This is a reprint from the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS 2022) which was originally published in Amsterdam, The Netherlands; the references to “Merck” or “Merck KGaA” within refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.

P693 For reactive Medical use only October 2022

UNIVERSITÄTSMEDIZIN GÖTTINGEN UMG

Evobrutinib, a Bruton's tyrosine kinase inhibitor, acts on microglia: implications in treatment of progressive mechanisms in multiple sclerosis

Anastasia Geladaris[1,2], Sebastian Torke[4], Philipp Haselmayer[5], Ursula Boschert[7], Wolfgang Brück[1], Martin S. Weber[1,2,3]

[1] Institute of Neuropathology, University Medical Center, Göttingen, Germany, [2] Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Göttingen, Germany, [3] Department of Neurology, University Medical Center, Göttingen, Germany,
[4] Experimental and Clinical Research Center of the Charité, Univeristiy Medical Center Berlin and Max-Delbrück-Center for Molecular Medicine in the Helmholtz Association, Berlin, Germany
[5] Merck KGaA, Darmstadt, Germany, [6]Ares Trading SA, Eysins, Switzerland, an affiliate of Merck KGaA

Background

In multiple sclerosis (MS), persisting disability can derive from acute relapses or alternatively, from slow and steady progression independent of relapse activity, termed chronic progression. Therapeutically controlling progression independent of relapses (PIRA) in MS, remains a major challenge. One promising strategy may be to reduce chronic neuroinflammation by the inhibition of the enzyme Bruton's tyrosine kinase (BTK), which is centrally involved in the activation of both B cells as well as myeloid cells, such as macrophages and microglia.

In the present study, we analysed the potential of the BTK inhibitor evobrutinib as a therapeutic strategy in halting progression in MS.

Methods

Primary micorglia were generated from newborn C57BL/6 mice. Furthermore, C57BL/6 mice were immunized with MOG peptide 35-55. Mice were treated with 10 mg/kg evobrutinib or vehicle control starting at day 40 post immunization. On day 70 cells were isolated and analyzed by flow cytometry. In a second approach mice were immunized with MOG peptide 35-55 and after 11-12 days, immunization draining lymph nodes were isolated and cultivated for 3 days in the presence of anti-IFN-gamma antibody, IL-12 and MOG Peptide 35-55. Subsequently, the pathogenic T cells were purified by the magnetic-bead associated removal of B cells and intraperitoneally injected into recipient mice. Recipients received evobrutinib or vehicle control starting 3 days prior to transfer. Microglial activation/modulation was assessed by ELISA and flow cytometry.

