Avelumab first-line maintenance for advanced urothelial carcinoma: long-term patient-reported outcomes in the phase 3 JAVELIN Bladder 100 trial

P. Grivas,[1] J.B. Aragon-Ching,[2] J. Bellmunt,[3] Y. Loriot,[4] S. S. Sridhar,[5] P.-J. Su,[6] S. H. Park,[7] Y. Yamamoto,[8] N. Jacob,[9] J. Hoffman,[10] M. Kearney,[9] M. Schlichting,[9] T. Powles[11]

[1]University of Washington; Fred Hutchinson Cancer Center, Seattle, WA, USA; [2]Inova Schar Cancer Institute, Fairfax, VA, USA; [3]Beth Israel Deaconess Medical Center, Harvard Medical School, Boston, MA, USA; [4]Gustave Roussy, INSERM U981, Université Paris-Saclay, Villejuif, France; [5]Princess Margaret Cancer Centre, University Health Network, Toronto, ON, Canada; [6]Chang Gung Memorial Hospital, Linkuo, Taiwan; [7]Samsung Medical Center, Sungkyunkwan University School of Medicine, Seoul, Korea; [8]Yamaguchi University Hospital, Ube, Yamaguchi, Japan; [9]the healthcare business of Merck KGaA, Darmstadt, Germany; [10]EMD Serono, Billerica, MA, USA; [11]Barts Cancer Institute, Experimental Cancer Medicine Centre, Queen Mary University of London, St Bartholomew's Hospital, London, UK

JAVELIN Bladder 100

CONCLUSIONS

- Long-term and exploratory analyses of patient-reported outcomes (PROs) in patients with advanced urothelial carcinoma (aUC) who received avelumab first-line (1L) switch maintenance + best supportive care (BSC) in the JAVELIN Bladder 100 trial showed that prolonged avelumab treatment, including in patients treated for ≥12 months, was associated with stable PROs, indicating preservation of health-related quality of life
 - These results complement previously reported results that compared PROs between study arms[1] and post hoc analyses showing the acceptable long-term safety profile of avelumab 1L maintenance, including in patients treated for ≥12 months[2]
 - These results are also consistent with a previous analysis showing that patients treated with avelumab 1L maintenance + BSC had a consistently longer quality-adjusted time without symptoms of disease or toxicity (Q-TWiST) than patients who received BSC alone, reflecting the safety profile of avelumab 1L maintenance in the context of an overall survival (OS) benefit[3]
 - Overall, these data suggest that patients receiving long-term avelumab treatment may have preserved health-related quality of life and control of cancer-related symptoms with manageable treatment-related toxicity
- PRO results from this trial further support the use of avelumab 1L maintenance until progression or unacceptable toxicity as standard of care in patients with aUC who are progression free after platinum-based chemotherapy

PLAIN LANGUAGE SUMMARY

- In the JAVELIN Bladder 100 study, avelumab switch maintenance treatment given after chemotherapy helped people with advanced urothelial cancer live longer
 - "Switch" maintenance treatment means giving a different treatment to people whose cancer disappeared, shrank, or stopped growing with chemotherapy
- In this new analysis, researchers looked at the long-term effects of avelumab treatment on people's quality of life
 - Quality of life is a measure of well-being. It includes how a person feels about their physical health, emotional well-being, ability to be active, and several other factors affecting everyday life
- Overall, avelumab treatment was found to maintain people's quality of life, and this was seen in both people treated with avelumab for any length of time and in people treated for at least 1 year
- Overall, these results support using avelumab switch maintenance (after chemotherapy) as a standard treatment for people with advanced urothelial cancer

BACKGROUND

- In the JAVELIN Bladder 100 phase 3 trial, avelumab 1L maintenance + BSC significantly prolonged OS and progression-free survival compared with BSC alone in patients with aUC that had not progressed with 1L platinum-based induction chemotherapy[4,5]
 - After ≥2 years of follow-up (data cutoff: June 4, 2021), median OS (from randomization) was 23.8 vs 15.0 months, respectively (hazard ratio [HR], 0.76 [95% CI, 0.63-0.91]; p=0.0036)[5]
 - In a post hoc exploratory analysis, median OS with avelumab 1L maintenance, measured from start of 1L platinum-based chemotherapy in this selected population without cancer progression was 29.7 months[6]
 - The long-term safety of avelumab 1L maintenance was also demonstrated[2,5]
- Based on results from JAVELIN Bladder 100, avelumab 1L maintenance has been approved in multiple countries worldwide[7,8] and is recommended as a standard of care in international guidelines[9-11]
- In a post hoc analysis, treatment with avelumab 1L maintenance + BSC resulted in a consistently longer Q-TWiST than BSC alone, indicating a net benefit in quality survival or well-being with avelumab[3]
- Initial analyses of PROs in JAVELIN Bladder 100 showed that avelumab 1L maintenance treatment resulted in stable health-related quality of life[1]
- Here, we report PRO data with long-term follow-up in the overall avelumab + BSC arm (any treatment duration) and in a subgroup with ≥12 months of avelumab treatment

METHODS

- JAVELIN Bladder 100 (NCT02603432) enrolled 700 patients with unresectable locally advanced or metastatic UC that had not progressed with 1L platinum-based chemotherapy
- Patients were randomized 1:1 to receive avelumab 1L maintenance + BSC or BSC alone
- The primary endpoint was OS
- PROs were a secondary endpoint and were assessed at baseline, on day 1 of each 4-week cycle, at end of treatment or withdrawal from the study, and up to 90 days post treatment
- PRO instruments used were the NCCN/FACT Bladder Symptom Index-18 (NFBlSI-18) and EuroQol EQ-5D-5L
 - NFBlSI-18 measures symptoms and quality of life in the past 7 days; score ranges are: NFBlSI-18 total score, 0-72; disease-related symptoms – physical (DRS-P), 0-36; disease-related symptoms – emotional (DRS-E), 0-8; treatment side effects (TSE), 0-20; and function/well-being (FWB), 0-8
 - EQ-5D-5L measures general health status based on mobility, self-care, usual activities, pain/discomfort, and anxiety/depression; EQ-5D-5L index score is calculated based on UK weights, and scores range from –0.594 to 1; EQ-5D-5L visual analog scale scores range from 0 to 100
- Descriptive and mixed-effect model analyses were conducted
- PROs were examined in all patients who received treatment in the avelumab + BSC arm for any duration and in a subgroup who had received ≥12 months of avelumab treatment
 - In this analysis, data were not evaluated in the BSC alone arm because few patients remained on study treatment at later time points, and comparative assessment was deemed not appropriate due to this attrition bias

RESULTS

- At data cutoff (June 4, 2021), median follow-up in the avelumab + BSC arm (n=350) was 38.0 months (≥2 years in all patients), and median duration of treatment was 5.8 months
- In patients treated for ≥12 months (n=118 [33.7%]), baseline characteristics were similar to those of patients in the overall avelumab + BSC arm, except for a higher proportion with ECOG performance status 0 (70.3% vs 60.9%) and PD-L1+ tumors (61.0% vs 54.0%) and a lower proportion with visceral metastases (47.5% vs 54.6%) (**Table 1**)
- In both populations, completion rates for both PRO instruments among evaluable patients were >80% at all time points during treatment (**Table 2**)

