Blood Biomarker Dynamics in Highly Active Relapsing Multiple Sclerosis Patients Treated with Cladribine Tablets: Results of the 2-Year MAGNIFY-MS Study

H. Wiendl[1], N. De Stefano[2], F. Barkhof[3,4], X. Montalban[5], A. Achiron[6], T. Derfuss[7], A. Chan[8], S. Hodgkinson[9], A. Prat[10], L. Leocani[11], K. Schmierer[12,13], F. Sellebjerg[14,15], P. Vermersch[16], H. Jin[17], E. Järvinen[17], A. Chudecka[18], L. Gardner[19]

[1]Department of Neurology, Institute of Translational Neurology, University of Münster, Münster, Germany; [2]Department of Medicine, Surgery and Neuroscience, University of Siena, Siena, Italy; [3]Department of Radiology, VU University Medical Center, Amsterdam, The Netherlands and [4]UCL Institute of Neurology, London, UK; [5]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitario Vall d'Hebron, Universitat Autonoma de Barcelona, Barcelona, Spain; [6]Multiple Sclerosis Center, Sheba Academic Medical Center, Ramat Gan, Israel; [7]Department of Neurology, University Hospital Basel, Basel, Switzerland; [8]Department of Neurology, Inselspital, Bern University Hospital, University of Bern, Bern, Switzerland; [9]Ingham Institute for Applied Medical Research, University of New South Wales Medicine, Sydney, NSW; Australia; [10]Department of Neurosciences, Université de Montréal, Montréal, QC, Canada; [11]Experimental Neurophysiology Unit, Vita-Salute San Raffaele University, Milan, Italy;[12]The Blizard Institute, Centre for Neuroscience, Surgery & Trauma, Barts and The London School of Medicine & Dentistry, Queen Mary University of London, London, UK and [13]Clinical Board Medicine (Neuroscience), The Royal London Hospital, Barts Health NHS Trust, London, UK; [14]Danish MS Center, Department of Neurology, Copenhagen University Hospital - Rigshospitalet, Glostrup, Denmark and [15]Department of Clinical Medicine, University of Copenhagen, Copenhagen, Denmark; [16]Univ. Lille, Inserm U1172 LilNCog, CHU Lille, FHU Precise, Lille, France; [17]the healthcare business of Merck KGaA, Darmstadt, Germany; [18]Cytel Inc., Geneva, Switzerland; [19]EMD Serono, Billerica, MA, USA

GET POSTER PDF
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors. QR codes are active only during the congress duration

CONCLUSIONS

- **Multifaceted immune changes were detected over a 2-year observation period following cladribine tablets treatment, including a novel observation regarding increase in CD16^{low}CD56^{bright} and NKp46 cells**
- **Reduction in pro-inflammatory B-cell (IL-6$^+$) and T-cell (GMCSF, TNFα, IFNγ) cytokines, and an increase in anti-inflammatory T-cell (IL-4$^+$) cytokines above baseline level were observed**
- **Serum IgG and IgM levels remained within normal limits for most patients**
- **Serum NfL levels decreased, indicating a positive response to cladribine tablets treatment**

INTRODUCTION

- The MAGNIFY-MS (NCT03364036) blood biomarker (BB) analysis aimed to characterize the effect of cladribine tablets (3.5 mg/kg cumulative dose over 2 years) on BBs in patients with highly active relapsing multiple sclerosis (RMS).
- Previously published data characterized the specific patterns of immune cell dynamics over 12 months following the first course of treatment with cladribine tablets in a subset of MAGNIFY-MS patients (n=57), and highlighted the pronounced reduction of immune cell subtypes from Month 2 onwards.[1]

OBJECTIVE

To describe BB dynamics over 2 years of treatment with cladribine tablets, including immune cell subsets, serum proteins, and intracellular cytokines.

METHODS

- Blood samples from patients in MAGNIFY-MS were collected at baseline and Months (M) 3, 6, 12, 15, 18, and 24 after treatment initiation for immune cell characterization, measurement of immunoglobulins (Ig), and serum neurofilament (sNfL) analysis.
- Intracellular cytokine analyses were performed for a subset of patients in the BB sub-study and required blood collection at additional time points (M1, 2, and 14).
- All analyses were performed without adjustment for multiplicity.
- Correlation analysis between change in immune cell subsets from Baseline to the M3/6 visit versus presence of MRI lesions during the Baseline period to M3/6 was assessed with Cramer's V, and included patients with both B-cell and MRI assessments from the main study (N=270).

Cladribine tablets 10 mg (3.5 mg/kg cumulative dose over 2 years) are indicated for the treatment of patients with MS in the United States (relapsing forms of MS, including relapsing-remitting disease and active secondary progressive disease, in adults).

RESULTS

- A total of 270 patients enrolled in the main study were analyzed (BB sub-study n=57, **Supplementary Table 1**).
- Quantitative immune cell changes were observed following each yearly treatment course of cladribine tablets, as summarized below.

B-cell Panel (FAS)

