Selection of the recommended phase 3 dose for the IAP antagonist xevinapant in combination with high-dose cisplatin with concurrent radiotherapy (CRT) in patients with high-risk locally advanced squamous cell carcinoma of the head and neck (LA SCCHN)

Y. Vugmeyster,[1] A. Ravula,[1] E. Rouits,[2] P. M. Diderichsen,[3] H. J. Kleijn,[3] K. Goteti,[1] A. Schroeder,[4] K. Venkatakrishnan[1]

[1]EMD Serono, Billerica, MA, USA; [2]Debiopharm International SA, Lausanne, Switzerland; [3]Certara Strategic Consulting, Breda, the Netherlands; [4]the healthcare business of Merck KGaA, Darmstadt, Germany

BACKGROUND

Squamous cell carcinoma of the head and neck (SCCHN) is the **eighth** most common cancer worldwide;[1] **~60%** of patients are diagnosed with locally advanced disease,[2] which in many cases cannot be removed by surgical resection[3]

High-dose cisplatin with concurrent radiotherapy (CRT) is the standard of care for patients with unresected locally advanced (LA) SCCHN[3,4]

Xevinapant is a first-in-class, potent, small molecule antagonist of **inhibitor of apoptosis proteins (IAPs)**, formulated as an **oral solution**

Xevinapant is designed to **restore sensitivity of cancer cells to apoptosis** and to **enhance the effects of other anticancer treatments**, such as chemotherapy and radiotherapy.[5-8] Xevinapant is thought to:

- **Restore sensitivity to apoptosis** in cancer cells by blocking X-linked IAP and cellular IAPs 1 and 2 (cIAP1/2) leading to activation of caspases downstream of intrinsic and extrinsic apoptotic pathways[5-8]
- **Enhance inflammatory antitumor responses** by immune cells of the tumor microenvironment by activating noncanonical NF-κB signaling via blocking cIAP1/2 effects downstream of the tumor necrosis factor receptor[6,7,9]

Clinical proof of concept and the RP2D of xevinapant were established in a phase 1/2 study, NCT02022098 (phase 1b dose escalation followed by a randomized phase 2 study; **Figure 1**)[8,10]

In the randomized, double-blind, multicenter, phase 2 study, **xevinapant + CRT significantly increased locoregional control (LRC; primary endpoint) at 18 months** vs placebo + CRT[8]

At **3-year follow-up median overall survival (OS) and progression-free survival (PFS) were significantly improved with xevinapant + CRT** vs placebo + CRT[11]

Figure 1. Clinical proof of concept and RP2D of xevinapant

Phase 1b[10]

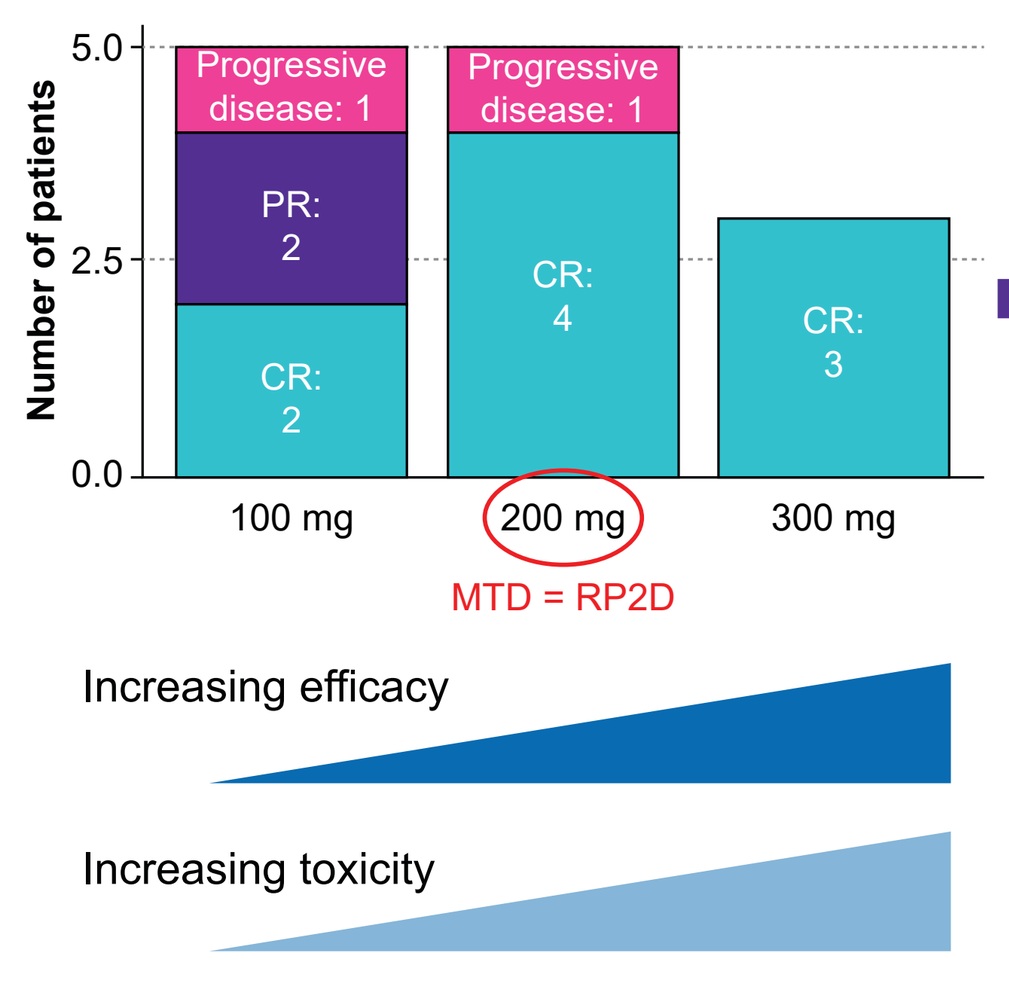
Number of patients
5.0
2.5
0.0
Progressive disease: 1
PR: 2
CR: 2
Progressive disease: 1
CR: 4
CR: 3
100 mg
200 mg
300 mg
MTD = RP2D
Increasing efficacy
Increasing toxicity

