 76
IO group 36 36 35 26 23 20 16

1L, first line; **IO,** immuno-oncology; **PBC,** platinum-based chemotherapy; **rwOS,** real-world overall survival; **rwPFS,** real-world progression-free survival.

Limitations

- Because this was a retrospective study, data availability to assess allocation to treatment and clinical effectiveness was limited to what was recorded in the patients' charts as part of routine clinical care
- Although steps were taken to ensure that complete and accurate information was obtained from the medical charts, we cannot fully exclude the potential for information bias if data were missing
- Differences in baseline patient characteristics, disease location, and disease behavior may contribute to variability in clinical effectiveness between the treatment groups studied
- Due to a very small sample size, the non-PBC group was not included in evaluation of clinical outcomes
- The rwOS results must be interpreted with caution due to immortal time bias being introduced during the inclusion period. Due to the low number of OS events, median rwOS is immature and calculated from start of 1L therapy to date of death or last contact
- The use of avelumab as maintenance therapy following 1L PBC in this study was limited due to eligibility criteria with end of PBC therapy in 2019 to September 2021 with ≥6 months of follow-up and the EC approval of avelumab granted in January 2021, which was late in the study period

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Stefan Machtens, stefan.machtens@gfo-kliniken-rhein-berg.de**

REFERENCES 1. Saginala K, et al. Med Sci (Basel). 2020;8(1):15. **2.** International Agency for Research on Cancer. Globocan 2020: Population fact sheets. Accessed December 3, 2023. https://gco.iarc.fr/today/data/factsheets/populations/276-germany-fact-sheets.pdf. **3.** Robert Koch Institut. Cancer in Germany 2017/2018. Accessed December 3, 2023. https://www.krebsdaten.de/Krebs/EN/Content/Publications/Cancer_in_Germany/cancer_in_germany.html. **4.** Cathomas R, et al. Eur Urol. 2022;81(1):95-103. **5.** Powles T, et al. Ann Oncol. 2022;33(3):244-58. **6.** Retz M, et al. Urologe A. 2016;55(9):1173-87. **7.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA, Darmstadt, Germany; 2022. **8.** Schlack K, et al. Ann Oncol. 2023;34(Suppl 2):Abstract 2388P. **DISCLOSURES S. Machtens** has served in consulting or advisory role for Amgen, AstraZeneca, Bayer, BD, Bebig, Janssen-Cilag, Merck & Co., Kenilworth, NJ, Pfizer, and Sanofi-Aventis; and has participated in speakers bureaus for Amgen, Apogepha, Astellas, AstraZeneca, Bayer, BD, Bebig, Ipsen, Janssen-Cilag, Pfizer, Merck & Co., Kenilworth, NJ, Novartis, Sanofi-Aventis, Takeda, and the healthcare business of Merck KGaA, Darmstadt, Germany. **K. Schlack** has served in speakers bureau and consulting or advisory roles for AAA, Amgen, Apogepha, Astellas, AstraZeneca, Bayer, Bristol Myers Squibb (personal and institutional), Eisai, EUSA-Pharma, Fosanis, Ipsen, Janssen Cilag, Merck & Co., Kenilworth, NJ, Novartis (personal and institutional), Pfizer (personal and institutional), Roche, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel, accommodations, and expenses from Astellas, AstraZeneca, Bayer, Janssen Cilag, Ipsen, Sanofi-Aventis, and the healthcare business of Merck KGaA, Darmstadt, Germany. **T. Kubin** has served in speakers bureau and consulting or advisory roles for AstraZeneca, BMS, Boehringer-Ingelheim, GSK, Janssen, Merck & Co., Kenilworth, NJ, Roche, and Takeda. **M. Ruhnke, C. Schulte,** and **A. Eisen** have nothing to disclose. **U. Osowaki** reports employment with Merck Healthcare Germany GmbH, Weiterstadt, Germany, an affiliate of Merck KGaA, Darmstadt, Germany. **S. Guenther** reported employment with the healthcare business of Merck KGaA, Darmstadt, Germany at time the study was conducted; and reports stock or other ownership with the healthcare business of Merck KGaA, Darmstadt, Germany. **M. Kearney** reports employment with the healthcare business of Merck KGaA, Darmstadt, Germany; and reports stock and other ownership interests with Novartis, the healthcare business of Merck KGaA, Darmstadt, Germany, and UCB. **R. Lipp** has served in consulting or advisory role for AstraZeneca, Gilead, Janssen-Cilag, Omnicare, Sanofi, Takeda, and the healthcare business of Merck KGaA, Darmstadt, Germany; has served in speakers bureau for the healthcare business of Merck KGaA, Darmstadt, Germany; and has provided project support for AbbVie, AstraZeneca, Bristol Myers Squibb, Gilead, Janssen-Cilag, Pfizer, and the healthcare business of Merck KGaA, Darmstadt, Germany. **S. Schmitz** has nothing to disclose. **ACKNOWLEDGMENTS** The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Editorial support was provided by Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 551. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.