Healthcare resource utilization and costs in patients with advanced cancer treated with PD-L1/PD-1 inhibitors who experienced select immune-related adverse events (irAEs)

S. George[1], Y. Zheng[2], E. J. Bell[3], N. M. Engel-Nitz[3], J. C. White[3], L. S. Lal[3], R. Kim[4], S. Krulewicz[4], J. Smith[2], F. X. Liu[2]

[1]Roswell Park Comprehensive Cancer Center, Buffalo, New York, USA; [2]EMD Serono, Inc.; a business of Merck KGaA, Darmstadt, Germany; [3]Optum, Eden Prairie, Minnesota, USA; [4]Pfizer, Inc., New York, New York, USA

SCOPE

- Assess the burden of irAEs in terms of irAE-related and all-cause healthcare resource utilization and costs

CONCLUSIONS

- Patients with irAEs that were severe enough to result in a healthcare encounter (i.e., a claim) had higher costs and higher risks of hospitalizations and emergency department (ED) visits compared with patients without irAEs
 - Inpatient stays were the driver for irAE-related medical costs; one-third of patients with an irAE ultimately required an irAE-related inpatient stay within a year of initiating a PD-L1/PD-1 inhibitor
 - Patients with irAEs had twice the risk of all-cause inpatient stays and all-cause ED visits compared with patients without irAEs
 - Patients with an irAE had 33% higher all-cause total healthcare costs over the 6 months after initiating a PD-L1/PD-1 inhibitor compared with those without an irAE

GET POSTER PDF

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster

Correspondence: Elizabeth Bell. **elizabeth.bell@optum.com**

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app to view a plain language summary of the accepted scientific abstract

INTRODUCTION

- Historically, AE-related hospitalizations were reported in >50% of patients on chemotherapy, while limited data exist for patients on PD-L1/PD-1 inhibitors.[1] Key gaps in economic burden of irAEs from PD-L1/PD-1 inhibitors remain:
 - Data on healthcare resource utilization associated with irAEs are sparse and not reported by cancer type[2-5]
 - Studies on costs associated with irAEs from immunotherapy have limited generalizability.[6-10] Most of these studies were:
 - Not conducted in the United States
 - Restricted to patients with metastatic melanoma or non-small cell lung cancer
 - Limited to select medications, which were sometimes not PD-L1/PD-1 inhibitors

METHODS

- This retrospective cohort study used medical and pharmacy claims from the US Optum Research Database to identify patients with select solid tumors (urothelial carcinoma [UC], renal cell carcinoma [RCC], non-small cell lung cancer [NSCLC], and Merkel cell carcinoma [MCC]) who were treated with PD-L1/PD-1 inhibitor monotherapy between September 1, 2014, and April 30, 2019 (**Table 1**)
- Twenty-one irAEs were chosen a priori based on National Comprehensive Cancer Network and American Society of Clinical Oncology guidelines
- Patients were followed for newly occurring irAEs, based on International Classification of Diseases (ICD)–9 and ICD-10-Clinical Modification (CM) codes from claims, from PD-L1/PD-1 inhibitor initiation to earliest date of any of the following: 6 months after discontinuation of initial line of PD-L1/PD-1 inhibitor therapy (to capture lingering effects of treatment), start of new treatment, death, disenrollment, or April 30, 2019
- Healthcare resource utilization and costs were calculated in the same time period and were considered irAE related if an ICD code for an irAE was present in fields 1 or 2 on the claim

