“This reprint might contain references to “Merck” or “Merck KGaA”, which refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.
There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.”

Development and Interrelation of Spatiotemporal Patterns of Brain Atrophy and Lesions During 5 Years' Treatment with Subcutaneous Interferon Beta-1a in Patients with a First Clinical Demyelinating Event in the REFLEX/ION Study

G. Gentile[1,2], R.M. Mattiesing[3], I. Brouwer[3], D. Jack[4], F. Barkhof[3,5], N. De Stefano[1], B.M.J. Uitdehaag[6], H. Vrenken[3], M. Battaglini[1,2]

[1]Department of Medicine, Surgery and Neuroscience, University of Siena, Siena, Italy; [2]SIENA imaging SRL, Siena, Italy; [3]Department of Radiology and Nuclear Medicine, Amsterdam UMC, Location VUmc, Amsterdam, The Netherlands; [4]Merck Serono Ltd, Feltham, UK (an affiliate of Merck KGaA); [5]Institutes of Neurology and Healthcare Engineering, UCL London, London, UK; [6]Department of Neurology, Amsterdam UMC, Location VUmc, Amsterdam, The Netherlands

GET POSTER PDF
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

CONCLUSIONS

Treatment modulates the concomitant spatial relation between inflammation and neurodegeneration.

Pseudoatrophy is more prominent in patients with signs of active inflammation processes.

NOTE:
Refer to Poster P430 for Further Analyses

INTRODUCTION

- Inflammation (lesions) and neurodegeneration (atrophy) are present early in the disease course of multiple sclerosis (MS).
- Whether these two processes result from independent pathological mechanisms remains to be elucidated.
- In the 5-year REFLEX/ION study, treatment with subcutaneous interferon β-1a was associated with overall magnetic resonance imaging (MRI) reduced activity in early MS.[1,2] However, how therapy influenced the inter-relation between changes in white matter lesions and brain atrophy was not investigated.

OBJECTIVES

To evaluate treatment influence on the concomitant regional relationship between inflammation and neurodegeneration in the early phase of MS in the REFLEX/ION study.

