The IAP antagonist xevinapant in combination with high-dose cisplatin chemoradiotherapy induces NF-κB and apoptotic pathway biomarkers in patients with high-risk, locally advanced squamous cell carcinoma of the head and neck

Luke Piggott, Bruno Gavillet, Franck Brichory, Kathrin Gollmer, Florilene Bouisset, Gregoire Vuagniaux

Debiopharm International SA, Lausanne, Switzerland

BACKGROUND

- Inhibitors of apoptosis proteins (IAPs) are a family of anti-apoptotic proteins that promote cancer cell survival and inhibit cell death
- IAPs represent promising therapeutic targets because they are overexpressed in various malignancies, including squamous cell carcinoma of the head and neck (SCCHN), and have been linked to tumor progression, treatment failure, and poor prognosis
- IAPs regulate apoptosis and modulate nuclear factor kappa B (NF-κB) signaling, which in turn drives the expression of genes involved in the regulation of apoptosis as well as immune and inflammatory responses
- Xevinapant (previously designated Debio 1143) is a first-in-class, potent, small-molecule antagonist of IAPs that restores the sensitivity of cancer cells to apoptosis (**Figure 1**)
- Xevinapant has shown antitumor activity in multiple models of human cancers, including SCCHN, in combination with chemotherapy or radiation therapy
- Xevinapant has also been shown to modulate NF-κB signaling, which may enhance antitumor response in immune cells in the tumor microenvironment[1]

