Real-world (RW) clinical outcomes in patients (pts) with locally advanced (LA) or metastatic Merkel cell carcinoma (mMCC) treated in United States (US) oncology clinical practices: results from SPEAR-Merkel

C. L. Cowey[1], F. X. Liu[2], R. Kim[3], M. Boyd[4], N. Fulcher[4], S. Krulewicz[5], J. Smith[2], A. Bhanegaonkar[2]

[1]The US Oncology Network, McKesson Life Sciences, Houston, Texas, USA; [2]EMD Serono, Inc., Rockland, Massachusetts, USA; a business of Merck KGaA, Darmstadt, Germany; [3]Pfizer, Inc, New York, New York, USA; [4]McKesson Life Sciences, The Woodlands, Texas, USA; [5]Pfizer, Inc, Collegeville, Pennsylvania, USA

SCOPE

- **The purpose of SPEAR-Merkel (Study informing treatment Pathway dEcisions in Merkel cell cARcinoma) was to understand RW clinical outcomes in pts with LA and mMCC initiating first-line (1L) treatment with avelumab, non-avelumab immuno-oncology (IO) therapy, or chemotherapy in the US community oncology practice setting**

CONCLUSIONS

- **The results of SPEAR-Merkel, a longitudinal retrospective study, indicate that pts with LA or mMCC treated with IO therapies targeting PD-1/PD-L1 showed clinical benefit vs chemotherapy in RW community-based oncology practices**
- **Results from SPEAR-Merkel contribute to the current knowledge about RW effectiveness of innovative therapies beyond the pivotal clinical studies and therefore inform clinicians of further treatment options for LA and mMCC**
- **To our knowledge, this is the first study to analyze pts with LA or mMCC treated with avelumab or non-avelumab IO alongside pts treated with chemotherapy in a RW setting. In addition, the study shows that 1L chemotherapy is still used in clinical practice in the US despite its limited durability, less favorable outcomes, and safety profile**

GET POSTER PDF

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster

Correspondence: Abhijeet Bhanegaonkar, **abhijeet.bhanegaonkar@emdserono.com**

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app to view a plain language summary of the accepted scientific abstract

BACKGROUND

- MCC is a rare, aggressive cancer[1] with considerable disease-associated mortality[2]
- Although MCC is sensitive to chemotherapy, response duration is low (2-4 months), and treatment has been associated with myelosuppression and neutropenia[3-6]
- MCC tumors and the surrounding microenvironment display elements of immune activation, including upregulation of the PD-L1/PD-1 pathway[5]
- Therapies targeting PD-L1 and PD-1 are increasingly becoming a standard of care,[6] based on response rates and durability
- US Food and Drug Administration (FDA) approval of avelumab (anti–PD-L1) was based on a long-term survival benefit and favorable objective response rate observed in JAVELIN Merkel 200 part A, in which pts received avelumab for disease progression after chemotherapy (avelumab approval date, 23 March 2017)[7-8]
 - After 44 months of follow-up, median overall survival (OS) was 12.6 months[9]
 - Part B showed a long-term survival benefit (median OS was 20.3 months after ≥15 months of follow-up) and safety in pts receiving 1L avelumab[10]
- FDA approval of pembrolizumab (anti–PD-1; approval date, 19 Dec 2018) was based on KEYNOTE-017 showing a long-term survival benefit among pts with recurrent LA or distant mMCC treated with 1L pembrolizumab[10]
- Nivolumab (anti–PD-1), although not FDA approved for MCC, was associated with a favorable objective response rate in the adjuvant setting in pts with stage IIa to IV resectable MCC in CheckMate-358[12]
- RW data on treatment patterns and clinical outcomes in MCC are limited due to the rarity of the disease[13]

METHODS

Study design and data source

- Retrospective, observational descriptive study of adult pts with LA or mMCC initiating 1L avelumab, non-avelumab IO, or chemotherapy between 1 Jan 2015 and 30 Jun 2019 identified from The US Oncology Network (USON) electronic healthcare record (EHR) database and followed through 30 Sep 2019. Data were obtained from EHR structured fields and supplemented by chart review
 - The USON is a community-based network of oncology practices comprising over 470 cancer treatment centers and 1,200 physicians in 25 states and treating over 1 million pts each year[14]

Statistical analysis

- Demographic and clinical characteristics and RW overall response rate (rwORR) were analyzed descriptively
- Kaplan-Meier methods were used to assess RW duration of response (rwDOR), RW progression-free survival (rwPFS), and OS
 - Pts who had not experienced an event during the study observation period were censored on the study end date or the last visit date recorded in the data set, whichever occurred first