Table 1. Patient baseline characteristics

	Overall avelumab + BSC arm (n=350)	Patients treated with avelumab for ≥12 months (n=118)
Age, median (range), years	68 (37-90)	69 (43-86)
Sex, n (%)		
Male	266 (76.0)	91 (77.1)
Female	84 (24.0)	27 (22.9)
Pooled geographic region, n (%)		
Europe	214 (61.1)	61 (51.7)
North America	12 (3.4)	6 (5.1)
Asia	73 (20.9)	32 (27.1)
Australasia	34 (9.7)	15 (12.7)
Rest of the world	17 (4.9)	4 (3.4)
ECOG performance status, n (%)		
0	213 (60.9)	83 (70.3)
1	136 (38.9)	35 (29.7)
2	1 (0.3)	0
PD-L1 status, n (%)		
Positive	189 (54.0)	72 (61.0)
Negative	139 (39.7)	39 (33.1)
Unknown	22 (6.3)	7 (5.9)
1L chemotherapy regimen, n (%)		
Gemcitabine + cisplatin	183 (52.3)	67 (56.8)
Gemcitabine + carboplatin	147 (42.0)	43 (36.4)
Gemcitabine + cisplatin or carboplatin*	20 (5.7)	8 (6.8)
Best response to 1L chemotherapy, n (%)		
CR	90 (25.7)	36 (30.5)
PR	163 (46.6)	51 (43.2)
SD	97 (27.7)	31 (26.3)
Site of metastasis at start of 1L chemotherapy, n (%)		
Visceral	191 (54.6)	56 (47.5)
Nonvisceral	159 (45.4)	62 (52.5)
Site of primary tumor, n (%)		
Upper tract	106 (30.3)	34 (28.8)
Lower tract	244 (69.7)	84 (71.2)

1L, first line; **BSC**, best supportive care; **CR**, complete response; **PR**, partial response; **SD**, stable disease.
*Patients who switched platinum regimens while receiving 1L chemotherapy.

Table 2. Completion rates for the NFBlSI-18 and EQ-5D-5L instruments

	NFBlSI-18, n completed/N eligible (%)		EQ-5D-5L, n completed/N eligible (%)	
	Overall avelumab + BSC arm (n=350)	Patients treated with avelumab for ≥12 months (n=118)	Overall avelumab + BSC arm (n=350)	Patients treated with avelumab for ≥12 months (n=118)
Baseline	333/342 (97)	114/117 (97)	339/346 (98)	117/118 (99)
Cycle 2/day 1	317/334 (95)	116/118 (98)	316/334 (95)	116/118 (98)
Cycle 5/day 1	211/227 (93)	111/118 (94)	214/227 (94)	111/118 (94)
Cycle 10/day 1	135/141 (96)	114/118 (97)	136/141 (96)	115/118 (97)
Cycle 15/day 1	108/114 (95)	108/114 (95)	108/114 (95)	108/114 (95)
Cycle 20/day 1	74/83 (89)	74/83 (89)	76/83 (92)	76/83 (92)
Cycle 25/day 1	69/72 (96)	69/72 (96)	69/72 (96)	69/72 (96)
Cycle 30/day 1	51/58 (88)	51/58 (88)	51/58 (88)	51/58 (88)
Cycle 35/day 1	33/36 (92)	33/36 (92)	33/36 (92)	33/36 (92)
Cycle 40/day 1	25/25 (100)	25/25 (100)	25/25 (100)	25/25 (100)
Cycle 45/day 1	16/18 (89)	16/18 (89)	16/18 (89)	16/18 (89)
End of treatment	227/291 (78)	54/70 (77)	227/291 (78)	54/70 (77)
Follow-up day 30	81/128 (63)	26/40 (65)	80/128 (63)	25/40 (63)
Follow-up day 60	70/99 (71)	23/30 (77)	70/99 (71)	23/30 (77)
Follow-up day 90	52/76 (68)	19/25 (76)	52/76 (68)	19/25 (76)

Rows showing individual cycles are truncated after the cycle when the total number of eligible patients was <20.
BSC, best supportive care; **NFBISI-18,** NCCN/FACT Bladder Symptom Index-18.

- On average, PRO scores remained stable throughout treatment, and no clinically important changes from baseline were reported (**Table 3**; **Figures 1** and **2**)
 - Mixed-model analyses, including data after treatment discontinuation, had similar results (data not shown)
- In the overall avelumab + BSC arm and in patients treated with avelumab for ≥12 months, approximately 75% of evaluable patients reported no change or a decrease in how much they were bothered by treatment side effects throughout 24 months of treatment (**Figure 3**)

Table 3. Mixed-effect model analysis of NFBISI-18 and EQ-5D-5L scores over all cycles prior to end of treatment

	Least-squares mean change from baseline (95% CI)	
	Overall avelumab + BSC arm (n=350)	Patients treated with avelumab for ≥12 months (n=118)
NFBISI-18 total score	−2.15 (−3.25, −1.04)	1.28 (0.08, 2.49)
DRS-P	−1.39 (−1.88, −0.89)	−0.05 (−0.63, 0.52)
DRS-E	0.27 (0.11, 0.44)	0.68 (0.46, 0.91)
Treatment side effects	−0.42 (−0.73, −0.11)	0.22 (−0.16, 0.59)
Function/well-being	−0.15 (−0.34, 0.04)	0.36 (0.09, 0.62)
EQ-5D-5L index score	−0.07 (−0.09, −0.05)	−0.02 (−0.04, 0.00)
Visual analog scale	−1.14 (−2.95, 0.67)	4.16 (2.09, 6.23)

Estimates of clinically important differences for NFBlSI-18 are: total score, 3-6; DRS-P, 2-3; treatment side effects, 1-2; and DRS-E and function/well-being, 1 each. Estimates of minimally important differences for EQ-5D-5L are: index score, 0.09-0.12 and visual analog scale, 7-12.
BSC, best supportive care; **DRS-E,** disease-related symptoms – emotional; **DRS-P,** disease-related symptoms – physical; **NFBISI-18,** NCCN/FACT Bladder Symptom Index-18.