Results

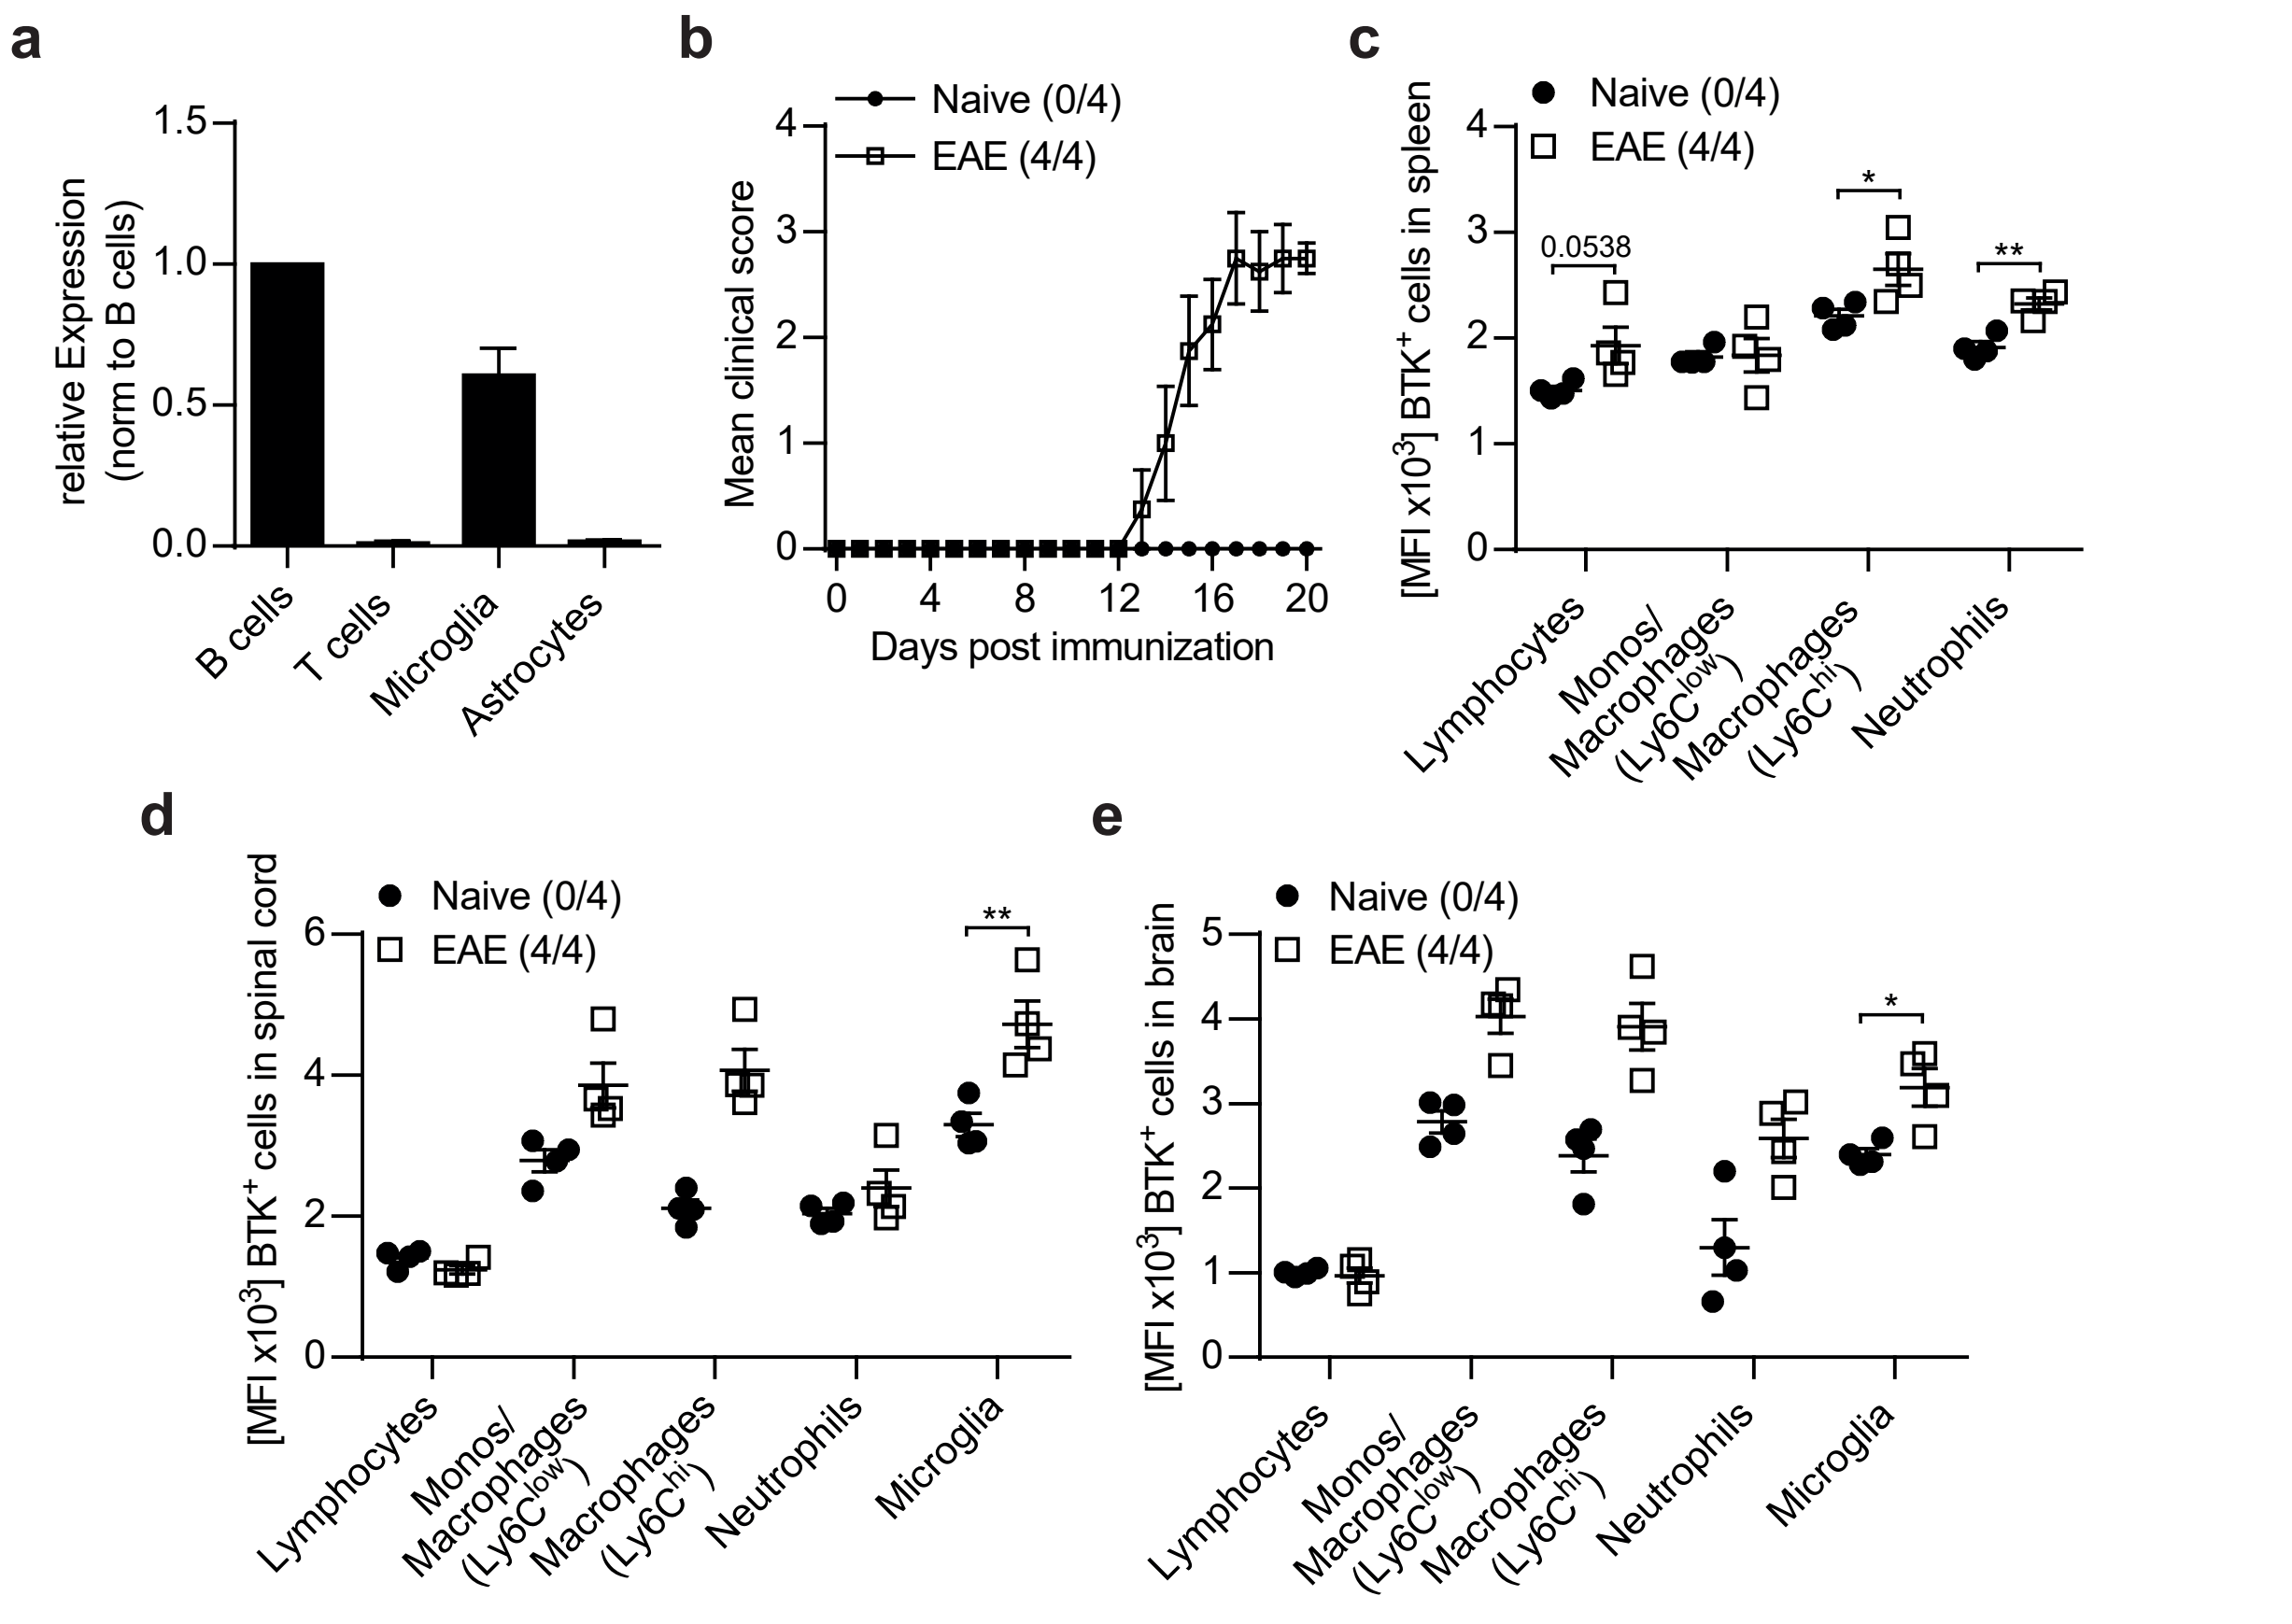
a
relative Expression (norm to B cells)
B cells
T cells
Microglia
Astrocytes
b
Naive (0/4)
EAE (4/4)
Mean clinical score
Days post immunization
c
[MFI x10³] BTK⁺ cells in spleen
0.0538
Lymphocytes
Monos/ Macrophages (Ly6Clow)
Macrophages (Ly6Chi)
Neutrophils
d
[MFI x10³] BTK⁺ cells in spinal cord
Microglia
e
[MFI x10³] BTK⁺ cells in brain

