A UK real-world observational study of avelumab + axitinib in advanced renal cell carcinoma (aRCC): outcomes at 36 months post treatment initiation

P. Nathan,[1] N. Charnley,[2] R. Frazer,[3] J. McGrane,[4] I. Muazzam,[5] M. Pillai,[6] S. Rudman,[7] A. Sharma,[1] R. Stevenson,[8] B. Venugopal,[9] J. Hickey,[10] A.R. Ritchie,[11] K. Liu[12]*

[1]Mount Vernon Cancer Centre, Northwood, UK; [2]Lancashire Teaching Hospitals, Preston, UK; [3]Velindre Cancer Centre, Cardiff, UK; [4]Royal Cornwall Hospital NHS Trust, Cornwall, UK; [5]Hull University Teaching Hospitals NHS Trust, Castle Hill Hospital, Cottingham, UK; [6]The Christie NHS Foundation Trust, Manchester, UK; [7]Guy's and St Thomas Hospital NHS Foundation Trust, London, UK; [8]Queen Elizabeth Hospital, Birmingham, UK; [9]The Beatson West of Scotland Cancer Centre, Glasgow, UK; [10]OPEN Health, London, UK; [11]Pfizer Ltd, Surrey, UK; [12]Merck Serono Ltd, Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany.

*Affiliation at the time the study was conducted.

CONCLUSIONS

- **This analysis assessed overall survival (OS), progression-free survival (PFS), objective response rate (ORR), and best response at 36 months in 130 patients with aRCC who had initiated first-line treatment with avelumab + axitinib (A + Ax) in routine clinical practice in the UK**
- **The OS rate at 36 months post initiation was 53%, in line with previously published figures from both real-world and clinical trial data[1-4]**
- **ORR at 36 months post initiation was 62%, broadly similar to published real-world and clinical trial data in UK patients with aRCC[1, 2]**
- **Overall, higher complete response (CR) and partial response (PR) rates were seen in good and intermediate prognostic groups**
- **OS, PFS, ORR, and best response results observed at 36 months were in line with findings from JAVELIN Renal 101 and other observational studies, with no newly emerging adverse events (AEs)**

PLAIN LANGUAGE SUMMARY

- **Based on clinical trial results, avelumab plus axitinib given in combination is an approved treatment for people with advanced renal cell cancer**
- **In this study, researchers looked at people in the UK with advanced renal cell cancer who received avelumab plus axitinib treatment in normal clinical practice, outside of a clinical trial**
- **53% of people were still alive 3 years after starting treatment**
- **On average, people lived for 13.5 months without their cancer getting worse**
- **62% of people had a treatment response, meaning that their cancer became undetectable or shrank during treatment**
- **Treatment responses occurred in people with different numbers of characteristics that are used to predict how long people might live with treatment (called risk factors)**
- **Overall, these results provide more evidence for using avelumab plus axitinib in people with advanced renal cell cancer**

BACKGROUND

- In patients with previously untreated aRCC, A + Ax combination therapy has shown superior PFS and ORR vs sunitinib across all International Metastatic RCC Database Consortium (IMDC) risk groups[1-4]
- A + Ax combination therapy for the first-line treatment of adult patients with aRCC was approved by the European Commission on October 29, 2019.[5] A + Ax combination therapy is approved and funded for the treatment of aRCC in the UK. The Scottish Medicines Consortium has funded A + Ax since October 12, 2020, and the National Institute for Health and Care Excellence via the Cancer Drugs Fund has funded it since July 30, 2020[6]
- A + Ax combination therapy was available to patients in the UK in August 2019 via an early access to medicines scheme (EAMS no. 11648/0002) prior to receiving marketing authorization[7]

METHODS

- This was a UK-based multicenter, noninterventional cohort study of adult patients with aRCC who initiated A + Ax on or after August 1, 2019
- Ethical review from a National Health Service research ethics committee was not required for this study as it was a retrospective observational study in which patient-level data were collected by members of the direct care team.[8] Informed consent was not required
- Inclusion criteria for the study were a diagnosis of aRCC, age ≥18 years, and initiation of A + Ax on or after August 1, 2019
- The primary endpoints were OS, PFS, ORR, and best response at 36 months post A + Ax initiation. Secondary endpoints included baseline demographics, clinical characteristics, history of RCC before A + Ax initiation, patterns of A + Ax use, and treatment-related safety events
- Data were analyzed descriptively

RESULTS

- A total of 130 patients from 9 UK sites with 36-month follow-up data were included in this analysis; baseline demographics and clinical characteristics are summarized in **Table 1**

Table 1. Baseline demographics and clinical characteristics

	N=130
Age, median (range), years	67.1 (35.8-87.0)
Sex, n	
Male	96
Female	34
BMI at treatment initiation, mean (SD), kg/m² (n=117)	28.2 (5.5)
Ethnicity, n (%)	
Asian/Asian British	5 (4)
White	90 (69)
Not recorded	35 (27)
Smoking status at treatment initiation, n (%)	
Current smoker	13 (10)
Former smoker	44 (34)
Never smoked	43 (33)
Not recorded	30 (23)
ECOG PS at treatment initiation, n (%)	
0	61 (47)
1	61 (47)
2	6 (5)
Not recorded	2 (2)
IMDC status at treatment initiation, n (%)	
Favorable	51 (39)
Intermediate	52 (40)
Poor	25 (19)
Not known	2 (2)
Histological subtype, n (%)	
RCC with clear cell component	115 (88)
Unclassified RCC	7 (5)
Xp11 translocation RCC	1 (1)
Collecting duct RCC (Bellini collecting duct RCC)	1 (1)
Papillary RCC	3 (2)
Chromophobe RCC	2 (2)
Not recorded	1 (1)
No. of metastatic sites at treatment initiation, median (range)	2.0 (1.0-5.0)
Metastatic sites at treatment initiation, n (%)	
Bone	28 (22)
Brain	5 (4)
Lung	87 (67)
Lymph node	43 (33)
Liver	18 (14)
Other	49 (38)
History of nephrectomy	
Yes, full	72 (55)
Yes, partial	16 (12)
No	41 (32)
Not recorded	1 (1)
Time between aRCC diagnosis and treatment initiation, median (range), months	2.5 (0.03-115.4)

BMI, body mass index; **ECOG PS,** Eastern Cooperative Oncology Group performance status; **IMDC,** International Metastatic RCC Database Consortium; **RCC,** renal cell carcinoma.

