TrilynX: A phase 3 trial of xevinapant + concurrent chemoradiotherapy (CRT) for locally advanced head and neck cancer

J. D. Schoenfeld,[1] E. E. W. Cohen,[2] C. Nutting,[3] L. F. Licitra,[4] B. Burtness,[5] M. Omar,[6] F. Bouisset,[6] H. Nauwelaerts,[6] Y. Urfer,[6] C. Zanna,[6] J. Bourhis[7]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]University of California, San Diego, La Jolla, CA, USA; [3]Royal Marsden NHS Foundation Trust, London, UK; [4]Fondazione IRCCS Istituto Nazionale dei Tumori and University of Milan, Milan, Italy; [5]Yale School of Medicine and Yale Cancer Center, New Haven, CT, USA; [6]Debiopharm International SA, Lausanne, Switzerland; [7]Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland

BACKGROUND

Squamous cell carcinoma of the head and neck (SCCHN) is the **eighth** most common cancer worldwide[1]

~60% of patients are diagnosed with locally advanced disease,[2] which in many cases cannot be removed by surgical resection[3]

High-dose cisplatin with concurrent radiotherapy (CRT) is the standard of care for patients with unresected locally advanced (LA) SCCHN[3,4]

Xevinapant is a first-in-class, potent, small molecule antagonist of inhibitor of apoptosis proteins (**IAPs**), formulated as an **oral solution**

Xevinapant is designed to **restore sensitivity of cancer cells to apoptosis** and to **enhance the effects of other anticancer treatments**, such as chemotherapy and radiotherapy[5-8] (**Figure 1**)