Figure 1: BTK is expressed in microglia but not astrocytes and upregulated under inflammation. a) Primary cell cultures were harvested and lysed for RNA extraction; relative expression normalized to GapDH ± SEM **b-f)** Mice were left untreated or immunized with MOG_{35-55}. On day 20, cells were isolated and stained for flow cytometry, **b)** Mean clinical score ± SEM **c-e)** Mean fluorescence intensity ± SEM; n=4; t-test *p<0.05

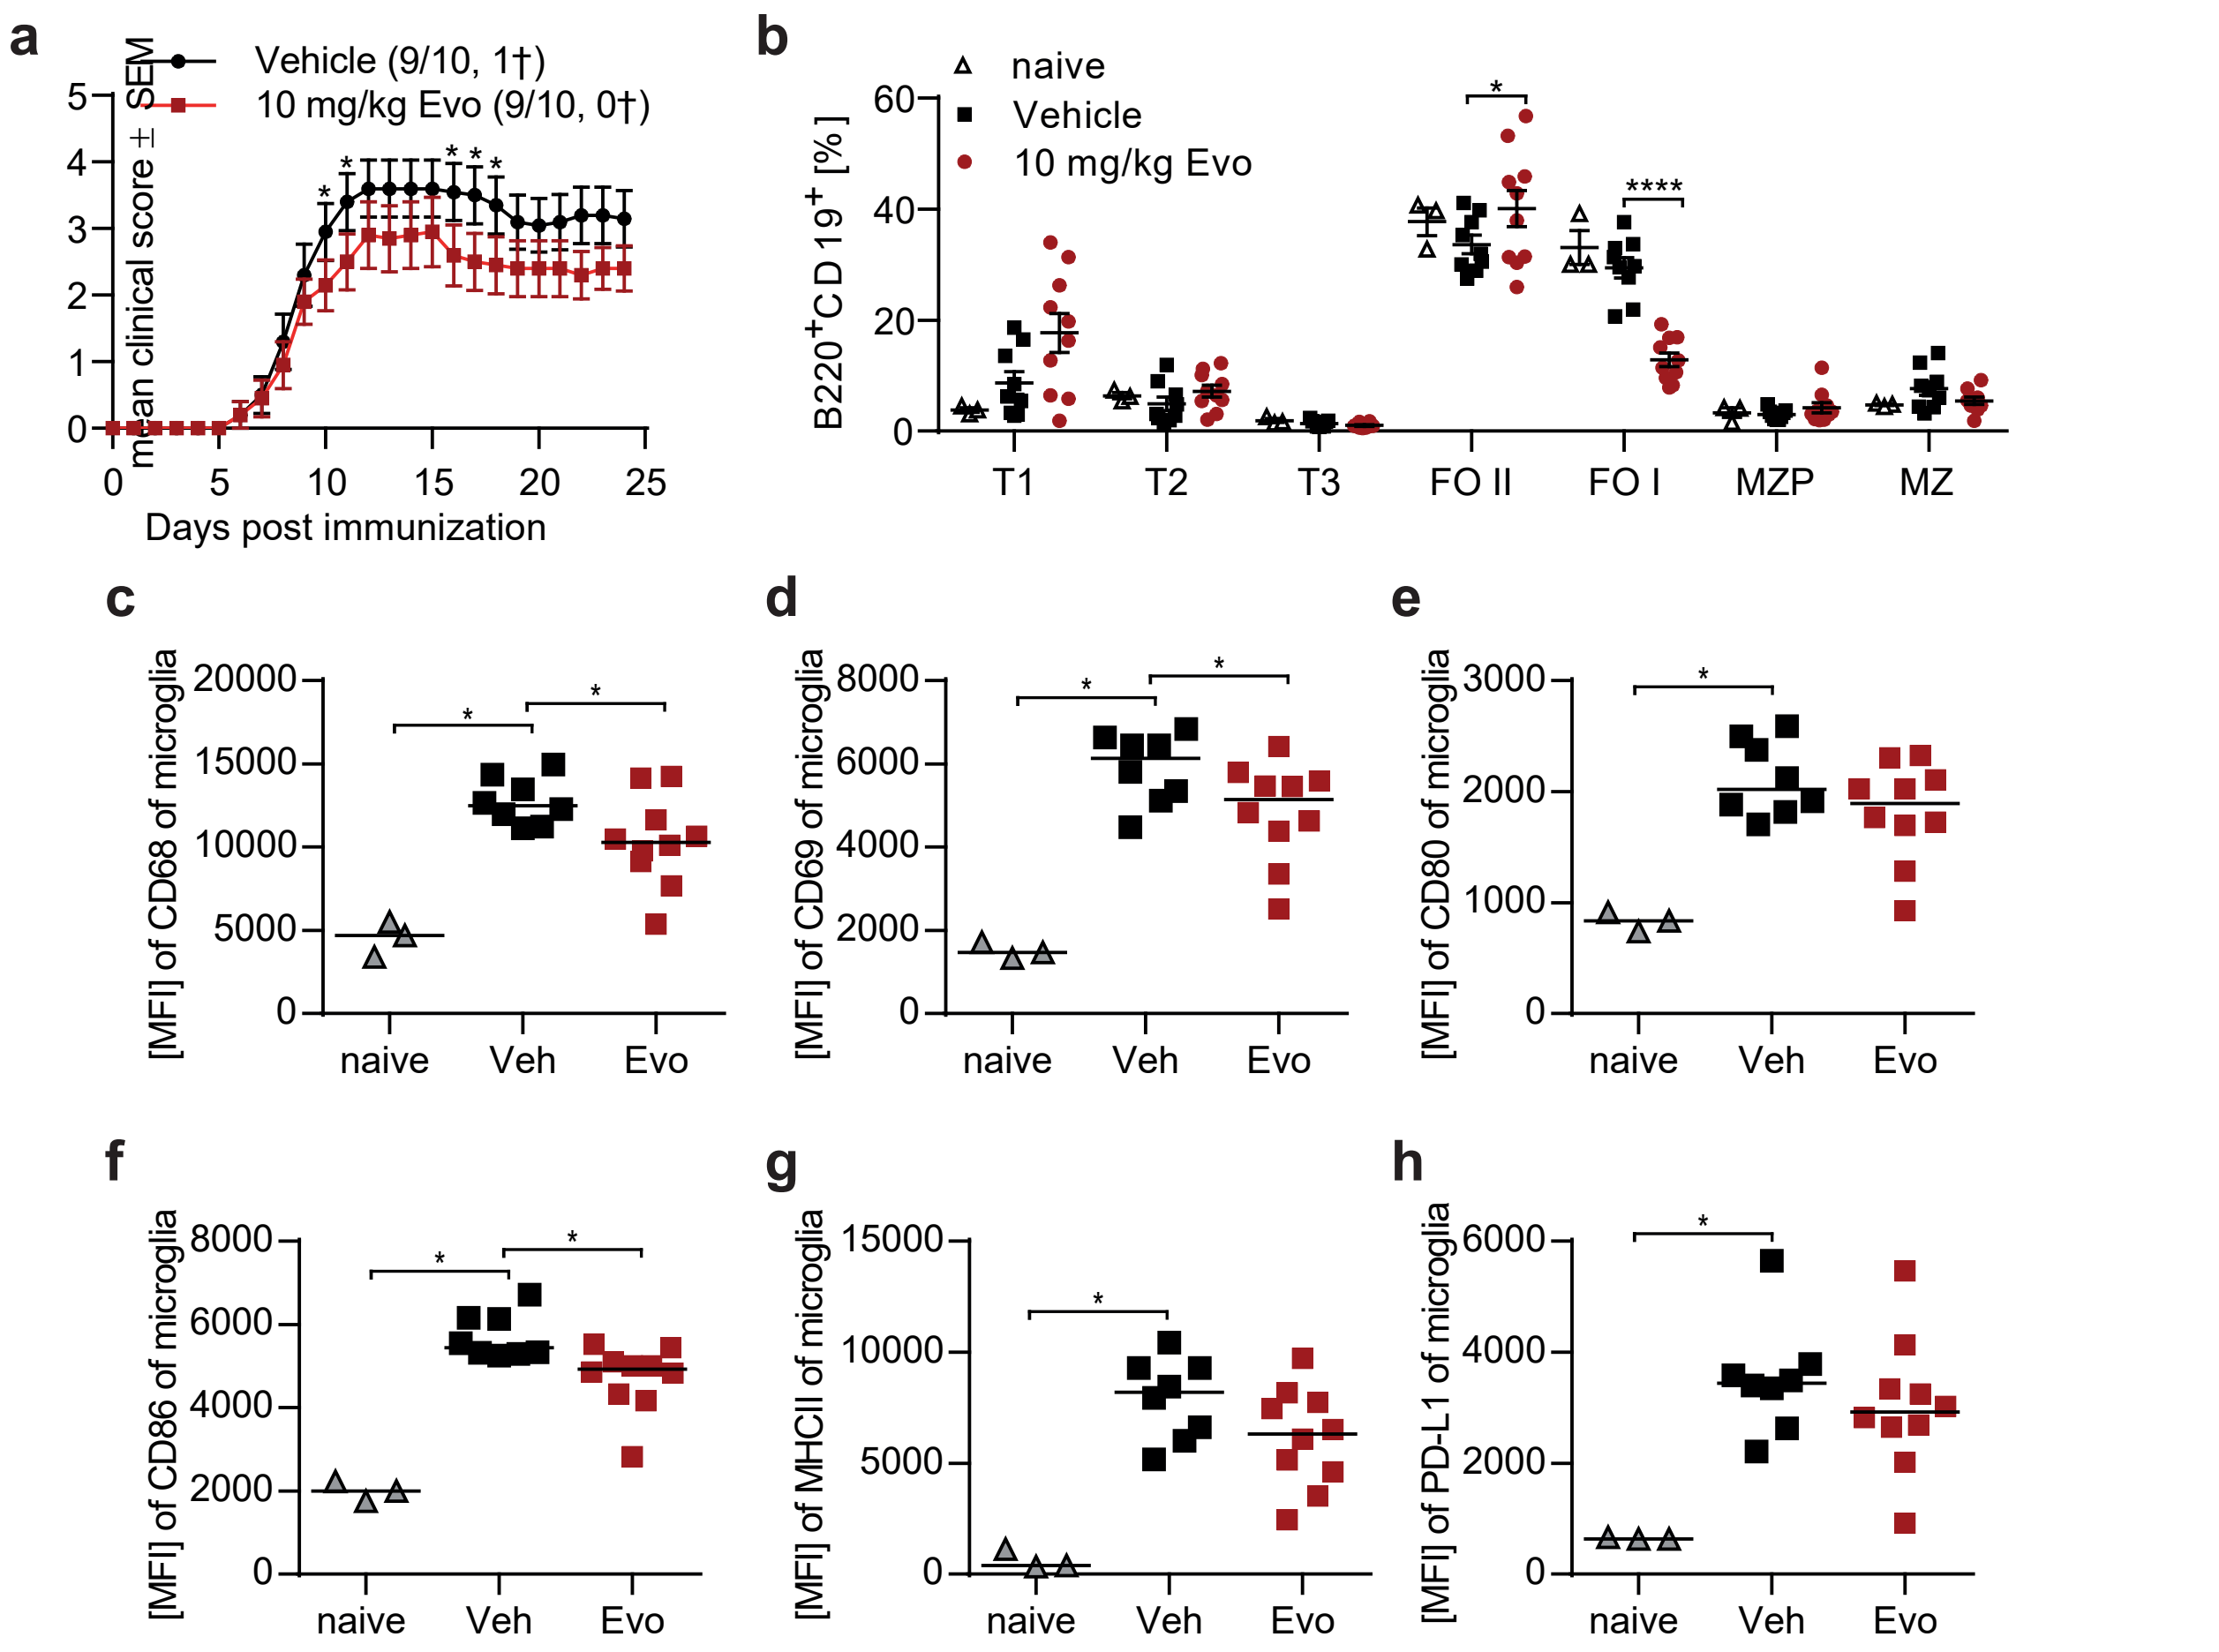
a
Vehicle (9/10, 1†)
10 mg/kg Evo (9/10, 0†)
mean clinical score ± SEM
Days post immunization
b
naive
Vehicle
10 mg/kg Evo
B220⁺CD19⁺ [%]
T1
T2
T3
FO II
FO I
MZP
MZ
c
[MFI] of CD68 of microglia
d
[MFI] of CD69 of microglia
e
[MFI] of CD80 of microglia
f
[MFI] of CD86 of microglia
g
[MFI] of MHCII of microglia
h
[MFI] of PD-L1 of microglia
naive
Veh
Evo

Figure 3: Adoptive transfer of pathogenic T cells lead to a strong microglia activation, a process that can be dampened by evobrutinib. C57BL/6 mice were immunized with 200 µg MOG_{35-55}. After 11-12 days, the inguinal lymph nodes were isolated and cultivated for 3 days at the density of 2-2.5x10^6 cells in the presence of 20 µg/ml anti-IFNγ, rIL-12 and 25 µg/ml MOG_{35-55}. Subsequently, T cells were purified by a magnetic-bead associated removal of B cells. Recipient mice, pre-treated for 3 days with evobrutinib or vehicle control, received 1.83x106 cells intraperitoneally. **a)** Mean clinical score ± SEM **b)** B cell maturation **c-h)** Microglia activation in the spinal cord, mean fluorescence intensity shown an median; n=9-10, unpaired t test; *p<0.05, **p<0.01.