RESULTS

- Among the 94 pts included in the study (28 avelumab, 26 non-avelumab IO, and 40 chemotherapy; **Figure 1**), median age was 73 years, 68.1% were male, and 83.0% were White (**Table 1**)
- At 1L initiation, 28.7% had LA MCC and 71.3% had mMCC.
 - The majority of pts were immunocompetent (>85% in each treatment cohort), and >50% had favorable (0 or 1) baseline Eastern Cooperative Oncology Group performance status (ECOG PS; **Table 1**)
- Median follow-up (months) was 11.2, 9.8, and 10.7 in avelumab, non-avelumab IO, and chemotherapy pts, respectively (**Table 1**)
- Median rwORR (physician assessed) was 64.3% in avelumab pts, 61.5% in non-avelumab IO pts, and 42.5% in chemotherapy pts (**Table 2**)
 - Median rwDOR was 15.5 months in avelumab pts, not reached (NR) in non-avelumab IO pts, and 44.5 months in chemotherapy pts. (**Table 2**)
 - rwDOR in chemotherapy pts was influenced by 2 long-term responders with 40+ months rwDOR. Median rwDOR with chemotherapy was 6.5 months without these 2 pts
- Median rwPFS from 1L treatment initiation was 11.4 months in avelumab pts, 8.1 months in non-avelumab IO pts, and 6.1 months in chemotherapy pts (**Figure 2**)
 - 6-month rwPFS rate: avelumab, 67.9%; non-avelumab IO, 57.7%; chemotherapy, 50.0%
- Median OS was 20.2 months in avelumab pts, NR in non-avelumab IO pts, and 14.7 months in chemotherapy pts (**Figure 3**)
 - 6-month OS rate: avelumab, 85.7%; non-avelumab IO, 69.2%; chemotherapy, 77.5%