Figure 1. Xevinapant mode of action

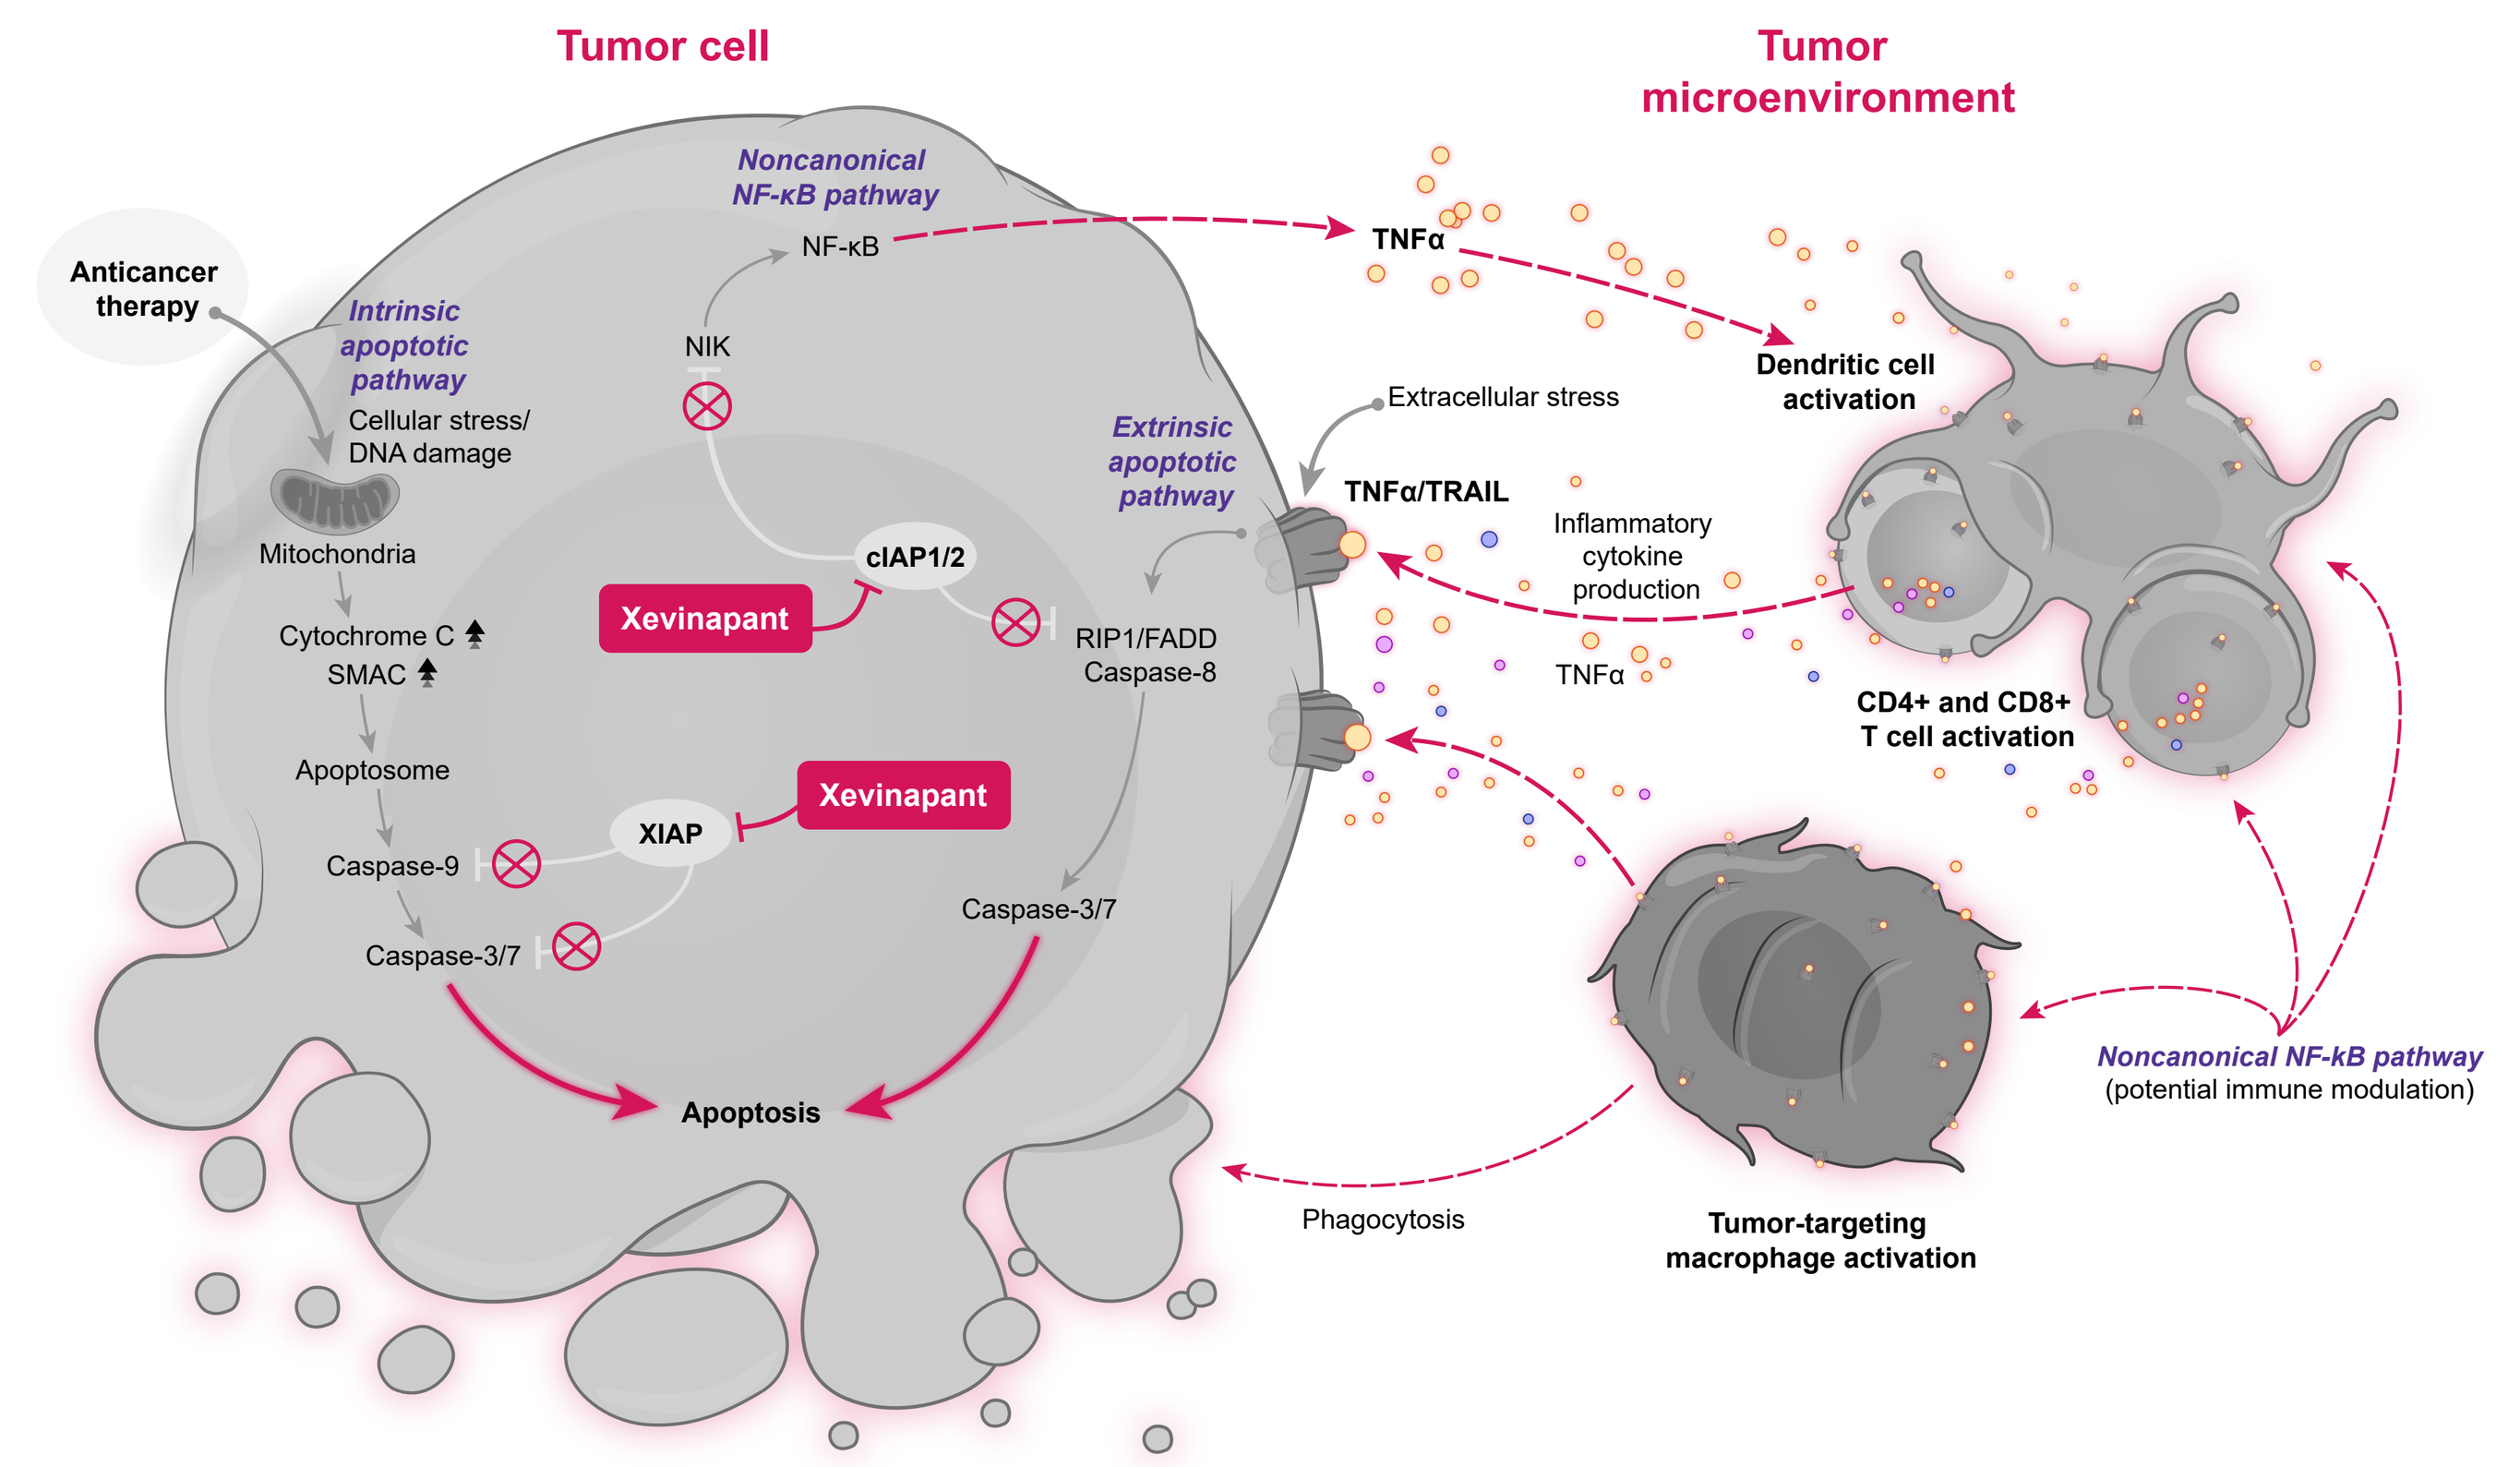
Tumor cell
Tumor microenvironment
Anticancer therapy
Intrinsic apoptotic pathway
Cellular stress/ DNA damage
Mitochondria
Cytochrome C
SMAC
Apoptosome
Caspase-9
Caspase-3/7
Noncanonical NF-κB pathway
NF-κB
NIK
cIAP1/2
Xevinapant
XIAP
Xevinapant
Extrinsic apoptotic pathway
RIP1/FADD Caspase-8
Caspase-3/7
Apoptosis
TNFα
Extracellular stress
TNFα/TRAIL
Inflammatory cytokine production
TNFα
Dendritic cell activation
CD4+ and CD8+ T cell activation
Noncanonical NF-κB pathway (potential immune modulation)
Tumor-targeting macrophage activation
Phagocytosis