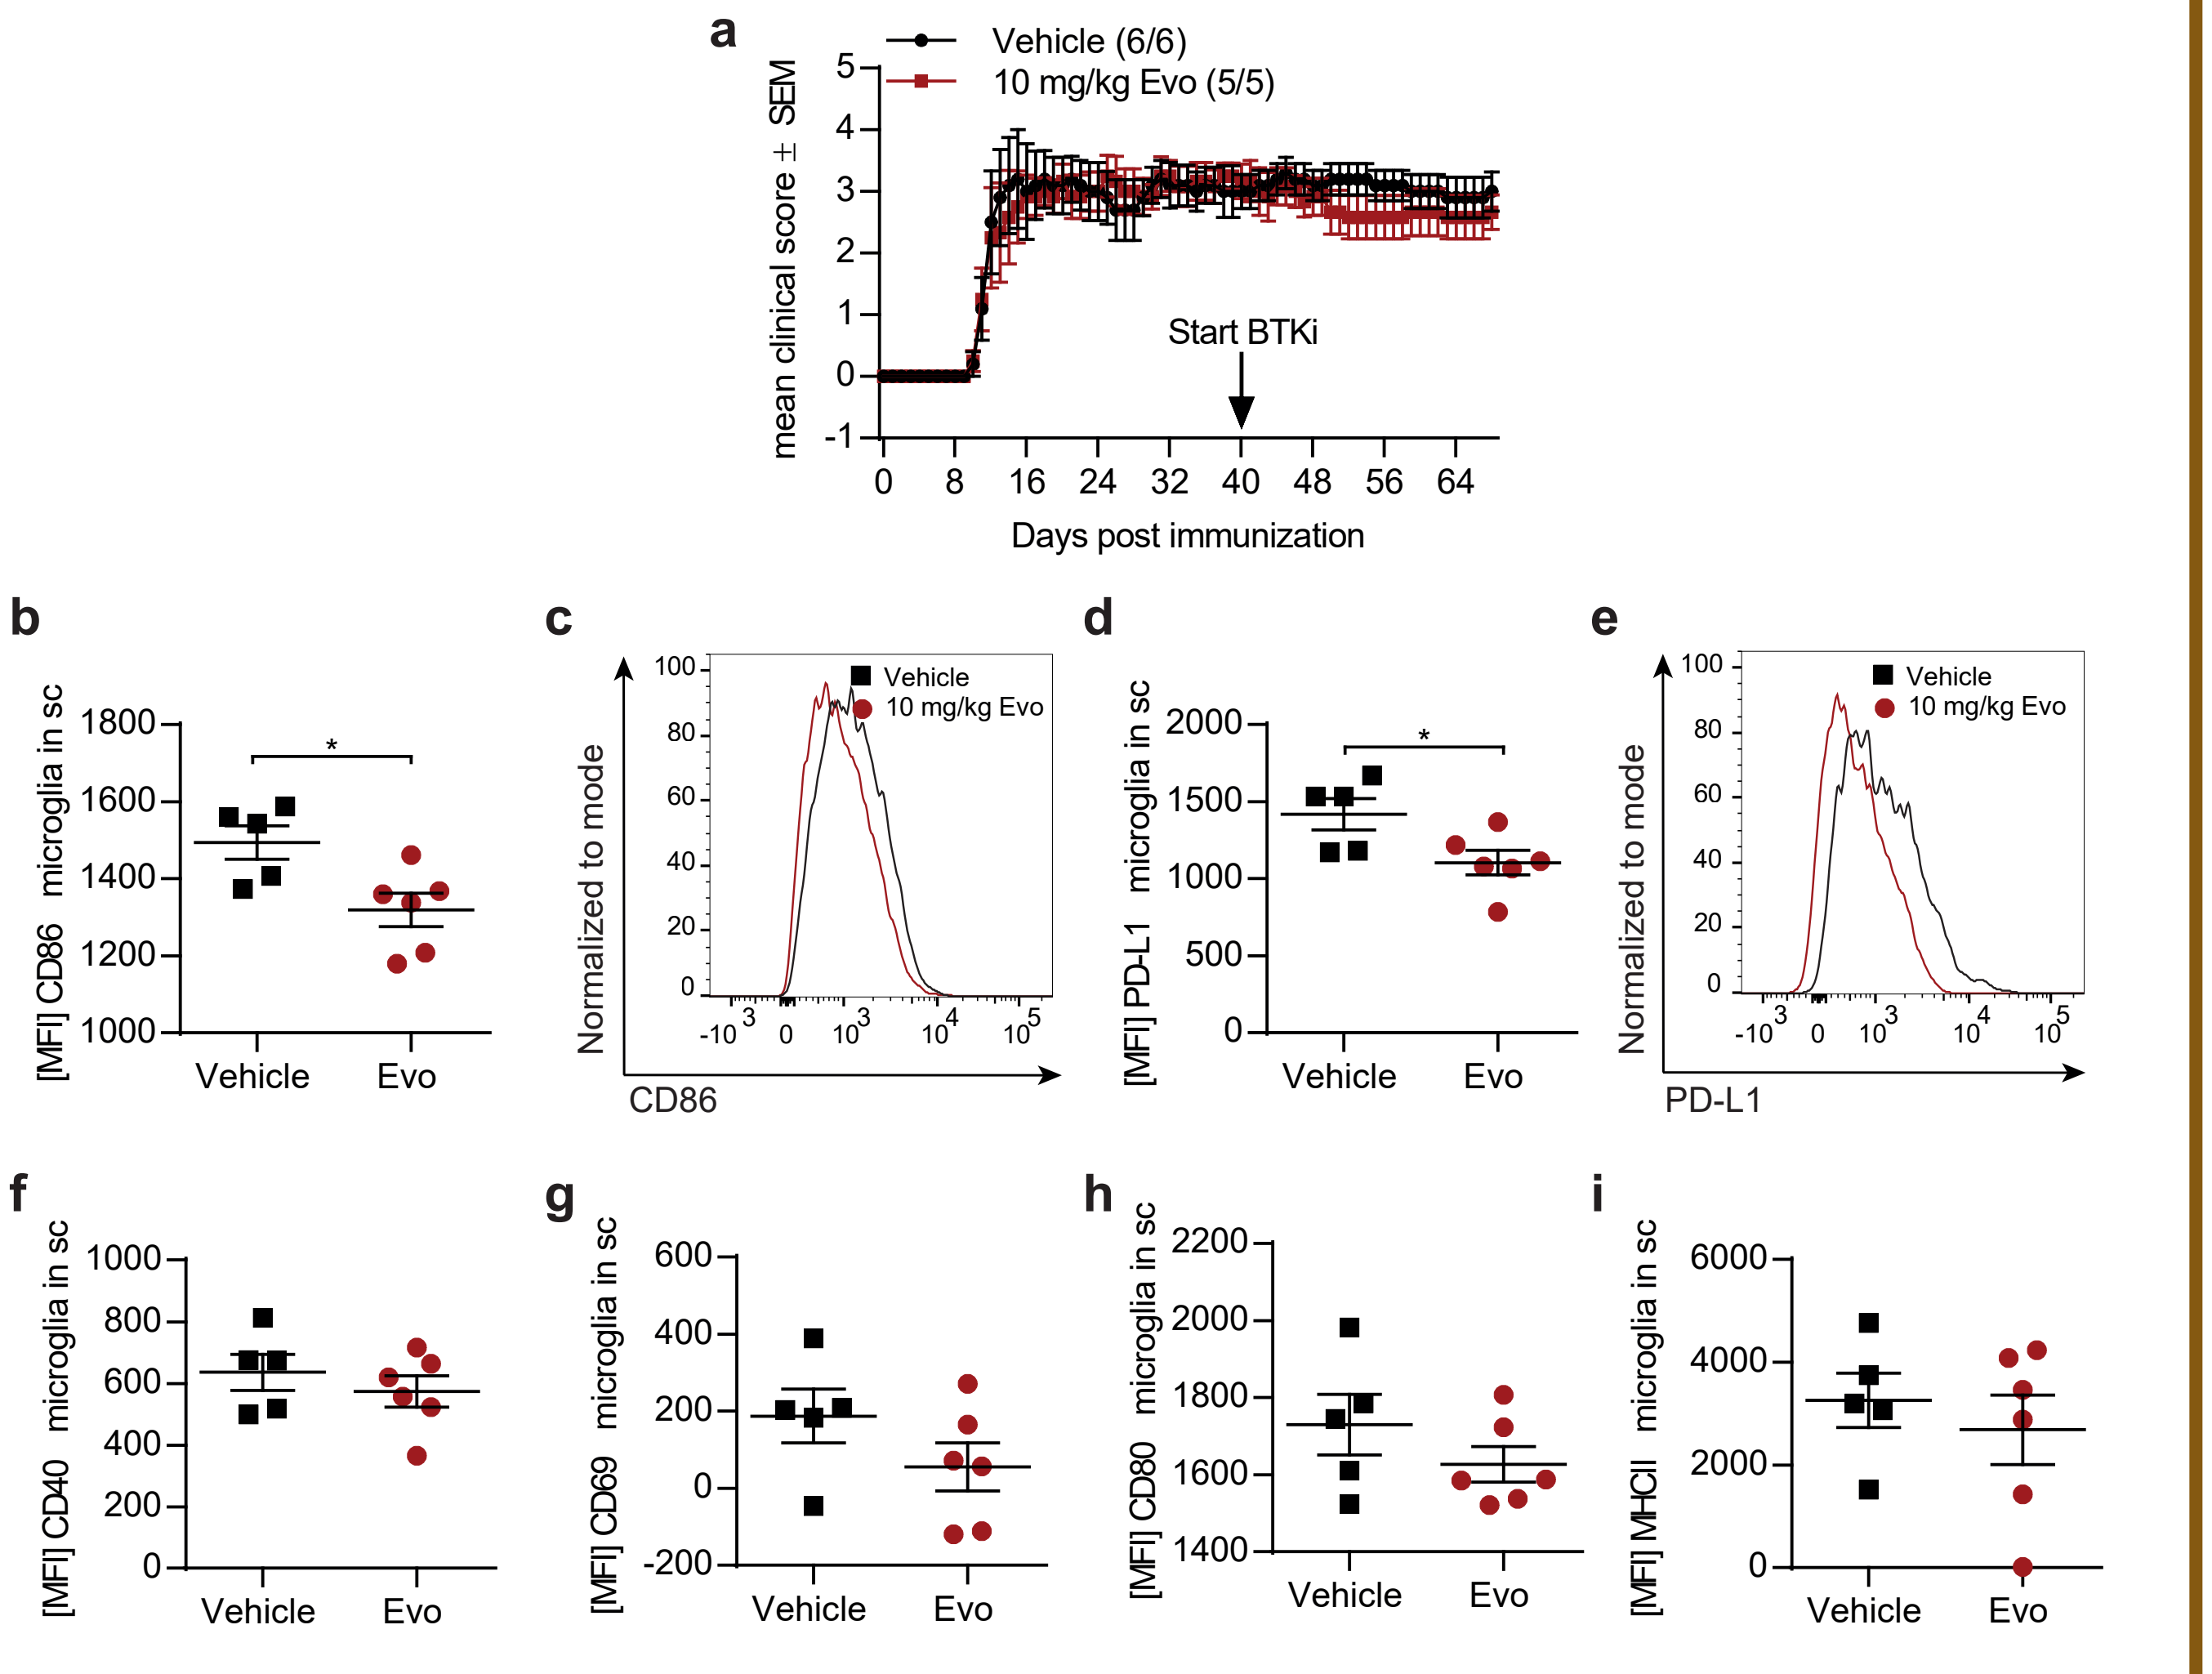
a
Vehicle (6/6)
10 mg/kg Evo (5/5)
mean clinical score ± SEM
Start BTKi
Days post immunization
b
[MFI] CD86 microglia in sc
c
Vehicle
10 mg/kg Evo
Normalized to mode
CD86
d
[MFI] PD-L1 microglia in sc
e
PD-L1
f
[MFI] CD40 microglia in sc
g
[MFI] CD69 microglia in sc
h
[MFI] CD80 microglia in sc
i
[MFI] MHCII microglia in sc
Vehicle
Evo

