“This reprint might contain references to “Merck” or “Merck KGaA”, which refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.
There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.”

Updated Post-Approval Safety of Cladribine Tablets in the Treatment of Multiple Sclerosis, with Particular Reference to Respiratory Viral Infections and COVID-19

G. Giovannoni, J. Berger, T. Leist, D. Jack, A. Galazka, A. Nolting, D. Damian

This study was sponsored by Merck KGaA, Darmstadt, Germany.

GG has received speaker honoraria and consulting fees from AbbVie, Actelion (Janssen/J&J), Almirall, Atara Bio, Bayer, Biogen, Celgene (BMS), FivePrime, GlaxoSmithKline, GW Pharma, Ironwood, Merck & Co., Merck KGaA (Darmstadt, Germany), Novartis, Pfizer Inc., Protein Discovery Laboratories, Roche, Sanofi-Genzyme, Teva Pharmaceutical Industries Ltd, UCB, and Vertex Pharmaceuticals; and has received research support unrelated to this study from Biogen, Ironwood, Merck & Co., Novartis, and Takeda.

JB has received honoraria as a Scientific Advisory Board member for Inhibikase and Novartis; has received consultancy fees from Amgen, Celgene (BMS), Dr. Reddy’s Laboratories, EMD Serono, Encycle, Excision Bio, Genzyme, Merck KGaA (Darmstadt, Germany), MAPI, Millennium/Takeda, Morphic, and Shire; and has received honoraria and institutional grants for consultancy from Biogen and Roche/Genentech.

TL has received consultancy fees or clinical research grants from Acorda, Bayer, Biogen, Daiichi, EMD Serono, Novartis, ONO, Pfizer, and Teva Neuroscience.

DJ and **AN** are employees of Merck KGaA, Darmstadt, Germany.

AG is an employee of Merck, Aubonne, Switzerland, a division of Merck KGaA, Darmstadt, Germany.

DD is an employee of EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, a business of Merck KGaA, Darmstadt, Germany.

Medical writing assistance was provided by Steve Winter of inScience Communications, Springer Healthcare Ltd, UK, and was funded by Merck KGaA, Darmstadt, Germany.
The CLARITY study: NCT00213135; the CLARITY Extension study: NCT00641537; the ORACLE study: NCT00725985; the PREMIERE registry: NCT01013350

Updated Post-Approval Safety of Cladribine Tablets in the Treatment of Multiple Sclerosis, with Particular Reference to Respiratory Viral Infections and COVID-19

G. Giovannoni[1], J. Berger[2], T. Leist[3], D. Jack[4], A. Galazka[5], A. Nolting[4], D. Damian[6]

[1]Blizard Institute, Barts and The London School of Medicine and Dentistry, Queen Mary University of London, London, UK; [2]Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [3]Division of Clinical Neuroimmunology, Jefferson University, Comprehensive MS Center, Philadelphia, PA, USA; [4]Merck KGaA, Darmstadt, Germany; [5]Merck, Aubonne, Switzerland, a division of Merck KGaA, Darmstadt, Germany; [6]EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, a business of Merck KGaA, Darmstadt, Germany

GET POSTER PDF

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

CONCLUSIONS

The pattern of AEs of special interest in the real-world (post-approval) safety data, cumulative to July 2020, was largely consistent with the clinical safety profile for cladribine tablets.

In the clinical development program, the estimated rates of respiratory viral infections in patients treated with cladribine tablets were similar to those treated with placebo.

Regarding COVID-19, the majority of patients had mild to moderate respiratory symptoms. None received mechanical ventilation; there was one fatality.

INTRODUCTION

- The safety of treatment with cladribine tablets was assessed in the clinical trial program, including the CLARITY,[1] CLARITY Extension,[2] and ORACLE-MS[3] studies.
- Integrated safety data for cladribine tablets, including findings for the PREMIERE registry, have previously been published.[4,5]
- There have been additional safety data obtained from use in clinical practice since the approval of cladribine tablets 10 mg (3.5 mg/kg cumulative dose over 2 years; now approved in >80 countries worldwide).[6]
 - In light of the COVID-19 pandemic, we present more detail on respiratory viral infections in the clinical trial program and post-approval setting.