Figure 1. NFBlSI-18 total scores over time

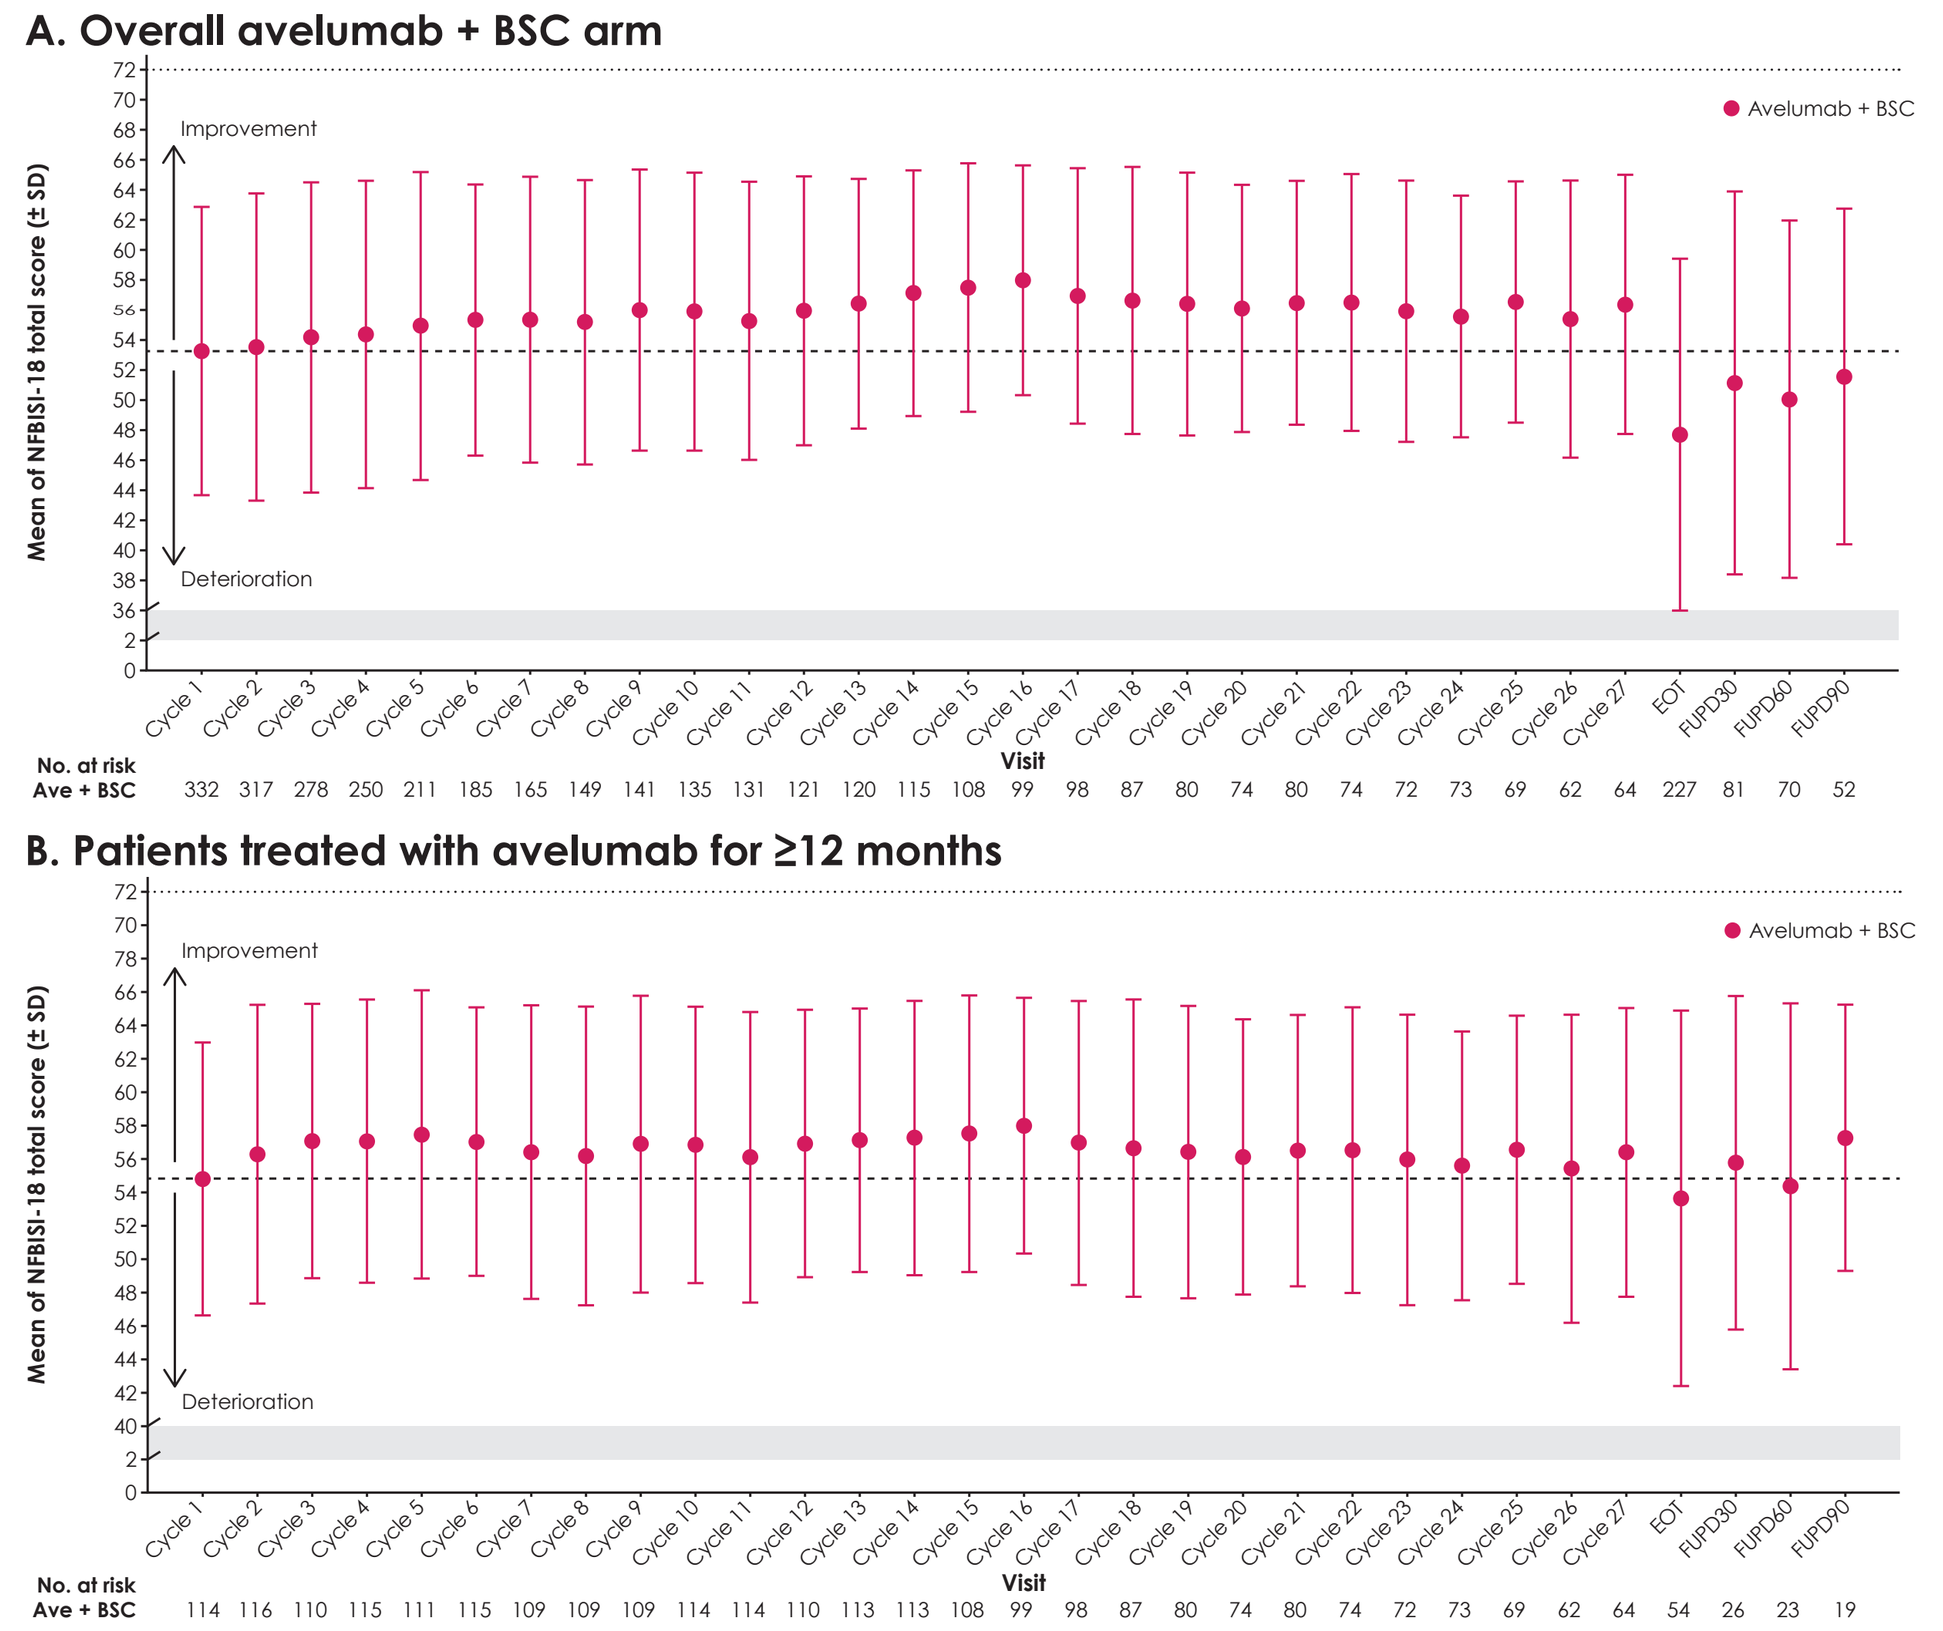
A. Overall avelumab + BSC arm
B. Patients treated with avelumab for ≥12 months
Mean of NFBISI-18 total score (± SD)
Improvement
Deterioration
Avelumab + BSC
Visit
No. at risk Ave + BSC
332 317 278 250 211 185 165 149 141 135 131 121 120 115 108 99 98 87 80 74 80 74 72 73 69 62 64 227 81 70 52
114 116 110 115 111 115 109 109 109 114 114 110 113 113 108 99 98 87 80 74 80 74 72 73 69 62 64 54 26 23 19