Xevinapant is thought to (A) restore apoptosis in cancer cells by blocking XIAP and cIAP1/2, leading to activation of caspases downstream of intrinsic mitochondrial and extrinsic TNF receptor pathways, respectively, and (B) enhance the inflammatory antitumor response in immune cells in the tumor microenvironment by activating noncanonical NF-κB signaling through blocking of cIAP1/2 effects downstream of the TNF receptor.

cIAP1/2, cellular IAPs 1 and 2; **FADD**, FAS-associated death domain protein; **IAP**, inhibitors of apoptosis proteins; **NF-κB**, nuclear factor kappa B; **NIK**, NF-κB–inducing kinase; **RIP1**, receptor-interacting serine/threonine-protein kinase 1; **SMAC**, second mitochondria-derived activator of caspase; **TNF**, tumor necrosis factor; **TRAIL**, TNF-related apoptosis-inducing ligand; **XIAP**, X-linked IAP.
Figure source: Bourhis J, et al. Future Oncol. 2022. Published online February 17, 2022.

- Results from a double-blind, multicenter, randomized, phase 2 trial (NCT02022098) in patients with high-risk, locally advanced SCCHN reported significant improvements in locoregional control (LRC) at 18 months.[2] After 3 years of follow-up, xevinapant + CRT significantly improved overall survival (OS; risk of death reduced by 51%) and progression-free survival (PFS; risk of disease progression or death reduced by 67%) vs placebo + CRT[3]
- Xevinapant + CRT treatment did not result in new safety signals and did not impact treatment compliance[2,3]
- Here, we analyzed pharmacodynamic (PD) biomarker time-concentration profiles and explored the association between PD changes over time and clinical efficacy and safety parameters

METHODS

Study design

- In this phase 2, double-blind study, 96 patients were randomized 1:1 to receive xevinapant 200 mg or placebo once daily in combination with standard-of-care CRT. Details of the study design have been reported previously[2]

Exploratory PD assessments

- We measured 3 serum biomarkers via ELISA at baseline and on days 1, 2, 5, 8, and 16 of cycle 1:
 - Caspase-cleaved fragment of cytokeratin 18 (CKM30), a biomarker of epithelial apoptosis
 - Monocyte chemoattractant protein-1 (MCP-1) and tumor necrosis factor α (TNF-α), downstream targets of the NF-κB pathway
- We tested time course changes in the levels of CKM30, MCP-1, and TNF-α measured by their areas under the curve (AUCs) to assess if these biomarkers were associated with safety and efficacy endpoints. We used linear mixed-effects models for continuous endpoints, logistic models for categorical endpoints, and Cox proportional hazards models for time-to-event endpoints
- Safety parameters included absolute neutrophil counts, alanine or aspartate aminotransferase increase, blood bilirubin increase, lipase or amylase increase, hemoglobin, anemia, weight loss, nausea, stomatitis, and mucosal inflammation
- Clinical response parameters included LRC at 18 months from end of treatment (EOT), PFS, duration of LRC, time to distant relapse, OS, and complete response (CR) 6 months after EOT

RESULTS

PD analysis set

- The exploratory biomarker analysis set comprised all patients with baseline PD assessments and ≥1 postbaseline measurement: n=35 for xevinapant + CRT; n=40 for placebo + CRT
- Consistent with the intent-to-treat population, improvements in LRC at 18 months, PFS, and OS were observed with xevinapant + CRT vs placebo + CRT in the PD analysis set (**Figure 2**)
- The safety profile in the PD population was consistent with that in the intent-to-treat population

Figure 2. Kaplan-Meier curves for duration of LRC (A), PFS (B), and OS (C) at 36 months in the PD analysis set