METHODS

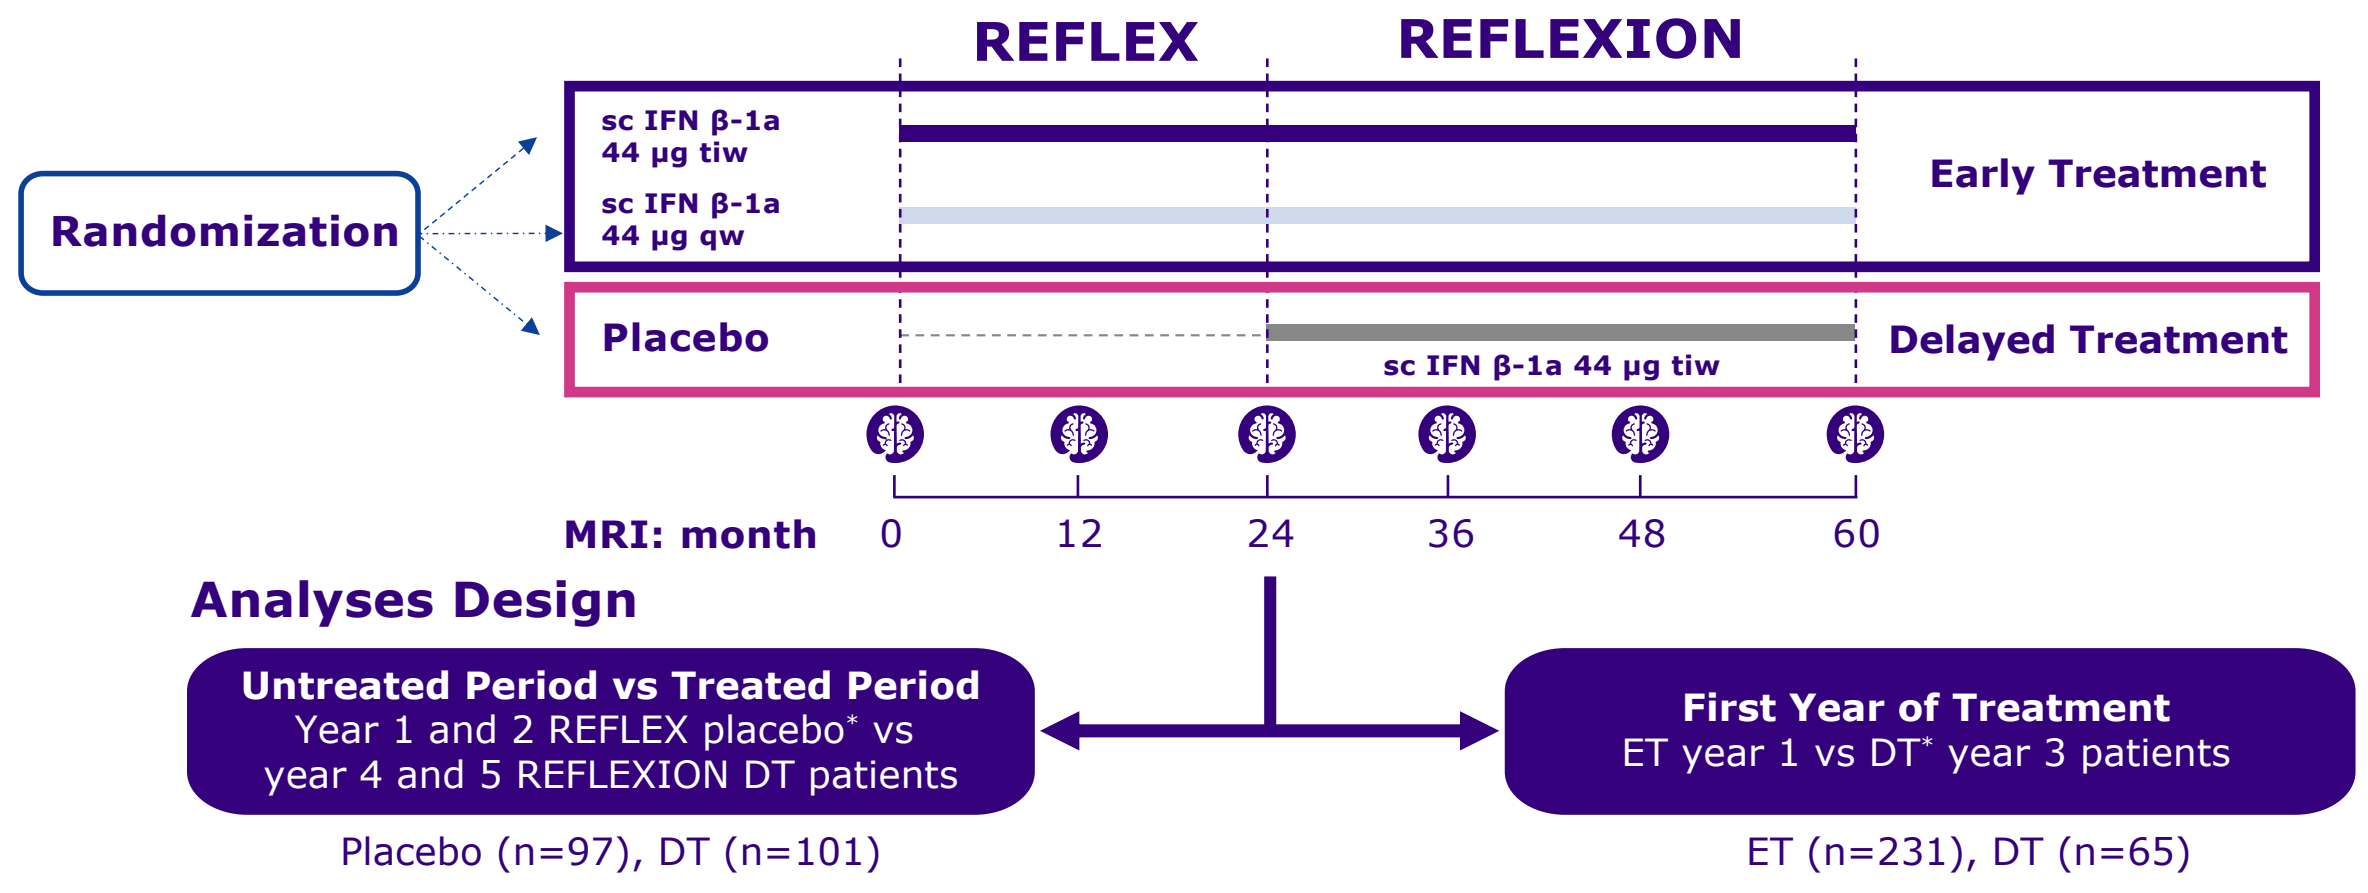
REFLEX
REFLEXION
Randomization
sc IFN β-1a 44 µg tiw
sc IFN β-1a 44 µg qw
Early Treatment
Placebo
sc IFN β-1a 44 µg tiw
Delayed Treatment
MRI: month
0
12
24
36
48
60
Analyses Design
Untreated Period vs Treated Period
Year 1 and 2 REFLEX placebo* vs year 4 and 5 REFLEXION DT patients
Placebo (n=97), DT (n=101)
First Year of Treatment
ET year 1 vs DT* year 3 patients
ET (n=231), DT (n=65)