Figure 1. Kaplan-Meier plot of OS at 36 months post treatment initiation

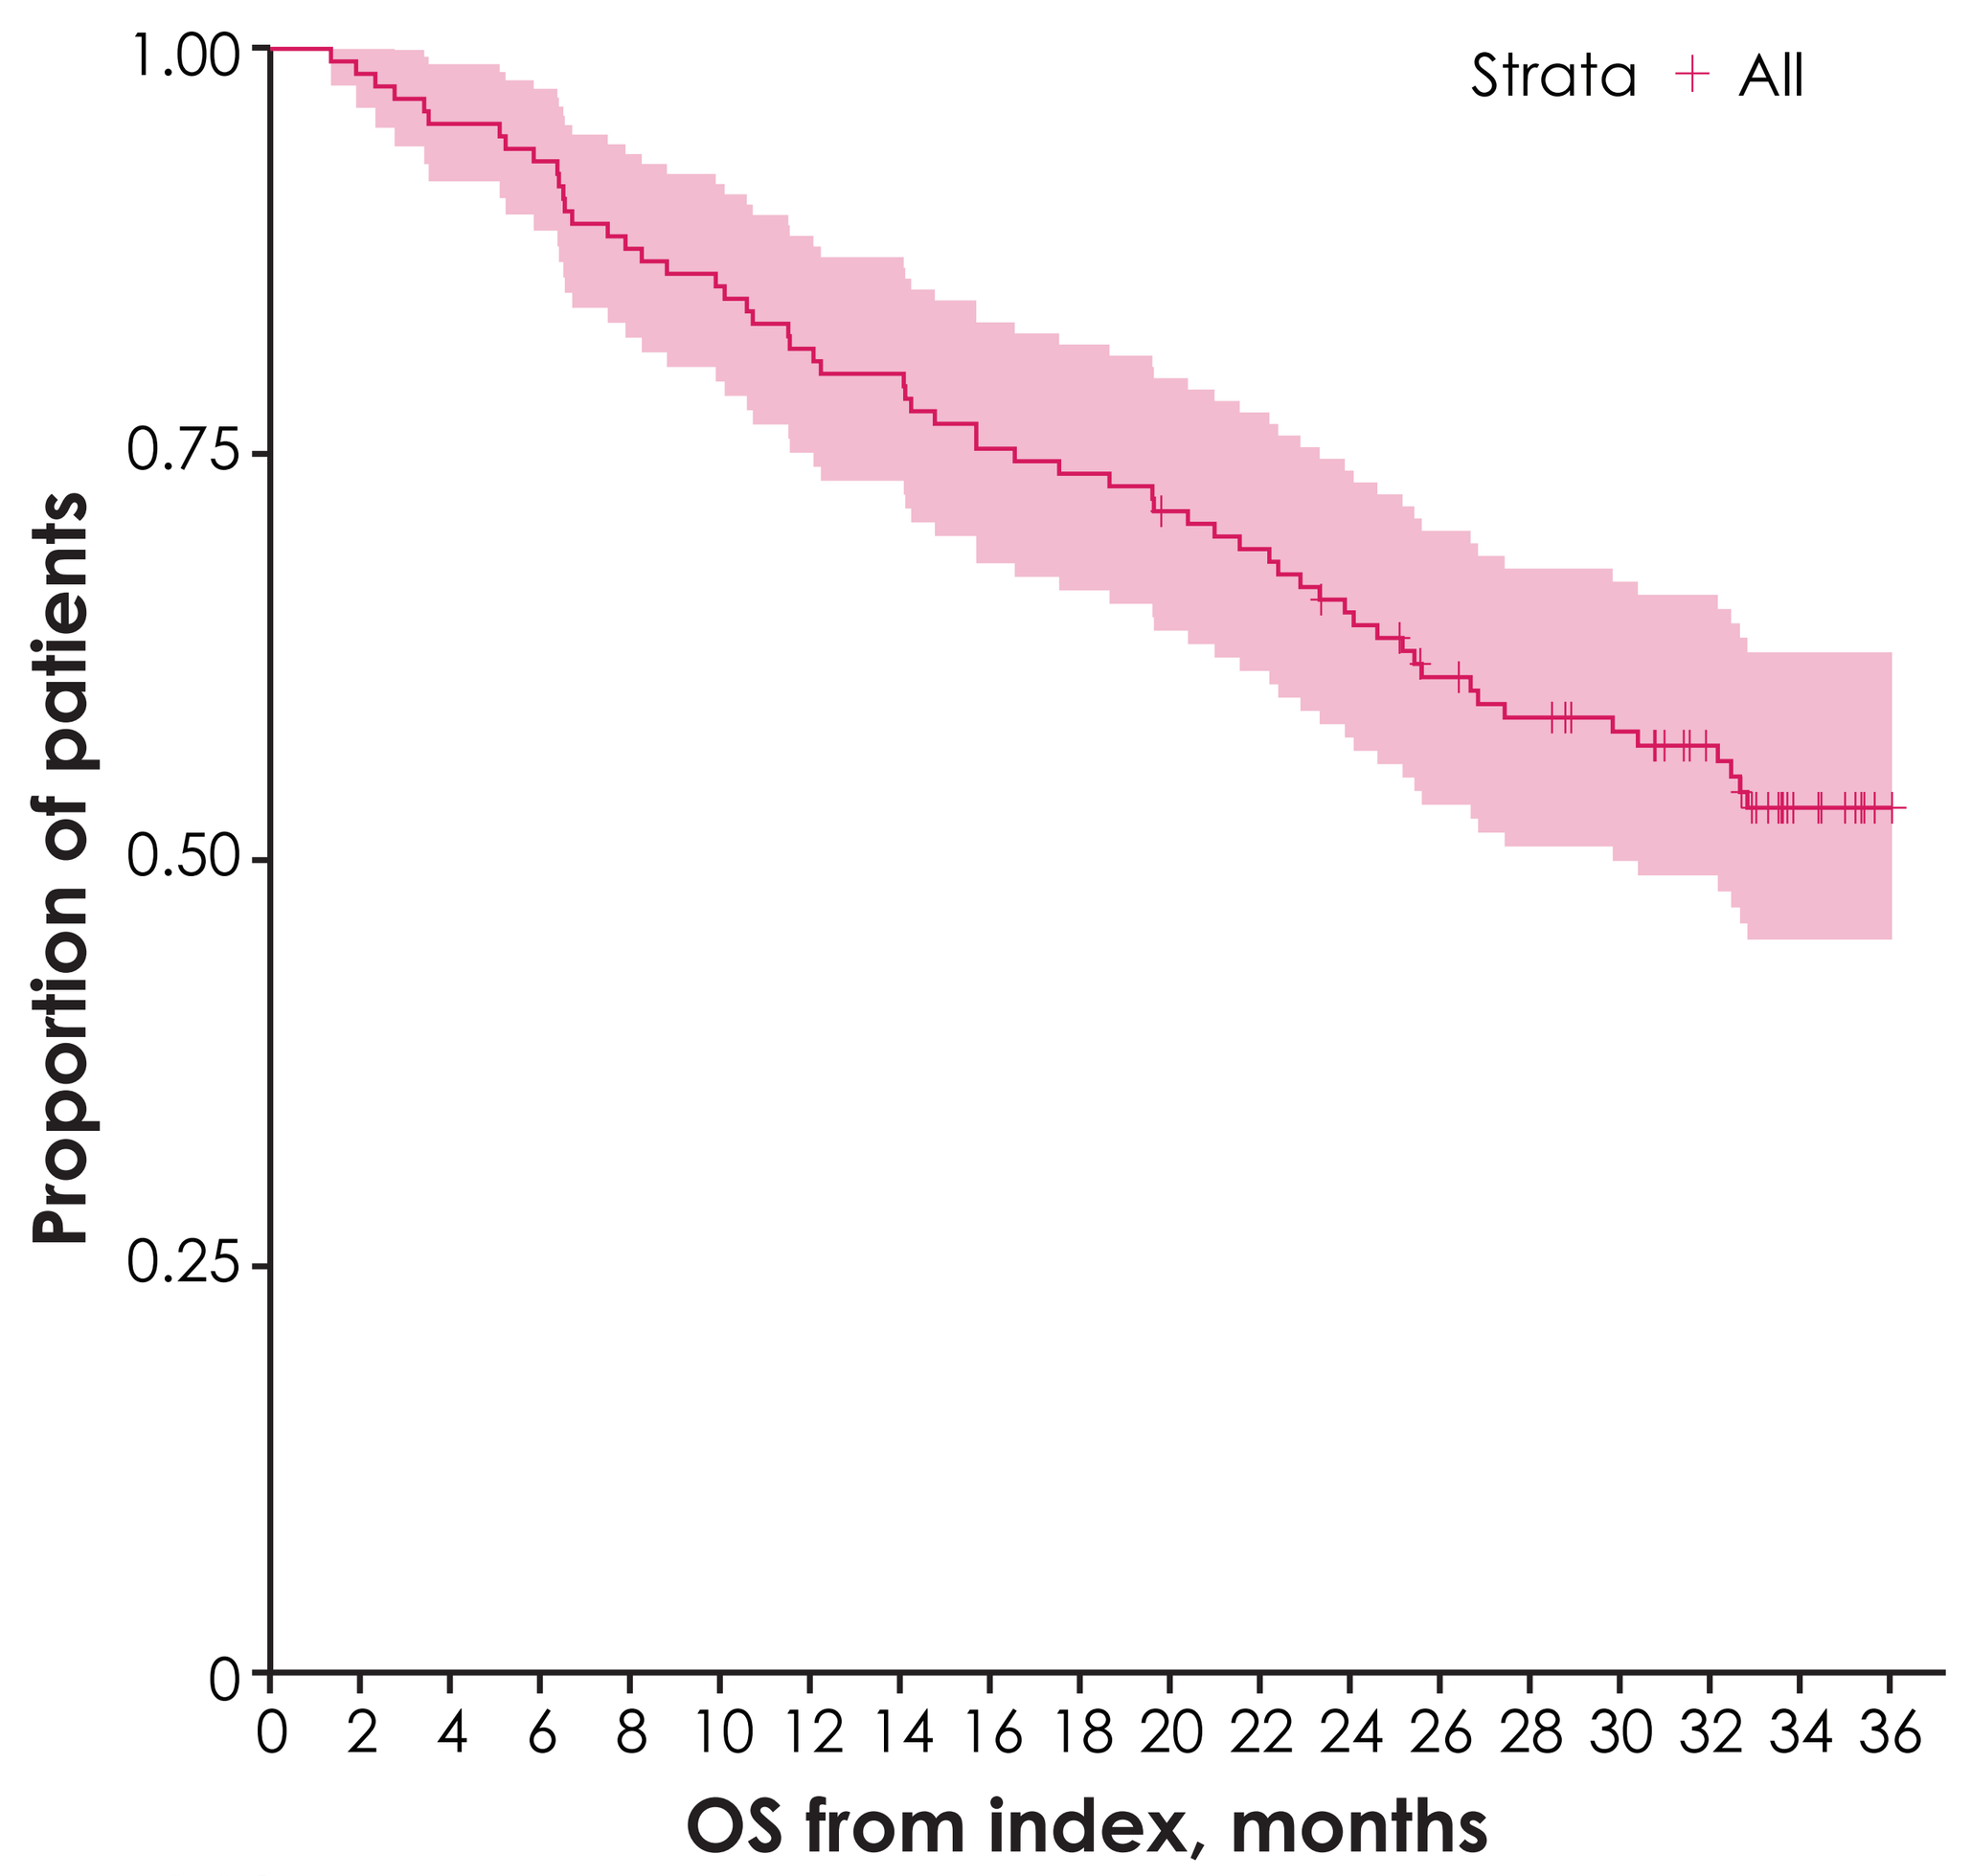

Strata + All
1.00
0.75
0.50
0.25
0
Proportion of patients
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36
OS from index, months

