“This reprint might contain references to “Merck” or “Merck KGaA”, which refer to (1) Merck KGaA, Darmstadt, Germany; (2) an affiliate of Merck KGaA, Darmstadt, Germany; or (3) one of the businesses of Merck KGaA, Darmstadt, Germany, which operate as EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada.
There are two different, unaffiliated companies that use the name “Merck”. Merck KGaA, Darmstadt, Germany, which is providing this content, uses the firm name “Merck KGaA, Darmstadt, Germany” and the business names EMD Serono in the healthcare, MilliporeSigma in the life science and EMD Electronics in the electronics business in the U.S. and Canada. The other company, Merck & Co., Inc. holds the rights in the trademark “Merck” in the U.S. and Canada. Merck & Co., Inc. is not affiliated with or related to Merck KGaA, Darmstadt, Germany, which owns the “Merck” trademark in all other countries of the world.”

For Reactive Use Only

Year 1 Performance of adveva®, a Patient Support Program for Patients Taking Cladribine Tablets for Highly Active Relapsing Remitting Multiple Sclerosis in the United Kingdom

K. Morgan, N. Lott, M. Lyons

This analysis was sponsored by Merck Serono Ltd, a business of Merck KGaA, Darmstadt, Germany

The adveva® nurse-led service was provided by Lloyds Pharmacy Clinical Homecare.
KM and **NL** are employees of Merck Serono Ltd, a business of Merck KGaA, Darmstadt, Germany. **ML** is a former employee of Merck Serono Ltd and is a current employee of Roche Products Ltd.
Editorial support for preparation of this poster was provided by Farah Johnson-May of inScience Communications, Springer Healthcare Ltd, UK, and was funded by Merck KGaA, Darmstadt, Germany.

Year 1 Performance of adveva®, a Patient Support Program for Patients Taking Cladribine Tablets for Highly Active Relapsing Remitting Multiple Sclerosis in the United Kingdom

K. Morgan[1], N. Lott[1], M. Lyons[2]

[1]Merck Serono Ltd, Feltham, Middlesex, UK; [2]Employee of Merck Serono Ltd, Feltham, Middlesex, UK, at the time of the analysis and current employee of Roche Products Ltd.

GET POSTER PDF
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

CONCLUSION

This is the first report in a post-approval and real world setting that suggests that the majority of patients find cladribine tablets simple-to-take.

100%

All 133 patients included in this assessment self-reported that they achieved the required cumulative 1.75 mg/kg dose of cladribine tablets in their first year of treatment.

A proactive telephone-based patient support program was of particular value to the 22 (17%) of patients who required additional support during their treatment days.

INTRODUCTION

- Merck provides a free patient support program (PSP), called adveva®, to support patients prescribed cladribine tablets to receive medication reminders during treatment days.
- The service also offers blood test reminders, a home-based phlebotomy service, and clinical appointment reminders (**Figure 1**).
- Adherence to disease-modifying therapy is a key determinant of treatment effectiveness, relating to drug regimen, patient knowledge, and patient-healthcare practitioner relationships.[1]