Figure 2: Evobrutinib alter the microglia phenotype in chronic EAE. C57BL/6 mice were immunized with 75 µg MOG_{35-55} peptide. Mice were treated with evobrutinib or vehicle starting at day 40 post immunization. On day 70 cells were isolated from the spinal cord and analyzed by flow cytometry. **a)** Mean clinical score ± SEM. **b,d,f-i)-** Microglia activation in the spinal cord; mean fluorescence intensity ± SEM; n=5-6. t test; *p<0.05, **p<0.01. **c, e)** Representative histograms.

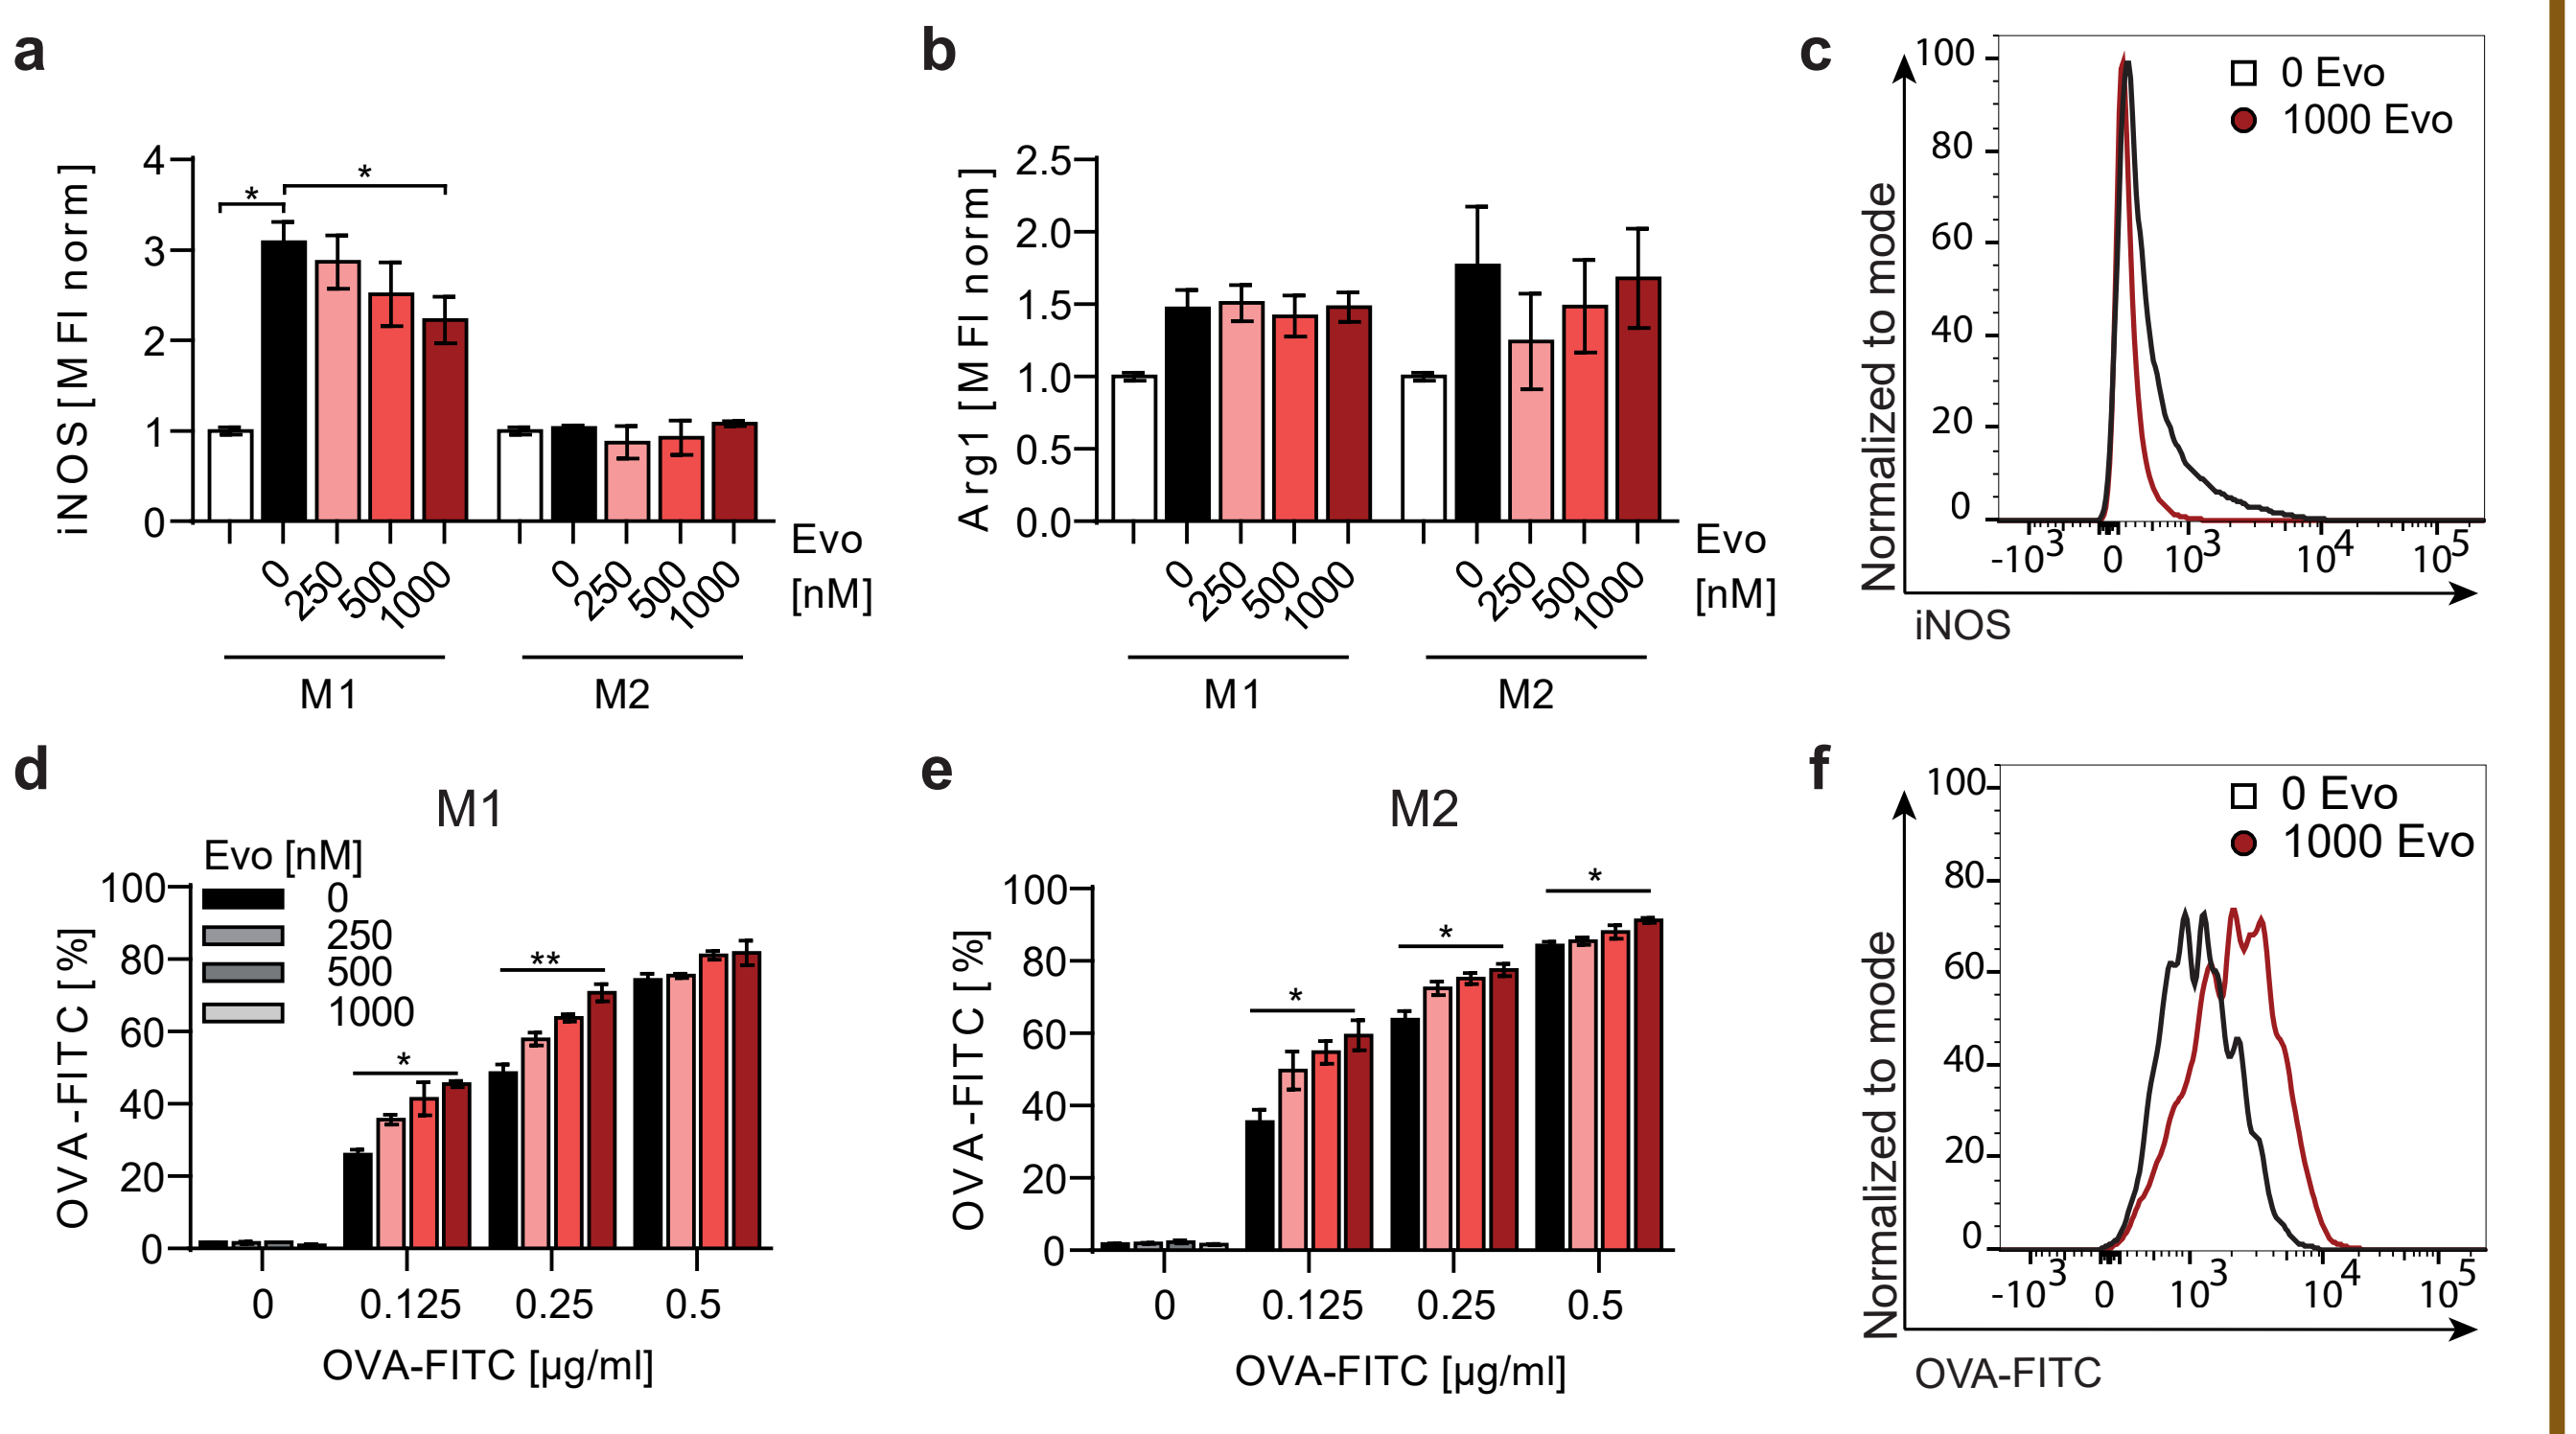
a
iNOS [MFI norm]
Evo [nM]
M1
M2
b
Arg1 [MFI norm]
c
0 Evo
1000 Evo
Normalized to mode
iNOS
d
M1
Evo [nM]
OVA-FITC [%]
OVA-FITC [µg/ml]
e
M2
f
OVA-FITC