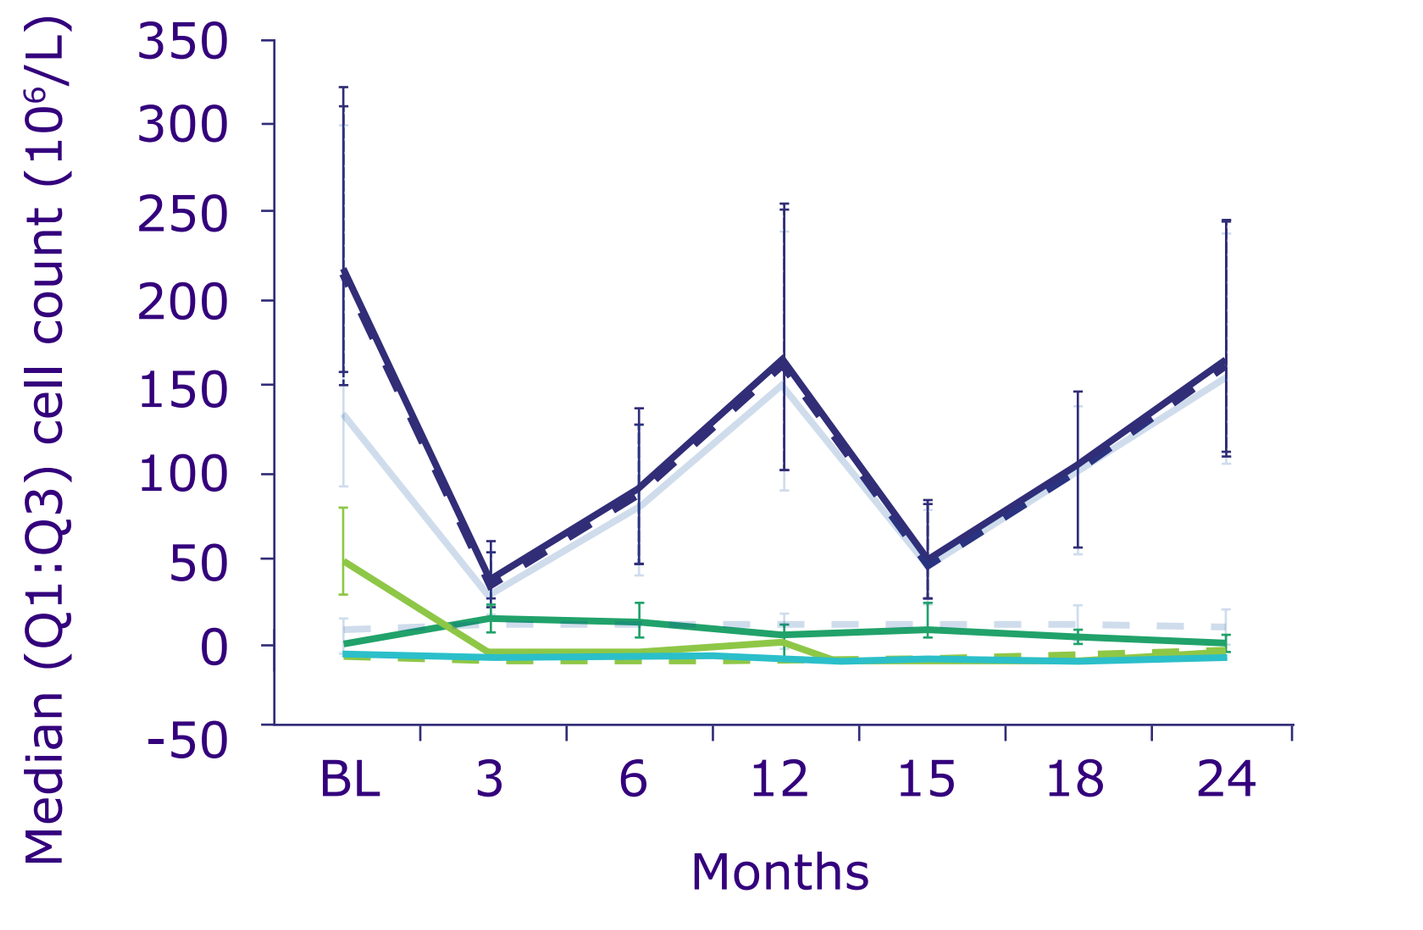
350
300
250
200
150
100
50
0
-50
Median (Q1:Q3) cell count (10⁶/L)
BL
3
6
12
15
18
24
Months
CD19+
CD20+
Memory
Activated CD69+
Plasma cells CD38+
Short-lived plasma
Naïve
Transitional
Regulatory

Median Percentage Change from Baseline

	M3	M6	M12	M15	M18	M24
CD19$^+$	-80.14	-60.60	-26.88	-77.24	-55.30	-27.65
CD20$^+$	-80.50	-60.32	-24.56	-77.11	-54.21	-24.77
Memory	-92.69	-91.56	-86.90	-96.47	-94.67	-89.29
Activated CD69$^+$	-74.02	-60.91	-28.82	-73.02	-51.87	-15.95
Plasma cells CD38$^+$	-66.62	-59.00	-54.75	-78.02	-72.39	-62.47
Short-lived plasma	-68.18	-56.55	-56.70	-82.96	-79.54	-70.10
Naïve	-75.87	-45.87	1.63	-69.17	-39.73	10.85
Transitional	-4.06	14.82	11.92	28.69	11.27	6.30
Regulatory	110.73	92.95	30.64	91.57	33.83	1.62

In the table, purple shading signifies value above Baseline level; green shading, highest value above Baseline level; blue shading, nadir value.
BL, baseline; **CD**, cluster of differentiation; **FAS**, full analysis set; **Q1:Q3**, quartile range; **M**, month

- Median percentage change from baseline at M24: regulatory (+1.62%), transitional (+6.3%), naïve (+10.85%), memory (-89.29%) B-cells; and total plasma cells: -62.47%.

T-cell Panel (FAS)