Table 1. Inclusion and exclusion criteria

	Patients Remaining		Patients Excluded	
	n	%	n	%
Patients with ≥2 nondiagnostic claims for UC, RCC, NSCLC, or MCC >30 days apart, in any position, from March 1, 2014, to April 30, 2019 (date of earliest cancer claim was considered the disease diagnosis date) **AND** ≥1 claim for a PD-L1/PD-1 inhibitor between September 1, 2014, and January 31, 2019, but after the disease diagnosis date. The date of the first claim was considered the index date	15,152			
Evidence of only 1 cancer type and no driver mutations for patients with NSCLC	14,162	93.46	990	6.53
≥18 years of age as of the index date (≥12 years old for MCC) and continuous health plan enrollment ≥6 months prior to and ≥3 months following the index date (unless less due to death) and between the disease diagnosis and index dates	9,909	69.97	4,253	30.03
No pregnancy, clinical trial participation, or other primary malignancies	7,507	75.76	2,402	24.24
No claims for UC, RCC, NSCLC, or MCC in the 90 days prior to disease diagnosis date (i.e., evidence of left truncation of data), no missing demographic data, and no evidence of use of anticancer agents other than PD-L1/PD-1 inhibitors	5,504	73.32	2,003	26.68
No preindex claims for any of the 21 irAEs	4,403	80.00	1,101	20.00
Final study sample was 4403 patients	Cohort Assignment			
	UC=417	RCC=436	NSCLC=3506	MCC=44

RESULTS

Patient characteristics

- Of 4,403 eligible patients included in the study, 955 patients (21.7%) developed an irAE after initiating a PD-L1/PD-1 inhibitor as monotherapy
- The mean (SD) age of patients at PD-L1/PD-1 inhibitor initiation was 71 (9) years, 57% were male, 72% had prior cancer treatment, and 30% had prior steroid use (**Table 2**)

Table 2. Patient demographics and clinical characteristics

Characteristic	Total (N=4,403)	Patients With a Select irAE (n=955)	Patients Without a Select irAE (n=3,448)
Age, mean (SD), years	71 (9)	71 (9)	71 (9)
Male, %	57	54	58
Prior cancer treatment, %	72	69	73
Systemic steroids within 6 months prior to PD-L1/PD-1 initiation, %	30	28	31

irAE-related healthcare resource utilization and costs among patients with an irAE

- Among the 955 patients treated with PD-L1/PD-1 inhibitor monotherapy who developed irAEs, the top healthcare resource utilization for irAEs per person per month was ambulatory visits, followed by inpatient stays (**Table 3**)
- Mean irAE-related medical costs were $2,359 per patient per month, driven by the cost of inpatient stays (**Figure 1**)
- 34% of patients who experienced an irAE had an irAE-related inpatient stay within 12 months after PD-L1/PD-1 inhibitor initiation (assessed using Kaplan-Meier method; data not shown)

Table 3. irAE-related healthcare resource utilization among patients who had an irAE subsequent to receiving PD-L1/PD-1 inhibitor monotherapy as treatment for cancer

	All Patients With irAEs (n=955)	UC (n=90)	RCC (n=127)	NSCLC (n=722)	MCC (n=16)
Healthcare resource utilization, mean visits per patient per month, n (SD)					
Ambulatory visit	0.23 (0.57)	0.30 (1.14)	0.21 (0.28)	0.23 (0.50)	0.28 (0.27)
ED visit	0.03 (0.14)	0.06 (0.22)	0.03 (0.11)	0.03 (0.13)	0.03 (0.06)
Inpatient stay	0.09 (0.23)	0.16 (0.43)	0.08 (0.16)	0.09 (0.20)	0.02 (0.05)

Figure 1. irAE-related medical costs among patients who had an irAE subsequent to receiving PD-L1/PD-1 inhibitor monotherapy as treatment for cancer