Figure 1. Patient selection

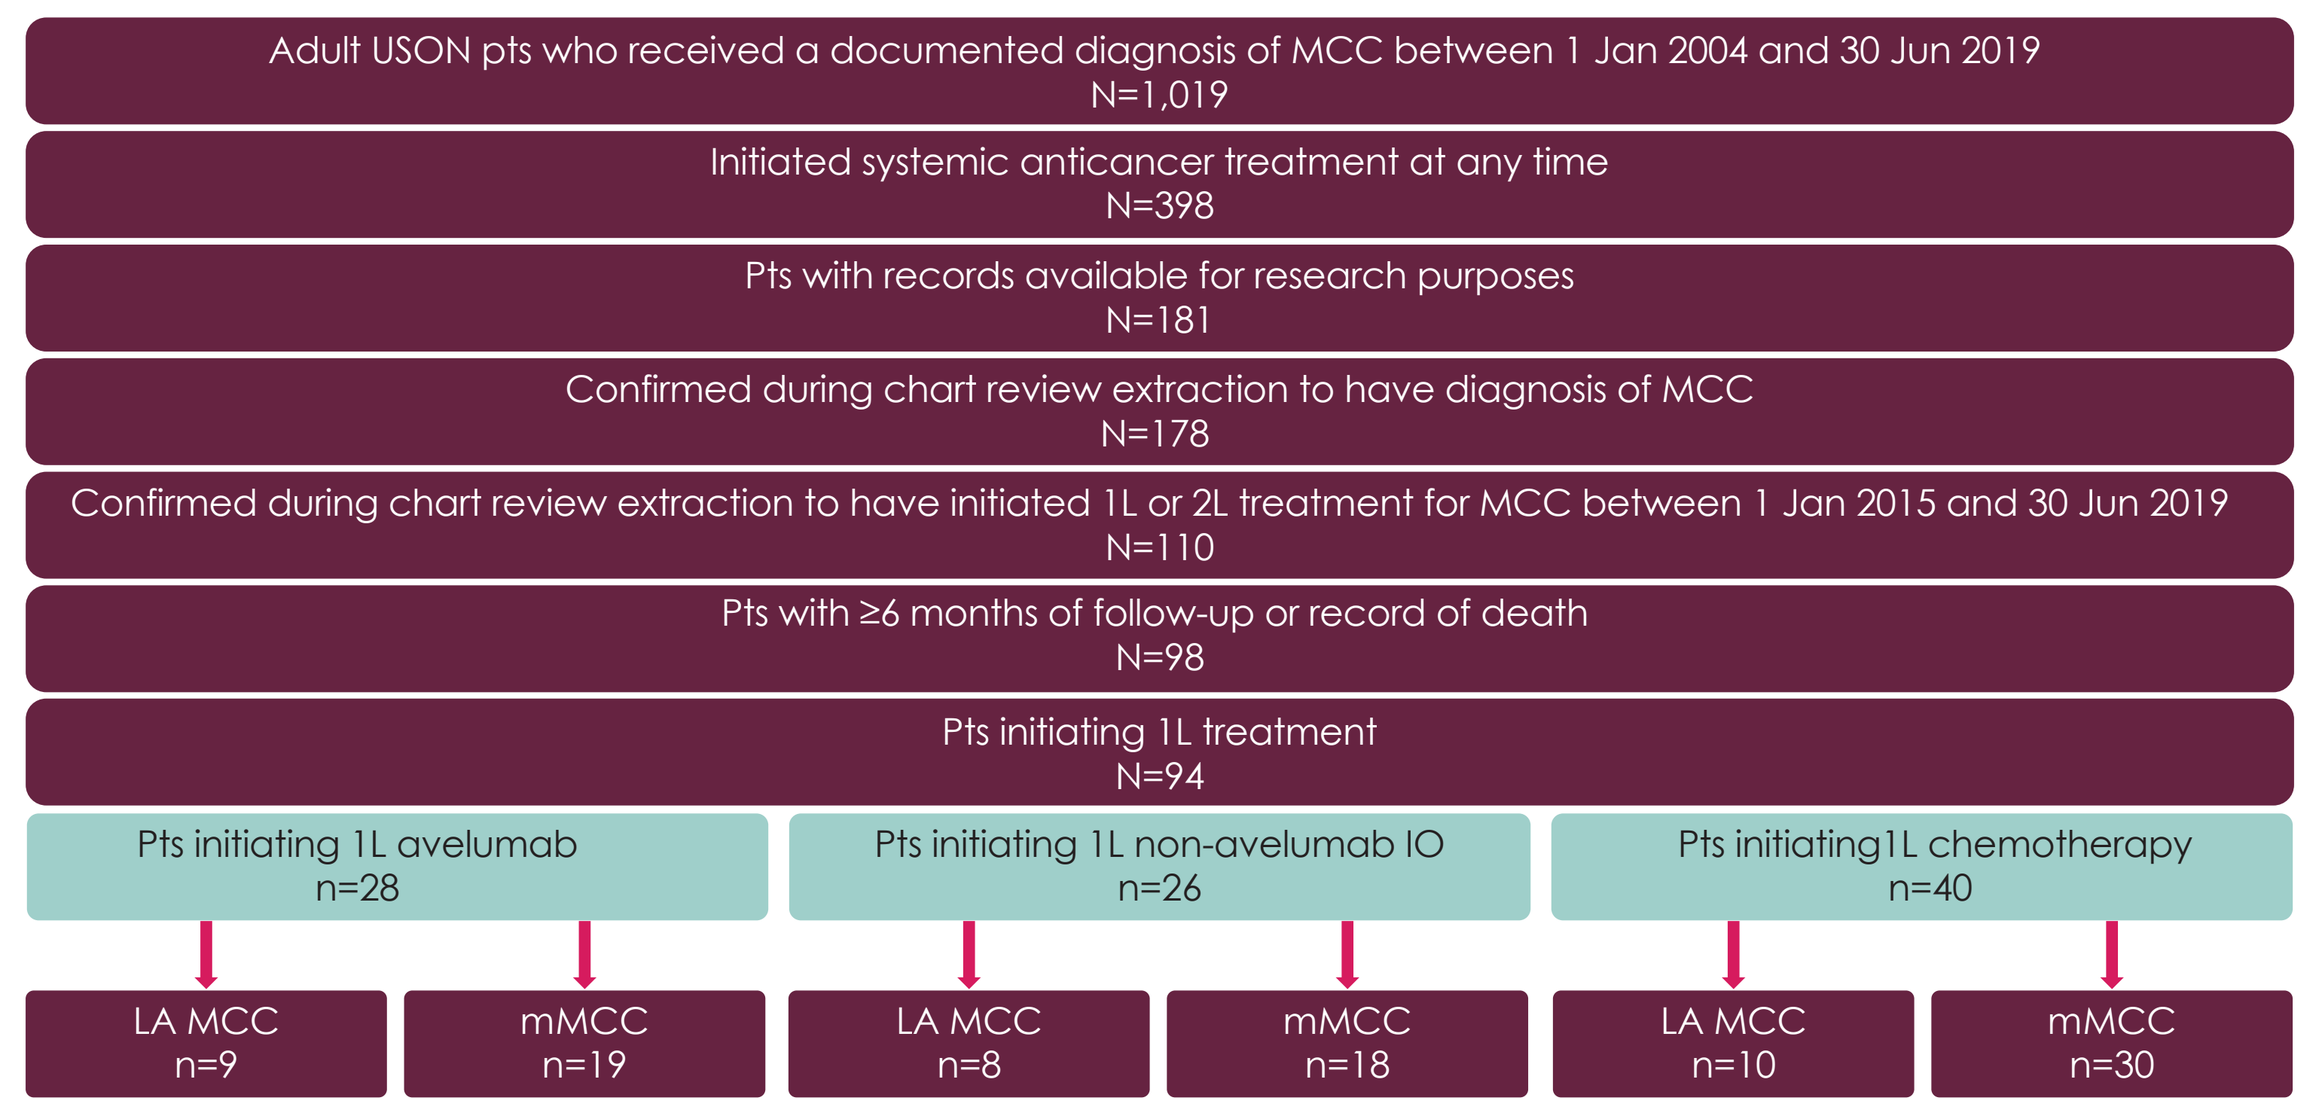
Adult USON pts who received a documented diagnosis of MCC between 1 Jan 2004 and 30 Jun 2019
N=1,019
Initiated systemic anticancer treatment at any time
N=398
Pts with records available for research purposes
N=181
Confirmed during chart review extraction to have diagnosis of MCC
N=178
Confirmed during chart review extraction to have initiated 1L or 2L treatment for MCC between 1 Jan 2015 and 30 Jun 2019
N=110
Pts with ≥6 months of follow-up or record of death
N=98
Pts initiating 1L treatment
N=94
Pts initiating 1L avelumab
n=28
Pts initiating 1L non-avelumab IO
n=26
Pts initiating1L chemotherapy
n=40
LA MCC
n=9
mMCC
n=19
LA MCC
n=8
mMCC
n=18
LA MCC
n=10
mMCC
n=30

2L, second-line treatment