*Patients who converted to MS during the first 2 years of the study were excluded (as per the study protocol, they received treatment upon conversion, which in their cases was earlier than month 24).

DT, delayed treatment; **ET**, early treatment; **MRI**, magnetic resonance imaging; **sc IFN β-1a 44 µg tiw/qw**, treatment with subcutaneous interferon β-1a three times/once per week.

Longitudinal Brain Activity Quantification

- Yearly total lesion volume change (TLVC) was determined by an automated method based on subtraction imaging.
- Yearly percentage brain volume change (PBVC) and percentage ventricular volume change (PVVC) obtained by FMRIB Software Library-Structural Image Evaluation using Normalization of Atrophy (FSL-SIENA) were used as measures of whole-brain and central atrophy, respectively.

Statistical Analyses

- Linear mixed models and the FSL Randomize tool were used for performing whole-brain and voxel-wise analyses, respectively.
- Statistical models employed PBVC/PVVC and yearly cerebral edge displacement maps as dependent variables. TLVC was the independent variable. All the analyses were corrected for age, sex and site. Volumes of significant voxels (V, mm^3) are reported.

RESULTS

Untreated vs Treated Period Whole-brain Analyses

- *REFLEX placebo patients (months 0–24):* A significant positive relation between PBVC and TLVC (B=0.072, SE=0.029, p=0.013), and a significant negative relation between PVVC and TLVC (B=-0.917, SE=0.306, p=0.003) were found.
- *REFLEXION delayed treatment (DT) patients (months 36–60):* No significant results.

Overall, a similar relation between white matter (WM) lesion activity and brain atrophy was observed through the untreated and treated periods.

Untreated vs Treated Period Voxel-wise Analyses

- *REFLEX placebo patients (months 0–24):* In Year 1, lower TLVC was associated with concomitant faster periventricular, parietal, infratentorial, and temporal lobe atrophy (volume of significant voxels [V]=16,272). In Year 2, higher TLVC was associated with concomitant faster periventricular atrophy (V=11,712).
- *REFLEXION DT patients (months 36–60):* In Years 4 and 5, lower TLVC was associated with concomitant faster atrophy in the frontal, temporal, and periventricular areas (Year 4, V=1784; Year 5, V=8000).

WM lesion activity and brain atrophy showed a different relation across the untreated and treated periods.

Figure 1. Voxel-wise Analyses Within the First Two Years of the Untreated Placebo Period