Figure 1. Xevinapant mode of action

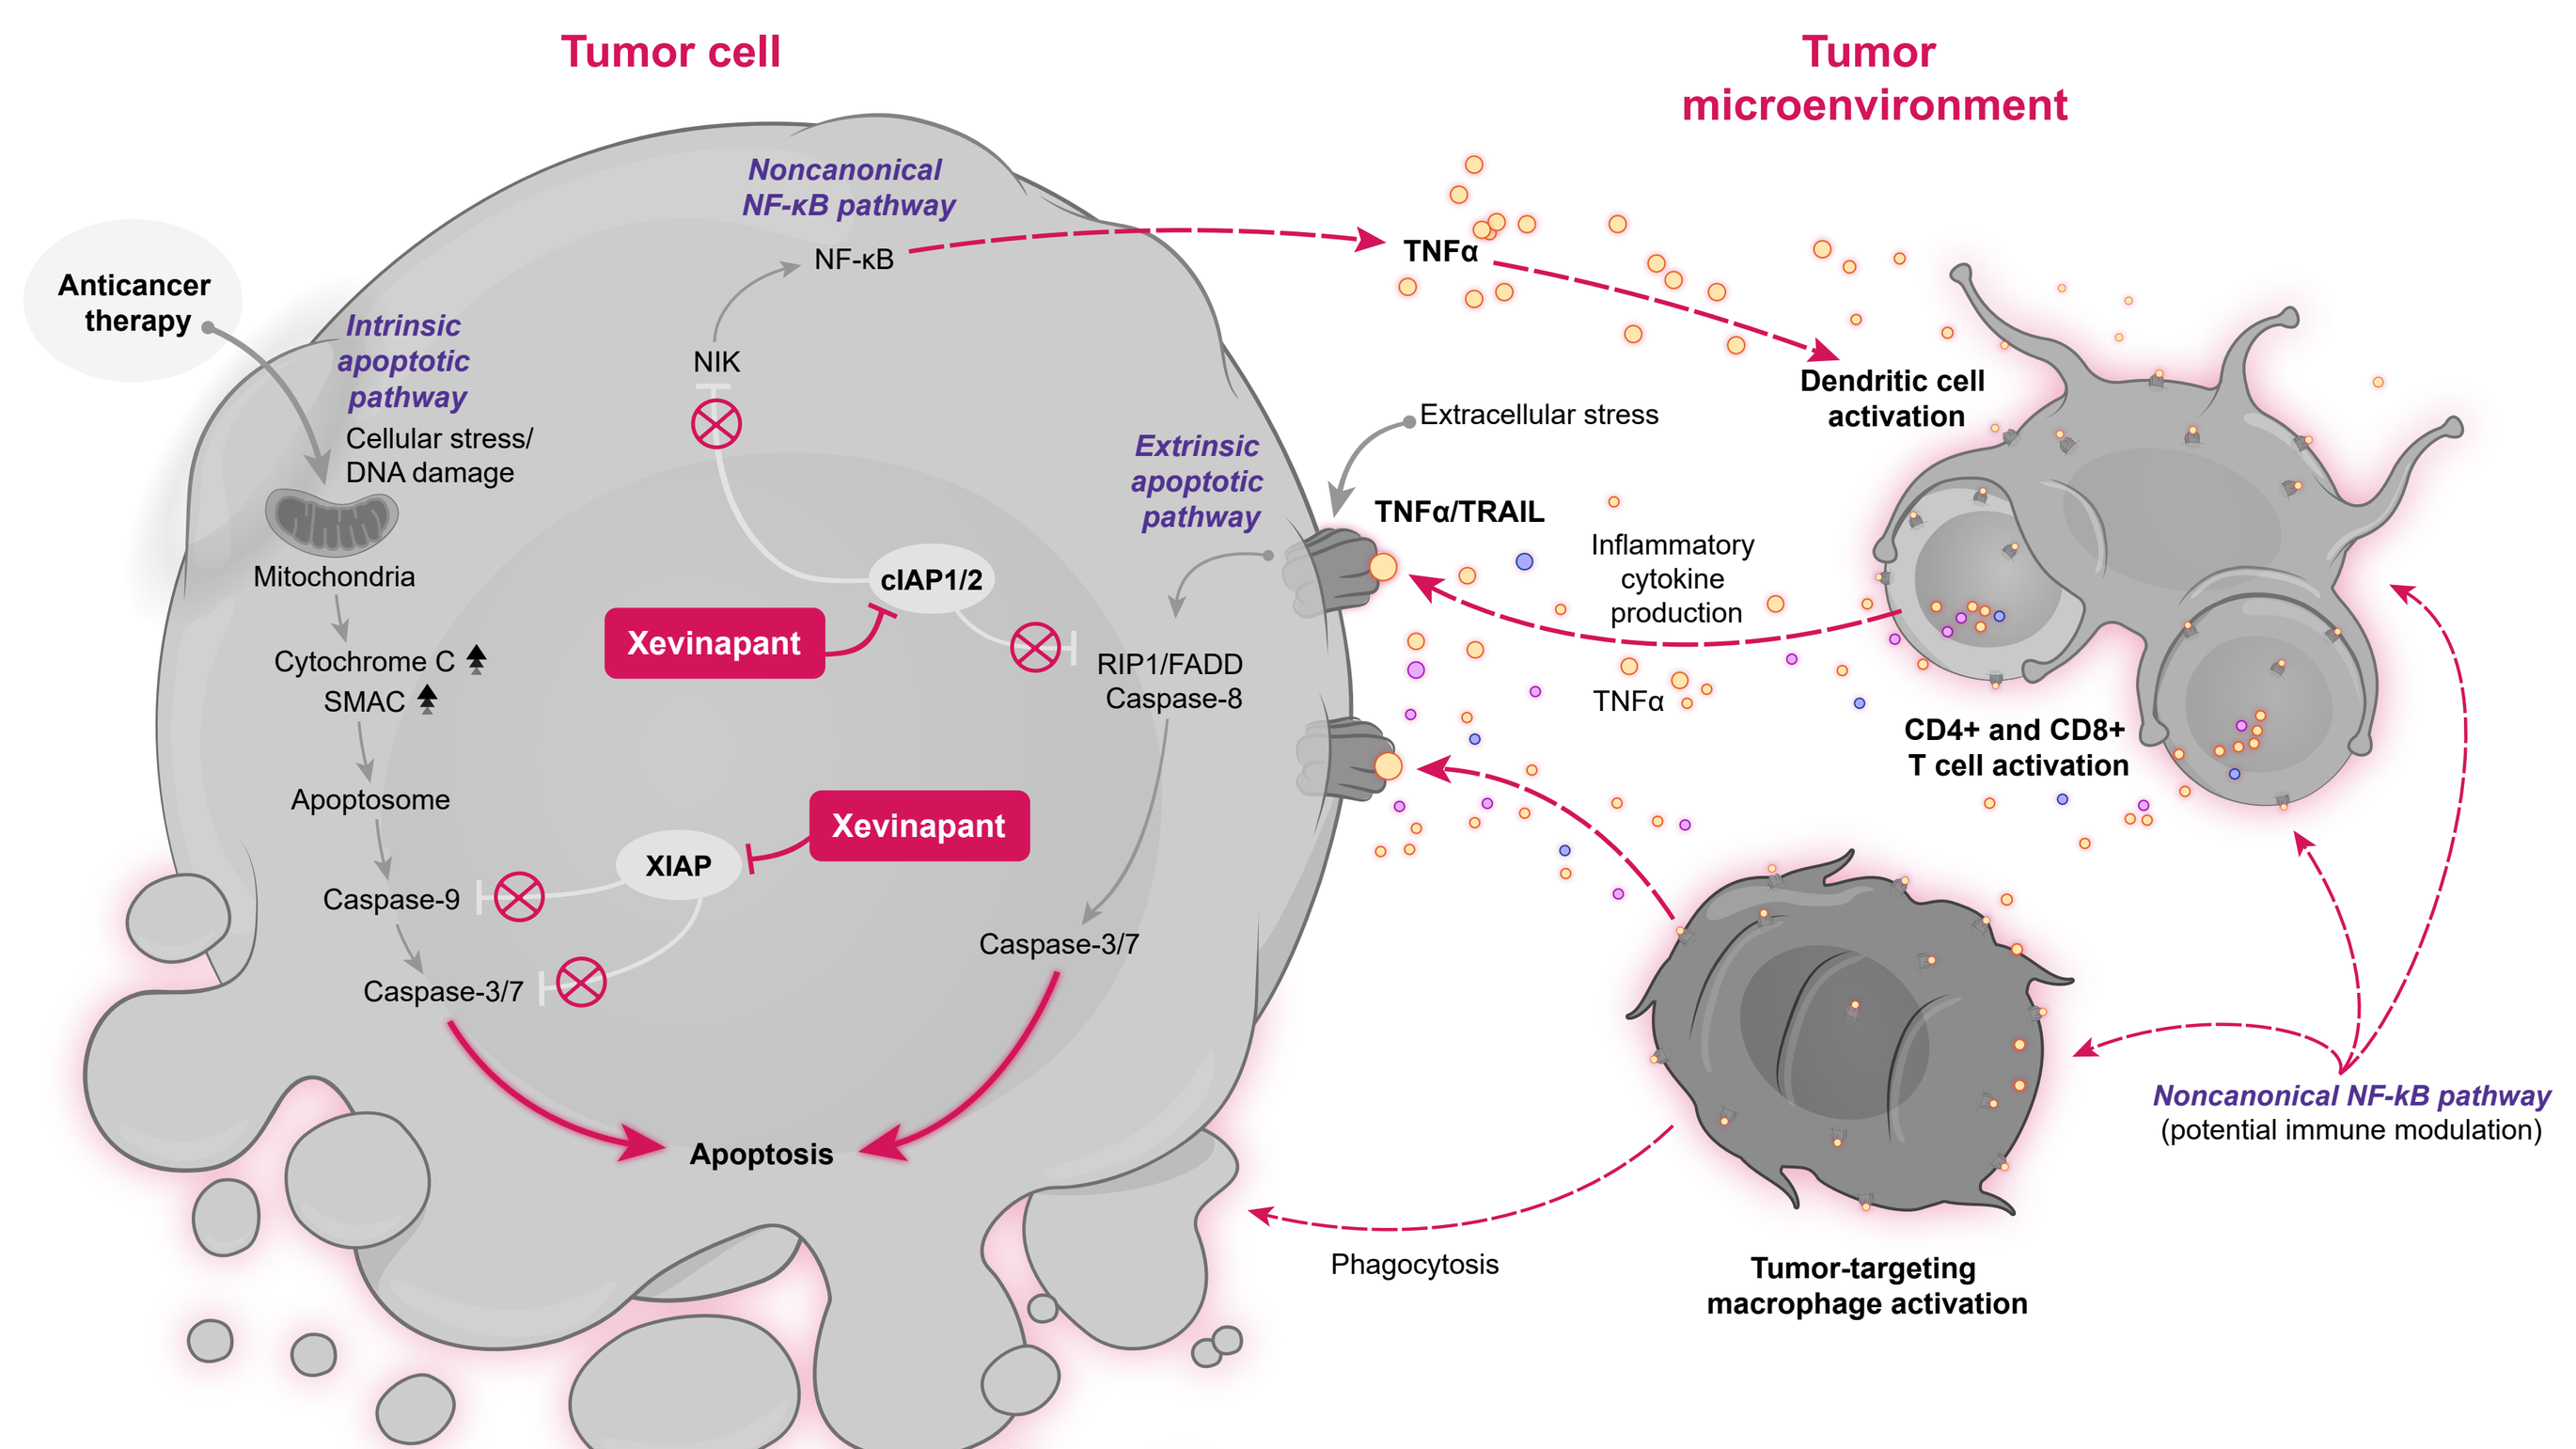

Tumor cell
Tumor microenvironment
Anticancer therapy
Noncanonical NF-κB pathway
NF-κB
TNFα
Intrinsic apoptotic pathway
Cellular stress/ DNA damage
NIK
Mitochondria
Cytochrome C
SMAC
Apoptosome
Caspase-9
Caspase-3/7
XIAP
Xevinapant
cIAP1/2
Xevinapant
Extrinsic apoptotic pathway
RIP1/FADD Caspase-8
Caspase-3/7
Apoptosis
Extracellular stress
TNFα/TRAIL
Inflammatory cytokine production
TNFα
Dendritic cell activation
CD4+ and CD8+ T cell activation
Noncanonical NF-κB pathway (potential immune modulation)
Phagocytosis
Tumor-targeting macrophage activation

Xevinapant is thought to:

- **Restore sensitivity to apoptosis** in cancer cells by blocking X-linked IAP and cellular IAPs 1 and 2 (cIAP1/2) leading to activation of caspases downstream of the intrinsic and extrinsic apoptotic pathways[5-8]
- **Enhance inflammatory antitumor responses** by immune cells of the tumor microenvironment by activating noncanonical NF-κB signaling via blocking cIAP1/2 downstream of the TNF receptor[6,7,9]

cIAP1/2, cellular IAPs 1 and 2; **FADD**, fas-associated protein with death domain; **IAP**, inhibitors of apoptosis proteins; **NF-κB**, nuclear factor kappa-light-chain-enhancer of activated B cells; **NIK**, NF-κB-inducing kinase; **RIP1**, receptor