Table 1. Baseline demographic and clinical characteristics

	Overall (N=94)	Avelumab (n=28)	Non-avelumab IO (n=26)	Chemotherapy (n=40)
Age at 1L treatment initiation, median (range), years	73 (21-90+)	78 (52-90+)	74 (43-90+)	69 (21-90+)
Age group, n (%)				
<65 years	20 (21.3)	3 (10.7)	4 (15.4)	13 (32.5)
65-74 years	34 (36.2)	7 (25.0)	9 (34.6)	18 (45.0)
≥75 years	40 (42.6)	18 (64.3)	13 (50.0)	9 (22.5)
Male, n (%)	64 (68.1)	24 (85.7)	15 (57.7)	25 (62.5)
Race, n (%)				
White	78 (83.0)	23 (82.1)	22 (84.6)	33 (82.5)
Not reported/not documented*	16 (17.0)	5 (17.9)	4 (15.4)	7 (17.5)
US practice region, n (%)				
South	40 (42.6)	16 (57.1)	12 (46.2)	12 (30.0)
West	42 (44.7)	11 (39.3)	10 (38.5)	21 (52.5)
Northeast	2 (2.1)	1 (3.6)	0	1 (2.5)
Midwest	10 (10.6)	0	4 (15.4)	6 (15.0)