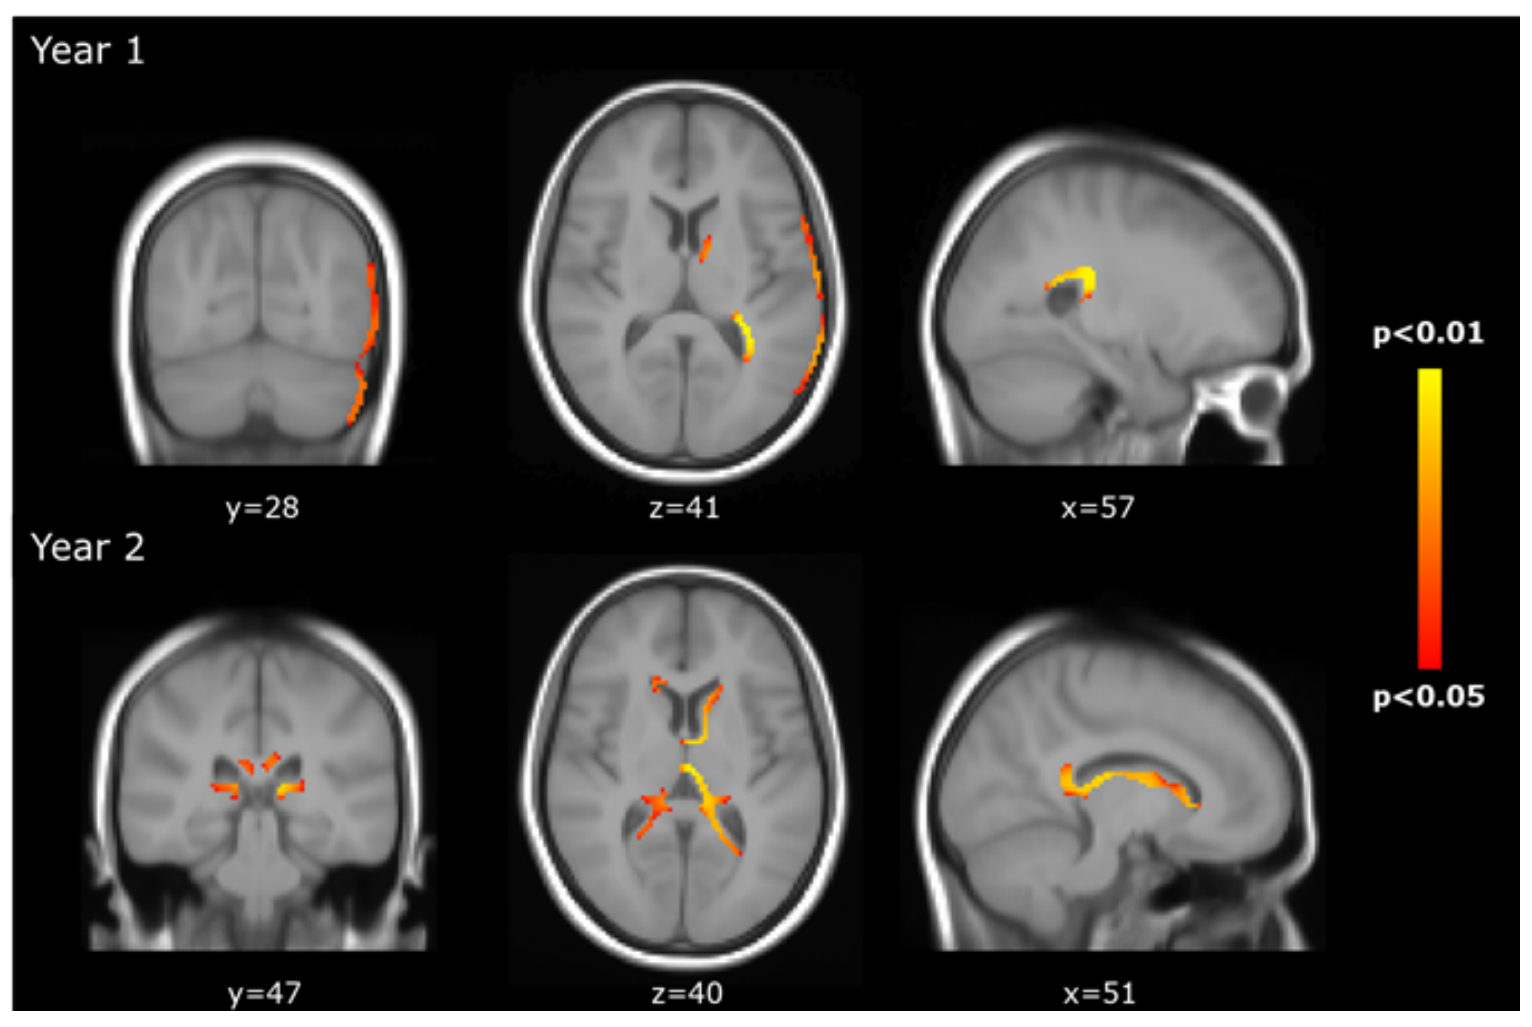
Year 1
y=28
z=41
x=57
Year 2
y=47
z=40
x=51
p<0.01
p<0.05

Yellow-orange show voxels of significant regions where lower TLVC was related to faster atrophy (top row) and where higher TLVC was related to faster atrophy (bottom row).

TLVC, total lesion volume change.

First Year of Treatment Early Treated (months 0–12) and Delay Treated (months 24–36)

- *Whole-brain analyses:* Faster whole (B=0.081, SE=0.027, p=0.003) and central (B=-1.08, SE=0.284, p<0.001) atrophy was associated with lower TLVC in patients who received early treatment. In DT patients, faster whole (B=-0.141, SE=0.065, p=0.032) and central atrophy (B=3.41, SE=0.677, p<0.001) was associated with higher TLVC.
- *Voxel-wise analyses:* In the first year of treatment, faster periventricular and frontal lobe atrophy was associated with lower TLVC (V=17,536) in ET patients. No significant relation was found for DT patients in Year 3.