Estimate of clinically important difference in the NFBlSI-18 total score is 3-6. **BSC**, best supportive care; **EOT**, end of treatment; **FUPD30**, follow-up day 30; **FUPD60**, follow-up day 60; **FUPD90**, follow-up day 90; **NFBISI-18**, NCCN/FACT Bladder Symptom Index-18.

Figure 2. EQ-5D-5L index scores over time

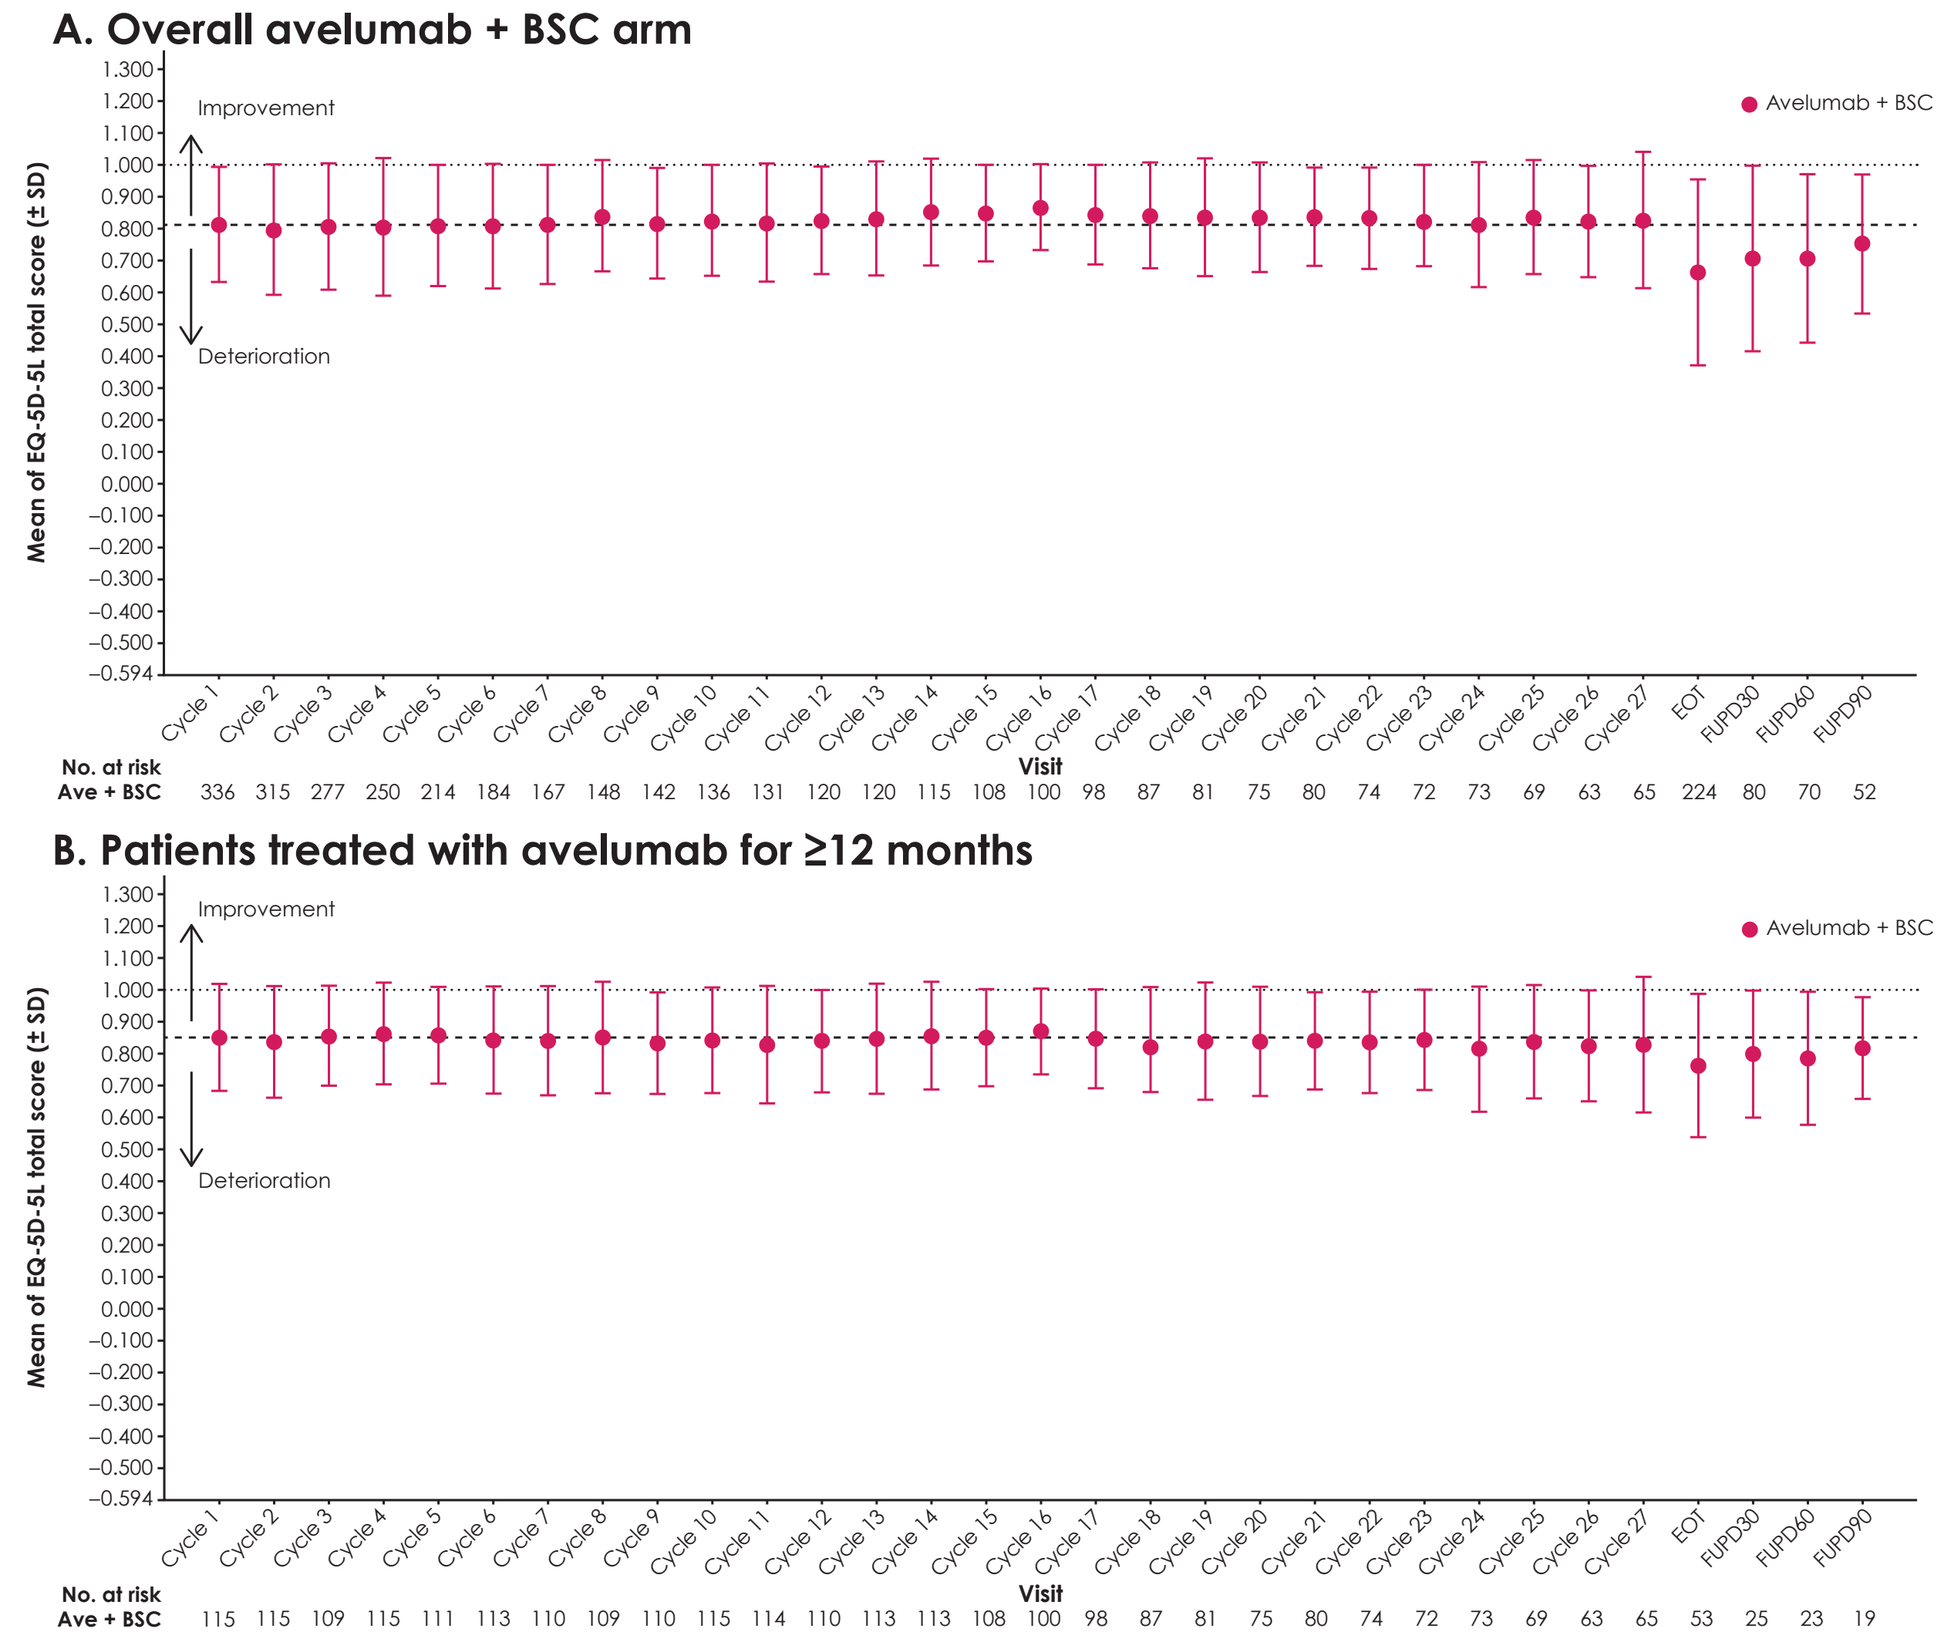
A. Overall avelumab + BSC arm
B. Patients treated with avelumab for ≥12 months
Mean of EQ-5D-5L total score (± SD)
Improvement
Deterioration
Avelumab + BSC
Visit
No. at risk Ave + BSC
336 315 277 250 214 184 167 148 142 136 131 120 120 115 108 100 98 87 81 75 80 74 72 73 69 63 65 224 80 70 52
115 115 109 115 111 113 110 109 110 115 114 110 113 113 108 100 98 87 81 75 80 74 72 73 69 63 65 53 25 23 19

Estimate of minimally important difference in EQ-5D-5L index score is 0.09-0.12. **BSC**, best supportive care; **EOT**, end of treatment; **FUPD30**, follow-up day 30; **FUPD60**, follow-up day 60; **FUPD90**, follow-up day 90.