Table 1. Baseline demographic and clinical characteristics (cont)

	Overall (N=94)	Avelumab (n=28)	Non-avelumab IO (n=26)	Chemotherapy (n=40)
Stage at initial MCC diagnosis, n (%)				
IA-IIIA	17 (18.1)	4 (14.3)	5 (19.2)	8 (20.0)
IIIB	12 (12.8)	4 (14.3)	3 (11.5)	5 (12.5)
IV	22 (23.4)	6 (21.4)	5 (19.2)	11 (27.5)
Not documented	43 (45.7)	14 (50.0)	13 (50.0)	16 (40.0)
Disease status at 1L initiation				
LA	27 (28.7)	9 (32.1)	8 (30.1)	10 (25.0)
Metastatic	67 (71.3)	19 (67.9)	18 (69.2)	30 (75.0)
Primary tumor location, n (%)				
Face	25 (26.6)	5 (17.9)	9 (34.6)	11 (27.5)
Lower limb	19 (20.2)	6 (21.4)	5 (19.2)	8 (20.0)
Upper limb	17 (18.1)	8 (28.6)	4 (15.4)	5 (12.5)
Trunk	10 (10.6)	4 (14.3)	3 (11.5)	3 (7.5)
Scalp and neck	8 (8.5)	4 (14.3)	1 (3.8)	3 (7.5)
Unknown primary/not documented/other	15 (16.1)	1 (3.6)	4 (15.4)	10 (25.0)
Immunocompromised, n (%)	7 (7.4)	1 (3.6)	3 (11.5)	3 (7.5)
ECOG PS, n (%)				
0	15 (16.0)	2 (7.1)	4 (15.4)	9 (22.5)
1	37 (39.4)	14 (50.0)	9 (34.6)	14 (35.0)
2	17 (18.1)	3 (10.7)	4 (15.4)	10 (25.0)
3	1 (1.1)	0	1 (3.8)	0
Not documented	24 (25.5)	9 (32.1)	8 (30.8)	7(17.5)
Metastatic sites recorded prior to 1L initiation, n (%)				
Pts with ≥1 recorded metastatic site	67 (71.2)	19 (57.6)	18 (69.2)	30 (75.0)
Lymph node†	38 (57.6)	10 (55.6)	11 (57.9)	17 (58.6)
Liver†	17 (25.4)	8 (42.1)	5 (27.8)	4 (13.3)
Bone†	16 (23.9)	3 (15.8)	4 (22.2)	9 (30.0)
Lung†	3 (4.5)	2 (10.5)	0	1 (3.3)
Brain†	3 (4.5)	0	1 (5.6)	2 (6.7)
Other†	15 (22.3)	4 (21.1)	4 (22.2)	7 (23.3)
Index year				
2015	20 (21.3)	0	0	20 (50.0)
2016	15 (16.0)	0	5 (19.2)	10 (25.0)
2017	22 (23.4)	7 (25.0)	8 (30.8)	7 (17.5)
2018	29 (30.9)	18 (64.3)	8 (30.8)	3 (7.5)
2019‡	8 (8.5)	3 (10.7)	5 (19.2)	0
Time from initial MCC diagnosis to la/mMCC status, median (range), weeks	5.3 (0.1-239.7)	5.1 (0.1-239.7)	6.1 (0.1-155.8)	4.0 (0.1-190.3)
Follow-up time from 1L initiation, median (range), months	11.0 (0.3-53.8)	11.2 (0.5-27.2)	9.8 (0.3-35.9)	10.7 (0.7-53.8)

* Not documented combined with not reportable due to n<5 per McKesson Life Sciences policy
† Percentage among all pts with metastases in treatment cohort
‡ Patient selection criteria requiring 6 months of follow-up (or death) limits 2019 index dates to on or before 31 Mar 2020

Table 2. RW response to 1L treatment

	Overall (N=94)	Avelumab (n=28)	Non-avelumab IO (n=26)	Chemotherapy (n=40)
rwORR (95% CI), %	54.3 (43.7-64.6)	64.3 (44.1-81.4)	61.5 (40.6-79.8)	42.5 (27.0-59.1)
Best overall response, n (%)				
Complete response	14 (14.9)	9 (32.1)	3 (11.5)	2 (5.0)
Response (NOS)	37 (39.4)	9 (32.1)	13 (50.0)	15 (37.5)
Stable disease	4 (4.3)	2 (7.1)	1 (3.8)	1 (2.5)
Mixed response	3 (3.2)	2 (7.1)	0	1 (2.5)
Progressive disease	13 (13.8)	2 (7.1)	5 (19.2)	6 (15.0)
Not evaluated	23 (24.5)	4 (14.3)	4 (15.4)	15 (37.5)
rwDOR, median (95% CI), months*	44.5 (8.3-NR)	15.5 (7.5-NR)	NR (4.2-NR)	44.5 (2.5-NR)
TTD, median (95% CI), months*	3.8 (3.0-5.6)	10.5 (5.3-14.3)	7.3 (2.5-18.2)	2.2 (1.9-3.5)