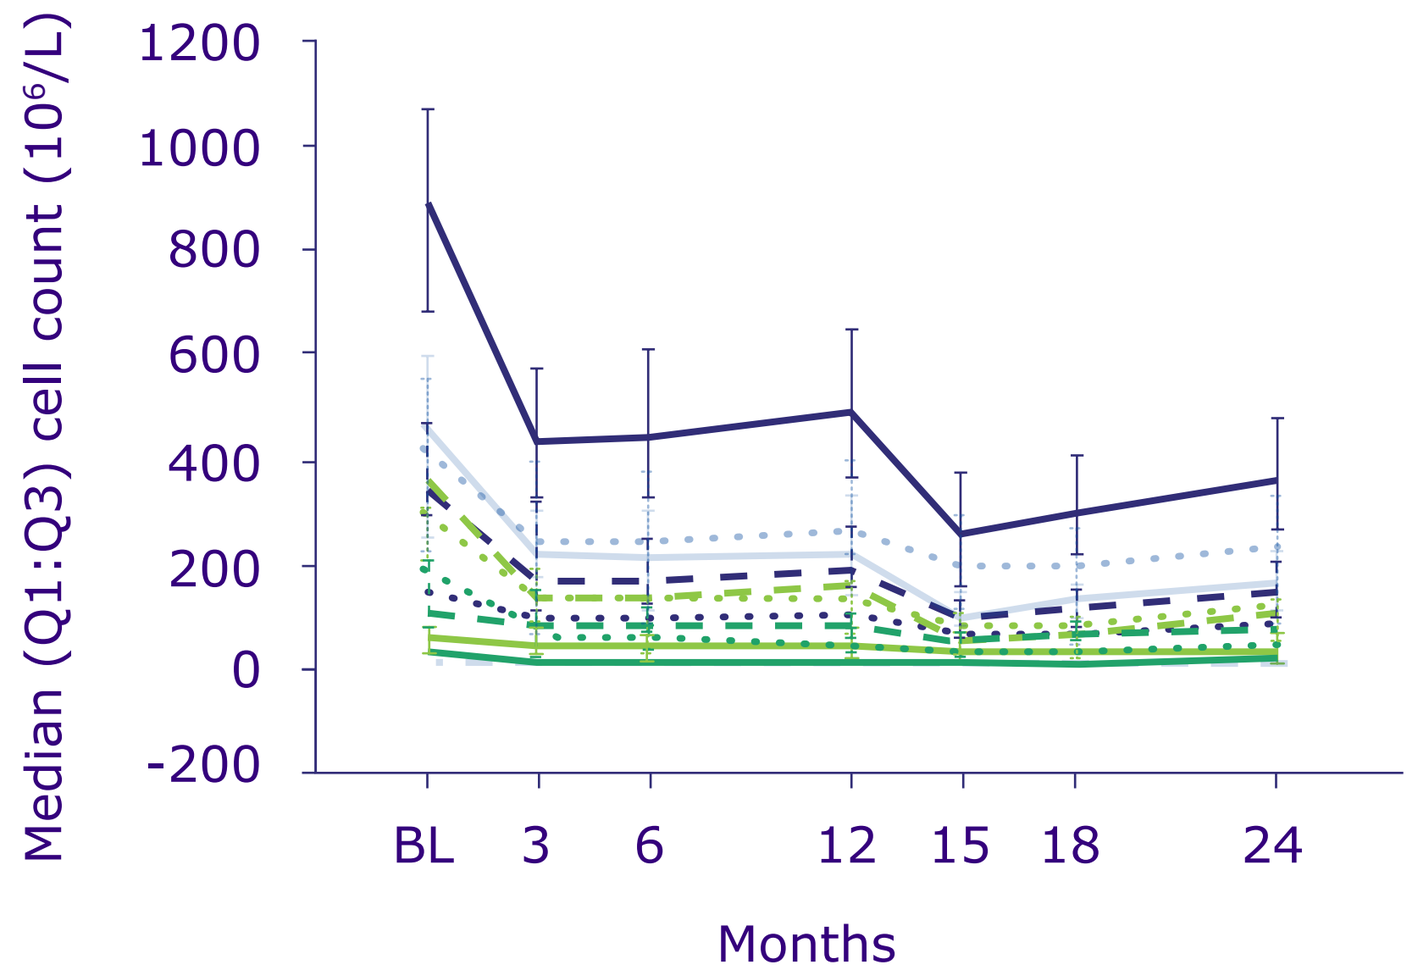
1200
1000
800
600
400
200
0
-200
Median (Q1:Q3) cell count (10⁶/L)
BL
3
6
12
15
18
24
Months
CD4+
CD4+ CM
CD4+ EM
CD4+ Th2
CD4+ Th17
CD8+
CD4+ Regulatory
CD4+ Naïve
CD4+ Th1
CD8+ CM
CD8+ EM
CD8+ Naïve

Median Percentage Change from Baseline

	M3	M6	M12	M15	M18	M24
CD4$^+$	-48.60	-47.18	-40.16	-69.04	-66.98	-57.51
CD4$^+$ CM	-49.83	-47.31	-39.97	-70.18	-65.91	-56.96
CD4$^+$ EM	-28.62	-26.81	-19.81	-46.05	-48.69	-38.56
CD4$^+$ reg	-25.98	-29.84	-25.60	-48.40	-48.73	-40.30
CD4$^+$ naïve	-60.43	-57.74	-50.50	-83.66	-78.50	-68.85
CD4$^+$ Th1	-44.35	-43.20	-35.55	-63.68	-63.01	-52.86
CD4$^+$ Th2	-50.89	-52.31	-47.80	-75.96	-71.82	-62.72
CD4$^+$ Th17	-33.09	-30.26	-18.39	-44.57	-44.77	-31.74
CD8$^+$	-42.33	-39.42	-36.28	-57.08	-54.44	-45.93
CD8$^+$ CM	-40.92	-43.30	-32.26	-62.74	-56.93	-43.26
CD8$^+$ EM	-26.20	-23.09	-22.26	-44.11	-44.18	-35.05
CD8$^+$ naïve	-67.58	-65.44	-58.31	-84.66	-77.74	-67.54

In the table, blue shading signifies nadir value.
CM, central memory; **EM**, effector memory; **FAS**, full analysis set; **Q1:Q3**, quartile range; **reg**, regulatory; **Th**, T helper cell type

- Reduction in most T cells is seen from M3 onward; no repopulation to baseline levels was observed during the trial.

Serum proteins (FAS)

Table 1. Median Serum IgM and IgG Levels Over Time (FAS)

	BL	M3	M6	M12	M15	M18	M24
IgM absolute value, g/L	1.14	1.1	1.01	0.95	0.79	0.8	0.77
IgM percentage change from BL	-	-6.98	-13.38	-19.61	-27.33	-30.65	-31.49
IgG absolute value, g/L	10.3	10.6	10.4	10.35	10.6	10.7	10.6
IgG percentage change from BL	-	5.00	3.71	1.05	4.90	2.40	2.99

BL, baseline; **FAS**, full analysis set; **Ig**, immunoglobulin; **M**, month

- Serum IgM levels decreased throughout the study (**Table 1**). There were no clinically relevant changes in IgG levels. The majority of patients had their individual values of IgM and IgG within reference ranges (IgG: 5.65-17.65 g/L; IgM: 0.40-2.30 g/L).
- Compared with baseline, sNfL was reduced at M12 (-25.22%) and M24 (-23.23%) (**Supplementary Figure 1**).