OBJECTIVES

To report post-approval safety data from worldwide sources, cumulative to July 2020, and to analyze rates of respiratory viral infections (including COVID-19) in patients treated with cladribine tablets.

METHODS

Summary of Data Included in the Monotherapy Oral Cohort from the Clinical Program

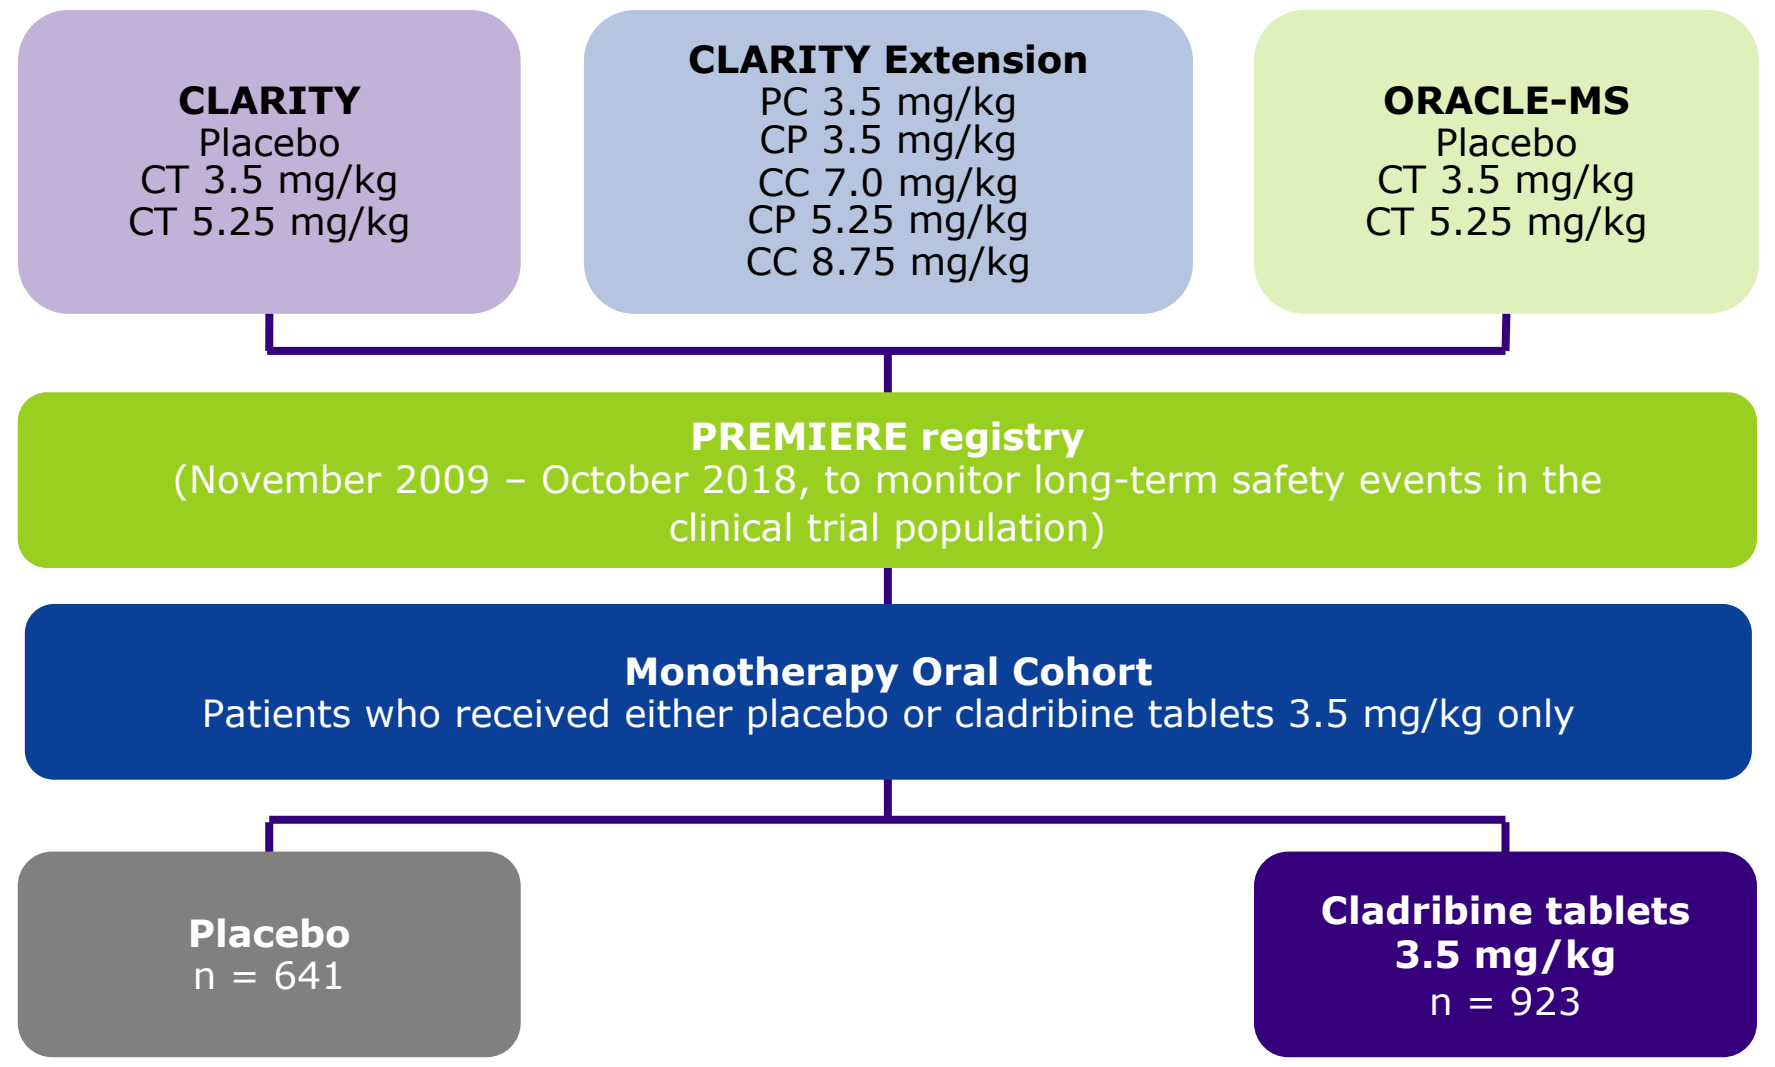
CLARITY
Placebo
CT 3.5 mg/kg
CT 5.25 mg/kg
CLARITY Extension
PC 3.5 mg/kg
CP 3.5 mg/kg
CC 7.0 mg/kg
CP 5.25 mg/kg
CC 8.75 mg/kg
ORACLE-MS
Placebo
CT 3.5 mg/kg
CT 5.25 mg/kg
PREMIERE registry
(November 2009 – October 2018, to monitor long-term safety events in the clinical trial population)
Monotherapy Oral Cohort
Patients who received either placebo or cladribine tablets 3.5 mg/kg only
Placebo
n = 641
Cladribine tablets
3.5 mg/kg
n = 923