NOS, not otherwise specified; **NR**, not reached; **TTD**, time to discontinuation
* Unadjusted Kaplan-Meier estimates

Figure 2. Kaplan-Meier plot of rwPFS from 1L initiation

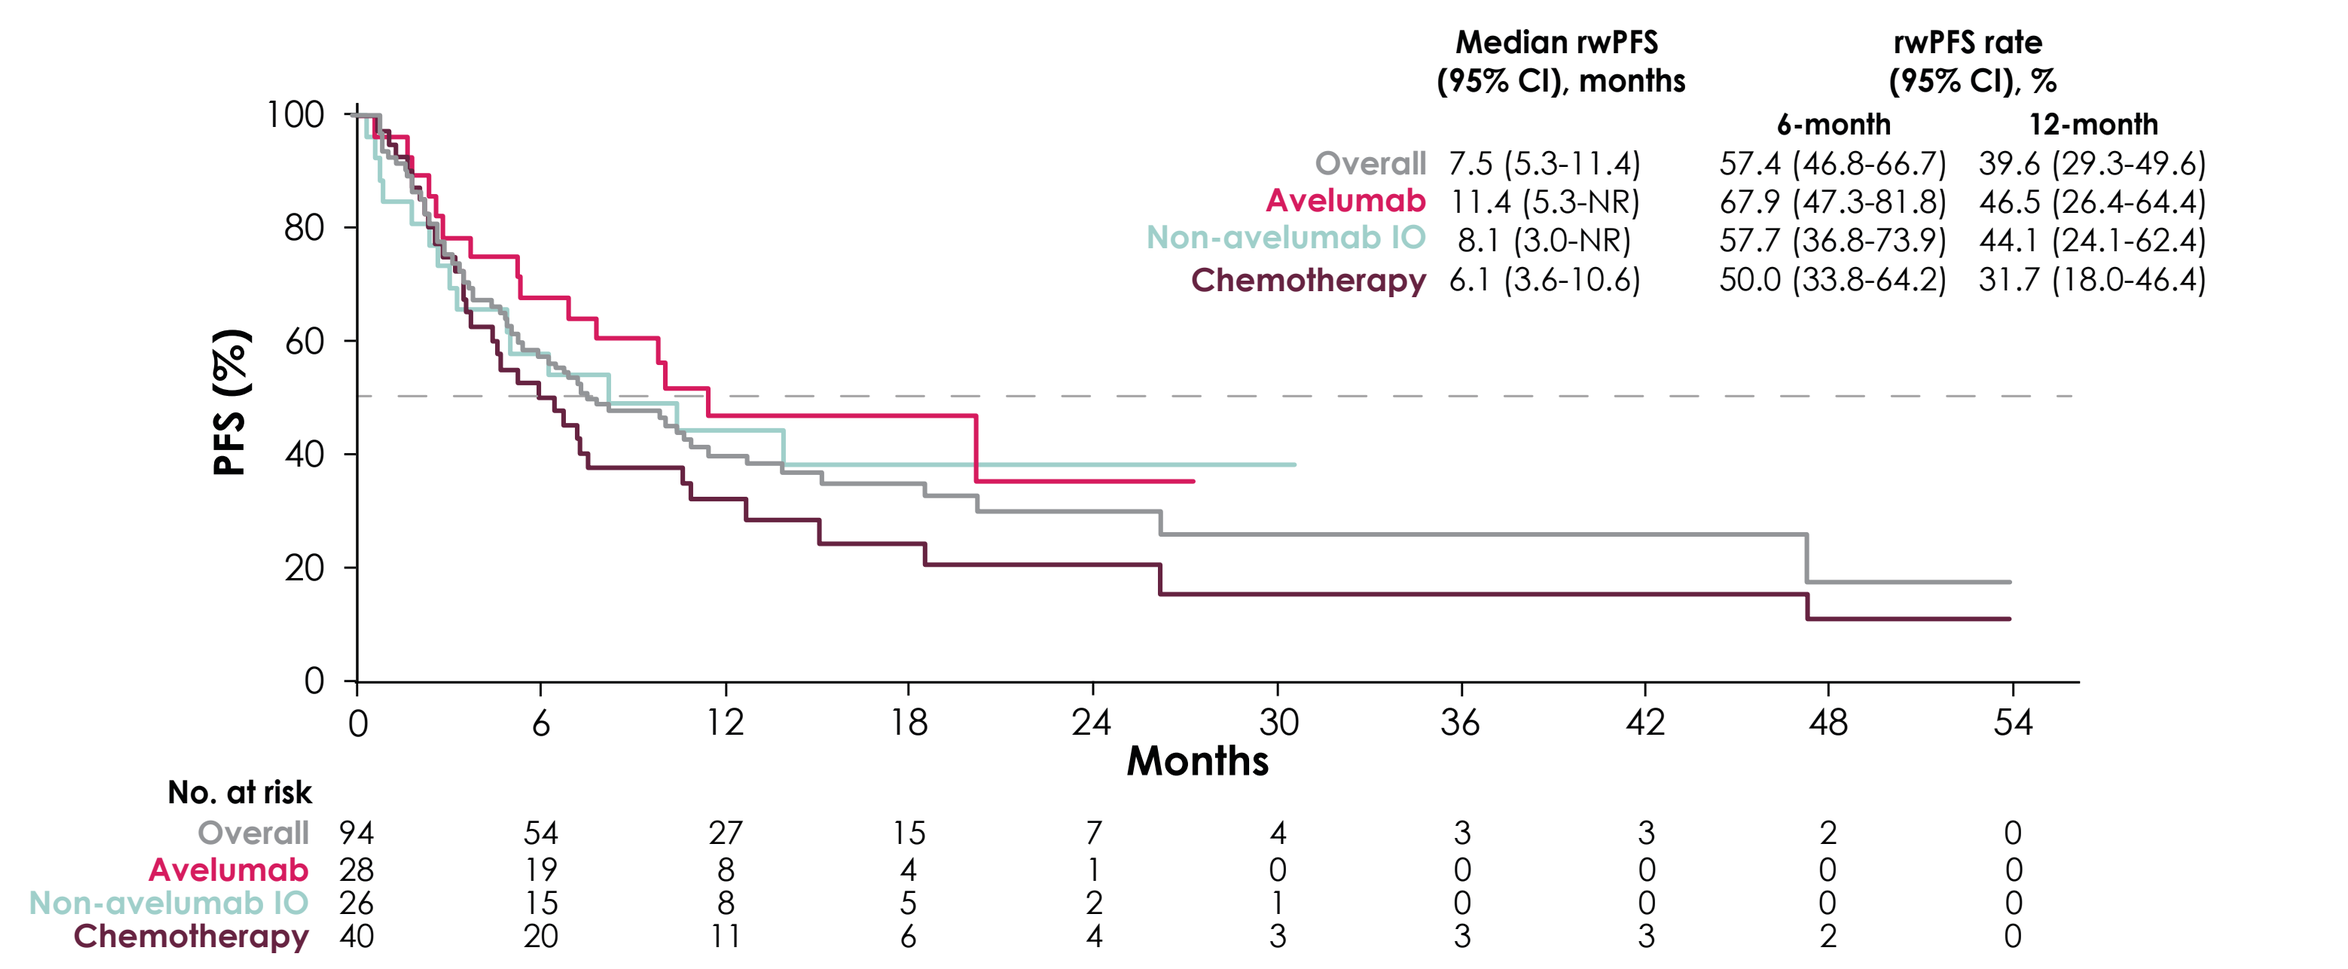
Median rwPFS (95% CI), months
rwPFS rate (95% CI), %
6-month
12-month
Overall 7.5 (5.3-11.4) 57.4 (46.8-66.7) 39.6 (29.3-49.6)
Avelumab 11.4 (5.3-NR) 67.9 (47.3-81.8) 46.5 (26.4-64.4)
Non-avelumab IO 8.1 (3.0-NR) 57.7 (36.8-73.9) 44.1 (24.1-62.4)
Chemotherapy 6.1 (3.6-10.6) 50.0 (33.8-64.2) 31.7 (18.0-46.4)
PFS (%)
Months
No. at risk
Overall 94 54 27 15 7 4 3 3 2 0
Avelumab 28 19 8 4 1 0 0 0 0 0
Non-avelumab IO 26 15 8 5 2 1 0 0 0 0
Chemotherapy 40 20 11 6 4 3 3 3 2 0