NK-cell Panel (FAS)

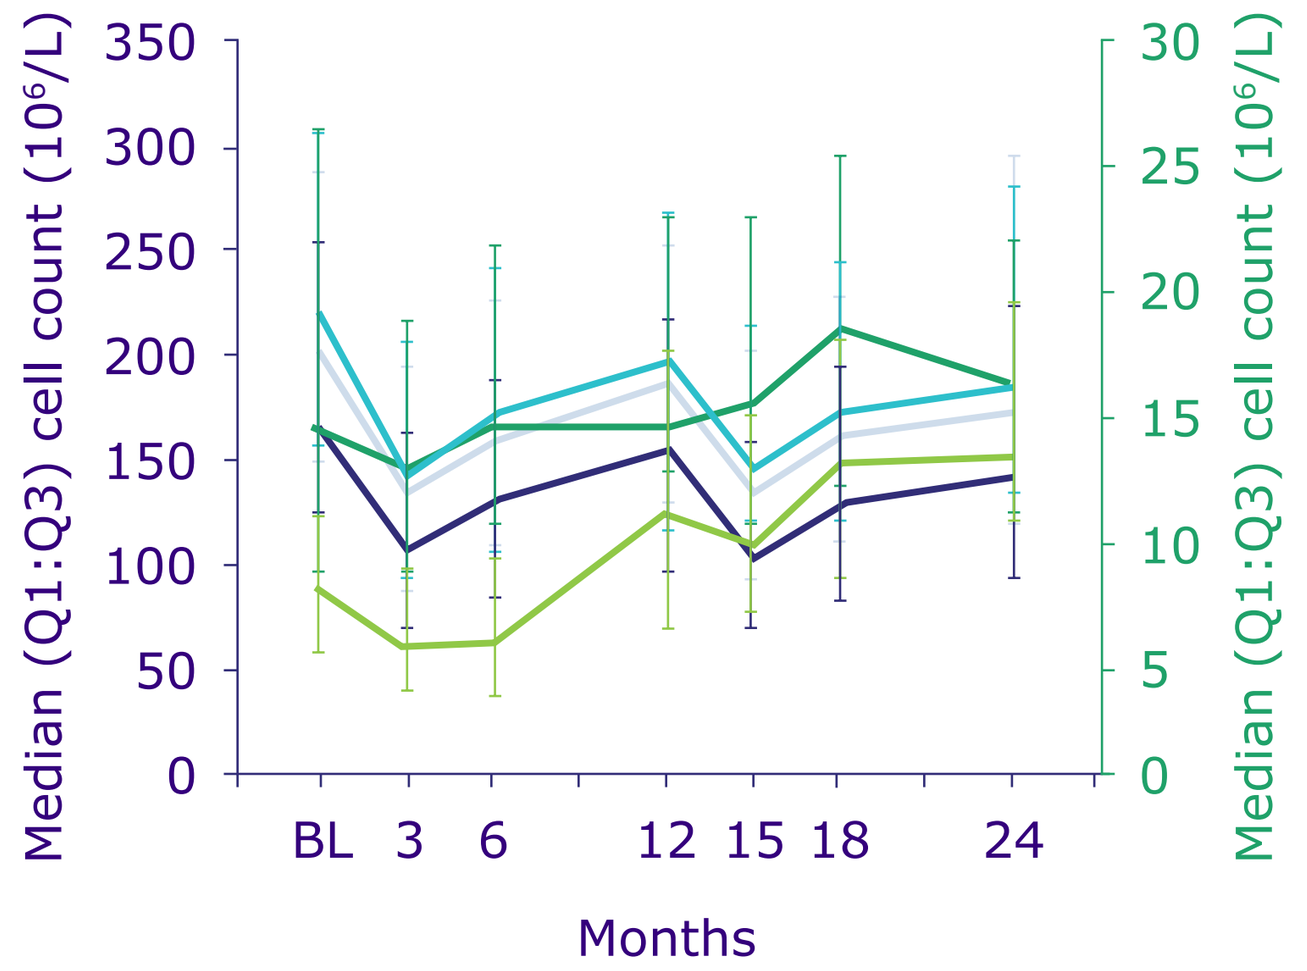
350
300
250
200
150
100
50
0
Median (Q1:Q3) cell count (10⁶/L)
30
25
20
15
10
5
0
Median (Q1:Q3) cell count (10⁶/L)
BL
3
6
12
15
18
24
Months
CD16bright CD56dim
CD16low CD56bright
NKp46
CD16+
NK cells

Median Percentage Change from Baseline

	M3	M6	M12	M15	M18	M24
CD16^{bright} CD56^{dim}	-36.13	-25.61	-11.05	-35.05	-24.99	-12.94
CD16^{low} CD56^{bright}	-8.94	3.72	2.56	4.77	30.13	17.21
NKp46	-20.85	-22.38	29.49	28.42	71.73	77.70
CD16$^+$	-32.50	-21.78	-8.10	-28.56	-21.47	-13.76
NK cells	-31.15	-22.18	-7.10	-27.70	-18.38	-13.05

In the table, purple shading signifies value above Baseline level; green shading, highest value above Baseline level; blue shading, nadir value.
BL, baseline; **CD**, cluster of differentiation; **FAS**, full analysis set; **M**, month; **NK**, natural killer; **Q1:Q3**, quartile range

- Increased levels above baseline from M12 onwards were detected for CD16^{low} CD56^{bright} (M24, +17.21%) and NKp46 cells (M24, +77.7%).

Intracellular Cytokines

- In the BB sub-study, reductions in *pro-inflammatory* cytokine-producing IL-6$^+$ B-cells and GMCSF$^+$, TNFα$^+$ and IFNγ$^+$ T-cells were observed. There were also increases in *anti-inflammatory* cytokine-producing IL-10$^+$ B cells and IL-4$^+$ CD4$^+$ and CD8$^+$ T cells (**Tables 2** and **3**).

Table 2. Median Absolute Change (%) from Baseline for Cytokine Producing B-cells (BB Sub-study)

MFI B-cell subset	Time point (months)					
	M3	M6	M12	M15	M18	M24
IL-10$^+$ B cells	1.9	1.11	-0.94	-1.6	-1.57	-0.78
IL-10$^+$ Memory B cells	-0.93	-1.35	-1.47	-2.49	-1.52	-1.44
IL-6$^+$ B cells	-7.77	-9.02	-16.33	-15.31	-15.62	-12.69
IL-6$^+$ Memory B cells	-8.72	-9.53	-12.02	-10.63	-11.56	-10.88

In the table, purple shading signifies value above Baseline level; green shading, highest value above Baseline level; blue shading, nadir value.
BB, blood biomarker; **IL**, interleukin; **M**, month; **MFI**, mean fluorescence intensity; **Q1:Q3**, quartile range

Table 3. Median Absolute Change (%) from Baseline for Cytokine Producing T-cells (BB sub-study)