No. at risk

All 130 128 124 121 114 111 106 104 98 96 92 89 83 75 71 67 60 47 40

OS, overall survival

Figure 2. Kaplan-Meier plot of PFS at 36 months post treatment initiation

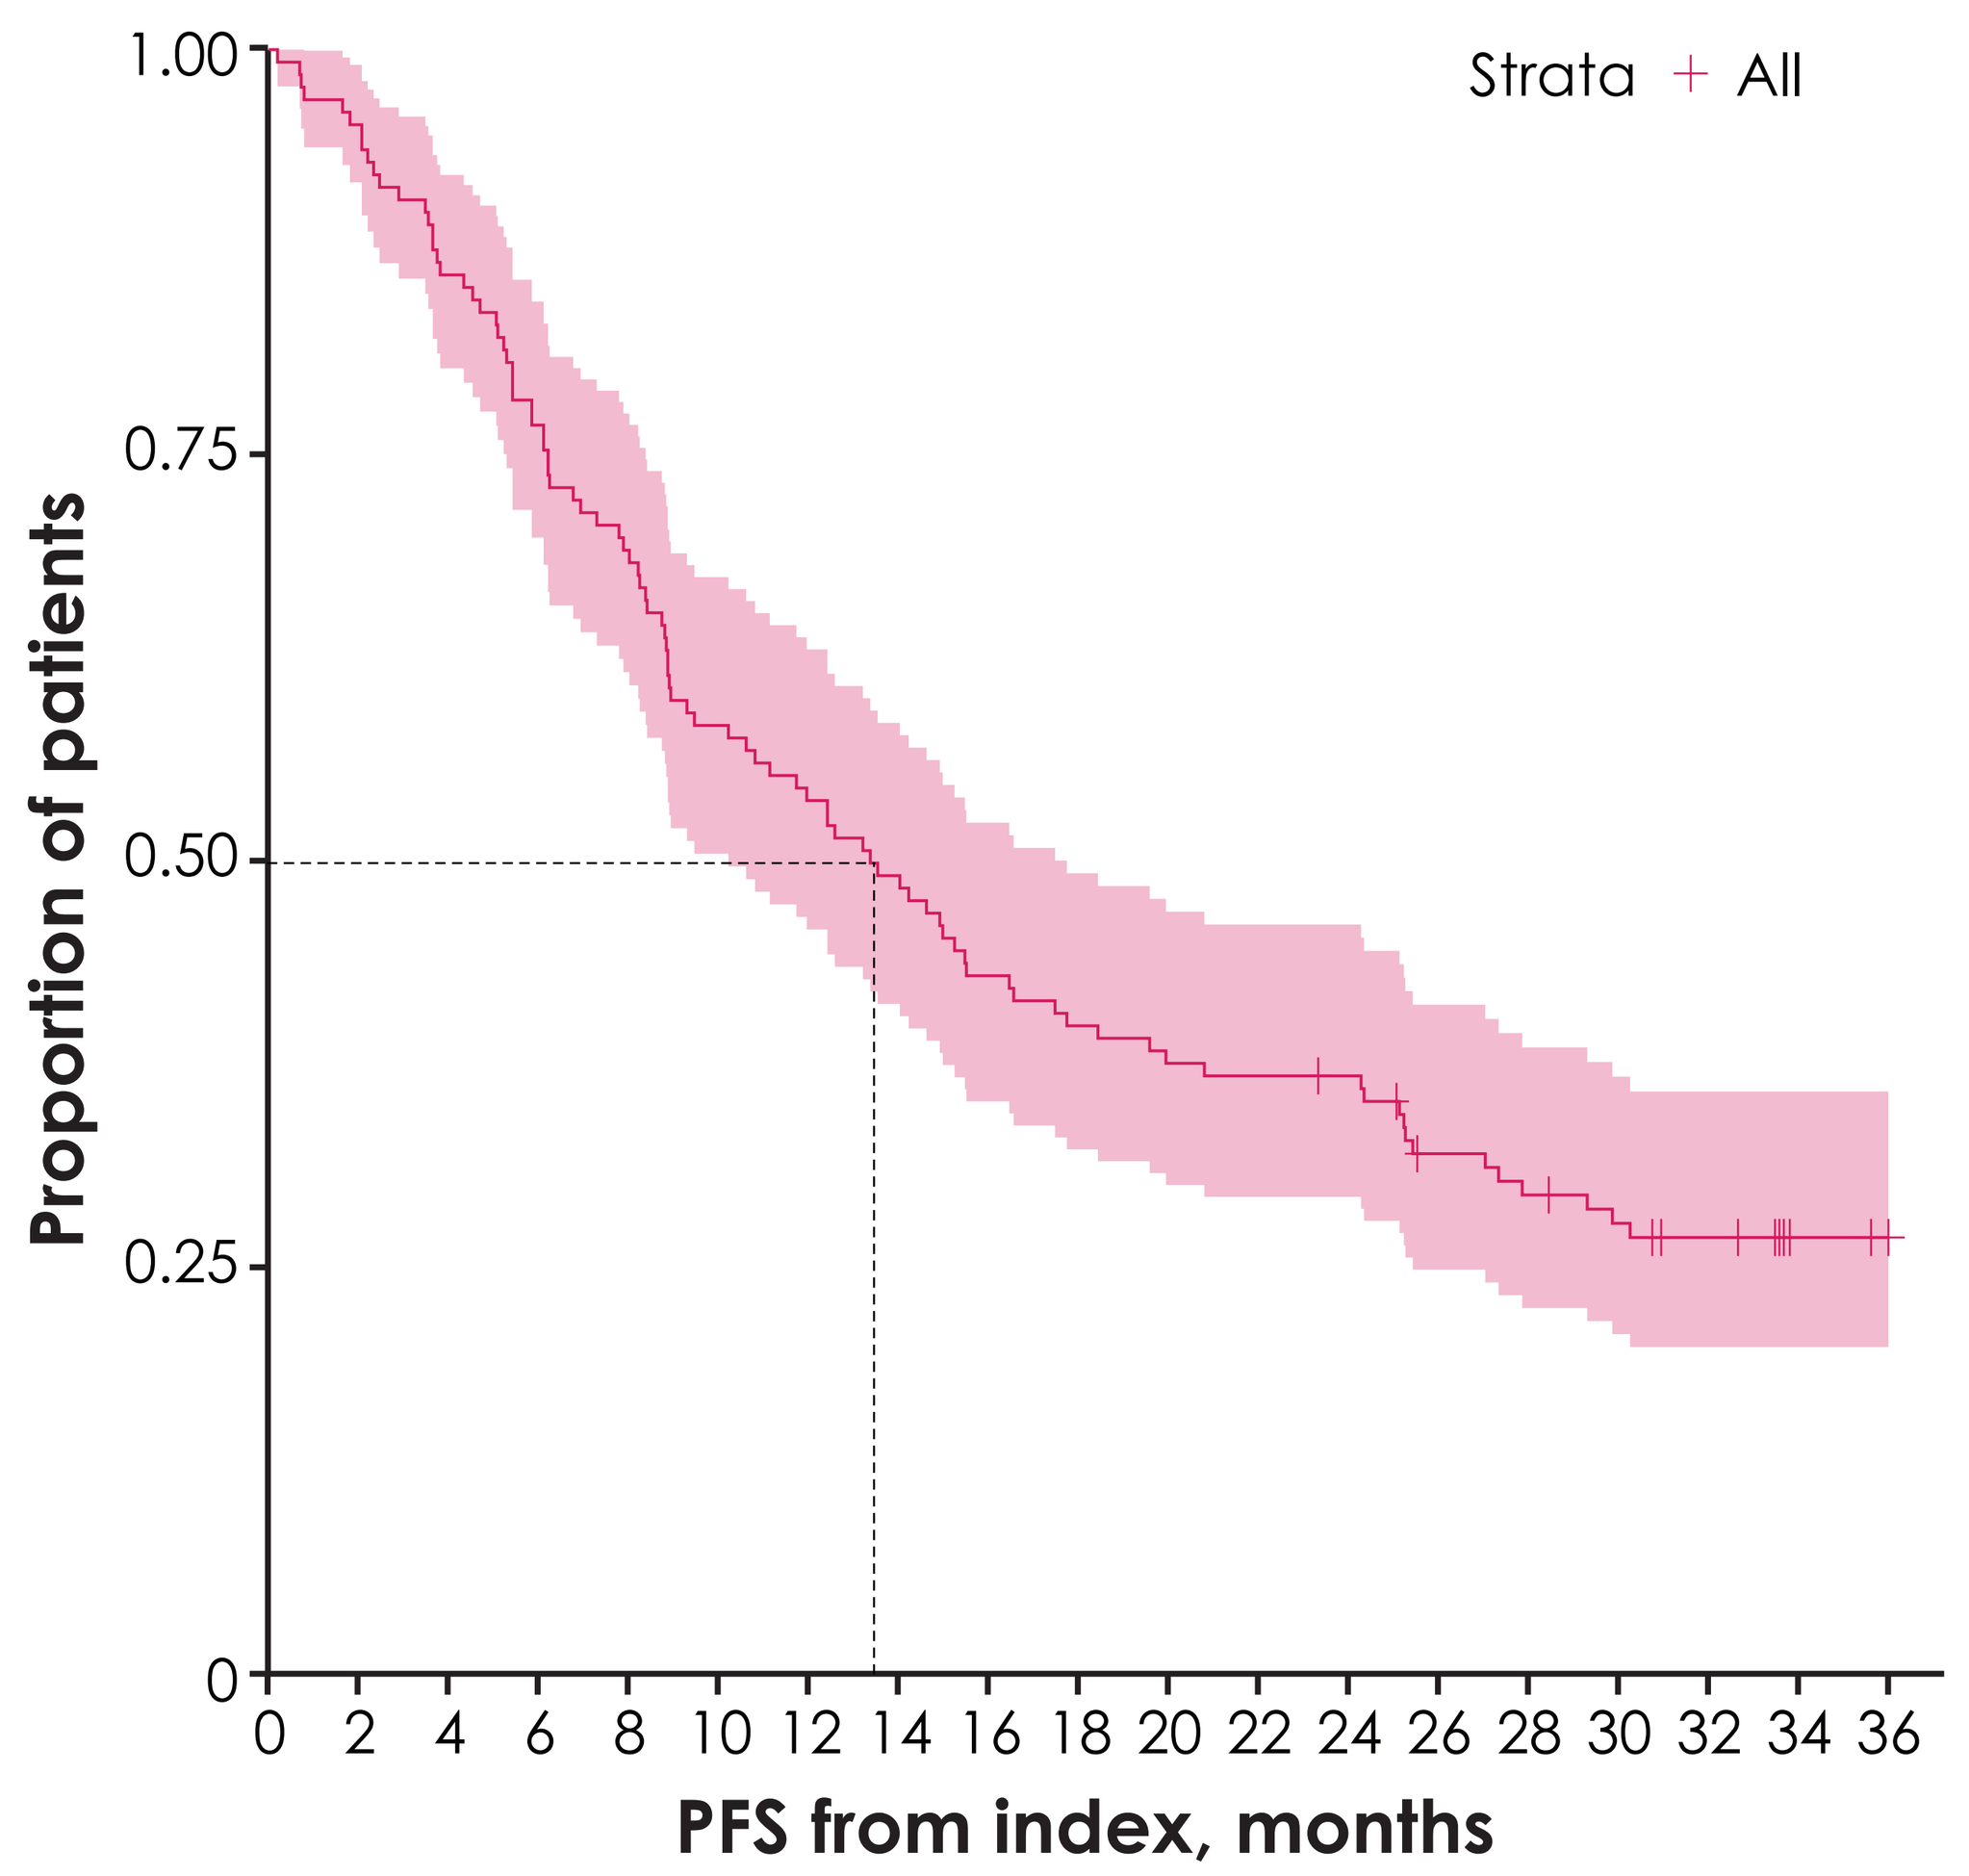

Strata + All
1.00
0.75
0.50
0.25
0
Proportion of patients
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36
PFS from index, months

No. at risk

All 130 124 112 100 90 76 70 64 56 52 49 48 47 38 35 32 29 24 23

PFS, progression-free survival

- The OS rate (95% CI) at 12, 24, and 36 months was 81.5% (75.1%-88.5%), 65.3% (57.6%-74.0%), and 53.3% (45.2%-62.9%), respectively (**Figure 1**)
- Median OS was not estimable
- The PFS rate (95% CI) at 12, 24, and 36 months was 53.1% (45.1%-62.5%), 36.4% (29.0%-45.8%), and 27.0% (20.3%-36.0%), respectively (**Figure 2**)
- Median PFS was 13.5 months (95% CI, 10.2-17.7 months)
- ORR (n=127; 3 not recorded) at 36 months was 62% (95% CI, 53.8%-70.6%), with
 - 5% (n=6) achieving CR
 - 57% (n=73) achieving PR
- ORR stratified by IMDC risk group is summarized in **Figure 3**
- The best responses within 36 months are summarized in **Table 2**

Table 2. Best responses within 36 months

Best response (n=127)	n (%)
Complete response	6 (5)
Partial response	73 (57)
Stable disease	39 (31)
Progressive disease	9 (7)
None recorded	3

Figure 3. Overall response rate by baseline IMDC risk group in evaluable patients (N=125)