interacting serine/threonine kinase 1; **SMAC**, second mitochondria-derived activator of caspase; **TNFα**, tumor necrosis factor alpha; **TRAIL**, TNF-related apoptosis-inducing ligand; **XIAP**, X-linked IAP.

BACKGROUND (CONT)

In a randomized double-blind, multicenter phase 2 study, **xevinapant** plus CRT significantly increased locoregional control (LRC; primary endpoint) at 18 months versus **placebo** plus CRT[8]

54% in the xevinapant arm had LRC

33% in the placebo arm had LRC

At **3-year follow-up** median overall survival (OS) and progression-free survival (PFS) were significantly improved with **xevinapant** plus CRT vs **placebo** plus CRT:[10]

51% **Reduction in risk of death with xevinapant (Hazard ratio [HR] 0.49, 95% CI:0.26-0.92; p=.0271)**

67% **Reduction in risk of disease progression or death with xevinapant (HR 0.33, 95% CI: 0.17-0.67; p=.0019)**

At the primary analysis, grade ≥3 adverse events were reported in 85% vs 87% of patients in the **xevinapant** and **placebo** arms, respectively.[8] At 3-year follow-up, late onset toxicities of grade ≥3 were reported in 14 patients (29%) in the xevinapant arm versus 15 (32%) in the placebo arm[10]

Based on the encouraging phase 2 data, the TrilynX study was designed to further assess the efficacy and safety of **xevinapant** in patients with LA SCCHN