Figure 1. Full Support Services of the PSP

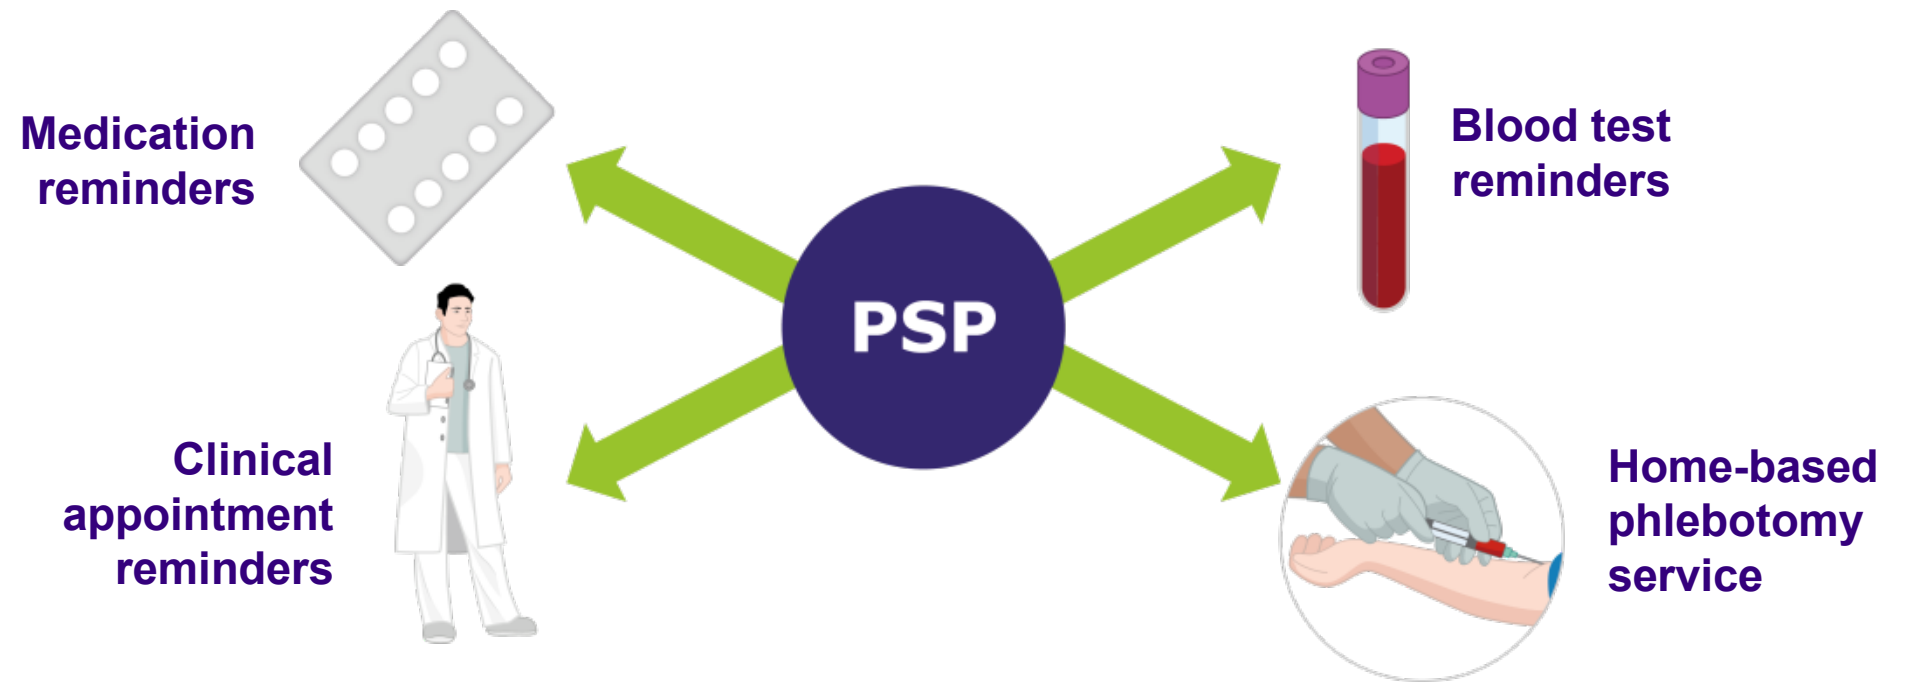
Medication reminders
Blood test reminders
PSP
Clinical appointment reminders
Home-based phlebotomy service

OBJECTIVE

To present year 1 results of a PSP for patients with highly active relapsing-remitting multiple sclerosis (RRMS) taking cladribine tablets.