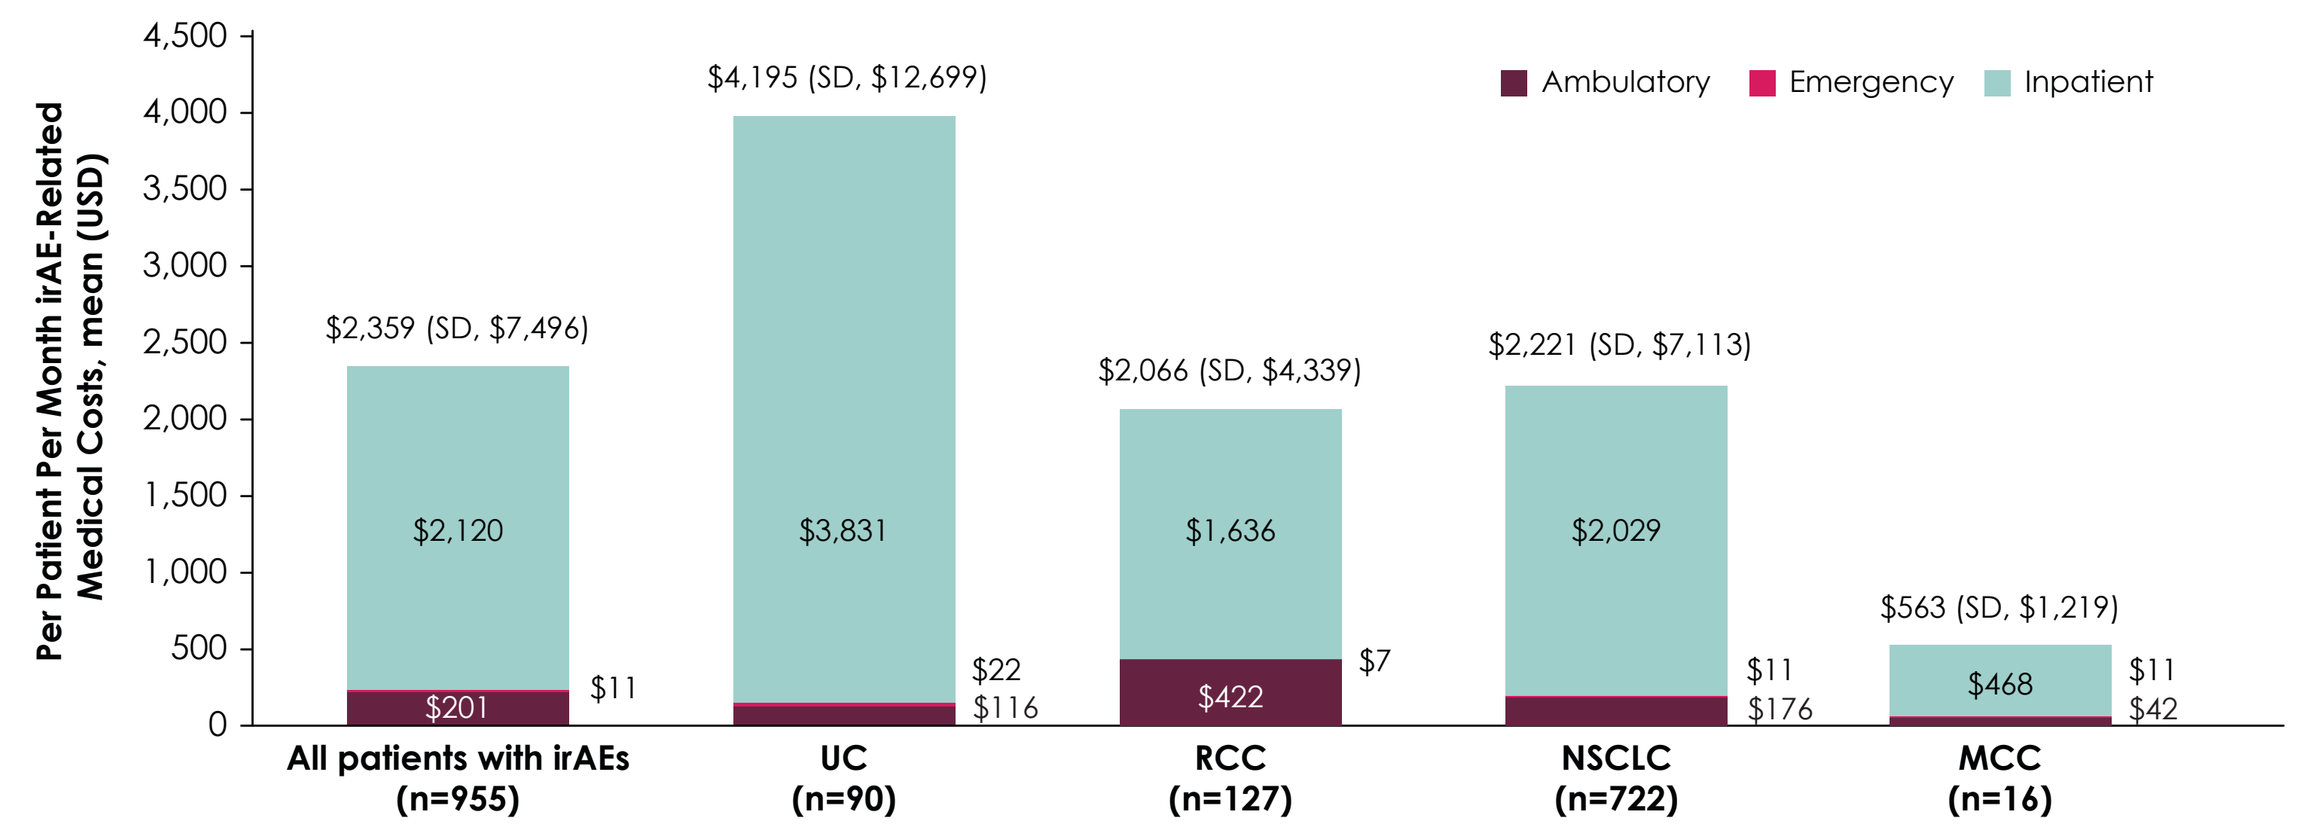
Per Patient Per Month irAE-Related Medical Costs, mean (USD)
4,500
4,000
3,500
3,000
2,500
2,000
1,500
1,000
500
0
Ambulatory
Emergency
Inpatient
$2,359 (SD, $7,496)
$2,120
$201
$11
$4,195 (SD, $12,699)
$3,831
$22
$116
$2,066 (SD, $4,339)
$1,636
$422
$7
$2,221 (SD, $7,113)
$2,029
$11
$176
$563 (SD, $1,219)
$468
$11
$42
All patients with irAEs (n=955)
UC (n=90)
RCC (n=127)
NSCLC (n=722)
MCC (n=16)