Figure 4: Evobrutinib specifically inhibits LPS-induced microglial M1 differentiation and promotes phagocytosis capacity. a-c) Primary microglia were pre-treated with the indicated evobrutinib concentrations for 30 minutes prior to differentiation into M1 (LPS) or M2 (rIL-4/10/13) phenotype. After 48h the cells were harvested and stained using the BD PhosFlow protocol. **a, b)** Mean fluorescence intensity ± SEM; n=4-8, pooled from 2-3 independent experiments; Kruskal-Wallis with Dunn's post hoc test; *p<0.05. d-f) Primary microglia were differentiated into M1 or M2 microglia. 30 min prior to phagocytosis assay the cells were treated with indicated concentrations of evobrutinib. Thereafter, the cells were incubated for 2h with indicated concentrations of OVA-FITC, harvested and stained for flow cytometry analysis. **d, e)** Frequency of OVA-FITC+ microglia cells ± SEM; n=3, Kruskal-Wallis with Dunn's post hoc test; *p<0.05.

Conclusion

BTK inhibition by evobrutinib downregulates inflammatory properties of microglia. These data highlight the therapeutic potential of BTK inhibition with evobrutinib in ameliorating underlying processes associated with chronic progression in MS.

Disclosure: A.G has nothing to disclose. S.T. recieves an intramural grant of the University Medical Center, Göttingen ("Startförderung") and travel support from EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, an affiliate of Merck KGaA. M.S.W. is serving as an editor for PLoS One. He receives research support from the National Multiple Sclerosis Society (NMSS; PP 1660), the Deutsche Forschungsgemeinschaft (DFG; WE 3547/4-1), from Novartis, EMD Merck Serono, TEVA, Biogen-Idec, Roche and the ProFutura Programm of the Universitätsmedizin Göttingen. The authors declare no conflicts of interest.

Copyright: Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.