“This reprint might contain references to “Merck” or “Merck KGaA”, which refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.

There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.”

P041

Suboptimal Treatment Outcomes in Employees with Multiple Sclerosis Initiating Disease-Modifying Drugs

C.M. Hersh,[1] R.A. Brook,[2] N.J. Rohrbacker,[3] I.A. Beren,[3] L. Lebson,[4] C. Henke,[5] A.L. Phillips[4]

[1]Cleveland Clinic, Lou Ruvo Center for Brain Health, Las Vegas, NV, US; [2]Better Health Worldwide, Inc, Newfoundland, NJ, US; [3]The HCMS Group, LLC, Cheyenne, WY, US; [4]EMD Serono, Inc., Rockland, MA, US; [5]Biopharma Business of Merck KGaA, Darmstadt, Germany

BACKGROUND

- Initiation of multiple sclerosis (MS) disease modifying drugs (DMDs) is associated with significant medical and indirect savings for employees with MS[1,2]
- DMD adherence is associated with a significantly lower rate of severe relapse and lower total costs over 2 years among employees with MS in the US[3,4]
- Some employees with MS may have challenges with their DMD treatment
 - Employees with MS may experience suboptimal treatment outcomes such as:
 - Continuing to relapse despite DMD treatment
 - Becoming non-adherent to their DMDs
 - Discontinuing DMD treatment with or without switching to an alternate therapy
- Little is known about the prevalence of employees with MS with suboptimal DMD treatment outcomes and the demographic and clinical characteristics of employees who have suboptimal DMD treatment outcomes
- A better understanding of suboptimal DMD treatment outcomes in employees with MS is an important aspect of optimizing patient care

OBJECTIVES

- To evaluate suboptimal treatment outcomes in employees with MS initiating self-injectable or oral DMD and to characterize employees with and without suboptimal treatment outcomes

METHODS

Data Source

- Employees with MS were from the Human Capital Management Services (HCMS) database
 - HCMS is a health benefits consultant for a number of large US employers with diverse salary, job type, employee age, sex, and location demographics
 - The HCMS Research Reference Database (RRDb) currently includes approximately 3.8 million employees (and insured spouses/dependents) who were employed at some point between January 1, 2001 and June 30, 2019

Patient Population

- Eligibility criteria were: employees aged 18–64 years with at least two medical claims with a diagnosis of MS over a period of longer than 30 days (International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM] code: 340.xx and ICD-10-CM code G35), and at least one prescription for a DMD after MS diagnosis
- The date of the first DMD prescription was defined as the index date
- Employees included in the study had either a self-injectable (i.e., subcutaneous or intramuscular interferon [IFN] β-1a, IFN β-1b, PEG-IFN β-1a, or glatiramer acetate) or an oral (i.e., dimethyl fumarate, fingolimod, or teriflunomide) DMD as their index DMD. Employees were excluded if their index DMD was an infusion DMD (i.e., alemtuzumab, mitoxantrone, ocrelizumab, or natalizumab) due to the challenges associated with accurately determining discontinuation with these DMDs
- Employees were required to have continuous eligibility for at least 6 months before the index date (i.e., eligible to receive health care benefits during the 6-month time period prior to initiating their first DMD) and 12 months after the index date

Study Outcomes

- Employees with MS with suboptimal DMD treatment outcomes (i.e., employees with MS who were not adherent to their DMD, discontinued DMD treatment altogether, switched DMDs, or continued to relapse despite DMD treatment) were compared to employees with optimal DMD treatment outcomes (i.e., employees who had none of the suboptimal DMD treatment outcome indicators)
- Relapse was defined as ≥1 MS-related hospitalization, emergency room visit, or outpatient visit with a corticosteroid claim ±7 days of the visit[5,6]
- Nonadherence was defined as the proportion of days covered (PDC) <80%, discontinuation was defined as a treatment gap >60 days, and switching was defined as initiating another DMD
- Demographic characteristics that were evaluated included: age at beginning of index (continuous), age group (categorical), sex, race, census region (categorical – Midwest, Northeast, South, and West), marital status, salary, and full-/part-time status
- Clinical characteristics that were evaluated included:
 - Comorbidities
 - Overall comorbidity as measured by the Charlson Comorbidity Index; and
 - Individual rates of hyperlipidemia, hypertension, gastrointestinal disease, depression, thyroid disease, anxiety, arthritis (rheumatoid arthritis or osteoarthritis), chronic lung disease, diabetes (type I and type II), and alcohol abuse. These comorbidities were selected as they are among the most common in patients with MS based on a review of the published literature[7]
 - Tobacco use
 - MS disease severity
 - Relapse rate during index; and
 - Baseline magnetic resonance imaging (MRI)