RP2D: oral xevinapant 200 mg/day days 1-14 every 3 weeks for 3 cycles

Phase 2[8]

Xevinapant RP2D in combination with CRT, with xevinapant dose reductions to 150 mg or 100 mg for the management of toxicities

Endpoint	Xevinapant + CRT (n=48)	Placebo + CRT (n=48)
LRC at 18 months, % (95% CI)	**54.2 (39.2-68.6)**	**33.3 (20.4-48.4)**
Odds ratio (95% CI); p value	2.69 (1.13-6.42); 0.026	
ORR at 6 months, %	66.7	47.9
PFS, HR (95% CI); p value[11]	0.33 (0.17-0.67); 0.0019	
OS, HR (95% CI); p value[11]	0.49 (0.26-0.92); 0.0271	

RP2D = RP3D in the phase 3 TrilynX study

Manageable toxicity, high relative dose intensity, low fraction of dose reduction = **acceptable safety profile**

CR, complete response; **CRT,** chemoradiotherapy; **HR,** hazard ratio; **LRC,** locoregional control; **MTD,** maximum tolerated dose; **ORR,** objective response rate; **OS,** overall survival; **PD,** progressive disease; **PFS,** progression-free survival; **PR,** partial response; **RP2D,** recommended phase 2 dose; **RP3D,** recommended phase 3 dose.

Based on the encouraging phase 2 data, the phase 3 TrilynX study was designed to further assess the efficacy and safety of xevinapant in combination with CRT in patients with LA SCCHN

We present **the integrated rationale for the recommended phase 3 dose (RP3D) of xevinapant (200 mg/day)** based on the available clinical and preclinical data, and modeling and simulation

TRILYNX STUDY DESIGN
A publication describing the TrilynX study design can be accessed through this **hyperlink** or QR code.

METHODS

Holistic integration of preclinical pharmacology, clinical efficacy, and safety profiles in phases 1 and 2, clinical pharmacokinetics (PK)/pharmacodynamics (PD) in phase 1, population PK (popPK), and exposure-response (E-R) analyses for efficacy and safety

Further detail for the methods can be accessed through this **hyperlink** or QR code.

1. A three-compartment linear popPK model with delayed absorption was developed using data from 2 clinical studies, NCT01078649 (solid tumors)[12] and NCT02022098 (LA SCCHN)[8,10]
2. PopPK/PD model simulations (500 virtual patients) were developed using data from study NCT01078649[12] to predict cIAP1 degradation for 100, 150, and 200 mg/day on days 1-14 every 3 weeks (**Figure 2**)
3. Individual exposures were derived using the popPK model. Exposure distributions/summary statistics for the different dosing regimens were generated using the popPK model and compared with preclinical in vitro and in vivo data (**Table 2A** and **2B,** respectively)
4. E-R analyses for efficacy and safety were conducted using the pooled data set: phase 1 dose-escalation and randomized phase 2 study (NCT02022098);[8,10] n=62 treated with xevinapant (**Figure 3A** and **3B**, respectively)