Adjusted association of irAEs with risk of all-cause inpatient stays and ED visits

- After adjustment for potential confounding variables, patients with irAEs had more than double the risk of an incident all-cause inpatient stay and an 82% higher risk of an incident all-cause ED visit than patients without irAEs (**Figure 2**)

Figure 2. Association between irAEs (modeled as time varying,[11] irAE vs no irAE) and risk of all-cause inpatient stays and ED visits among patients who received PD-L1/PD-1 inhibitor monotherapy as treatmentr cancer

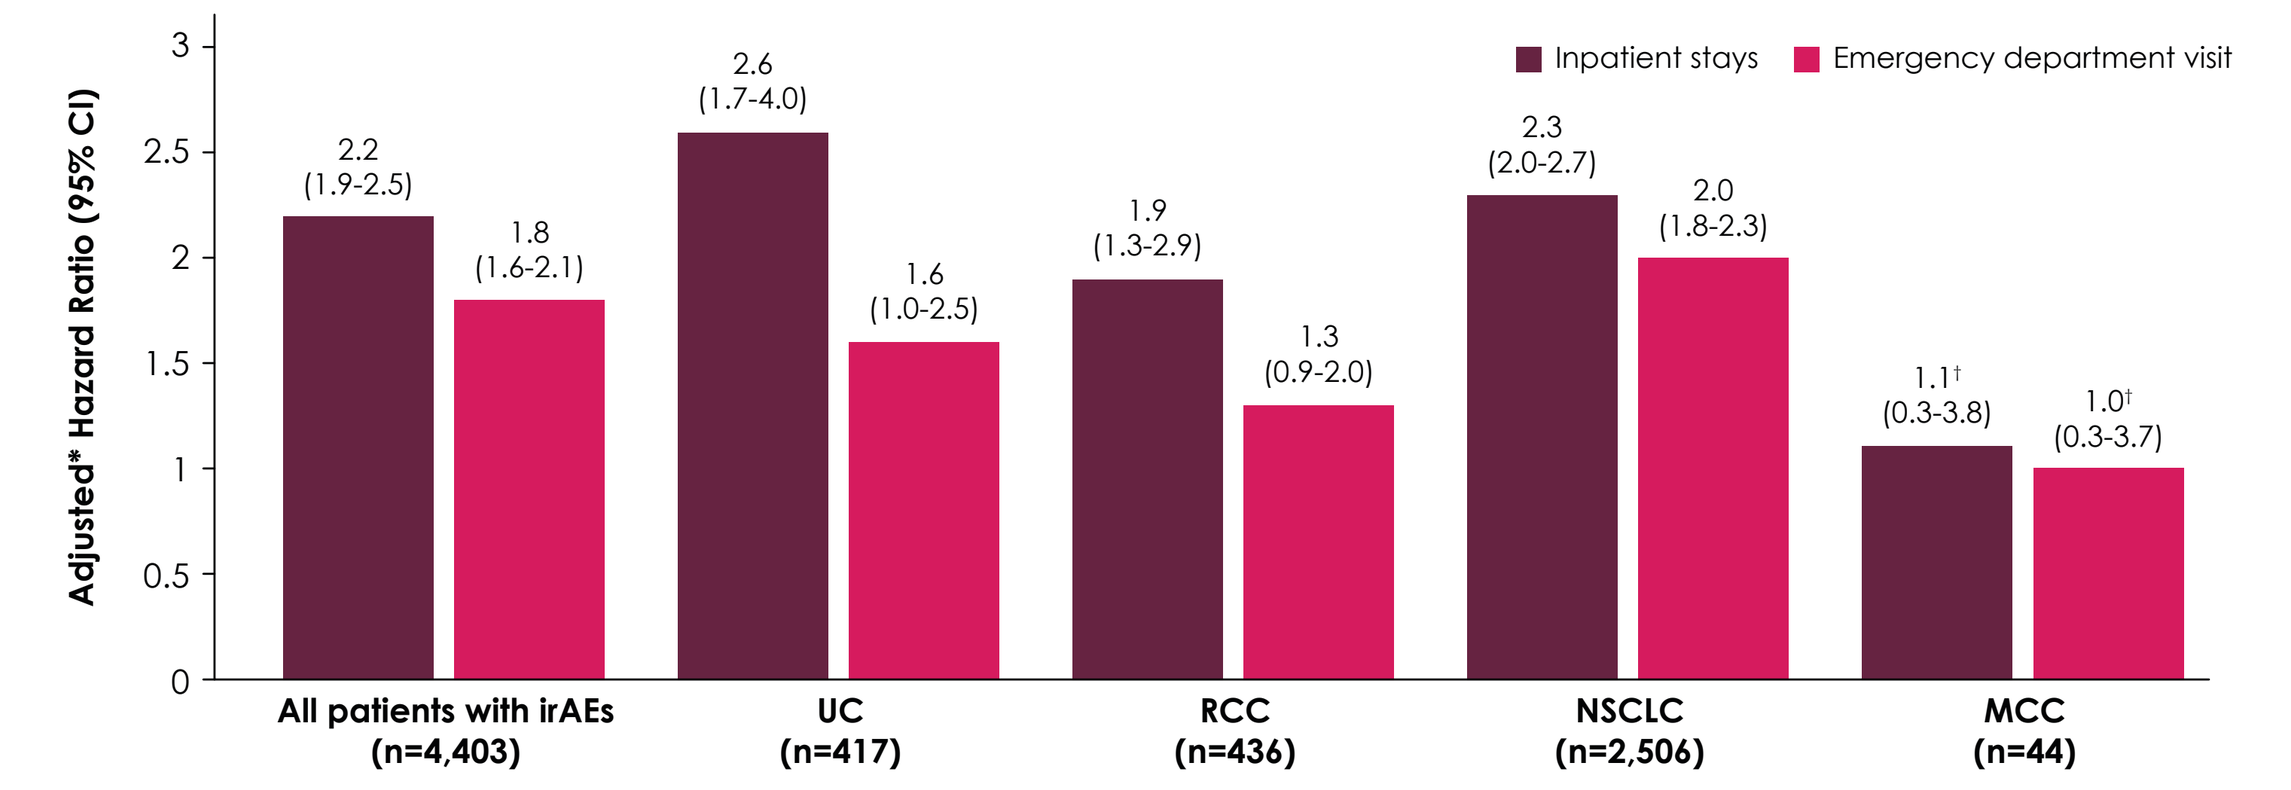
Adjusted* Hazard Ratio (95% CI)
3
2.5
2
1.5
1
0.5
0
Inpatient stays
Emergency department visit
2.2 (1.9-2.5)
1.8 (1.6-2.1)
2.6 (1.7-4.0)
1.6 (1.0-2.5)
1.9 (1.3-2.9)
1.3 (0.9-2.0)
2.3 (2.0-2.7)
2.0 (1.8-2.3)
1.1† (0.3-3.8)
1.0† (0.3-3.7)
All patients with irAEs (n=4,403)
UC (n=417)
RCC (n=436)
NSCLC (n=2,506)
MCC (n=44)