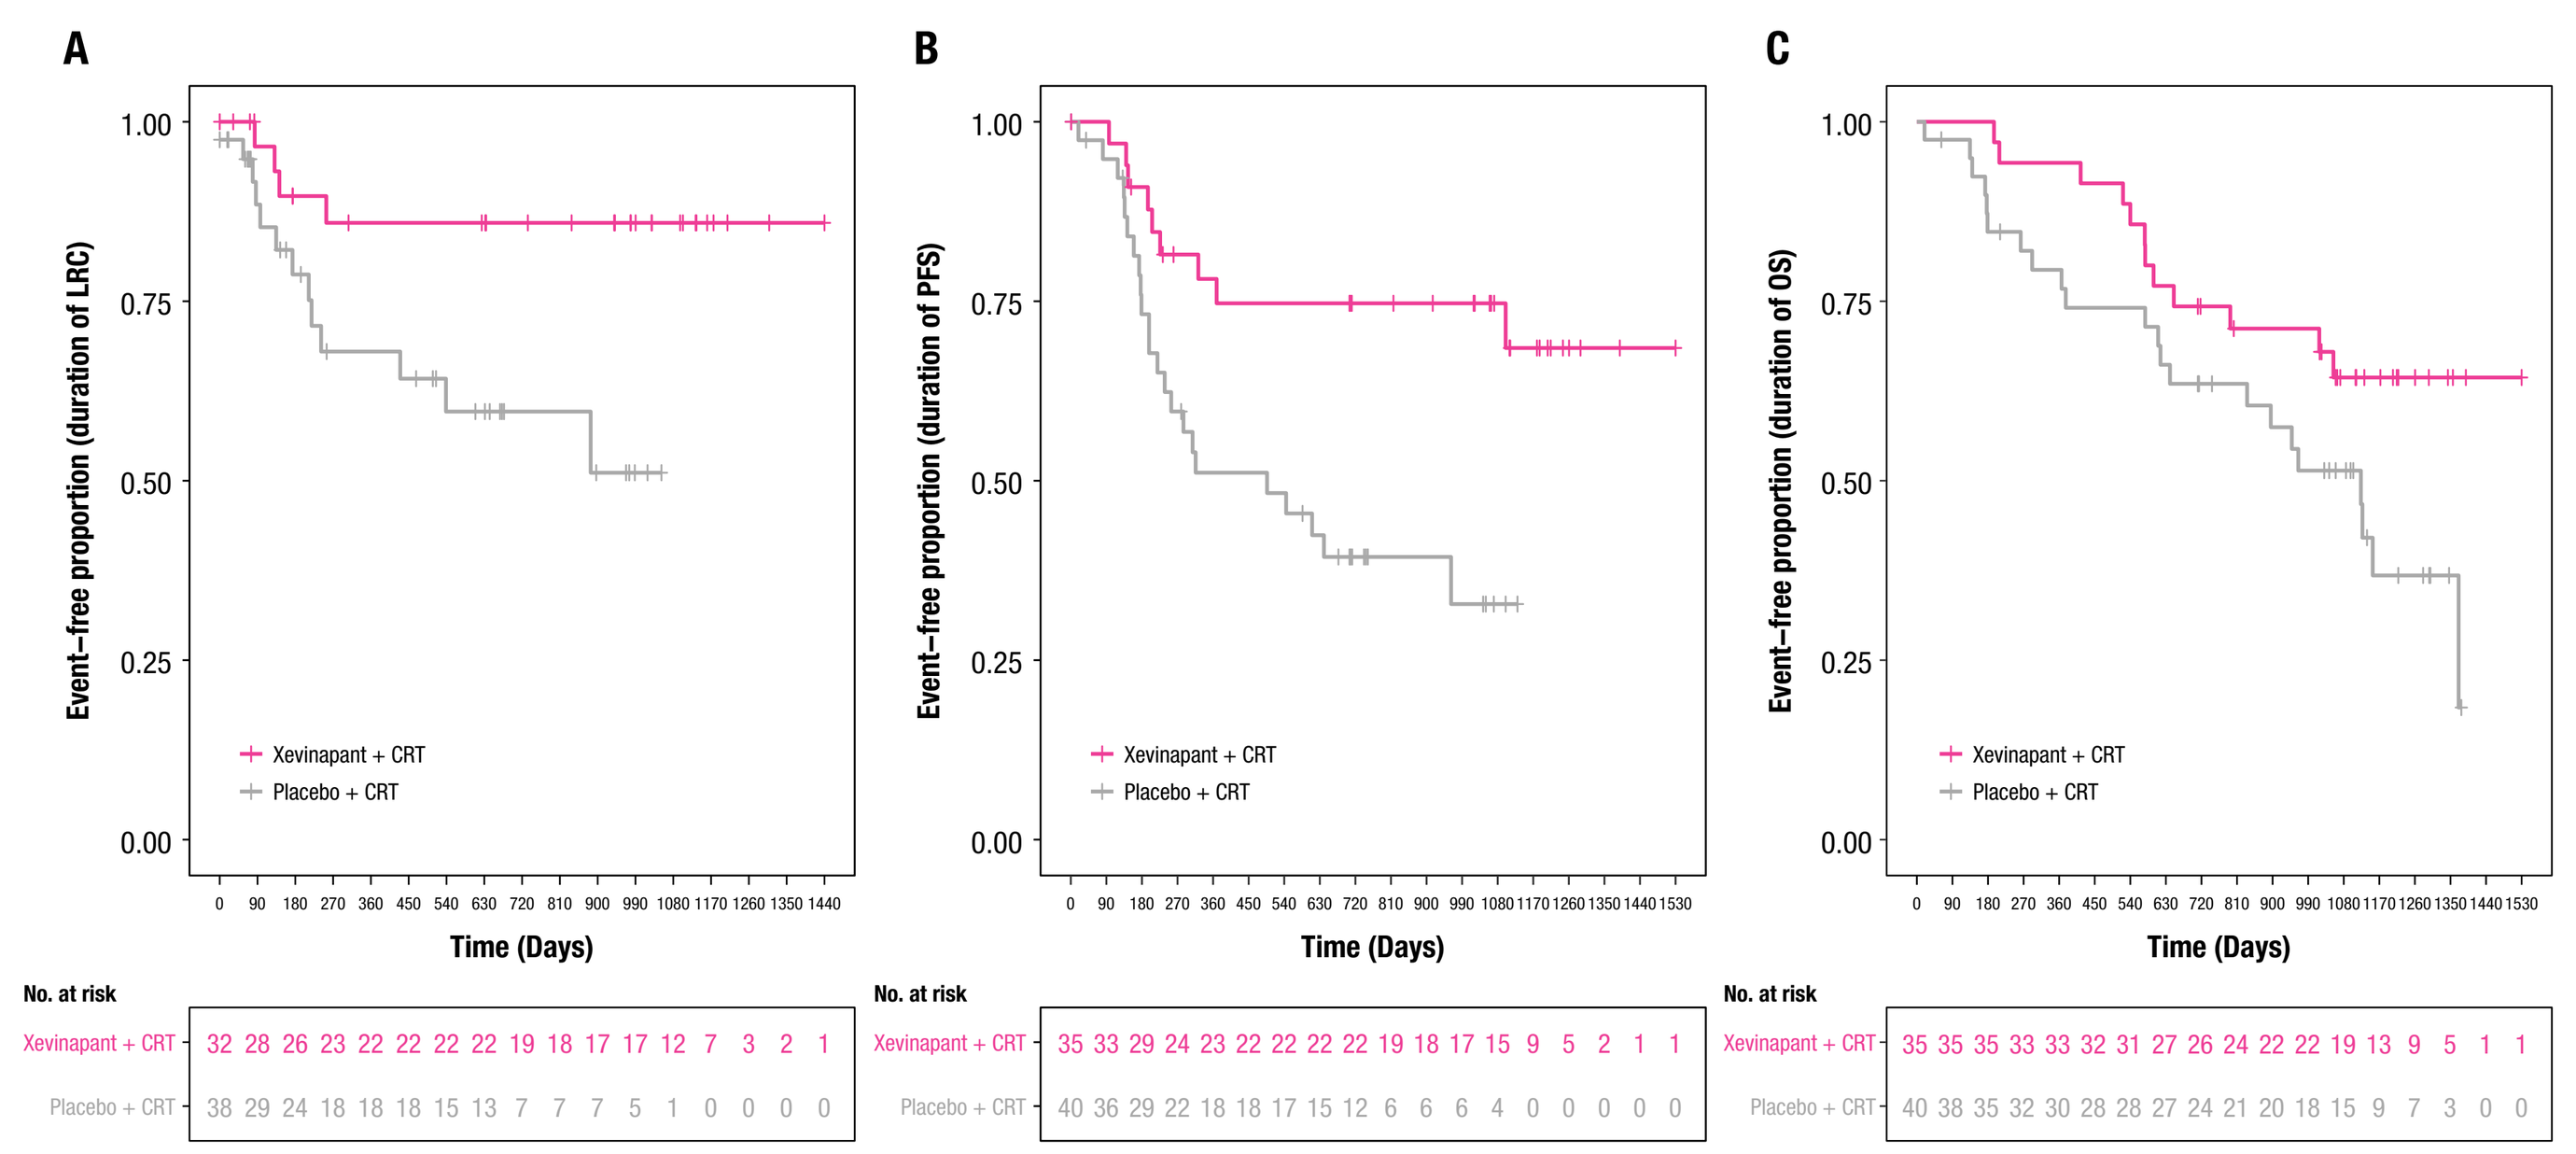
A
B
C
Event-free proportion (duration of LRC)
Event-free proportion (duration of PFS)
Event-free proportion (duration of OS)
Xevinapant + CRT
Placebo + CRT
Time (Days)
No. at risk
Xevinapant + CRT 32 28 26 23 22 22 22 22 19 18 17 17 12 7 3 2 1
Placebo + CRT 38 29 24 18 18 18 15 13 7 7 7 5 1 0 0 0 0
Xevinapant + CRT 35 33 29 24 23 22 22 22 22 19 18 17 15 9 5 2 1 1
Placebo + CRT 40 36 29 22 18 18 17 15 12 6 6 6 4 0 0 0 0 0
Xevinapant + CRT 35 35 35 33 33 32 31 27 26 24 22 22 19 13 9 5 1 1
Placebo + CRT 40 38 35 32 30 28 28 27 24 21 20 18 15 9 7 3 0 0