Table 1. E-R analysis approach

Endpoint	Analysis	Exposure metric
Efficacy: LRC at 18 months, OR, CR	Logistic regression	AUC_{1C}
Efficacy: PFS, OS, duration of LRC	Time to event	
Safety: Grade ≥3 TEAEs, TEAEs leading to discontinuation, grade ≥3 ALT elevation, grade ≥2 AST elevation, grade ≥2 amylase elevation, grade ≥2 lipase elevation, grade ≥3 neutropenia, grade ≥3 skin toxicity, grade ≥3 mucositis, grade ≥3 dysphagia, composite endpoints: grade ≥3 mucositis and/or dysphagia, grade ≥3 dysphagia and/or odynophagia	Logistic regression	AUC_{3C}
Safety: incidence and time to first dose reduction	Graphical	

ALT, alanine aminotransferase; **AST,** aspartate aminotransferase; AUC_{1c}, area under the concentration-time curve in plasma over cycle 1; AUC_{3c}, area under the concentration-time curve in plasma over cycles 1-3; **CR,** complete response; **LRC,** locoregional control; **OR,** objective response; **OS,** overall survival; **PFS,** progression-free survival; **TEAE,** treatment-emergent adverse event

RESULTS

- Maximal (≈90%) cIAP1 degradation in peripheral blood mononuclear cells during dosing (days 1-14) with 100 to 200 mg/day (**Figure 2**)
 - 100 to 200 mg/day is a pharmacologically active dose range, supporting individual dose reductions to 150 and 100 mg/day as needed to manage adverse events in individual patients
- Partial degradation in the off-window (days 15-20) with a trend of dose response at day 20 over the 100- to 200-mg/day dose range (**Figure 2**)