*Using Cox regression, adjusted for potential confounding variables: Treatment line number of PD-L1/PD-1 inhibitor therapy, age at PD-L1/PD-1 inhibitor initiation, sex, presence of metastatic disease at PD-L1/PD-1 inhibitor initiation, region, insurance type, cardiovascular disease at PD-L1/PD-1 inhibitor initiation, radiation during the 6 months prior to PD-L1/PD-1 inhibitor initiation, hypertension at PD-L1/PD-1 inhibitor initiation, dyslipidemia at PD-L1/PD-1 inhibitor initiation, inpatient stay or ED visit during the 6 months prior to PD-L1/PD-1 inhibitor initiation

†Due to low sample size, unadjusted estimates are presented

Adjusted association between irAEs and all-cause total (medical and pharmacy) healthcare costs

- After adjustment for potential confounding variables, patients in the irAE group incurred an average of $99,037 in all-cause total healthcare costs in the 6 months after initiation of PD-L1/PD-1 monotherapy (**Figure 3**)
 - This finding represents a mean of $24,301 more in all-cause total healthcare costs than the cohort without an irAE over 6 months (95% CI: $18,828-$29,774, p< 0.001; **Figure 3**)

Figure 3. Adjusted* all-cause total (medical and pharmacy) costs by irAE group among patients who received PD-L1/PD-1 inhibitor monotherapy