Study Analyses

- All study variables were analyzed descriptively
 - Categorical and binary variables were summarized using frequencies and percentages
 - Continuous variables were summarized using means (with confidence intervals), standard deviations, and medians
- Baseline demographic and clinical characteristics were compared between employees with MS and suboptimal versus optimal DMD treatment outcomes

RESULTS

- Of 2173 employees with ≥2 MS diagnoses, 1281 (59.0%) were using a DMD and 488 (22.5%) were using a DMD and had 6 months pre- and 12 months post-index continuous eligibility (**Figure 1**)

Figure 1. Patient Selection from the Human Capital Management Services (HCMS) Research Reference Database (RRDb)

2,543,265 patients in the database from January 1, 2001-December 31, 2018

↓ 2,541,092 without 2 diagnoses of MS at least 30 days apart

2173 with 2 diagnoses of MS at least 30 days apart

↓ 892 not using a DMD

1281 using a DMD (1st day of DMD initiation was index date)

↓ 793 without 6 months pre- and 12 months post-index eligibility

488 with 6 months pre- and 12 months post-index eligibility

Suboptimal Treatment Outcomes

- Half of the employees with MS meeting eligibility criteria (n=247; 50.6%) had suboptimal treatment outcomes (indicators not mutually exclusive; **Figure 2**):
 - 39.5% nonadherence
 - 9.8% discontinuation
 - 10.9% switching
 - 20.7% relapse

Figure 2. Optimal and Suboptimal Treatment Outcomes Among Employees with MS Treated with DMDs