CRT, chemoradiotherapy; **LRC**, locoregional control; **OS**, overall survival; **PFS**, progression-free surival.

RESULTS (CONT)

- Serum CKM30 levels increased 24 hours after treatment initiation in both arms. After day 2, levels of CKM30 decreased mostly to baseline, with significant differences observed between treatment arms (**Figure 3A**)
- Serum MCP-1 levels increased 6 hours after treatment initiation in both arms. After day 1, MCP-1 levels remained above baseline levels in the xevinapant + CRT arm but returned closer to baseline levels in the placebo + CRT arm (**Figure 3B**)
- Change in serum TNF-α levels over time was similar to that in MCP-1 levels, but the difference was not statistically significant between the 2 treatment arms (**Figure 3C**)

Figure 3. Fold changes in CKM30 (A), MCP-1 (B), and TNF-α (C) over time

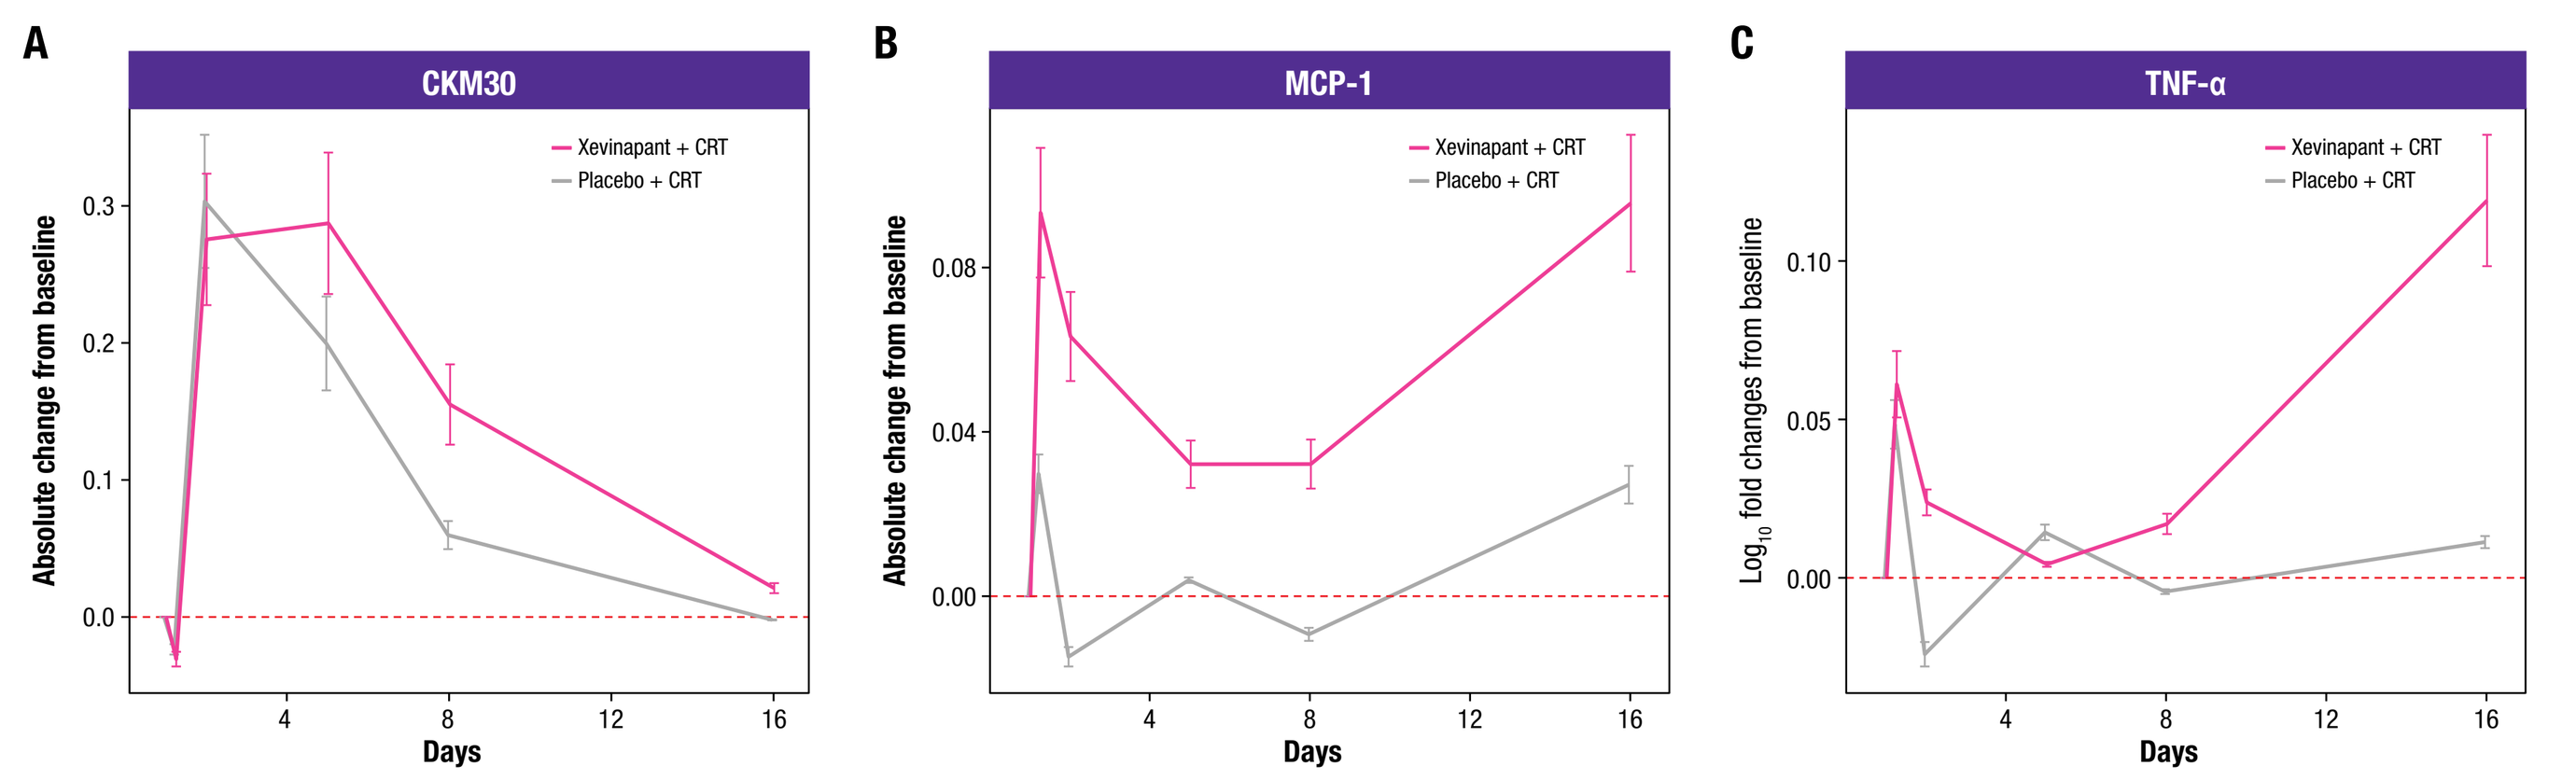
A
CKM30
B
MCP-1
C
TNF-α
Xevinapant + CRT
Placebo + CRT
Absolute change from baseline
Log10 fold changes from baseline
Days

Error bars represent standard errors.
CKM30, caspase-cleaved fragment of cytokeratin 18; **CRT**, chemoradiotherapy; **MCP-1**, monocyte chemoattractant protein-1; **TNF-α**, tumor necrosis factor α.

Association between PD biomarkers and efficacy and safety

- CKM30 AUC was associated with LRC at 18 months after EOT (missing as failure; p=0.0103) and with CR at 6 months after EOT (missing as failure; p=0.0511) (**Table 1**)
- No significant associations were observed between efficacy endpoints and MCP-1 and TNF-α

Table 1. Association between efficacy endpoints and PD biomarker AUCs

Marker	Endpoint	p value	False discovery rate
CKM30	**LRC at 18 months after EOT (missing as failure)**	**0.0103**	**4%**
	CR at 6 months after EOT (missing as failure)	**0.0511**	**20%**
	CR at 6 months after EOT (missing excluded)	0.0600	24%
	OS	0.0729	29%
	LRC at 18 months after EOT (missing excluded)	0.0764	31%
	Time to distant relapse	0.4063	73%
	PFS	0.4935	94%
	Duration of LRC	0.6122	82%
MCP-1	LRC at 18 months after EOT (missing as failure)	0.0740	15%
	Duration of LRC	0.1051	21%
	PFS	0.1362	54%
	CR at 6 months after EOT (missing excluded)	0.1872	37%
	LRC at 18 months after EOT (missing excluded)	0.1898	38%
	OS	0.2457	49%
	Time to distant relapse	0.2719	73%
	CR at 6 months after EOT (missing as failure)	0.3277	66%
TNF-α	Duration of LRC	0.0832	21%
	CR at 6 months after EOT (missing excluded)	0.2794	37%
	OS	0.4946	66%
	CR at 6 months after EOT (missing as failure)	0.5188	69%
	Time to distant relapse	0.5453	73%
	LRC at 18 months after EOT (missing excluded)	0.6026	60%
	LRC at 18 months after EOT (missing as failure)	0.9213	92%
	PFS	0.9803	98%

CKM30, caspase-cleaved fragment of cytokeratin 18; **CR**, complete response; **EOT**, end of treatment; **LRC**, locoregional control; **MCP-1**, monocyte chemoattractant protein-1; **OS**, overall survival; **PFS**, progression-free survival.; **TNF-α**, tumor necrosis factor α.