METHODS

TrilynX (NCT04459715) is a randomized, double-blind, placebo-controlled, international, phase 3 study comparing **xevinapant** plus CRT with **placebo** plus CRT in ~700 patients (**Figure 2**)

- The primary endpoint of the study is event-free survival (EFS), as assessed by a blinded independent review committee (BIRC)

Eligible patients are aged ≥18 years, have histologically confirmed, unresected cancer of the oropharynx (p16-negative by immunohistochemistry [IHC]), hypopharynx or larynx, and are suitable for definitive CRT (see **Table 1** for a summary of eligibility criteria)

- Patients with cisplatin-related toxicity after the first dose of cisplatin may be switched to carboplatin (AUC, area under the curve = 5 or 4 on day 2 of each subsequent cycle, depending on the toxicity observed)

Figure 2. TrilynX clinical trial design

Histologically confirmed unresected locally advanced SCCHN

Stage III, IVa, or IVb tumor of the oropharynx, hypopharynx, or larynx

ECOG PS 0 or 1

N≈700

R 1:1*

Combination treatment (xevinapant arm)
- **Xevinapant** 200 mg/day (days 1-14, Q3W for 3 cycles)
- **Cisplatin** 100 mg/m² Q3W for 3 cycles at day 2†
- **IMRT** 70 Gy (2 Gy/day, 5 days/week for 7 weeks)

→ **Monotherapy**
- **Xevinapant** 200 mg/day (days 1-14, Q3W for 3 cycles)