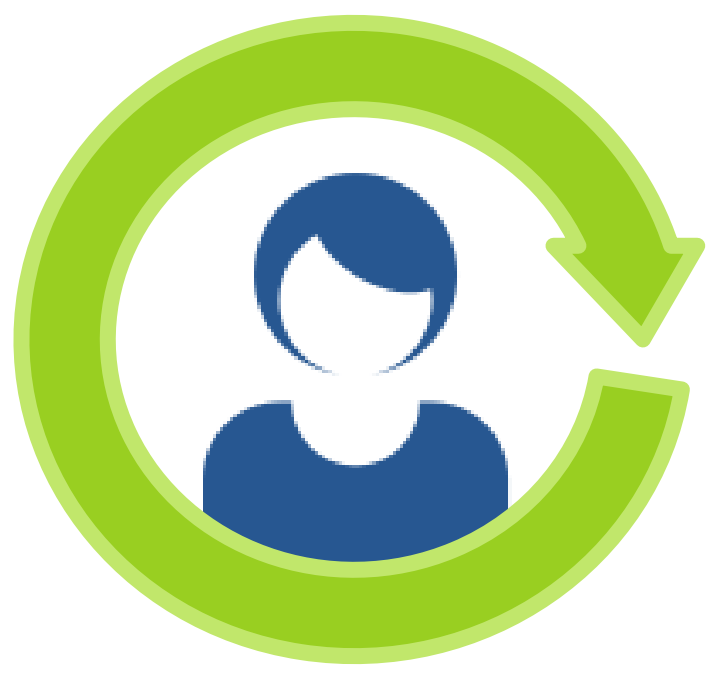

PATIENT

METHODS

- Aggregated and anonymized telephone conversation records were reviewed between December 2017 and August 2018 (**Figure 2**).
- The analysis determined whether patient-reported adherence, recorded within the PSP data collection system, was achieved and if basic support or additional support was required to achieve the full first year cladribine tablets cumulative dose of 1.75 mg/kg.
- All potential adverse events (AEs) are reported to Merck KGaA, the manufacturer, in accordance with local reporting timelines.

Figure 2. PSP Support Between Analysis Time Points

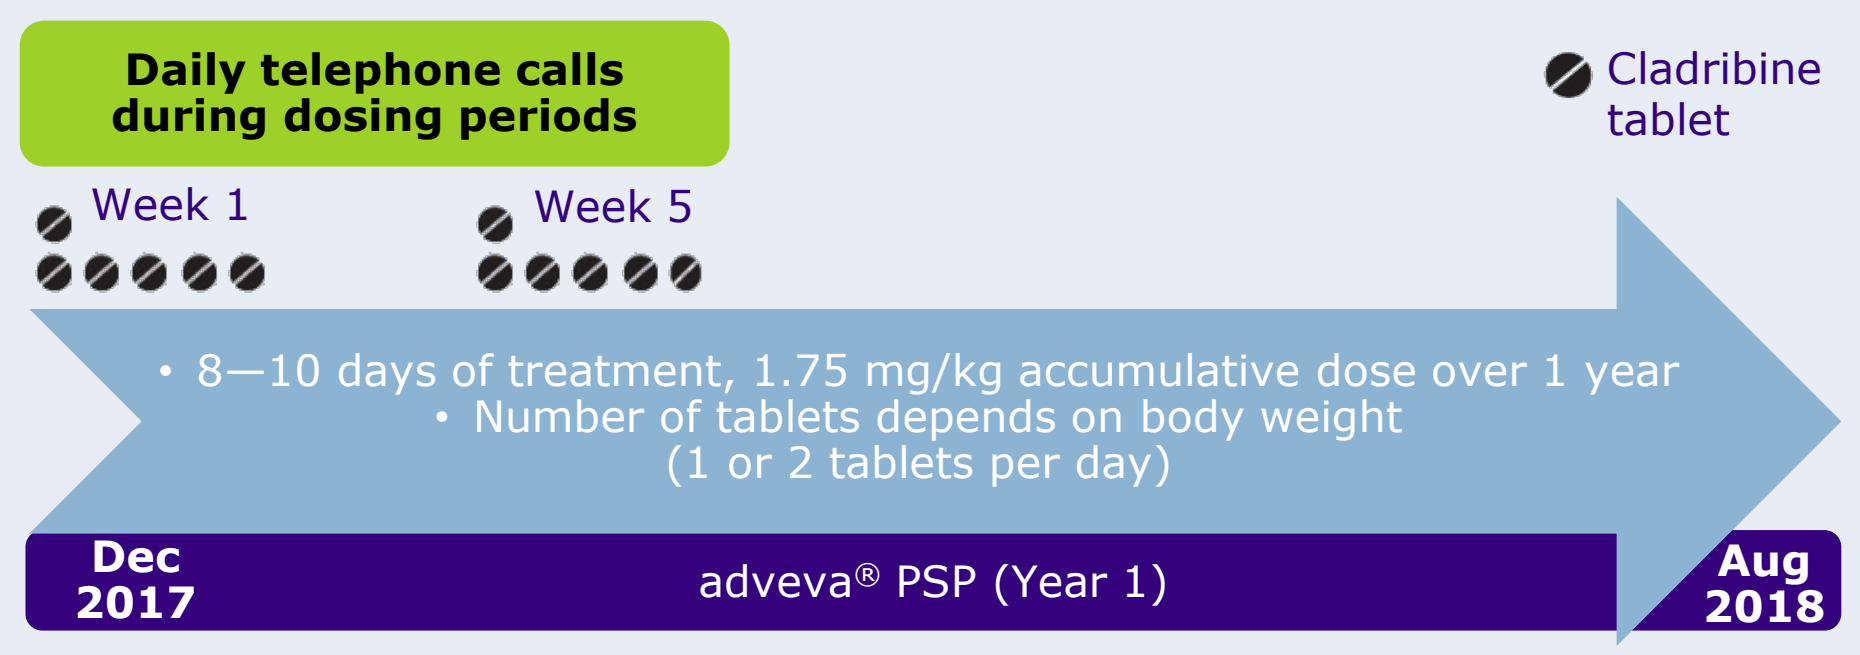
Daily telephone calls during dosing periods
Cladribine tablet
Week 1
Week 5
• 8—10 days of treatment, 1.75 mg/kg accumulative dose over 1 year
• Number of tablets depends on body weight (1 or 2 tablets per day)
Dec 2017
adveva® PSP (Year 1)
Aug 2018