Figure 2. PopPK/PD modeling of phase 1 data of xevinapant

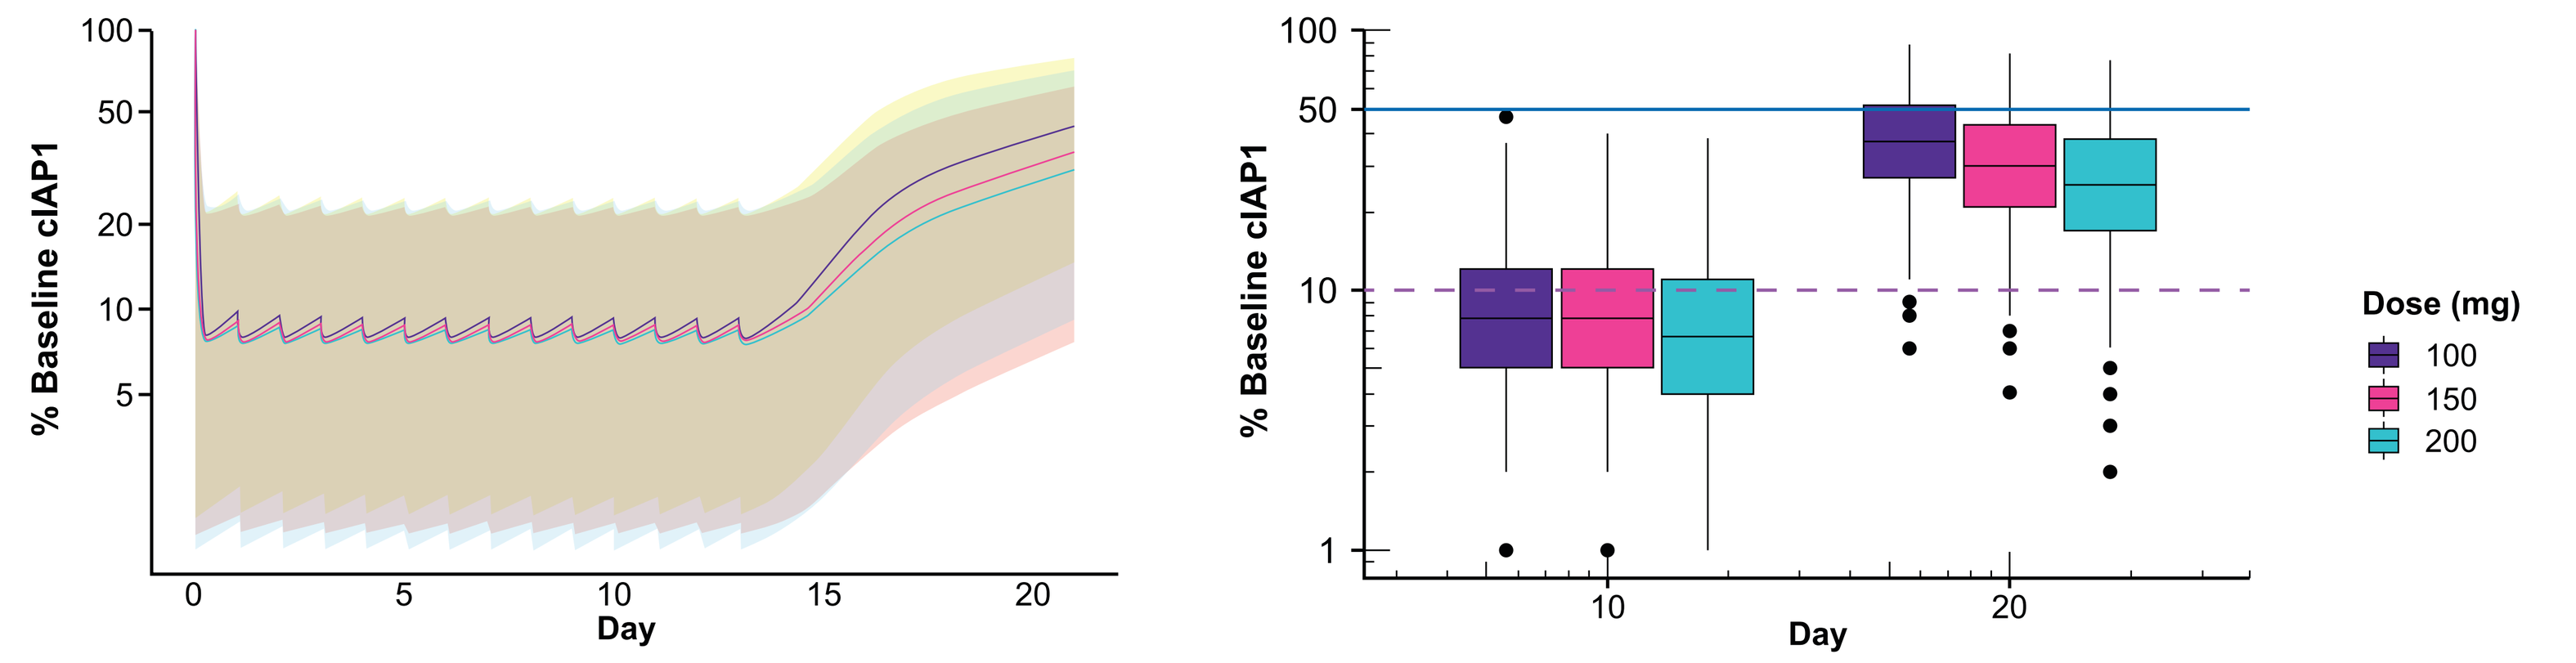
% Baseline cIAP1
Day
Dose (mg)
100
150
200

Typical response is represented by solid lines; 95% prediction intervals are the shaded areas
cIAP1, cellular IAP 1; **popPK,** population pharmacokinetics.

- With 200 mg/day dose
 - 95.2% of the patients had popPK-derived trough concentration at steady state ($C_{trough,ss}$) above in vitro IC_{50} (90 nM) for cIAP1 degradation (**Table 2A**)
 - Free average daily concentration at steady state ($C_{avg,ss}$) in humans overlapped with that at efficacious dose range in the mouse SCCHN model (**Table 2B**)

Table 2. Nonclinical/translational pharmacology

A. Percentage of patients with popPK-derived $C_{trough,ss}$ above in vitro IC_{50} for cIAP1 degradation

Daily dose	Participants, %
100 mg	69.2
150 mg	88.2
200 mg	95.2
300 mg	99.4
400 mg	99.8