MFI T-cell subset	Time point (months)					
	M3	M6	M12	M15	M18	M24
GMCSF$^+$ CD4$^+$ T cells	-3.25	-1.61	-5.71	-6.67	-5.41	-3.93
IFNγ$^+$ CD4$^+$ T cells	-3.63	-4.10	-6.13	-9.20	-6.97	-5.15
TNFα$^+$ CD4$^+$ T cells	-12.15	-16.30	-25.85	-46.30	-48.40	-28.45
IL-4$^+$ CD4$^+$ T cells	0.79	0.84	0.61	0.92	2.30	1.70
IL-10$^+$ CD4$^+$ T cells	1.78	0.53	-0.30	-0.87	-1.16	-0.94
GMCSF$^+$ CD8$^+$ T cells	-3.77	-2.80	-10.48	-11.60	-9.63	-4.70
IFNγ$^+$ CD8$^+$ T cells	-0.3	-3.05	-14.72	-18.78	-15.06	-11.85
TNFα$^+$ CD8$^+$ T cells	-13.93	-15.40	-27.65	-44.65	-48.48	-28.05
IL-4$^+$ CD8$^+$ T cells	0.40	0.74	0.18	0.52	1.19	0.61
IL-10$^+$ CD4$^+$ T cells	-0.01	-0.23	-0.68	-1.19	-1.01	-0.89

In the table, purple shading signifies value above Baseline level; green shading, highest value above Baseline level; blue shading, nadir value.
BB, blood biomarker; **CD**, cluster of differentiation; **GMCSF**, granulocyte-macrophage colony-stimulating factor; **IFNγ**, interferon gamma; **IL**, interleukin; **M**, month; **MFI**, mean fluorescence intensity; **TNFα**, tumor necrosis factor alpha

Correlations

- No correlation was found between memory or regulatory B cells and CUA, T1 Gd+ and active T2 lesions at M3 and M6 (**Supplementary Figure 2**).

For patient demographics, sNfL, and correlation data please scan QR code

Reference: 1. Wiendl H, et al. *Neurol Neuroimmunol Neuroinflamm*. 2023;10(1):e200048.

Disclosures: **HW** is member of scientific advisory boards/steering committees for Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva. He received speaker honoraria and travel support from Bayer, Biogen, CSL Behring, EMD Serono, Fresenius Medical Care, the healthcare business of Merck KGaA, Darmstadt, Germany, Omniamed, Novartis, Sanofi, and Teva. He received compensation as a consultant from Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Omniamed, Roche, and Sanofi. He has received research support from Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Sanofi, and Teva, as well as German Ministry for Education and Research (BMBF), German Research Foundation (DFG), Else Kröner Fresenius Foundation, Fresenius Foundation, Hertie Foundation, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, NRW Ministry of Education and Research, Interdisciplinary Center for Clinical Studies (IZKF) Münster, and RE Children's Foundation. **NDS** is a consultant for Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva; has grants or grants pending from FISM and Novartis, is on the speakers' bureaus of Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva; and has received travel funds from the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva. **FB** is supported by the NIHR Biomedical Research Centre at UCLH and is a consultant to Biogen, Combinostics, IXICO, the healthcare business of Merck KGaA, Darmstadt, Germany, and Roche. **XM** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with AbbVie, Actelion (Janssen/J&J), Alexion, Biogen, Celgene (BMS), EMD Serono, Immunic, Janssen, MedDay, the healthcare business of Merck KGaA, Darmstadt, Germany, Mylan, Nervgen, Novartis, Roche, Sandoz, Sanofi, Teva, TG Therapeutics, Excemed, MSIF, and NMSS. **AA** has received honoraria or consulting fees from Bayer, Biogen, Celgene (BMS), the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi; and research support from Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Roche, and Sanofi. **TD** serves on scientific advisory boards for Bayer, Actelion (Janssen/J&J), Biogen, Celgene (BMS), GeNeuro, MedDay, the healthcare business of Merck KGaA, Darmstadt, Germany, Mitsubishi Pharma, Novartis, Roche, and Sanofi; has received funding for travel and/or speaker honoraria from Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi; and receives research support from Biogen, the European Union, Novartis, Roche, the Swiss MS Society, and the Swiss National Foundation. **AC** has received speaker/advisory board honoraria from Actelion (Janssen/J&J), Almirall, Bayer, Biogen, Celgene (BMS), the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva, all for hospital research funds. He has received research support from Biogen, Sanofi, and UCB, the European Union, and the Swiss National Foundation. He serves as associate editor of the European Journal of Neurology, on the editorial board for Clinical and Translational Neuroscience, and as topic editor for the Journal of International Medical Research. **SH** serves on advisory boards for Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi. She has received money for travel and speaker honoraria from Bayer, Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, and Sanofi. **AP** has received honoraria and operating grants from pharmaceutical companies. **LL** has received honoraria for consulting services or speaking activities from Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, and Roche; and research support from Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, and Novartis. **KS** has received research support from Biogen, the healthcare business of Merck KGaA, Darmstadt, Germany, and Novartis; speaking honoraria from, and/or served in an advisory role for, Amgen-Gensenta, Biogen, EMD Serono, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva; and remuneration for teaching activities from AcadeMe, Medscape, and the Neurology Academy. FS has served on scientific advisory boards, been on the steering committees of clinical trials, served as a consultant, received support for congress participation, received speaker honoraria, or received research support for his laboratory from Biogen, Celgene (BMS), EMD Serono, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva. **PV** has received honoraria or consulting fees from AB Science, Biogen, Celgene (BMS), Imcyse, the healthcare business of Merck KGaA, Darmstadt, Germany, Novartis, Roche, Sanofi, and Teva; and research support from Novartis, Roche, and Sanofi. **HJ** and **EJ** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **ACHu** is an employee of Cytel Inc., Geneva Branch, Switzerland, funded by the healthcare business of Merck KGaA, Darmstadt, Germany to perform statistical analyses for this study. **LG** is an employee of EMD Serono, Billerica, MA, USA

Medical writing support was provided by Joe Ward of inScience Communications, Springer Healthcare Ltd, UK, and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Presented at ACTRIMS Forum 2023 | 23–25 February | San Diego, CA, USA

For reactive Medical use only

February 2023

The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)

RESULTS

MAGNIFY MS
MS 700568-0022

Supplementary Table 1. Patient Demographics

	Main study population (N=270)	Sub-study population (n=57)
Female, n (%)	180 (66.7)	35 (61.4)
Age >40 – <65 years, n (%)	118 (43.7)	22 (38.6)
Time since onset of MS in months, mean ± SD	84.90 ±85.472	84.94 ± 93.385
Time since diagnosis in months, mean ± SD	60.87 ±74.489	52.54 ± 67.413
Time since first relapse in months, mean ± SD	54.44 ±72.583	52.63 ± 80.704
Number of relapses within 12 months prior to Baseline, n (%)		
0	3 (1.1)	2 (3.5)
1	102 (37.8)	15 (26.3)
2	133 (49.3)	29 (50.9)
>2	32 (11.9)	11 (19.3)
EDSS score ≤3 at Baseline, n (%)	204 (75.6)	42 (73.7)
Median EDSS at Baseline (range)	2.0 (0.0–5.0)	2.5 (0.0–5.0)
Number of previous DMTs, n (%)		
0	117 (43.3)	30 (52.6)
1	88 (32.6)	13 (22.8)
2	50 (18.5)	10 (17.5)
>2	15 (5.6)	4 (7.0)