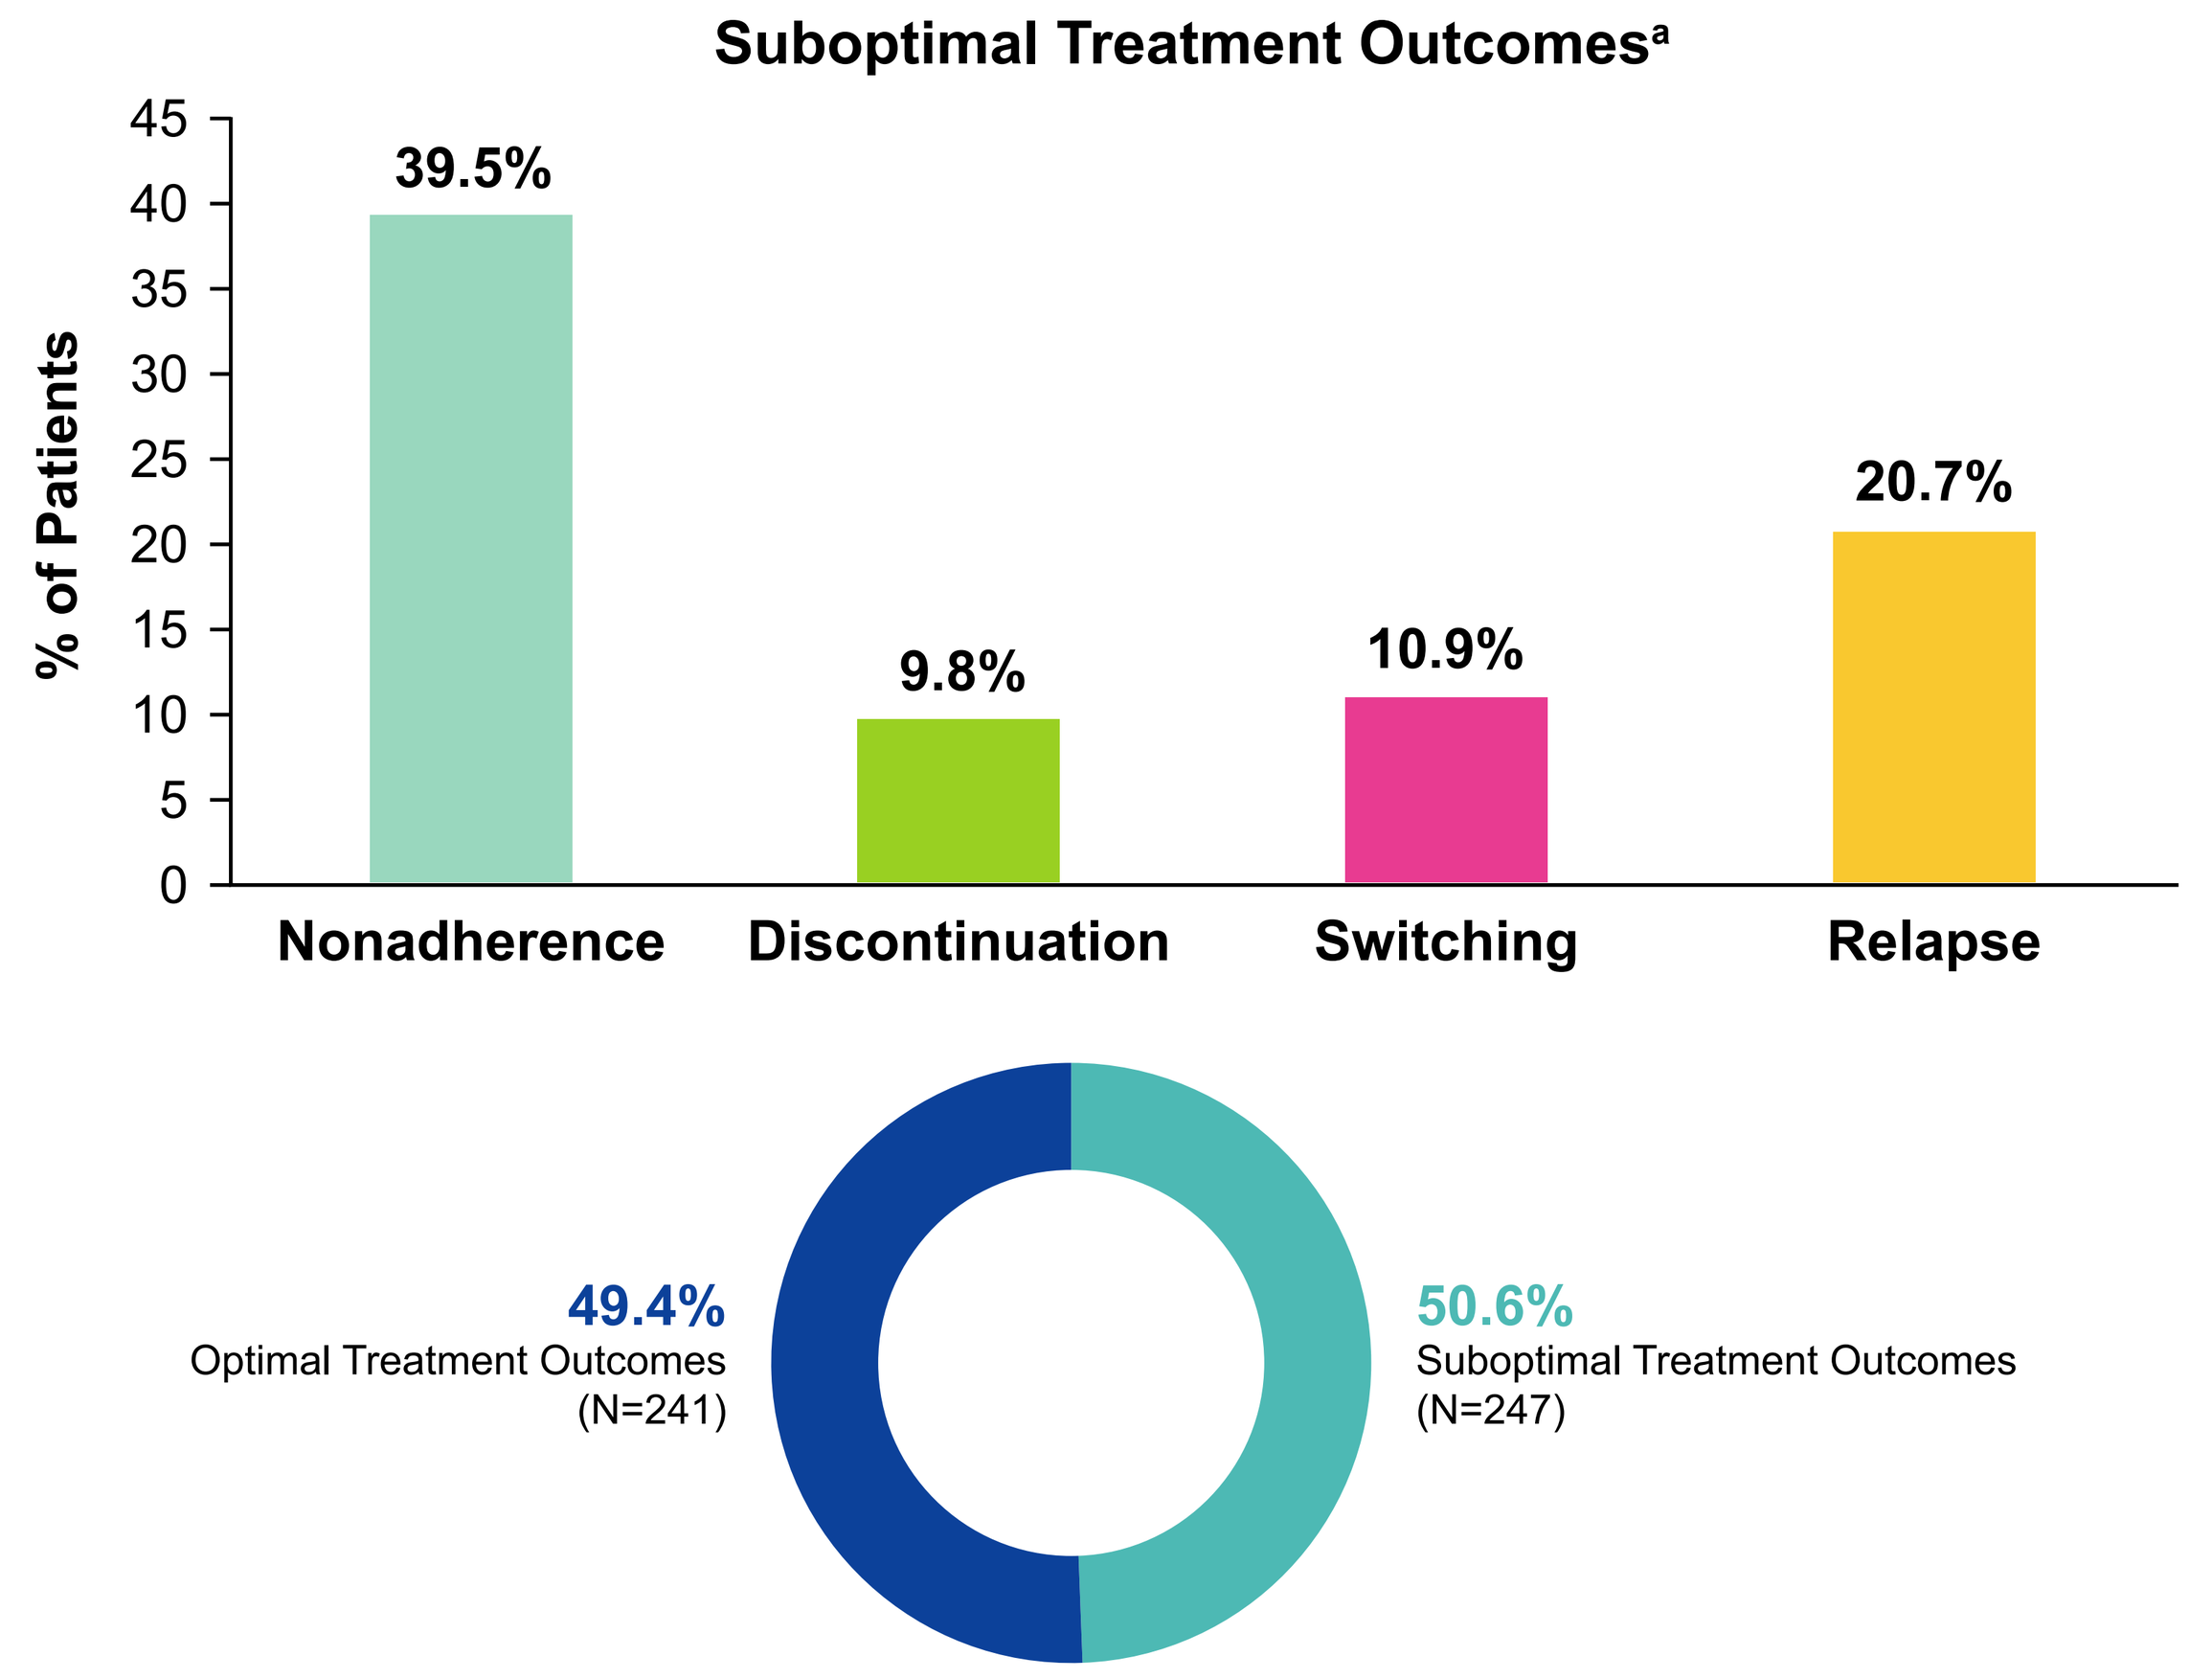
Suboptimal Treatment Outcomes[a]
% of Patients
0
5
10
15
20
25
30
35
40
45
39.5%
9.8%
10.9%
20.7%
Nonadherence
Discontinuation
Switching
Relapse
49.4%
Optimal Treatment Outcomes
(N=241)
50.6%
Suboptimal Treatment Outcomes
(N=247)