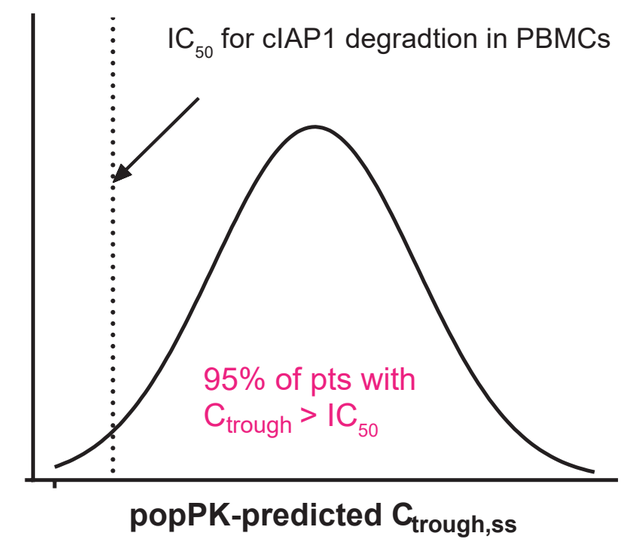
IC_{50} for cIAP1 degradtion in PBMCs
95% of pts with C_{trough} > IC_{50}
popPK-predicted $C_{trough,ss}$

cIAP1, cellular IAP 1; $C_{trough,ss}$, trough concentration at steady state; IC_{50}, half maximal inhibitory concentration; **PBMC,** peripheral blood mononuclear cell; **popPK,** population pharmacokinetics.

IC_{50}=90 nM

B. Human popPK-derived vs mouse free $C_{avg,ss}$

Species	Dose/day	Free in vivo $C_{avg,ss}$, nM
Human Median (5th-95th percentile)	200 mg	125 (60-252)
	150 mg	93 (45-190)
	100 mg	62 (30-127)
Mouse*	30-100 mg/kg	71-237

*calculated using mean concentration from n=3 mice/per time point

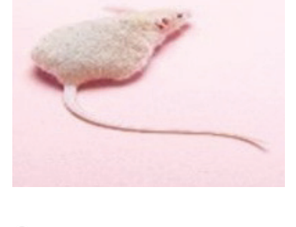

SCCHN xenograft model (SQ20B)
Xevinapant + 2Gy RT for 14 days

Human and mouse fraction unbound estimates of 13.8% and 18.4%, respectively, were used to calculate free exposures. Efficacious dose range of 30 to 100 mg/kg/day in mice in combination with RT was based on tumor growth inhibition and survival compared with RT alone. Mouse PK is from Cai et al, 2011.[13]

$C_{avg,ss}$, average daily concentration at steady state; **popPK,** population pharmacokinetics; **RT,** radiotherapy; **SCCHN,** squamous cell carcinoma of the head and neck.