DMT, disease-modifying therapy; **EDSS**, Expanded Disability Status Scale; **MS**, multiple sclerosis; **SD**, standard deviation

For reactive Medical use only

February 2023

Presented at ACTRIMS Forum 2023 | 23–25 February | San Diego, CA, USA

The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)

RESULTS

MAGNIFY MS
MS 700568-0022

Supplementary Figure 1. sNfL Levels Over Time (FAS)

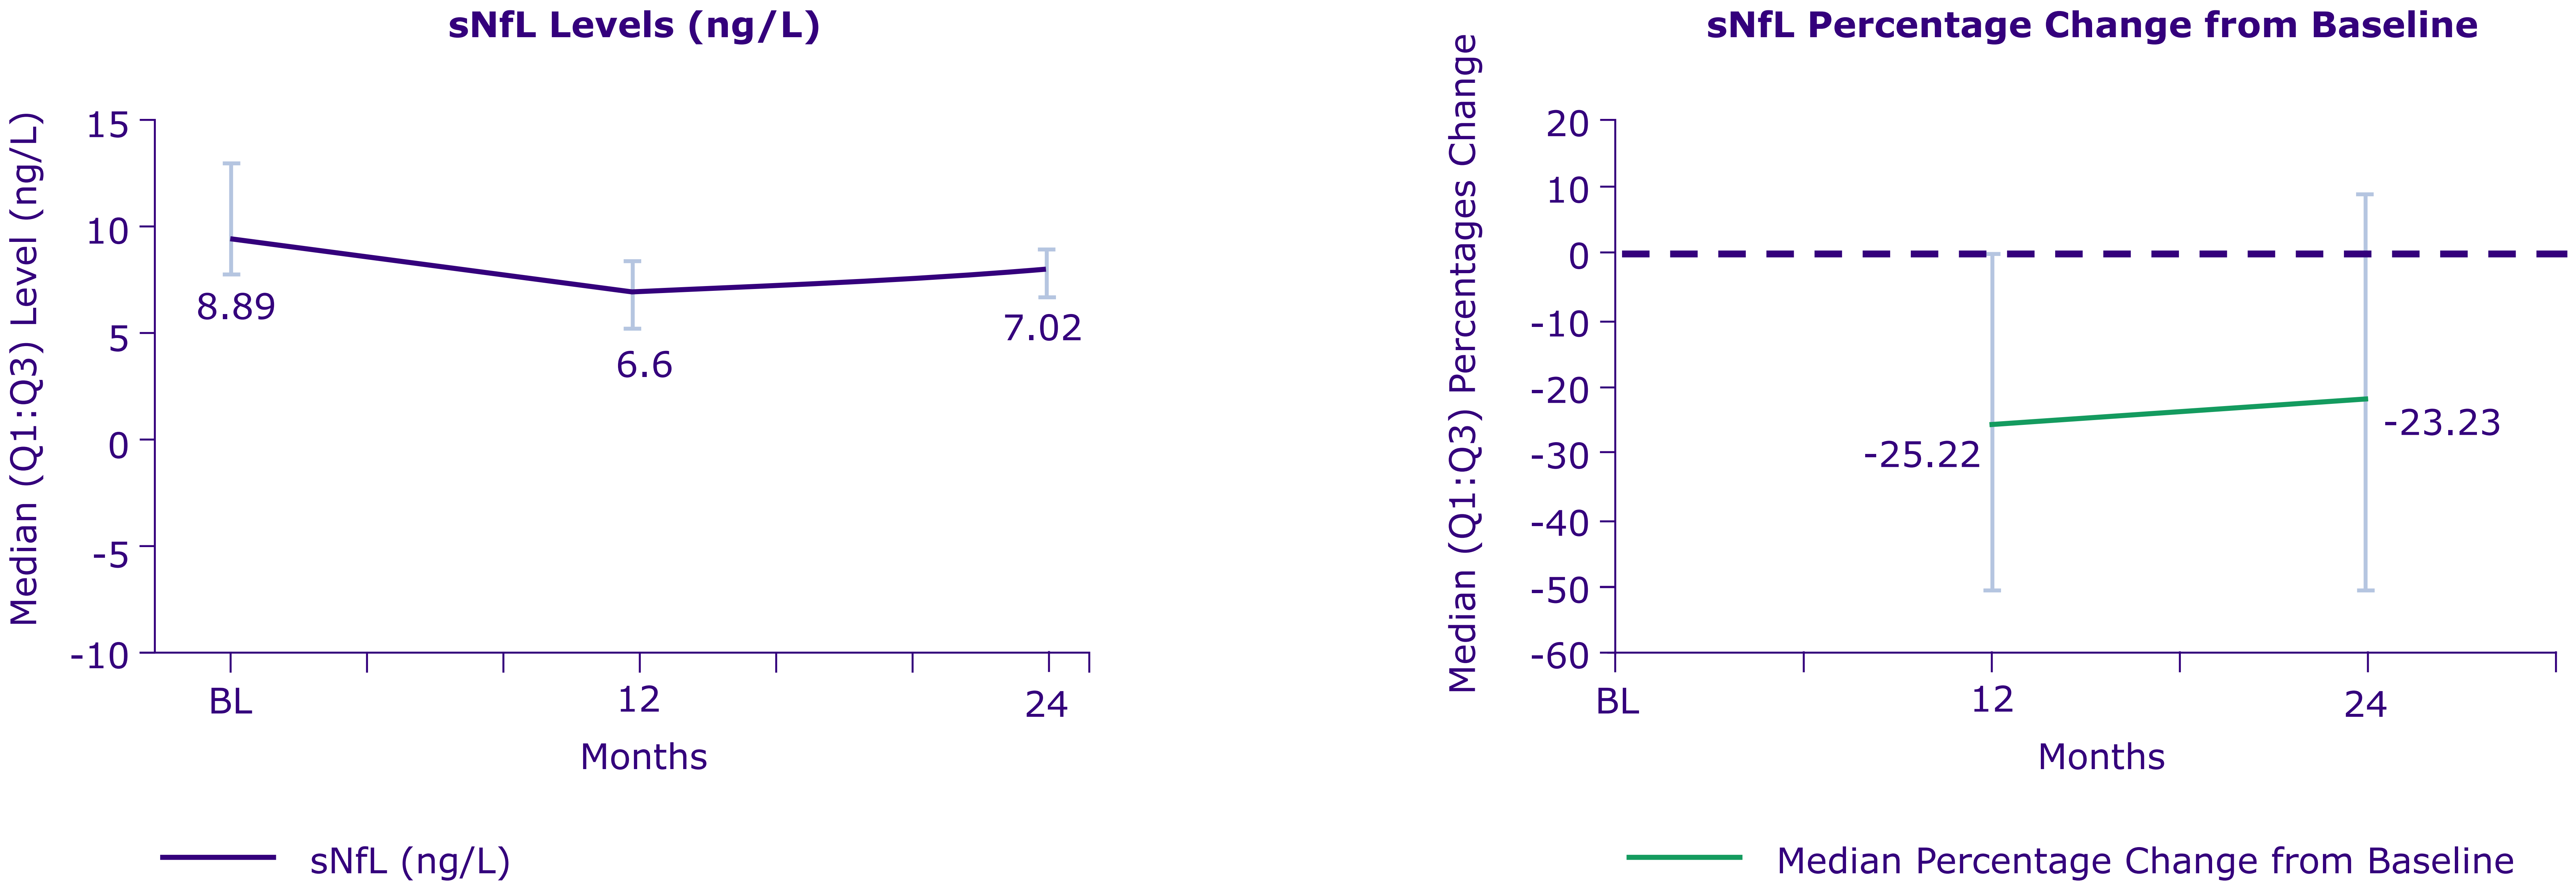

sNfL Levels (ng/L)
Median (Q1:Q3) Level (ng/L)
15
10
5
0
-5
-10
8.89
6.6
7.02
BL
12
24
Months
sNfL (ng/L)
sNfL Percentage Change from Baseline
Median (Q1:Q3) Percentages Change
20
10
0
-10
-20
-30
-40
-50
-60
-25.22
-23.23
BL
12
24
Months
Median Percentage Change from Baseline

BL, baseline; **FAS**, full analysis set; **M**, month; **Q1:Q3**, quartile range; **sNfL**, serum neurofilament light chain

For reactive Medical use only

February 2023

Presented at ACTRIMS Forum 2023 | 23–25 February | San Diego, CA, USA

The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)