RESULTS

- 262 patients were registered with adveva® between December 2017 and August 2018 (**Figure 3**).
- Of these, 186 patients were registered as fully supported patients (daily phone calls on treatment days) (**Figure 3**).
- 133 patients who self-reported to have completed both treatment weeks in year 1 at the time of assessment (**Figure 3**); 53 patients had not yet reached week 5 at the time of assessment.

Figure 3. Patient Flow

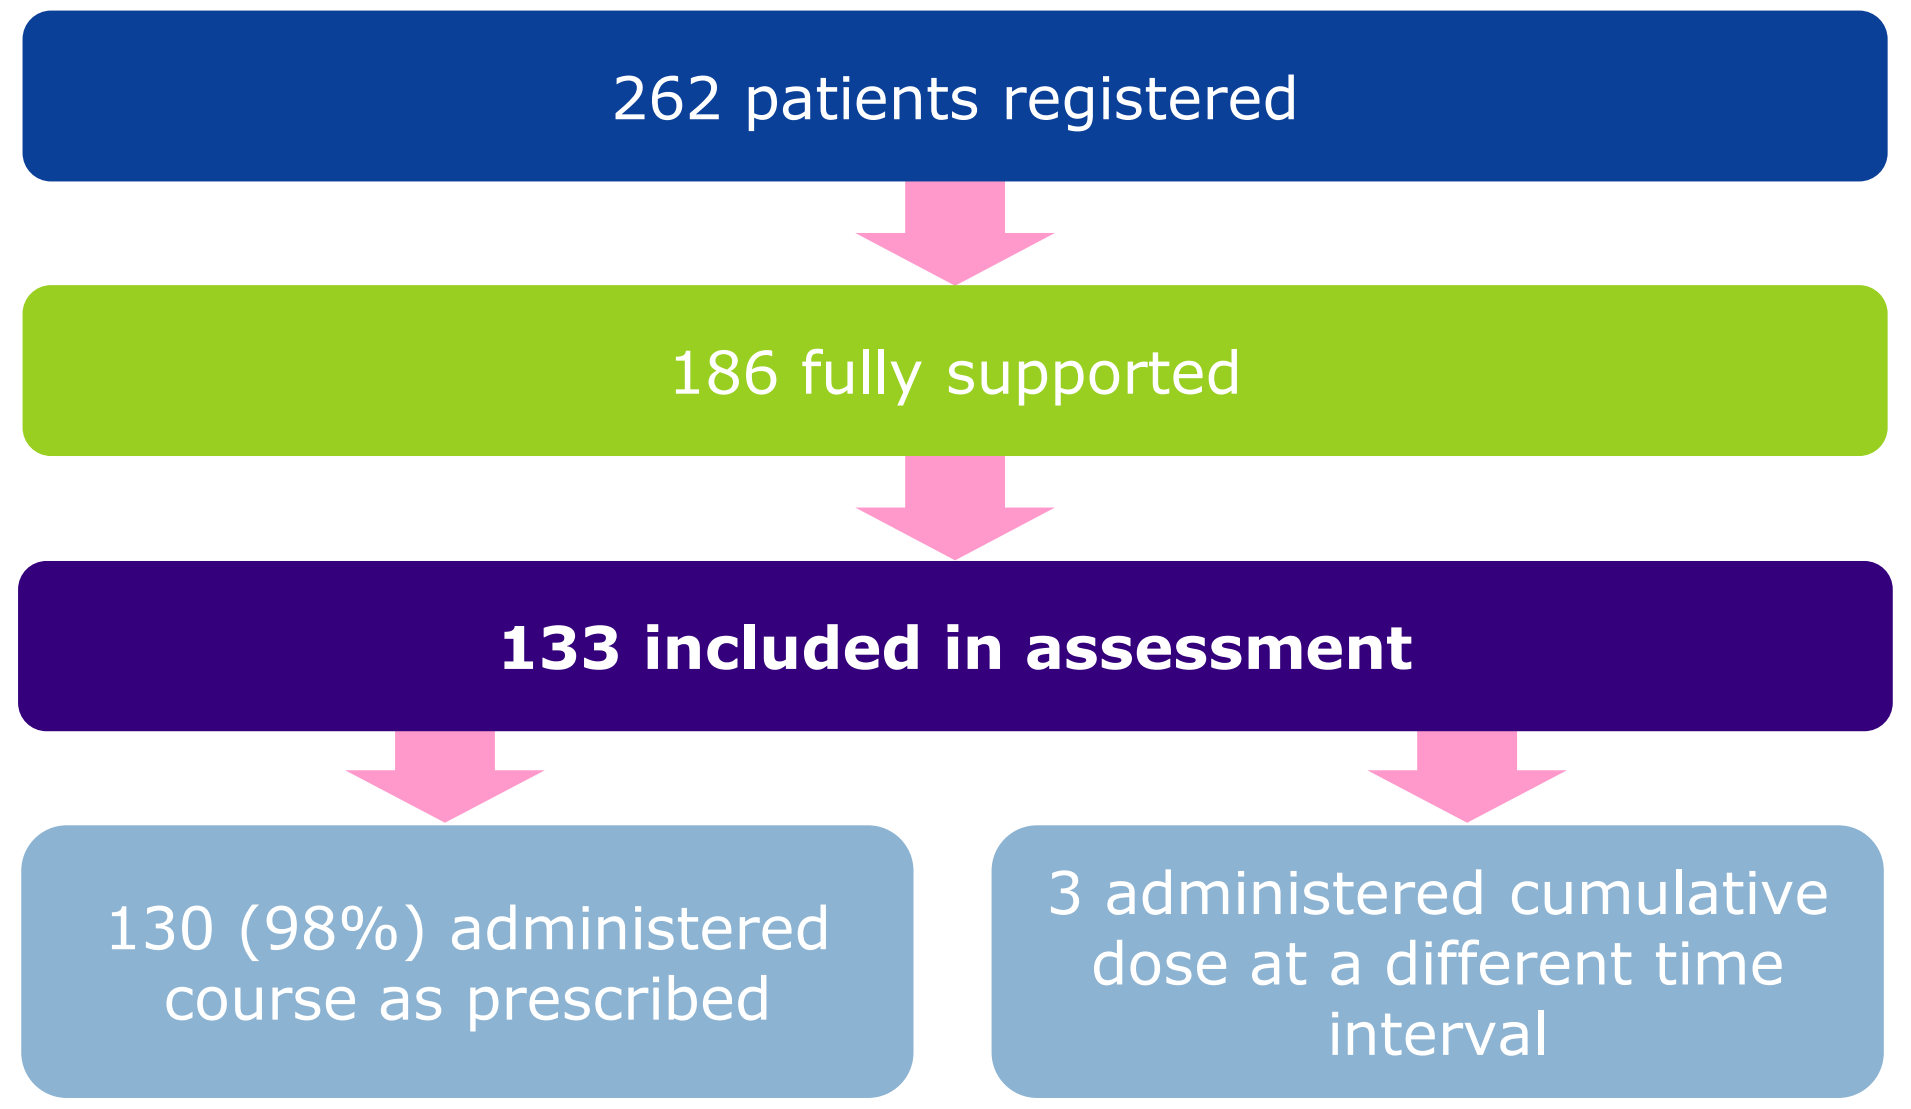
262 patients registered
186 fully supported
133 included in assessment
130 (98%) administered course as prescribed
3 administered cumulative dose at a different time interval

- 100% of patients analyzed successfully took the cumulative dose of 1.75 mg/kg in the first year.
 — Of the three patients who did not achieve this within the recommended time frame, one patient had a 1-day extension, one took the last dose 1 week late, and one administered the second treatment week 1 week early.
- 22 (17%) patients required additional support from the adveva® call handlers during their treatment days.
 — Discussion topics for these patients (may involve more than one topic) included: treatment counselling (to support patients using approved scripts) (41%), dosing advice (30%), and liaising over AEs (30%) (**Figure 4**).