Figure 3. Distribution of patients over time who reported being bothered by treatment side effects in NFBlSI-18 instrument

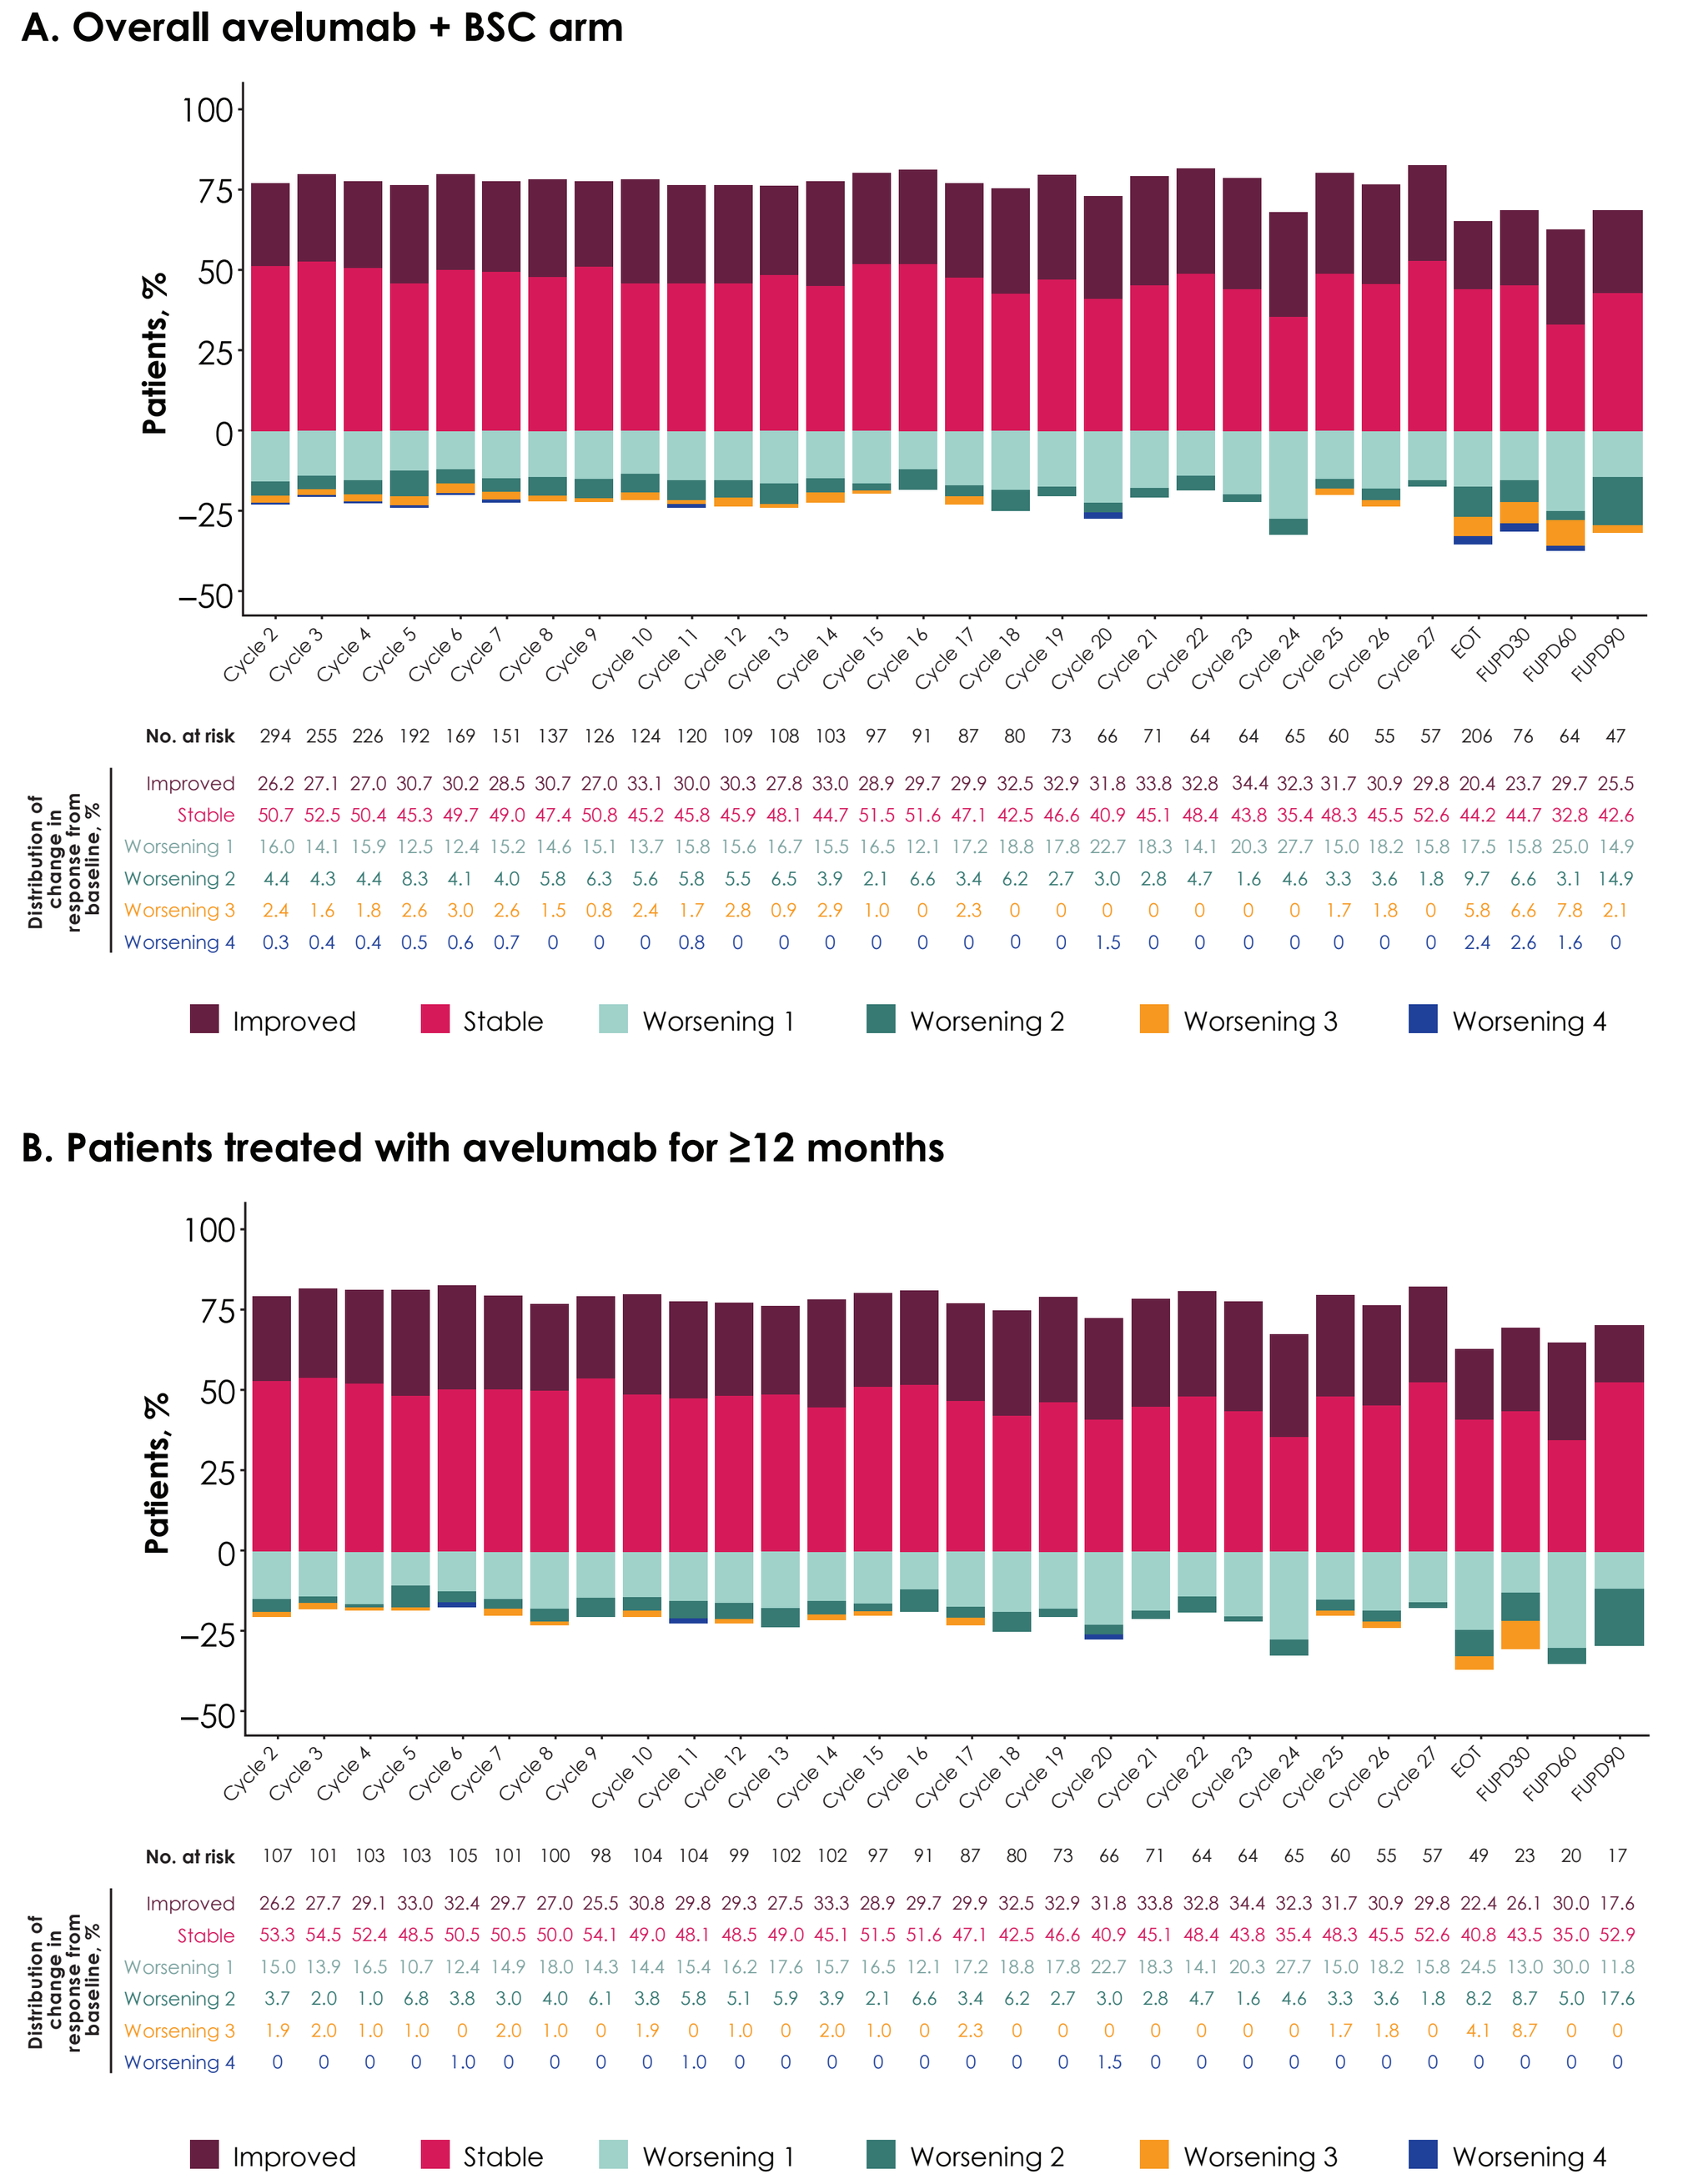
A. Overall avelumab + BSC arm
Patients, %
No. at risk 294 255 226 192 169 151 137 126 124 120 109 108 103 97 91 87 80 73 66 71 64 64 65 60 55 57 206 76 64 47
Distribution of change in response from baseline, %
Improved 26.2 27.1 27.0 30.7 30.2 28.5 30.7 27.0 33.1 30.0 30.3 27.8 33.0 28.9 29.7 29.9 32.5 32.9 31.8 33.8 32.8 34.4 32.3 31.7 30.9 29.8 20.4 23.7 29.7 25.5
Stable 50.7 52.5 50.4 45.3 49.7 49.0 47.4 50.8 45.2 45.8 45.9 48.1 44.7 51.5 51.6 47.1 42.5 46.6 40.9 45.1 48.4 43.8 35.4 48.3 45.5 52.6 44.2 44.7 32.8 42.6
Worsening 1 16.0 14.1 15.9 12.5 12.4 15.2 14.6 15.1 13.7 15.8 15.6 16.7 15.5 16.5 12.1 17.2 18.8 17.8 22.7 18.3 14.1 20.3 27.7 15.0 18.2 15.8 17.5 15.8 25.0 14.9
Worsening 2 4.4 4.3 4.4 8.3 4.1 4.0 5.8 6.3 5.6 5.8 5.5 6.5 3.9 2.1 6.6 3.4 6.2 2.7 3.0 2.8 4.7 1.6 4.6 3.3 3.6 1.8 9.7 6.6 3.1 14.9
Worsening 3 2.4 1.6 1.8 2.6 3.0 2.6 1.5 0.8 2.4 1.7 2.8 0.9 2.9 1.0 0 2.3 0 0 0 0 0 0 0 1.7 1.8 0 5.8 6.6 7.8 2.1