Combination treatment (placebo arm)
- **Matched placebo** (days 1-14, Q3W for 3 cycles)
- **Cisplatin** 100 mg/m² Q3W for 3 cycles at day 2†
- **IMRT** 70 Gy (2 Gy/day, 5 days/week for 7 weeks)

→ **Monotherapy**
- **Matched placebo** (days 1-14, Q3W for 3 cycles)

*Stratification factors: region (North America vs Western Europe vs rest of the world); primary tumor site (larynx vs other); lymph node involvement (N0-1 vs N2 vs N3); and tumor size (T4 vs other).
†All patients will receive ≥1 cycle of cisplatin; for patients not eligible to continue with cisplatin after cycle 1, a switch to carboplatin (AUC 5) is allowed.
ECOG PS, European Cooperative Oncology Group performance status; **IMRT**, intensity-modulated radiotherapy; **Q3W**, every 3 weeks; **R**, randomization; **SCCHN**, squamous cell carcinoma of the head and neck.

METHODS (CONT)

Table 1. Trial endpoints

Table 1. Trial endpoints
Primary endpoint
• **EFS by BIRC:** Time from randomization to the occurrence of death, clinical or radiological progression, primary treatment failure, radiological or clinical relapse after achieving a loco-regional CR, or the occurrence of secondary cancers unless pathological findings exclude squamous histology
Secondary endpoints
• **OS:** Time from randomization to death due to any cause
• **DOR:** Time from the first evidence of response (CR or PR, per BIRC according to RECIST v1.1) to the first occurrence of progression (radiological or clinical, per BIRC) or death from any cause
• **PFS:** Time from randomization to radiological or clinical disease progression or death from any cause, per BIRC
• **ORR:** Proportion of patients with CR or PR by RECIST v1.1, per BIRC
• **Locoregional control:** Time from randomization to the first occurrence of progression at the site of the primary tumor or the locoregional lymph nodes per RECIST v1.1 or clinical assessment by BIRC
• **Safety:** Incidence and severity of AEs, serious AEs and AEs of special interest, changes in laboratory values, vital signs, and electrocardiograms according to NCI-CTCAE v5.0
• **Health related quality of life:** Assessed using the EORTC quality-of-life core questionnaire (QLQ-C30), with head and neck specific symptoms, such as pain and swallowing, assessed using the EORTC head-and-neck module questionnaire (QLQ-H&N35)
Exploratory endpoints
• Pharmacokinetics
• Proportion of patients with definitive tracheostomy
• Proportion of patients with enteral and parenteral feeding
• Patient-reported outcomes
• Healthcare resource utilization
• Biomarker assessments
• Time to distant metastasis, assessed by BIRC, according to RECIST v1.1

AE, adverse event; **BIRC**, blinded independent review committee; **CR**, complete response; **DOR**, duration of response; **EORTC**, European Organisation for Research and Treatment of Cancer; **EFS**, event-free survival; **NCI-CTCAE**, National Cancer Institute Common Terminology Criteria for Adverse Events; **ORR**, objective response rate; **OS**, overall survival; **PFS**, progression-free survival; **PR**, partial response; **RECIST**, Response Evaluation Criteria in Solid Tumors.

Table 2. Summary of eligibility criteria

Table 2. Summary of eligibility criteria
Key inclusion criteria
• Age ≥18 years
• Histologically confirmed, unresected, LA SCCHN, suitable for definitive CRT in at least one of the following sites; oropharynx*, hypopharynx, larynx
• ECOG PS 0 or 1
• Evaluable tumor burden based on RECIST v1.1
• Adequate hematologic function (ANC ≥1500 cells/μL, platelets ≥100,000 cells/μL, hemoglobin ≥9.0 g/dL)
• Adequate renal function (eGFR ≥60 mL/min/1.73m² using the CKD-EPI creatinine formula)[11]
• Adequate hepatic function (ALT and AST ≤3.0 x ULN, total bilirubin ≤1.5 x ULN†)
Key exclusion criteria
• Primary tumor of nasopharyngeal, paranasal sinuses, nasal or oral cavity, salivary, thyroid or parathyroid gland pathologies, skin or unknown primary site
• Prior definitive or adjuvant RT and/or radical surgery to the head and neck region that may jeopardize the primary tumor irradiation plan, or any other prior SCCHN systemic treatment
• Metastatic disease