Figure 4. Breakdown of Support Provided to Patients Enrolled on the PSP

Level of Support Patients Received

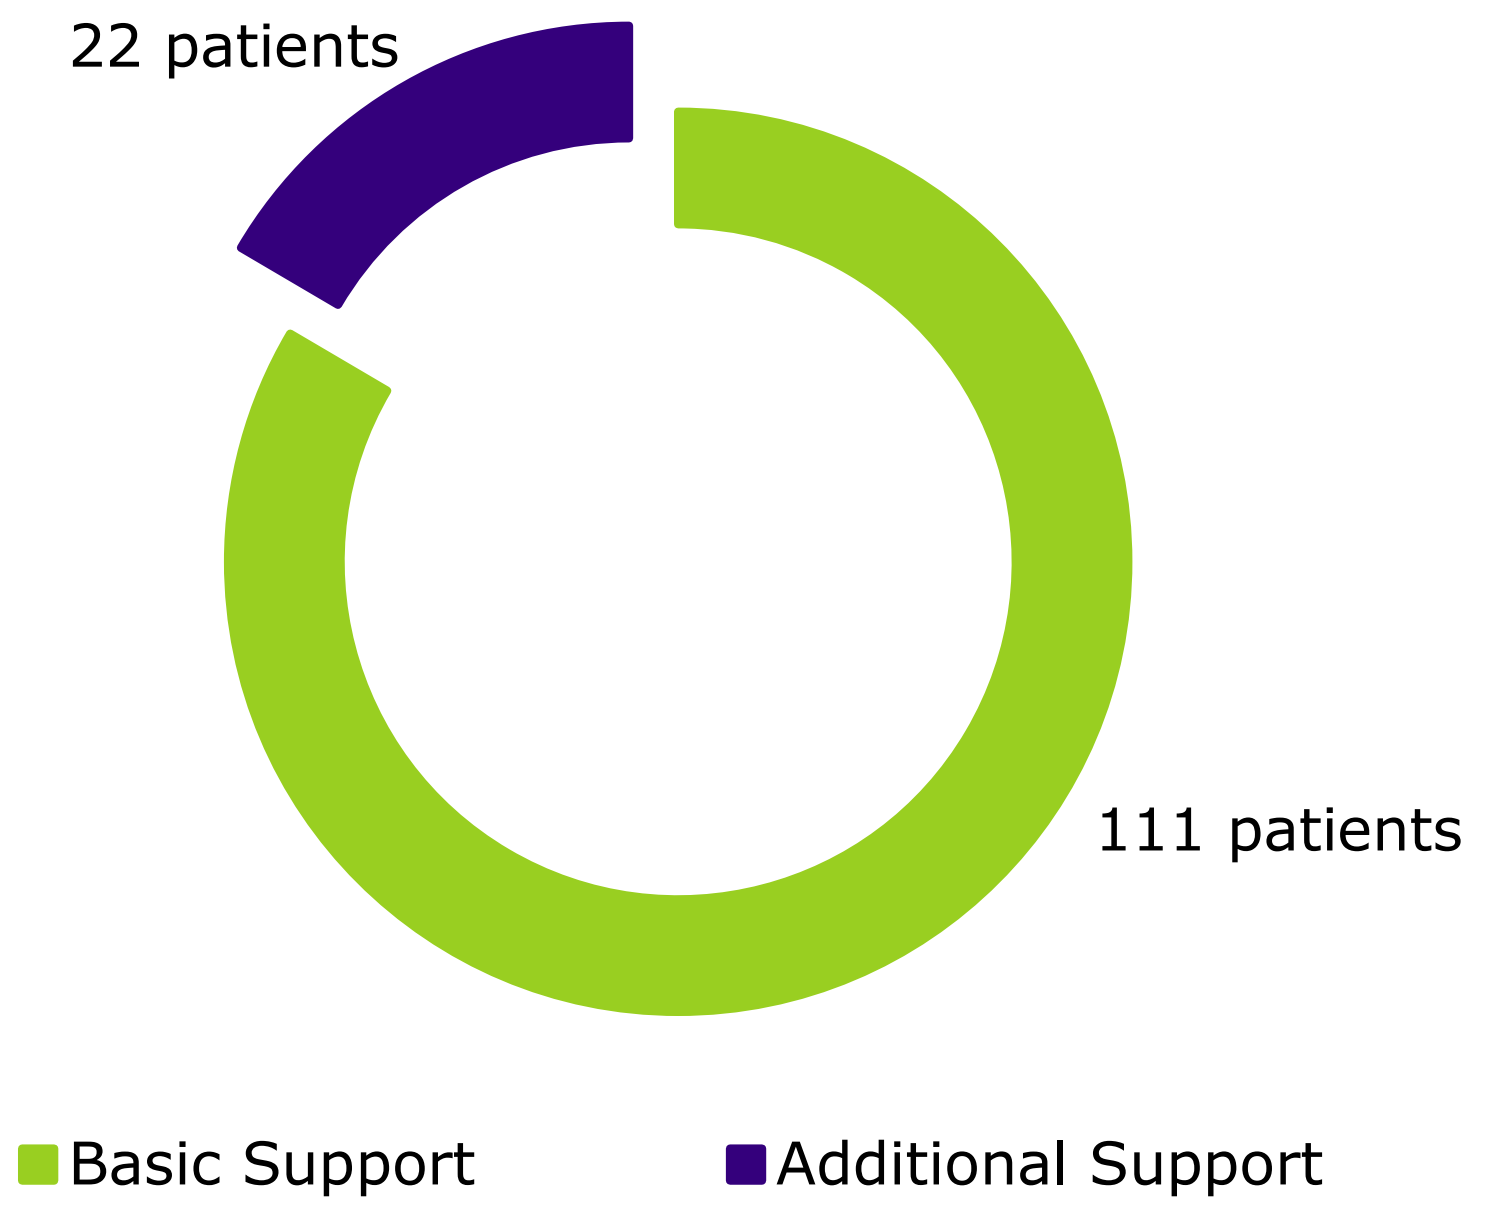
22 patients
111 patients
Basic Support
Additional Support