This figure presents the unadjusted Kaplan-Meier curves of PFS from 1L initiation to earliest date of first progression (as defined by physician progress note or imaging report), recurrent disease, or death, censoring pts who were progression free or alive at last contact

Figure 3. Kaplan-Meier plot of OS from 1L initiation

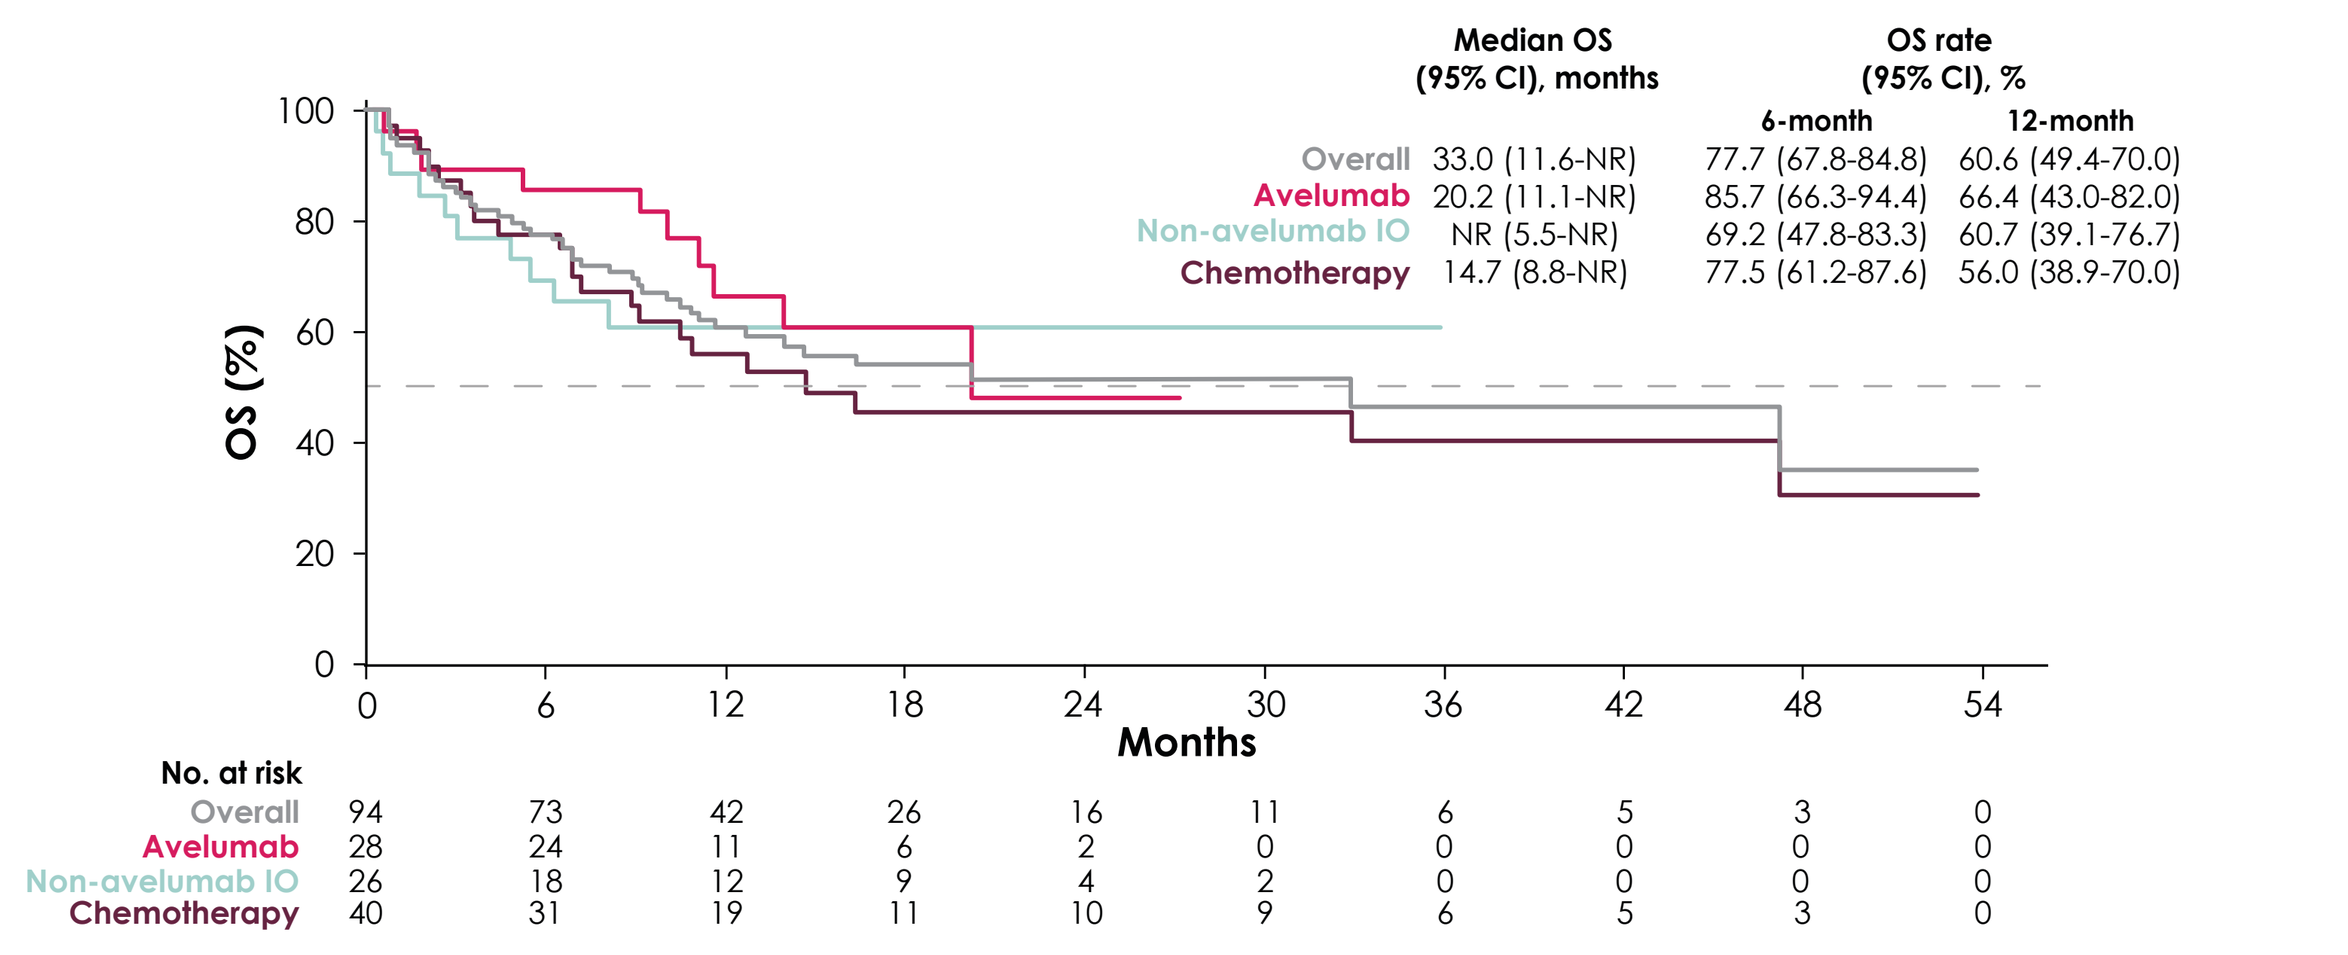
Median OS (95% CI), months
OS rate (95% CI), %
6-month
12-month
Overall 33.0 (11.6-NR) 77.7 (67.8-84.8) 60.6 (49.4-70.0)
Avelumab 20.2 (11.1-NR) 85.7 (66.3-94.4) 66.4 (43.0-82.0)
Non-avelumab IO NR (5.5-NR) 69.2 (47.8-83.3) 60.7 (39.1-76.7)
Chemotherapy 14.7 (8.8-NR) 77.5 (61.2-87.6) 56.0 (38.9-70.0)
OS (%)
Months
No. at risk
Overall 94 73 42 26 16 11 6 5 3 0
Avelumab 28 24 11 6 2 0 0 0 0 0
Non-avelumab IO 26 18 12 9 4 2 0 0 0 0
Chemotherapy 40 31 19 11 10 9 6 5 3 0