RESULTS (CONT)

- For the 2 endpoints that had significant associations with CKM30 AUC (LRC at 18 months after EOT and CR at 6 months after EOT), patients in the placebo + CRT arm who responded had lower CKM30 AUC values than those who did not respond (**Figure 4**)
- In the xevinapant + CRT arm, average CKM30 AUC levels were similar between responders and nonresponders (**Figure 4**)
- PD biomarkers were not associated with any of the clinical safety parameters

Figure 4. Association between CKM30 and LRC at 18 months after EOT (A) and CR at 6 months after EOT (B) by treatment response

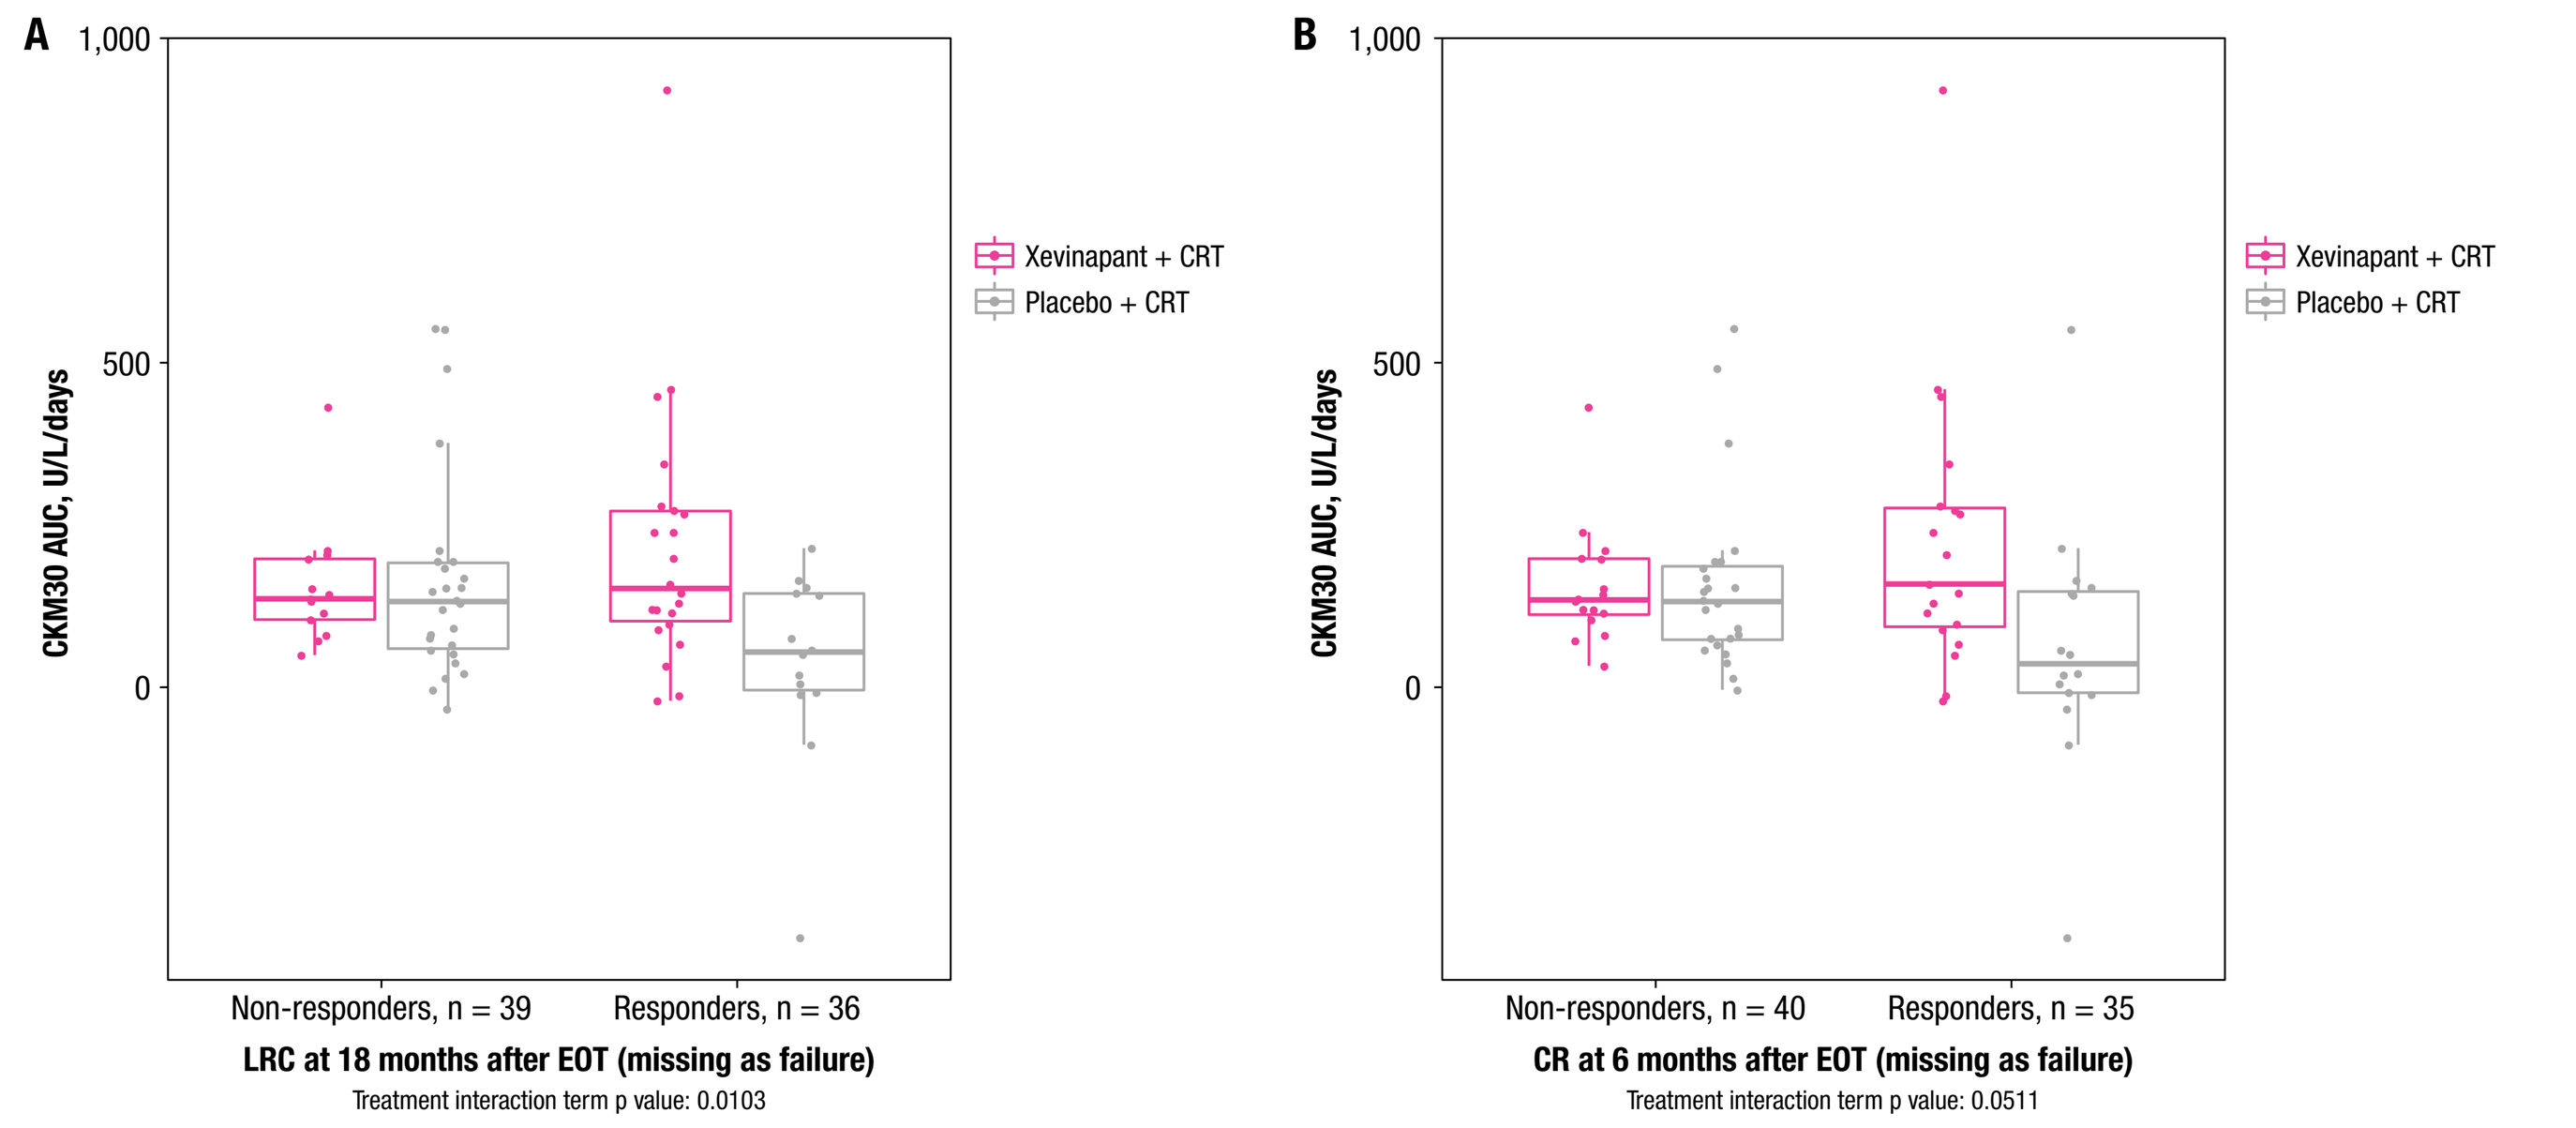
A
B
CKM30 AUC, U/L/days
Xevinapant + CRT
Placebo + CRT
Non-responders, n = 39
Responders, n = 36
LRC at 18 months after EOT (missing as failure)
Treatment interaction term p value: 0.0103
Non-responders, n = 40
Responders, n = 35
CR at 6 months after EOT (missing as failure)
Treatment interaction term p value: 0.0511