CP 3.5 mg/kg, cladribine tablets 3.5 mg/kg in CLARITY followed by placebo in CLARITY Extension; **CP 5.25 mg/kg**, cladribine tablets 5.25 mg/kg in CLARITY followed by placebo in CLARITY Extension; **CC 7.0 mg/kg**, cladribine tablets 3.5 mg/kg in CLARITY followed by cladribine tablets 3.5 mg/kg in CLARITY Extension; **CC 8.75 mg/kg**, cladribine tablets 5.25 mg/kg in CLARITY followed by cladribine tablets 3.5 mg/kg in CLARITY Extension; **PC 3.5 mg/kg**, placebo in CLARITY followed by cladribine 3.5 mg/kg in CLARITY Extension

METHODS

Monotherapy Oral Cohort Data

- The Monotherapy Oral Cohort comprised patients from the CLARITY, CLARITY Extension, and ORACLE-MS trials, and the PREMIERE registry.
 - 923 patients received cladribine tablets 3.5 mg/kg.
 - 641 patients received placebo.

Post-Approval Data

- Adverse event reports (serious and non-serious) from post-approval worldwide sources are presented, cumulative to July 2020.
- COVID-19 findings were sourced from the Merck KGaA Global Patient Safety Database (to 15 January 2021).

RESULTS

Table 1. Baseline Characteristics of the Monotherapy Oral Cohort from the Clinical Program

	Placebo (n = 641)	Cladribine tablets 3.5 mg/kg (n = 923)
Patient-years	2422	3937
Mean time on study, years (SD)	3.78 (2.66)	4.27 (2.53)
Time on study, ≥96–480 weeks [~2–9 years], n (%)	493 (76.9)	784 (84.9)
Time on study, ≥192–480 weeks [~4–9 years], n (%)	204 (31.8)	431 (46.7)
Median age, years[a] (range)	36.5 (18.1, 64.2)	37.6 (18.2, 66.1)
Age >40 years, n (%)	245 (38.2)	383 (41.5)
Female, n (%)	424 (66.1)	612 (66.3)
Prior use of DMT, n (%)	131 (20.4)	184 (19.9)

[a] As reported at first dosing date.

Table 2. Adverse Events of Special Interest in the Clinical Program and Post-Approval

	Monotherapy Oral Cohort cladribine tablets 3.5 mg/kg (n = 923)		Post-Approval Information cladribine tablets 3.5 mg/kg (n = 18,463)	
	n	AE rate (crude incidence, %)	n	AE rate (crude incidence, %)
Severe lymphopenia	24	2.60	39	0.21
Herpes zoster	28	3.03	197	1.07
Tuberculosis	2[b]	0.22	10	0.05
Severe infection	29	3.14	227	1.23
PML	0	0	0	0
Opportunistic infection[a]	10	1.08	7	0.04
Malignancy	10	1.08	42	0.23
Congenital anomaly	0[c]	0	0[d]	0

Post-Approval Information concerns adverse event reports (from post-approval sources) among the first 18,463 patients treated with cladribine tablets from August 2017 to July 2020; [a] Majority of opportunistic infections were superficial dermal and mucosal fungal infections that resolved on standard treatments. Opportunistic infections that could be life-threatening were not observed; [b] Both cases of tuberculosis were serious (one coded as tuberculosis, one coded as pulmonary tuberculosis); [c] No cases of congenital anomaly occurred in pregnancies during cladribine treatment or within 6 months after the last dose; [d] In one case of maternal exposure during pregnancy reported by a Health Authority (#E2B_90073559), an elective termination was performed due to a congenital anomaly of the fetus (not further specified). Exposure to cladribine tablets occurred in the first trimester. The patient was on concomitant medication with terbutaline and ferrous sulfate which could adversely impact fetal development.

Post-Approval Data

- A total of 3357 AEs were reported in the first 18,463 patients who received cladribine tablets post-approval.
- In total, 435 (13%) of the 3357 AEs were classified as serious.
- The pattern of AEs of special interest was largely consistent with the clinical safety profile for cladribine tablets.

AE, adverse event; **DMT**, disease-modifying therapy; **n**, number of events; **PML**, progressive multifocal leukoencephalopathy; **SD**, standard deviation

Table 3. Respiratory Viral Infections in the Clinical Program and Post-Approval

	Monotherapy Oral Cohort				Post-Approval Information cladribine tablets 3.5 mg/kg (n = 18,463)	
	Placebo (n = 641)		Cladribine tablets 3.5 mg/kg (n = 923)			
	n	AE rate (crude incidence, %)	n	AE rate (crude incidence, %)	n	AE rate (crude incidence, %)
H1N1 influenza	0	0	1	0.11	2	0.01
Influenza	51	7.96	88	9.53	126	0.68
Laryngitis viral	0	0	1	0.11	0	0
Respiratory tract infection viral	13	2.03	26	2.82	1	0
Viral infection	12	1.87	13	1.41	49	0.27
Viral pharyngitis	1	0.16	1	0.11	0	0
Viral upper respiratory tract infection	96	14.98	158	17.12	7	0.04

Post-Approval Information concerns adverse event reports (from post-approval sources) among the first 18,463 patients treated with cladribine tablets from August 2017 to July 2020.