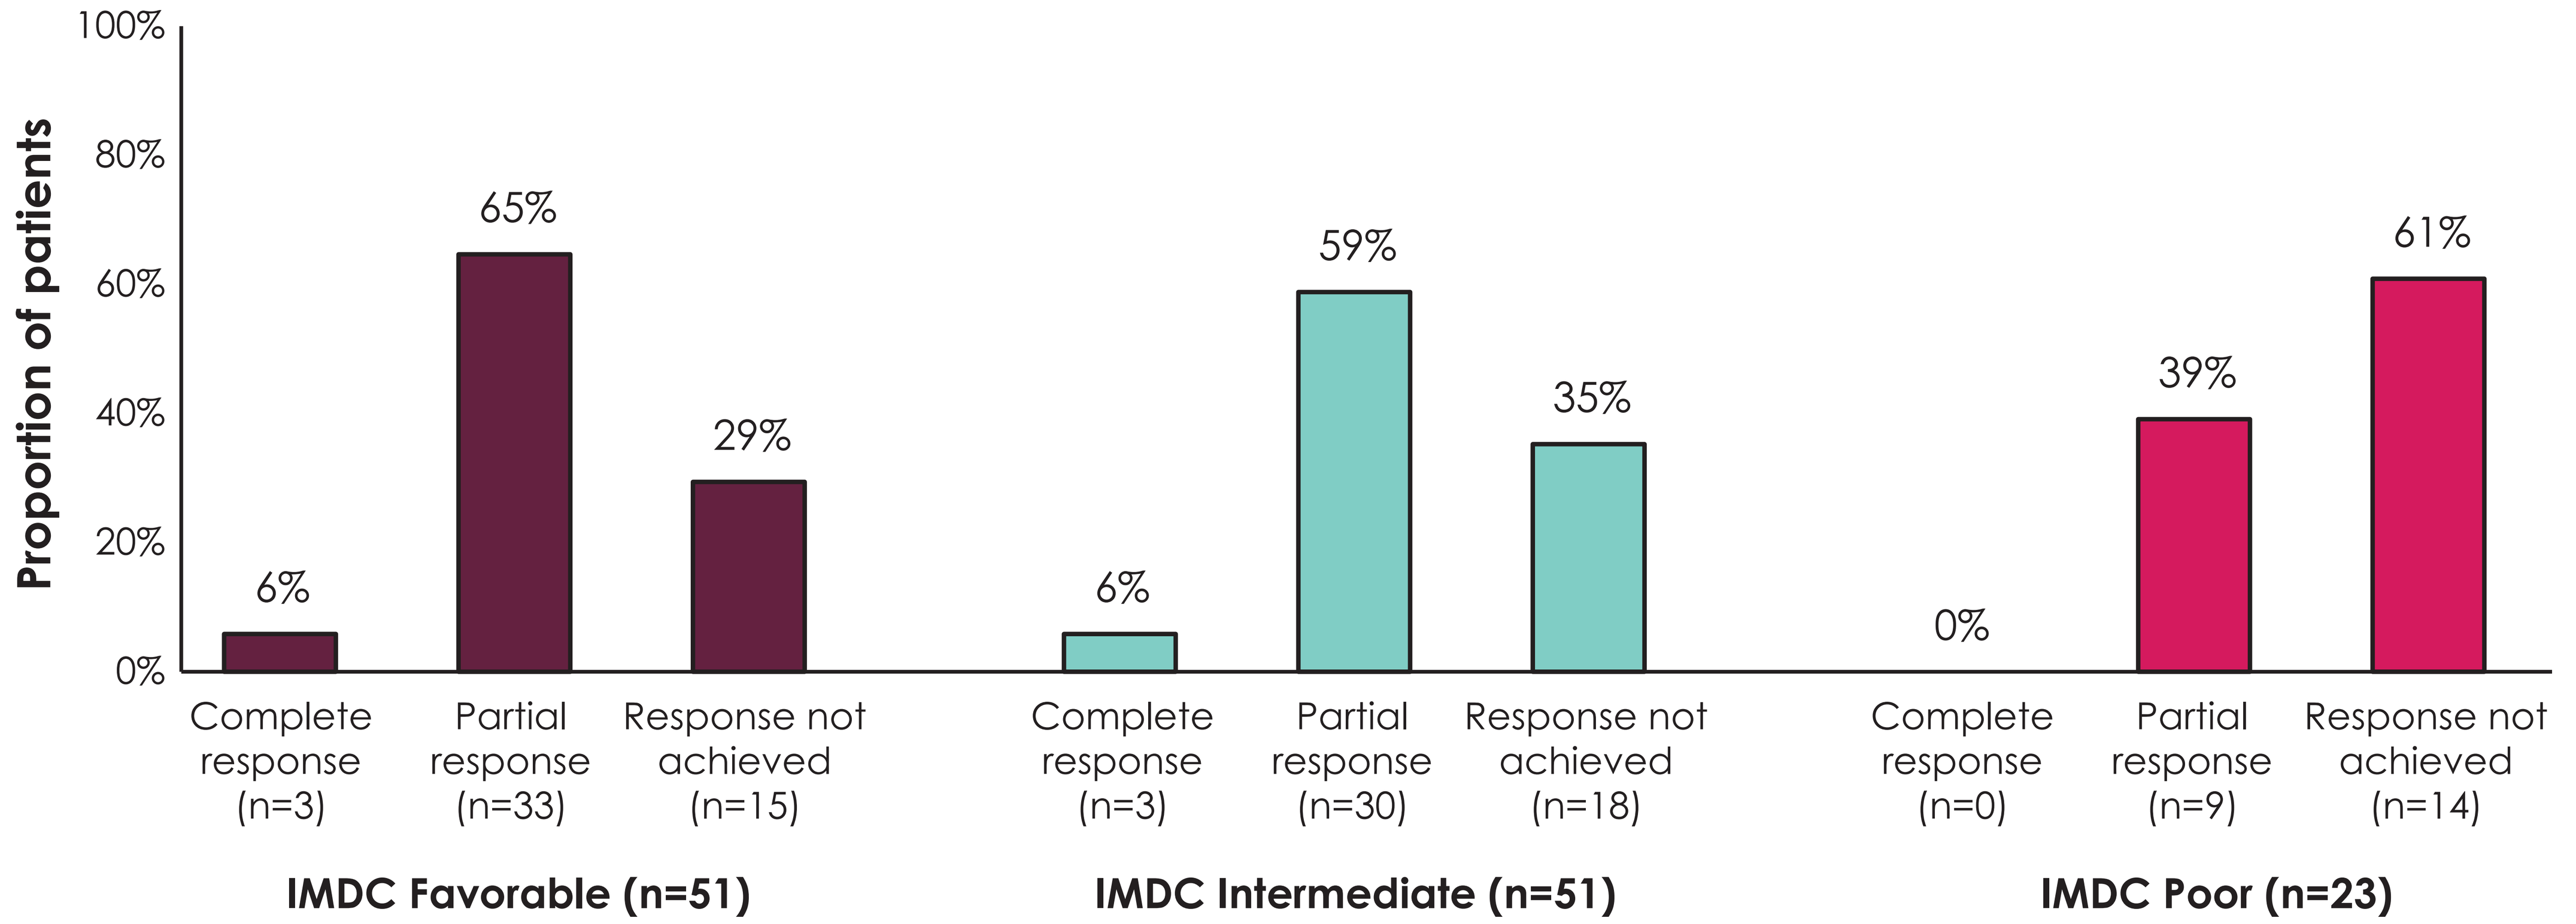

100%
80%
60%
40%
20%
0%
Proportion of patients
6%
65%
29%
Complete response (n=3)
Partial response (n=33)
Response not achieved (n=15)
IMDC Favorable (n=51)
6%
59%
35%
Complete response (n=3)
Partial response (n=30)
Response not achieved (n=18)
IMDC Intermediate (n=51)
0%
39%
61%
Complete response (n=0)
Partial response (n=9)
Response not achieved (n=14)
IMDC Poor (n=23)

IMDC, International Metastatic RCC Database Consortium.