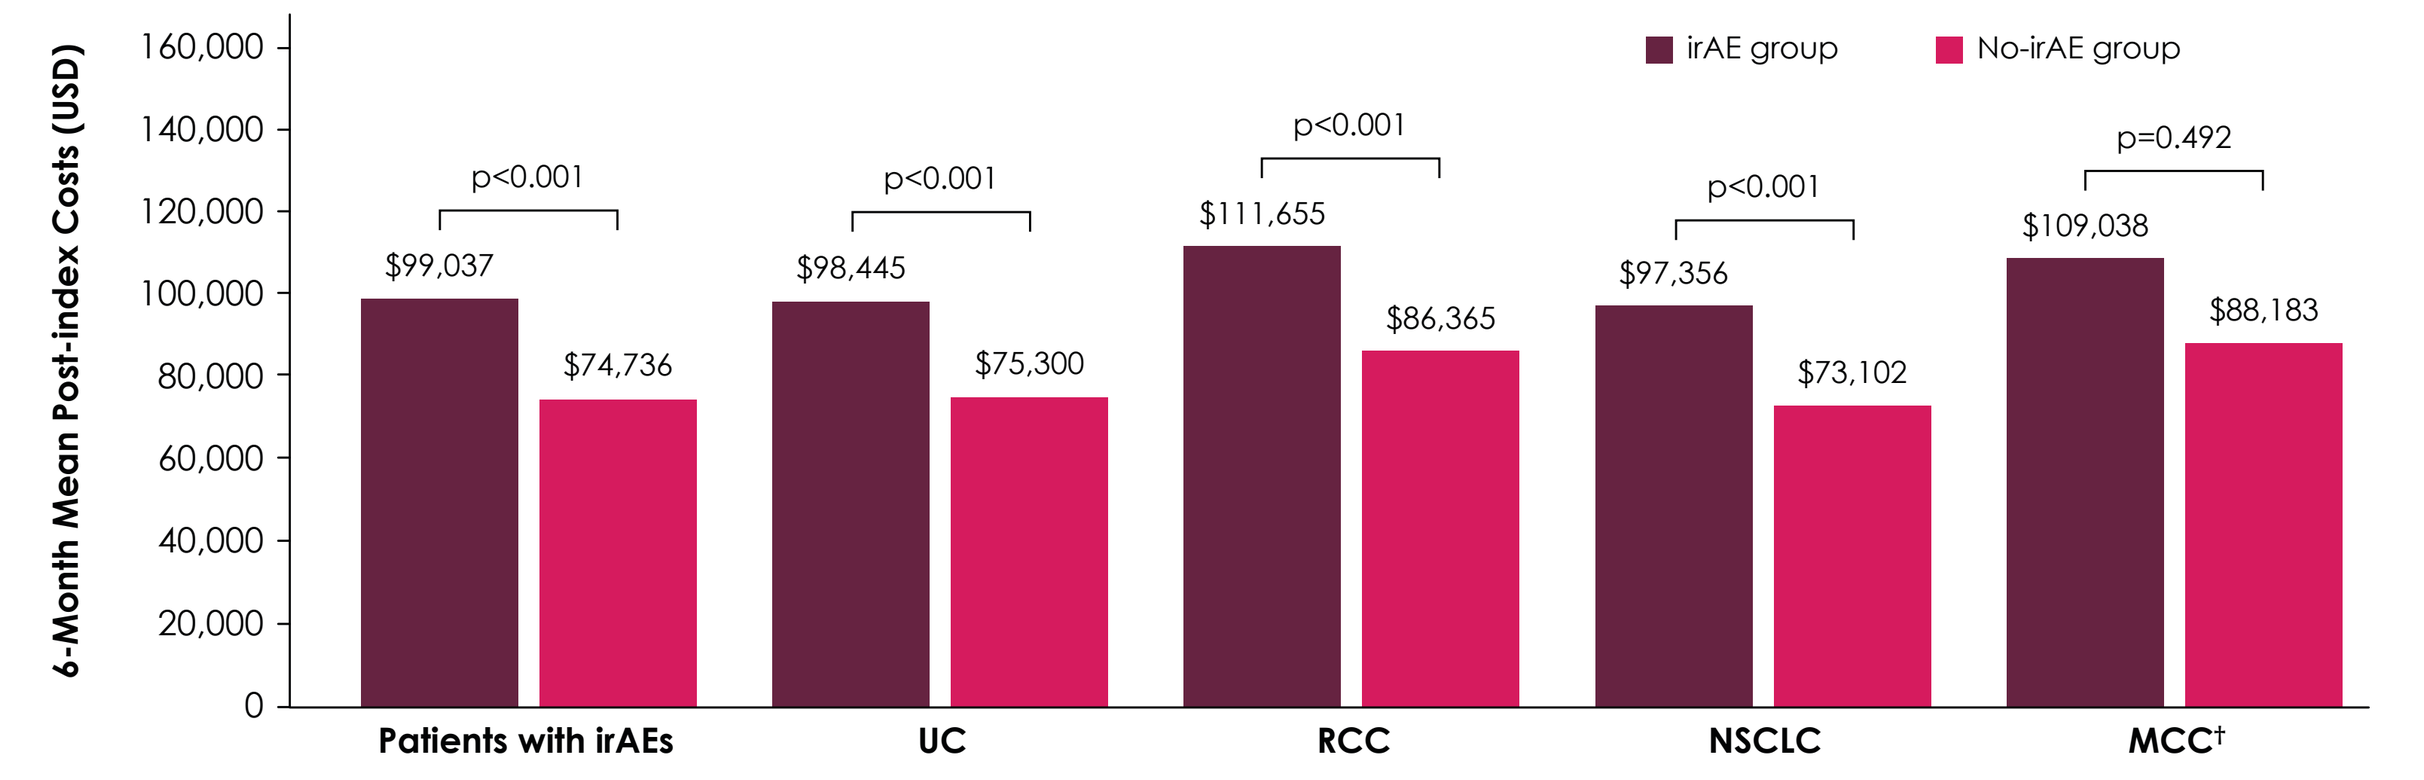
6-Month Mean Post-index Costs (USD)
160,000
140,000
120,000
100,000
80,000
60,000
40,000
20,000
0
irAE group
No-irAE group
p<0.001
$99,037
$74,736
p<0.001
$98,445
$75,300
p<0.001
$111,655
$86,365
p<0.001
$97,356
$73,102
p=0.492
$109,038
$88,183
Patients with irAEs
UC
RCC
NSCLC
MCC†