- Respiratory viral infections were seen at similar rates in patients treated with cladribine tablets compared with placebo.
- The pattern of respiratory viral infections was also consistent in the real-world (post-approval) setting.

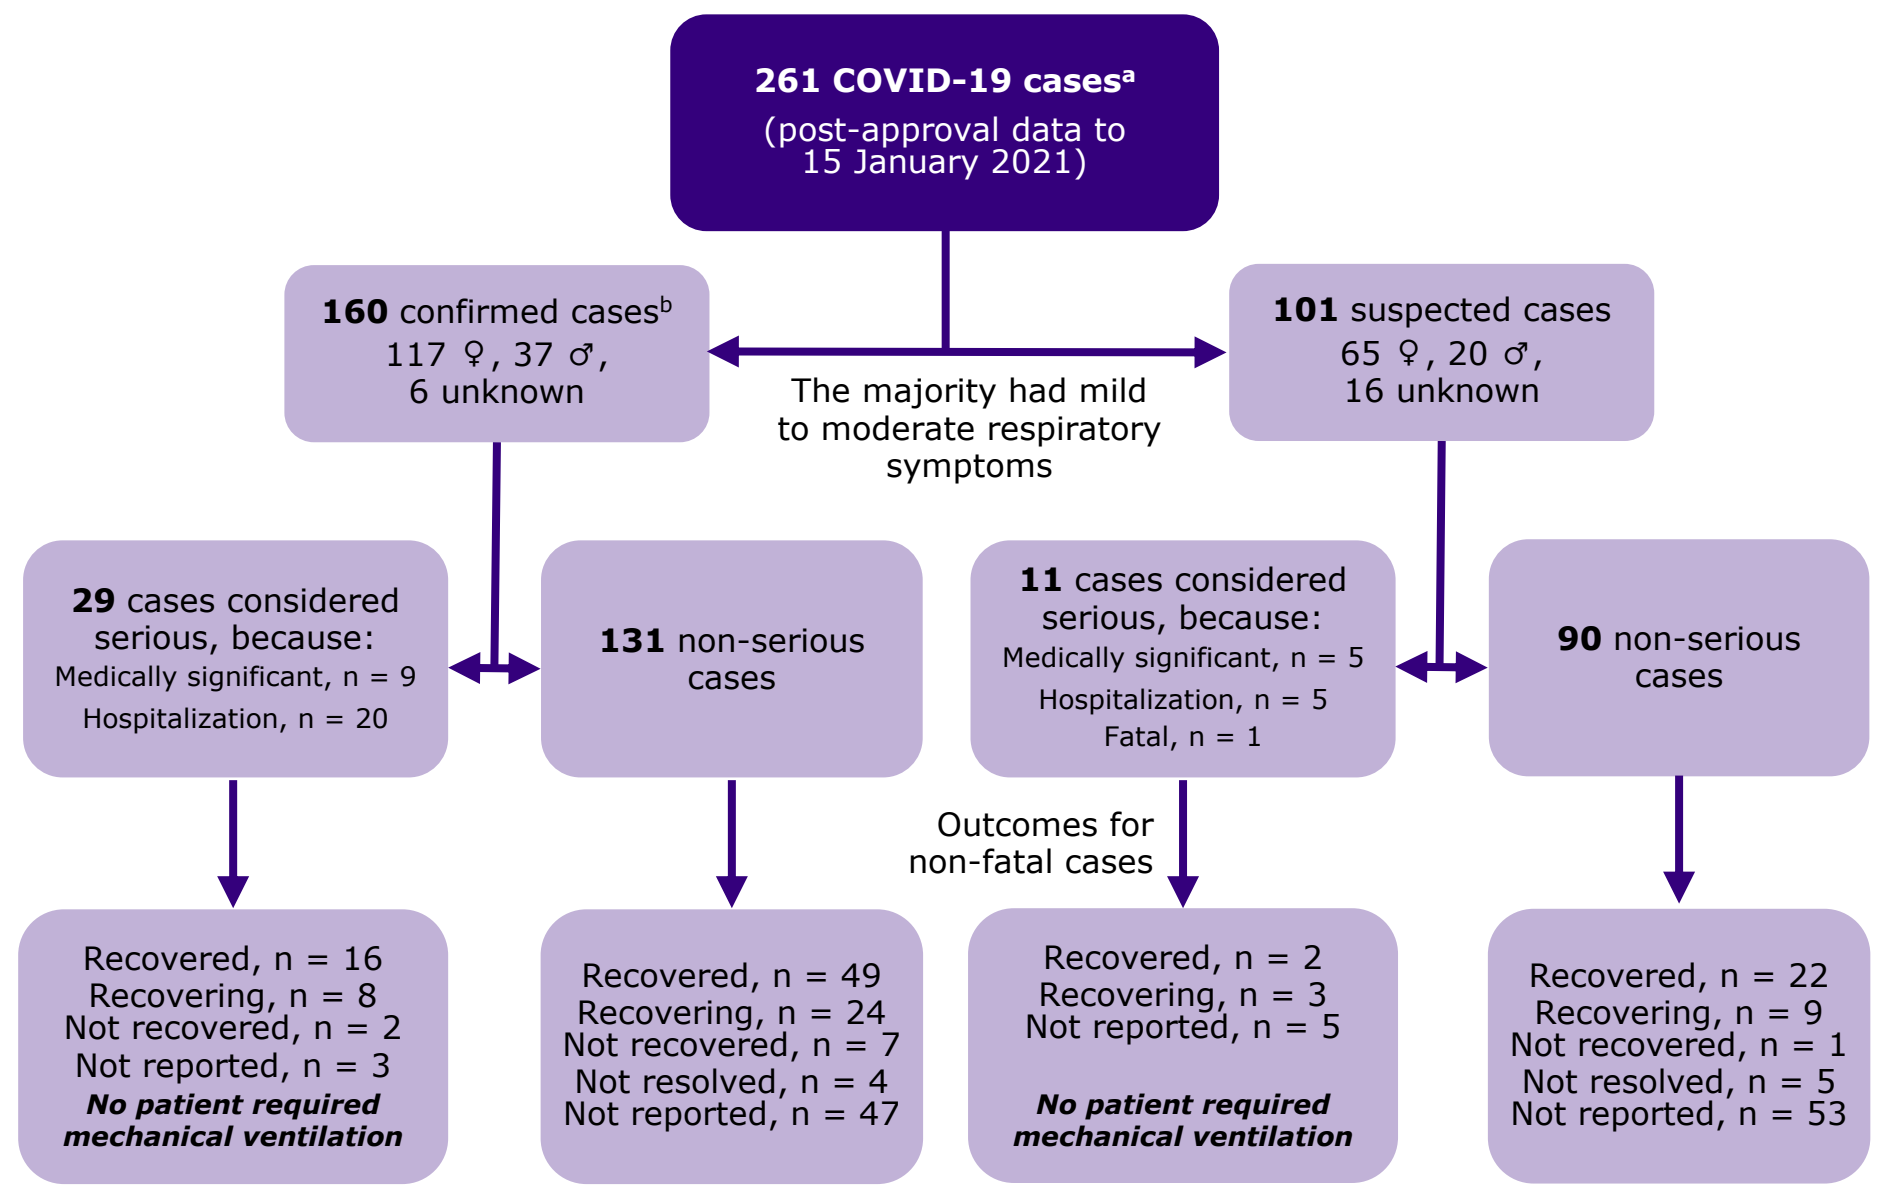
261 COVID-19 cases[a]
(post-approval data to 15 January 2021)
160 confirmed cases[b]
117 ♀, 37 ♂, 6 unknown
101 suspected cases
65 ♀, 20 ♂, 16 unknown
The majority had mild to moderate respiratory symptoms
29 cases considered serious, because:
Medically significant, n = 9
Hospitalization, n = 20
131 non-serious cases
11 cases considered serious, because:
Medically significant, n = 5
Hospitalization, n = 5
Fatal, n = 1
90 non-serious cases
Outcomes for non-fatal cases
Recovered, n = 16
Recovering, n = 8
Not recovered, n = 2
Not reported, n = 3
No patient required mechanical ventilation
Recovered, n = 49
Recovering, n = 24
Not recovered, n = 7
Not resolved, n = 4
Not reported, n = 47
Recovered, n = 2
Recovering, n = 3
Not reported, n = 5
No patient required mechanical ventilation
Recovered, n = 22
Recovering, n = 9
Not recovered, n = 1
Not resolved, n = 5
Not reported, n = 53

[a] An additional 11 patients had compatible COVID-19 symptoms but not evaluated further given negative PCR tests; [b] Includes 18 patients who were asymptomatic (2 cases considered serious).