RESULTS

Supplementary Figure 2. Correlation of B-cells with MRI Findings

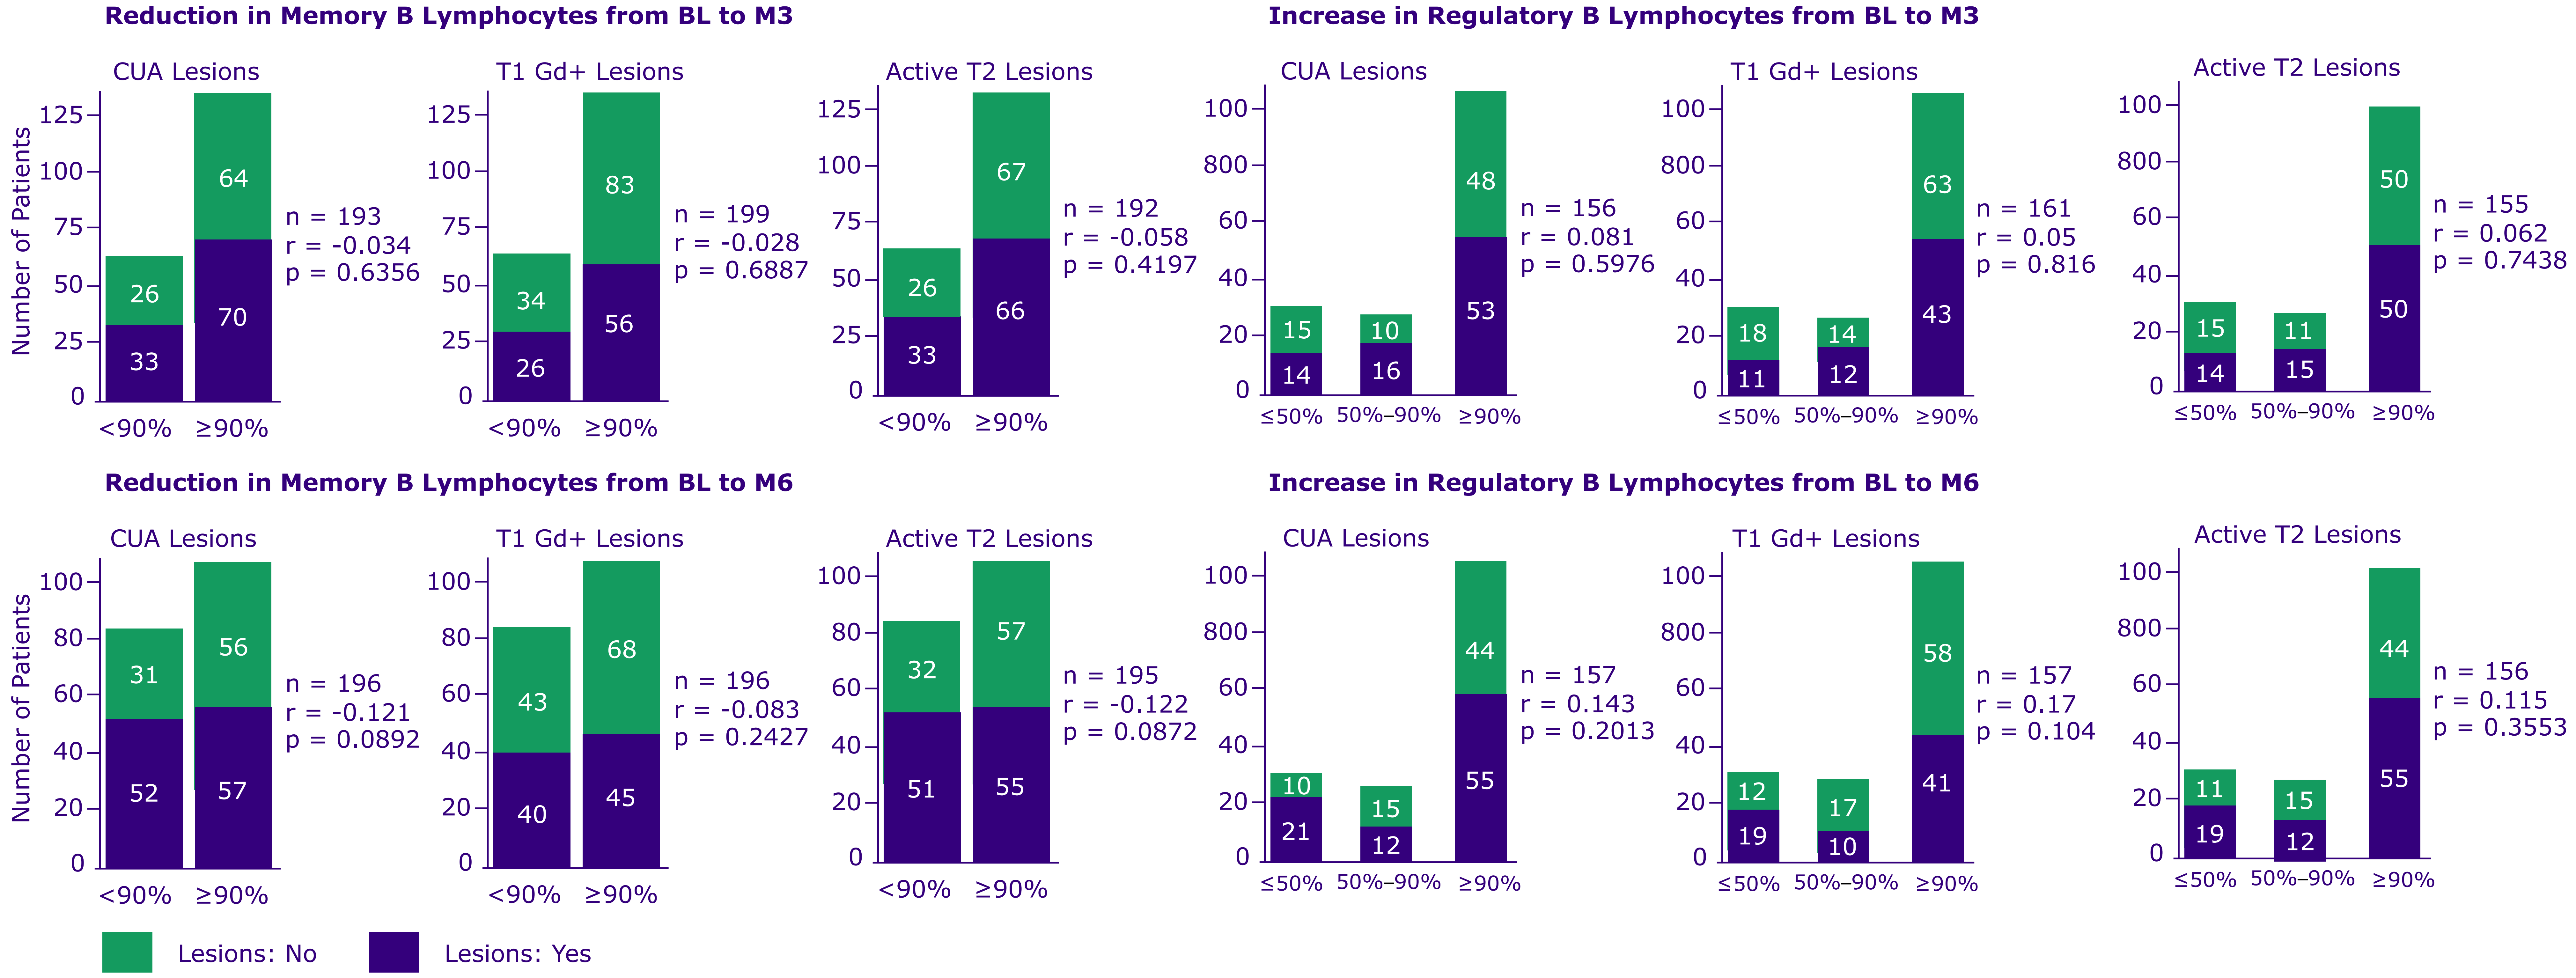
Reduction in Memory B Lymphocytes from BL to M3
CUA Lesions
Number of Patients
125
100
75
50
25
0
26
33
64
70
<90%
≥90%
n = 193
r = -0.034
p = 0.6356
T1 Gd+ Lesions
34
26
83
56
n = 199
r = -0.028
p = 0.6887
Active T2 Lesions
26
33
67
66
n = 192
r = -0.058
p = 0.4197
Increase in Regulatory B Lymphocytes from BL to M3
CUA Lesions
100
800
60
40
20
0
15
14
10
16
48
53
≤50%
50%–90%
≥90%
n = 156
r = 0.081
p = 0.5976
T1 Gd+ Lesions
18
11
14
12
63
43
n = 161
r = 0.05
p = 0.816
Active T2 Lesions
15
14
11
15
50
50
n = 155
r = 0.062
p = 0.7438
Reduction in Memory B Lymphocytes from BL to M6
CUA Lesions
Number of Patients
100
80
60
40
20
0
31
52
56
57
n = 196
r = -0.121
p = 0.0892
T1 Gd+ Lesions
43
40
68
45
n = 196
r = -0.083
p = 0.2427
Active T2 Lesions
32
51
57
55
n = 195
r = -0.122
p = 0.0872
Increase in Regulatory B Lymphocytes from BL to M6
CUA Lesions
10
21
15
12
44
55
n = 157
r = 0.143
p = 0.2013
T1 Gd+ Lesions
12
19
17
10
58
41
n = 157
r = 0.17
p = 0.104
Active T2 Lesions
11
19
15
12
44
55
n = 156
r = 0.115
p = 0.3553
Lesions: No
Lesions: Yes

Cramer's V is a measure of association between two nominal variables. It ranges from 0 to 1, where 0 indicates no association between the two variables and 1 indicates a perfect association between the two variables. *p* < 0.05 is considered significant.

BL, baseline; **CUA**, combined unique active; **Gd+**, gadolinium enhancing; **M**, month; **MRI**, magnetic resonance imaging; **n**, number of patients with both B-cell and MRI assessments; **r**, Cramer’s V correlation coefficient

For reactive Medical use only

February 2023

Presented at ACTRIMS Forum 2023 | 23–25 February | San Diego, CA, USA

The study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)