*Using Lin's regression, adjusted for potential confounding variables: Treatment line number of PD-L1/PD-1 inhibitor therapy, age at PD-L1/PD-1 inhibitor initiation, sex, metastatic disease at PD-L1/PD-1 inhibitor initiation, region, insurance type, cardiovascular disease at PD-L1/PD-1 inhibitor initiation, radiation during the 6 months prior to PD-L1/PD-1 inhibitor initiation, hypertension at PD-L1/PD-1 inhibitor initiation, dyslipidemia at PD-L1/PD-1 inhibitor initiation, inpatient stay or ED visit during the 6 months prior to PD-L1/PD-1 inhibitor initiation

†Due to low sample size, unadjusted estimates are presented

Limitations

- Potential exists for residual confounding due to observational study design
- Steroid use prior to PD-L1/PD-1 inhibitor initiation was common, as it can be used for symptom management, premedication prior to chemo administration and other, non-cancer indications; thus, patients may have already been immunocompromised
- All-cause healthcare resource utilization and costs may have been due to factors other than the irAE or tumor
- Because claims data rely upon diagnosis codes and healthcare encounters to indicate the presence of irAEs, irAEs coded incorrectly or not considered severe enough to warrant an encounter would have been missed

REFERENCES: 1. Prince RM, et al. JAMA Oncol. 2015;1(9):1333-9. 2. Balaji A, et al. J Oncol Pract. 2019;15(9):e825-34. 3. Nuhait MA, et al. J Clin Oncol. 2019;37(15 suppl):e14082. 4. El Majzoub I, et al. Ann Emerg Med. 2019;73(1):79-87. 5. Reynolds KL, et al. J Clin Oncol. 2018;36(5 suppl):127. 6. Mennini FS, et al. Clin Drug Investig. 2018;38(10):967-76. 7. Chouaid C, et al. Clinicoecon Outcomes Res. 2017;9:443-9. 8. Vouk K, et al. J Med Econ. 2016;19(9):900-12. 9. Yousaf N, et al. Melanoma Res. 2015;25(3):259-64. 10. Engel-Nitz NM, et al. J Manag Care Spec Pharm. 2020;26(6):729-40. 11. Kleinbaum DG, Klein M. Extension of the Cox proportional hazards model for time-dependent variables. In: Survival Analysis: a Self-Learning Text. 3rd ed. Springer-Verlag New York. 2012:241-88.

DISCLOSURES: SG has served as a consultant for or received research funding from Agensys, Bayer, Bristol-Myers Squibb, Calithera Biosciences, Corvus Pharmaceuticals, Eisai, EMD Serono, Exelixis, Genentech/Roche, Immunomedics, Merck & Co, Inc., Novartis, Pfizer, Sanofi/Genzyme, and Seattle Genetics/Astellas. **YZ** and **JS** are employees of EMD Serono. **FL** is an employee of EMD Serono and owns stock in Merck & Co, Inc. **EJB**, **JCW**, and **LSL** are employees of Optum. **NMEN** is an employee of Optum and owns stock in UnitedHealth Group. **RK** is employed by Pfizer and owns stock in Bristol-Myers Squibb and Exelixis. **SK** is an employee of Pfizer and owns stock in Abbott Laboratories, Amgen, Bristol-Myers Squibb, CVS Health Corporation, Gilead, GlaxoSmithKline, Medtronic, and Pfizer. Deja Scott-Shemon (Optum, Eden Prairie) provided medical writing services, which was funded by EMD Serono. **ACKNOWLEDGMENTS:** This study was sponsored by Merck KGaA, Darmstadt, Germany as part of an alliance between Merck KGaA and Pfizer.

Abstract No. 2858. Presented at the ESMO Virtual Congress 2020, 19–21 September 2020.