- The time to onset of COVID-19 from start of most recent course of cladribine tablets was available for 139/261 patients; median of 162 days (i.e. ~5 months).

1. Giovannoni G, et al. *N Engl J Med.* 2010;362:416–426. 2. Giovannoni G, et al. *Mult Scler.* 2018;24:1594–1604. 3. Leist T, et al. *Lancet Neurol.* 2014;13:257–267. 4. Cook S, et al. *Mult Scler Relat Disord.* 2019;29:157–167. 5. Cook S, et al. *Mult Scler.* 2018;24(S2):465–466. 6. Periodic Benefit-Risk Evaluation Report (PBRER 6) (data on file, Merck KGaA, Darmstadt, Germany).

Presented at the AAN 2021 Virtual Congress | 17–22 April

This study was sponsored by Merck KGaA, Darmstadt, Germany.

METHODS

Summary of Data Included in the Monotherapy Oral Cohort from the Clinical Program

CLARITY
Placebo
CT 3.5 mg/kg
CT 5.25 mg/kg

CLARITY Extension
PC 3.5 mg/kg
CP 3.5 mg/kg
CC 7.0 mg/kg
CP 5.25 mg/kg
CC 8.75 mg/kg

ORACLE-MS
Placebo
CT 3.5 mg/kg
CT 5.25 mg/kg

PREMIERE registry
(November 2009 – October 2018, to monitor long-term safety events in the clinical trial population)

Monotherapy Oral Cohort
Patients who received either placebo or cladribine tablets 3.5 mg/kg only

Placebo
n = 641

Cladribine tablets 3.5 mg/kg
n = 923

CP 3.5 mg/kg, cladribine tablets 3.5 mg/kg in CLARITY followed by placebo in CLARITY Extension; **CP 5.25 mg/kg**, cladribine tablets 5.25 mg/kg in CLARITY followed by placebo in CLARITY Extension; **CC 7.0 mg/kg**, cladribine tablets 3.5 mg/kg in CLARITY followed by cladribine tablets 3.5 mg/kg in CLARITY Extension; **CC 8.75 mg/kg**, cladribine tablets 5.25 mg/kg in CLARITY followed by cladribine tablets 3.5 mg/kg in CLARITY Extension; **PC 3.5 mg/kg**, placebo in CLARITY followed by cladribine 3.5 mg/kg in CLARITY Extension

RESULTS

261 COVID-19 cases[a]
(post-approval data to
15 January 2021)

160 confirmed cases[b]
117 ♀, 37 ♂,
6 unknown

101 suspected cases
65 ♀, 20 ♂,
16 unknown

The majority had mild
to moderate respiratory symptoms

29 cases considered
serious, because:
Medically significant, n = 9
Hospitalization, n = 20

Recovered, n = 16
Recovering, n = 8
Not recovered, n = 2
Not reported, n = 3
No patient required mechanical ventilation

131 non-serious
cases

Recovered, n = 49
Recovering, n = 24
Not recovered, n = 7
Not resolved, n = 4
Not reported, n = 47

11 cases considered
serious, because:
Medically significant, n = 5
Hospitalization, n = 5
Fatal, n = 1

Outcomes for
non-fatal cases

Recovered, n = 2
Recovering, n = 3
Not reported, n = 5
No patient required mechanical ventilation

90 non-serious
cases

Recovered, n = 22
Recovering, n = 9
Not recovered, n = 1
Not resolved, n = 5
Not reported, n = 53

The time to onset of COVID-19 from start of most recent course of cladribine tablets was available for 139/261 patients; median of 162 days (i.e. ~5 months).

[a] An additional 11 patients had compatible COVID-19 symptoms but not evaluated further given negative PCR tests; [b] Includes 18 patients who were asymptomatic (2 cases considered serious).