[a]Nonadherence, discontinuation, switching, and relapse are not mutually exclusive

Demographic and Clinical Characteristics

- Baseline demographic characteristics were similar for employees with versus without suboptimal treatment outcomes (**Table 1**):
 - Mean age: 42.60 vs. 43.87
 - Female: 73.7% vs. 71.4%
 - White / black / Hispanic: 32.0% / 5.3% / 3.6% vs. 29.5% / 5.8% / 6.6%
 - Married: 23.1% vs. 27.4%
 - Mean annual salary: $61,898 vs. $68,737, respectively
- Baseline clinical characteristics were also similar for employees with versus without suboptimal treatment outcomes (**Table 2**):
 - Hypertension: 14.6% vs. 16.6%
 - Hyperlipidemia: 11.7% vs. 12.9%
 - Gastrointestinal disease: 16.6% vs. 12.9%
 - Tobacco use: 3.2% vs. 2.5%
 - With baseline MRI: 57.9% vs. 58.9%; and
 - With baseline relapse: 22.7% vs. 19.9%, respectively

LIMITATIONS

- The ICD-9-CM and ICD-10-CM codes for MS do not distinguish between different types of MS, such as relapsing-remitting or primary progressive MS
- Potential limitations of administrative data include the risk of clerical inaccuracies, recording bias secondary to financial incentives, temporal changes in billing codes, and a lack of clinically relevant variables (e.g., MRI results)
- Adherence to DMD was assessed based on dispensed medications. It is not known whether the employees with MS actually took their medications
- Relapse was determined by a validated algorithm used for administrative claims data.[5,6] Relapses may have been underestimated as only relapses requiring an outpatient visit with steroid use, ER visit, or inpatient stay were captured
- These administrative claims data are derived from employees with commercial health insurance, and the data may not be generalizable to patients who do not have health insurance from their employers. Also, these results cannot be generalized to employees initiating infusion DMDs as these treatments were excluded from the analyses

Table 1. Baseline Demographic Characteristics for Employees with and without Suboptimal Treatment Outcomes

	Both groups (N=488[a])		Without indicators of suboptimal DMD treatment outcomes (N=241[b])		With indicators of suboptimal DMD treatment outcomes (N=247[c])		Comparison	
Variable	**Mean**	**SE**	**Mean**	**SE**	**Mean**	**SE**	**Difference**	**P-value**
Employee age, years	43.22	0.45	43.87	0.61	42.60	0.66	1.27	0.1596
Female	72.5%	2.0%	71.4%	2.9%	73.7%	2.8%	-2.3%	0.5667
Marital Status								
Married	25.2%	2.0%	27.4%	2.9%	23.1%	2.7%	4.3%	0.2730
Not married	18.6%	1.8%	16.6%	2.4%	20.6%	2.6%	-4.1%	0.2508
Missing marital status	56.1%	2.2%	56.0%	3.2%	56.3%	3.2%	-0.3%	0.9541
Race								
White	30.7%	2.1%	29.5%	2.9%	32.0%	3.0%	-2.5%	0.5459
Black	5.5%	1.0%	5.8%	1.5%	5.3%	1.4%	0.5%	0.7920
Hispanic	5.1%	1.0%	6.6%	1.6%	3.6%	1.2%	3.0%	0.1335
Other race	0.2%	0.2%	0.0%	0.0%	0.4%	0.4%	-0.4%	0.3228
Not specified	58.4%	2.2%	58.1%	3.2%	58.7%	3.1%	-0.6%	0.8907
Annual salary	$65,242	$2075	$68,737	$3123	$61,898	$2738	$6839	0.0996
Full-time	79.7%	1.8%	79.3%	2.6%	80.2%	2.5%	-0.9%	0.8029

[a]All but Annual salary (N=405); [b]All but Annual salary (N=198); [c]All but Annual salary (N=207).
DMD, Disease-modifying drug; **SE**, standard error