Worsening 4 0.3 0.4 0.4 0.5 0.6 0.7 0 0 0 0.8 0 0 0 0 0 0 0 0 1.5 0 0 0 0 0 0 0 2.4 2.6 1.6 0
Improved Stable Worsening 1 Worsening 2 Worsening 3 Worsening 4
B. Patients treated with avelumab for ≥12 months
Patients, %
No. at risk 107 101 103 103 105 101 100 98 104 104 99 102 102 97 91 87 80 73 66 71 64 64 65 60 55 57 49 23 20 17
Improved 26.2 27.7 29.1 33.0 32.4 29.7 27.0 25.5 30.8 29.8 29.3 27.5 33.3 28.9 29.7 29.9 32.5 32.9 31.8 33.8 32.8 34.4 32.3 31.7 30.9 29.8 22.4 26.1 30.0 17.6
Stable 53.3 54.5 52.4 48.5 50.5 50.5 50.0 54.1 49.0 48.1 48.5 49.0 45.1 51.5 51.6 47.1 42.5 46.6 40.9 45.1 48.4 43.8 35.4 48.3 45.5 52.6 40.8 43.5 35.0 52.9
Worsening 1 15.0 13.9 16.5 10.7 12.4 14.9 18.0 14.3 14.4 15.4 16.2 17.6 15.7 16.5 12.1 17.2 18.8 17.8 22.7 18.3 14.1 20.3 27.7 15.0 18.2 15.8 24.5 13.0 30.0 11.8
Worsening 2 3.7 2.0 1.0 6.8 3.8 3.0 4.0 6.1 3.8 5.8 5.1 5.9 3.9 2.1 6.6 3.4 6.2 2.7 3.0 2.8 4.7 1.6 4.6 3.3 3.6 1.8 8.2 8.7 5.0 17.6
Worsening 3 1.9 2.0 1.0 1.0 0 2.0 1.0 0 1.9 0 1.0 0 2.0 1.0 0 2.3 0 0 0 0 0 0 0 1.7 1.8 0 4.1 8.7 0 0
Worsening 4 0 0 0 0 1.0 0 0 0 0 1.0 0 0 0 0 0 0 0 0 1.5 0 0 0 0 0 0 0 0 0 0 0
Improved Stable Worsening 1 Worsening 2 Worsening 3 Worsening 4