- Median duration of response was 14.9 months (95% CI, 12.0-23.5)
- Median time to discontinuation of A or Ax was 11.7 months (95% CI, 9.0-17.6) and for A + Ax combined was 14.7 months (95% CI, 11.0-24.4)
- AEs were reported in 68% of patients (88/129), who had a total of 519 nonserious AEs due to A + Ax treatment; 13% of patients (17/129) had a total of 27 serious AEs
- The most common AEs were diarrhea (n=49), fatigue (n=33), and oral mucositis (n=24); 9 patients discontinued A and/or Ax due to AEs, including diarrhea in 3 patients

GET POSTER PDF

Copies of this poster obtained through this Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the corresponding author of this poster. Per ASCO requirements you will be redirected to the ASCO meeting site.

Correspondence: **Anand Sharma,** anand.sharma3@nhs.net

REFERENCES 1. Allison J, et al. J Clin Oncol. 2021;39(Suppl 6): abstract 294. **2.** Choueiri RK, et al. Ann Oncol. 2020;31(8):1030-9. **3.** Tomita Y, et al. J Clin Oncol. 2021;39(Suppl 15): abstract 4548. **4.** Motzer RJ, et al. N Engl J Med. 2019;380(12):1103-15. **5.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA, Darmstadt, Germany; 2022. **6.** National Institute for Health and Care Excellence. Accessed January 12, 2023. https://www.nice.org.uk/guidance/ta645/documents/final-appraisal-determination-document. **7.** Medicines and Healthcare Products Regulatory Agency. Accessed January 12, 2023. https://assets.publishing.service.gov.uk/government/uploads/system/uploads/attachment_data/file/844195/Avelumab_EAMS_-_UK_PAR.pdf. **8.** Health Research Authority. Accessed January 12, 2023. https://s3.eu-west-2.amazonaws.com/www.hra.nhs.uk/media/documents/GAfREC_Final_v2.1_July_2021_Final.pdf.

DISCLOSURES P. Nathan: reports providing a consulting or advisory role for AstraZeneca, Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Immunocore, Pfizer, Pierre Fabre, Novartis, GSK, Ipsen, 4SC, and the healthcare business of Merck KGaA, Darmstadt, Germany; has provided speaker services for Bristol Myers Squibb, Novartis, Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has received reimbursement for travel and accommodations expenses from Bristol Myers Squibb and Merck & Co., Kenilworth, NJ. **N. Charnley:** has received research funding from Bristol Myers Squibb; has provided an advisory role for Bristol Myers Squibb; and has provided speaker services for Pfizer and Ipsen. **R. Frazer:** reports providing an advisory role for Bristol Myers Squibb, Novartis, Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has provided speaker services for Pierre-Fabre, Sanofi, Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Servier, Ipsen, EUSA, Eisai, and the healthcare business of Merck KGaA, Darmstadt, Germany. **J. McGrane:** reports providing an advisory role for Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany, Bristol Myers Squibb, and Pfizer; owns stocks/shares in GSK; and has provided speaker services for Bristol Myers Squibb, Ipsen, Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany. **I. Muazzam:** has spoken on behalf of and received expenses from Ipsen and EUSA Pharma. **M. Pillai:** has no conflicts of interest to declare. **S. Rudman:** reports providing an advisory role for Pfizer and owns stocks/shares in Pfizer. **A. Sharma:** reports providing an advisory role for Bristol Myers Squibb, Merck & Co., Kenilworth, NJ, Eisai, EUSA, and Pfizer; and has provided speaker services for Bristol Myers Squibb, Ipsen, Chugai, Novartis, EUSA, and Pfizer. **R. Stevenson:** reports providing an advisory role for Bayer, Astellas, and the healthcare business of Merck KGaA, Darmstadt, Germany; and has provided speaker services for AstraZeneca, Amgen, Bayer, Bristol Myers Squibb, Chugai, EUSA, Janssen, Merck & Co., Kenilworth, NJ, and the healthcare business of Merck KGaA, Darmstadt, Germany, and Pfizer. **B. Venugopal:** reports having received research support from Merck & Co., Kenilworth, NJ, Ipsen, Calithera, Pfizer, and Exelixis; has spoken on behalf of and received expenses from Ipsen, Merck & Co., Kenilworth, NJ, Pfizer, the healthcare business of Merck KGaA, Darmstadt, Germany, and Bristol Myers Squibb; and has participated on advisory boards for Bristol Myers Squibb, EUSA Pharma, Merck & Co., Kenilworth, NJ, and Eisai. **J. Hickey:** is an employee of OPEN Health. **A.R. Ritchie:** is an employee of Pfizer. **K. Liu:** is an employee of Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS** This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and was previously conducted under an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer. Medical writing support was provided by Will Cottam, PhD (OPEN Health), and editorial support was provided by Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. 386. Presented at the ASCO Genitourinary Cancers Symposium, January 25-27, 2024; San Francisco, CA.