Pseudoatrophy effect was present in the first year of therapy for ET patients only. No pseudoatrophy was detected in DT patients, probably related to a lack of WM lesion activity.

Figure 2. Voxel-wise Analyses Within the First Year of Treatment of ET Patients (Year 1)

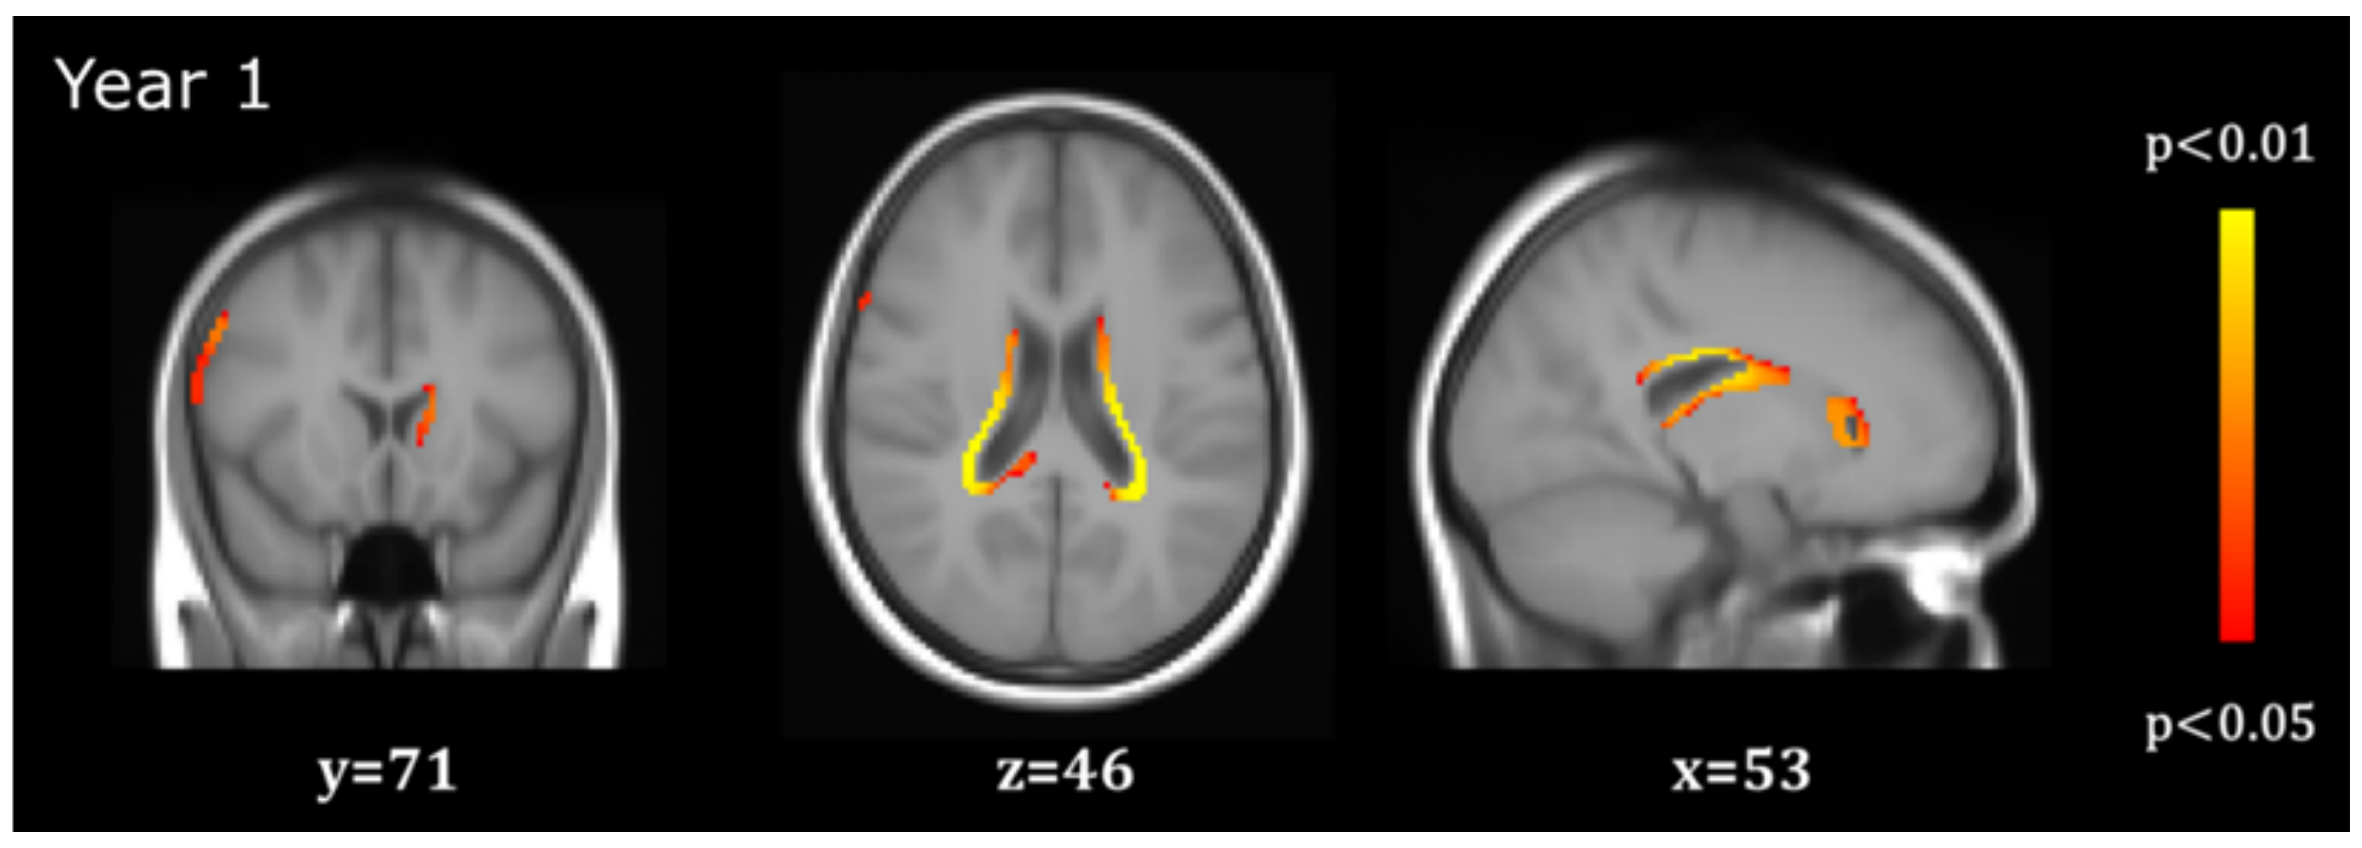
Year 1
y=71
z=46
x=53
p<0.01
p<0.05

Yellow-orange show voxels of significant regions where lower TLVC was related to faster atrophy.

ET, early treated; **TLVC**, total lesion volume change.

REFERENCES
1. Comi G, et al. *Lancet Neurol.* 2012; 11(1):33–41. 2. Comi G, et al. *J Neurol Neurosurg Psychiatry*. 2017;88:285–294.

Disclosures: RMM has received research support from Merck. **IB** has received research support from Merck, Novartis, Teva, and the Dutch MS Research Foundation. **DJ** is an employee of Merck Serono Ltd, Feltham, UK (an affiliate of Merck KGaA). **FB** is supported by the NIHR Biomedical Research Centre at UCLH and is a consultant to Biogen, Combinostics, IXICO, Merck, and Roche. **NDS** is a consultant for Biogen, Merck, Novartis, Sanofi-Genzyme, Roche, and Teva; has grants or grants pending from FISM and Novartis, is on the speakers' bureaus of Biogen, Merck, Novartis, Roche, Sanofi-Genzyme, and Teva; and has received travel funds from Merck, Novartis, Roche, Sanofi-Genzyme, and Teva. **BMJU** has received consultancy fees from Biogen, Merck, Novartis, Roche, Sanofi-Genzyme, and Teva. **HV** has received research support from Merck, Novartis, Pfizer, and Teva, consulting fees from Merck and speaker honoraria from Novartis; all funds were paid to his institution. **GG** and **MB** have nothing to disclose.

Editorial support was provided by Clara Garcia-Gorro of inScience Communications, Springer Healthcare Ltd, UK, and was funded by Merck Healthcare KGaA, Darmstadt, Germany.

ClinicalTrials.gov identifier: NCT00813709.

Presented at ECTRIMS 2021 Virtual Congress | 13–15 October

This study was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945)

FFOR REACTIVE MEDICAL USE ONLY