Figure 3A. Xevinapant exposure-efficacy analysis

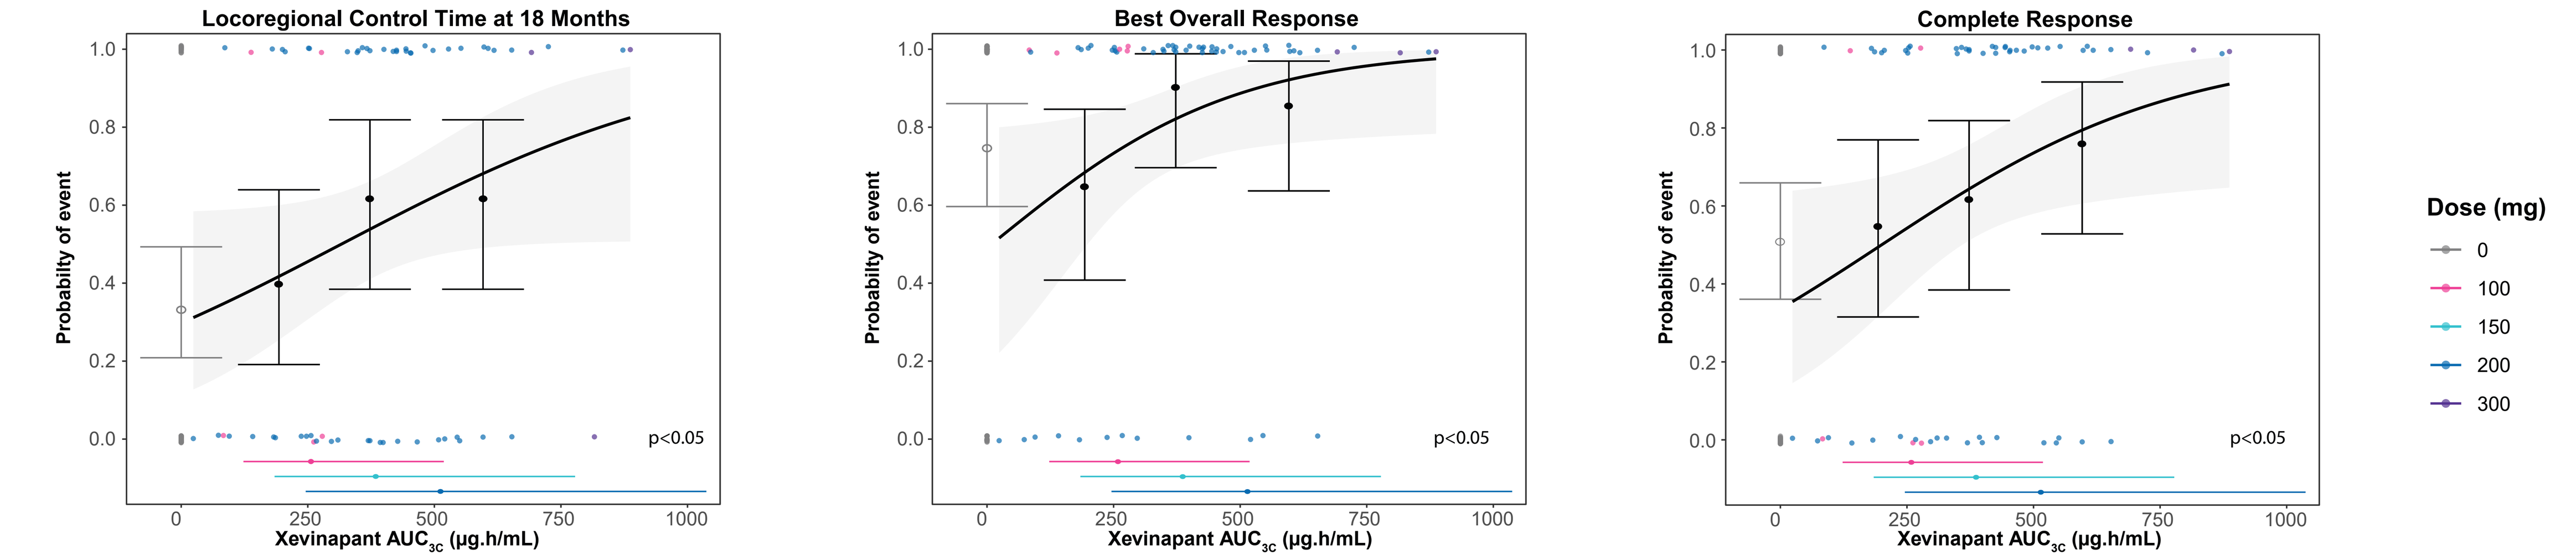
Locoregional Control Time at 18 Months
Best Overall Response
Complete Response
Probability of event
p<0.05
Xevinapant AUC_{3C} (µg.h/mL)
Dose (mg)
0
100
150
200
300

Pink, **teal**, and **blue** dots and horizontal error bars: the typical predictions and 95% prediction interval for AUC_{3C} from the popPK model for 100-, 150-, and 200-mg/day doses, assuming full compliance. **Gray**, **pink**, **blue**, and **purple** dots: individual data with 0 (placebo; n=47) or 100 (n=5), 200 (n=53), or 300 (n=5) mg/day, respectively, for which AUC_{3C} was derived using actual administered doses. The predictions of the binary logistic regression model (solid black line) and 95% CI (shaded gray area) for LRC at 18 months, ORR, and CR probabilities are compared with the observed incidence data by tertiles of AUC_{3C}. Closed and open circles and error bars represent median and 95% CI in exposure tertiles of xevinapant-treated patients and the control group, respectively.
AUC_{3C}, area under the concentration-time curve in plasma in cycle 3; **CR,** complete response; **LRC,** locoregional control; **ORR,** objective response; **OS,** overall survival; **popPK,** population pharmacokinetics.

- LRC at 18 months, OR, and CR incidence showed a statistically significant increase with increasing exposures (**Figure 3A**)

Note: No statistically significant E-R relationship was detected for duration of LRC, PFS, OS; but time to event was not yet mature at the data cutoff