Table 2. Baseline Clinical Characteristics for Employees with and without Suboptimal Treatment Outcomes

	Both groups (N=488)		Without indicators of suboptimal DMD treatment outcomes (N=241)		With indicators of suboptimal DMD treatment outcomes (N=247)		Comparison	
Variable	**Mean**	**SE**	**Mean**	**SE**	**Mean**	**SE**	**Difference**	**P-value**
CCI	0.32	0.04	0.36	0.06	0.28	0.04	0.08	0.2987
Comorbidities								
Alcohol disorder	0.6%	0.4%	0.4%	0.4%	0.8%	0.6%	-0.4%	0.5770
Anxiety	8.6%	1.3%	6.2%	1.6%	10.9%	2.0%	-4.7%	0.0638
Arthritis	2.9%	0.8%	1.7%	0.8%	4.0%	1.3%	-2.4%	0.1140
Depression	9.2%	1.3%	7.1%	1.7%	11.3%	2.0%	-4.3%	0.1021
Diabetes	4.7%	1.0%	4.1%	1.3%	5.3%	1.4%	-1.1%	0.5616
Hyperlipidemia	12.3%	1.5%	12.9%	2.2%	11.7%	2.1%	1.1%	0.7059
Hypertension	15.6%	1.6%	16.6%	2.4%	14.6%	2.2%	2.0%	0.5379
Thyroid disease	8.4%	1.3%	6.6%	1.6%	10.1%	1.9%	-3.5%	0.1656
Chronic lung disease	3.5%	0.8%	4.6%	1.3%	2.4%	1.0%	2.1%	0.1984
Gastrointestinal disease	14.8%	1.6%	12.9%	2.2%	16.6%	2.4%	-3.7%	0.2446
Tobacco use	2.9%	0.8%	2.5%	1.0%	3.2%	1.1%	-0.7%	0.6201
MRI	58.4%	2.2%	58.9%	3.2%	57.9%	3.1%	1.0%	0.8181
Relapse	21.3%	1.9%	19.9%	2.6%	22.7%	2.7%	-2.8%	0.4575

CCI, Charlson Comorbidity Index; **DMD,** Disease-modifying drug; **MRI,** magnetic resonance imaging; **SE,** standard error

CONCLUSIONS

- **Half of employees (50.6%) with MS meeting eligibility criteria had evidence of suboptimal treatment outcomes in the first year of treatment (i.e., nonadherence, discontinuation, switching, or relapse)**
- **There were no differences in baseline characteristics between employees with and without suboptimal treatment outcomes**
- **A better understanding of nonadherence and relapse and their impact on cost may help in optimizing care in employees with MS**

REFERENCES

1. Birnbaum HG, et al. *Curr Med Res Opin.* 2009;25(4):869–77.
2. Rajagopalan K, et al. *Curr Med Res Opin.* 2011;27(1):179–88.
3. Ivanova JI, et al. *J Med Econ.* 2012;15:601–9.
4. Yermakov S, et al. *J Med Econ.* 2015;18(9):711–20.
5. Chastek BJ, et al. *J Med Econ.* 2010;13(4):618–25.
6. Capkun G, et al. *Curr Med Res Opin.* 2015;31: 1029–39.
7. Marrie RA, et al. *Mult Scler.* 2015;21(3):263–81.

ACKNOWLEDGMENTS

This study was sponsored by EMD Serono, Inc., a business of Merck KGaA, Darmstadt, Germany. Writing and editorial support for the preparation of this poster was provided by Jenna Steere, PhD, and Nick White of Ashfield (New York, NY, US); funding was provided by EMD Serono. EMD Serono reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content

DISCLOSURES

CMH: Received speaking and consulting fees from Genzyme, Genentech, EMD Serono, Biogen, and Novartis. Received research support paid to her institution by Genentech, Biogen, and PCORI. **RAB:** Employee of Better Health Worldwide, Inc. Better Health Worldwide, Inc. received funding from EMD Serono, Inc. to conduct the study. **NJR and IAB:** Employees of The HCMS Group, LLC. The HCMS Group, LLC received funding from EMD Serono, Inc. to conduct the study. **LL and ALP:** Employees of EMD Serono, Inc., Rockland, MA, US (a business of Merck KGaA, Darmstadt, Germany). **CH:** Employee of Biopharma Business of Merck KGaA, Darmstadt, Germany

Copies of this poster obtained using the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at ACTRIMS 2020; February 27–29, West Palm Beach, Florida, US