*Patients with oropharyngeal tumors only must be HPV-negative as determined by p16 expression using IHC.
†Up to 2.0 × ULN if the direct bilirubin level is normal and elevation is limited to indirect bilirubin.
ALT, alanine aminotransferase; **ANC**, absolute neutrophil count; **AST**, aspartate aminotransferase; **CKD-EPI**, Chronic Kidney Disease Epidemiology Collaboration; **CRT**, chemoradiotherapy; **ECOG PS**, Eastern Cooperative Oncology Group performance status; **eGFR**, estimated glomerular filtration rate; **HPV**, human papilloma virus; **IHC**, immunohistochemistry; LA, locally advanced; **RECIST**, response evaluation criteria in solid tumors; **RT**, radiotherapy; **SCCHN**, squamous cell carcinoma of the head and neck; **ULN**, upper limit of normal.

METHODS (CONT)

Assessments and statistical analysis

- All patients will be assessed at the end of therapy visit (20 ± 1 weeks) using 18F-fluorodeoxyglucose positron emission tomography/computed tomography scans. Computed tomography scans or MRI will be assessed every 3 months up to 3 years, then every 6 months up to 5 years to determine overall clinical outcomes and the need for nodal dissection
- Safety will be assessed throughout the study and graded using National Cancer Institute Common Terminology Criteria for Adverse Events v5.0
- HPV-status will be assessed by p16 IHC in oropharyngeal tumors only
- The primary analysis population for analyses of efficacy and health-related quality of life will include all randomized patients
- The safety analysis population will include all patients who receive ≥1 dose of study medication
- The population size (~700 patients) of the study is driven by the primary endpoint of EFS
 - 90% power, with a one-sided type I error of 2.5%, to detect prolonged EFS (assessed by BIRC) with xevinapant plus CRT
 - Estimated treatment effect HR of 0.73; assuming a median EFS in the control group of 17 months, 429 EFS events will be required
- Secondary endpoints will be tested with a hierarchical strategy that preserves the type I error rate at 2.5% (one-sided)

Current enrollment status (February 2022)