Figure 3B. Xevinapant exposure-safety analysis

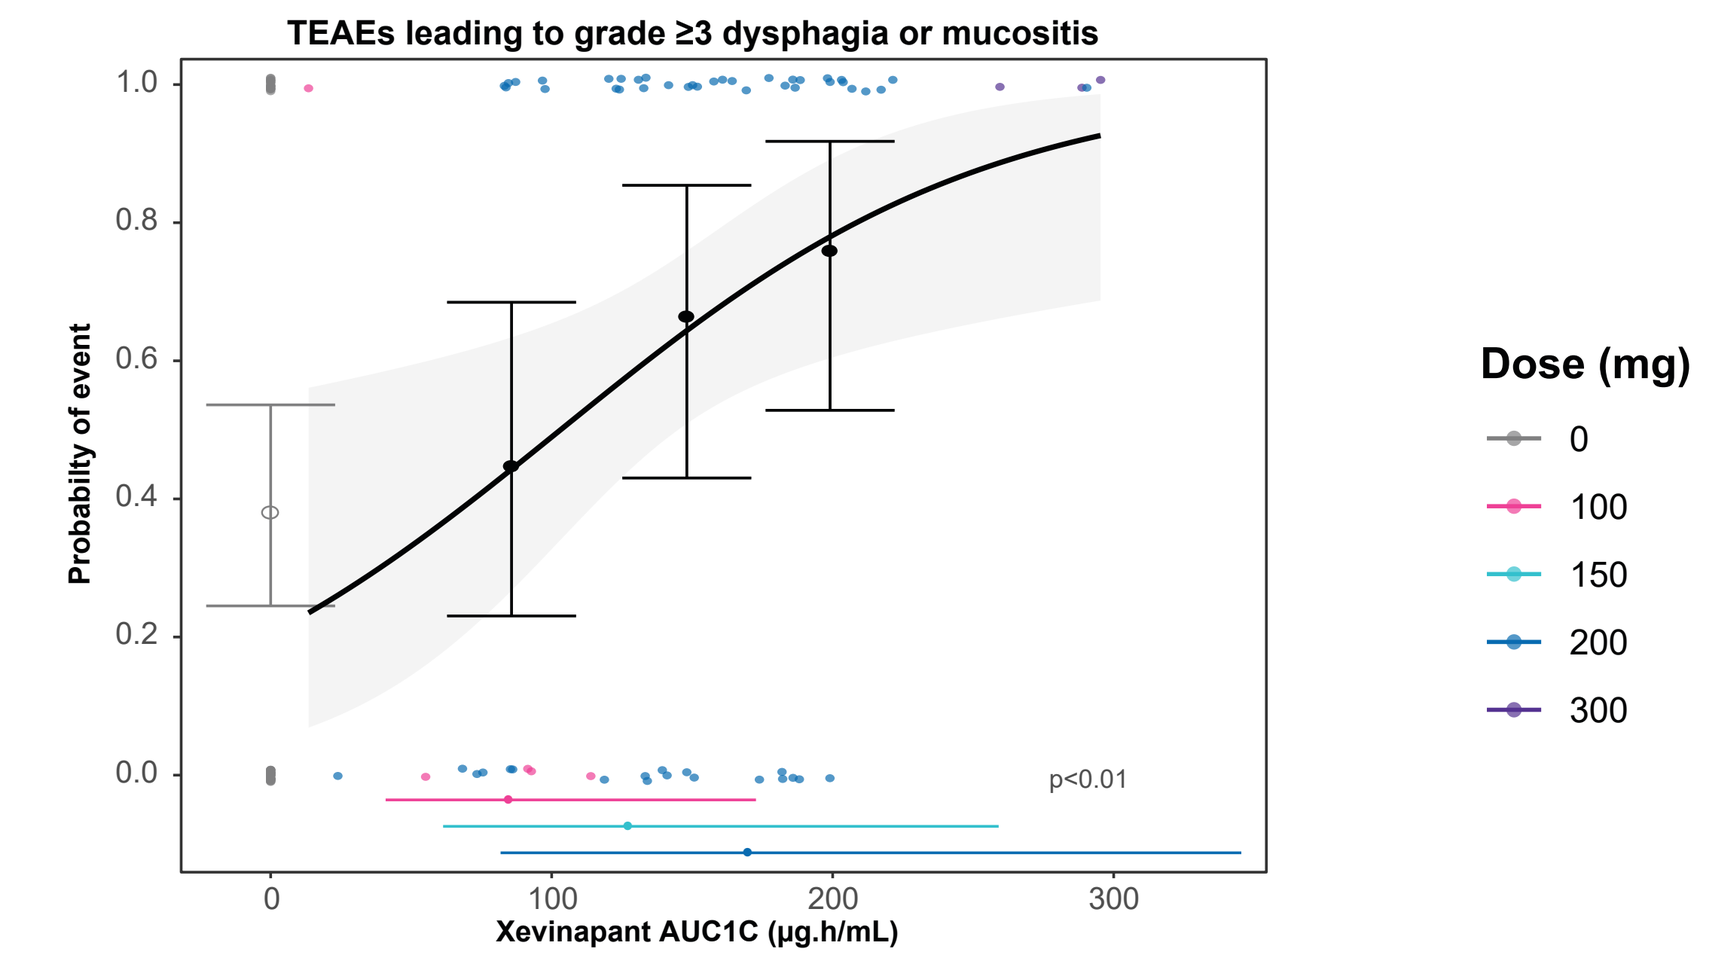
TEAEs leading to grade ≥3 dysphagia or mucositis
Probability of event
p<0.01
Xevinapant AUC1C (µg.h/mL)
Dose (mg)
0
100
150
200
300