Reasons for Additional Support*

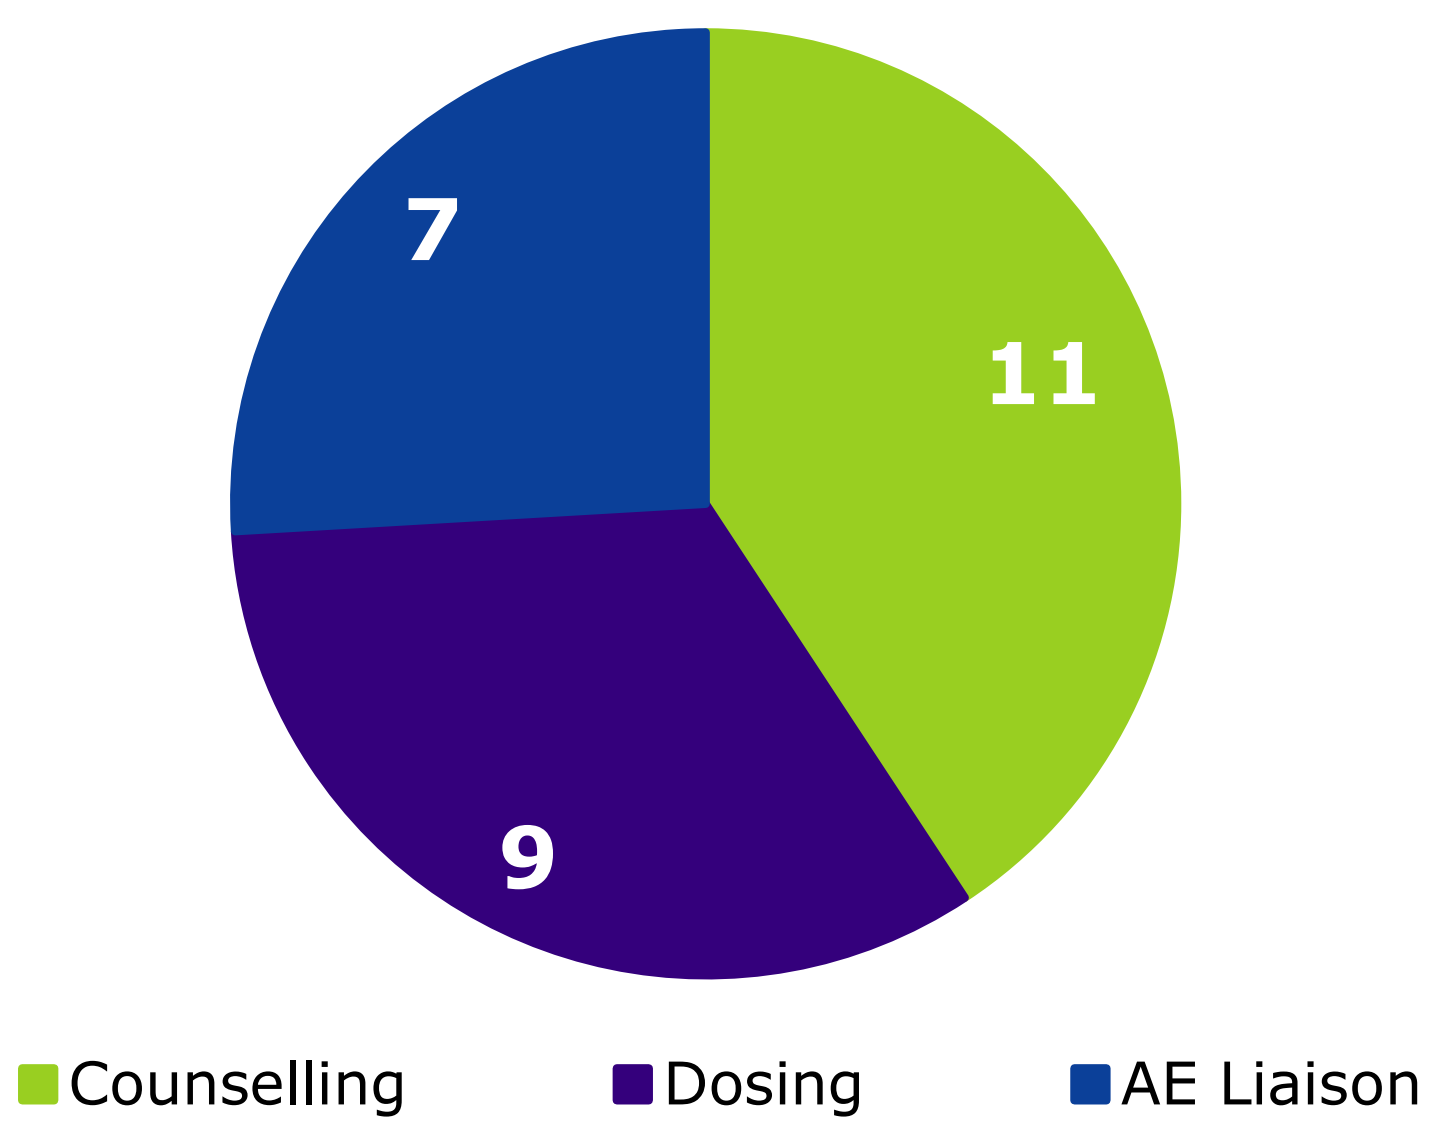
7
11
9
Counselling
Dosing
AE Liaison

*While 22 patients required additional support, some support calls covered multiple topics.

1. Fernandes M, et al. *Int J Clin Pharm.* 2014;36:86–91.

Cladribine tablets 10 mg (3.5 mg/kg cumulative dose over 2 years) are indicated for the treatment of patients with MS in the United States (relapsing forms of MS, including relapsing-remitting disease and active secondary progressive disease, in adults)

The adveva® nurse-led service was provided by Lloyds Pharmacy Clinical Homecare.

KM and **NL** are employees of Merck Serono Ltd, a business of Merck KGaA, Darmstadt, Germany. **ML** is a former employee of Merck Serono Ltd and is a current employee of Roche Products Ltd. Editorial support for preparation of this poster was provided by Farah Johnson-May of inScience Communications, Springer Healthcare Ltd, UK, and was funded by Merck KGaA, Darmstadt, Germany.

Presented at the American Academy of Neurology (AAN) 2020 Virtual Congress | 25 April – 1 May

For Reactive Use Only

This analysis was sponsored by Merck Serono Ltd, a business of Merck KGaA, Darmstadt, Germany