This figure presents the unadjusted Kaplan-Meier curves of OS from 1L initiation to death, censoring pts who were alive at last contact

Limitations

- The small sample sizes of the cohorts, likely due to the rarity of the disease, limit the drawing of conclusions
- The study data were originally collected for clinical instead of research purposes. Coding errors of omission and commission could not be confirmed or corrected, leading to misclassification bias
- Because SPEAR-Merkel was a retrospective observational study, pts were not randomized to the treatment cohorts, which could have led to clinical differences that may have altered treatment outcomes
- In SPEAR-Merkel, pts receiving 1L chemotherapy had longer follow-up periods compared with the other cohorts, which may have influenced results in the 1L chemotherapy cohort
- There may have been differences in RW clinical outcomes compared with clinical trials due to variations across the patient populations, as well as types and frequency of assessments (eg, Response Evaluation Criteria in Solid Tumors as opposed to physician assessments)
- The USON encourages the use of evidence-based guidelines, which might not be followed across all oncology practices, limiting generalizability to pts treated at other community oncology practices or academic centers

Discussion

- The results of SPEAR-Merkel indicate that pts with LA or mMCC treated with IO therapies had clinical benefit vs chemotherapy in RW community oncology practices
- The patient populations in pivotal clinical trials and RW studies had some similarities in baseline demographic characteristics compared with those in SPEAR-Merkel (older, male, and/or white)[9,10,11,13]
- Some clinical outcomes in SPEAR-Merkel were also consistent with those trials and RW studies
 - In the SPEAR-Merkel 1L avelumab cohort vs JAVELIN Merkel 200 part B, median OS was 20.2 vs 20.3 months, and the 12-month OS rate was 66.4% vs 60%[10]
 - In the SPEAR-Merkel 1L non-avelumab cohort vs KEYNOTE-017 (recurrent locoregional or distant mMCC treated with 1L pembrolizumab), median OS was NR in either cohort[11]
- In a RW study of 1L chemotherapy for distant mMCC, the ORR was 55%, median PFS was 3 months, and median OS was 9.5 months from chemotherapy initiation.[4] In SPEAR-Merkel in the 1L chemotherapy cohort, the rwORR was 42.5%, median rwPFS was 6.1 months, and median OS was 14.7 months

REFERENCES 1. Toker C. Arch Dermatol. 1972;105(1):107-10. 2. Surveillance, Epidemiology, and End Results Program. https://seer.cancer.gov/archive/csr/1975_2016/. Accessed 24 Aug 2020. 3. Cowey CL, et al. Future Oncol. 2017;13(19):1699-710. 4. Iyer JG, et al. Cancer Med. 2016;5(9):2294-301. 5. Lipson EJ, et al. Cancer Immunol Res. 2013;1(1):54-63. 6. National Comprehensive Cancer Network. https://www.nccn.org/professionals/physician_gls/pdf/mcc.pdf. Accessed 24 Aug 2020. 7. US Food and Drug Administration. https://www.fda.gov/drugs/resources-information-approved-drugs/avelumab-bavencio. Accessed 24 Aug 2020. 8. Kaufman HL, et al. J Immunother Cancer. 2018;6(1):7. 9. D'Angelo SP, et al. J Immunother Cancer. 2020;8(1):e000674. 10. D'Angelo SP, et al. SITC 2019 [abstract P362]. 11. Nghiem P, et al. J Clin Oncol. 2019;37(9):693-702. 12. Topalian SL, et al. J Clin Oncol. 2020;38(22):2476-87. 13. Walker JW, et al. J Immunother Cancer. 2020;8(1):e000313. 14. The US Oncology Network. https://www.usoncology.com/our-company. Accessed 1 Jul 2020. **DISCLOSURES: Charles. L. Cowey** is an employee of US Oncology. **Ruth Kim** is an employee of Pfizer and owns stock in Bristol Myers Squibb. **Marley Boyd** and **Nicole Fulcher** are employees of McKesson Life Sciences. **Stan Krulewicz** is an employee of and owns stock in Pfizer, and owns stock in Medtronic, Abbott Laboratories, Amgen, Bristol Myers Squibb, CVS Health, GlaxoSmithKline, and Gilead Sciences. **Jodi Smith**, **Frank X. Liu**, and **Abhijeet Bhanegaonkar** are employees of and own stock in EMD Serono Inc., a business of Merck KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS:** This study was sponsored by EMD Serono, Inc., a business of Merck KGaA, Darmstadt, Germany, as part of an alliance between Merck KGaA and Pfizer.

Abstract No. 1822. Presented at the ESMO Virtual Congress 2020, 19–21 September 2020.