CONCLUSIONS

- Consistent with previous reports,[1] xevinapant appears to modulate NF-κB signaling; we observed significant differences in a marker of apoptosis (CKM30) and a downstream target of the NF-κB pathway (MCP-1) between patients who received xevinapant + CRT and those who received placebo + CRT
- While CKM30 serum levels returned mostly to baseline levels in both arms at day 2, the elevated levels of MCP-1 in the xevinapant + CRT arm at day 1 suggest that xevinapant may augment downstream CRT-induced apoptosis
- CKM30 AUC changes correlated with 2 response endpoints and may be a potential surrogate for response to xevinapant + CRT
- No associations were observed between any of the biomarkers and the safety endpoints explored
- Xevinapant in combination with CRT is currently being evaluated in the ongoing phase 3, randomized, international TrilynX study in patients with unresected, locally advanced SCCHN (NCT04459715)[4]. A publication summarising the study design can be accessed via the QR code below.

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from AACR and the author of this poster

Correspondence: Luke Piggott, **Luke.Piggott@debiopharm.com**

TRILYNX STUDY DESIGN

A publication describing the TrilynX study design can be accessed through this **hyperlink** or QR code.

References: 1. Gomez-Roca C, et al. Clin Transl Sci. 2022;15(1):55-62. 2. Sun X-S, et al. Lancet Oncol. 2020;21(9):1173-1187. **3.** Tao Y, et al. Manuscript in development. **4.** Bourhis J, et al. Future Oncol. 2022. Published online February 17, 2022. **Acknowledgments:** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. Medical writing support was provided by ClinicalThinking, and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) in accordance with Good Publication Practice guidelines (http://www.ismpp.org/gpp3).
Disclosures: L. Piggott, B. Gavillet, F. Brichory, K. Gollmer, F. Bouisset, and **G. Vuagniaux** are employees of Debiopharm International SA.

Abstract No. 603. Presented at the AACR Annual Meeting 2022, April 8-13, 2022; New Orleans, LA, and Virtual.