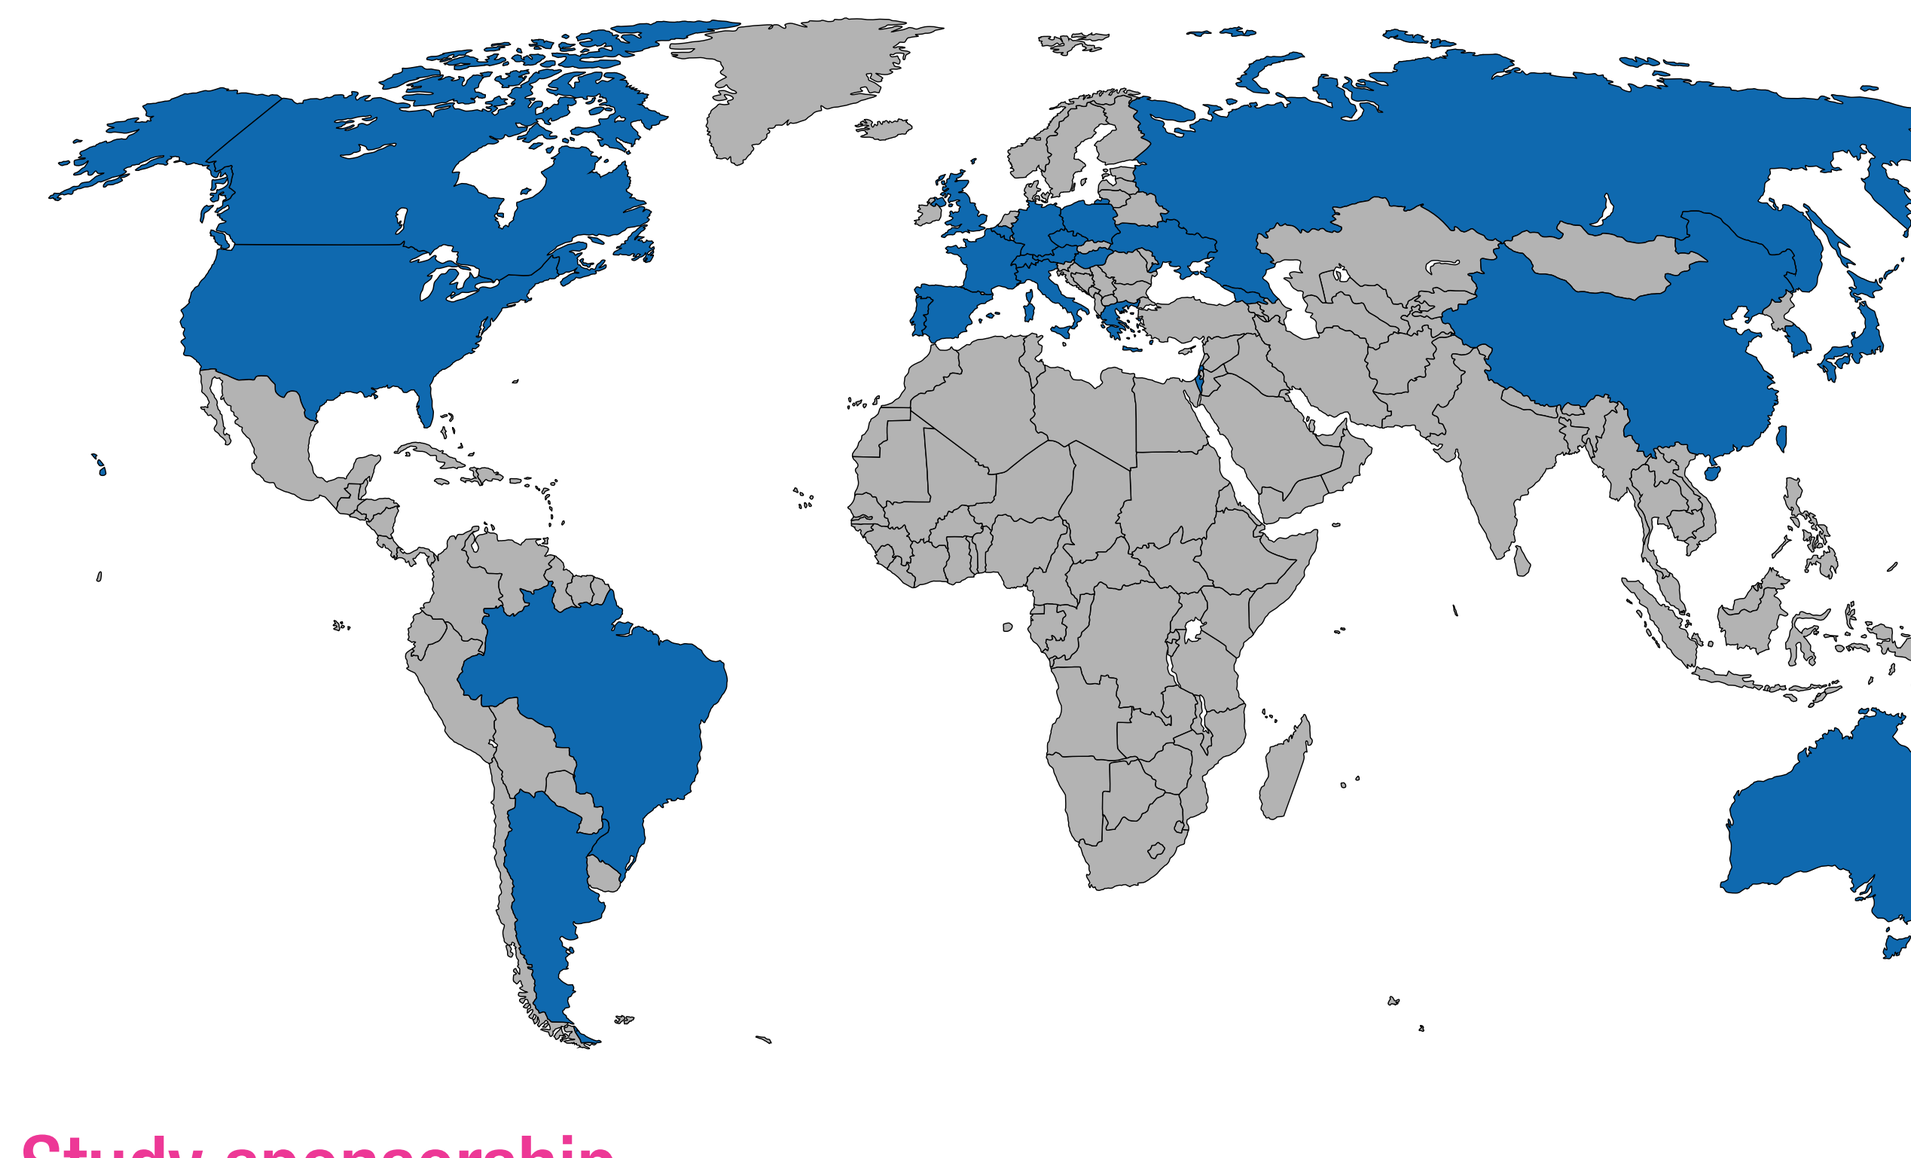

The study is ongoing and recruiting in 26 countries worldwide:
Argentina, Australia, Austria, Belgium, Brazil, Canada, China, Czech Republic, France, Georgia, Germany, Greece, Hungary, Israel, Italy, Japan, Poland, Portugal, Republic of Korea, Russian Federation, Spain, Switzerland, Taiwan, Ukraine, United Kingdom, and United States