BSC, best supportive care; **EOT**, end of treatment; **FUPD30**, follow-up day 30; **FUPD60**, follow-up day 60; **FUPD90**, follow-up day 90; **NFBISI-18**, NCCN/FACT Bladder Symptom Index-18.

GET POSTER PDF AND VIEW VIDEO PRESENTATION

Copies of this poster and any associated video or audio files obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Petros Grivas**, pgrivas@uw.edu

REFERENCES 1. Grivas P, et al. Eur Urol 2023;83:320-8. **2.** Bellmunt J, et al. J Clin Oncol 2023;41(Suppl 16):Abstract 4516. **3.** Powles T, et al. J Clin Oncol 2023;41(Suppl 16):Abstract 4515. **4.** Powles T, et al. N Engl J Med 2020;383:1218-30. **5.** Powles T, et al. J Clin Oncol 2023;41:3486-92. **6.** Sridhar SS, et al. J Clin Oncol 2023;41(Suppl 6):Abstract 508. **7.** Bavencio (avelumab). Prescribing information. EMD Serono, Rockland, MA, USA; 2023. **8.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA; 2023. **9.** NCCN Clinical Practice Guidelines in Oncology: Bladder Cancer. V3.2023. **10.** Powles T, et al. Ann Oncol 2022;33:244-58. **11.** Cathomas R, et al. Eur Urol 2022;8:95-103.
DISCLOSURES P. Grivas has served in consulting or advisory roles for 4D Pharma, Aadi Bioscience, Astellas Pharma, Asieris Pharmaceuticals, AstraZeneca, BostonGene, Bristol Myers Squibb, CG Oncology, Dyania Health, Fresenius Kabi, G1 Therapeutics, Genentech/Roche, Gilead Sciences, Guardant Health, ImmunityBio, Janssen, Lucence, Merck & Co., Kenilworth, NJ, Pfizer, PureTech Health, Seagen, Silverback Therapeutics, Strata Oncology, the healthcare business of Merck KGaA, Darmstadt, Germany, and UroGen Pharma; and has received institutional research funding from Acrivon Therapeutics, ALX Oncology, Bavarian Nordic, Bristol Myers Squibb, Clovis Oncology, Debiopharm Group, G1 Therapeutics, Gilead Sciences, GSK, Merck & Co., Kenilworth, NJ, Mirati Therapeutics, Pfizer, QED Therapeutics, and the healthcare business of Merck KGaA, Darmstadt, Germany. **J. B. Aragon-Ching** has served in consulting or advisory roles for Algeta/Bayer, Amgen, AstraZeneca, AstraZeneca/MedImmune, AVEO, Bayer, Dendreon, Exelixis, Immunomedics, Janssen Biotech, Merck & Co., Kenilworth, NJ, Pfizer, Pfizer/Myovant, Sanofi, Seagen, and the healthcare business of Merck KGaA, Darmstadt, Germany; has reported speakers services for Astellas Pharma, Bristol Myers Squibb, Janssen-Ortho, and Seagen/Astellas; has received travel and accommodation expenses from Algeta/Bayer, Astellas Pharma, Bristol Myers Squibb, Dendreon, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received honoraria from Astellas Scientific and Medical Affairs Inc, Bristol Myers Squibb, and the healthcare business of Merck KGaA, Darmstadt, Germany. **J. Bellmunt** has served in consulting or advisory roles for Astellas Pharma, AstraZeneca/MedImmune, Bristol Myers Squibb, Genentech, Novartis, Pfizer, Pierre Fabre, and the healthcare business of Merck KGaA, Darmstadt, Germany; has received travel and accommodation expenses from Ipsen, Merck & Co., Kenilworth, NJ, and Pfizer; reports patents, royalties, and other intellectual property from UpToDate; reports stock and other ownership interests in Rainier Therapeutics; has received honoraria from UpToDate; and has received institutional research funding from Millennium, Pfizer, Sanofi, and the healthcare business of Merck KGaA, Darmstadt, Germany. **Y. Loriot** has served in consulting or advisory role for Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Immunomedics, Janssen, Janssen (Inst), Loxo/Lilly, Merck & Co., Kenilworth, NJ, Merck & Co., Kenilworth, NJ (Inst), Pfizer/the healthcare business of Merck KGaA, Darmstadt, Germany, Roche, Seagen, and Taiho Pharmaceutical; has received institutional research funding from Astellas Pharma, AstraZeneca, Basilea, Bristol Myers Squibb, Exelixis, Gilead Sciences, Incyte, Janssen Oncology, Merck & Co., Kenilworth, NJ, Nektar, Pfizer, Roche, Sanofi, Seagen, Taiho Pharmaceutical, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel, accommodations, and expenses from Astellas Pharma, AstraZeneca, Janssen Oncology, Merck & Co., Kenilworth, NJ, Roche, and Seagen. **S. S. Sridhar** has served in consulting or advisory roles for Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Immunomedics, Janssen, Merck & Co., Kenilworth, NJ, Pfizer, Roche/Genentech, Seagen, and Taiho Pharmaceutical; has received institutional research funding from Astellas Pharma, AstraZeneca, Basilea, Bristol Myer Squibb, Exelixis, Gilead Sciences, Incyte, Janssen Oncology, Merck & Co., Kenilworth, NJ, Nektar, Pfizer, Roche, Sanofi, Seagen, Taiho Pharmaceutical, the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel and accommodation expenses from Astellas Pharma, AstraZeneca, Janssen Oncology, Merck & Co., Kenilworth, NJ, Roche, and Seagen. **P-J Su** has nothing to disclose. **S. H. Park** has received honoraria from the healthcare business of Merck KGaA, Darmstadt, Germany, Pfizer, Ono Pharmaceutical; has served in consulting or advisory roles for Janssen Oncology; and has received research funding from Merck & Co., Kenilworth, NJ (Inst). **Y. Yamamoto** has received honoraria from Astellas, AstraZeneca, Bristol Myers Squibb, Janssen, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Sanofi, Takeda, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received institutional research funding from Pfizer and Takeda. **N. Jacob** reports employment by the healthcare business of Merck KGaA, Darmstadt, Germany. **J. Hoffman** reports employment by EMD Serono and stock in Merck & Co., Kenilworth, NJ, Pfizer, and the healthcare business of Merck KGaA, Darmstadt, Germany. **M. Kearney** and **M. Schlichting** report employment by the healthcare business of Merck KGaA, Darmstadt, Germany. **T. Powles** has served in consulting or advisory roles for Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Eisai, Exelixis, Incyte, Ipsen, Johnson & Johnson, Merck & Co., Kenilworth, NJ, Novartis, Pfizer, Roche, Seagen, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received travel and accommodations expenses from Pfizer. **ACKNOWLEDGMENTS** This study was sponsored by Pfizer and was previously conducted under an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and Pfizer. This analysis was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany. Medical writing support was provided by Hiba Al-Ashtal of Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 581. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.