Pink, **teal**, and **blue** dots and horizontal error bars: the typical predictions and 95% prediction interval for AUC_{1C} from the popPK model for 100-, 150-, and 200-mg/day doses, assuming full compliance. **Gray**, **pink**, **blue**, and **purple** dots: individual data with 0 (placebo; n=47) or 100 (n=5), 200 (n=53), or 300 (n=5) mg/day, respectively, for which AUC_{1C} was derived using actual administered doses. The predictions of the binary logistic regression model (solid black line) and 95% CI (shaded gray area) for probabilities of AE are compared with the observed incidence data by tertiles of AUC_{3C}. Closed and open circles and error bars represent median and 95% CI in exposure tertiles of xevinapant-treated patients and the control group, respectively.
AE, adverse event; AUC_{1C}, area under the concentration-time curve in plasma in cycle 1; AUC_{3C}, area under the concentration-time curve in plasma in cycle 3; **popPK,** population pharmacokinetics; **TEAE,** treatment emergent adverse event.

- Probability of adverse events for the composite safety endpoint (grade ≥3 mucositis or dysphagia) showed a statistically significant increase with increasing exposures (**Figure 3B**)
 - This endpoint is directly linked to quality of life
 - Consistent with the radiosensitizing mode of action of xevinapant
- E-R was not discernable for all other binary safety endpoints examined (p>0.05)
 - A trend was observed for mucositis inflammation (p=0.065)
- A relatively small fraction of patients had dose reductions
 - 20.8% in xevinapant arm vs 14.9% in placebo
- There was no association of exposure and incidence of dose reduction or time to first dose reduction

CONCLUSIONS

The holistic integration of preclinical pharmacology, clinical efficacy and safety profiles, clinical PK/PD, popPK, and E-R analyses support the RP3D selection of xevinapant at 200 mg/day administered on days 1 to 14 every 3 weeks with concurrent CRT, allowing for successive dose reductions to 150 mg and 100 mg for the management of toxicities

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from AACR and the author of this poster

Correspondence: Yulia Vugmeyster, **yulia.vugmeyster@emdserono.com**

References: 1. Sung H, et al. *CA Cancer J Clin.* 2021;71(3):209-49. **2.** Ang KK, et al. *Oncologist.* 2008;13(8):899-910. **3.** Machiels JP, et al. *Ann Oncol.* 2020;31(11):1462-75. **4.** NCCN Clinical Practice Guidelines in Oncology. Head and Neck Cancers. V1. 2021. **5.** Brunckhorst MK, et al. *Cancer Biol Ther.* 2012;13(9):804-11. **6.** Matzinger O, et al. *Radiother Oncol.* 2015;116(3):495-503. **7.** Gomez-Roca C, et al. *Cancer Res.* 2019;79(Suppl 13). Abstract 5001. **8.** Sun XS, et al. *Lancet Oncol.* 2020;21(9):1173-87. **9.** Dougan M, et al. J *Exp Med.* 2010;207(10):2195-2206. **10.** Le Tourneau C, et al. *Clin Cancer Res.* 2020;26(24):6429- 36. **11.** Tao Y, et al. Manuscript in development. **12.** Hurwitz H, et al. *Cancer Chemother Pharmacol.* 2015;75(4):851-9. **13.** Cai Q, et al. *J Med Chem.* 2011;54(8):2714-26. **Acknowledgments:** The authors would like to thank the patients and their families, the investigators, coinvestigators, and study teams at each of the participating centers. Medical writing support was provided by ClinicalThinking, and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). **Disclosures: Y. Vugmeyster, A. Ravula, K. Goteti, and K. Venkatakrishnan** are employees of EMD Serono, Billerica, MA, USA. **E. Rouits** is an employee of Debiopharm International SA, Lausanne, Switzerland. **P. M. Diderichsen and H. J. Kleijn** are employees of Certara Strategic Consulting, Breda, the Netherlands. **A. Schroeder** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **Correspondence:** Yulia Vugmeyster, **Yulia.Vugmeyster@emdserono.com**

Abstract No. 5424. Presented at the AACR Annual Meeting 2022, April 8-13, 2022; New Orleans, LA, and Virtual.