Study sponsorship

The principal investigator of this study is Dr Jean Bourhis. This study is sponsored by EMD Serono Research & Development Institute, Inc., an affiliate of Merck KGaA, Darmstadt, Germany (Clinical Lead: **almudena.rodriguez@emdgroup.com**).
Details can be found on clinicaltrials.gov.

GET POSTER PDF

Copies of this poster obtained through this hyperlink or quick response (QR) code are for personal use only and may not be reproduced without permission from ASTRO MDT and the author of this poster

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app or click here to view a plain language summary of the accepted scientific abstract

References: 1. Sung H, et al. CA *Cancer J Clin.* 2021;71(3):209-49. **2.** Ang KK, et al. *Oncologist.* 2008;13(8):899-910. **3.** Machiels JP, et al. *Ann Oncol.* 2020;31(11):1462-475. **4.** NCCN Clinical Practice Guidelines in Oncology. Head and Neck Cancer. v1.2021. **5.** Brunckhorst MK, et al. *Cancer Biol Ther.* 2012;13(9):804-11. **6.** Matzinger O, et al. *Radiother Oncol.* 2015;116(3):495-503. **7.** Gomez-Roca C, et al. *Cancer Research.* 2019;79(suppl_13):Abstract 5001. **8.** Sun XS, et al. *Lancet Oncol.* 2020;21(9):1173-87. **9.** Dougan M, et al. *J Exp Med.* 2010;207(10):2195-206. **10.** Sun XS, et al. Manuscript in preparation. **11.** Levey AS, et al. *Ann Intern Med* 2009;150(9);604-12.

Acknowledgments: The authors would like to thank the patients and their families, the investigators, coinvestigators, and study teams at each of the participating centers. Medical writing support was provided by Sophie Saunders of ClinicalThinking, and was funded by Merck KGaA, Darmstadt, Germany. **Disclosures: J. D. Schoenfeld** reports serving in consulting or advisory roles for ACI Clinical, Catenion, Debiopharm, Immunitas, LEK, Tilos Therapeutics, Astellas and Stimit; receiving research funding from BMS, the healthcare business of Merck KGaA, Darmstadt, Germany and Regeneron; and providing expert testimony for Heidell, Pittoni, Murphy & Bach, Kline & Specter P.C. Equity: Doximity. **E. E. W. Cohen** reports serving in consulting or advisory roles for ALX Oncology, Ascendis Pharma, Bayer, BioLineRx, BMS, Debiopharm, Dynavax Technologies, the healthcare business of Merck KGaA, Darmstadt, Germany, Merck & Co., Kenilworth, NJ, Regeneron Pharmaceuticals, and Sanofi. **C. Nutting** reports receiving institutional research funding from Cancer Research UK; and owns stock in Advanced Oncotherapy. **L. F. Licitra** reports serving in consulting or advisory roles for AstraZeneca, Bayer, BMS, Eisai, Merck & Co., Kenilworth, NJ, the healthcare business of Merck KGaA, Darmstadt, Germany, Boehringer Ingelheim, Hoffmann-La Roche Ltd, Novartis, Roche, Debiopharm, Sobi, Incyte Biosciences Italy srl, Doxa Pharma srl, Amgen, Nanobiotix and GSK. **B. Burtness** reports serving in consulting or advisory roles for Debiopharm and the healthcare business of Merck KGaA, Darmstadt, Germany. **M. Omar, F. Bouisset, H. Nauwelaerts, Y. Urfer** and **C. Zanna** are employees of Debiopharm. **J. Bourhis** reports serving in consulting or advisory roles for AstraZeneca, BMS, Debiopharm, the healthcare business of Merck KGaA, Darmstadt, Germany, Merck & Co., Kenilworth, NJ, Nanobiotix, and Roche. **Correspondence:** Jonathon D. Schoenfeld, **Jonathan_Schoenfeld@dfci.harvard.edu**

Abstract No. 116. Presented at the 2022 Multidisciplinary Head and Neck Cancers Symposium, February 24-26, 2022; Phoenix, AZ, USA.